An international, non-interventional, post-authorization long-term safety study of LUTATHERA®, in patients with unresectable or metastatic, well-differentiated, somatostatin receptor positive gastroenteropancreatic neuroendocrine tumours (SALUS study).

CLICK TO GO BACK TO KIOSK MENU

J. Strosberg[1], M. Caplin[2], S. Navalkissoor[2], J. Erion[3], P. Santoro[3], B. Polack[3], A. Luria[3], E. Sztangret[3], V. Di Gialleonardo[3], V. Nassiri[3], I. Badoi[4], L. Ravasi[4], M. Mariani[4], G. Gericke[4], E. Krenning[5]

[1]Moffitt Cancer Center, Tampa, FL 33612, USA; [2]Royal Free Hospital, London, United Kingdom; [3]Advanced Accelerator Applications, a Novartis Company, New York, NY, USA; [4]Advanced Accelerator Applications, a Novartis Company, Geneva, Switzerland; [5]Erasmus Medical Center, Rotterdam, Netherlands

BACKGROUND

After market authorization, FDA / EMA recommended to assess the long-term safety profile of LUTATHERA® according to the label indication (SmPC/USPI)

Active substance	**INN: lutetium (^{177}Lu) oxodotreotide** **USAN: lutetium Lu 177-DOTATATE** **ATC code: V10XX04**
Medicinal product	LUTATHERA® 370 MBq/mL solution for infusion
Marketing authorization holder	Advanced Accelerator Applications, a Novartis Company 4 rue de la tour de l'île 1204 Geneva- Switzerland

METHODS

Primary research-objective

- **incidence assessment and nature of potential long term second primary malignancies**, including solid tumours and haematological neoplasia, in patients with **unresectable** or **metastatic**, **well-differentiated**, somatostatin receptor positive **GEP-NET**

Secondary research-objectives

- **Risk Management Plan** (RMP) for:
 - renal dysfunction
 - myelosuppression/cytopenias
 - myelodysplastic syndrome
 - hypogonadism
 - sexual dysfunction
 - drug-drug interaction SST
 - tumour cell lysis-related hormone release-induced crises
 - hepatotoxicity, radiotoxicity
- Detection of **potential new risks** overall, and potential risks in patients under-represented in the clinical trial, including elderly patients, patients with renal and liver impairment, reduced bone-marrow reserve, exposure in breast-feeding women, accidental foetal and child exposure.
- Description of **drug patterns utilisation** that may add knowledge about the safety of LUTATHERA®

Inclusion criteria

- Adult patients with **unresectable** or **metastatic**, **well-differentiated**, somatostatin receptor positive **GEP-NET** treated with LUTATHERA®, regardless of the quantity and number of doses administered and whatever the reasons for ending

Exclusion criteria

- Hypersensitivity to LUTATHERA®
- Presence of established or suspected pregnancy or pregnancy not excluded
- Presence of kidney failure with creatinine clearance < 30 mL/min

Follow up of 4 years for prospective study and 4-7 years for retrospective + prospective

All patients will be documented for their second primary tumour and living status during follow-up period from **4 - 7 years**, *via* investigators and/or contact with the patients

Sites and cohort size

~1000 patients

- US : 8 sites
- Europe : 13 sites

An international, non-interventional, post-authorization long-term safety study of LUTATHERA®, in patients with unresectable or metastatic, well-differentiated, somatostatin receptor positive gastroenteropancreatic neuroendocrine tumours (SALUS study).

J. Strosberg[1], M. Caplin[2], S. Navalkissoor[2], J. Erion[3], P. Santoro[3], B. Polack[3], A. Luria[3], E. Sztangret[3], V. Di Gialleonardo[3], V. Nassiri[3], I. Badoi[4], L. Ravasi[4], M. Mariani[4], G. Gericke[4], E. Krenning[5]

[1]Moffitt Cancer Center, Tampa, FL 33612, USA; [2]Royal Free Hospital, London, United Kingdom; [3]Advanced Accelerator Applications, a Novartis Company, New York, NY, USA; [4]Advanced Accelerator Applications, a Novartis Company, Geneva, Switzerland; [5]Erasmus Medical Center, Rotterdam, Netherlands

Data analysis

Descriptive statistics will be presented for the overall full analysis set, and by potential risk factors including

- gender
- age
- bone metastasis
- prior therapies
- prior external-bean radiation therapy

- radiolabeled-SST analogue tumour uptake
- tumour load
- type and grade of toxicity during treatment
- baseline blood counts
- renal function

Continuous variables will be summarized by the number of observations, mean, standard deviation, median, minimum, and maximum.

Categorical variables will be summarized by frequency counts and percentages for each category. Baseline will be the last assessment of the variable under consideration prior to the intake of the first dose of LUTATHERA®.

Planned Milestones

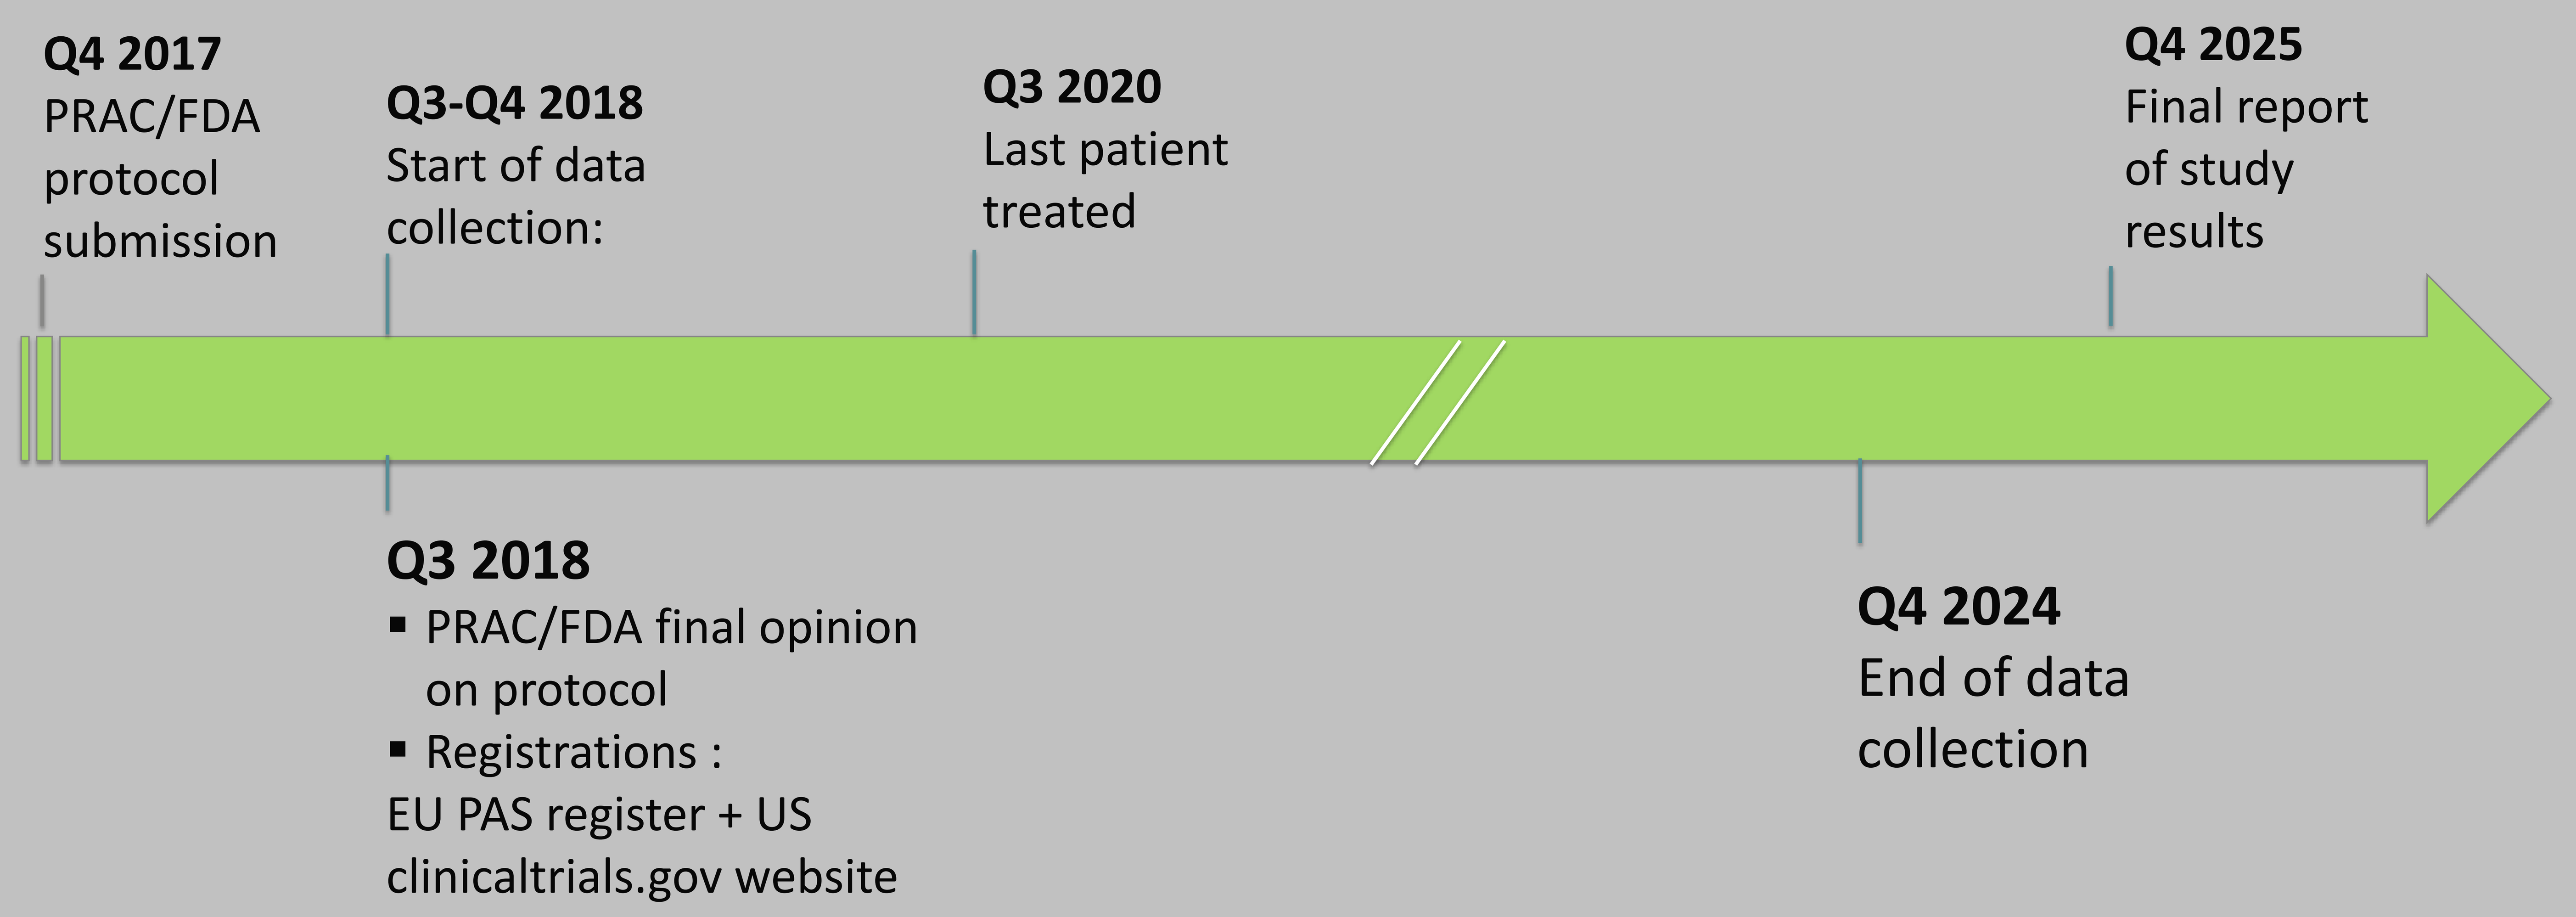
Q4 2017
PRAC/FDA protocol submission
Q3-Q4 2018
Start of data collection:
Q3 2020
Last patient treated
Q4 2025
Final report of study results
Q3 2018
▪ PRAC/FDA final opinion on protocol
▪ Registrations :
EU PAS register + US clinicaltrials.gov website
Q4 2024
End of data collection

Final report to be submitted within 12 months of the end of data collection.

RESULTS

Primary Safety Outcome

The primary safety outcome is **incidence** and type of a second primary cancer (a solid tumour or an haematological cancer).
This outcome will be systematically asked for, searched for in the patient's chart, and collected in the eCRF during each ''data collection point'' over the follow-up.

A **second primary cancer** will be considered as an outcome when the diagnosis is confirmed with appropriate procedures according to the participating physician (data on diagnosis characteristics and procedures not collected).

CONCLUSIONS

The Salus registry shall provide important real world data on long term safety profile and benefits of LUTATHERA®