Long-term safety/tolerability of lanreotide autogel/depot in neuroendocrine tumor patients with carcinoid syndrome: the ELECT long-term open-label extension

Aaron Vinik,[1] George A. Fisher, Jr,[2] Pamela L. Kunz,[2] Nilani Liyanage,[3] Beloo Mirakhur,[4] Susan Pitman Lowenthal,[4] Rodney F. Pommier,[5] Montaser Shaheen,[6] Edward M. Wolin,[7] on behalf of ELECT Study Investigators

[1]Eastern Virginia Medical School, Norfolk, VA, USA; [2]Stanford University School of Medicine, Stanford, CA, USA; [3]Ipsen, Boulogne-Billancourt, France; [4]Ipsen, Basking Ridge, NJ, USA; [5]Oregon Health & Science University, Portland, OR, USA; [6]University of New Mexico Cancer Center, Albuquerque, NM, USA; [7]Montefiore Einstein Center for Cancer Care, Bronx, NY, USA.

Introduction

- Mainstay treatments for the symptoms of carcinoid syndrome (CS) are long-acting somatostatin analogs (SSAs), supplemented with short-acting SSAs for breakthrough symptoms.[1,2]
- The efficacy and safety of the SSA lanreotide depot (also known as autogel) for CS symptoms have been demonstrated previously in European studies[3,4] and the treatment has been licensed for this use in Europe for more than 10 years.
- The double-blind (DB) phase of the ELECT study recently demonstrated that use of the short-acting SSA octreotide as rescue medication for flushing and/or diarrhea events was reduced with lanreotide depot 120 mg every 4 weeks compared with placebo.[5]
- Symptomatic control was sustained subsequently in patients continuing into the single-arm (lanreotide) initial open-label (IOL) phase (NCT00774930).
- Here, we present results from the final phase, the long-term open-label extension (LTOLE), which evaluated long-term safety/tolerability.

Methods

Patients

- Key inclusion criteria for the ELECT study included:
 - Adults (≥18 years).
 - Somatostatin receptor-positive, histopathologically confirmed neuroendocrine tumor (NET), or a tumor of unknown location with liver metastases.
 - A history of CS.
 - SSA-naïve or responsive to octreotide long-acting release (LAR; ≤30 mg/4 weeks) or short-acting octreotide (≤600 µg/daily).
 - No evidence of tumor progression on two sequential scans within 6 months prior to entry.[5]
- Key exclusion criteria included:
 - History of CS refractory to conventional SSA doses.
 - Treatment with interferon, chemotherapy, and/or peptide receptor radionuclide therapy, and/or tumor debulking <3 months before study entry.
 - History of hepatic arterial embolization or chemoembolization and/or selective internal radiation therapy <6 months before study entry.
 - Life expectancy <1 year.

Study design and treatment

- The ELECT study (NCT00774930; EudraCT 2010-019066-92) consisted of a screening period, conducted up to 4 months before randomization, followed by three phases:
 - A 16-week, DB, randomized, placebo-controlled phase,
 - A 32-week IOL phase and,
 - An LTOLE phase in countries where lanreotide had not been approved for the treatment of symptoms of carcinoid syndrome.
- The LTOLE phase ended after the last patient completed the IOL phase, or after marketing approval was obtained in the respective country (whichever occurred first).

Figure 1. Patient flow (ITT population).

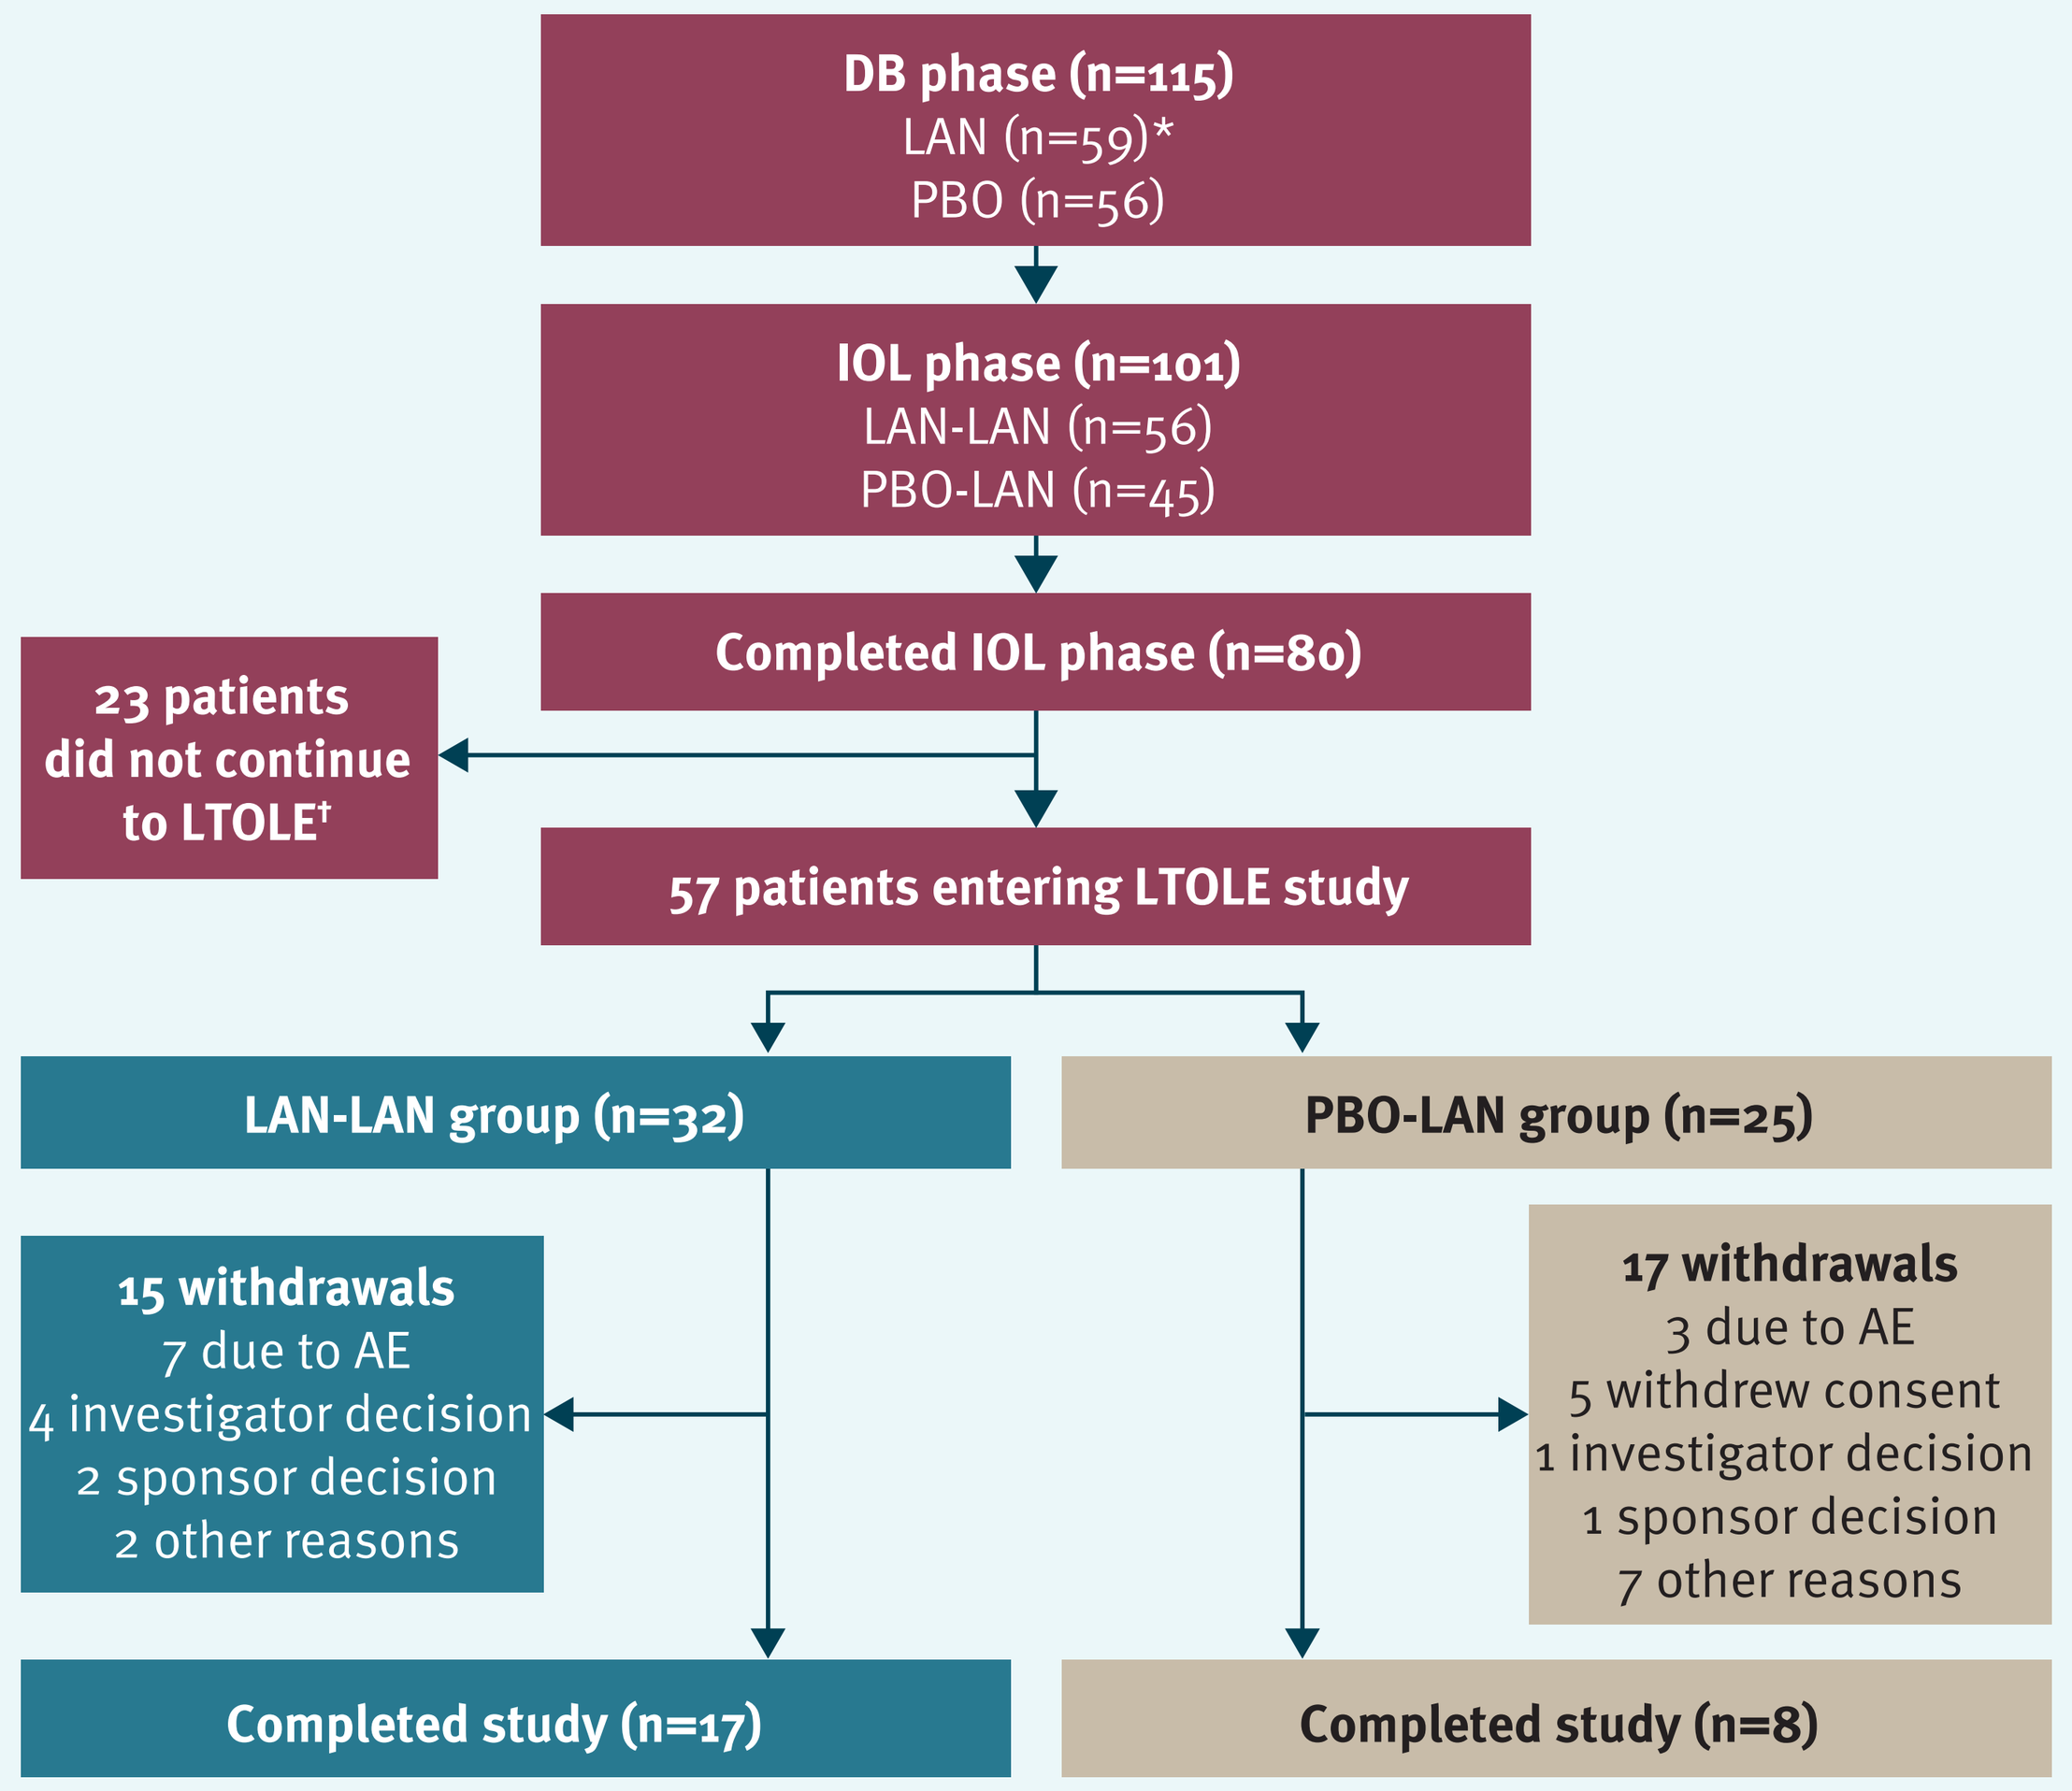
DB phase (n=115)
LAN (n=59)*
PBO (n=56)
IOL phase (n=101)
LAN-LAN (n=56)
PBO-LAN (n=45)
Completed IOL phase (n=80)
23 patients did not continue to LTOLE†
57 patients entering LTOLE study
LAN-LAN group (n=32)
PBO-LAN group (n=25)
15 withdrawals
7 due to AE
4 investigator decision
2 sponsor decision
2 other reasons
17 withdrawals
3 due to AE
5 withdrew consent
1 investigator decision
1 sponsor decision
7 other reasons
Completed study (n=17)
Completed study (n=8)

*One patient was randomly assigned to the lanreotide group received placebo in error and was included in the lanreotide group for the ITT population but the placebo group for the safety population. †Patients did not continue as sites in countries where lanreotide depot is already approved for CS symptom control did not participate in the LTOLE phase, or patients completed IOL before the LTOLE phase was created. AE, adverse event; DB, double-blind; ITT, intention-to-treat; LAN-LAN, patients initially receiving lanreotide depot continuing to receive lanreotide depot; LAN, lanreotide depot 120 mg; LTOLE, long-term open-label extension; PBO, placebo; PBO-LAN, patients initially receiving placebo switching to lanreotide depot.

Table 1. Demographic and disease characteristics at baseline of the DB phase for patients continuing in the LTOLE phase (ITT population).

	LAN-LAN (n=32)	PBO-LAN (n=25)	All patients (n=57)
Mean (SD) age, years	56 (11)	60 (12)	58 (11)
Men	13 (41)	8 (32)	21 (37)
Ethnicity			
White	21 (66)	16 (64)	37 (65)
Multiracial	6 (19)	4 (16)	10 (18)
Asian	3 (9)	3 (12)	6 (11)
Black/African American	2 (6)	2 (8)	4 (7)
Mean (SD) BMI, kg/m²	26 (6)	27 (4)	27 (5)
Time since first symptoms to treatment start			
<1 year	8 (25)	11 (44)	19 (33)
≥1 year	24 (75)	14 (56)	38 (67)
Time since first diagnosis to treatment start			
<1 year	12 (38)	13 (52)	25 (44)
≥1 year	20 (63)	12 (48)	32 (56)
Region and prior SSA therapy at any time			
USA	9 (28)	5 (20)	14 (25)
Prior therapy	7 (22)	5 (20)	12 (21)
Not USA	23 (72)	20 (80)	43 (75)
Prior therapy	4 (13)	7 (28)	11 (19)
Any prior SSA therapy within 3 months of screening	9 (28)	10 (40)	19 (33)
Use of short-acting octreotide during screening	16 (50)	15 (60)	31 (54)

Data are number (%) unless stated otherwise.
BMI, body mass index; DB, double-blind; ITT, intention-to-treat; LAN, lanreotide depot 120 mg; LAN-LAN, patients who received lanreotide depot in double-blind, IOL, and LTOLE phases; LTOLE, long-term open-label extension; PBO, placebo; PBO-LAN, patients who received placebo in the DB phase and lanreotide depot in the IOL and LTOLE phases; SD, standard deviation; SSA, somatostatin analog.

- During the LTOLE phase, patients received lanreotide depot 120 mg every 4 weeks, with short-acting octreotide (100–600 µg/day in 2–4 divided doses) for control of breakthrough CS symptoms used, when needed, at the patient's discretion.

Study endpoints and assessments (LTOLE phase)

- The objective was to evaluate the long-term safety of lanreotide depot 120 mg every 4 weeks.
- Study visits were performed every 4 weeks, up to week 264, with a follow-up visit every 6 months.
- Adverse events (AEs; including diarrhea and flushing, reported once worsened from the IOL phase) and concomitant medication use (including short-acting octreotide use) were recorded at each study visit.
- Gallbladder echography was performed at weeks 96, 144, 192, and 240, and also if a patient developed gallbladder symptoms.

Statistical analyses

- Summary statistics were calculated according to the treatments received:
 - Lanreotide in the DB phase, continuing into IOL and LTOLE phases (lanreotide [LAN]-LAN).
 - Placebo in the DB phase, switching to lanreotide in IOL and LTOLE phases (placebo [PBO]-LAN).
- Analyses were conducted using the intention-to-treat (ITT) population (all randomized patients) for baseline characteristics and the safety population (all randomized patients who received at least one injection of lanreotide 120 mg) for safety data.

Results

Patient disposition and baseline characteristics

- A total of 115 patients took part in the DB phase (lanreotide, n=59; placebo, n=56) and 101 continued into the IOL phase; of these, 57 continued into the LTOLE phase (Figure 1).
- Demographic and disease characteristics at baseline of the DB phase for patients continuing in the LTOLE phase (ITT population) are summarized in Table 1.
 - The majority of US patients had prior SSA therapy, while most non-US patients were SSA-naïve.

Safety/tolerability

- The median treatment exposure for all patients receiving lanreotide in the ELECT study was 72 weeks (interquartile range: 48–166) (Table 2).
- A summary of AE incidence by study phase is presented in Table 2 and the most common AEs are listed by class and preferred term in Table 3.

Double-blind phase

- During the 16-week DB phase (as previously published[5]), similar proportions of patients in both groups experienced AEs; most were mild to moderate in severity (Table 2).
- Few patients experienced serious AEs (SAEs), with one SAE considered related to treatment occurring in each group.
- There were no deaths in the lanreotide group.
- The most frequently reported class of AE was gastrointestinal disorders (Table 3).
- Lanreotide treatment had no notable effect on variables relating to vital signs and electrocardiogram (ECG) functions, but there were reported incidences of weight decreased.

Table 2. Summary of safety data by study phase (safety population).

	DB phase (16 weeks)*		IOL phase (32 weeks)*			LTOLE phase (≥2 years)			All phases
	DB LAN (n=58)	DB PBO (n=57)†	IOL LAN-LAN (n=56)	IOL PBO-LAN (n=45)	All patients (n=101)	LTOLE LAN-LAN (n=32)	LTOLE PBO-LAN (n=25)	All patients (n=57)	All patients receiving LAN (n=103)
Median treatment exposure, weeks (IQR)	16 (16–16)	16 (8–16)	32 (32–33)	32 (31–33)	32 (31–33)	125 (76–152)	87 (48–120)	110 (64–135)	72 (48–166)
Any AE, n (%)	31 (53)	34 (60)	39 (70)	32 (71)	71 (70)	27 (84)	21 (84)	48 (84)	85 (83)
Mild	13 (21)	16 (28)	13 (23)	10 (22)	23 (23)	2 (6)	5 (20)	7 (12)	16 (16)
Moderate	13 (22)	14 (25)	17 (30)	19 (42)	36 (36)	13 (41)	10 (40)	23 (40)	41 (40)
Severe	5 (10)	4 (7)	9 (16)	3 (7)	12 (12)	12 (38)	6 (24)	18 (32)	28 (27)
TRAEs, n (%)	15 (26)	11 (19)	15 (27)	11 (24)	26 (26)	7 (22)	6 (24)	13 (23)	39 (38)
Severe	2 (3)	1 (2)	2 (4)	0	2 (2)	1 (3)	1 (4)	2 (4)	6 (6)
SAEs, n (%)	2 (3)	5 (9)	7 (13)	1 (2)	8 (8)	10 (31)	5 (20)	15 (26)	22 (21)
Treatment-related	1 (2)	1 (2)	0	0	0	1 (3)	1 (4)	2 (4)	3 (3)
AEs with death as outcome, n (%)	0	2 (4)	0	0	0	4 (13)	1 (4)	5 (9)	5 (5)
Treatment-related	0	1 (2)	0	0	0	0	0	0	0
AEs leading to discontinuation, n (%)	1 (2)	1 (2)	0	1 (2)	1 (1)	3 (9)	2 (8)	5 (9)	7 (7)
Treatment-related	0	1 (2)	0	0	0	0	0	0	0

Data are for patients completing each respective phase. Direct comparison of AE incidence between phases is not appropriate as not all patients entered the IOL and LTOLE phases. *Diarrhea and flushing were recorded as part of efficacy assessments and not as safety assessments during the DB and IOL phases, and as AEs in the LTOLE phase. Diarrhea and flushing were reported in the LTOLE phase once worsened from the last record in the patient's diary during IOL phase. †One patient was randomized to the lanreotide group but received placebo in error and was included in the lanreotide group for the ITT population but the placebo group for the safety population.
AE, adverse event; DB, double-blind; IQR, interquartile range; ITT, intention-to-treat; LAN, lanreotide depot 120 mg; LAN-LAN, patients who received lanreotide depot in double-blind, IOL, and LTOLE phases; LTOLE, long-term open-label extension; IOL, initial open-label; PBO, placebo; PBO-LAN, patients who received placebo in the DB phase and lanreotide depot in the IOL and LTOLE phases; SAE, serious adverse event; SD, standard deviation; TRAE, treatment-related adverse event.

- Two clinically significant out of range laboratory values (recorded as AEs) were considered related to treatment: blood viscosity increased (lanreotide group) and blood glucose increased (placebo group).

Initial open-label phase

- During the 32-week IOL phase, 39 (70%) LAN-LAN patients compared with 32 (71%) PBO-LAN patients experienced AEs.
- Most AEs were mild to moderate in severity, 12 (12%) patients experienced a severe AE (Table 2).

Table 3. Incidence of AEs with LAN treatment during each phase of the ELECT study.

	DB phase (LAN, n=58)	IOL phase (All patients*, n=101)	LTOLE phase (All patients*, n=57)	All phases (All patients receiving LAN, n=103)
Most common† AEs by class				
GI events	19 (33)	37 (37)	30 (53)	53 (52)
General/administration site	9 (16)	24 (24)	26 (46)	46 (45)
Infection/infestation	9 (16)	18 (18)	23 (40)	39 (38)
Musculoskeletal and connective tissue	8 (14)	25 (25)	19 (33)	36 (35)
Nervous system disorders	13 (22)	19 (19)	12 (21)	34 (33)
Investigations	3 (5)	9 (9)	13 (23)	31 (30)
Metabolism/nutrition	3 (5)	17 (17)	18 (32)	29 (28)
Respiratory, thoracic, and mediastinal	2 (3)	15 (15)	12 (21)	26 (25)
Most common‡ individual AEs				
Abdominal pain	5 (9)	11 (11)	14 (25)	21 (20)
Headache	7 (12)	5 (11)	5 (9)	17 (17)
Dizziness	4 (7)	1 (2)	3 (5)	11 (11)
Nausea	5 (9)	8 (8)	4 (7)	16 (16)
Fatigue	2 (3)	10 (10)	9 (16)	16 (16)
Vomiting	4 (7)	6 (6)	5 (9)	13 (13)
Decreased appetite	1 (2)	4 (9)	5 (9)	13 (13)
Arthralgia	0	7 (7)	6 (11)	12 (12)
Back pain	1 (2)	5 (5)	8 (14)	11 (11)
Constipation	2 (3)	3 (7)	5 (9)	11 (11)
Hypertension	0	3 (7)	5 (9)	11 (11)
Hyperglycemia	0	4 (4)	8 (14)	10 (10)
Asthenia	2 (3)	2 (2)	6 (11)	10 (10)
Diarrhea§	0	0	8 (14)	8 (8)
Most common# TRAEs				
Cholelithiasis	0	3 (3)	4 (7)	6 (6)
Nausea	3 (5)	3 (3)	0	6 (6)
Abdominal pain	1 (2)	3 (3)	1 (2)	5 (5)
Fatigue	2 (3)	3 (3)	2 (4)	5 (5)
Hyperglycemia	0	2 (2)	2 (4)	3 (3)

Data are number (%) of patients. Direct comparison of AE incidence between phases is not appropriate as not all patients entered the IOL and LTOLE phases. *PBO-LAN and LAN-LAN groups. †Occurring in ≥20% of patients (in LTOLE or All phases). ‡Occurring in ≥10% of patients (in LTOLE or All phases). §Diarrhea and flushing were recorded as part of efficacy assessments and not as safety assessments during the DB and IOL phases, and as AEs in the LTOLE phase. Diarrhea and flushing were reported in the LTOLE phase once worsened from the last record in the patient's diary during IOL phase. #Occurring in ≥4% of patients (in LTOLE or All phases).
AE, adverse event; DB, double-blind; GI, gastrointestinal; IOL, initial open-label; LAN, lanreotide depot 120 mg; LAN-LAN, patients who received lanreotide depot in double-blind, IOL, and LTOLE phases; LTOLE, long-term open-label extension; PBO, placebo; PBO-LAN, patients who received placebo in the DB phase and lanreotide depot in the IOL and LTOLE phases; TRAE, treatment-related adverse event.

- 26 (26%) patients experienced treatment-related AEs (TRAEs), of these two experienced severe TRAEs.
- Eight patients experienced SAEs but none were treatment-related and there were no deaths.
- Consistent with the DB phase, the most frequently reported class of AE was gastrointestinal disorders (Table 3).
- Similarly, there was no effect on vital signs or ECG functions in the IOL phase, but AEs of weight decreased were reported.
- One AE of change in laboratory values considered related to treatment occurred in the PBO-LAN group: gamma glutamyl transpeptidase increased.
- Cholelithiasis occurred in three patients (3%) during the IOL phase.

Long-term open-label extension phase

- Median treatment exposure in the LTOLE phase was 110 weeks (interquartile range: 64–135).
- A total of 29 (51%) of patients received short-acting octreotide during the LTOLE phase.
- In the LAN-LAN group, 27 (84%) patients compared with 21 (84%) patients in the PBO-LAN group experienced AEs.
- Ten (31%) patients in the LAN-LAN group experienced a total of 19 SAEs, and five (20%) in the PBO-LAN group experienced a total of 11 SAEs.
- One patient in each group experienced an SAE that was considered to be related to treatment.
- Five patients had study drug withdrawn owing to an AE, and five AEs with an outcome of death occurred (none of these events were treatment-related).
- Consistent with the previous phases, the most frequently reported class of AE was gastrointestinal disorders (Table 3).
- As in previous phases, there were no AEs related to vital signs or ECG functions, but incidences of weight decrease occurred.
- One AE of increase in blood triglycerides (LAN-LAN group) considered related to treatment was the only clinically relevant change in laboratory parameters.
- Cholelithiasis occurred in four patients (7%) during the LTOLE.

Conclusions

- Overall, new safety concerns were not identified during the ELECT LTOLE phase and long-term safety data are consistent with the established safety/tolerability profile of lanreotide autogel/depot 120 mg every 4 weeks.
- These data add to the evidence supporting the long term use of lanreotide in patients with NETs.[4–7]

Acknowledgments

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield company, for medical writing support in preparing this poster.

References

1. Kulke MH *et al.* Neuroendocrine tumors, version 1.2015. *J Natl Compr Canc Netw* 2015;13:78–108.
2. Pavel M *et al.* ENETS consensus guidelines update for the management of distant metastatic disease of intestinal, pancreatic, bronchial neuroendocrine neoplasms (NEN) and NEN of unknown primary site. *Neuroendocrinology* 2016;103:172–85.
3. Ruszniewski P *et al.* Rapid and sustained relief from the symptoms of carcinoid syndrome: results from an open 6-month study of the 28-day prolonged-release formulation of lanreotide. *Neuroendocrinology* 2004;80:244–51.
4. Ruszniewski P *et al.* Patient-reported outcomes with lanreotide Autogel/Depot for carcinoid syndrome: An international observational study. *Dig Liver Dis* 2016;48:552–8.
5. Vinik AI *et al.* Evaluation of lanreotide depot/autogel efficacy and safety as a carcinoid syndrome treatment (Elect): A randomized, double-blind, placebo-controlled trial. *Endocr Pract* 2016;[Epub ahead of print] doi: 10.4158/EP151172.OR
6. Caplin ME *et al.* Lanreotide in metastatic enteropancreatic neuroendocrine tumors. *N Engl J Med* 2014;371:224–33.
7. Caplin ME *et al.* Anti-tumour effects of lanreotide for pancreatic and intestinal neuroendocrine tumours: the CLARINET open-label extension study. *Endocr Relat Cancer* 2016;23:191–9.

Presented at the 2016 NANETS Annual Symposium, 30 September–1 October 2016, Jackson, WY, USA.

This study is sponsored by Ipsen