ElevatION NET-201: An Open-Label Phase II Study to Evaluate the Efficacy and Safety of PDR001 in Patients With Advanced or Metastatic, Well-Differentiated, Non-Functional NET of Pancreatic, Gastrointestinal (GI), or Thoracic Origin or Poorly-Differentiated Gastroenteropancreatic Neuroendocrine Carcinoma (GEP-NEC) Who Have Progressed on Prior Treatment

J.C. Yao[1], N. Fazio[2], M.E. Pavel[3], J. Strosberg[4], E. Bergsland[5], P. Ruszniewski[6], M. Voi[7], C. Wu[7], E. Degtyarev[8], P. Aimone[8], S. Singh[9]

[1]University of Texas/MD Anderson Cancer Center, Houston, Texas, USA; [2]European Institute of Oncology, Milan, Italy; [3]Dept. of Medicine 1, Friedrich-Alexander University Erlangen-Nürnberg, Erlangen, Germany; [4]Department of Medicine, Moffitt Cancer Center, Tampa, Florida, USA; [5]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, USA; [6]Gastroenterology and Pancreatology Department, Beaujon Hospital, Clichy, France; [7]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA; [8]Novartis Pharma AG, Basel, Switzerland; [9]Sunnybrook Health Sciences Centre, Toronto, Ontario, Canada

Introduction

- Monoclonal antibody (mAb) inhibitors of immune checkpoints, including anti-programmed cell death protein-1 (PD-1) and its ligands PD-L1/2, have become established treatment options in various solid tumors.[1]
- Although many advances with checkpoint inhibitors have been made in other tumor types, the data generated in neuroendocrine tumors (NETs) are limited.
- PD-1 is a receptor, expressed on activated T cells and other lymphocytes, which negatively regulates T-cell function.[2]
 - Binding of PD-1 by PD-L1 or PD-L2 results in the inhibition of T-cell function, and the protection of tumor cells from attack by cytotoxic T cells[3]
 - Tumors can evade the immune system by upregulating expression of PD-L1[4]
- PD-L1 overexpression has been associated with poorer prognosis in some tumor types.[5-8]
- In GEP-NET tumors PD-L1 expression was found to be significantly associated with high grade tumors (Grade 3).[9]
- In preclinical models, PD-1 blockade has been shown to restore effector T-cell function, leading to T-cell proliferation, interferon-γ secretion, and increased cytolytic function.[10,11]
- PDR001 is a high-affinity, humanized IgG4 antibody directed against PD-1 that blocks the binding of PD-L1 and PD-L2 to PD-1.[12-16] (**Figure 1**).
- In a phase I trial of PDR001 (mAb checkpoint inhibitor targeting PD-1) conducted in patients with multiple solid tumor types, a patient with histologically confirmed metastatic atypical pulmonary carcinoid demonstrated a partial response with (according to RECIST v1.1) resolution of liver and pleural metastases.[16]

Figure 1. Mechanism of Action of PDR001

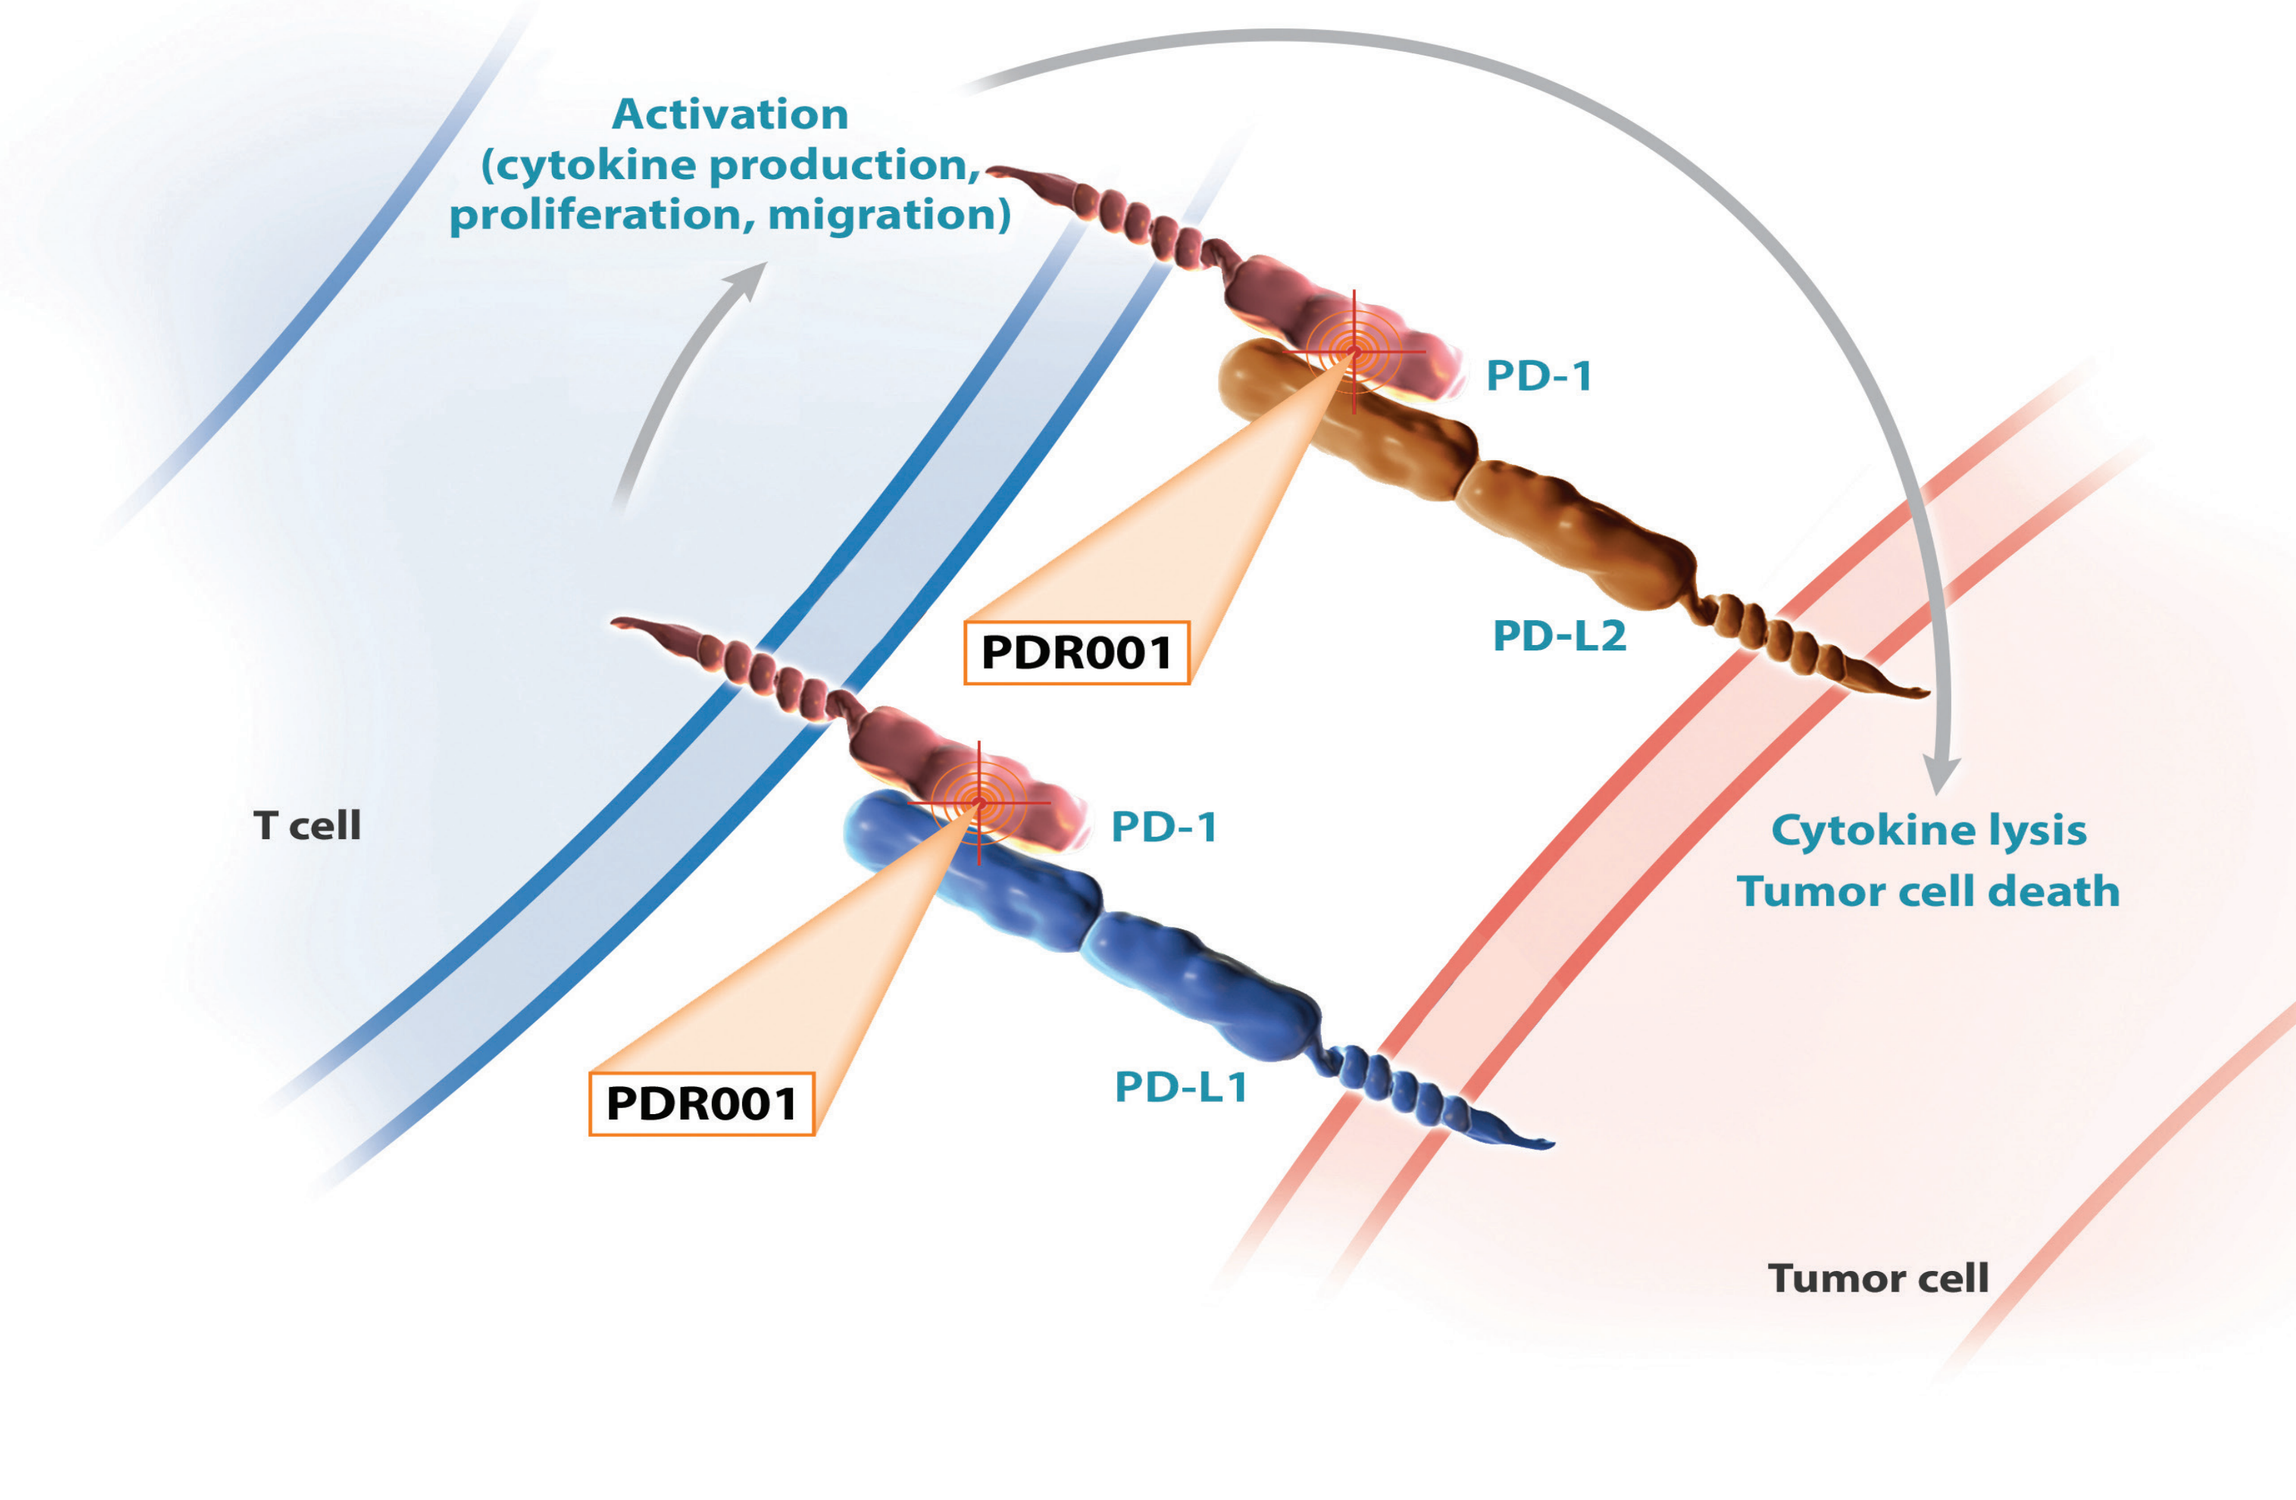

Activation
(cytokine production, proliferation, migration)
PD-1
PDR001
PD-L2
T cell
PD-1
Cytokine lysis
Tumor cell death
PDR001
PD-L1
Tumor cell

Methods

Study Design

- This is a phase II (NCT02955069), single-arm, open-label, multicenter study in patients with advanced or metastatic, well-differentiated, non functional, NET of pancreatic, gastrointestinal (GI) or throacic origin or poorly-differentiated gastroenteropancreatic neuroendocrine carcinoma (GEP-NEC) who have progressed on or after their last treatment (**Figure 2**).
- Patients will receive PDR001 (400 mg, once every 4 weeks) administered via intravenous (IV) infusion over 30 minutes until disease progression, unacceptable toxicity, or patient withdrawal.

Figure 2. Study Design

Patients:

- Advanced, non functional, well-differentiated NET of pancreatic or GI origin (grade 1 or 2) or thoracic (typical or atypical carcinoid) origin

OR

- Poorly-differentiated GEP-NEC
- ECOG status 0-2
- Measurable disease as per RECIST 1.1

→

Total (N=110)

GI Cohort (n=30)

Pancreatic Cohort (n=30)

Thoracic Cohort (n=30)

GEP-NEC Cohort (n=20)

→

PDR001 400 mg Q4W, flat dose until:

- Confirmed PD per RECIST 1.1
- Toxicity
- Patient withdrawal

No dose modifications allowed

Dose may be interrupted for up to 12 weeks

ECOG, Eastern Cooperative Oncology Group; GEP-NEC, gastroenteropancreatic neuroendocrine carcinoma; GI, gastrointestinal; PD, progressive disease; pNET, pancreatic neuroendocrine tumor; Q4W, every 4 weeks; RECIST, Response Evaluation Criteria In Solid Tumors.

Study Endpoints

Primary endpoint

- Overall Response Rate (ORR) per RECIST 1.1 and Blinded Independent Review Committee (BIRC)

Key secondary endpoint

- Duration of Response (DOR) per RECIST 1.1 and BIRC

Additional secondary endpoints

- Disease Control Rate (DCR) per RECIST 1.1 and BIRC
- Time to Response (TTR) per RECIST 1.1 and BIRC
- Progression-free Survival (PFS) per RECIST 1.1 and BIRC
- 1-year and 2-year overall survival rates
- Efficacy endpoints based on immune response criteria per immune-related RECIST and BIRC (irORR, irDOR, irDCR, irTTR, irPFS)
- Changes from baseline in chromogranin A and neuron specific enolase
- Pharmacokinetic (PK) parameters
- Global health status/Quality of Life (QoL) score of the EORTC QLQ-C30 and the index score of the EQ-5D-5L
- Antidrug antibodies (ADA) prevalence at baseline and ADA incidence on-treatment
- Frequency and severity of adverse events (AEs)

Key Inclusion Criteria

- Pathologically confirmed, advanced (unresectable or metastatic):
 - Well-differentiated (Grade 1 or Grade 2) based on local pathology report, non-functional NET of GI or pancreatic origin or thoracic (including lung and thymus) origin (typical or atypical carcinoid)
 - Poorly-differentiated GEP-NEC based on local pathology report
- No active symptoms related to carcinoid syndrome during the last 3 months prior to start of study treatment.
- Patients must have received prior treatment for advanced disease:

Well-differentiated NET group

Thoracic (lung and thymus origin) cohort	GI cohort
• Thymus origin: at least one prior systemic therapy according to investigator's choice • Lung origin: at least one prior systemic therapy is required, which must include everolimus	• At least two prior systemic regimens, which must include everolimus

pNET cohort

- At least two prior systemic regimens, which must include everolimus and/or sunitinib.

Poorly-differentiated GEP-NEC group

GEP-NEC cohort

- At least one prior chemotherapy regimen according to Investigator's choice

- Tumor biopsy material must be provided for all patients for the purpose of biomarkers analysis:
 - Well-differentiated NET: Biopsy material must be provided following the diagnosis of metastatic disease. The tumor sample must be collected from a metastatic site not previously irradiated and should preferably be taken within 6 months but not more than 24 months prior to start of study treatment.
 - Poorly-differentiated GEP-NEC: Biopsy material must be collected from the primary tumor or from a metastatic site not previously irradiated, taken not more than 24 months prior to start of study treatment.
- Radiological documentation of disease progression:
 - Well-differentiated NET: disease progression while on/or after the last treatment; progression must have been observed within 6 months prior to start of study treatment (i.e. maximum of 24 weeks from documentation of progression until study entry). Disease must show evidence of progression based on scans performed not more than one year apart.
 - Poorly-differentiated GEP-NEC: disease progression while on/or after prior treatment
- At least one measurable lesion assessed by CT and/or MRI according to RECIST 1.1.

Key Exclusion Criteria

- Well-differentiated, Grade 3 neuroendocrine tumors; poorly differentiated neuroendocrine carcinoma of any origin (other than GEP-NEC) including NEC of unknown origin, adenocarcinoid, and goblet cell carcinoid
- Pretreatment with interferon-γ as last treatment prior to start of study treatment
- Prior treatment for study indication with:
 - Antibodies or immunotherapy within 6 weeks before the first dose of study treatment
 - PRRT administered within 6 months of the first dose
 - Systemic antineoplastic therapy (including cytotoxic chemotherapy, and toxin immune-conjugates) or any experimental therapy within 14 days or 5 half-lives
 - Tyrosine kinase inhibitors within 14 days or 5 half-lives, before the first dose of study treatment
 - Prior PD-1– or PD-L1–directed therapy
 - Cryoablation, radiofrequency ablation, or trans arterial embolization of hepatic metastases within 2 months before the first dose of study treatment
- History of severe hypersensitivity reactions to other mAb which in the opinion of the investigator may pose an increased risk of a serious infusion reaction
- Known history or current interstitial lung disease or non-infections pneumonitis
- Use of somatostatin analogs or any other medications administered to control active symptoms related to carcinoid syndrome during the last 3 months prior to start of study treatment

Study Flow

Screen period: Day –28 to Day –1

↓

Treatment period: PDR001 400 mg Q4W

↓

End of treatment (EOT)

↓

Safety follow-up period: 30, 60, 90, 120, 150 days | **Efficacy follow-up: Until PD per irRECIST; unless PD prior to EOT**

↓

End of post treatment follow-up

↓

Survival follow-up

Statistical Analysis

The primary efficacy variable of the study is ORR, defined as the proportion of patients with best overall response (BOR) of complete response (CR) or partial response (PR), as per blinded independent central review. ORR will be evaluated according to RECIST 1.1. ORR will be calculated separately within well-differentiated NET and poorly-differentiated GEP-NEC groups. No formal hypothesis testing will be conducted.

The response rate within each group with 95% two-sided confidence interval (exact method) will be computed.

Study Enrollment

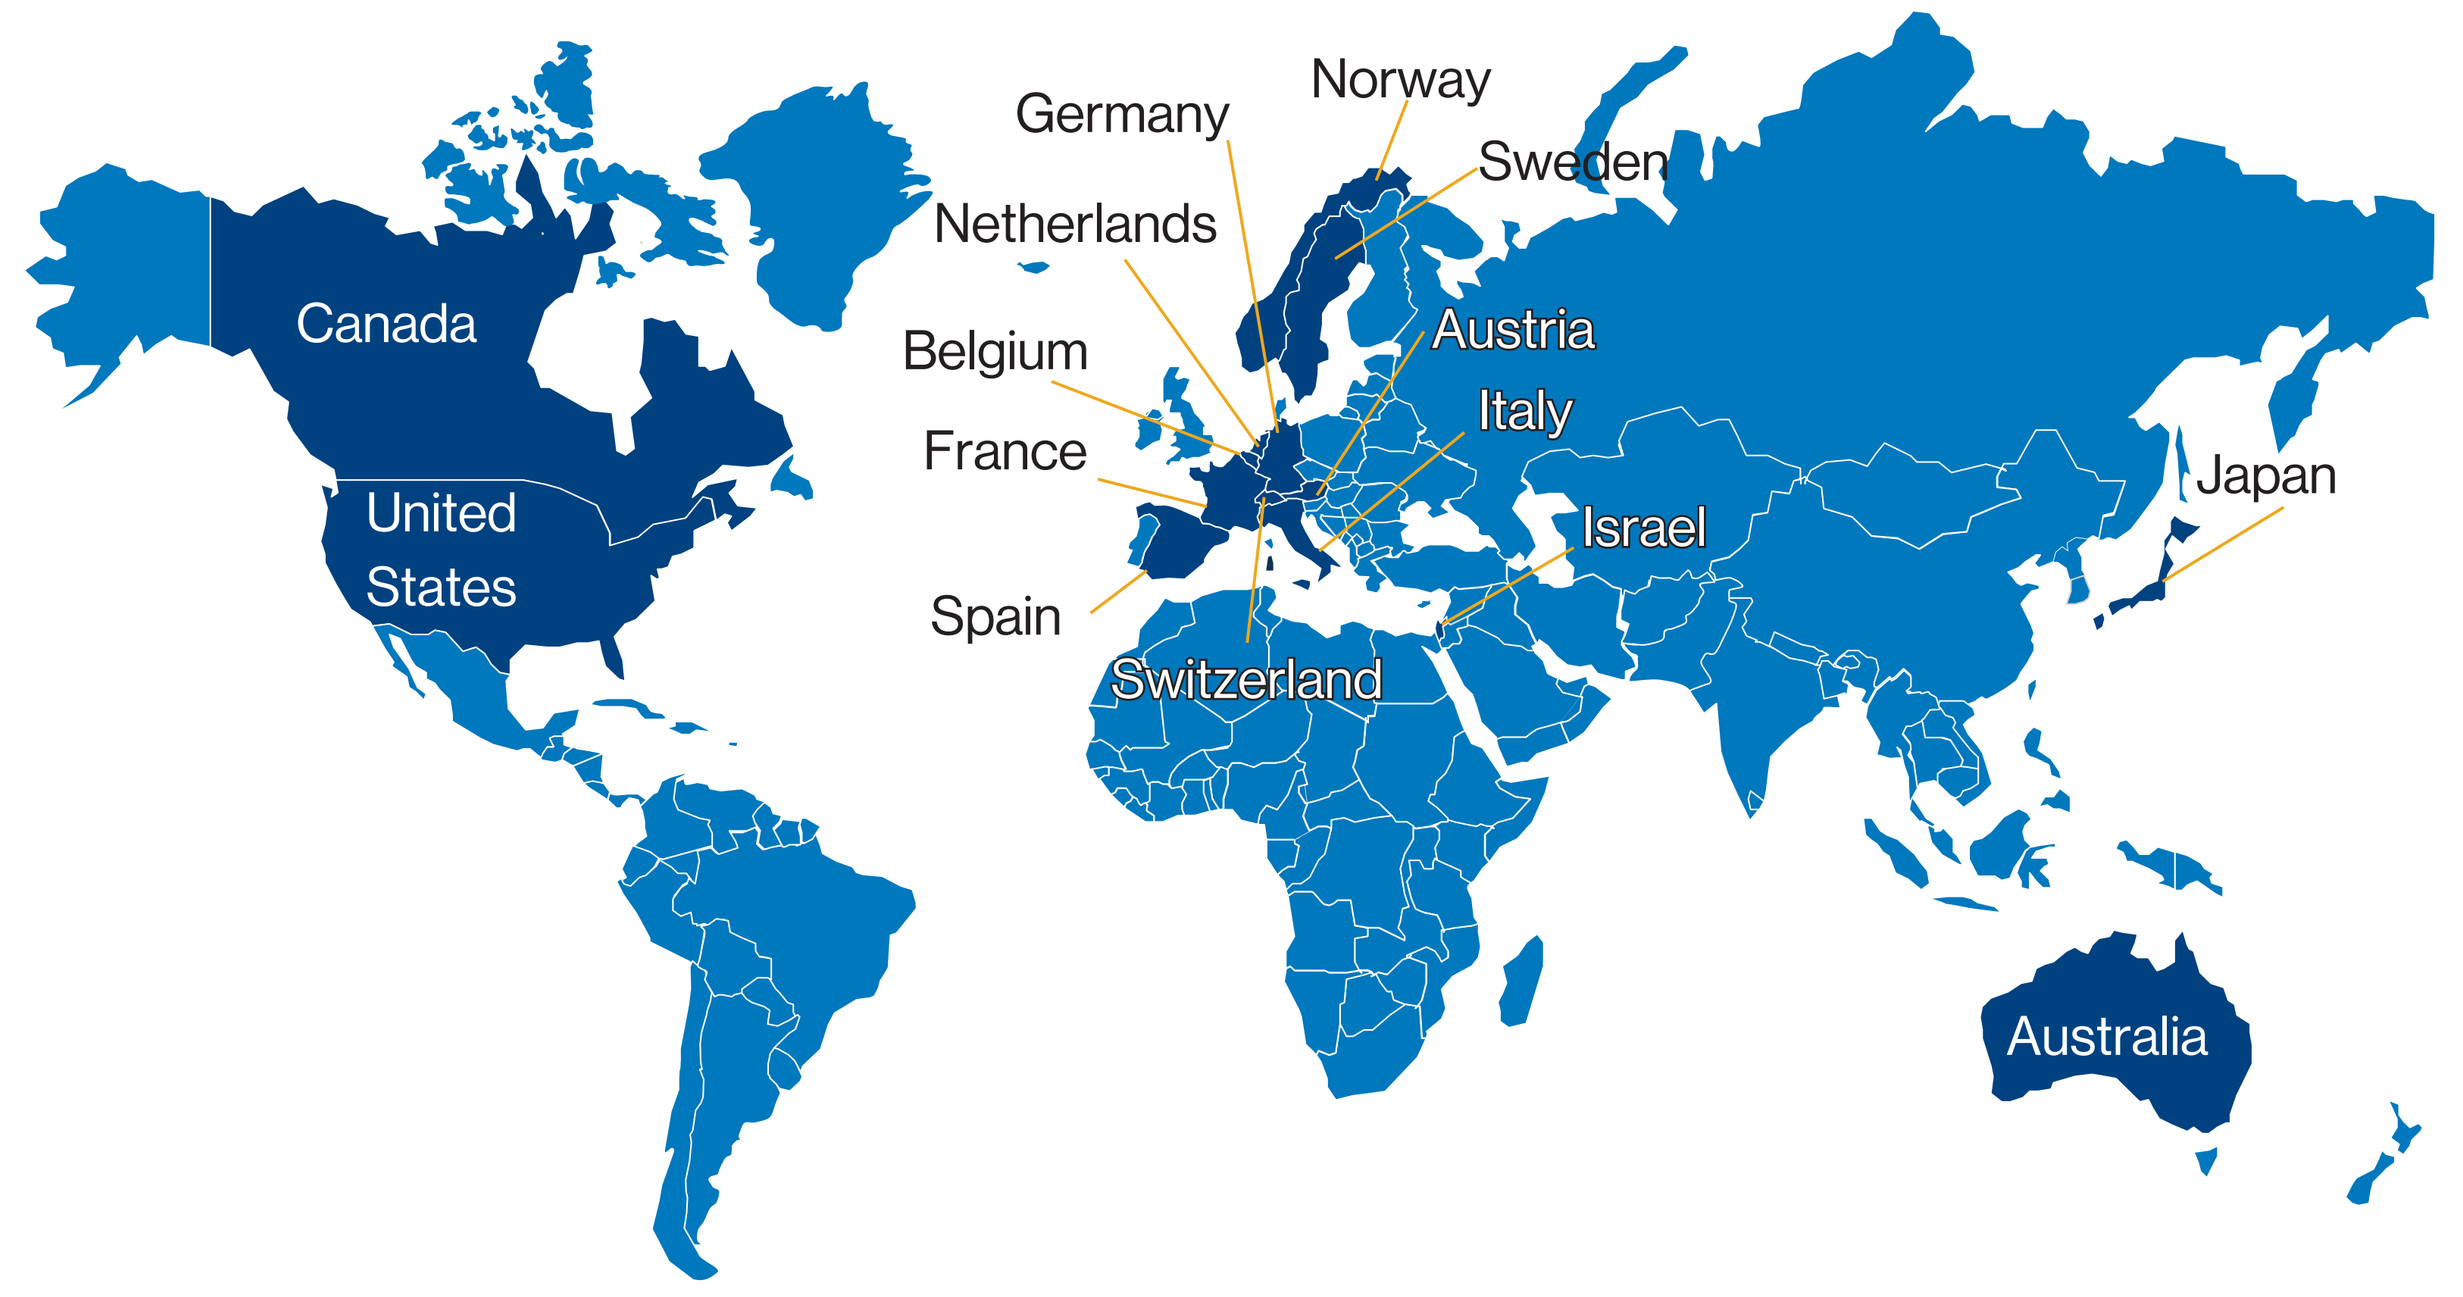

Canada
United States
Germany
Norway
Sweden
Netherlands
Belgium
France
Spain
Austria
Italy
Israel
Switzerland
Japan
Australia

Conclusions

- This phase II, open-label, multicenter study has enrolled patients in United States, Europe, Canada, Australia, Israel, and Japan (NCT02955069) to investigate the role of immunotherapy in NET after prior treatment.

References

1. Topalian SL, et al. *N Engl J Med*. 2012;366:2443-2454.
2. Pardoll DM. *Nat Rev Cancer*. 2012;12:252-264.
3. Chen L, et al. *J Clin Invest*. 2015;125:3384-3391.
4. Dong H, et al. *Nat Med*. 2002;8:793-800.
5. Gao Q, et al. *Clin Cancer Res*. 2009;15:971-979.
6. Thompson RH, et al. *Proc Natl Acad Sci USA*. 2004;101:v17174-v17179.
7. Hino R, et al. *Cancer*. 2010;116:1757-1766.
8. Fan Y, et al. *Onco Targets Ther*. 2016;9:6075-6082.
9. Kim ST, et al. *J Cancer*. 2016;7:1044-1048.
10. Wong RM, et al. *Int Immuno*l. 2007;19:1223-1234.
11. Zhou Q, et al. *J Immunol*. 2010;185:5082-5092.
12. Wang W, et al. *Int Immunol*. 2009;21:1065-1077.
13. Mangsbo SM, et al. *J Immunother*. 2010;33:225-235.
14. Mkrtichyan M, et al. *Eur J Immunol*. 2011;41:2977-2986.
15. Rosenblatt J, et al. *J Immunother*. 2011;34:409-418.
16. Naing A, et al. *J Clin Oncol*. 2016;34:suppl;abstr 3060.

Acknowledgments

We would like to thank the patients and their caregivers for participating in the study. We thank Anamika Gulati from Novartis Healthcare Pvt. Ltd. for providing medical editorial assistance with the poster.

Disclosures

JCY reports consultation fee from Novartis and Ipsen. NF and PR reports no conflicts of interest. MEP reports receiving of research support from Novartis and Ipsen. MEP also served as consultant for Novartis, Ipsen, Lexicon, and Pfizer. EB received research support from Novartis and Merck. JS received honoraria from Novartis, Ipsen, and Lexicon. MV, CW, ED, and PA are employees of Novartis. SS received research support and honoraria from Novartis.

Presented at the North American Neuroendocrine Tumor Society (NANETS) 10th Annual Symposium, October 19-21, 2017, Philadelphia, PA, USA.

This study was funded by Novartis Pharmaceuticals Corporation

Text: Q82a61
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=82a61

Scan this QR code

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.