Telotristat Ethyl in Carcinoid Syndrome: Safety and Efficacy Results of an Open-label Extension of the TELECAST Phase 3 Clinical Trial

Poster 213

Marianne Pavel[1,2], Marta Benavent[3], Petros Perros[4], Raj Srirajaskanthan[5], Richard R.P. Warner[6], Matthew H. Kulke[7], Lowell B. Anthony[8], Pamela L. Kunz[9], Dieter Hörsch[10], Pablo Lapuerta[11], Rosanna Fleming[11], David J. Gross[12]

[1]Charité–Universitätsmedizin, Berlin, Germany; [2]Current affiliation: Friedrich-Alexander-Universität Erlangen-Nürnberg, Erlangen, Germany; [3]Hospital Universitario Virgen del Rocío, Instituto de Biomedicina de Sevilla, Sevilla, Spain; [4]Royal Victoria Infirmary, Newcastle Upon Tyne, UK; [5]Kings College Hospital, Institute of Liver Studies, London, UK; [6]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [7]Dana-Farber Cancer Institute, Boston, MA, USA; [8]University of Kentucky, Lexington, KY, USA; [9]Stanford University, Palo Alto, CA, USA; [10]Zentralklinik Bad Berka, Bad Berka, Germany; [11]Lexicon Pharmaceuticals, Inc., The Woodlands, TX, USA; [12]Hadassah-Hebrew University Medical Center, Jerusalem, Israel

Background

- Telotristat ethyl is a novel, small-molecule tryptophan hydroxylase inhibitor that decreases urinary 5-hydroxyindoleacetic acid (u5-HIAA) levels and bowel movement (BM) frequency in patients with carcinoid syndrome (CS) and is approved by the US Food and Drug Administration and European Commission for the treatment of CS diarrhea in combination with somatostatin analog (SSA) therapy in adults whose symptoms are inadequately controlled by SSA therapy.[1–3]
- The Phase 3 TELECAST study, the companion to the Phase 3 TELESTAR study,[4] included patients with CS with <4 BMs per day and patients not taking SSAs; it met its primary safety and efficacy endpoints, and the results of the Open-label Extension (OLE) period are presented.[5]

Objective

To provide additional OLE data on the effect of telotristat ethyl, compared with placebo, on the incidence of treatment-emergent adverse events (TEAEs) and on u5-HIAA levels.

Methods

Study treatment and design

Telotristat Etiprate for Carcinoid Syndrome Therapy (TELECAST) is a Phase 3, randomized, multicenter, parallel-group, double-blind, placebo-controlled study (ClinicalTrials.gov identifier: NCT02063659,[6] **Figure 1**).

FIGURE 1: TELECAST Study Design

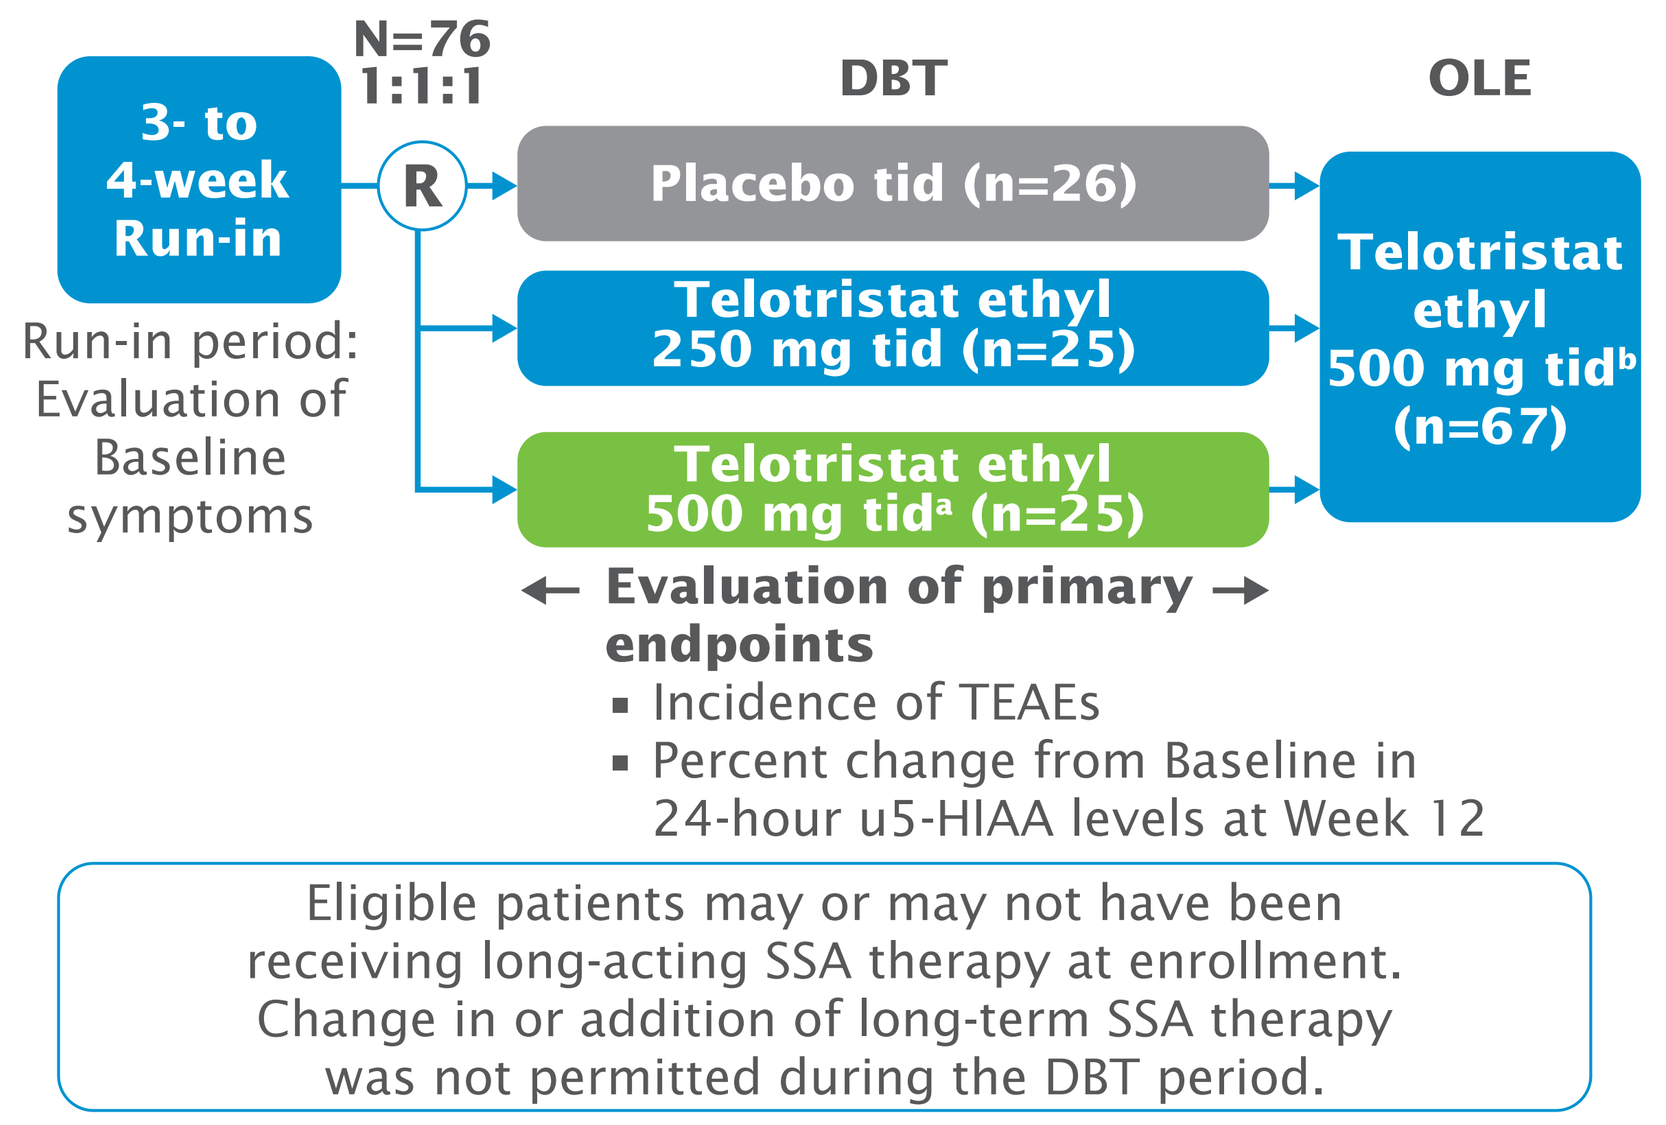
3- to 4-week Run-in
Run-in period: Evaluation of Baseline symptoms
N=76
1:1:1
R
DBT
OLE
Placebo tid (n=26)
Telotristat ethyl 250 mg tid (n=25)
Telotristat ethyl 500 mg tid[a] (n=25)
Telotristat ethyl 500 mg tid[b] (n=67)
Evaluation of primary endpoints
Incidence of TEAEs
Percent change from Baseline in 24-hour u5-HIAA levels at Week 12
Eligible patients may or may not have been receiving long-acting SSA therapy at enrollment. Change in or addition of long-term SSA therapy was not permitted during the DBT period.

[a]Including a blinded titration step of 1 week of telotristat ethyl 250 mg tid and [b]for patients on placebo during the DBT
DBT, Double-blind Treatment; OLE, Open-label Extension; R, randomization; SSA, somatostatin analog; TEAEs, treatment-emergent adverse events; tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid

Key inclusion criteria

Patients must have been ≥18 years of age with histopathologically confirmed, well-differentiated metastatic neuroendocrine tumors and a documented history of CS. Patients on SSA therapy at enrollment had <4 BMs per day AND at least 1 sign/symptom, including daily stool form/consistency ≥5 on the Bristol Stool Form Scale,[7] average daily flushing episode frequency of ≥2, nausea present ≥20% of days, or u5-HIAA > upper limit of normal (ULN). Patients not taking stable-dose SSAs at enrollment had ≥1 of the above-described signs/symptoms or ≥4 BMs per day.

Key exclusion criteria

Patients treated with tumor-directed therapy <12 weeks prior to the Run-in period or with presence of enteric infection, diarrhea attributed to any conditions other than CS, or a Karnofsky Performance Status ≤60%.

Efficacy and safety assessments

Primary safety and efficacy endpoints were the incidence of TEAEs and percentage change from Baseline in 24-hour u5-HIAA levels at Week 12.[4]

Results

- Baseline demographics were similar between groups (**Table 1**).
- The majority of patients completed the DBT period, 22 (88%) in each of the telotristat ethyl groups and 24 (92.3%) in the placebo group.
 - In total, 67 (88.2%) patients continued into the OLE period (24, 22, and 21 patients from the placebo and 250 mg tid and 500 mg tid telotristat ethyl groups, respectively).
- Patients entering the OLE period received a mean of 30.31 additional weeks of telotristat ethyl exposure (median 36 weeks) over Weeks 13–48, compared with a mean of 11.45 weeks (median 12 weeks) during the DBT period.
- During the OLE period, 20 patients discontinued treatment: 9 due to withdrawal of consent, 7 due to adverse events (AEs), and 1 each due to lack of efficacy, physician decision, resolution of CS, or Week 48 visit mistakenly not done.

TABLE 1: Demographic and Baseline Characteristics

	DBT			OLE
	Placebo	Telotristat ethyl		
Patient characteristics	tid (n=26)	250 mg tid (n=25)	500 mg tid (n=25)	500 mg tid (n=67)
Mean age, years (SD)	62.2 (10.3)	63.6 (12.6)	62.7 (12.0)	63.3 (11.4)
Male, n (%)	13 (50)	14 (56)	15 (60)	39 (58.2)
u5-HIAA at randomization, n (%)[a]				
≤ULN	9 (34.6)	5 (20.0)	8 (32.0)	19 (28.4)
>ULN	17 (65.4)	18 (72.0)	17 (68.0)	46 (68.7)
Unknown	0	2 (8.0)	0	2 (3.0)
Mean u5-HIAA at Baseline, mg/day (median)	82.0 (31.1)	86.3 (84.0)	66.0 (40.0)	78.2 (43.1)
Mean BM frequency, counts/day (SD)[b]	2.2 (0.7)	2.5 (1.2)	2.8 (1.6)	2.4 (1.1)
Mean weekly stool consistency score (SD)[b]	5.0 (0.9)	5.1 (0.8)	5.3 (0.8)	5.1 (0.9)
Mean number of flushing episodes/day (SD)[b]	3.7 (4.1)	2.7 (3.7)	1.8 (2.2)	2.6 (3.1)
Mean weekly abdominal pain rating (SD)[b]	1.7 (1.7)	1.2 (1.5)	1.8 (1.7)	1.5 (1.7)
SSA therapy at study entry, n (%)				
Octreotide	12 (46.2)	17 (68.0)	16 (64.0)	40 (59.7)
Lanreotide	14 (53.8)	5 (20.0)	3 (12.0)	20 (29.9)
None[c]	0	3 (12.0)	6 (24.0)	7 (10.5)

[a]Reference range: normal u5-HIAA 0–15 mg per 24 hours[8]
[b]Baseline diary endpoints for placebo (n=25) and OLE (n=66)
[c]Includes patients listed as "unknown" or "not applicable" for patient not receiving SSA therapy at Baseline
BM, bowel movement; DBT, Double-blind Treatment; OLE, Open-label Extension; SD, standard deviation, SSA, somatostatin analog; tid, 3 times per day; u5-HIAA, urinary 5-hydroxyindoleacetic acid; ULN, upper limit of normal

Primary safety endpoint

- For the OLE period, the types of safety-related events, incidences of overall TEAEs, and incidences of TEAEs leading to study discontinuation were generally similar to those in the DBT period, considering the increase in exposure to telotristat ethyl (**Table 2**).

TABLE 2: Incidence of TEAEs

	DBT			OLE
	Placebo	Telotristat ethyl		
Category, n (%)	tid (n=26)	250 mg tid (n=25)	500 mg tid (n=25)	500 mg tid (n=67)
Any TEAE	21 (80.8)	25 (100)	22 (88.0)	61 (91.0)
Treatment-related TEAEs	7 (26.9)	10 (40)	11 (44)	28 (41.8)
Serious TEAEs	5 (19.2)	1 (4.0)	3 (12.0)	17 (25.4)
Serious treatment-related TEAEs	0	0	0	2 (3.0)
Study discontinuation due to TEAEs	1 (3.8)	2 (8.0)	0	7 (10.4)
TEAE resulting in death	0	0	0	0

DBT, Double-blind Treatment; OLE, Open-label Extension; TEAE, treatment-emergent adverse event; tid, 3 times per day

- The most common TEAEs were reported for the class of Gastrointestinal disorders (**Table 3**).
- The incidences of TEAEs in patients who remained on telotristat ethyl and those who crossed over from placebo to telotristat ethyl in the OLE period were similar.

TABLE 3: TEAEs Occurring in ≥10% of Patients in Any Group

	DBT			OLE
	Placebo	Telotristat ethyl		
Preferred term, n (%)	tid (n=26)	250 mg tid (n=25)	500 mg tid (n=25)	500 mg tid (n=67)
Nausea	4 (15.4)	3 (12.0)	2 (8.0)	14 (20.9)
Abdominal pain	4 (15.4)	8 (32.0)	1 (4.0)	12 (17.9)
Diarrhea	5 (19.2)	4 (16.0)	2 (8.0)	9 (13.4)
Flushing	2 (7.7)	3 (12.0)	0	9 (13.4)
Constipation	1 (3.8)	4 (16.0)	3 (12.0)	8 (11.9)
Depression	0	0	1 (4.0)	8 (11.9)
Vomiting	1 (3.8)	1 (4.0)	1 (4.0)	7 (10.4)
Asthenia	2 (7.7)	1 (4.0)	0	7 (10.4)
Fatigue	2 (7.7)	3 (12.0)	2 (8.0)	7 (10.4)
Pyrexia	0	3 (12.0)	0	6 (9.0)
Abdominal pain upper	3 (11.5)	1 (4.0)	2 (8.0)	5 (7.5)
Abdominal distension	0	3 (12.0)	1 (4.0)	4 (6.0)
Urinary tract infection	0	3 (12.0)	0	4 (6.0)
Dizziness	3 (11.5)	0	2 (8.0)	3 (4.5)

DBT, Double-blind Treatment; OLE, Open-label Extension; TEAE, treatment-emergent adverse event; tid, 3 times per day

- The incidence of AEs of special interest (AESIs) did not increase during the OLE period when accounting for treatment duration (**Table 4**).
- All depression-related AESIs were mild or moderate in severity. During the OLE period, 1 patient required modification of an existing antidepressant regimen.
- AESIs related to hepatic enzyme abnormalities were rated on the Drug-Induced Liver Injury Network scale (DILIN) as mild (1+), and over half resolved.

TABLE 4: Adverse Events of Interest

		DBT			OLE
		Placebo	Telotristat ethyl		
Category, n (%)		tid (n=26)	250 mg tid (n=25)	500 mg tid (n=25)	500 mg tid (n=67)
Abdominal pain	Mild	1 (3.8)	3 (12.0)	0	5 (7.5)
	Moderate	3 (11.5)	3 (12.0)	0	6 (9.0)
	Severe	0	2 (8.0)	1 (4.0)	1 (1.5)
Diarrhea	Mild	2 (7.7)	2 (8.0)	1 (4.0)	3 (4.5)
	Moderate	3 (11.5)	2 (8.0)	1 (4.0)	4 (6.0)
	Severe	0	0	0	2 (3.0)
Nausea	Mild	4 (15.4)	2 (8.0)	2 (8.0)	8 (11.9)
	Moderate	0	1 (4.0)	0	5 (7.5)
	Severe	0	0	0	1 (1.5)
Constipation	Mild	0	3 (12.0)	3 (12.0)	5 (7.5)
	Moderate	1 (3.8)	1 (4.0)	0	2 (3.0)
	Severe	0	0	0	1 (1.5)
Depressed mood	Mild	2 (7.7)	1 (4.0)	0	2 (3.0)
	Moderate	0	0	0	0
	Severe	0	0	0	0
Decreased interest	Mild	1 (3.8)	0	0	NR
	Moderate	0	0	0	NR
	Severe	0	0	0	NR
Depression	Mild	0	0	0	4 (6.0)
	Moderate	0	0	1 (4.0)	4 (6.0)
	Severe	0	0	0	0
Elevated GGT	Mild	0	1 (4.0)	0	1 (1.5)
	Moderate	0	0	1 (4.0)	1 (1.5)
	Severe	0	0	0	2 (3.0)
Elevated ALT	Mild	0	1 (4.0)	0	2 (3.0)
	Moderate	0	0	0	0
	Severe	0	0	0	1 (1.5)
Elevated AST	Mild	NR	NR	NR	2 (3.0)
	Moderate	NR	NR	NR	0
	Severe	NR	NR	NR	1 (1.5)

Color indicates AE category (gastrointestinal, gray; depression, blue; hepatic enzymes, green).
ALT, alanine aminotransferase; AST, aspartate aminotransferase; DBT, Double-blind Treatment; GGT, gamma-glutamyl transferase; NR, not reported; OLE, Open-label Extension; TEAE, treatment-emergent adverse event; tid, 3 times per day

Primary efficacy endpoint

Significant reductions in u5-HIAA levels were observed in both telotristat ethyl groups at Week 12. Reductions were generally sustained over the OLE period (**Figure 2**).

FIGURE 2: Percent Change From Baseline in u5-HIAA

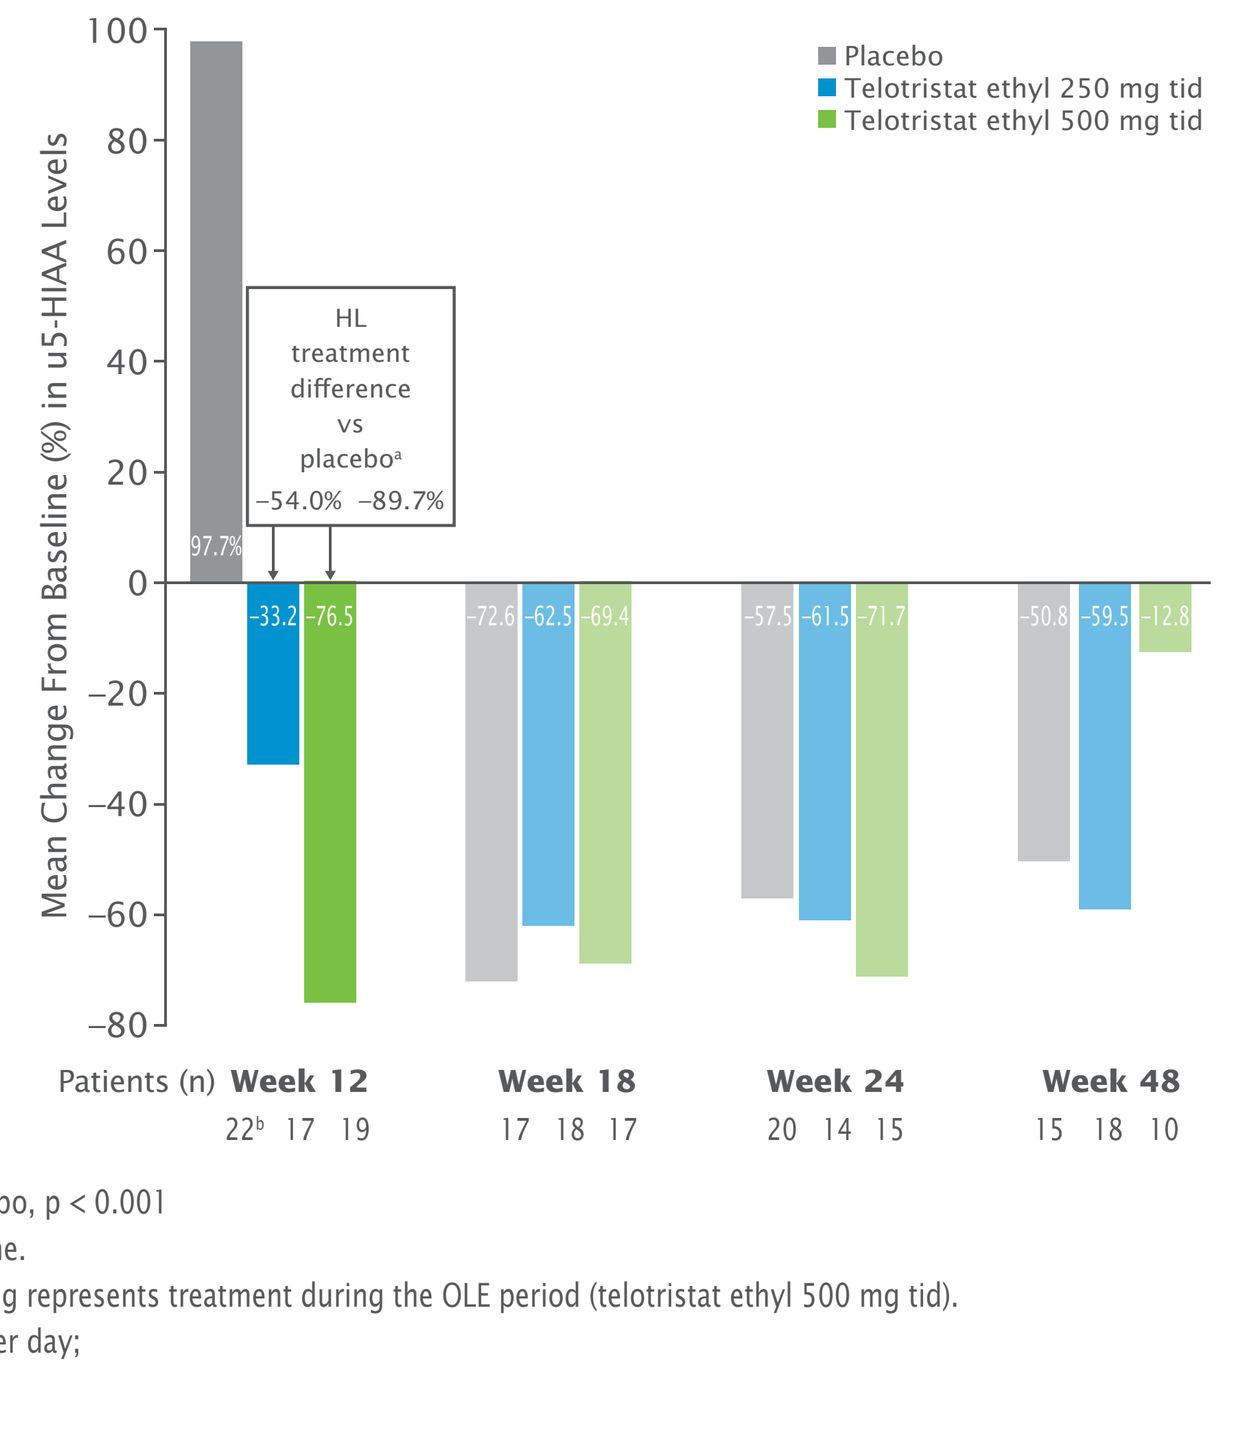
Mean Change From Baseline (%) in u5-HIAA Levels
100
80
60
40
20
0
−20
−40
−60
−80
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid
97.7%
HL treatment difference vs placebo[a]
−54.0% −89.7%
−33.2 −76.5
−72.6 −62.5 −69.4
−57.5 −61.5 −71.7
−50.8 −59.5 −12.8
Patients (n) Week 12 Week 18 Week 24 Week 48
22[b] 17 19
17 18 17
20 14 15
15 18 10

[a]Hodges–Lehmann (HL) estimation of treatment difference versus placebo, p < 0.001
[b]Data include 1 patient who experienced a 1864% increase from Baseline.
Color represents treatment group during the DBT period; lighter shading represents treatment during the OLE period (telotristat ethyl 500 mg tid).
DBT, Double-blind Treatment; OLE, Open-label Extension; tid, 3 times per day;
u5-HIAA, urinary 5-hydroxyindoleacetic acid

Secondary efficacy endpoints

- Over the DBT period, mean BMs per day were significantly reduced from Baseline in the 250 mg tid (–0.452, p = 0.004) and 500 mg tid (–0.535, p < 0.001) telotristat ethyl groups, compared with placebo.
 - Reduction in mean BMs per day continued through the OLE period, with mean decreases of –0.584 at Week 24 (n=56) and –0.473 at Week 48 (n=38).
- Improvements from Baseline for European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30 (EORTC QLC-C30)[9] diarrhea scores were observed at Week 12 and continued for all treatment groups; however, differences versus placebo were not statistically significant (**Figure 3**).

FIGURE 3: Changes From Baseline in EORTC QLQ-C30 Diarrhea Scores

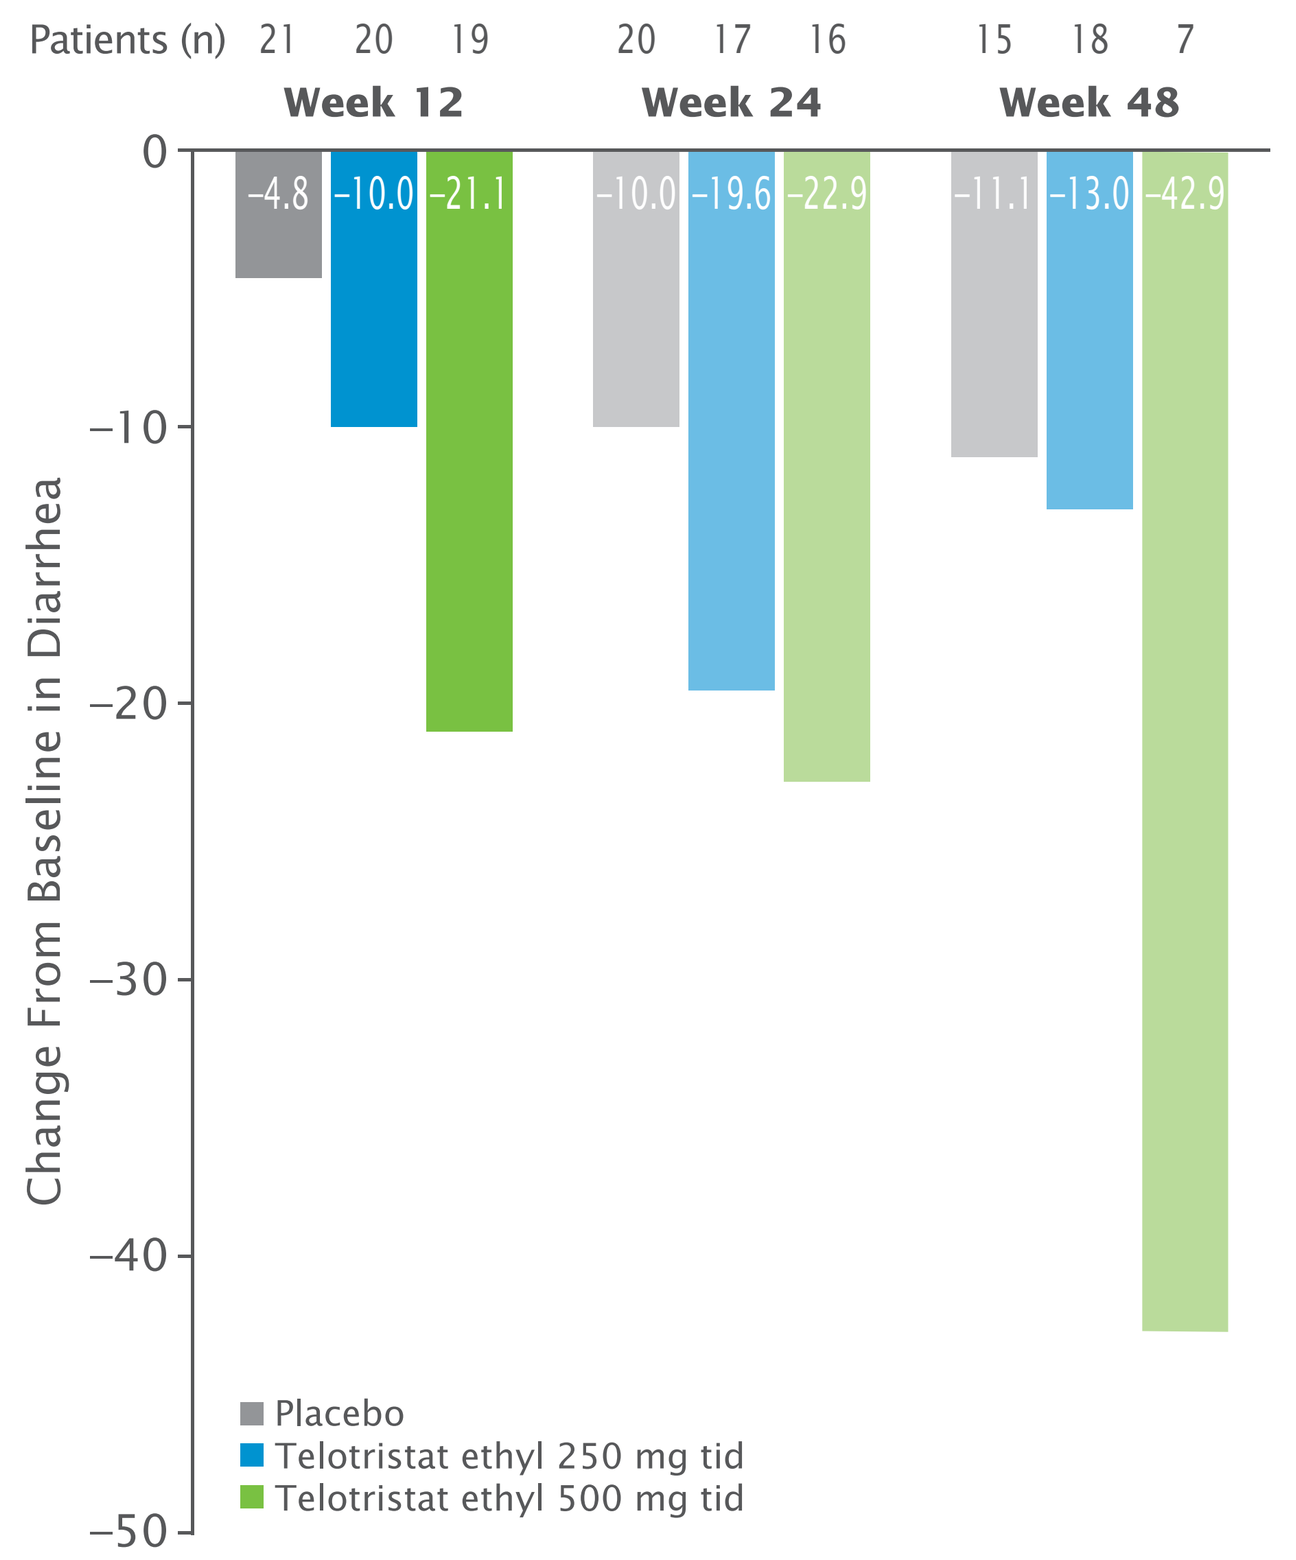
Patients (n) 21 20 19
20 17 16
15 18 7
Week 12 Week 24 Week 48
Change From Baseline in Diarrhea
0
−10
−20
−30
−40
−50
−4.8 −10.0 −21.1
−10.0 −19.6 −22.9
−11.1 −13.0 −42.9
Placebo
Telotristat ethyl 250 mg tid
Telotristat ethyl 500 mg tid

Color represents treatment group during the DBT period; lighter shading represents treatment during the OLE period (telotristat ethyl 500 mg tid).
EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30; tid, 3 times per day

Conclusions

- Data from the 48-week OLE period suggest that telotristat ethyl was generally well tolerated, with few discontinuations due to AEs.
- No notable increase in the rate of TEAEs was observed for patients who crossed from placebo to telotristat in the OLE period, and TEAE incidence was similar to that of the DBT period.
- There was a statistically significant reduction in u5-HIAA levels at Week 12, and reductions were sustained through Week 48.
- Improvement in EORTC-QLQ diarrhea scores was observed at Week 12 with telotristat ethyl and maintained through Week 48.

REFERENCES

1. National Comprehensive Cancer Network. Neuroendocrine Tumors (Version 3.2017). 2017. **2.** Xermelo® (telotristat ethyl) US Highlights of Prescribing Information. Lexicon Pharmaceuticals, Inc. February 2017. **3.** European Commission. Commission Implementing Decision of 18.9.2017 granting marketing authorisation under Regulation (EC) No 726/2004 of the European Parliament and of the Council for "Xermelo - telotristat", an orphan medicinal product for human use. 2017. http://ec.europa.eu/health/documents/community-register/2017/20170918138652/dec_138652_en.pdf. Accessed October 2, 2017. **4.** Kulke MH, et al. *J Clin Oncol.* 2017;35(1):14–23. **5.** Pavel M, et al. Poster presented at the North American Neuroendocrine Tumor Society Annual Symposium; September 30–October 1, 2016; Jackson, WY; Poster #174. **6.** ClinicalTrials.gov. NCT02063659: Telotristat etiprate for carcinoid syndrome therapy (TELECAST). https://clinicaltrials.gov/ct2/show/NCT02063659. Accessed February 6, 2017. **7.** Longstreth GF, et al. *Gastroenterology.* 2006;130(5):1480–1491. **8.** Tellez MR, Mamikunian G, O'Dorisio TM, Vinik AI, Woltering EA. *Pancreas.* 2013;42(3):405–410. **9.** Cocks K, et al. *Eur J Cancer.* 2012;48(11):1713–1721.

ACKNOWLEDGEMENTS

We thank the patients and Investigators for participating in the study. We thank James Banigan, PhD (Chameleon Communications International with funding from Lexicon Pharmaceuticals, Inc.) for editorial assistance in the preparation of this report. The TELECAST study was sponsored by Lexicon Pharmaceuticals, Inc.

DISCLOSURES

MP: Ipsen Bioscience, Novartis Pharmaceuticals, Lexicon Pharmaceuticals, Inc., and Pfizer, Inc. PP: Lexicon Pharmaceuticals, Inc. RS: Ipsen Pharmaceuticals, Inc., and Novartis Pharmaceuticals. RRPW: Lexicon Pharmaceuticals, Inc. MHK: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, and Novartis Pharmaceuticals. LBA: Lexicon Pharmaceuticals, Inc. PLK: Merck & Co., Genentech, Advanced Accelerator Applications, Lexicon Pharmaceuticals, Inc., Oxygen, Esanex, Dicerna Pharmaceuticals, Ipsen Biopharmaceuticals, and Novartis Pharmaceuticals. DH: Lexicon Pharmaceuticals, Inc., Ipsen Bioscience, Novartis Pharmaceuticals, and Pfizer, Inc. PL and RF: current employees of Lexicon Pharmaceuticals, Inc., and may own common stock or may have been granted stock options or other equity incentive awards. MB and DJG: nothing to disclose.

To view this poster online, snap a photo of this QR code with your smart phone (QR reader required).

Presented at the NANETS 2017 Annual Symposium; October 19–21, 2017; Philadelphia, PA, USA.

Lexicon pharmaceuticals