Phase 1/2 open-label trial to assess the safety and preliminary efficacy of ^{177}Lu-OPS201 as peptide receptor radionuclide therapy in patients with somatostatin receptor-positive, progressive neuroendocrine tumors

Guillaume Nicolas,[1,2] Lisa Bodei,[3] Richard P. Baum,[4] Ken Herrmann,[5] Michael Lassmann,[6] Rodney J. Hicks,[7] Shaunak Navalkissoor,[2] Alexander R. Haug,[8] Heike Oberwittler,[9] Tiffany Wang,[10] Damian Wild[1]

[1]University of Basel Hospital, Basel, Switzerland; [2]Royal Free Hospital, London, UK; [3]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [4]Zentralklinik Bad Berka, Bad Berka, Germany; [5]UCLA, Los Angeles, CA, USA; [6]University Hospital Würzburg, Würzburg, Germany; [7]Peter MacCallum Cancer Centre, East Melbourne, Victoria, Australia; [8]Medical University of Vienna, Vienna, Austria; [9]Ipsen, Les Ulis, France; [10]Ipsen, Cambridge, MA, USA

Scan here to view a PDF of this poster

Copies of this poster obtained through this quick response code are for personal use only and may not be reproduced without written permission from the authors

Background

- Radiolabeled somatostatin receptor (SSTR) agonists are currently used in peptide receptor radionuclide therapy (PRRT) for neuroendocrine tumors (NETs) in Europe.[1]
- Internalization of radiolabeled receptor agonists into tumor cells and the resulting radionuclide accumulation was previously thought to be critical for efficient tumor targeting.[1]
- This paradigm has been challenged by evidence that SSTR antagonists have higher tumor binding, tumor uptake, longer residence time and consequently higher tumor-radiation dose compared with SSTR agonists despite limited or no internalization.[2,3]
- ^{177}Lu-OPS201 (DOTA-JR11) is a radiolabeled SSTR antagonist that is selective for, and binds with high affinity to SSTR2 (expressed for example by gastroenteropancreatic [GEP]-NETs,[4] lung NETs,[5] pheochromocytomas and paragangliomas,[6] medullary thyroid cancer, small cell lung cancer, breast cancer, non-Hodgkin lymphoma, renal cell carcinoma[7]).

Table 1. Key inclusion and exclusion criteria.

Inclusion criteria
• Patients (≥18 years) with a histologically confirmed diagnosis of an unresectable grade 1/2 SSTR-positive GEP-NET or lung NET; or an SSTR-positive, malignant, unresectable pheochromocytoma or paraganglioma
• Karnofsky performance score of ≥60
• Life expectancy of ≥6 months
• PD during the previous 6 months despite antitumor therapy
• Glomerular filtration rate ≥55 mL/min
• Leukocytes ≥4 × 10^9/L; erythrocytes ≥3.5 × 10^{12}/L; platelets ≥100 × 10^9/L; albumin >30 g/L; alanine transaminase, aspartate transaminase and alkaline phosphatase ≤5 × ULN; bilirubin ≤2 × ULN
Exclusion criteria
• Active infection at screening or within the previous 6 weeks
• Diagnosis of thymic NET
• Chemotherapy or extensive radiotherapy ≤3 months before enrollment, or any previous PRRT

GEP-NET, gastroenteropancreatic neuroendocrine tumor; NET, neuroendocrine tumor; PD, progressive disease; PRRT, peptide receptor radionuclide therapy; SSTR, somatostatin receptor; ULN, upper limit of normal.

Aims

- The aims of this phase 1/2 trial are to evaluate the safety, tolerability, biodistribution, pharmacokinetics (PK), dosimetry, preliminary efficacy and quality-of-life (QoL) effect of ^{177}Lu-OPS201 administered in three consecutive cycles to 45 adults with unresectable progressive SSTR-positive GEP-NETs, lung NETs, pheochromocytomas, and paragangliomas.

Methods

Patients

- Inclusion and exclusion criteria are shown in **Table 1**.
- Enrollment of 45 patients from 15 participating centers with experience in PRRT (or other radionuclide therapy) in Europe, USA and Australia is planned.

Study design and treatment

- This is a phase 1/2 international single-arm open-label study evaluating ^{177}Lu-OPS201 for the treatment of NETs (EudraCT 2015-002867-41; NCT02592707).
- The study consists of a core trial (6–9 months) and a long-term follow-up period (2 years).
- The core trial has two phases, phase A (low activity) and phase B (high activity) (**Figure 1**).

 Phase A (low activity, n=15)
 - Patients receive three cycles of ^{177}Lu-OPS201 (250–300 µg peptide) at an activity of 4.5 GBq by intravenous infusion every 8 weeks.
 - A safety review committee (SRC; composed of study investigators, a dosimetry expert, and at least one Ipsen representative) will monitor safety and dosimetry data and adjust the administered activity of ^{177}Lu-OPS201 accordingly.

Figure 1. Study design.

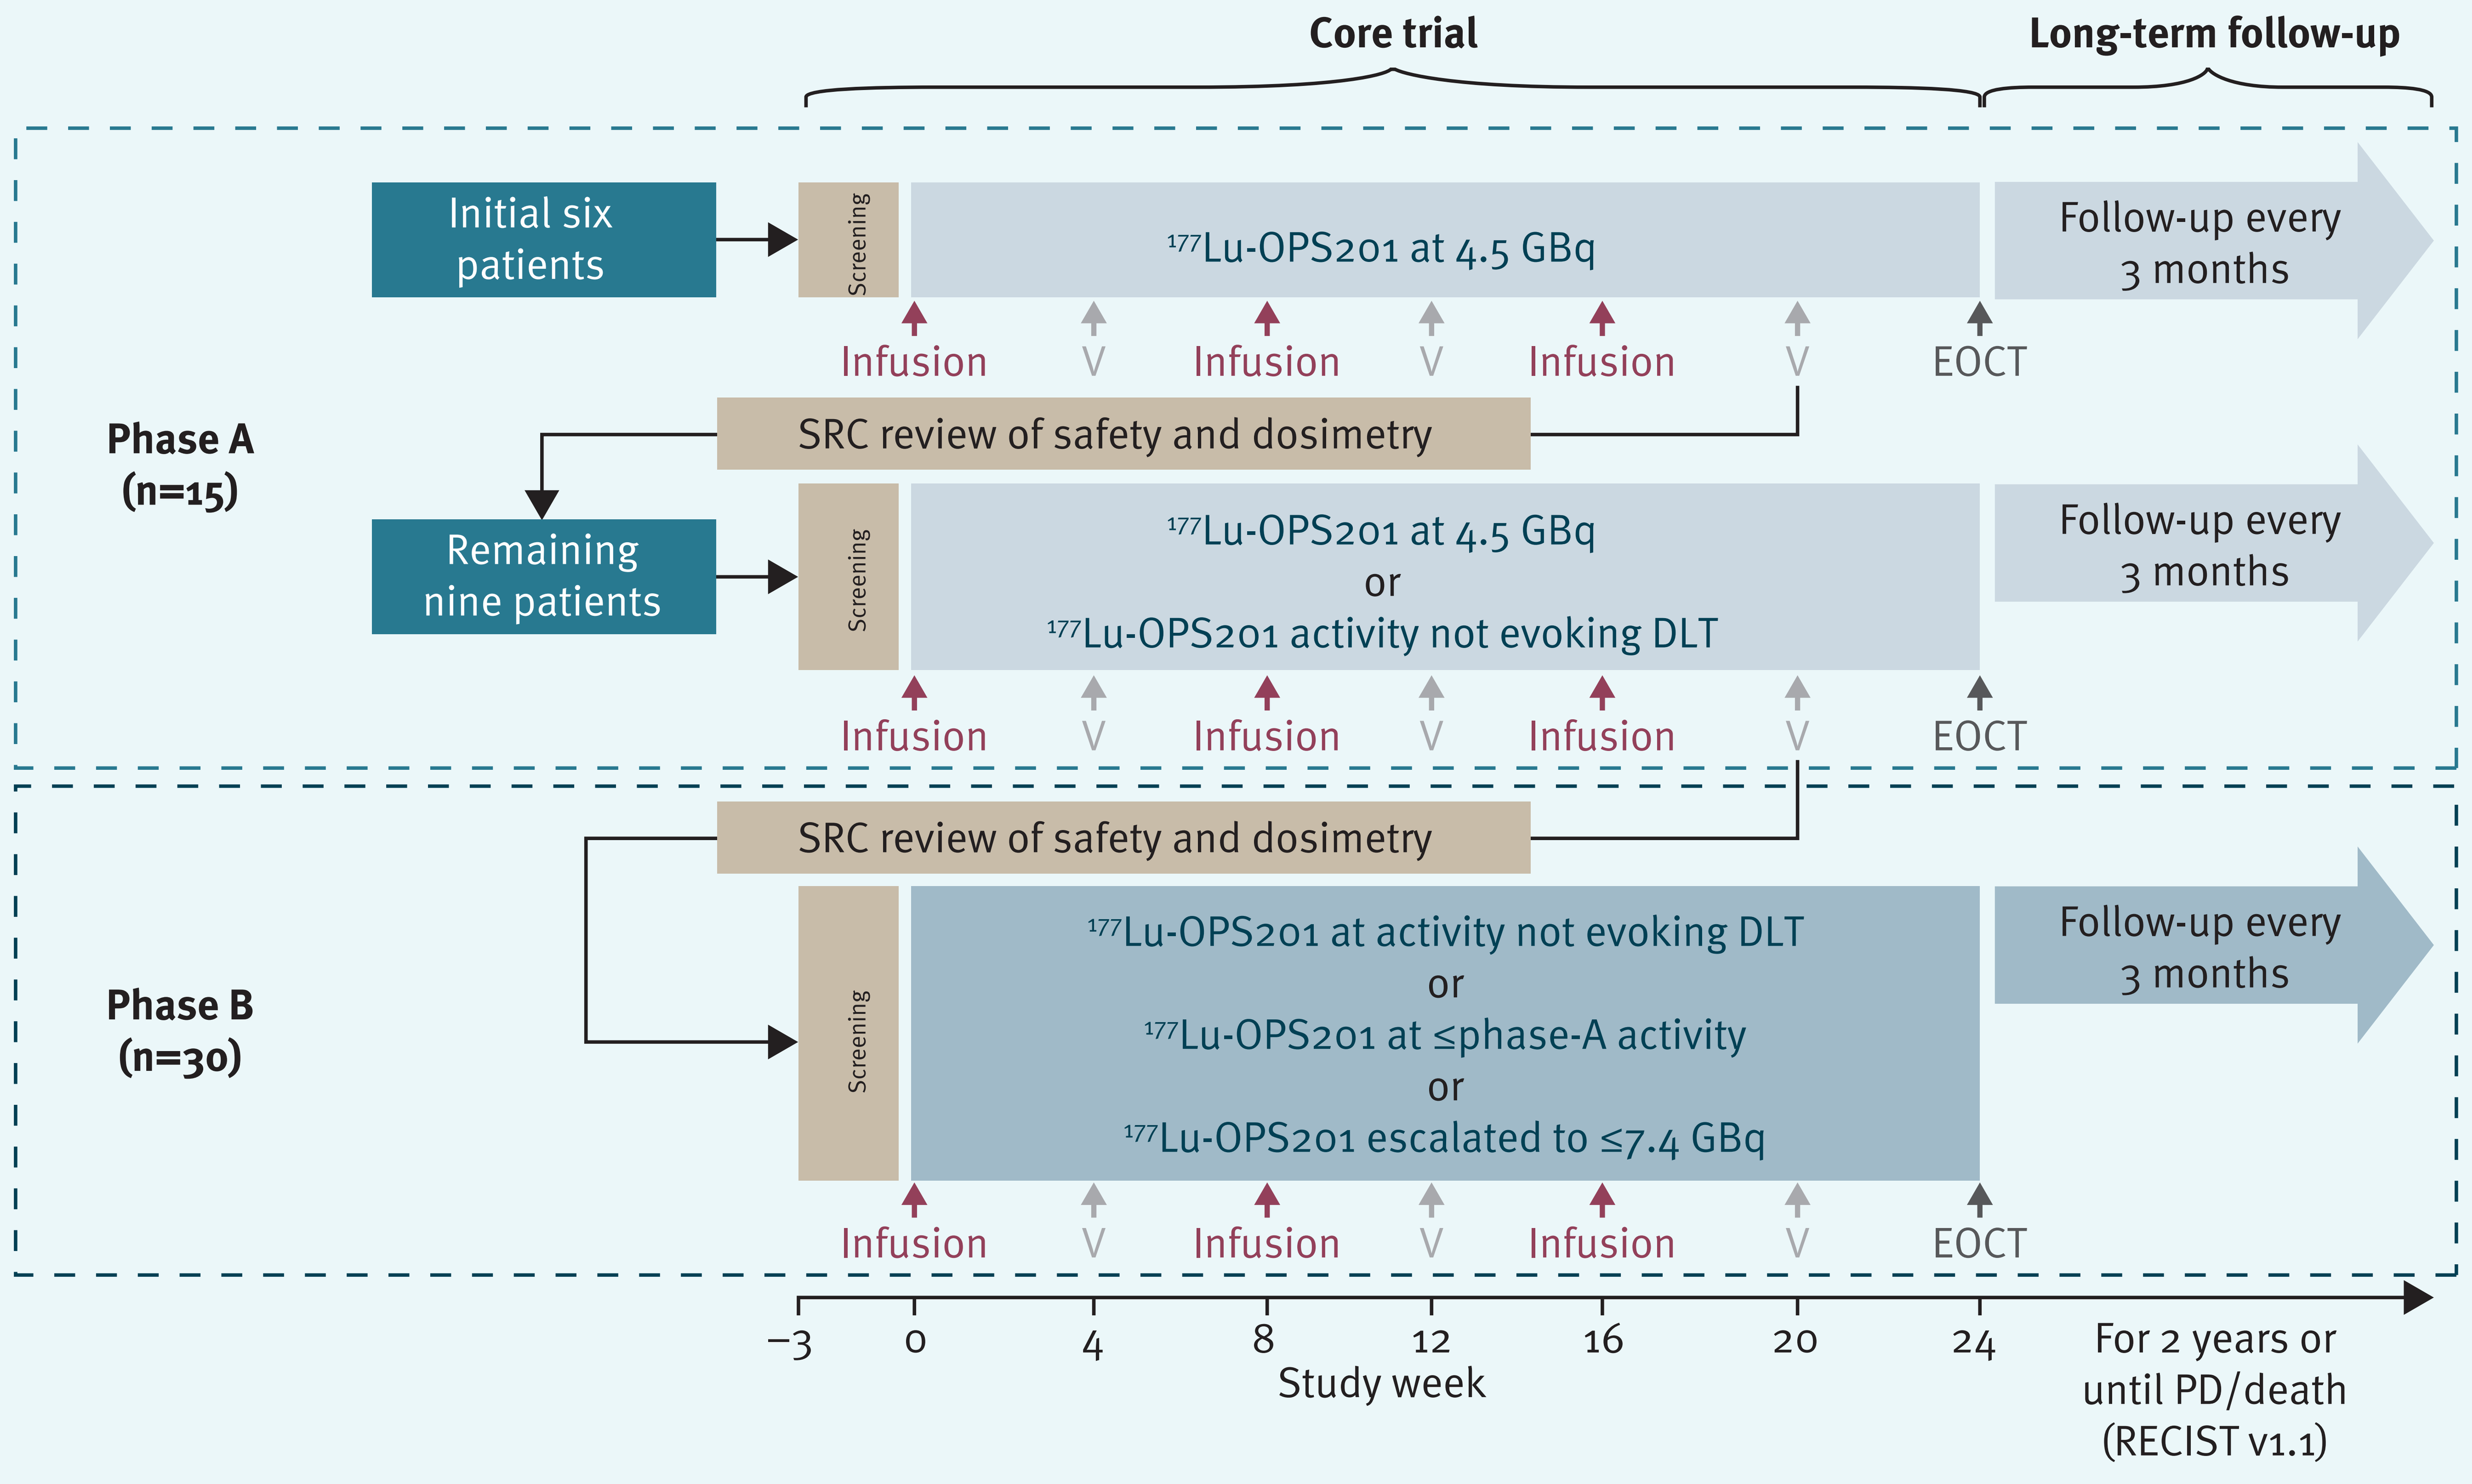

Core trial
Long-term follow-up
Phase A (n=15)
Initial six patients
Screening
177Lu-OPS201 at 4.5 GBq
Follow-up every 3 months
Infusion
V
EOCT
SRC review of safety and dosimetry
Remaining nine patients
177Lu-OPS201 at 4.5 GBq or 177Lu-OPS201 activity not evoking DLT
Phase B (n=30)
177Lu-OPS201 at activity not evoking DLT or 177Lu-OPS201 at ≤phase-A activity or 177Lu-OPS201 escalated to ≤7.4 GBq
−3 0 4 8 12 16 20 24
Study week
For 2 years or until PD/death (RECIST v1.1)

DLT, dose-limiting toxicity; EOCT, end-of-core-trial visit; Infusion, administration of ^{177}Lu-OPS201; PD, progressive disease; RECIST, response evaluation criteria in solid tumors; SRC, safety review committee; V, visit (safety and efficacy assessments will be performed at these time points).

Figure 2. Study endpoints.

Primary endpoint

Safety and tolerability	AEs, physical examination, vital signs, electrocardiogram, clinical laboratory measurements, DLTs, concomitant medication, pituitary markers, and bone-marrow aspirate for persisting toxicities ≥ grade 3

Secondary endpoints

Biodistribution and PK	• Maximal uptake (%) in target lesion and organs • ^{177}Lu-OPS201 AUC in target lesion, organs, and blood • Terminal half-life of activity concentrations in blood
Radiation dosimetry	• Organs receiving the highest absorbed dose • Specific absorbed dose per target lesion and organ • Cumulative absorbed organ doses
Preliminary efficacy	• Tumor response (RECIST v1.1) • PFS (RECIST v1.1)
QoL	• EORTC QLQ-C30 and QLQ-GI.NET21 questionnaires

Exploratory endpoints

- OPS201 PK (phase B only)
- Radioactive metabolites in plasma
- Exploratory efficacy
- Exploratory safety biomarkers
- Biobanking blood (optional)

All AEs will be reported using the National Cancer Institute common toxicology criteria classification (v4.03) and coded according to the medical dictionary for regulatory activities v18.0 or later. Concomitant medication will be coded using the World Health Organization drug dictionary. AE, adverse event; AUC, area under the concentration–time curve; DLT, dose-limiting toxicity; EORTC QLQ-C30, European Organisation for the Treatment of Cancer core quality-of-life questionnaire core 30; EORTC QLQ-GI.NET21, European Organisation for the Treatment of Cancer quality-of-life questionnaire for gastro-intestinal neuroendocrine tumors; PFS, progression-free survival; PK, pharmacokinetics; QoL, quality of life; RECIST, response evaluation criteria in solid tumors.

Phase B (high activity, n=30)
- A separate cohort of patients (n=30) will receive three consecutive cycles of ^{177}Lu-OPS201 (300 µg peptide) at activities up to 7.4 GBq dependent on the phase-A safety profile (the maximum dose will be decided by the SRC).

- Follow-up computed tomography/magnetic resonance imaging (centrally reviewed) will be performed every 3 months from the end-of-core-trial visit for 2 years or until progressive disease/death according to RECIST v1.1.

Study endpoints

- Primary, secondary, and exploratory endpoints are summarized in **Figure 2**.

Current status

- Three centers are now open for recruitment and treatment in phase A is underway.

Conclusion

- **This study will provide important information regarding the safety, tolerability, biodistribution, dosimetry, preliminary efficacy and quality-of-life effect of ^{177}Lu-OPS201 peptide receptor radionuclide therapy in patients with progressive, somatostatin receptor-positive GEP-NETs, lung NETs, pheochromocytomas, and paragangliomas.**

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical for medical writing support in preparing this poster.

Disclosures

G.N. has received research funding from Octreopharm/Ipsen Group; consultancy fees from Novartis, Bayer and Ipsen; member of Ipsen advisory board. **L.B.** has received consultancy fees from Ipsen and Advanced Accelerator Applications. **R.P.B.** has received research support from Octreopharm/Ipsen Group; consultancy fees from Ipsen. **K.H.** has received research funding from Octreopharm/Ipsen Group; co-founder of SurgicEye GmbH; consultant for Siemens, Sofie Biosciences and Ipsen. **M.L.** has received research funding from Octreopharm/Ipsen Group. **R.H.** has received research funding from Octreopharm/Ipsen Group; consultancy fees from Ipsen. **S.N.** has nothing to disclose. **A.R.H.** has received research funding from Ipsen. **H.O.** and **T.W.** are employees of Ipsen. **D.W.** has received research funding from Octreopharm/Ipsen and Siemens; consultancy fees from Ipsen; and Ipsen advisory board member.

References

1. Cescato R, *et al. J Nucl Med* 2006;47:502–11.
2. Fani M, *et al. J Nucl Med 2017*;58:61S–66S.
3. Nicolas G, *et al. J Nucl Med* 2017;58:1435–41.
4. Papotti M, *et al. Virchows Arch* 2002;440:461–75.
5. Kanakis G, *et al. Neuroendocrinology* 2015;101:211–22.
6. Elston M, *et al. Hum Pathol* 2015;46:390–6.
7. Reubi J, *et al. J Nucl Med* 2017;58:300–6.

Presented at the 10th NANETS Annual Meeting, Philadelphia, PA, USA, October 19–21, 2017.

This study is sponsored by Ipsen.