Efficacy and Safety of Telotristat Ethyl in Patients With Carcinoid Syndrome Inadequately Controlled by Somatostatin Analogs: Analysis of the Completed TELESTAR Extension Period

Poster 214

Dieter Hörsch[1], Matthew H. Kulke[2], Martyn E. Caplin[3], Lowell B. Anthony[4], Emily Bergsland[5], Kjell Öberg[6], Richard R. P. Warner[7], Pamela L. Kunz[8], Enrique Grande[9], Juan W. Valle[10], Joseph S. Dillon[11], Pablo Lapuerta[12], Phillip Banks[12], Shanna Jackson[12], Marianne Pavel[13,14]

[1]Zentralklinik Bad Berka, Bad Berka, Germany; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]Royal Free Hospital, London, UK; [4]University of Kentucky, Lexington, KY, USA; [5]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA, USA; [6]Uppsala University, Uppsala, Sweden; [7]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [8]Stanford University, Palo Alto, CA, USA; [9]Hospital Ramón y Cajal, Madrid, Spain; [10]University of Manchester/The Christie NHS Foundation Trust, Manchester, UK; [11]University of Iowa, Iowa City, IA, USA; [12]Lexicon Pharmaceuticals, Inc., The Woodlands, TX, USA; [13]Charité–Universitätsmedizin, Berlin, Germany; [14]Current affiliation: Friedrich-Alexander-Universität Erlangen-Nürnberg, Erlangen, Germany

Background

- Carcinoid syndrome (CS) occurs in <20% of patients with neuroendocrine tumors (NETs) and is characterized by an overproduction of serotonin (5-HT), which causes diarrhea, abdominal pain, and carcinoid heart disease.[1–4]
- Somatostatin analogs (SSAs) are the standard treatment for patients with CS, but patients may develop recurrent symptoms.[5,6]
- Telotristat ethyl is a novel, small-molecule tryptophan hydroxylase inhibitor that decreases urinary 5-hydroxyindoleacetic acid (u5-HIAA) levels and bowel movement (BM) frequency in patients with CS and is approved by the US Food and Drug Administration and European Commission for the treatment of CS diarrhea in combination with SSA therapy in adults whose symptoms are inadequately controlled by SSA therapy.[7–9]

Objective

- To examine the efficacy and safety of telotristat ethyl from the full 48 weeks of the TELESTAR study, including the open-label extension (OLE) period.

Methods

Study design

- TELESTAR is a Phase 3, randomized, multicenter, parallel-group, double-blind, placebo-controlled study consisting of a 12-week double-blind treatment (DBT) period and a 36-week OLE period (ClinicalTrials.gov identifier: NCT01677910, **Figure 1**).[10]

Key inclusion criteria

- Age ≥18 years old with histopathologically confirmed, well-differentiated metastatic NETs and a documented history of CS
- Was receiving stable-dose SSAs (long-acting release, depot, or infusion pump) for ≥3 months prior to enrollment and had ≥4 BMs per day (BMs/day)

Key exclusion criteria

- Had undergone tumor-directed therapy <4 weeks before screening; >12 watery BMs/day associated with volume contraction, dehydration, or hypotension; evidence of enteric infection; Karnofsky Performance Status ≤60%; or a history of short bowel syndrome

FIGURE 1: TELESTAR Study Design

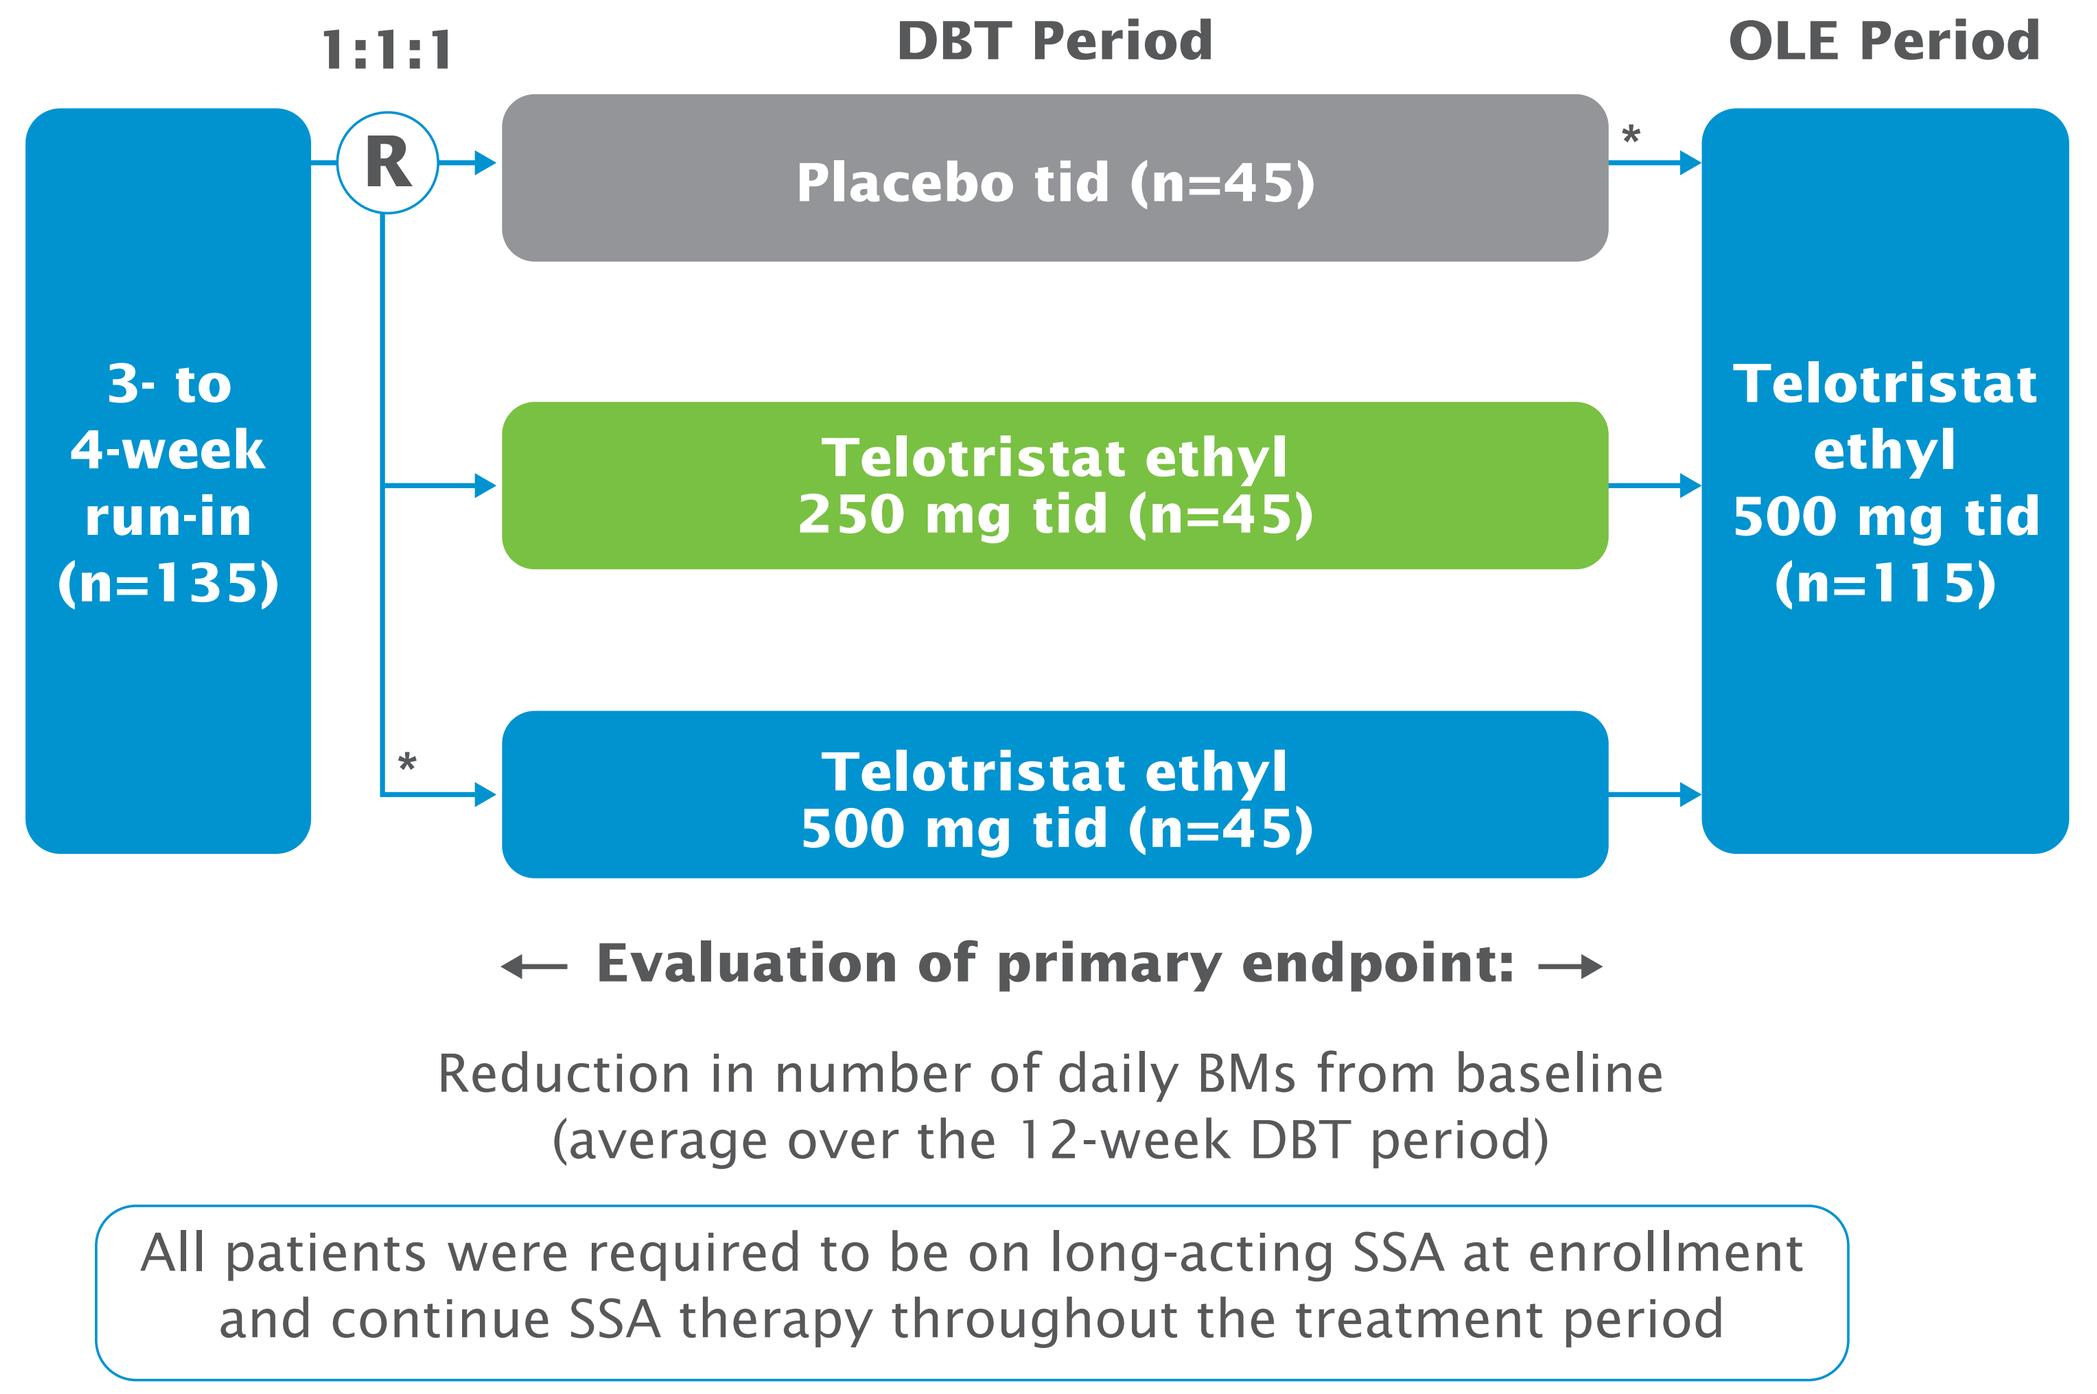
1:1:1
DBT Period
OLE Period
3- to 4-week run-in (n=135)
R
Placebo tid (n=45)
Telotristat ethyl 250 mg tid (n=45)
Telotristat ethyl 500 mg tid (n=45)
Telotristat ethyl 500 mg tid (n=115)
*
*
← Evaluation of primary endpoint: →
Reduction in number of daily BMs from baseline (average over the 12-week DBT period)
All patients were required to be on long-acting SSA at enrollment and continue SSA therapy throughout the treatment period

*Including a blinded titration step of 1 week of 250 mg tid

BMs, bowel movements; DBT, double-blind treatment; OLE, open-label extension; R, randomization, SSA, somatostatin analog; tid, 3 times per day

Efficacy assessments

- Changes from baseline in BM frequency, u5-HIAA, and European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30 (EORTC QLQ-C30) scores

Safety assessments

- Incidence of treatment-emergent adverse events (TEAEs)
- Changes from baseline in clinical laboratory results, vital signs results, physical examinations, and electrocardiogram findings

Results

- 118 of 135 patients completed the DBT period (38, 42, and 38 patients on placebo tid, telotristat ethyl 250 mg tid, and telotristat ethyl 500 mg tid, respectively).
- 115 patients participated and 79 patients completed the OLE period.
- 36 patients discontinued the OLE period, primarily for adverse events (15 patients), withdrawal of consent (9 patients), and lack of efficacy (5 patients).
- Cumulative exposure to telotristat ethyl for patients who entered the OLE period was 38.4 weeks (standard deviation 11.4 weeks).

TABLE 1: Patient Demographics

Patient characteristics	DBT Placebo (n=45)	DBT Telotristat ethyl 250 mg tid (n=45)	DBT Telotristat ethyl 500 mg tid (n=45)	OLE Telotristat ethyl 500 mg tid (n=115)
Mean age, years (SD)	63.3 (8.7)	62.4 (9.1)	64.9 (9.1)	63.2 (9.3)
Median age, years (range)	64 (42–80)	64 (37–83)	65 (44–88)	64 (37–88)
Male, n (%)	24 (53.3)	21 (46.7)	25 (55.6)	59 (51.3)
SSA therapy at study entry, n (%)				
Octreotide	30 (66.7)	40 (88.9)	33 (73.3)	86 (74.8)
Lanreotide	15 (33.3)	5 (11.1)	12 (26.7)	29 (25.2)
Mean BM frequency at baseline, counts/day (SD)	5.2 (1.4)	6.1 (2.1)	5.8 (2.0)	5.7 (1.8)
u5-HIAA at baseline, n (%)[a]				
≤ULN	12 (26.7)	12 (26.7)	12 (26.7)	33 (28.7)
>ULN	26 (57.8)	26 (57.8)	26 (57.8)	65 (56.5)
Unknown	7 (15.6)	7 (15.6)	7 (15.6)	17 (14.8)
Mean u5-HIAA at baseline, mg/day (SD)	81.0 (161.0)	92.6 (114.9)	89.5 (144.5)	72.2 (115.5)

[a]Reference range: normal u5-HIAA 0–15 mg per 24 hours[11]

BM, bowel movement; DBT, double-blind treatment; OLE, open-label extension; SD, standard deviation; SSA, somatostatin analog; tid, 3 times per day; u5-HIAA, urinary 5-hydroxindoleacetic acid; ULN, upper limit of normal

FIGURE 2: Primary Efficacy: Mean Change From Baseline in BMs (counts/day)

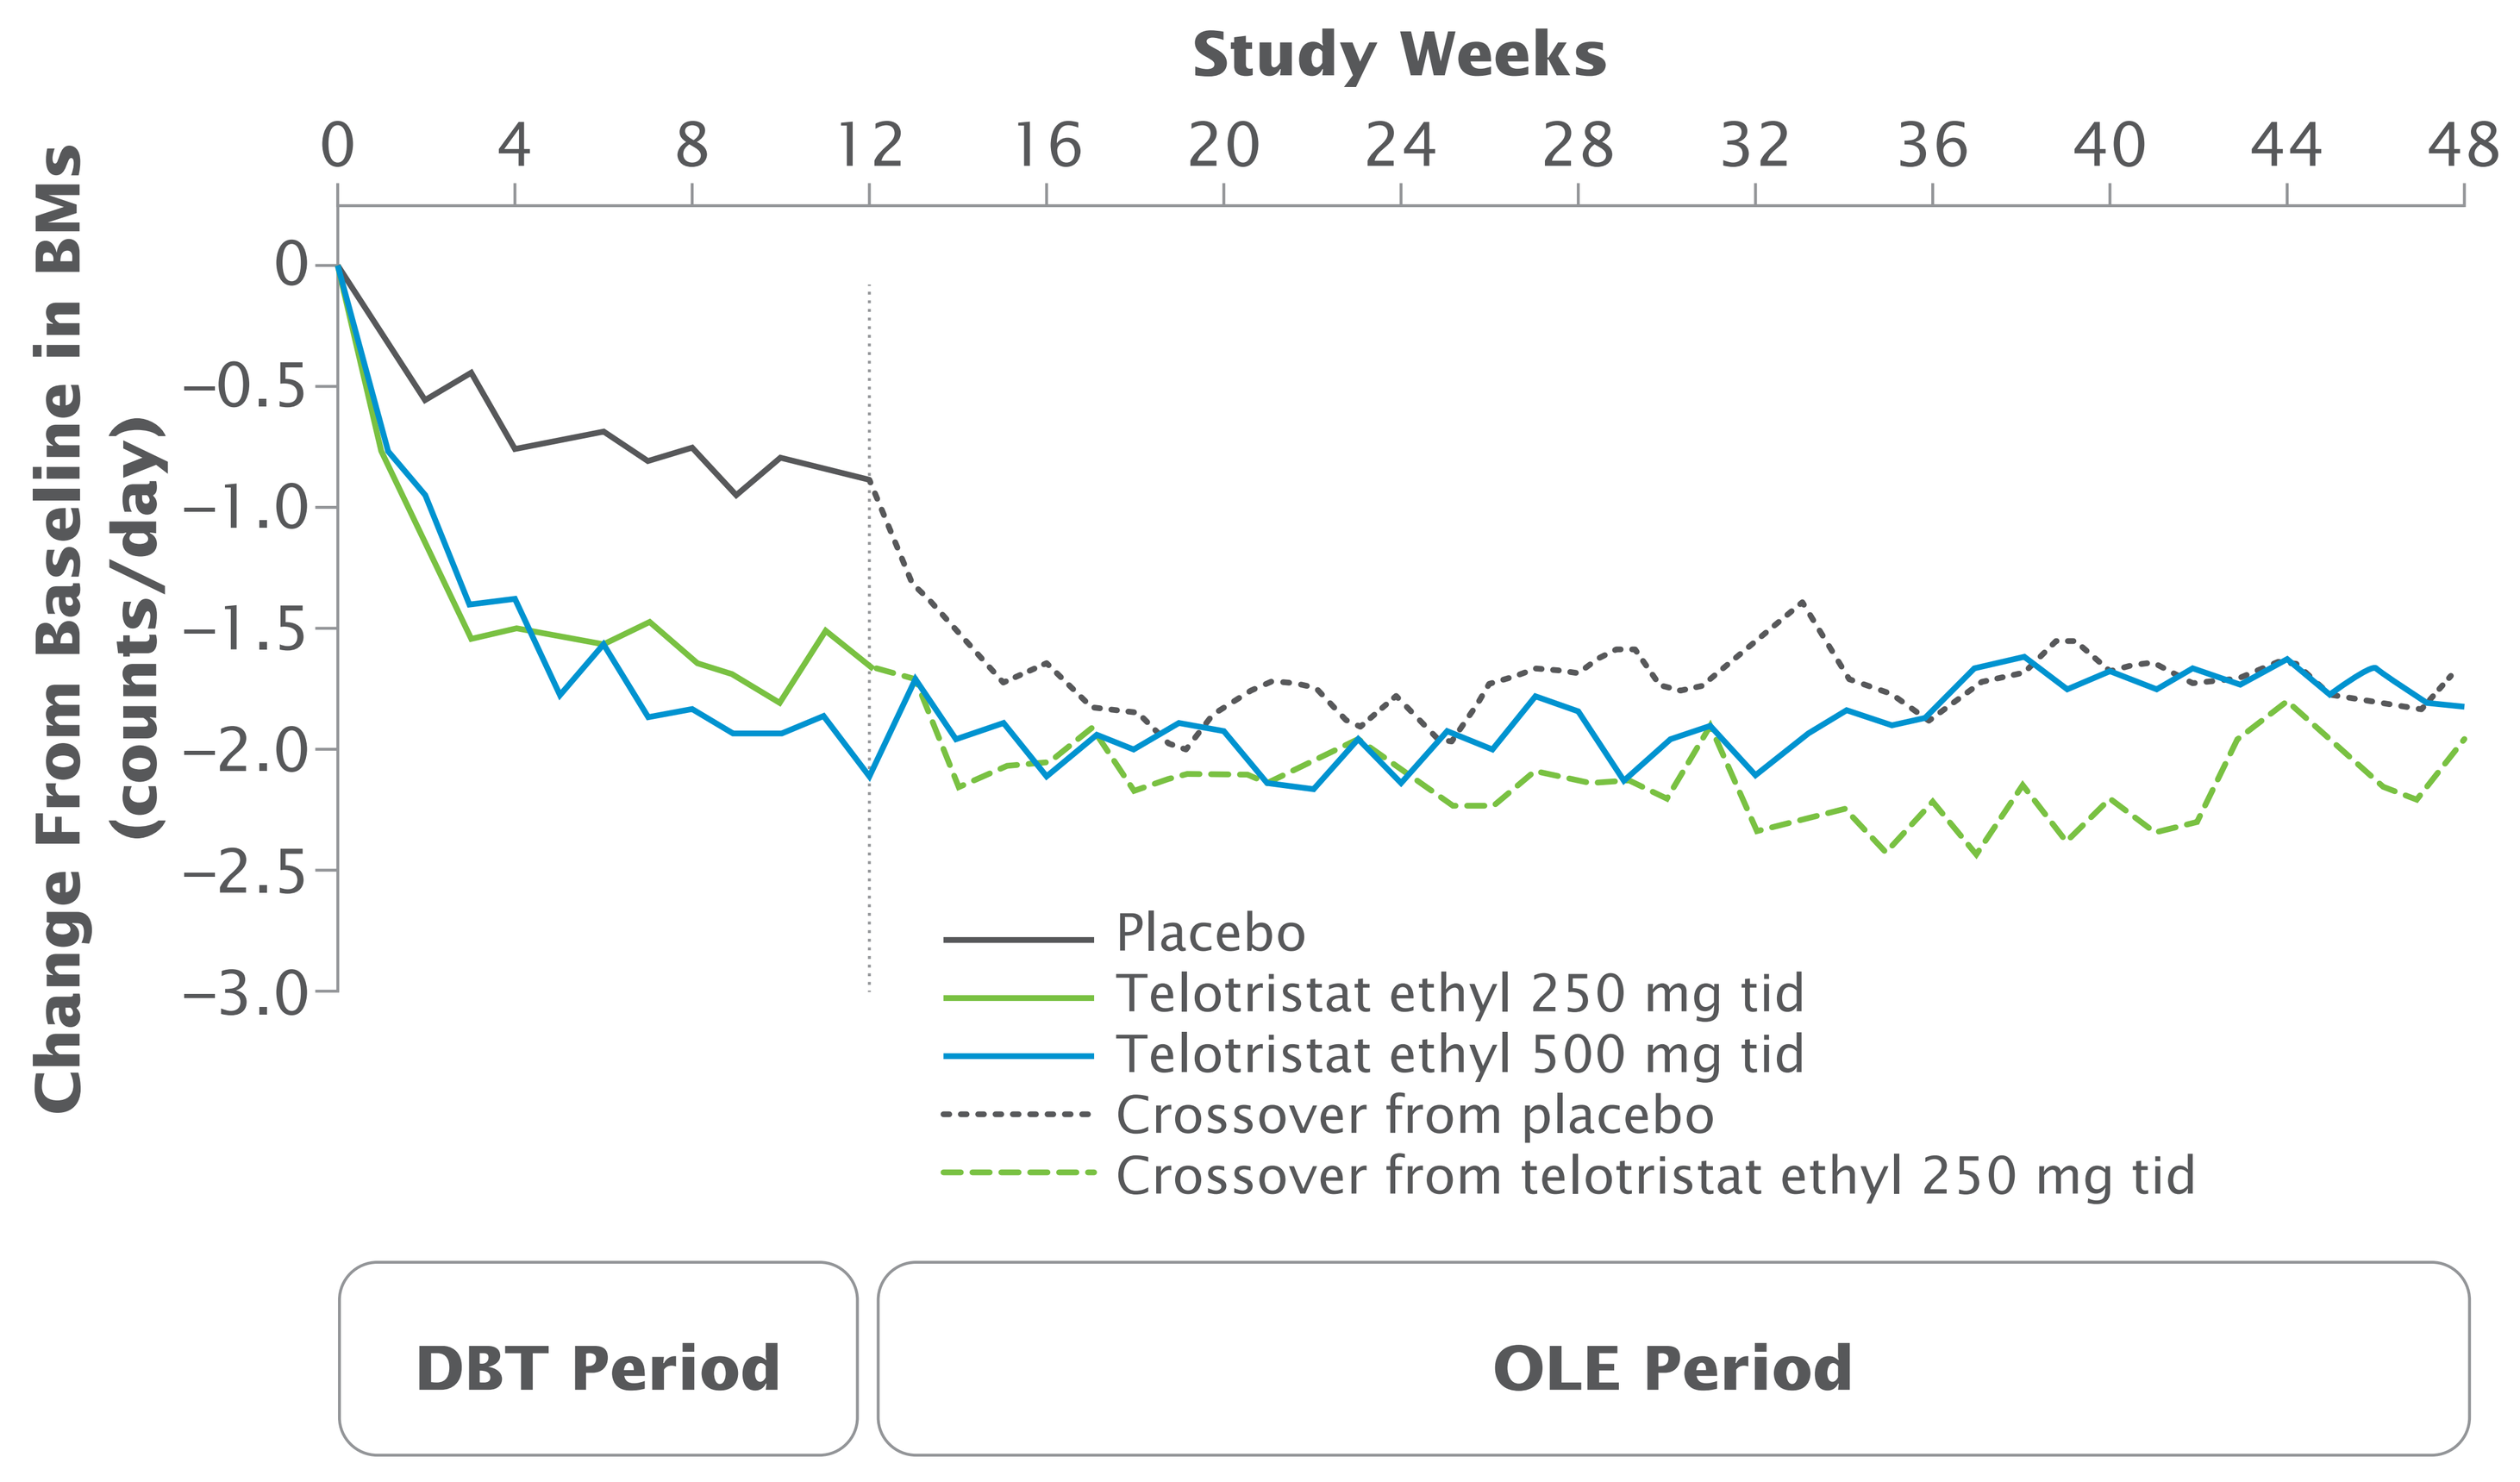
Study Weeks
0
4
8
12
16
20
24
28
32
36
40
44
48
Change From Baseline in BMs (counts/day)
0
−0.5
−1.0
−1.5
−2.0
−2.5
−3.0
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
Crossover from placebo
Crossover from telotristat ethyl 250 mg tid
DBT Period
OLE Period

Color represents treatment group during the DBT period.
BM, bowel movement; DBT, double-blind treatment; OLE, open-label extension; tid, 3 times per day

- Reductions of ~2 BMs/day with telotristat ethyl treatment persisted during the OLE period.

FIGURE 3: Mean Change From Baseline in u5-HIAA Levels (mg/24 hours)

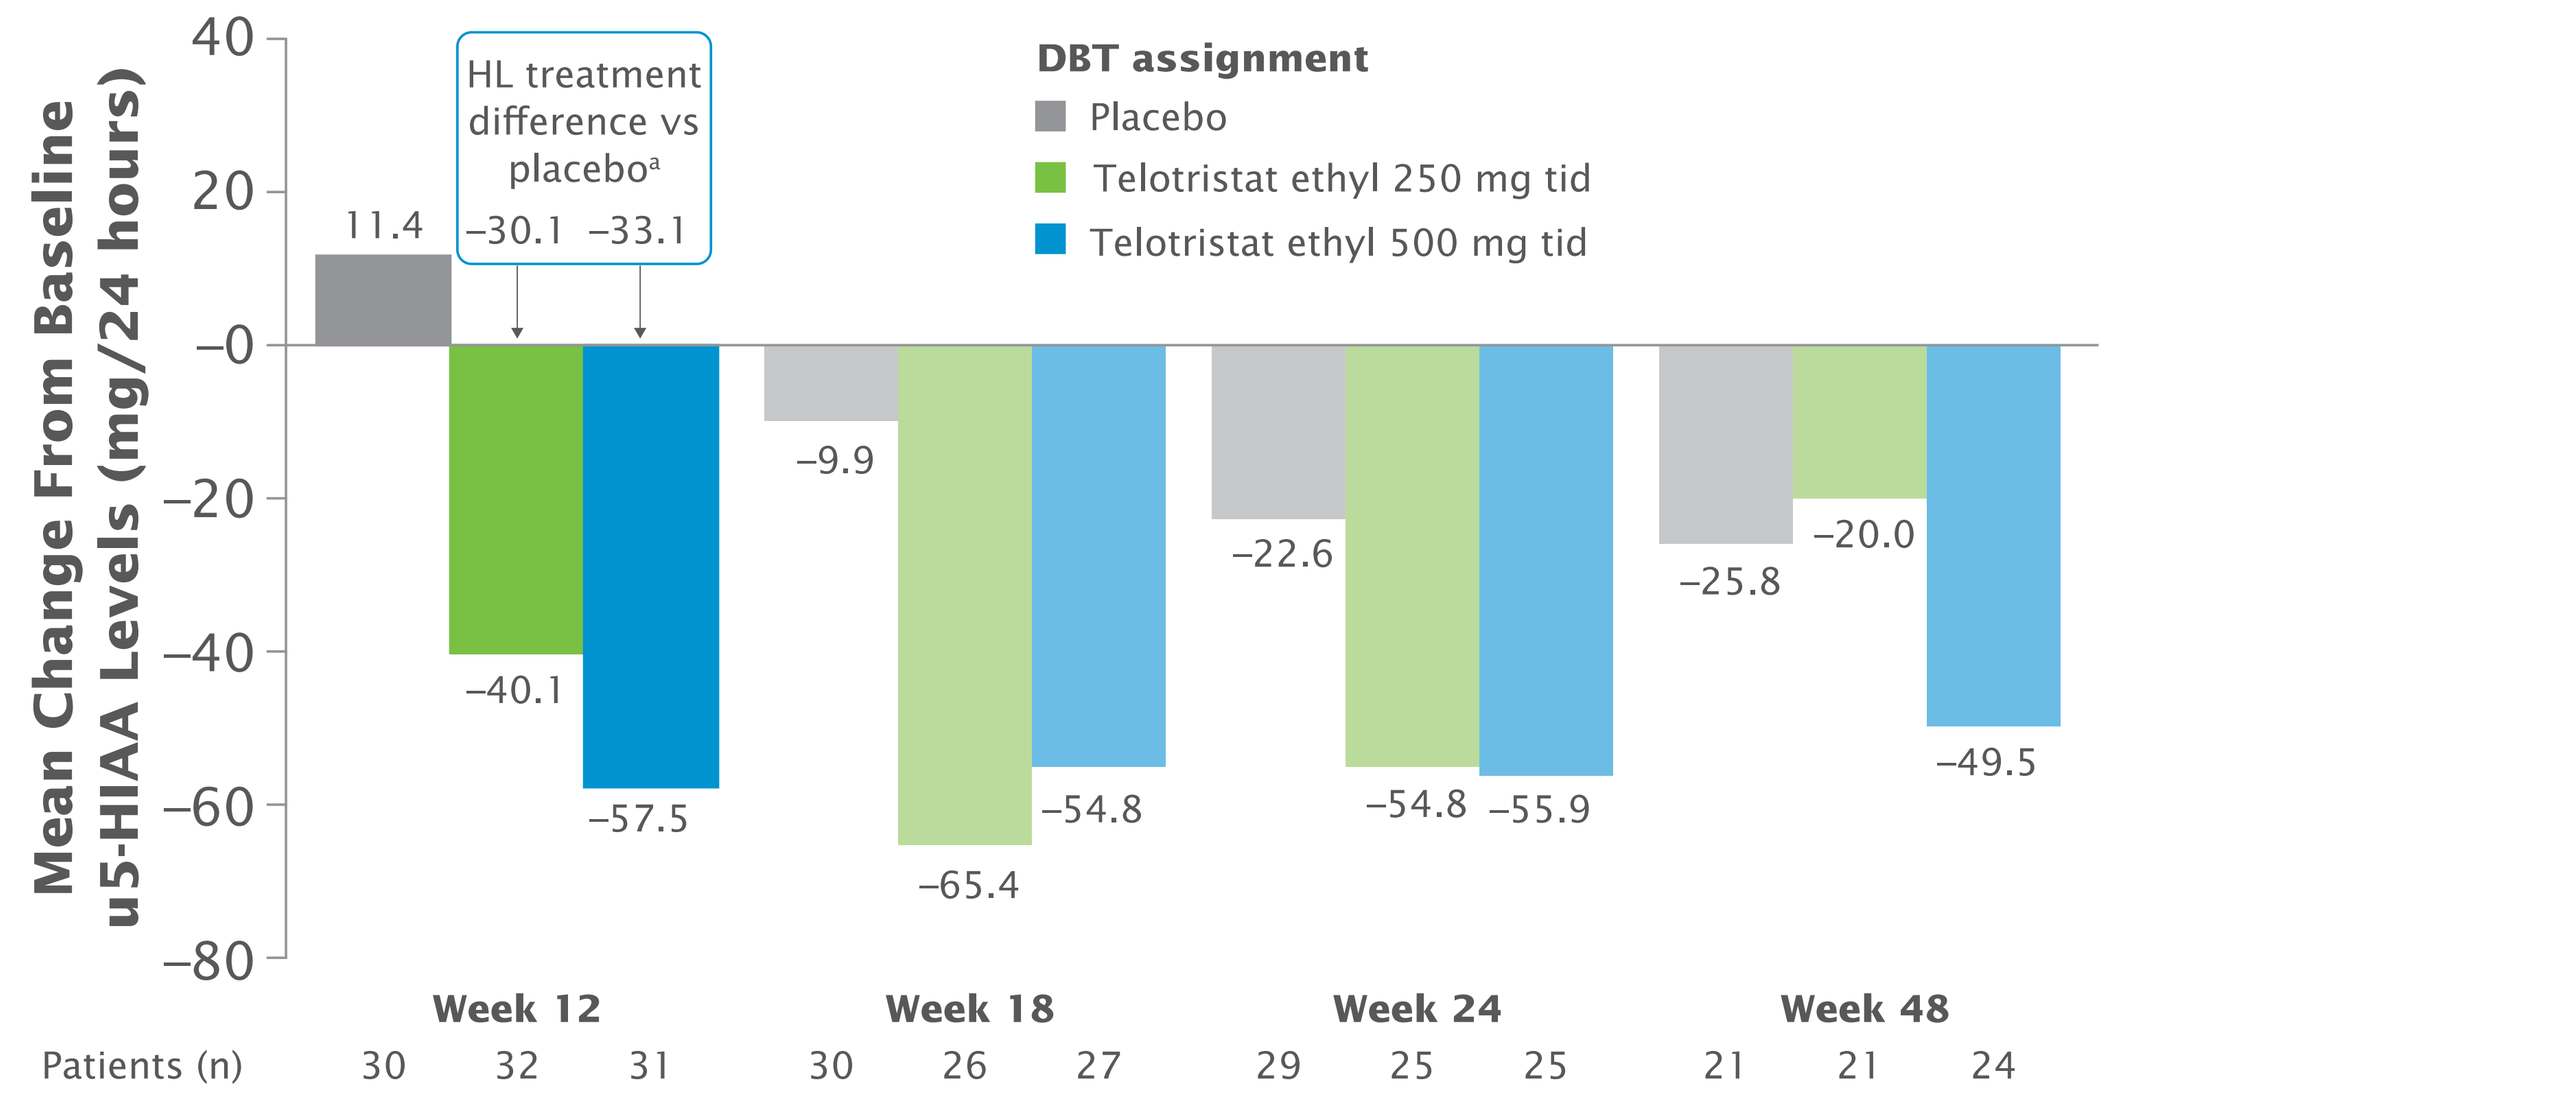
Mean Change From Baseline u5-HIAA Levels (mg/24 hours)
40
20
0
−20
−40
−60
−80
HL treatment difference vs placebo[a]
−30.1 −33.1
DBT assignment
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
11.4
−40.1
−57.5
−9.9
−65.4
−54.8
−22.6
−54.8
−55.9
−25.8
−20.0
−49.5
Week 12
Week 18
Week 24
Week 48
Patients (n) 30 32 31 30 26 27 29 25 25 21 21 24

[a]Hodges–Lehmann (HL) estimation of treatment difference versus placebo, p<0.001 Color represents treatment group during the DBT period. Lighter colors represent treatment during the OLE period (telotristat ethyl 500 mg tid).

DBT, double-blind treatment; tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid

- Reductions in mean u5-HIAA levels persisted during the OLE period.

FIGURE 4: Mean Change From Baseline in EORTC QLQ-C30 Diarrhea Score

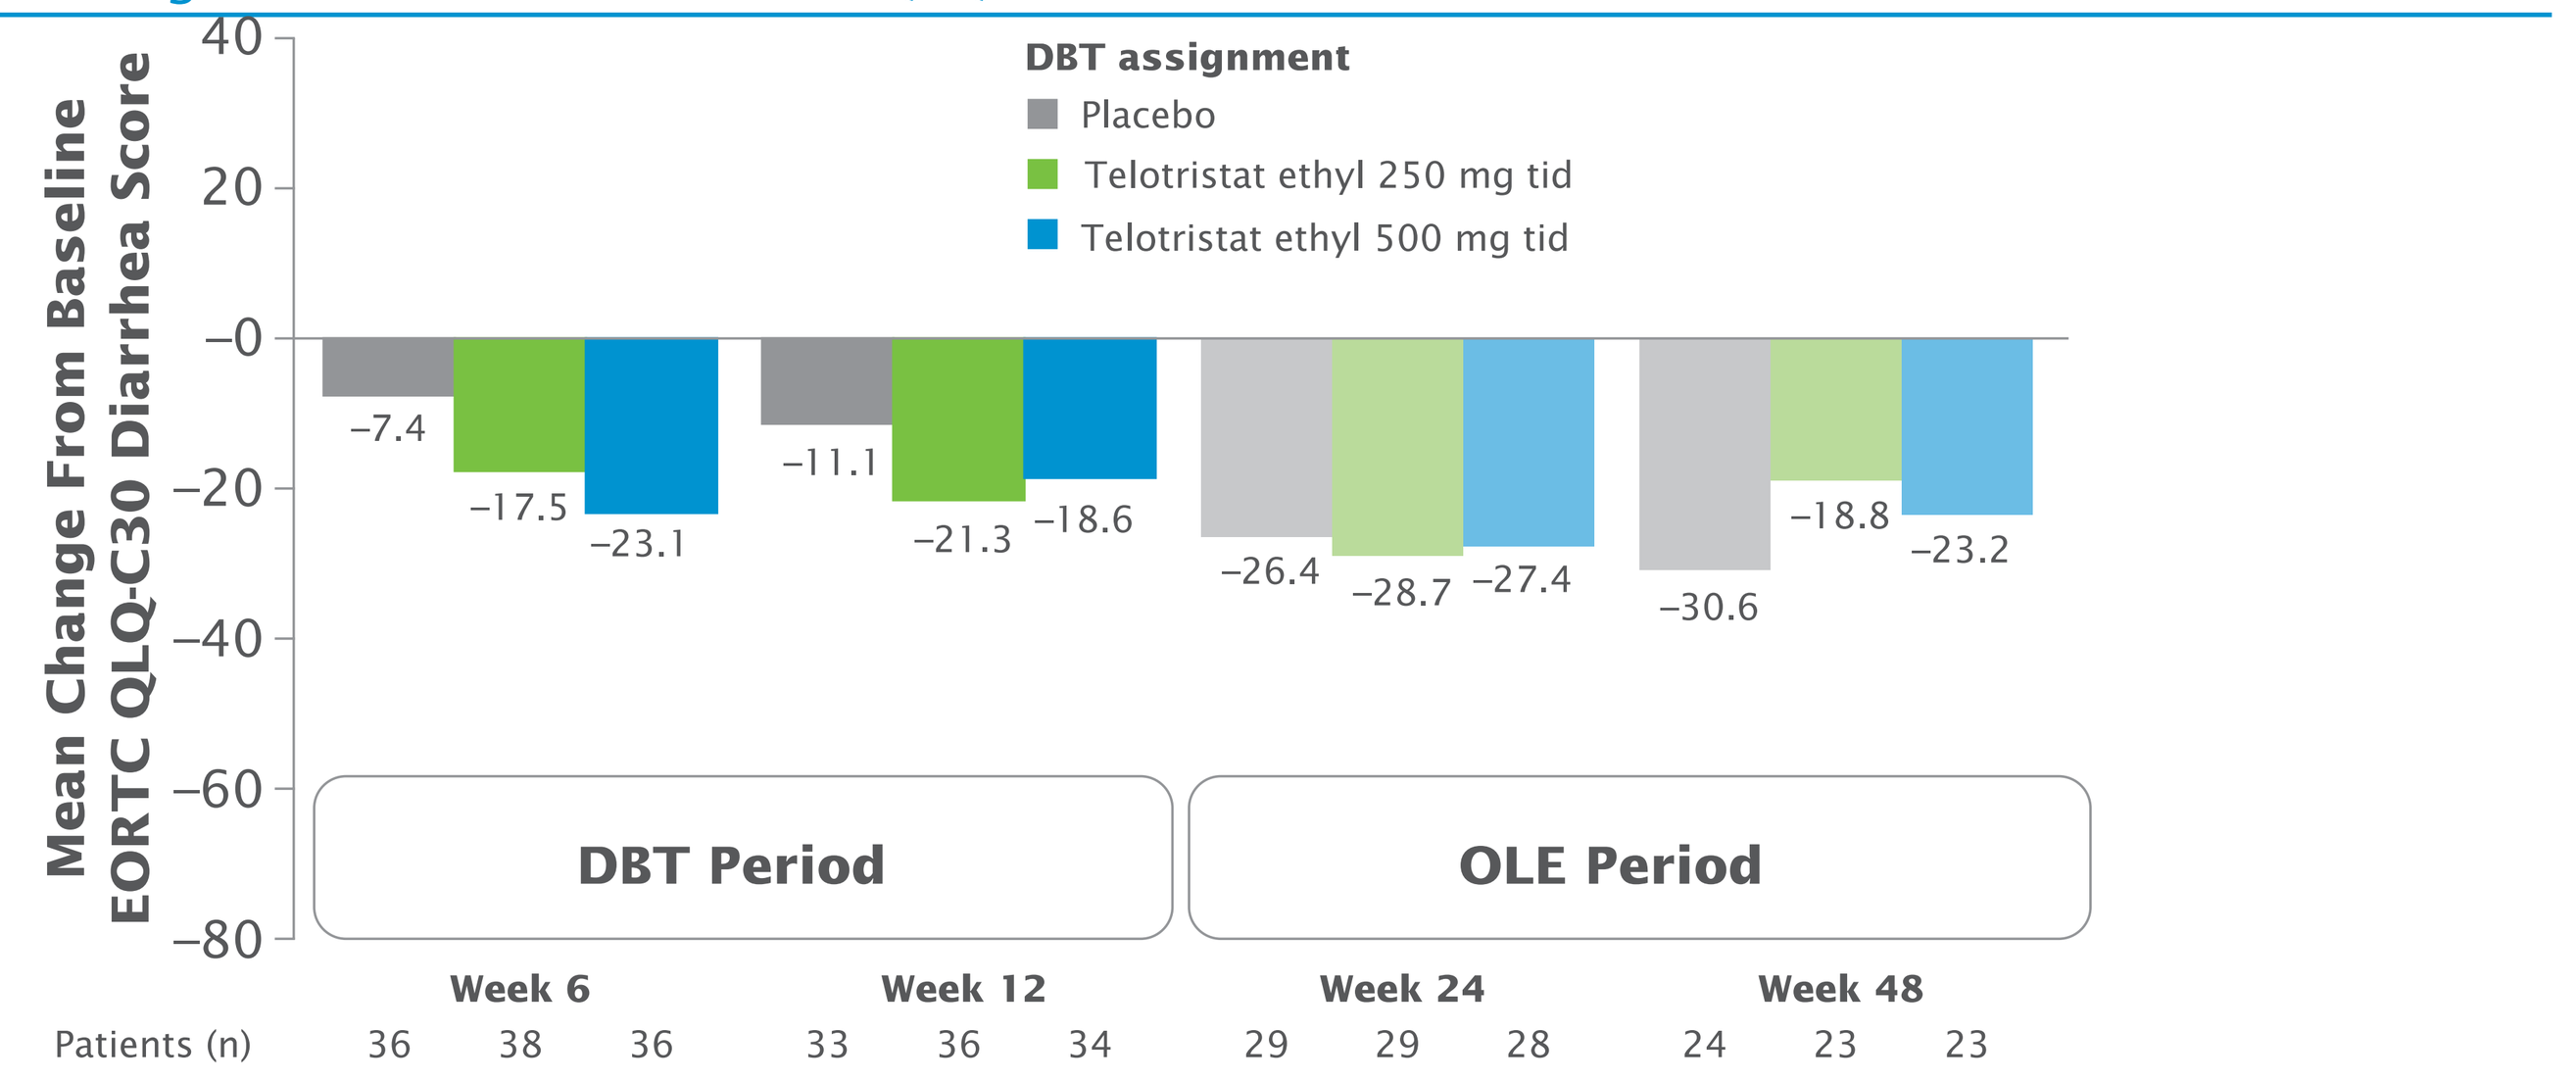
Mean Change From Baseline EORTC QLQ-C30 Diarrhea Score
40
20
0
−20
−40
−60
−80
DBT assignment
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
−7.4
−17.5
−23.1
−11.1
−21.3
−18.6
−26.4
−28.7
−27.4
−30.6
−18.8
−23.2
DBT Period
OLE Period
Week 6
Week 12
Week 24
Week 48
Patients (n) 36 38 36 33 36 34 29 29 28 24 23 23

Color represents treatment group during the DBT period; lighter colors represent treatment during the OLE period (telotristat ethyl 500 mg tid).

DBT, double-blind treatment; EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30; OLE, open-label extension; tid, 3 times per day

- Reductions in EORTC QLQ-C30 diarrhea score persisted during the OLE period.

Safety

- Throughout the OLE period, the overall incidences of TEAEs and incidences of TEAEs leading to study discontinuation were similar to those in the DBT period, despite the longer exposure to telotristat ethyl.
- The incidences of TEAEs in patients who crossed over from placebo to telotristat ethyl (253 incidences in 37/38 patients) were similar to those of patients who remained on telotristat ethyl (631 incidences in 73/77 patients) during the OLE period.

TABLE 2: Summary of TEAEs

Category, n (%)	DBT Placebo (n=45)	DBT Telotristat ethyl 250 mg tid (n=45)	DBT Telotristat ethyl 500 mg tid (n=45)	OLE Telotristat ethyl 500 mg tid (n=115)
Any TEAE	39 (86.7)	37 (82.2)	42 (93.3)	110 (95.7)
Any treatment-related TEAE	12 (26.7)	16 (35.6)	30 (66.7)	58 (50.4)
Any serious TEAE	7 (15.6)	7 (15.6)	8 (17.4)	37 (32.2)
Any treatment-related serious TEAE	1 (2.2)	0	2 (4.4)	8 (7.0)
Study drug discontinuations due to TEAEs	6 (13.3)	3 (6.7)	5 (11.1)	18 (15.7)
TEAEs resulting in death	3 (6.7)	1 (2.2)	1 (2.2)	9 (7.8)

DBT, double-blind treatment; OLE, open-label extension; TEAE, treatment-emergent adverse event; tid, 3 times per day

During the OLE period, the most frequent TEAEs leading to discontinuation (by system organ class) were gastrointestinal disorders (6 patients), general disorders and administration site conditions (3 patients), and 2 patients each for the following TEAEs: infections and infestations, metabolism and nutrition disorders, nervous system disorders, and renal and urinary disorders.

TABLE 3: TEAEs of All Grades Experienced by ≥10% of Patients

System organ class preferred term, n (%)	DBT Placebo (n=45)	DBT Telotristat ethyl 250 mg tid (n=45)	DBT Telotristat ethyl 500 mg tid (n=45)	OLE Telotristat ethyl 500 mg tid (n=115)
Cardiac disorders	5 (11.1)	2 (4.4)	1 (2.2)	14 (12.2)
Gastrointestinal disorders	23 (51.1)	20 (44.4)	25 (55.6)	77 (67.0)
Nausea	5 (11.1)	6 (13.3)	14 (31.1)	27 (23.5)
Abdominal pain	8 (17.8)	5 (11.1)	10 (22.2)	35 (30.4)
Vomiting	4 (8.9)	2 (4.4)	5 (11.1)	16 (13.9)
Abdominal pain upper	0	3 (6.7)	5 (11.1)	13 (11.3)
Abdominal distension	3 (6.7)	2 (4.4)	1 (2.2)	13 (11.3)
General disorders and administration site conditions	18 (40.0)	16 (35.6)	14 (31.1)	50 (43.5)
Fatigue	4 (8.9)	4 (8.9)	7 (15.6)	13 (11.3)
Infections and infestations	6 (13.3)	10 (22.2)	12 (26.7)	38 (33.0)
Injury, poisoning and procedural complications	0	1 (2.2)	1 (2.2)	13 (11.3)
Investigations	8 (17.8)	11 (24.4)	9 (20.0)	35 (30.4)
Metabolism and nutrition disorders	10 (22.2)	12 (26.7)	12 (26.7)	29 (25.2)
Decreased appetite	2 (4.4)	3 (6.7)	7 (15.6)	13 (11.3)
Hypokalemia	3 (6.7)	3 (6.7)	5 (11.1)	8 (7.0)
Musculoskeletal and connective tissue disorders	5 (11.1)	6 (13.3)	11 (24.4)	35 (30.4)
Neoplasms benign, malignant, and unspecified (including cysts and polyps)	1 (2.2)	1 (2.2)	2 (4.4)	15 (13.0)
Nervous system disorders	11 (24.4)	8 (17.8)	15 (33.3)	32 (27.8)
Headache	2 (4.4)	5 (11.1)	5 (11.1)	12 (10.4)
Psychiatric disorders	5 (11.1)	6 (13.3)	11 (24.4)	36 (31.3)
Depression	3 (6.7)	2 (4.4)	7 (15.6)	10 (8.7)
Renal and urinary disorders	1 (2.2)	4 (8.9)	3 (6.7)	12 (10.4)
Respiratory, thoracic, and mediastinal disorders	3 (6.7)	5 (11.1)	12 (26.7)	15 (13.0)
Dyspnea	0	2 (4.4)	5 (11.1)	7 (6.1)
Skin and subcutaneous tissue disorders	6 (13.3)	6 (13.3)	4 (8.9)	17 (14.8)
Vascular disorders	2 (4.4)	7 (15.6)	6 (13.3)	18 (15.7)

DBT, double-blind treatment; OLE, open-label extension; TEAE, treatment-emergent adverse event; tid, 3 times per day

Summary and Conclusions

- Statistically significant reductions in BM frequency were observed at Week 12 for both telotristat ethyl 250 mg tid and telotristat ethyl 500 mg tid, and reductions persisted through Week 48.
- Statistically significant reductions in u5-HIAA levels for both telotristat ethyl 250 mg tid and telotristat ethyl 500 mg tid at Week 12 were sustained through Week 48.
- Improvements in EORTC QLQ-C30 diarrhea scores observed with telotristat ethyl at Week 12 were maintained through Week 48.
- Safety data from the 48-week study suggest that telotristat ethyl was well tolerated, with few discontinuations due to adverse events.
- There was no notable increase in the incidence of TEAEs in patients who crossed over from placebo to telotristat ethyl, and TEAE incidence during the OLE period was similar to that in the DBT period.

REFERENCES

1. Mamikunian G, et al. *Neuroendocrine Tumors - A Comprehensive Guide to Diagnosis and Management*. Fourth Edition. Inglewood, CA: Inter Science Institute; 2009. **2.** National Cancer Institute. Gastrointestinal Carcinoid Tumors Treatment (PDQ®)–Health Professional Version. 2015. **3.** Mocellin S, Nitti D. *Ann Oncol*. 2013;24(12):3040-3044. **4.** Davar J, et al. *J Am Coll Cardiol*. 2017;69(10):1288-1304. **5.** Boudreaux JP, et al. *Pancreas*. 2010;39(6):753-766. **6.** Pavel M, et al. *Neuroendocrinology*. 2017; doi: 10.1159/000471880. **7.** National Comprehensive Cancer Network. Neuroendocrine Tumors (Version 3.2017). 2017. **8.** Xermelo® (telotristat ethyl) US Highlights of Prescribing Information. Lexicon Pharmaceuticals, Inc. February 2017. **9.** European Commission. Commission Implementing Decision of 18.9.2017 granting marketing authorisation under Regulation (EC) No 726/2004 of the European Parliament and of the Council for "Xermelo - telotristat", an orphan medicinal product for human use. 2017. http://ec.europa.eu/health/documents/community-register/2017/20170918138652/dec_138652_en.pdf. Accessed October 2, 2017. **10.** Kulke MH, et al. *J Clin Oncol*. 2017;35(1):14-23. **11.** Tellez MR, et al. *Pancreas*. 2013;42(3):405-410.

ACKNOWLEDGMENTS

We thank the patients and Investigators for participating in the study. We thank James Banigan, PhD (Chameleon Communications International with funding from Lexicon Pharmaceuticals, Inc.) for editorial assistance in the preparation of this report. The TELESTAR study was sponsored by Lexicon Pharmaceuticals, Inc.

DISCLOSURES

DH: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, Novartis Pharmaceuticals, and Pfizer, Inc. MHK: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, and Novartis Pharmaceuticals. MEC: Lexicon Pharmaceuticals, Inc., Novartis Pharmaceuticals, and Ipsen Biopharmaceuticals. LBA: Lexicon Pharmaceuticals, Inc. EB: Ipsen Bioscience, Lexicon Pharmaceuticals, Inc., and Novartis Pharmaceuticals. PLK: Merck & Co., Genentech, Advanced Accelerator Applications, Lexicon Pharmaceuticals, Inc., Oxygen, Esanex, Dicerna Pharmaceuticals, Ipsen Biopharmaceuticals, and Novartis Pharmaceuticals. JSD: Lexicon Pharmaceuticals, Inc. PL, PB, and SJ: employees of Lexicon Pharmaceuticals, Inc. MP: Ipsen Bioscience, Novartis Pharmaceuticals, Lexicon Pharmaceuticals, Inc., and Pfizer, Inc. KO, RRPW, EG, and JWV: nothing to disclose.

To view this poster online, snap a photo of this QR code with your smart phone (QR reader required).

Presented at the NANETS 2017 Annual Symposium; October 19–21, 2017; Philadelphia, PA, USA