Sign-off sheet

Job number: 8286

Markup document title: 17-10-16 Dillon Time to BM Response NANETS poster draft 1b.pdf

Returning document title: 8286_LEX_NANETS BM response poster 03.pdf

Tasks carried out:

- [x] All comments/amends addressed
- [] Comments/amends partially addressed
- [] Additional tasks carried out
- [] Some or all of copy has been retyped, editorial check required

Signed: Jan

Date: 17.10.17

This form is part of our continuous improvement initiative.
If you have any feedback, suggestions, or think we have missed anything important please let us know.

Internal use only. Please remove this page from PDF before marking up further amends or sending on.

Entwine

Time to Sustained Improvements in Bowel Movement Frequency With Telotristat Ethyl: Analysis of the Phase 3 TELESTAR Study

Poster 255

Joseph S. Dillon[1], Matthew H. Kulke[2], Marianne Pavel[3,4], Dieter Hörsch[5], Lowell B. Anthony[6], Richard R. P. Warner[7], Emily Bergsland[8], Staffan Welin[9], Thomas M. O'Dorisio[1], Chad McKee[10], Pablo Lapuerta[10]

[1]University of Iowa, Iowa City, IA, USA; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]Charité–Universitätsmedizin, Berlin, Germany; [4]Current affiliation: Friedrich-Alexander-Universität Erlangen-Nürnberg, Erlangen, Germany; [5]Zentralklinik Bad Berka, Bad Berka, Germany; [6]University of Kentucky, Lexington, KY, USA; [7]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [8]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA, USA; [9]Uppsala University Hospital, Uppsala, Sweden; [10]Lexicon Pharmaceuticals, Inc., The Woodlands, TX, USA

Background

- Telotristat ethyl is a novel, small-molecule tryptophan hydroxylase inhibitor that decreases urinary 5-hydroxyindoleacetic acid (u5-HIAA) levels and bowel movement (BM) frequency in patients with carcinoid syndrome (CS). It is approved by the US Food and Drug Administration and European Commission for the treatment of CS diarrhea in combination with somatostatin analog (SSA) therapy in adults whose symptoms are inadequately controlled by SSA therapy.[1,2]
- SSAs and telotristat ethyl have differing mechanisms of action that, although complementary, may have different time courses of effect.[1,3]

OBJECTIVE

- To examine the time to sustained improvement (reduction) in BM frequency during the 12-week Double-blind Treatment (DBT) period in the Phase 3 TELESTAR study.

METHODS

Study design

- TELESTAR was a Phase 3, randomized, multicenter, parallel-group, double-blind, placebo-controlled study (ClinicalTrials.gov identifier: NCT01677910, **Figure 1**).[4]

FIGURE 1: TELESTAR Study Design

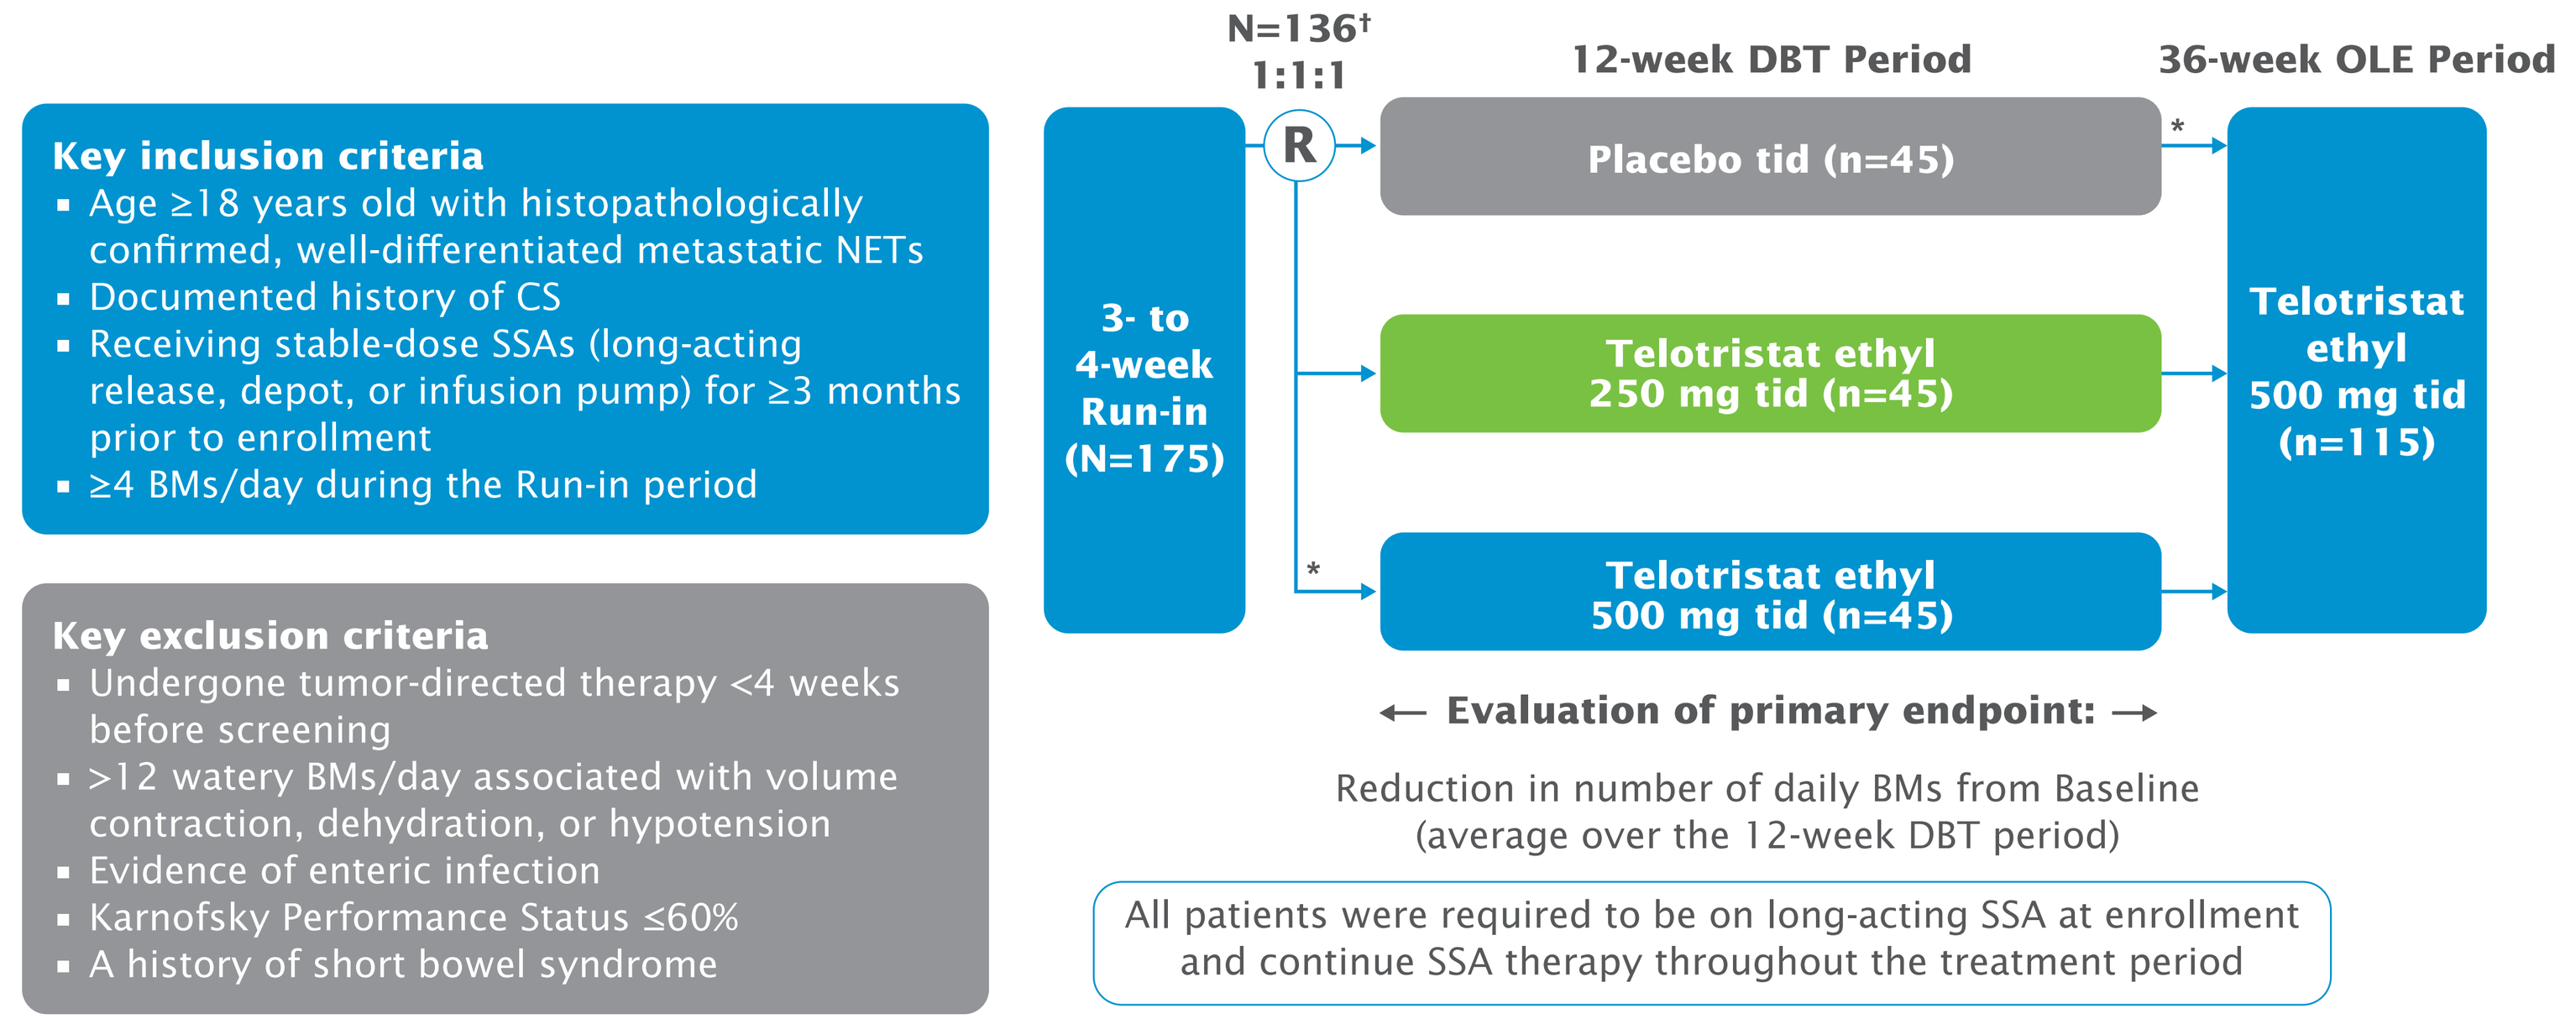
Key inclusion criteria
Age ≥18 years old with histopathologically confirmed, well-differentiated metastatic NETs
Documented history of CS
Receiving stable-dose SSAs (long-acting release, depot, or infusion pump) for ≥3 months prior to enrollment
≥4 BMs/day during the Run-in period
Key exclusion criteria
Undergone tumor-directed therapy <4 weeks before screening
>12 watery BMs/day associated with volume contraction, dehydration, or hypotension
Evidence of enteric infection
Karnofsky Performance Status ≤60%
A history of short bowel syndrome
N=136†
1:1:1
R
12-week DBT Period
36-week OLE Period
3- to 4-week Run-in (N=175)
Placebo tid (n=45)
Telotristat ethyl 250 mg tid (n=45)
Telotristat ethyl 500 mg tid (n=45)
Telotristat ethyl 500 mg tid (n=115)
*
*
← Evaluation of primary endpoint: →
Reduction in number of daily BMs from Baseline (average over the 12-week DBT period)
All patients were required to be on long-acting SSA at enrollment and continue SSA therapy throughout the treatment period

*Including a blinded titration step of 1 week of 250 mg tid
†136 were randomized; 135 were treated.
BMs, bowel movements; CS, carcinoid syndrome; DBT, Double-blind Treatment; NETs, neuroendocrine tumors; OLE, Open-label Extension; R, randomization, SSA, somatostatin analog; tid, 3 times per day

Assessments

- A sustained improvement (reduction) in BM frequency was defined as the time from the first dose to the first day of a continuous 14-day period in which a patient had ≥30% reduction from Baseline in BM frequency.
- The time to the first sustained BM response in the TELESTAR study was examined among treatment groups and analyzed using Cox regression to analyze hazard ratios and the log-rank test for treatment comparisons.

RESULTS

- A total of 135 patients received placebo, telotristat ethyl 250 mg 3 times per day (tid), or telotristat ethyl 500 mg tid during the DBT period.
- The mean Baseline BM frequency was 5.8 counts per day (range 5.2–6.1) (**Table 1**).

TABLE 1: Patient Characteristics

Characteristic	Statistic	Placebo (n=45)	Telotristat ethyl 250 mg tid (n=45)	Telotristat ethyl 500 mg tid (n=45)
u5-HIAA (mg/24 hours)	n Mean (SD)	44 81.0 (161.0)	42 92.6 (114.9)	44 89.5 (144.5)
BMs (counts/day)	n Mean (SD)	45 5.2 (1.4)	45 6.1 (2.1)	45 5.8 (2.0)
Stool form score[a]	n Mean (SD)	45 5.9 (0.7)	45 5.9 (0.5)	45 6.0 (0.7)

[a]Bristol Stool Form Scale[5]
BMs, bowel movements; SD, standard deviation; tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid

- Sustained improvement in BM frequency over the 12-week DBT period occurred in more than two-thirds of the patients receiving telotristat ethyl (**Table 2**).
- Median time to sustained improvements in BM frequency was about 3–4 weeks for patients on telotristat ethyl.
- Treatment with telotristat ethyl increased the likelihood of sustained improvement in BM frequency by more than 2-fold compared with placebo.

TABLE 2: Analysis of Time to First Occurrence of Sustained ≥30% Improvement in BM Frequency

	Placebo (n=45)	Telotristat ethyl 250 mg tid (n=45)	Telotristat ethyl 500 mg tid (n=45)
Patients with a sustained response, n	19	34	31
25th percentile, days (95% CL)	18.0 (4.0, 50.0)	5.0 (1.0, 12.0)	10.0 (1.0, 15.0)
Median percentile, days (95% CL)	NR	19.0 (9.0, 44.0)	27.0 (13.0, 44.0)
75th percentile, days (95% CL)	NR	73.0 (NC)	NR (NC)
Hazard ratio (telotristat ethyl/placebo) (95% CL)	–	2.29 (1.32, 4.10)	2.15 (1.22, 3.89)
P-value	–	0.004	0.009

BM, bowel movement; CL, confidence limits; NC, not calculated; NR, not reached; tid, 3 times per day

- The timing of the responses varied widely among patients; the earliest time of sustained improvements in BM frequency occurred within 5 days of treatment initiation, and the latest was at 73 days (**Figure 2**).
- The reasons for the variability of responses, including responses on placebo, are unclear.

FIGURE 2: Time to First Occurrence of Sustained ≥30% Improvement From Baseline in BM Frequency During the DBT Period

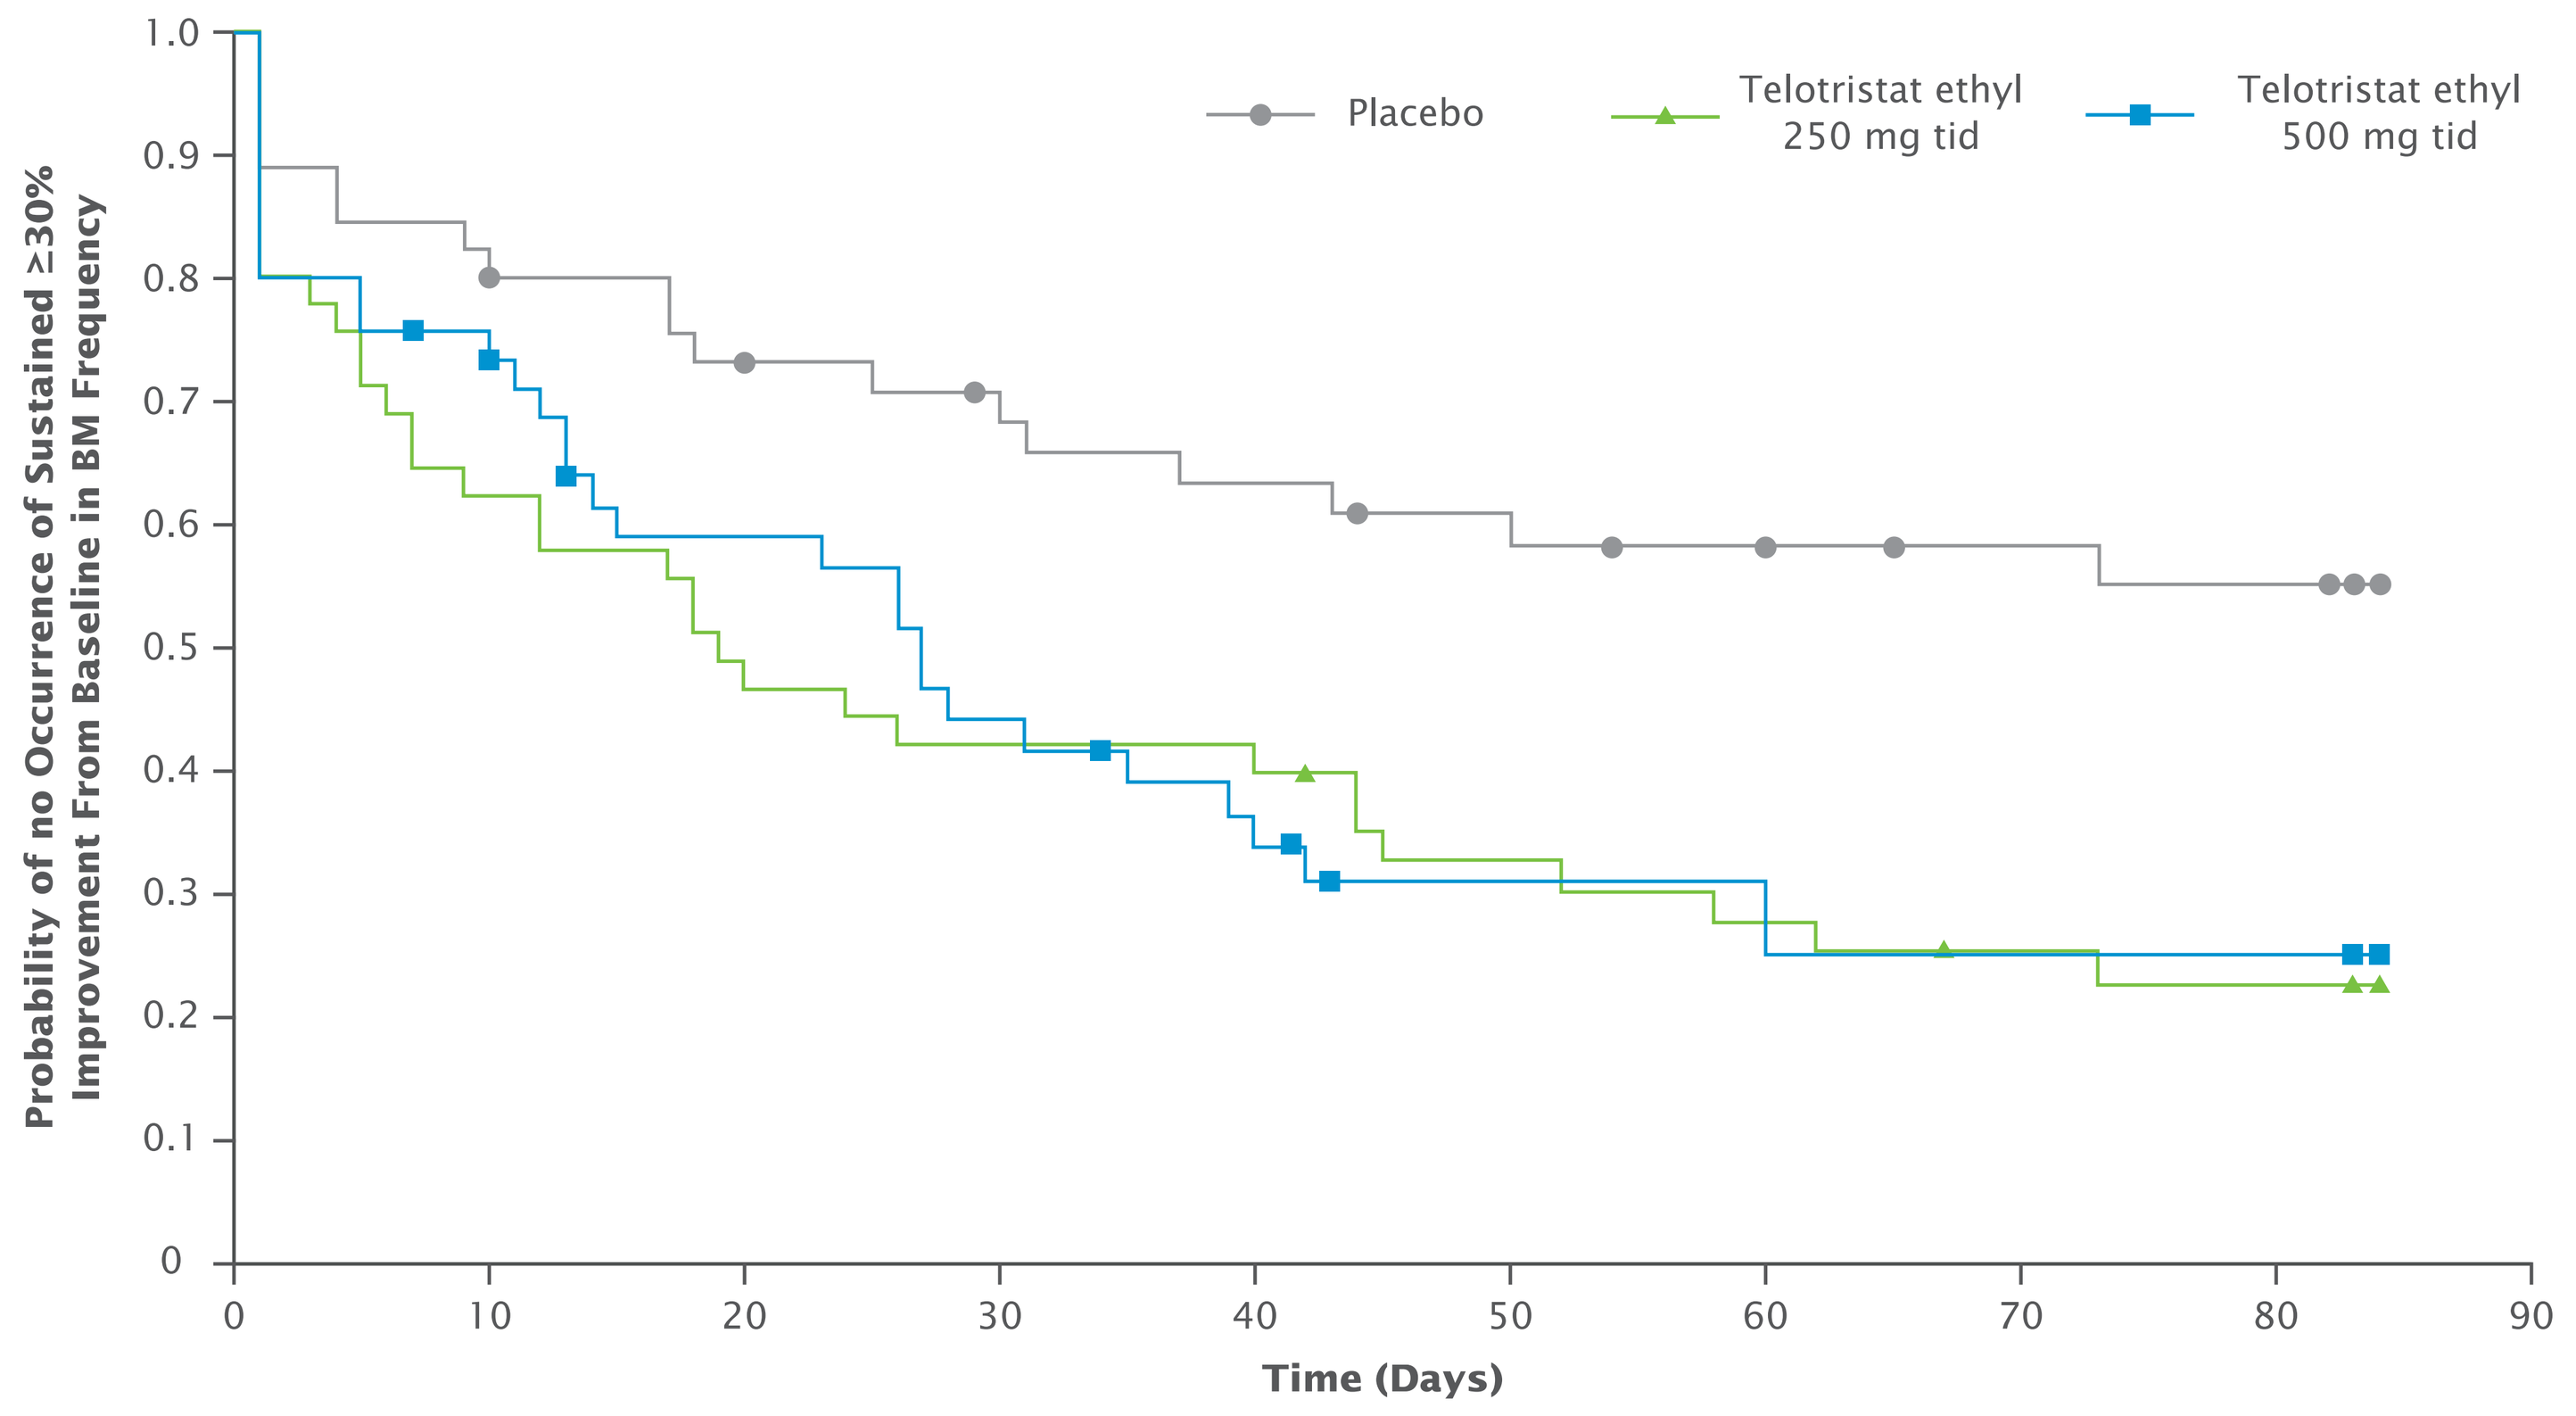
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
Probability of no Occurrence of Sustained ≥30% Improvement From Baseline in BM Frequency
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
0
10
20
30
40
50
60
70
80
90
Time (Days)

BM, bowel movement; DBT, Double-blind Treatment; tid, 3 times per day

Summary and Conclusions

- Understanding the mean and range of time of onset of sustained BM frequency improvement is important when considering the use of telotristat ethyl in patients with CS diarrhea.
- The time to sustained clinical benefit with telotristat ethyl may vary between patients.
- The time to sustained BM frequency reduction on telotristat ethyl occurred between 3 and 4 weeks after treatment initiation in approximately half of the patients treated, with a range of onset between 5 and 73 days; this was superior to the time to sustained reduction in BM frequency observed in the placebo group.

REFERENCES

1. National Comprehensive Cancer Network. Neuroendocrine Tumors (Version 3.2017). 2017. https://www.nccn.org/professionals/physician_gls/pdf/neuroendocrine.pdf. Accessed October 2, 2017.
2. European Commission. Commission Implementing Decision of 18.9.2017 granting marketing authorisation under Regulation (EC) No 726/2004 of the European Parliament and of the Council for "Xermelo - telotristat", an orphan medicinal product for human use. 2017. http://ec.europa.eu/health/documents/community-register/2017/20170918138652/dec_138652_en.pdf. Accessed October 2, 2017.
3. Grozinsky-Glasberg S, Shimon I, Korbonits M, Grossman AB. Somatostatin analogues in the control of neuroendocrine tumours: efficacy and mechanisms. *Endocr Relat Cancer.* 2008;15(3):701-720.
4. Kulke MH, Horsch D, Caplin ME, et al. Telotristat ethyl, a tryptophan hydroxylase inhibitor for the treatment of carcinoid syndrome. *J Clin Oncol.* 2017;35(1):14-23.
5. Longstreth GF, Thompson WG, Chey WD, Houghton LA, Mearin F, Spiller RC. Functional bowel disorders. *Gastroenterology.* 2006;130(5):1480-1491.

ACKNOWLEDGMENTS

We thank the patients and Investigators for participating in the study. We thank James Banigan, PhD (Chameleon Communications International, with funding from Lexicon Pharmaceuticals, Inc.), for editorial assistance in the preparation of this poster. The TELESTAR study was sponsored by Lexicon Pharmaceuticals, Inc.

DISCLOSURES

JSD: Lexicon Pharmaceuticals, Inc. MHK: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, and Novartis Pharmaceuticals. MP: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, and Novartis Pharmaceuticals. DH: Lexicon Pharmaceuticals, Inc., Ipsen Biopharmaceuticals, Inc., Novartis Pharmaceuticals, and Pfizer, Inc. LBA: Lexicon Pharmaceuticals, Inc. RRPW: Lexicon Pharmaceuticals, Inc. EB: UpToDate, Lexicon Pharmaceuticals, Inc., Ipsen. CM and PL: employee of Lexicon Pharmaceuticals, Inc. Authors who are employees of Lexicon Pharmaceuticals, Inc., may have common stock or may have been granted stock options or other equity incentive awards. SW and TOD: nothing to disclose.

To view this poster online, snap a photo of this QR code with your smart phone (QR reader required).

Presented at the 2017 NANETS Symposium; October 19–21, 2017; Philadelphia, PA, USA

Lexicon pharmaceuticals