Integrated Safety Analysis of Telotristat Ethyl in Patients With Carcinoid Heart Disease

Poster 212

Pablo Lapuerta[1], Matthew H. Kulke[2], Marianne Pavel[3,4], Talia Biran[1], Rosanna Fleming[1], Jerome S. Zacks[5], Philipp Hoffmanns[6], Richard R.P. Warner[5]

[1]Lexicon Pharmaceuticals, Inc., The Woodlands, TX, USA; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]Charité–Universitätsmedizin, Berlin, Germany; [4]Current affiliation: Friedrich-Alexander-Universität Erlangen-Nürnberg, Erlangen, Germany; [5]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [6]IPSEN, Boulogne-Bilancourt, Paris, France

Background

- Up to 50% of patients with carcinoid syndrome (CS) may eventually develop carcinoid heart disease (CaHD).[1]
- Telotristat ethyl is a novel, small-molecule tryptophan hydroxylase inhibitor that decreases bowel movement (BM) frequency and urinary 5-hydroxyindoleacetic acid (u5-HIAA) levels in patients with CS and is approved by the US Food and Drug Administration and European Commission for the treatment of CS diarrhea in combination with somatostatin analog (SSA) therapy in adults whose symptoms are inadequately controlled by SSA therapy.[2–4]
- Here we report a post hoc subgroup analysis of the safety of telotristat ethyl in patients with CaHD.

Methods

Subgroup analysis objective

To examine the safety of telotristat ethyl in a subgroup of patients with CS with a medical history of CaHD, using a combined analysis of safety data from both the Double-blind Treatment (DBT) and Open-label Extension (OLE) periods of TELESTAR[5] and TELECAST.[6] OLE data were based on interim data extractions when the OLE periods were still ongoing.

Study designs

FIGURE 1: TELESTAR and TELECAST Study Designs

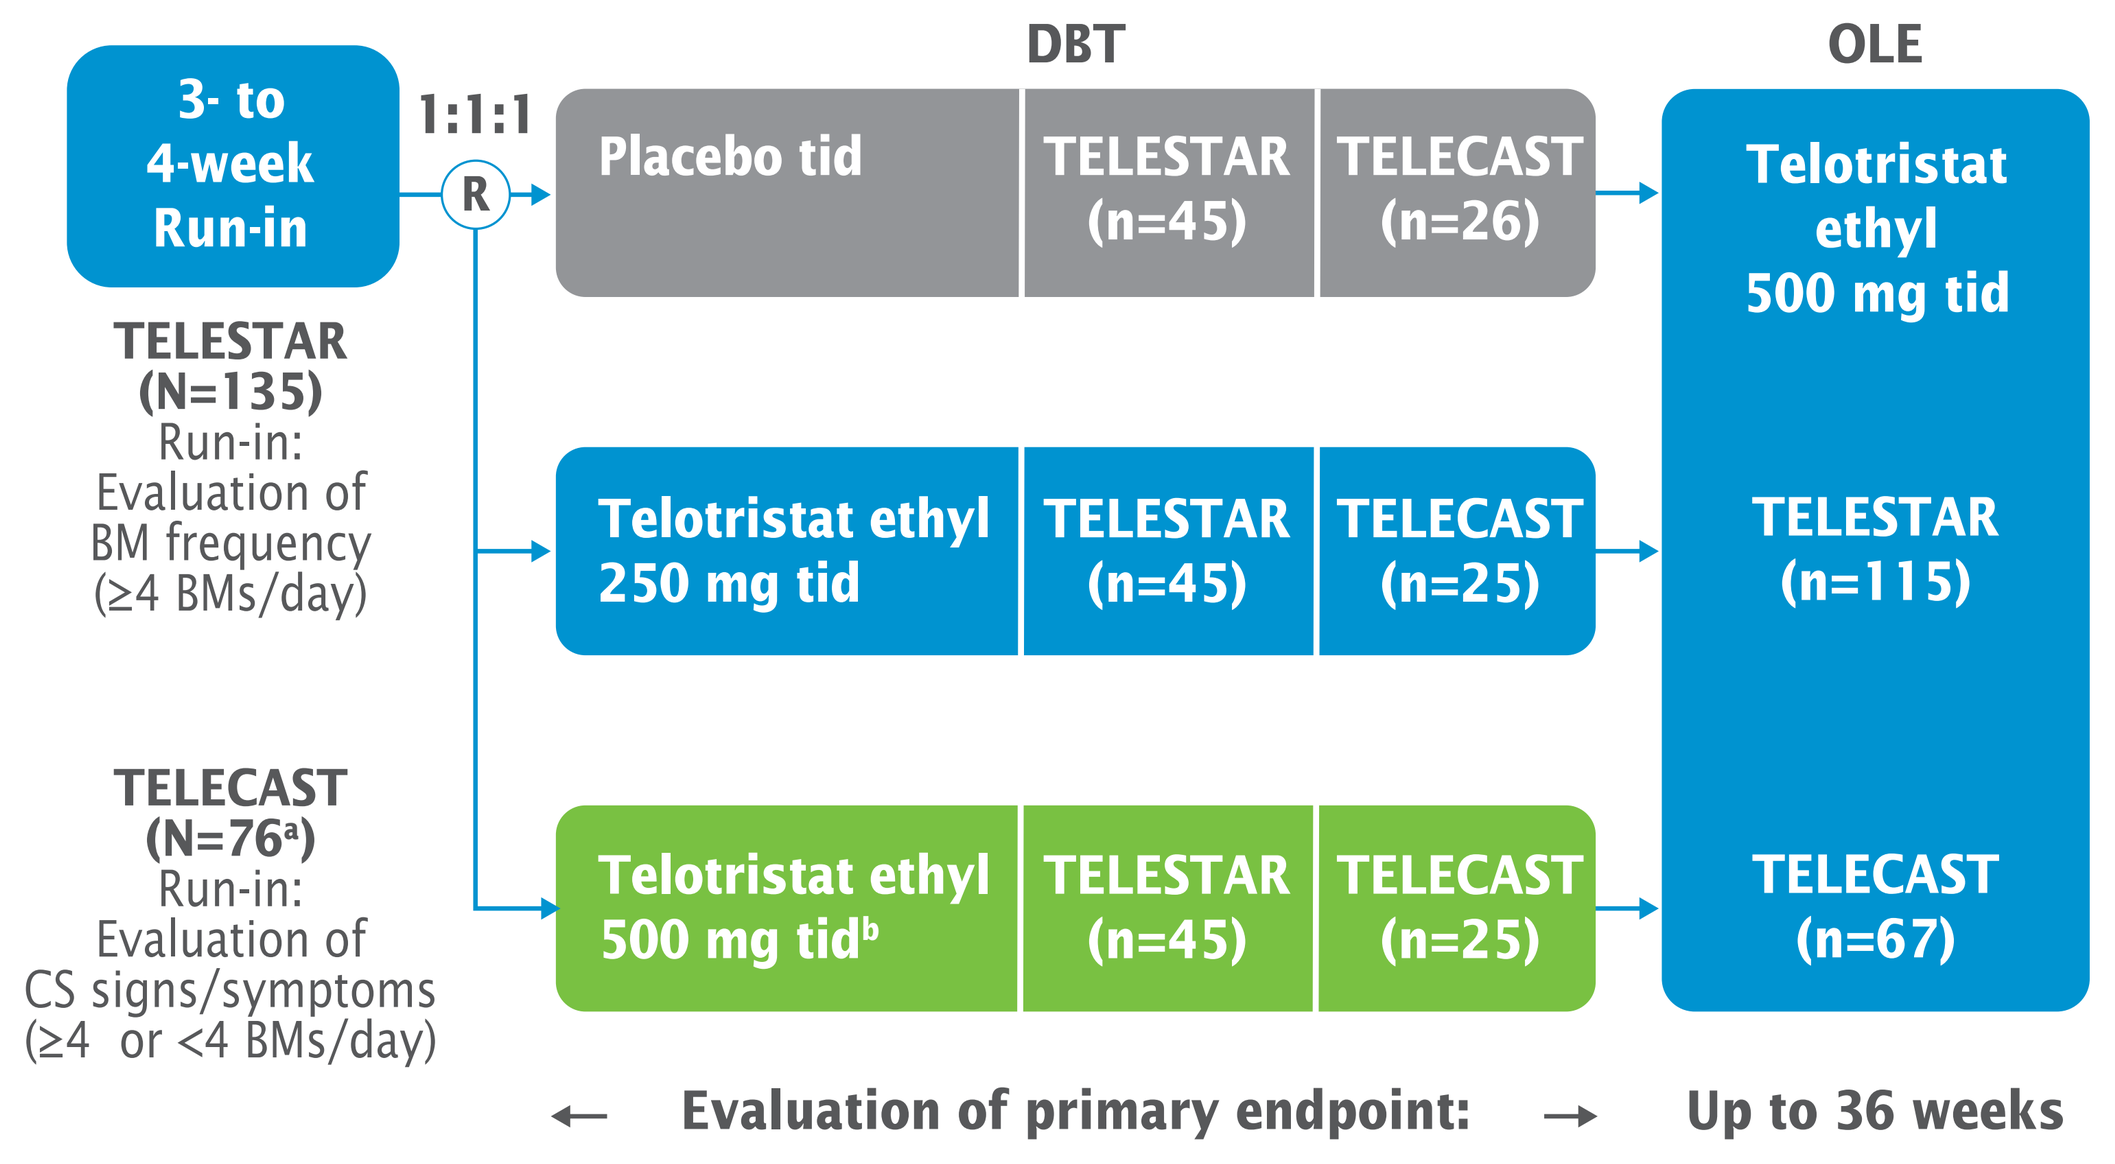
DBT
OLE
3- to 4-week Run-in
1:1:1
R
Placebo tid
TELESTAR (n=45)
TELECAST (n=26)
Telotristat ethyl 500 mg tid
TELESTAR (N=135)
Run-in: Evaluation of BM frequency (≥4 BMs/day)
Telotristat ethyl 250 mg tid
TELESTAR (n=45)
TELECAST (n=25)
TELESTAR (n=115)
TELECAST (N=76[a])
Run-in: Evaluation of CS signs/symptoms (≥4 or <4 BMs/day)
Telotristat ethyl 500 mg tid[b]
TELESTAR (n=45)
TELECAST (n=25)
TELECAST (n=67)
Evaluation of primary endpoint:
Up to 36 weeks

TELESTAR: Reduction in number of daily BMs from Baseline (averaged over the 12-week DBT period)

TELECAST: Reduction from Baseline in u5-HIAA levels at Week 12 and incidence of TEAEs

TELESTAR: All patients **were required to be on long-acting SSA therapy at enrollment** and continued SSA therapy throughout the treatment period.

TELECAST: Eligible patients **may have been receiving long-acting SSA therapy at enrollment.** SSA status remained unchanged throughout the treatment period.

[a]All patients randomly assigned to placebo in the TELECAST study were receiving long-acting SSA therapy, and only 2 (8.0%) and 4 (16.0%) of patients receiving telotristat ethyl 250 mg and 500 mg tid, respectively, were **not** receiving long-acting SSA therapy.
[b]Including a 1-week blinded titration step for patients randomly assigned to receive telotristat ethyl 500 mg tid
BM, bowel movement; CS, carcinoid syndrome; DBT, Double-blind Treatment; OLE, Open-label Extension; R, randomization; SSA, somatostatin analog; TEAEs, treatment-emergent adverse events; tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid

Key inclusion criteria

- In both studies: Documented history of CS and well-differentiated metastatic neuroendocrine tumors (NETs), plus:
 - In TELESTAR: All patients were receiving stable-dose somatostatin analog (SSA) therapy and had ≥4 BMs per day.
 - In TELECAST: Patients on SSA therapy at enrollment had <4 BMs per day AND at least 1 sign/symptom, including daily stool form/consistency ≥5 on the Bristol Stool Form Scale for ≥50% of the days during the Run-in period, average daily flushing episode frequency of ≥2, nausea present ≥20% of days, or u5-HIAA >upper limit of normal (ULN).
- Patients who were not on stable-dose SSAs at enrollment had ≥1 of the above-described signs/symptoms or ≥4 BMs per day.

Key exclusion criteria

In both studies: >12 BMs per day, evidence of *Clostridium difficile* or other enteric infection, previous tumor-directed therapy <4 weeks before screening, or history of short bowel syndrome

Patient population

All patients had metastatic NETs with CS. Fifty-three patients with CaHD were identified in the overall safety analysis (N=211) by manually searching the medical history listings for "carcinoid heart disease," "Hedinger syndrome," and terms that contain "valve," which would indicate cardiac valve insufficiency in the context of CS (**Table 1**).

Safety assessments

- Safety data for the DBT periods of both Phase 3 studies were pooled by treatment arm (placebo, telotristat ethyl 250 mg tid, and telotristat ethyl 500 mg tid) as of their extraction dates (TELESTAR: September 24, 2015; TELECAST: October 30, 2015); interim safety data for the OLE periods of both Phase 3 studies at these extraction dates were pooled by study (treatment arms combined).
- DBT + OLE includes patients who received at least 1 dose of telotristat ethyl during the DBT period, the OLE period, or both.

TABLE 1: Medical History Related to Cardiac Disorders in the CaHD Patient Population

Patient characteristics	Placebo (n=17)	Telotristat ethyl 250 mg tid (n=17)	Telotristat ethyl 500 mg tid (n=19)
Cardiac disorders, n (%)			
Tricuspid valve incompetence	5 (29.4)	8 (47.1)	4 (21.1)
Mitral valve incompetence	3 (17.6)	3 (17.6)	3 (15.8)
Aortic valve incompetence	3 (17.6)	0	1 (5.3)
Carcinoid heart disease	2 (11.8)	1 (5.9)	6 (31.6)
Tricuspid valve disease	0	0	2 (10.5)
Heart valve incompetence	1 (5.9)	1 (5.9)	3 (15.8)
Cardiac valve disease	2 (11.8)	0	2 (10.5)
Pulmonary valve incompetence	2 (11.8)	0	2 (10.5)
Aortic valve disease	2 (11.8)	0	0
Pulmonary valve stenosis	0	1 (5.9)	0
Tricuspid valve stenosis	0	0	1 (5.3)
Aortic valve stenosis	0	0	1 (5.3)
Surgical and medical procedures, n (%)			
Pulmonary valve replacement	2 (11.8)	4 (23.5)	1 (5.3)
Tricuspid valve replacement	4 (23.5)	6 (35.3)	3 (15.8)
Heart valve replacement	0	1 (5.9)	1 (5.3)
Mitral valve repair	0	0	1 (5.3)

CaHD, carcinoid heart disease; tid, 3 times per day

Results

- Across both studies, Baseline demographics for the CaHD subset were similar across treatment arms.
- The majority of patients presented with metabolism and nutrition disorders (61% in TELESTAR, 60% in TELECAST), including cachexia (TELESTAR only) and malnutrition (TELECAST only), and general disorders and administration site conditions (70% in TELESTAR, 55% in TELECAST), including fatigue and weakness/asthenia.

TABLE 2: Baseline Characteristics of Patients With CS and CaHD in TELESTAR and TELECAST Studies

Patient characteristics	Placebo (n=17)	Telotristat ethyl 250 mg tid (n=17)	Telotristat ethyl 500 mg tid (n=19)	DBT + OLE (all treatment arms) (n=51)
Age, mean (years)	63.6	56.7	65.6	62.3
Male sex, n (%)	8 (47.1)	10 (58.8)	12 (63.2)	29 (56.9)
Body mass index, mean (kg/m²)	22.3	25.2	23.2	23.7
Mean u5-HIAA, mg/day (min, max)				
TELESTAR	92 (0, 289), n=8	141 (11, 638), n=11	115 (10, 454), n=14	NA
TELECAST	77 (8, 225), n=9	93 (18, 196), n=6	124 (3, 332), n=5	NA

CaHD, carcinoid heart disease; CS, carcinoid syndrome; DBT, Double-blind Treatment; NA, not available; OLE, Open-label Extension; tid, 3 times per day

Efficacy assessment

Median change from Baseline to Week 12 in u5-HIAA in patients with CaHD showed reductions in both telotristat ethyl groups for TELESTAR and TELECAST (**Figure 2**).

FIGURE 2: Median Percent Change in u5-HIAA Levels From Baseline to Week 12

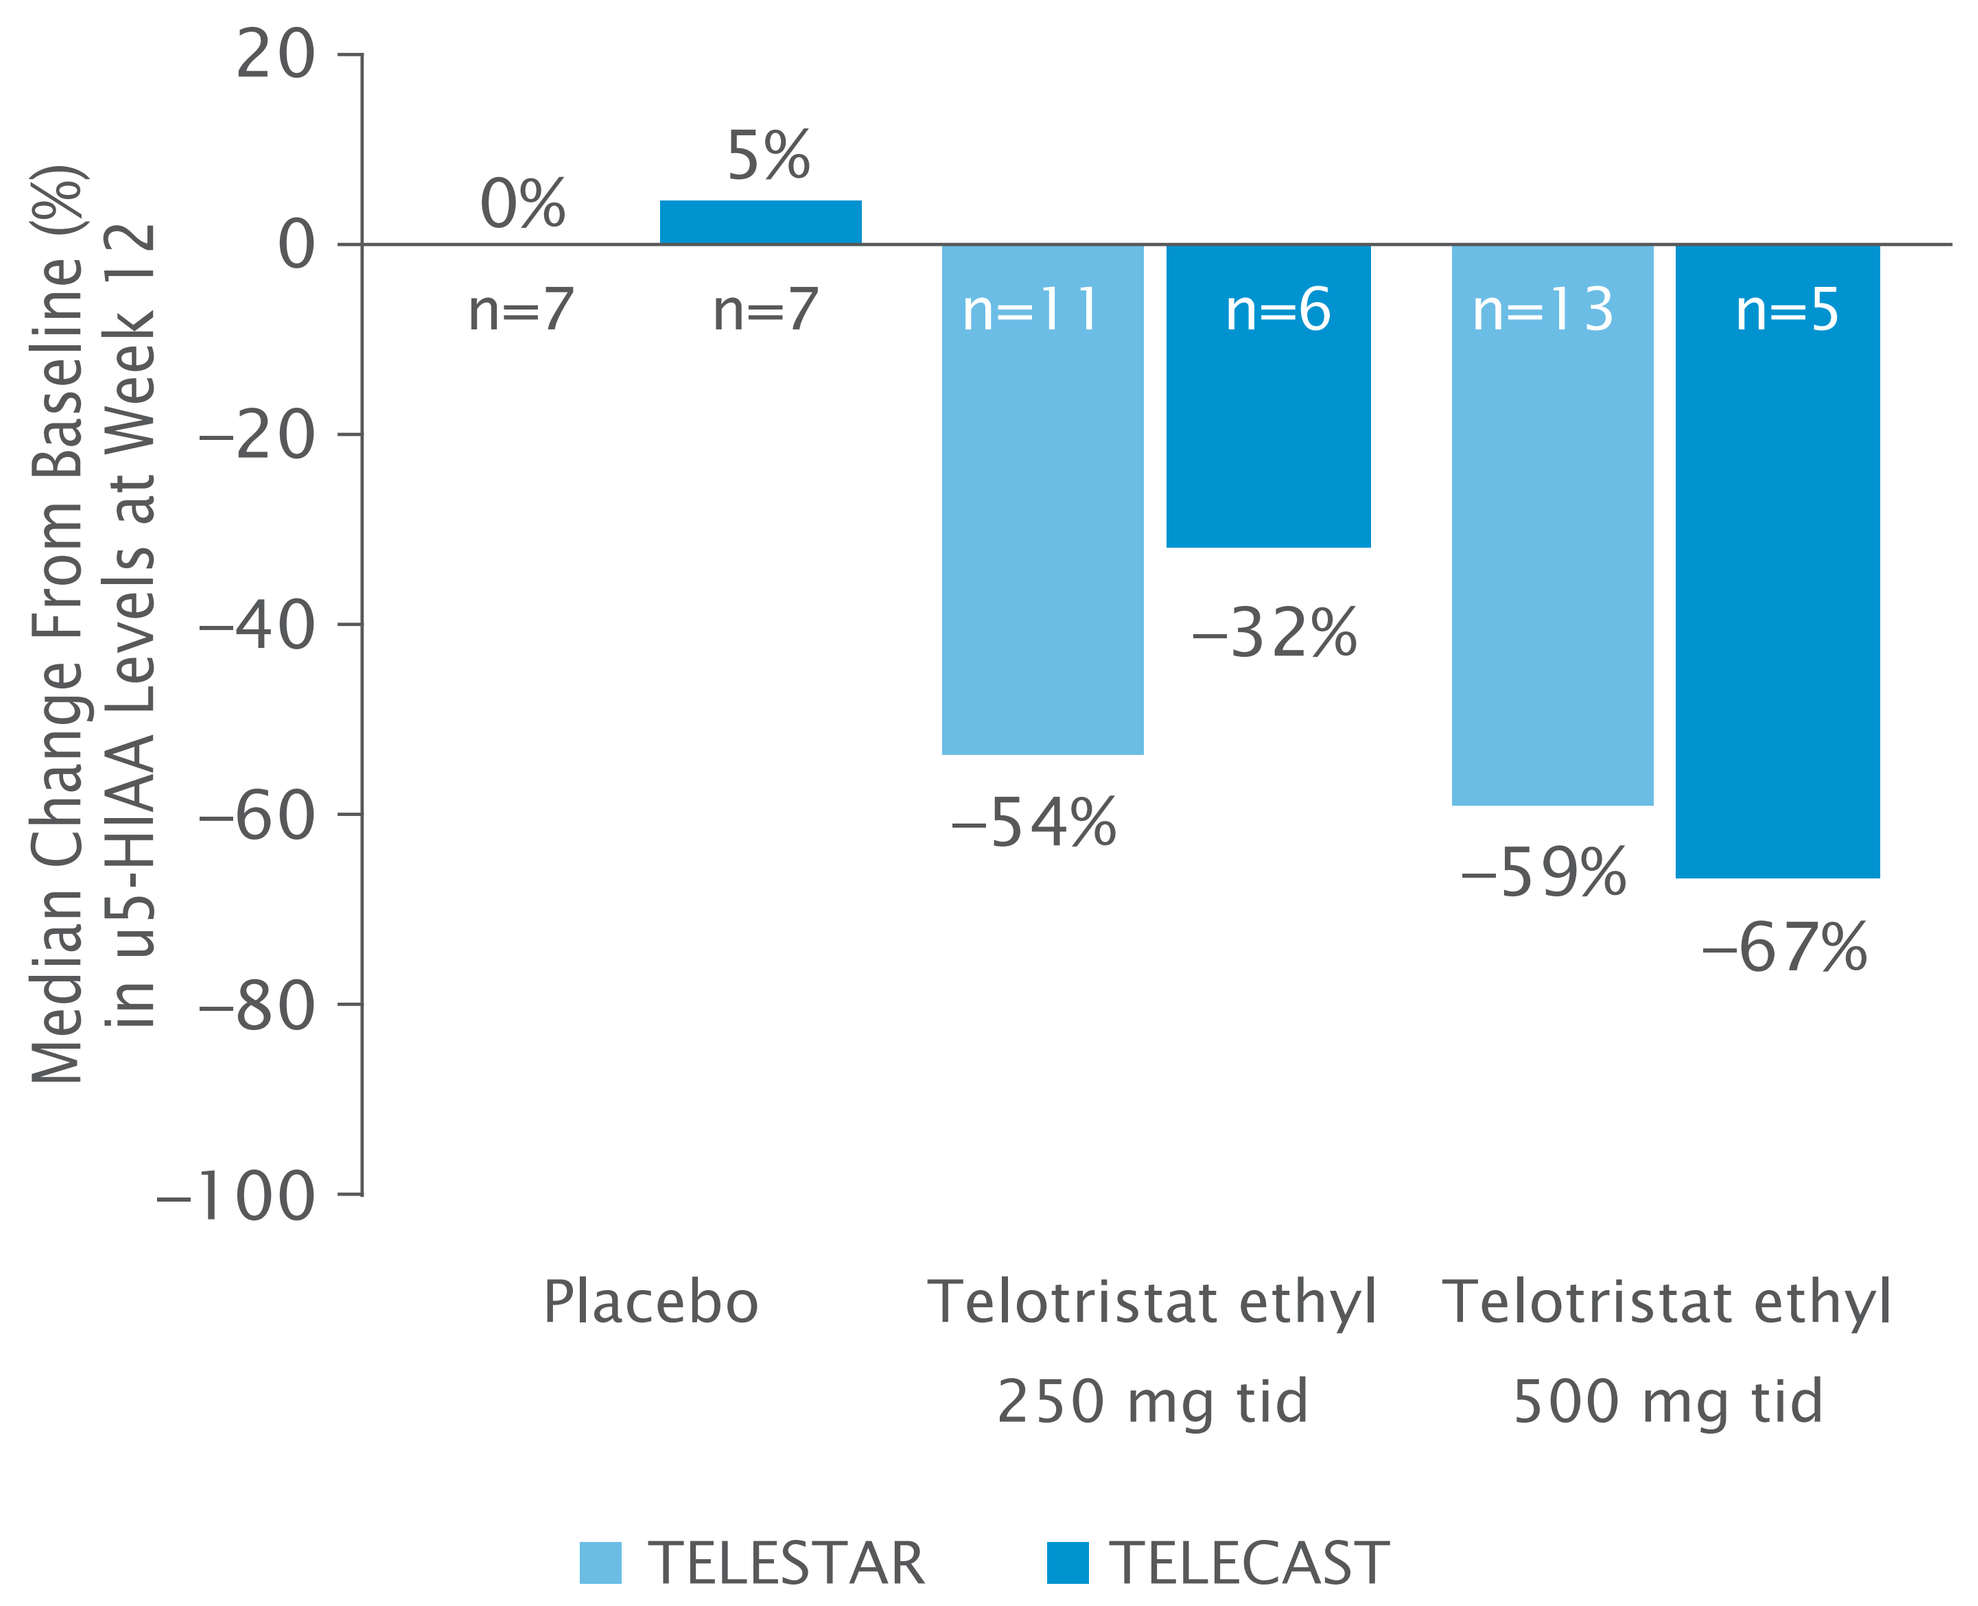
Median Change From Baseline (%) in u5-HIAA Levels at Week 12
20
0
−20
−40
−60
−80
−100
0%
5%
n=7
n=7
n=11
n=6
n=13
n=5
−54%
−32%
−59%
−67%
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
TELESTAR
TELECAST

tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid

Overall combined safety analysis

- The safety experience was similar between the DBT and DBT + OLE groups (**Table 3**).
- No treatment-related TEAEs resulting in death were observed in either the DBT or the DBT + OLE group.

TABLE 3: Overall Combined Safety Summary in Patients With CaHD in the TELESTAR and TELECAST Studies

Patient characteristics	Placebo (n=17)	Telotristat ethyl 250 mg tid (n=17)	Telotristat ethyl 500 mg tid (n=19)	DBT + OLE (all treatment arms) (n=51)[a]
Patients with any TEAE	15 (88.2)	15 (88.2)	17 (89.5)	49 (96.1)
Patients with any SAE	6 (35.3)	4 (23.5)	2 (10.5)	25 (49.0)
Patients with any severe TEAE	4 (23.5)	0	1 (5.3)	23 (45.1)
Patients with any treatment-related TEAE	5 (29.4)	8 (47.1)	12 (63.2)	33 (64.7)
Patients with any severe treatment-related TEAE	0	0	0	8 (15.7)
Patients with any TEAE leading to study drug discontinuation	2 (11.8)	3 (17.6)	4 (21.1)	26 (51.0)
Patients with any TEAE resulting in death	1 (5.9)	0	0	6 (11.8)
Patients with any treatment-related TEAE resulting in death	0	0	0	0

[a]TEAEs on placebo during the DBT period were not included in this column.
CaHD, carcinoid heart disease; DBT, Double-blind Treatment; OLE, Open-label Extension; SAE, serious adverse event; TEAE, treatment-emergent adverse event; tid, 3 times per day

- The most common TEAEs (≥15%) in the CaHD subgroup in the DBT + OLE groups while on telotristat ethyl were nausea (39.2%), abdominal pain (29.4%), abdominal distension (15.7%), vomiting (15.7%), asthenia (15.7%), fatigue (15.7%), decreased appetite (15.7%), and depression (15.7%).
- No differences have been noted between the adverse event profile for telotristat ethyl in the CaHD population compared with the overall study populations.[7–9]

Adverse events of special interest

The incidence of new cardiac events was low in both the DBT and DBT + OLE groups (**Table 4**).

TABLE 4: Incidence of Adverse Events of Special Interest in Patients With CaHD

Adverse event, n (%)	Placebo (n=17)	Telotristat ethyl 250 mg tid (n=17)	Telotristat ethyl 500 mg tid (n=19)	DBT + OLE (all treatment arms) (n=51)[a]
Gastrointestinal disorders				
Constipation	0	2 (11.8)	1 (5.3)	7 (13.7)
Cardiac disorders				
Acute myocardial infarction	0	0	0	2 (3.9)
Cardiac failure	0	0	0	2 (3.9)
Heart valve incompetence	0	0	0	2 (3.9)
Tachycardia	0	0	0	2 (3.9)
Arrhythmia supraventricular	0	0	0	1 (2.0)
Bradycardia	0	0	0	1 (2.0)
Carcinoid heart disease[b]	0	0	0	1 (2.0)
Cardiovascular disorder	0	0	0	1 (2.0)
Tachyarrhythmia	0	0	0	1 (2.0)
Mitral valve incompetence	1 (5.9)	0	0	0
Pulmonary valve incompetence	0	0	0	0
Pulmonary valve stenosis	0	0	0	0
Tricuspid valve incompetence	0	0	0	0

[a]TEAEs on placebo during the DBT period were not included in this column.
[b]MedDRA preferred term
CaHD, carcinoid heart disease; DBT, Double-blind Treatment; MedDRA, Medical Dictionary for Regulatory Activities; OLE, Open-label Extension; tid, 3 times per day

Conclusions

- The safety profile for telotristat ethyl was similar in patients with CaHD and the overall safety population.[5,7–9]
- The safety experience was also similar between patients in the DBT and DBT + OLE groups.
- The incidence of new cardiac disorders was low in both the DBT and DBT + OLE groups for the CaHD population.
- Gastrointestinal disorders were relatively common and included constipation.

REFERENCES
1. Fox DJ, Khattar RS. *Heart*. 2004;90(10):1224–1228. **2.** National Comprehensive Cancer Network. Neuroendocrine Tumors (Version 3.2017). 2017. **3.** Xermelo® (telotristat ethyl) US Highlights of Prescribing Information. Lexicon Pharmaceuticals, Inc. February 2017. **4.** European Commission. Commission Implementing Decision of 18.9.2017 granting marketing authorisation under Regulation (EC) No 726/2004 of the European Parliament and of the Council for "Xermelo - telotristat", an orphan medicinal product for human use. 2017. http://ec.europa.eu/health/documents/community-register/2017/20170918138652/dec_138652_en.pdf. Accessed October 2, 2017. **5.** Kulke MH, et al. *J Clin Oncol*. 2017;35(1):14–23. **6.** Pavel M, et al. Presented at North American Neuroendocrine Tumor Society (NANETS) Annual Meeting; September 30–October 1, 2016; Jackson, WY. **7.** Horsch D, et al. Presented at European Neuroendocrine Tumor Society (ENETS) Annual Meeting; March 9–11, 2016; Barcelona, Spain. **8.** Kulke MH, et al. Presented at European Society for Medical Oncology (EMSO) Annual Meeting; October 7–11, 2016; Copenhagen, Denmark. **9.** Pavel M, et al. Presented at European Neuroendocrine Tumor Society (ENETS) Annual Meeting; March 8–10, 2017; Barcelona, Spain.

ACKNOWLEDGEMENTS
We thank the patients and Investigators for participating in the studies. We thank James Banigan, PhD (Chameleon Communications International with funding from Lexicon Pharmaceuticals, Inc.) for editorial assistance in the preparation of this report. The TELESTAR and TELECAST studies were sponsored by Lexicon Pharmaceuticals, Inc.

DISCLOSURES
PL, TB, and RF: current employees of Lexicon Pharmaceuticals, Inc., and may own common stock or may have been granted stock options or other equity incentive awards. MHK: Lexicon Pharmaceuticals, Inc., Ipsen Biopharmaceuticals, and Novartis Pharmaceuticals. MP: Ipsen Biopharmaceuticals, Novartis Pharmaceuticals, Lexicon Pharmaceuticals, Inc., and Pfizer. JSZ: Lexicon Pharmaceuticals, Inc.

To view this poster online, snap a photo of this QR code with your smart phone (QR reader required).

Presented at the NANETS 2017 Annual Symposium; October 19–21, 2017; Philadelphia, PA, USA.

Lexicon pharmaceuticals