249

Phase I, Open-label, Dose-escalation Study of SNX-5422 plus Everolimus in Neuroendocrine Tumors (NETs)

Martin E. Gutierrez,[1] Giuseppe Giaccone[2], Stephen V. Liu[2], Arun Rajan[3], Udayan Guha[3], Thorvardur R. Halfdanarson[4], Pamela L. Kunz[5], James M. Hinson, Jr.[6], Everardus O. Orlemans[7]

[1]The John Theurer Cancer Center, Hackensack, NJ; [2]Lombardi Comprehensive Cancer Center, Georgetown University, Washington DC; [3]Thoracic and GI Oncology Branch, National Cancer Institute, Bethesda, MD; [4]Mayo Clinic Cancer Center, Rochester, MN; [5]Stanford University School of Medicine, Palo Alto, CA; [6]Unicorn Pharma Consulting, Nashville, TN; [7]Esanex Inc., Indianapolis, IN

BACKGROUND

- SNX-5422 is an orally bioavailable pro-drug of SNX-2112, a highly potent and selective inhibitor of heat shock protein 90 (Hsp90).[1]
- SNX-5422 has specific activity, targeting tumors via multiple signaling networks with numerous potential biomarkers identified.[2]

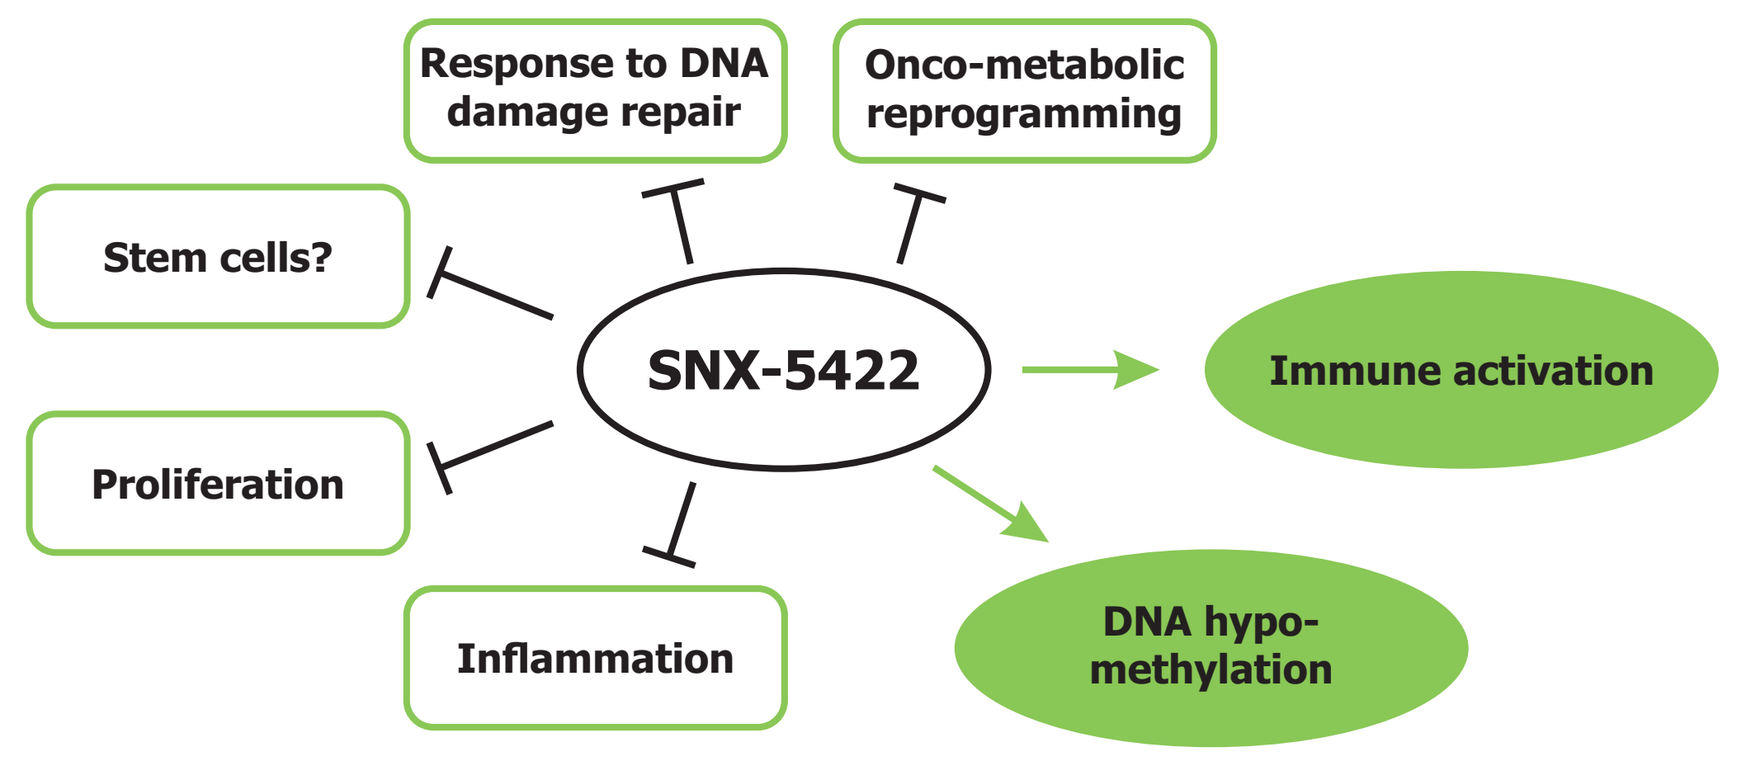
SNX-5422
Response to DNA damage repair
Onco-metabolic reprogramming
Stem cells?
Proliferation
Inflammation
Immune activation
DNA hypo-methylation

- Neuroendocrine tumors (NETs) are a heterogeneous group of rare neoplasms, accounting for 0.46% of all gastrointestinal (GI) and pulmonary malignancies. They are most frequently seen in the bronchopulmonary tree and small intestine, though other areas of the GI tract as well as the pancreas are also common sites.[3]
- Preclinical data have suggested that targeted inhibition of Hsp90 may have an antiproliferative effect on neuroendocrine tumor cells.[4-7]
- Previously, at doses of 42-100 mg/m^2 of SNX-5422 every other day, 2 of 3 subjects with refractory NETs achieved stable disease for ≥8 cycles.[8]
- Everolimus (EVR), an mTOR (mammalian target of rapamycin) kinase inhibitor, is used for the treatment of progressive pancreatic NETs as well as progressive, well-differentiated, nonfunctional GI and pulmonary NETs that are unresectable, locally advanced or metastatic.[9]
- In preclinical studies, the effects of SNX-2112 and EVR appear at least additive.[2]
- In this study, we investigated the dose limiting toxicity of SNX-5422 + EVR during the first cycle of treatment as well as the safety of the combination and its effect on tumor response (RECIST 1.1 criteria).

References

1. Chandarlapaty S, et al. *Oncogene*. 2010;29(3):325-334.
2. Data on file. Esanex, Inc.
3. Hallet J, et al. *Cancer.* 2015;121(4):589-597.
4. Andersson E, et al. *Mod Pathol.* 2016;29(6):616-629.
5. Zitzmann K, et al. *Int J Oncol.* 2013;43(6):1824-1832.
6. Mayer P, et al. *Endocr Relat Cancer*. 2012;19(3):217-232.
7. Gilbert JA, et al. *Endocr Relat Cancer.* 2010;17(3):623-636.
8. Infante JR, et al. *Eur J Cancer.* 2014;50(17):2897-2904.
9. Afinitor® (everolimus) prescribing information, 2016.

METHODS

Study Design

- Phase 1, open-label, multicenter, 3+3 dose-escalation study. Each dosing cycle was 28 days.
- SNX-5422 dosing started at 50 mg/m^2. The actual dose administered was based on body surface area, calculated using height at Screening and body weight measured at the start of each cycle. The calculated dose was then rounded to nearest mg on the dosing chart. SNX-5422 was available in 5, 20, 75, and 150 mg capsules.
- EVR was dosed at 10 mg once daily; dose reduction was allowed based on EVR toxicity.

Dosing Schedule and Times

- For each 28-day dosing cycle:
 - **SNX-5422:** Once daily by mouth every other day (QOD) in the morning for 21 days (total of 11 doses) followed by a 7-day drug-free period; no food 2 hours before and 1 hour after dosing.
 - **EVR:** Once daily by mouth every evening (at least 8 hours after SNX-5422 dosing) for 28 out of 28 days, and taken consistently either with or without food.

Key Inclusion Criteria

- Adults ≥18 years-of-age with pathologic evidence of locally advanced or metastatic NET of gastro-entero-pancreatic, pulmonary (other than small cell lung cancer), or thymic origin.
- No more than 4 prior lines of systemic anti-cancer therapy.
- Karnofsky performance score ≥70, life expectancy ≥3 months, adequate baseline laboratory values.
- All adverse events from previous anti-cancer therapy (with the exception of alopecia) recovered to ≤ Grade 1 toxicity.

Key Exclusion Criteria

- Contraindication to EVR, or prior failed treatment with mTOR inhibitors (current EVR use permitted for subjects still receiving treatment benefit).
- Prior treatment with any Hsp90 inhibitor.
- Clinically significant interstitial lung disease, or obstructive disease without sufficient reserve (Forced Expiratory Volume in one second <50% of predicted).
- Neuroendocrine tumor with uncontrolled hormone-related symptoms, particularly symptoms that may mimic SNX-5422 toxicity such as diarrhea (diarrhea ≥4 stools per day).
- Neuroendocrine cancer of the thyroid.
- Conventional chemotherapy or radiation within 4 weeks or palliative radiation within 2 weeks before study start.
- Treatment with other anti-cancer drugs (except EVR or somatostatin analogues) or any other investigational drug within 30 days prior to the first dose of SNX-5422 and throughout the study.
- GI diseases or conditions (including chronic diarrhea) that could affect drug absorption or alter safety assessments.

RESULTS

Table 1. Baseline Demographics

Parameter	N=20
Age, median (range), years	59 (36-70)
Age >65 years, n (%)	5 (25%)
Males, n (%)	10 (50%)
Race, n (%)	
White	13 (65%)
Black	2 (10%)
Asian	1 (5%)
Other	4 (20%)
Not Hispanic or Latino, n (%)	14 (70%)

Table 2. Baseline Disease Characteristics

Characteristic	N=20
Tumor location/type	n (%)
Lung	10 (50%)
Small Cell Lung Cancer (SCLC)[a]	3
Neuroendocrine carcinoma	3
Atypical carcinoid	1
Neuroendocrine tumor	2
Carcinoid	1
Gastrointestinal, n (%)	5 (25%)
Neuroendocrine carcinoma	1
Neuroendocrine tumor	2
Carcinoid	2
Pancreas, n (%)	3 (15%)
Neuroendocrine carcinoma	1
Neuroendocrine tumor	1
Unknown	1
Unknown, n (%)	2 (10%)
Carcinoid	2
Number of prior anti-cancer regimens, n (%)	
0	1 (5%)
1 or 2	11 (55%)
3	2 (10%)
≥5	6 (30%)
Duration of diagnosis, mean (SD), years	1.9 (1.91)

Note: Neuroendocrine carcinoma: Ki67 >20% (or high-grade).
[a]These 3 subjects were enrolled prior to SCLC being an exclusion criterion, and are not included in the efficacy evaluable population.

Table 3. SNX-5422 Dosing Cohorts

Cohort	SNX-5422 Dose (mg/m^2)	Number of Subjects Assigned to Cohort (N=20)
1	50	3
2	75	17

Dose Limiting Toxicity (DLT)

- One subject experienced a DLT of Grade 3 diarrhea after 5 doses of SNX-5422 75 mg/m^2 QOD and daily EVR 10 mg. This subject continued in the study with SNX-5422 dose de-escalation to 50 mg/m^2 QOD and EVR dose reduction to 5 mg daily. No further SNX-5422 dose escalation was attempted in the study due to the observed adverse GI events with combined therapy.

Figure 1. Best Response: All Subjects

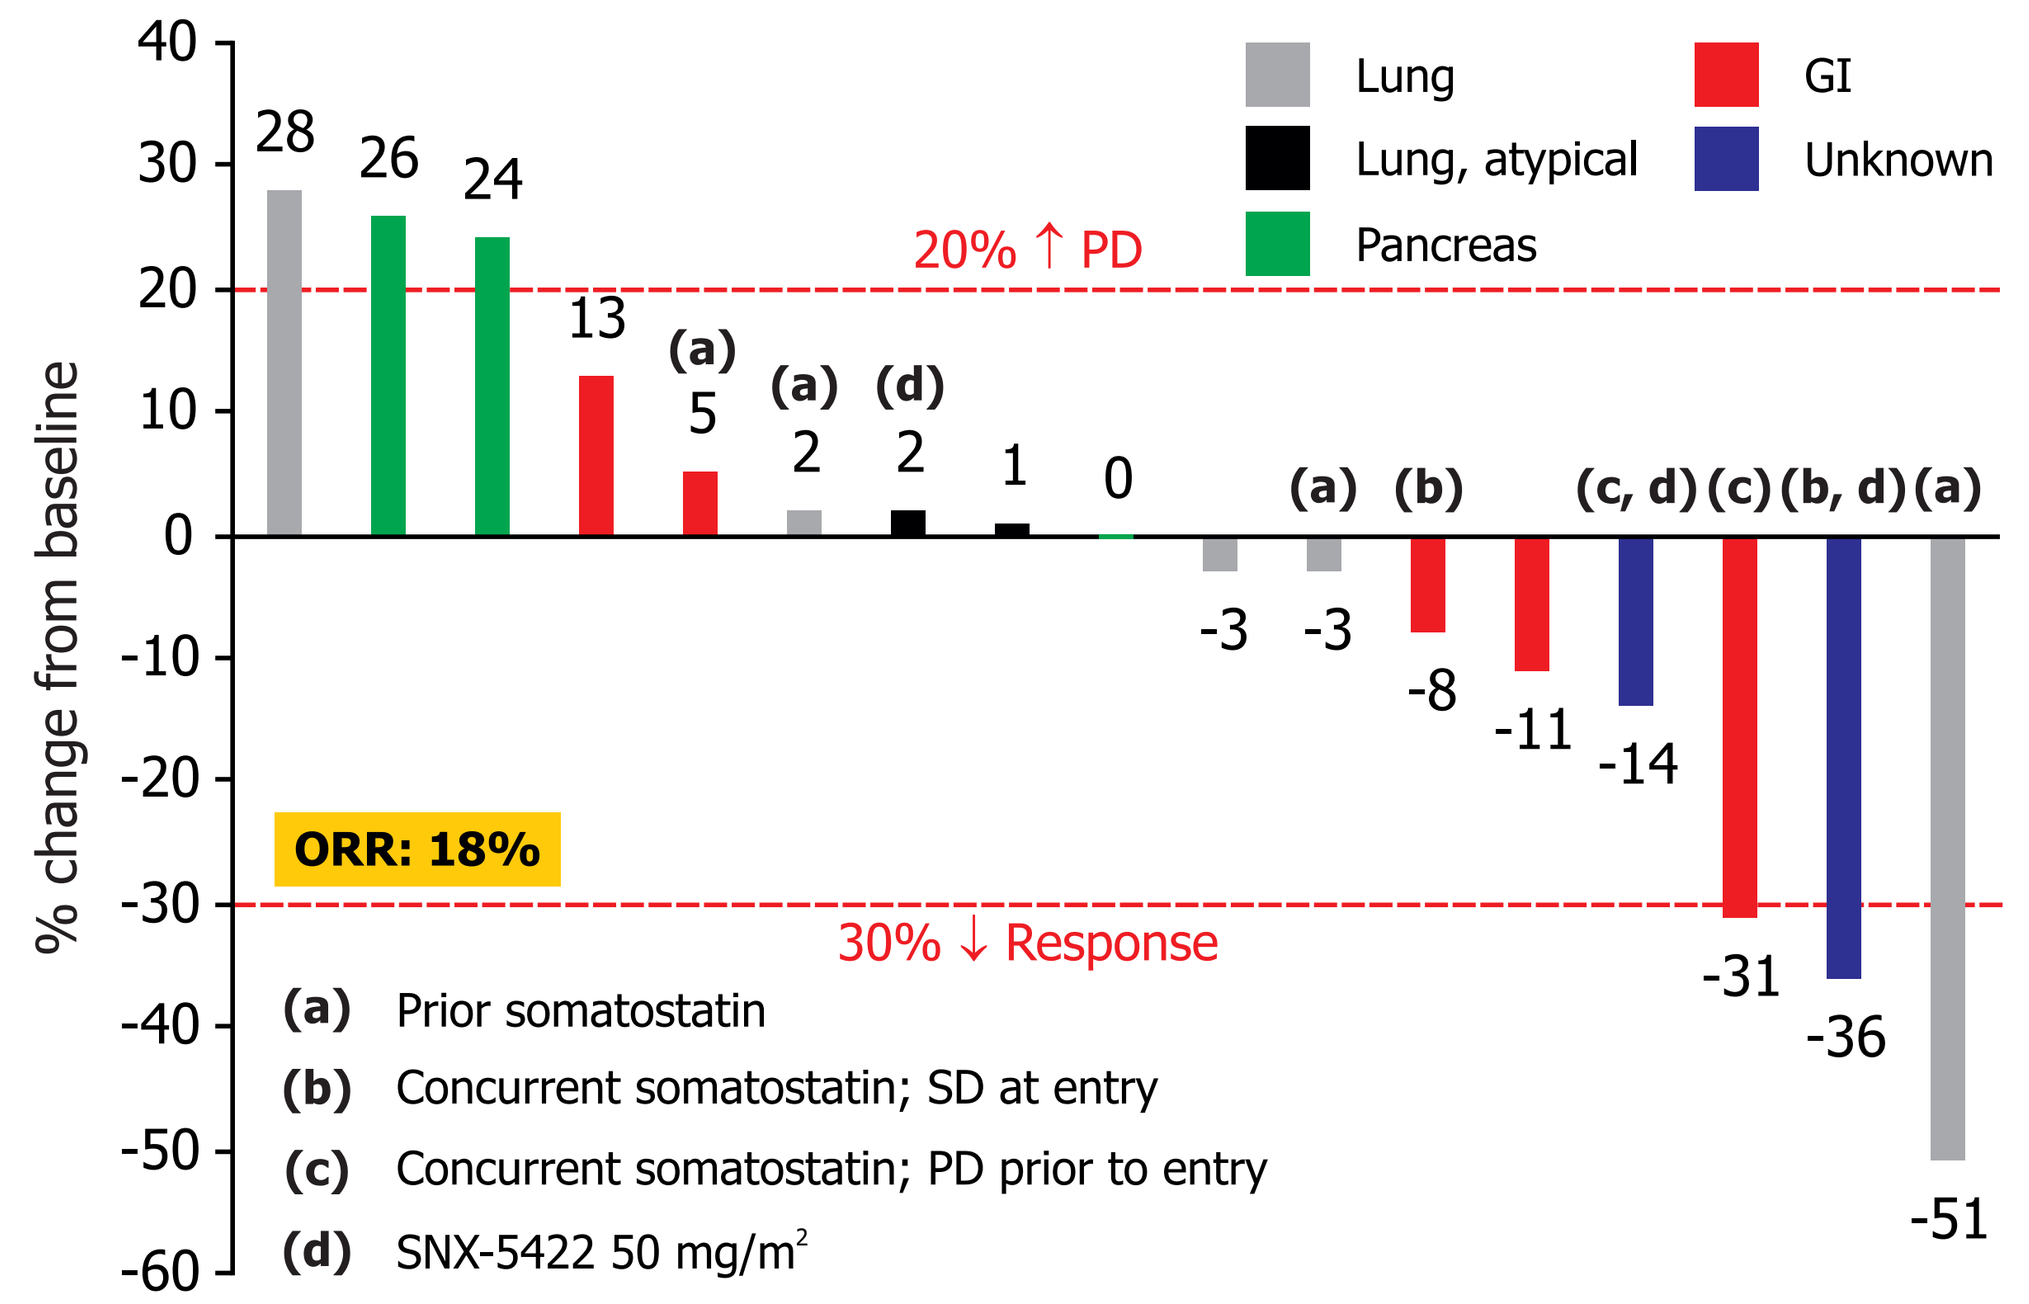
% change from baseline
40
30
20
10
0
-10
-20
-30
-40
-50
-60
28
26
24
13
(a) 5
(a) 2
(d) 2
1
0
(a)
(b)
(c, d)
(c)
(b, d)
(a)
-3
-3
-8
-11
-14
-31
-36
-51
20% ↑ PD
30% ↓ Response
ORR: 18%
Lung
Lung, atypical
Pancreas
GI
Unknown
(a) Prior somatostatin
(b) Concurrent somatostatin; SD at entry
(c) Concurrent somatostatin; PD prior to entry
(d) SNX-5422 50 mg/m2

- Tumor response in the 17 subjects evaluable for efficacy:
 - Partial response: 3 subjects
 - Stable disease: 11 subjects
 - Progressive disease: 3 subjects

Figure 2. Disease Characteristics and Treatment Duration

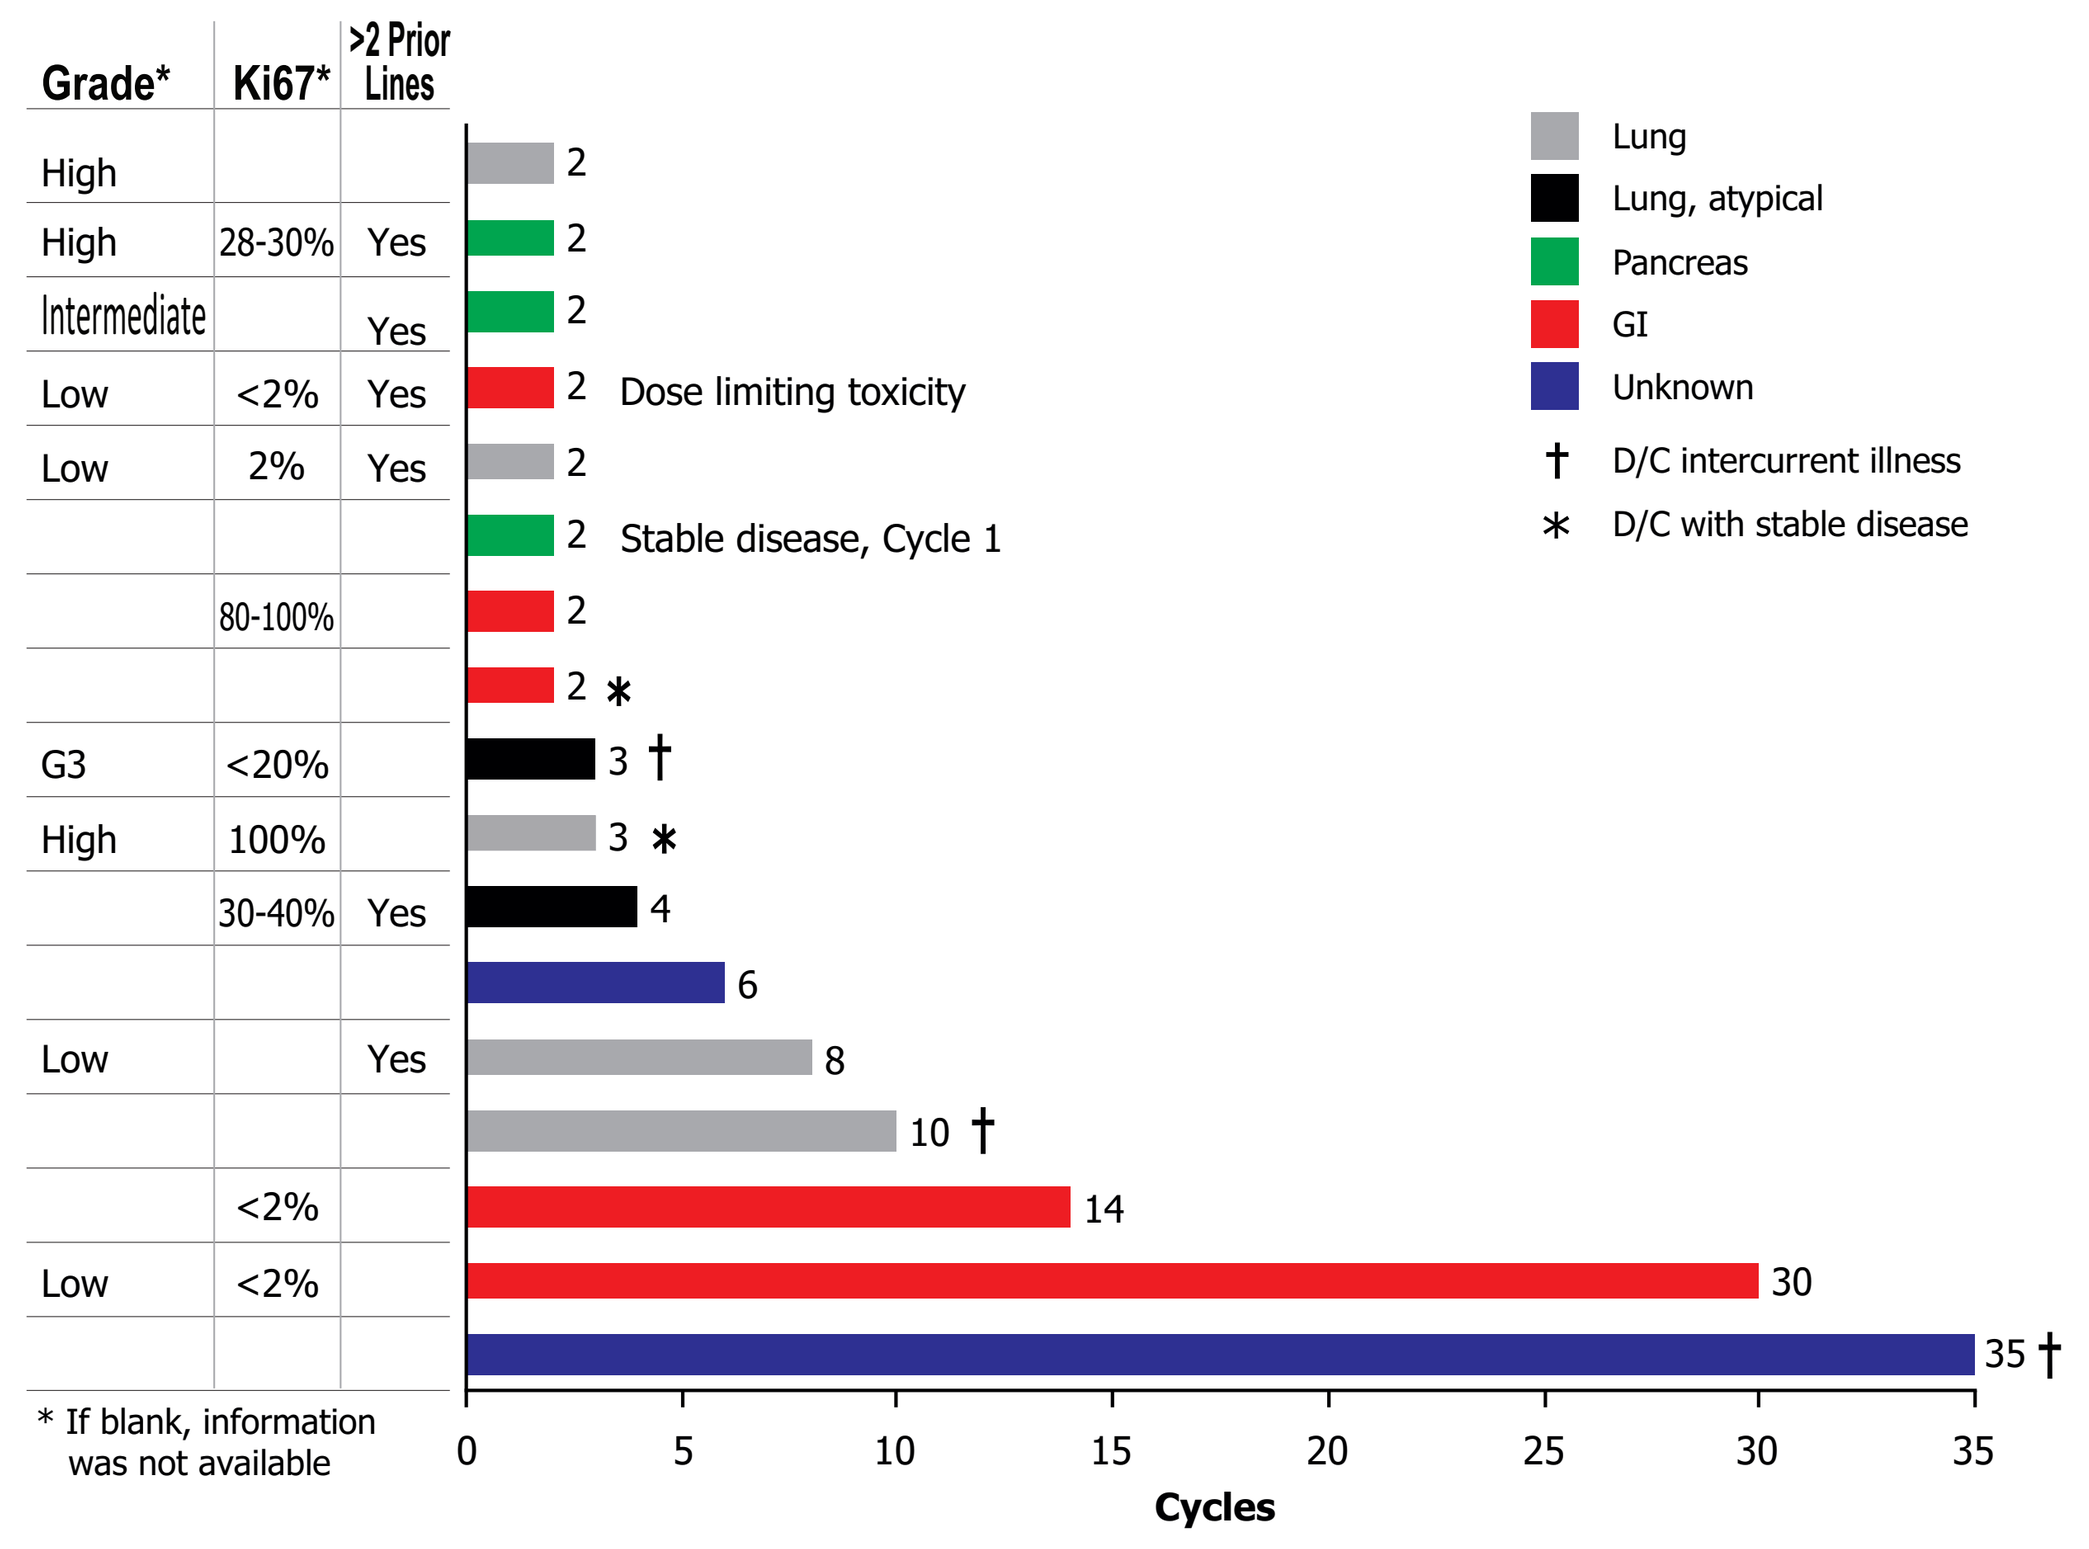
Grade*
Ki67*
>2 Prior Lines
High
High 28-30% Yes
Intermediate Yes
Low <2% Yes
Low 2% Yes
80-100%
G3 <20%
High 100%
30-40% Yes
Low Yes
<2%
Low <2%
2
2
2
2 Dose limiting toxicity
2
2 Stable disease, Cycle 1
2
2 ∗
3 †
3 ∗
4
6
8
10 †
14
30
35 †
* If blank, information was not available
0
5
10
15
20
25
30
35
Cycles
Lung
Lung, atypical
Pancreas
GI
Unknown
† D/C intercurrent illness
∗ D/C with stable disease

Figure 3. Best Response: Subjects with ≤2 Prior Lines of Anti-Cancer Therapy, Excluding Ki67 >20%

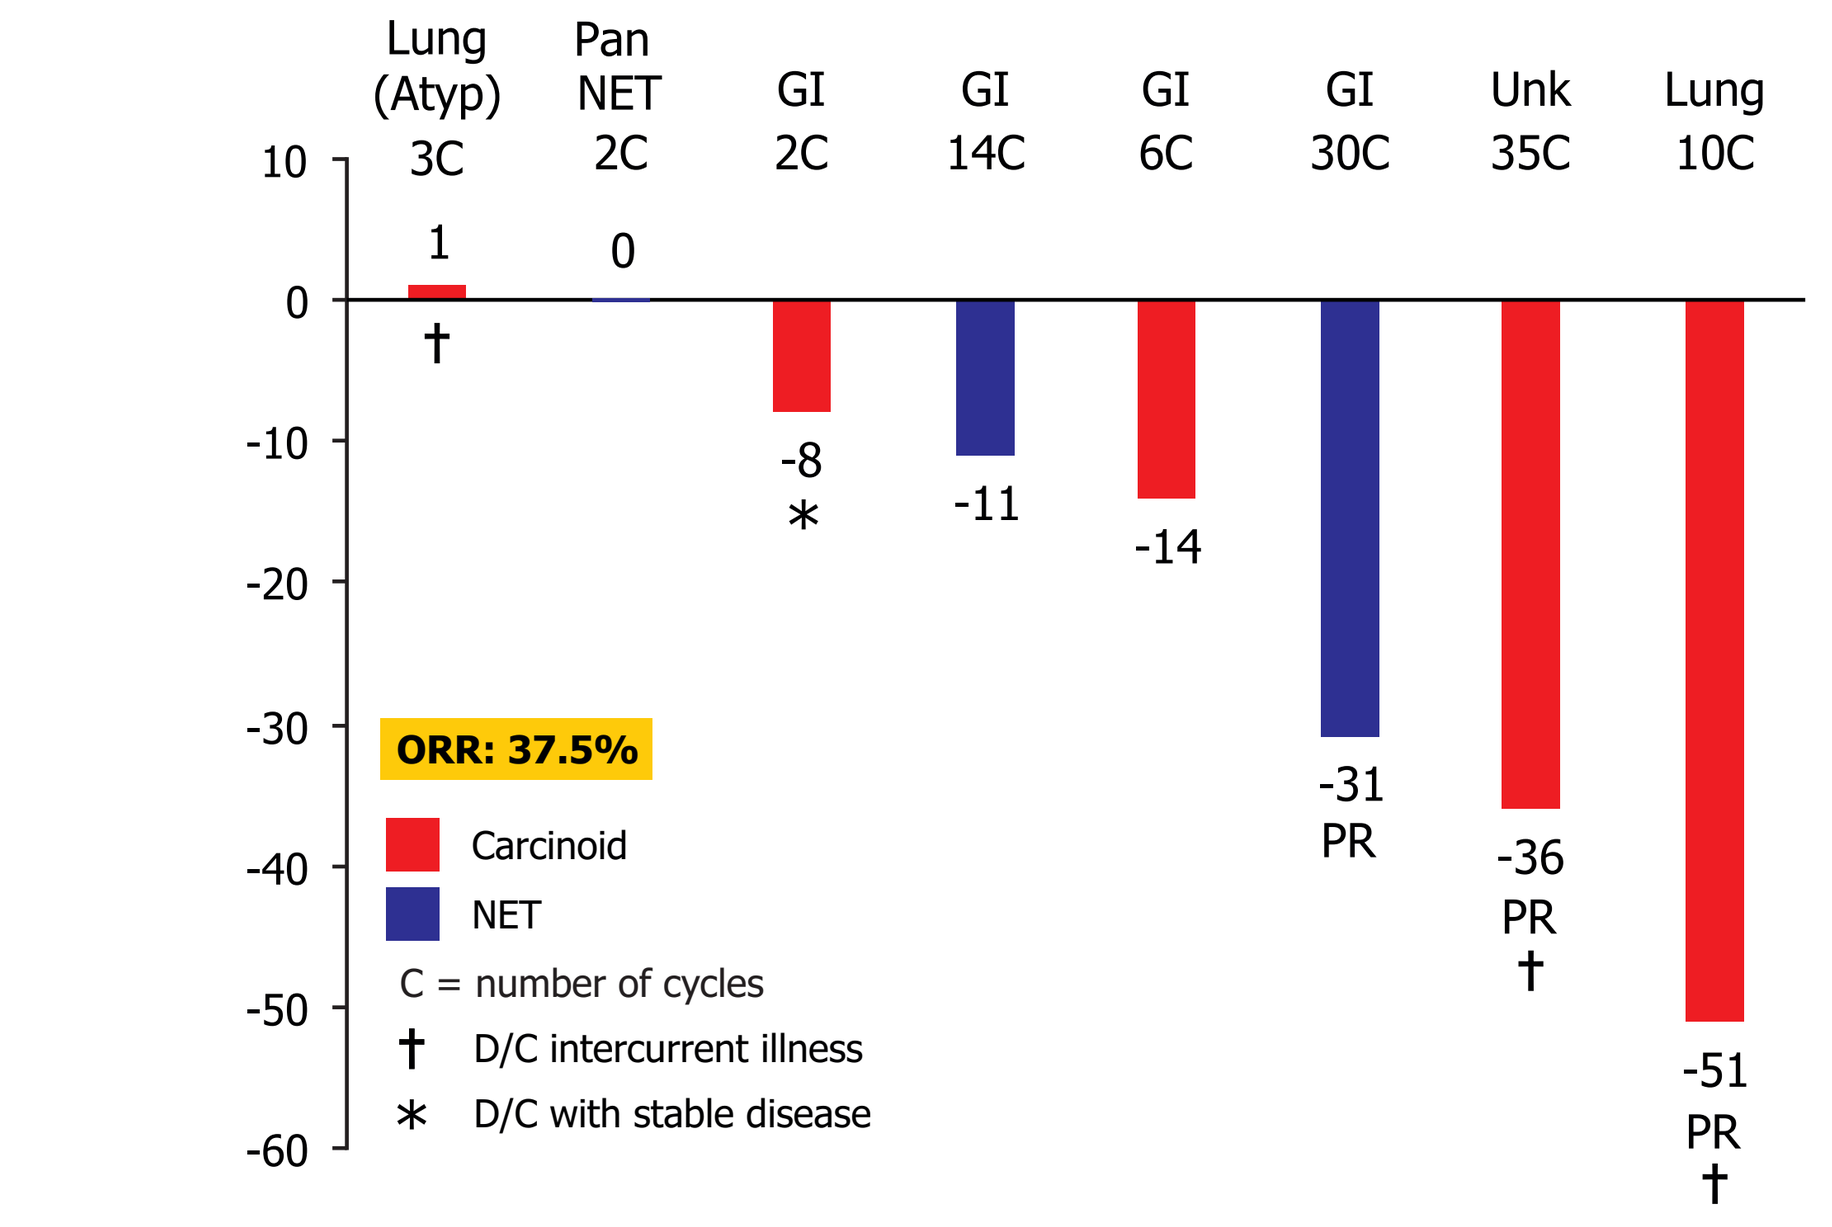
Lung (Atyp) 3C
Pan NET 2C
GI 2C
GI 14C
GI 6C
GI 30C
Unk 35C
Lung 10C
10
0
-10
-20
-30
-40
-50
-60
1 †
0
-8 ∗
-11
-14
-31 PR
-36 PR †
-51 PR †
ORR: 37.5%
Carcinoid
NET
C = number of cycles
† D/C intercurrent illness
∗ D/C with stable disease

- Best response to SNX-5422 and EVR combination was observed in subjects with:
 - ≤2 prior lines of anti-cancer therapy
 - Ki67 <20%

Figure 4. Best Response: Subjects with GI/Lung/Unknown Location Tumor and ≤2 Prior Lines of Anti-Cancer Therapy, Excluding Ki67 >20%

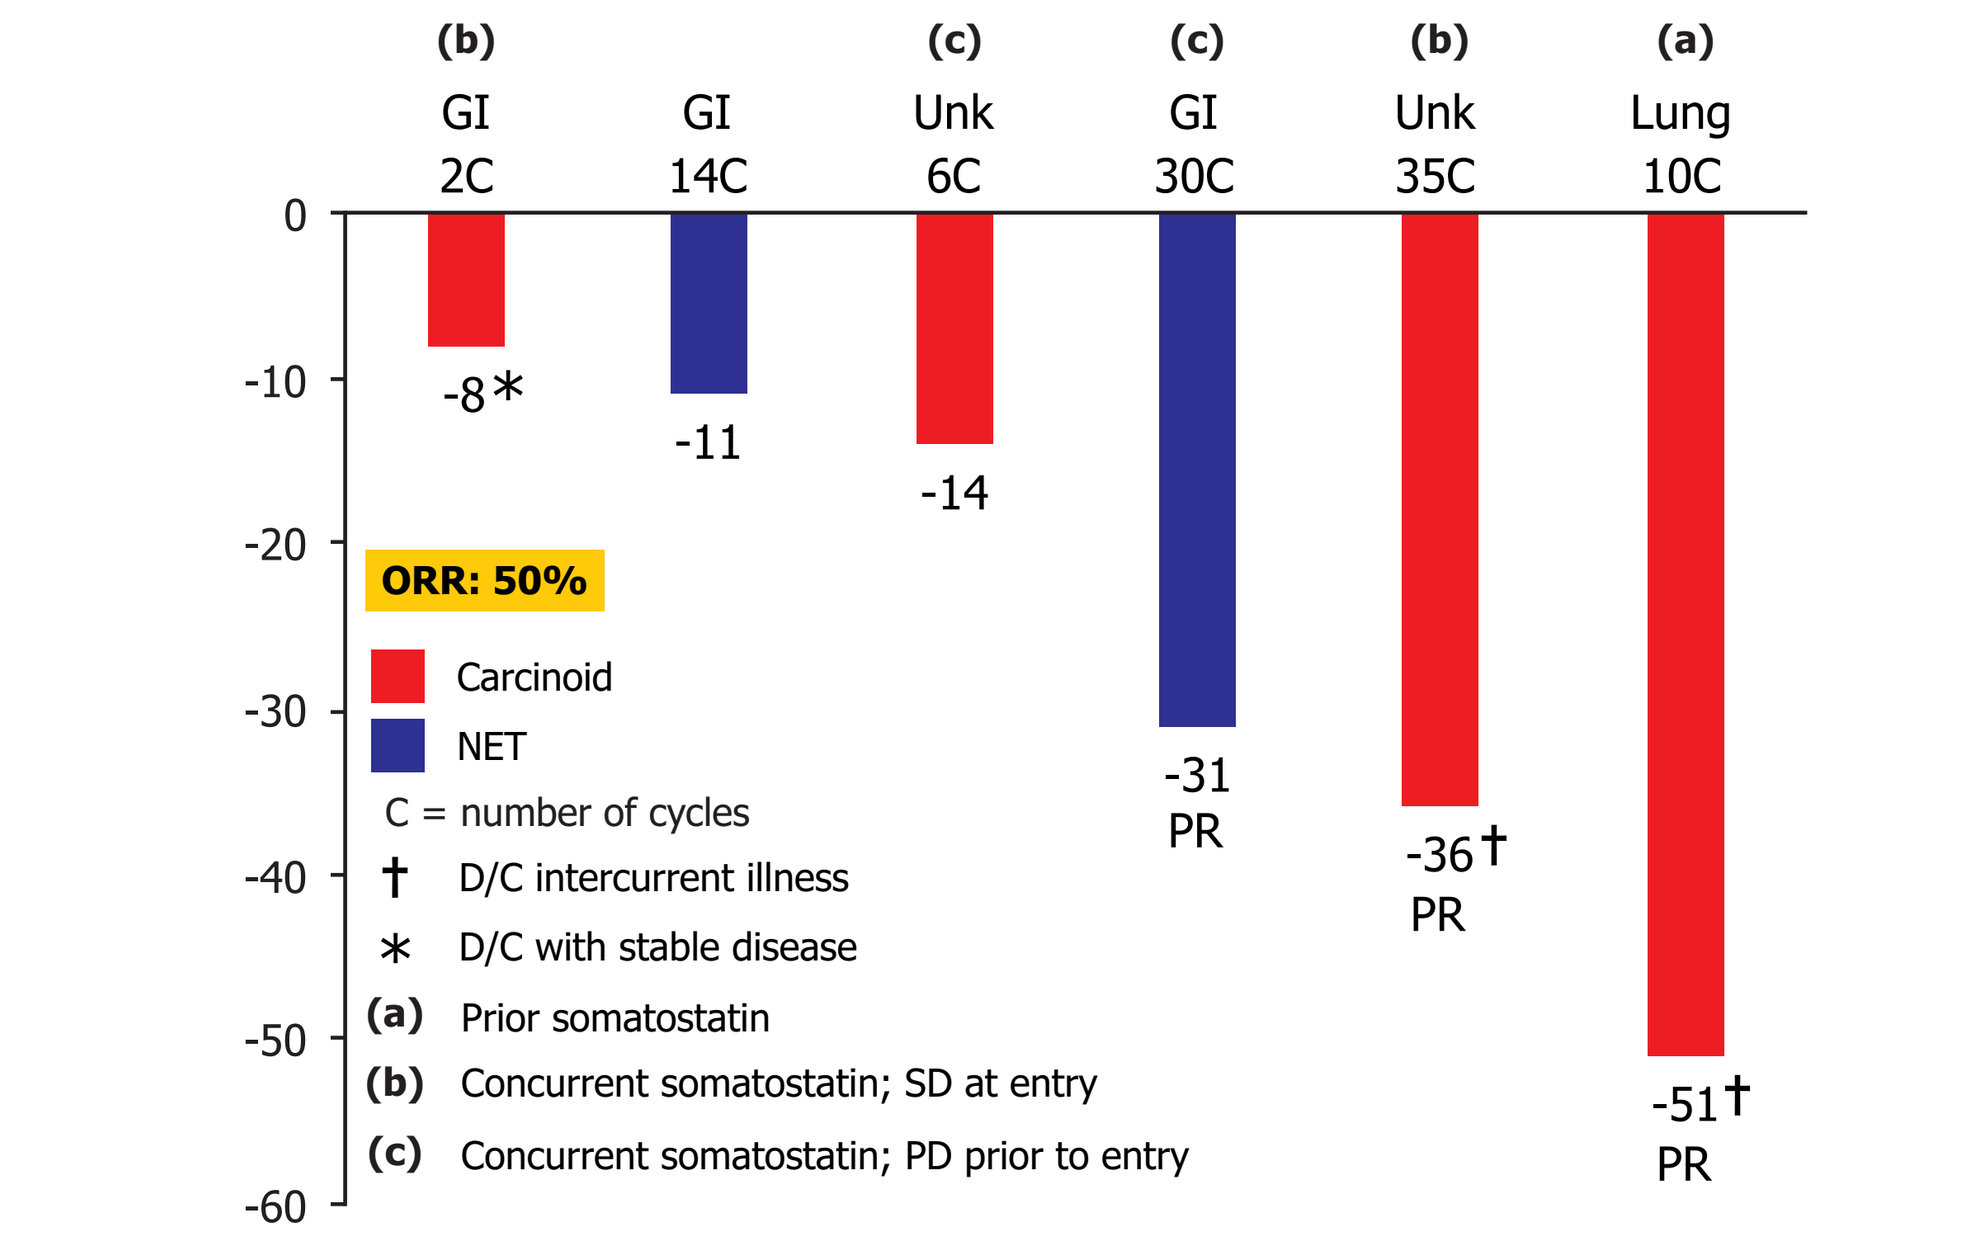
(b) GI 2C
GI 14C
(c) Unk 6C
(c) GI 30C
(b) Unk 35C
(a) Lung 10C
0
-10
-20
-30
-40
-50
-60
-8∗
-11
-14
-31 PR
-36† PR
-51† PR
ORR: 50%
Carcinoid
NET
C = number of cycles
† D/C intercurrent illness
∗ D/C with stable disease
(a) Prior somatostatin
(b) Concurrent somatostatin; SD at entry
(c) Concurrent somatostatin; PD prior to entry

- Six subjects with tumors in the GI tract/lung/unknown carcinoid had ≤2 prior lines of anti-cancer therapy and Ki67 <20%.
- Three of these subjects had partial responses.

Tumor Type	Percent Tumor Reduction	Number of Cycles	Reason for Discontinuation
Lung carcinoid	51%	10	Unrelated intercurrent illness
Unknown carcinoid	36%	35	Intercurrent illness possibly related to everolimus
GI NET	31%	30	Progressive disease

Table 4. Most Common Adverse Events with SNX-5422 plus EVR Combined Therapy*

Preferred term	SNX-5422 50 mg/m^2 N=3		SNX-5422 75 mg/m^2 N=17		Overall N=20	
	Number of Subjects (%)					
	All Cause	Related	All Cause	Related	All Cause	Related
Any TEAE	3 (100)	3 (100)	17 (100)	12 (71)	20 (100)	15 (75)
Diarrhea	2 (67)	1 (33)	14 (82)	12 (71)	16 (80)	13 (65)
Hyponatremia	1 (33)	0	9 (53)	2 (12)	10 (50)	2(10)
Anemia	*2 (67)*	*0*	*8 (48)*	*4 (24)*	*10 (50)*	*4 (20)*
Vomiting	0	0	8 (48)	3 (18)	8 (40)	3 (15)
Nausea	1 (33)	0	6 (35)	4 (24)	7 (35)	4 (20)
Hypokalemia	0	0	6 (35)	1 (6)	6 (30)	1 (5)
AST increased	*0*	*0*	*6 (35)*	*1 (6)*	*6 (30)*	*1 (5)*
Platelet count decreased	*0*	*0*	*6 (35)*	*2 (12)*	*6 (30)*	*2 (10)*
Fatigue	*1 (33)*	*0*	*5 (29)*	*4 (24)*	*6 (30)*	*4 (20)*
Weight decreased	0	0	6 (35)	0	6 (30)	0

TEAE=treatment-emergent adverse event.
* TEAEs occurring in 6 or more subjects overall. Note: TEAEs associated with everolimus are in italics.

- All subjects reported at least 1 TEAE.
- Diarrhea was the most commonly reported TEAE, occurring in 80% of subjects overall. In 13 subjects (65%), the event was considered related to the combination of SNX-5422 and EVR.
- Adverse events associated with EVR included anemia, AST increased, platelet count decreased, and fatigue.

CONCLUSIONS

- SNX-5422 in combination with everolimus showed promising signs of clinical activity in subjects with advanced NETs, with the greatest response seen in those who had ≤2 prior lines of anti-cancer therapy and Ki67 <20%. These subjects also remained on the combination for prolonged periods of time, including 2 subjects for ≥30 cycles.
- SNX-5422 at doses up to 75 mg/m^2 QOD was generally well-tolerated when combined with everolimus.
- Diarrhea was the most common adverse event during SNX-5422 + everolimus combined therapy.

Acknowledgements
The authors would like to thank the study participants and their families as well as the study investigators and study team members.
Content development support provided by Lorraine R. Baer, PharmD, Baer PharMed Consulting, Ltd.

Funding
This study was sponsored by Esanex Inc.

Poster presented at the North American Neuroendocrine Tumor Society (NANETS) Annual Symposium in Philadelphia, PA, October 19-21, 2017.