Final progression-free survival analyses for lanreotide autogel/depot 120 mg in metastatic enteropancreatic neuroendocrine tumors: the CLARINET extension study

Edward M. Wolin,[1] Marianne E. Pavel,[2] Jarosław B. Ćwikła,[3] Alexandria T. Phan,[4] Markus Raderer,[5] Eva Sedláčková,[6] Guillaume Cadiot,[7] Jaume Capdevila,[8] Lucy Wall,[9] Guido Rindi,[10] Catherine Lombard-Bohas,[11] Nilani Liyanage,[12] Xuan-Mai Truong Thanh,[13] Philippe B. Ruszniewski,[14] Martyn E. Caplin[15]; CLARINET Investigators

[1]Montefiore Einstein Center for Cancer Care, Bronx, NY, USA; [2]Universitätsklinikum Erlangen, Erlangen, Germany; [3]University of Warmia and Mazury, Olsztyn, Poland; [4]University of New Mexico Cancer Center, Albuquerque, NM, USA; [5]University Hospital, Vienna, Austria; [6]Department of Oncology of the First Faculty of Medicine and General Teaching Hospital, Prague, Czech Republic; [7]Robert-Debré Hospital, Reims, France; [8]Vall d'Hebron University Hospital, Barcelona, Spain; [9]Western General Hospital, Edinburgh, UK; [10]Università Cattolica del Sacro Cuore, Rome, Italy; [11]Edouard-Herriot Hospital, Hospices Civils de Lyon, Lyon, France; [12]Ipsen, Les Ulis, France; [13]Ipsen, Boulogne-Billancourt, France; [14]Beaujon Hospital, Clichy, France; [15]Royal Free Hospital, London, UK.

Scan here to view a PDF of this poster

Copies of this poster obtained through this quick response code are for personal use only and may not be reproduced without written permission from the authors

Background

- In the CLARINET core study, lanreotide autogel (depot in the USA) 120 mg injected every 4 weeks significantly improved progression-free survival (PFS) vs. placebo in 204 patients with metastatic grade 1 or 2 (Ki-67 <10%) enteropancreatic neuroendocrine tumors (NETs).[1]
- An open-label extension (OLE) to the core study evaluated long-term safety and efficacy.[2,3] Although the vast majority of the core-study population (96%) had stable disease (SD) at baseline, the OLE included patients with progressive disease (PD) while receiving placebo during the core study, and patients with SD at the end of the core study who had been receiving lanreotide or placebo.[2]
 - Both the pre-planned interim and final analyses of the OLE demonstrated acceptable long-term safety/tolerability of lanreotide autogel 120 mg.[2–4]
 - Here, we report the final analysis of PFS for lanreotide autogel, and also the PFS within subgroups according to tumor origin and prior therapy.

Methods

Patients

- Patients were enrolled into the core study if they had metastatic, well-/moderately differentiated, grade 1 or 2 (Ki-67 <10%), non-functioning somatostatin receptor-positive NETs, originating in the pancreas, midgut or hindgut, or of unknown origin.
- Patients were eligible for the OLE if they had:
 - SD at the end of the 96-week core study after receiving lanreotide or placebo, or withdrawn as a result of PD while receiving placebo.
 - A World Health Organization performance status score ≤2.
- Not all eligible patients continued into the OLE, as participation was optional and some centers did not participate.

Study design and treatment

- The OLE was a single-arm non-randomized international study (ClinicalTrials.gov: NCT00842348; EudraCT: 2008-004019-36).
- All patients received lanreotide autogel 120 mg every 4 weeks until PD/death, or early withdrawal.

Objectives and endpoints

- OLE objectives: to investigate the long-term safety (primary) and further investigate the efficacy (secondary) of lanreotide autogel 120 mg.[2]
- Here, we provide, in brief, safety information (adverse events [AEs]) from the OLE for patients continuing into the OLE after completing the 96-week core study receiving lanreotide autogel with SD.
 - AEs were recorded at 4-weekly visits.
- We report the PFS of lanreotide autogel 120 mg using data from patients continuing treatment with lanreotide in the OLE appended to data from patients originally randomized to lanreotide in the core study. PFS data were additionally calculated for subgroups according to tumor origin and presence/absence of previous therapy for nonfunctioning NETs at the start of the core study.
 - In the core study imaging scans were performed every 12 weeks in year 1 and every 24 weeks in year 2; these scans were centrally assessed (RECIST v.1.0). In the OLE imaging scans were performed at OLE baseline, every 24 weeks, and at the time of early withdrawal/PD; the scans were assessed locally for signs of PD (response evaluation criteria in solid tumors [RECIST] v.1.0).
 - In all cases, PFS was defined as the time from randomization in the core study to the first occurrence of PD (RECIST v.1.0) or death in the core study or the OLE.

Statistical analyses

- Descriptive statistics: calculated for demographic and disease characteristics and AEs using the safety population (all patients who received at least one dose of lanreotide autogel in the OLE).
- Efficacy analyses: based on the intent-to-treat population (all patients randomly allocated to treatment in the core study, regardless of whether they continued into the OLE) or subsets of that population.
- PFS data: described using Kaplan–Meier plots. Data are presented in months approximated to 4 weeks.

Results

- A total of 89 patients were enrolled into the OLE. Three quarters of these had pancreatic or midgut NETs (38% in each case).
- A total of 42 patients were included after completing the core study receiving lanreotide autogel (LAN–LAN group).
- A total of 26 patients were withdrawn. Reasons for withdrawal: PD (n=17), consent withdrawn (n=2), AEs (n=2), protocol violation (n=1), other reasons (n=4). One of the patients withdrawn had been enrolled in the OLE before PD in the core study had been confirmed; therefore, 41 patients with SD entered OLE and contributed to the PFS analysis.
- Sixteen patients completed the OLE without experiencing PD.
- Baseline characteristics are shown in Table 1.
- The median (range) exposure to lanreotide autogel, for core and OLE studies combined, was 59.0 (26.0–102.3) months.
- Although not the focus of analyses presented here, the OLE also included 47 patients who had previously received placebo during the core study (PBO–LAN group; 15 with SD and 32 with PD).

Safety

- During the OLE, 40% of the 42 patients in the LAN–LAN group experienced treatment-related AEs (in comparison, 47% of 47 patients in the PBO–LAN group experienced treatment-related AEs).
- In the LAN–LAN group, cholelithiasis and diarrhea were the most common treatment-related AEs (seven patients [16.7%] and four patients [9.5%], respectively) (corresponding frequencies for the PBO–LAN group were six (12.8%) and 12 (25.5%), respectively). Other treatment-related AEs occurred in <10% of patients. Two patients in the LAN–LAN group experienced serious treatment-related AEs (cholelithiasis in both cases); none of the patients withdrew due to treatment-related AEs.
- Overall, no new safety concerns were identified.

Efficacy

- The median [95% CI] PFS for lanreotide autogel (n=101) was 38.5 [30.9; 59.4] months (Figure 1).
 - Survival estimates were based on 51 events; 32 events during the core study and 19 events during the OLE (including one death and 18 PD events). Of the 18 patients experiencing PD, one patient died as a consequence (only the PD event for this patient is included in the PFS analyses).

Table 1. Demographic and disease characteristics (safety population).

Characteristic	LAN–LAN group (n=42)
Men	19 (45.2)
Age, mean (SD) in years*	64.8 (10.8)
Time since diagnosis, mean (SD) in months	36.1 (58.1)
WHO performance status score*	
0 – normal activity	36 (85.7)
1 – restricted activity	6 (14.3)
2 – in bed ≤50% of the time	0
Prior NET treatment	6 (14.3)
NET origin	
Pancreas	12 (28.6)
Midgut	17 (40.5)
Hindgut	5 (11.9)
Other/unknown	8 (19.0)
Tumor progression at baseline of:	
Core study	0
OLE study	1 (2.3)†
Tumor grade	
1 (Ki-67 0–2%)	30 (71.4)
2 (Ki-67 3–10%)§	12 (28.6)
Hepatic tumor load	
0%	9 (21.4)
>0–10%	19 (45.2)
>10–25%	2 (4.8)
>25–50%	10 (23.8)
>50%	2 (4.8)

Data are for assessments at baseline of either the CLARINET core study or *OLE and are n (%) unless stated otherwise. †Enrolled by investigator before communication of results of central assessment (progressive disease) in the core study; patient withdrawn from the OLE on receipt of assessment result. §Tumor proliferation index (Ki-67) thresholds are stated in accordance with WHO 2010 classification, but with values >2 to ≤10% assigned to tumor grade 2. LAN–LAN, patients receiving lanreotide autogel 120 mg in the core study who continued into the OLE; OLE, open-label extension; NET, neuroendocrine tumor; WHO, World Health Organization.

Figure 1. PFS for patients receiving lanreotide autogel 120 mg during the core study and OLE and PFS for patients receiving placebo in the core study (ITT population).

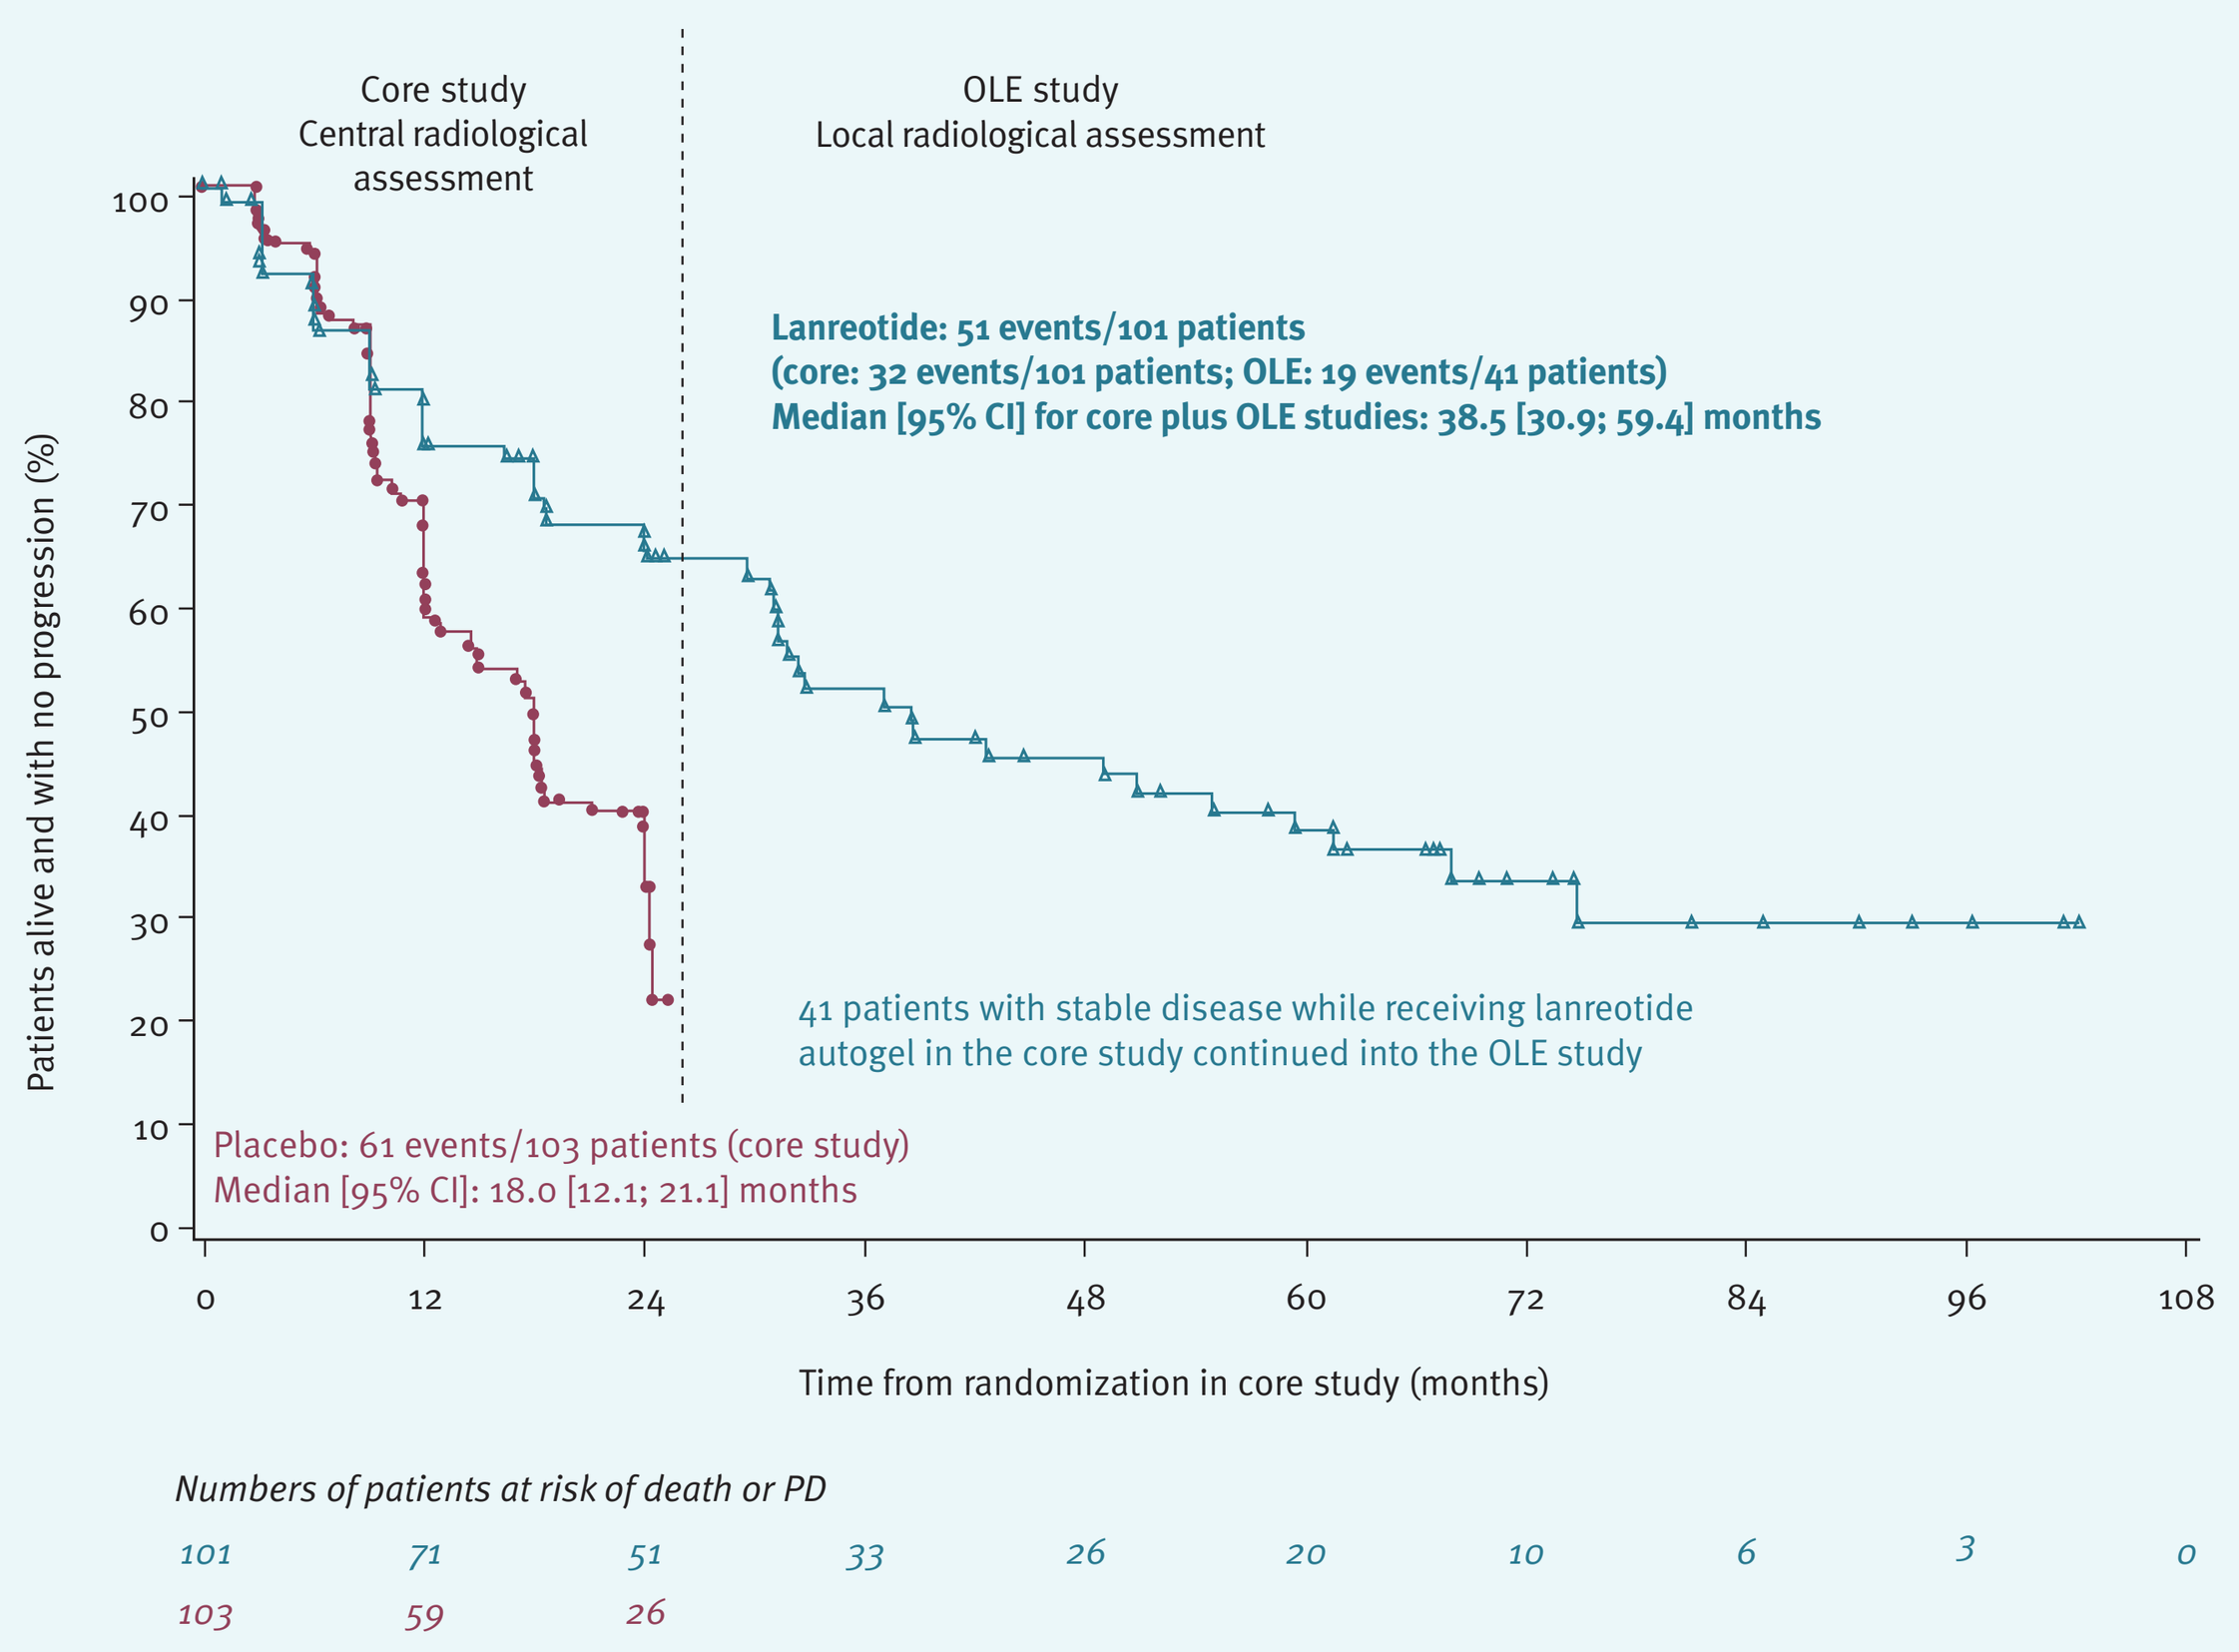
Core study
Central radiological assessment
OLE study
Local radiological assessment
Lanreotide: 51 events/101 patients
(core: 32 events/101 patients; OLE: 19 events/41 patients)
Median [95% CI] for core plus OLE studies: 38.5 [30.9; 59.4] months
41 patients with stable disease while receiving lanreotide autogel in the core study continued into the OLE study
Placebo: 61 events/103 patients (core study)
Median [95% CI]: 18.0 [12.1; 21.1] months
Patients alive and with no progression (%)
Time from randomization in core study (months)
Numbers of patients at risk of death or PD
101 71 51 33 26 20 10 6 3 0
103 59 26

Data are presented in months approximated to 4 weeks. The data from the OLE (n=41) were appended to data from the core study (n=101) for lanreotide autogel. ITT, intent-to-treat; OLE, open-label extension; PFS, progression-free survival.

Figure 2. PFS during the core study and the OLE for patients randomized to lanreotide autogel 120 mg in the core study (compared with the PFS for patients randomized to placebo in the core study) for subgroups according to tumor origin and prior therapy (ITT population).

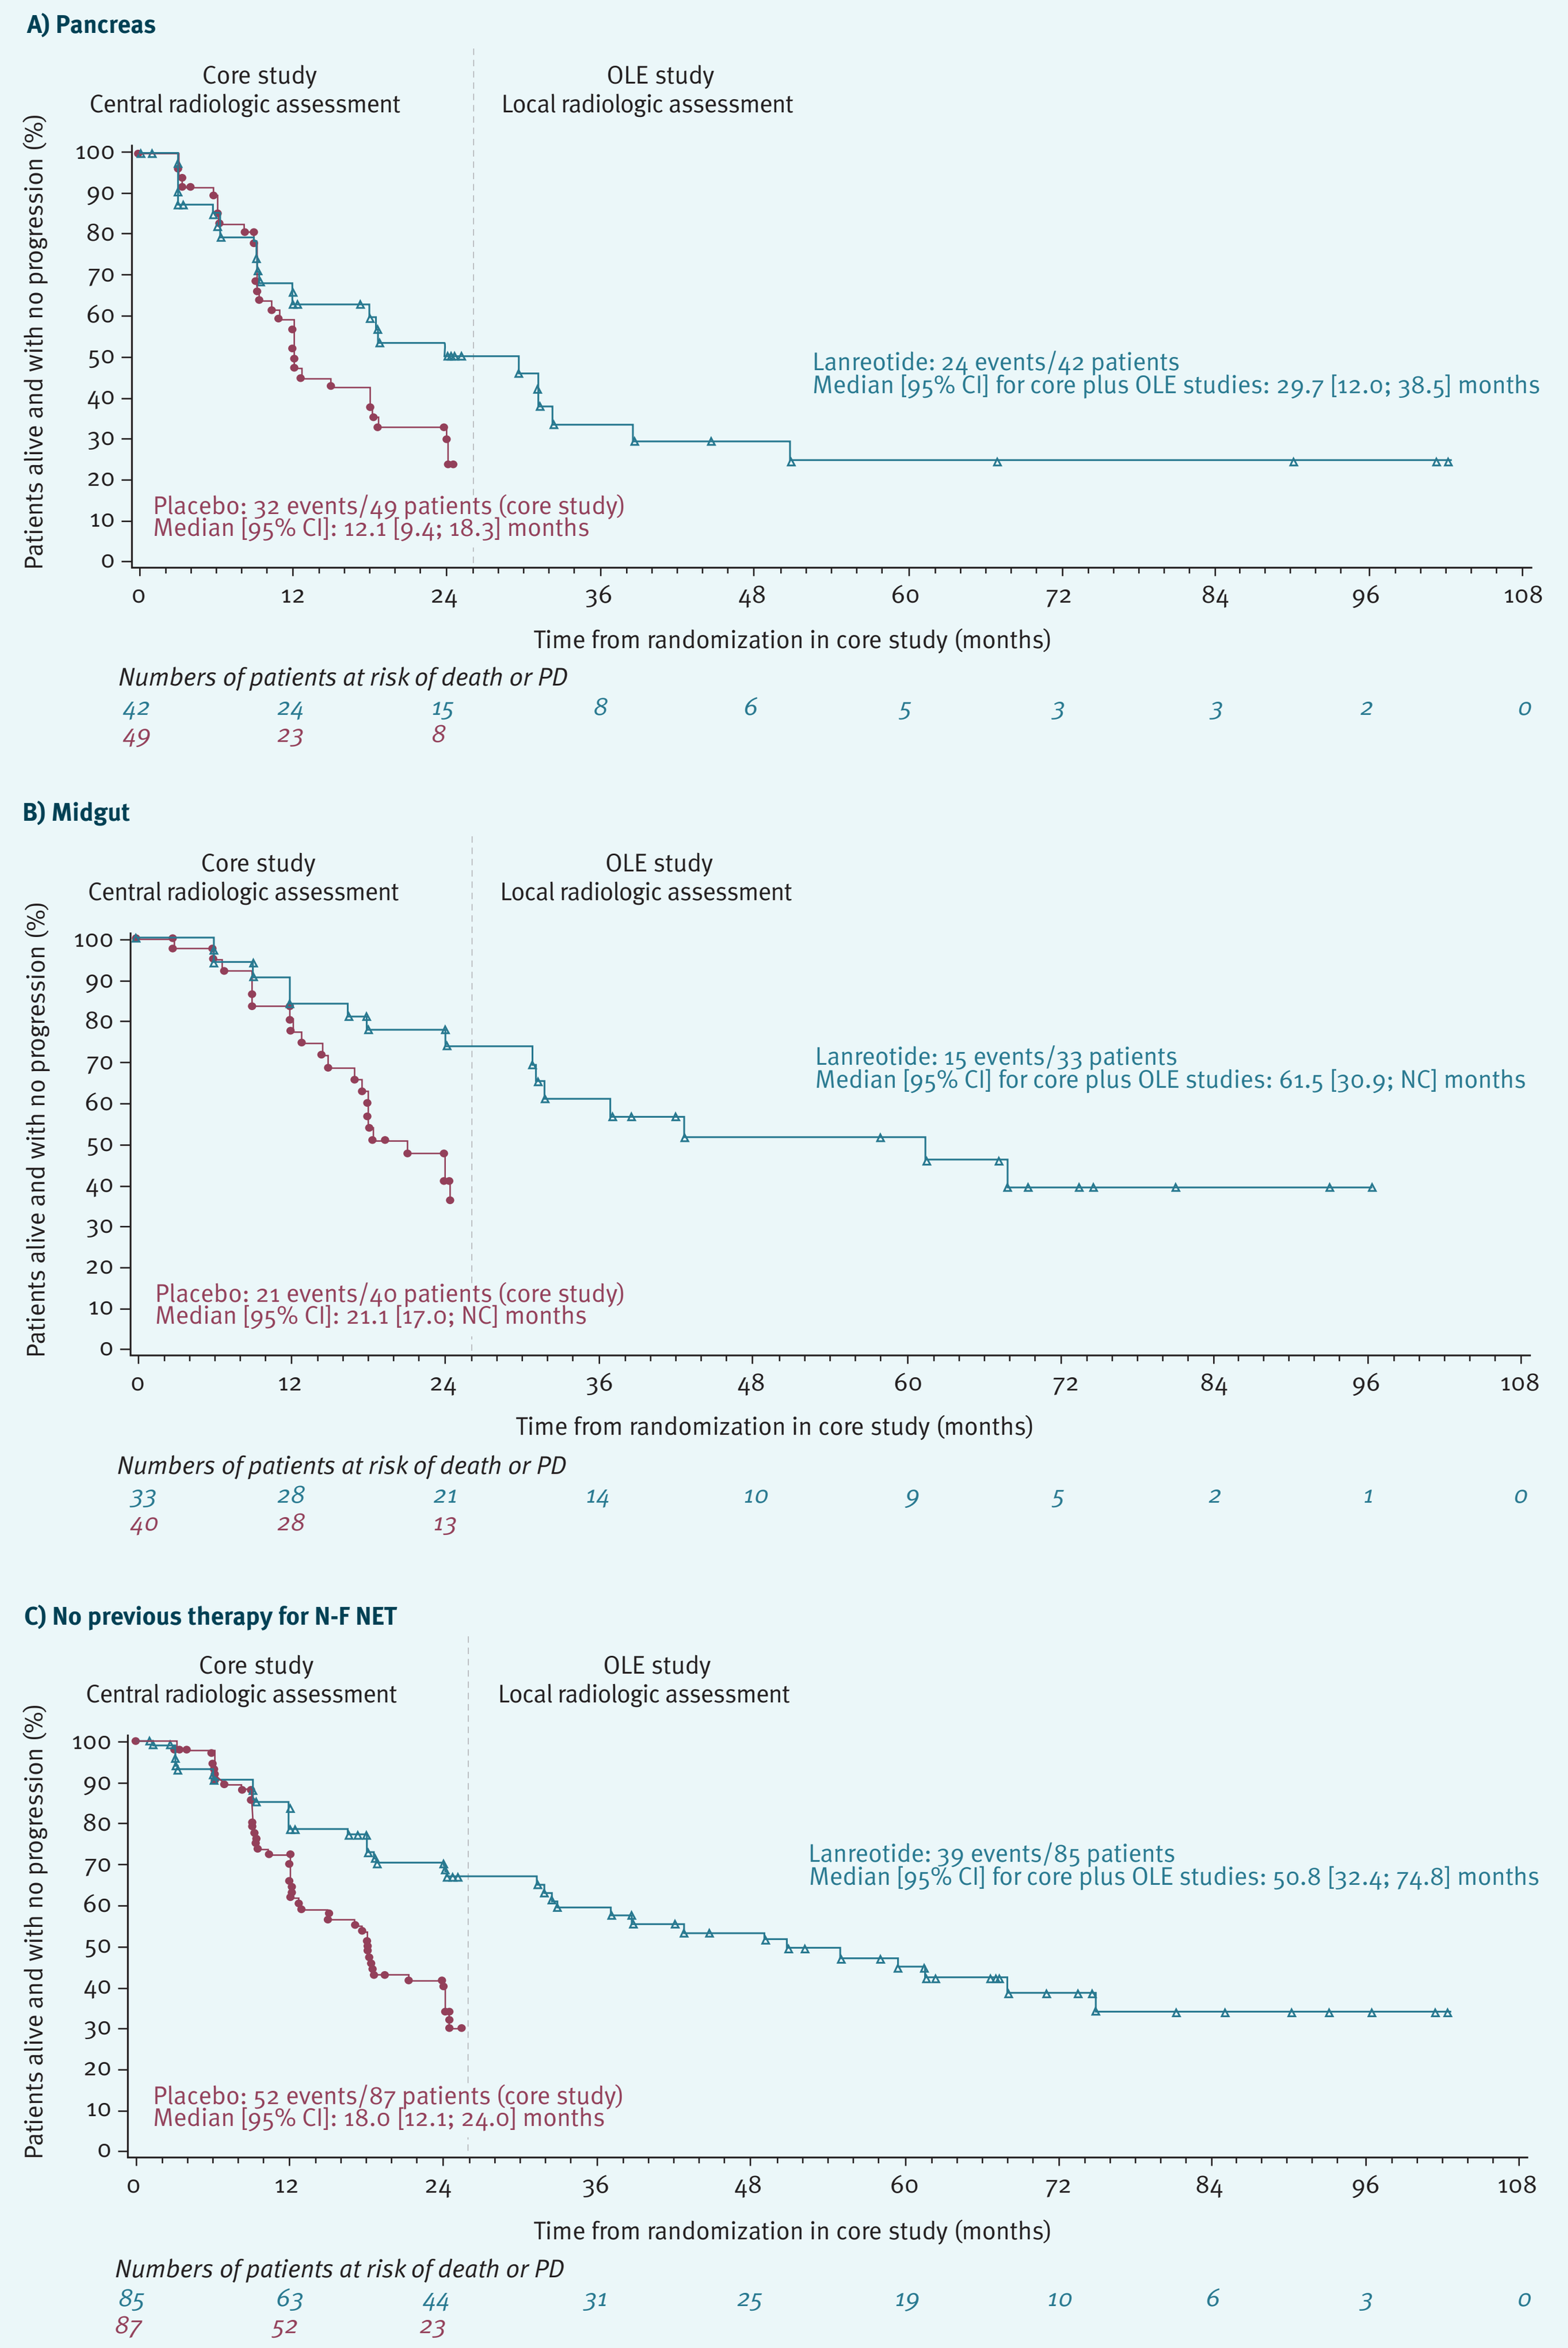
A) Pancreas
Core study
Central radiologic assessment
OLE study
Local radiologic assessment
Lanreotide: 24 events/42 patients
Median [95% CI] for core plus OLE studies: 29.7 [12.0; 38.5] months
Placebo: 32 events/49 patients (core study)
Median [95% CI]: 12.1 [9.4; 18.3] months
Patients alive and with no progression (%)
Time from randomization in core study (months)
Numbers of patients at risk of death or PD
42 24 15 8 6 5 3 3 2 0
49 23 8
B) Midgut
Lanreotide: 15 events/33 patients
Median [95% CI] for core plus OLE studies: 61.5 [30.9; NC] months
Placebo: 21 events/40 patients (core study)
Median [95% CI]: 21.1 [17.0; NC] months
33 28 21 14 10 9 5 2 1 0
40 28 13
C) No previous therapy for N-F NET
Lanreotide: 39 events/85 patients
Median [95% CI] for core plus OLE studies: 50.8 [32.4; 74.8] months
Placebo: 52 events/87 patients (core study)
Median [95% CI]: 18.0 [12.1; 24.0] months
85 63 44 31 25 19 10 6 3 0
87 52 23

D) Other subgroups*

	Median PFS [95% CI] (no. patients), months	
	LAN (core study and OLE)	PBO (core study)
Tumor origin		
Hindgut	55.0 [2.9; NC] (11)	24.4 [12.0; 24.4] (3)
Other/unknown	59.4 [32.8; 74.8] (15)	15.0 [6.3; NC] (11)
Previous therapy for N-F NET		
Yes	29.7 [6.0; 31.3] (16)	12.0 [3.3; NC] (16)

Data are presented in months approximated to 4 weeks in all cases. The data from the OLE study were appended to data from the core study for lanreotide autogel. ITT, intent-to-treat; LAN, lanreotide autogel 120 mg; N-F, non-functioning; NC, not calculable; NET, neuroendocrine tumor; OLE, open-label extension; PBO, placebo; PD, progressive disease; PFS, progression-free survival.

- Median PFS varied across subgroups based on tumor origin and prior therapy status (Figure 2).
- Patients with a NET of midgut origin had a higher median PFS with long-term lanreotide treatment than those with a NET of pancreatic origin (Figures 2a and b). Patients who had received no previous therapy for non-functioning NETs had a higher median PFS than those receiving previous therapy (Figures 2c and d).

Conclusion

- **The final analysis of the CLARINET OLE showed that long-term treatment with lanreotide autogel 120 mg every 4 weeks has a generally acceptable tolerability profile in patients with SD continuing with lanreotide after the core study.**
- **CLARINET OLE suggests there are sustained antitumor effects with lanreotide autogel 120 mg every 4 weeks in patients with metastatic grade 1 or 2 (Ki-67 <10%) enteropancreatic NETs compared with placebo (core study), irrespective of tumor origin, and that there may be benefits as an early treatment.**

References

1. Caplin *et al. N Engl J Med* 2014;371:224–33.
2. Caplin *et al. Endocr Relat Cancer* 2016;23:191–9.
3. Phan *et al.* Presented at the 14th Annual ENETS Conference, March 8–10, 2017, Barcelona, Spain: abst K18.
4. Caplin *et al. Ann Oncol* 2016;27: abst 439P.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support in preparing this poster.

Disclosures

E.M.W. received research funding from Ipsen and Novartis; consulting/advisory fees from Ipsen, Novartis, Pfizer and Celgene; and honoraria from Ipsen and Novartis. **M.E.P.** received research funding from Novartis; and consulting/advisory fees from Ipsen, Novartis, Pfizer and Lexicon. **J.B.Ć.** received research funding from Ipsen. **A.T.P.** received research funding from Ipsen, Novartis, Lexicon, Sanofi and Incyte; consulting/advisory fees from Ipsen and Novartis; and speaker fees from Lilly, Genentech, Celgene, Novartis and Ipsen. **M.R.** received honoraria from Novartis, Ipsen, Roche, Pfizer, Bayer and Celgene; and speaker fees from Novartis, Ipsen, Roche, Pfizer, Bayer and Celgene. **E.S.** and **L.W.** have no conflicts of interest. **G.C.** received research funding from Ipsen; and consulting/advisory fees from Ipsen, Novartis and Keocyt. **J.C.** received consulting/advisory fees from Ipsen, Novartis and Pfizer. **G.R.** received speaker fees from Ipsen. **C.L-B.** received consulting/advisory fees from Ipsen, Novartis and Pfizer. **N.L.** and **X-M.T.T.** are employees of Ipsen. **P.B.R.** received research funding from Ipsen and Novartis; speaker fees from Ipsen and Novartis; consulting/advisory fees from Ipsen; and honoraria from Ipsen and Novartis. **M.E.C.** received consulting/advisory fees from Ipsen.

CLARINET core study and OLE† investigators:*

Austria M. Raderer*; **Belgium** I. Borbath*†, D. Ysebaert*†; **Czech Republic** P. Holeckova†, E. Sedláčková*†, P. Vítek*†; **Denmark** H. Grønbæk*; **France** A. Adenis*†, L. Buscail*, G. Cadiot*†, S. Dominguez*†, JF. El Hajbi†, M. Ducreux*, C. Lombard-Bohas*†, E. Mitry*, P. Ruszniewski*†, J.F. Seitz*; **Germany** N. Begum*, I. Harsch*, M. Pavel*, C. Schöfl*, M. Weber*, B. Wiedenmann*; **India** M. Mallath*†, P. Patil*†, K. Sambasivaiah*, R. Saxena*†; **Italy** E. Bajetta*†, A. Buonadonna*†, R. Buzzoni*†, R. Cannizzaro*†, A. Colao*†, C. De Angelis*†, P. Tomassetti*; **Poland** J. Ćwikła*†, B. Kos-Kudła*; **Slovakia** T. Salek*†; **Spain** J. Capdevila*†, G. Soler*, J.M. Tabernero*, A. Teulé Vega†; **Sweden** H. Ahlman*, M. Kjellman*; **UK** G. Aithal*†, A. Anthoney*, M. Caplin*†, A. Grossman*, J. Newell-Price*, J. Ramage*, N. Reed*†, A. Rees*†, W. Steward*, L. Wall*†; **USA** M. Choti*†, A. Hendifart, T. Pawlik†, A.T. Phan*, E.M. Wolin*†.

Presented at the 10th NANETS Annual Meeting, Philadelphia, PA, USA, October 19–21, 2017.

This study is sponsored by Ipsen.