Co-administration of rA1M during ^{177}Lu-octreotate treatment does not interfere with the therapeutic effect

Charlotte Andersson[1], Emman Shubbar[1], Bo Åkerström[2], Magnus Gram[2,3], and Eva Forssell-Aronsson[1]

[1]Dept of Radiation Physics, Inst of Clinical Sciences, Sahlgrenska Cancer Center, Sahlgrenska Academy at the University of Gothenburg, Gothenburg, Sweden
[2]Lund University, Dept of Clinical Sciences, Infection Medicine, Lund, Sweden
[3]A1M Pharma AB, Lund, Sweden

TAP TO GO BACK TO KIOSK MENU

Background

PRRT with ^{177}Lu-octreotate has yielded promising results in treatment of patients with metastasized NET, but complete tumor remission is scarce. An approach to achieve better tumor control with increased administered activity is to use radioprotectors that limits the side effects on risk organs. A pharmaceutical candidate of alpha-1-microglobulin (rA1M, RMC-035), a human radical scavenger and antioxidant, with the ability to protect tissues from oxidative stress is a conceivable kidney and hematologic protector during PRRT. When introducing changes in a treatment protocol it is crucial to make sure that the changes do not limit the overall effect of the treatment. Adding a radiation protecting agent may result in protective effects not only on the normal tissue but also on the tumor tissue. This study examines co-infusion of rA1M and ^{177}Lu-octreotate in NET-bearing mice with the aim to investigate if rA1M affects therapeutic response to ^{177}Lu-octreotate administration.

Conclusion

Administration of rA1M simultaneously with ^{177}Lu-octreotate does not interfere with the therapeutic effects of ^{177}Lu-octreotate.
rA1M is a promising radioprotector, and further studies should be performed in order to investigate protective renal and hematologic effects of rA1M during PRRT.

Material & Methods (Click)

The biodistribution of ^{177}Lu after injection of either ^{177}Lu-octreotate, or ^{177}Lu-octreotate and rA1M were studied in mice with human medullary thyroid carcinoma, GOT2.
The possible effects of rA1M on tumor volume was studied on mice bearing human small intestine NET, GOT1. Mice were injected with: rA1M or ^{177}Lu-octreotate or both combined.

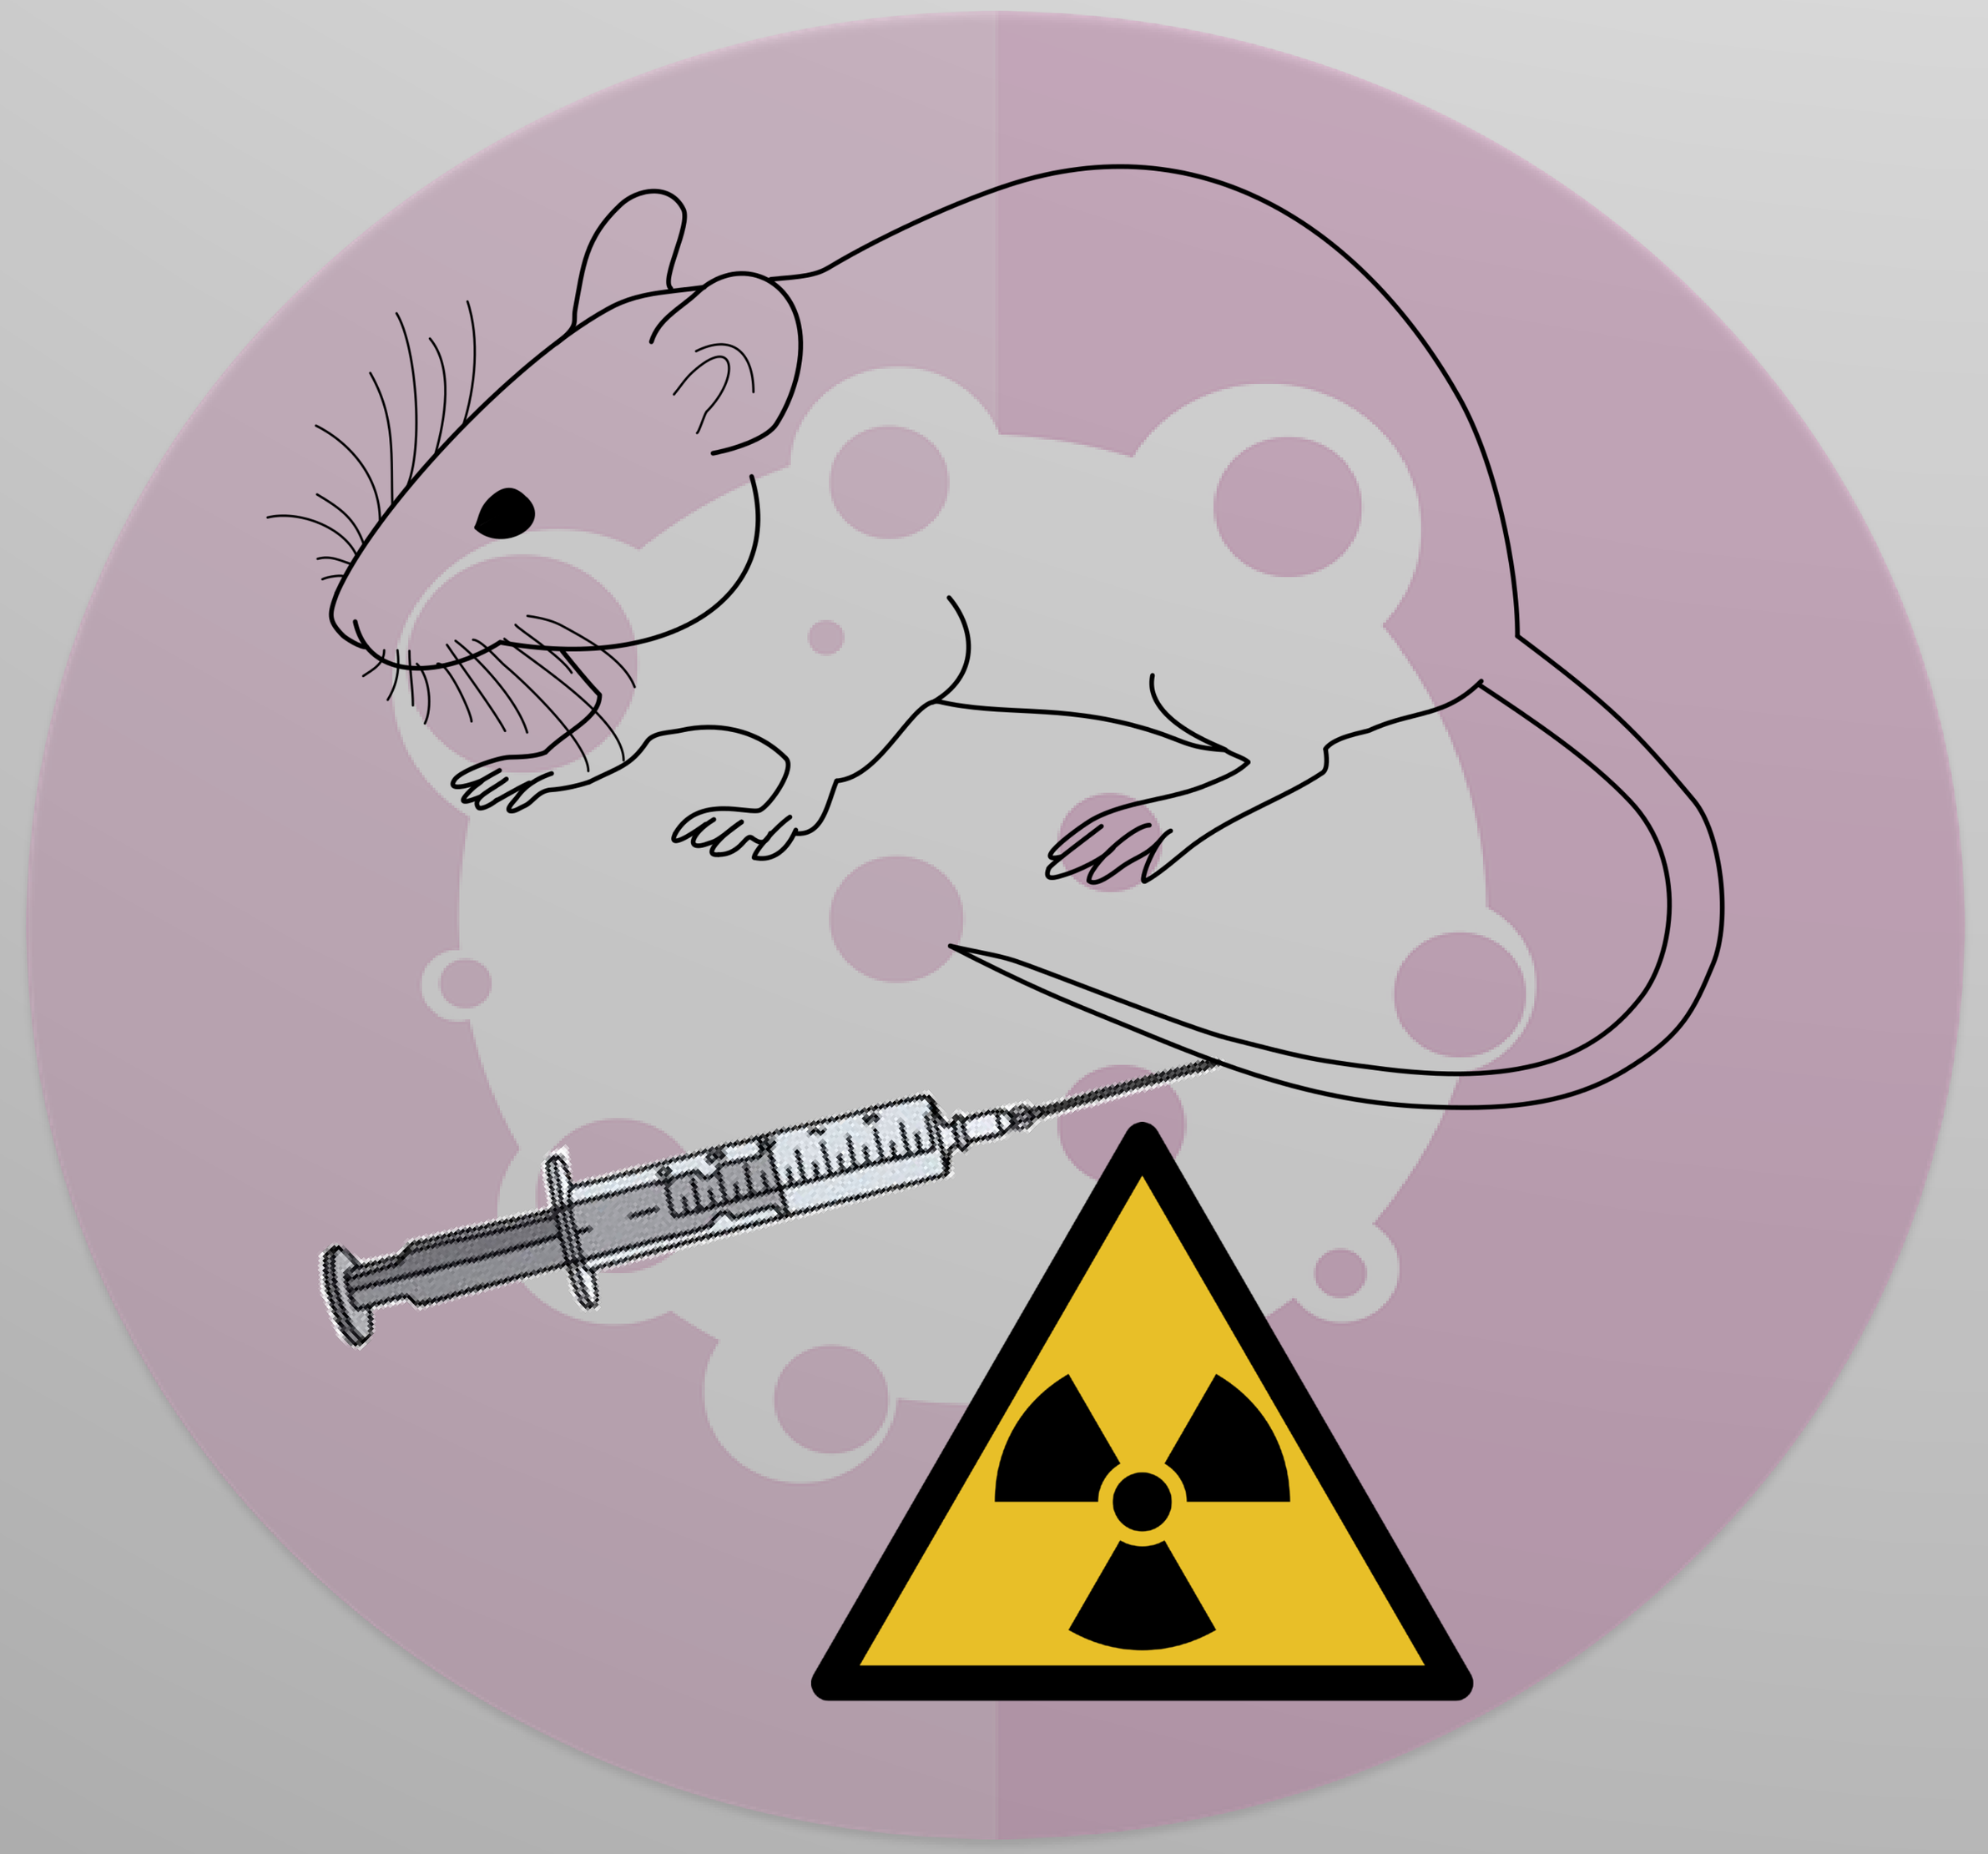

Results (Click)

rA1M showed no negative effect on the:

- biodistribution of ^{177}Lu-octreotate
- therapeutic response of ^{177}Lu-octreotate

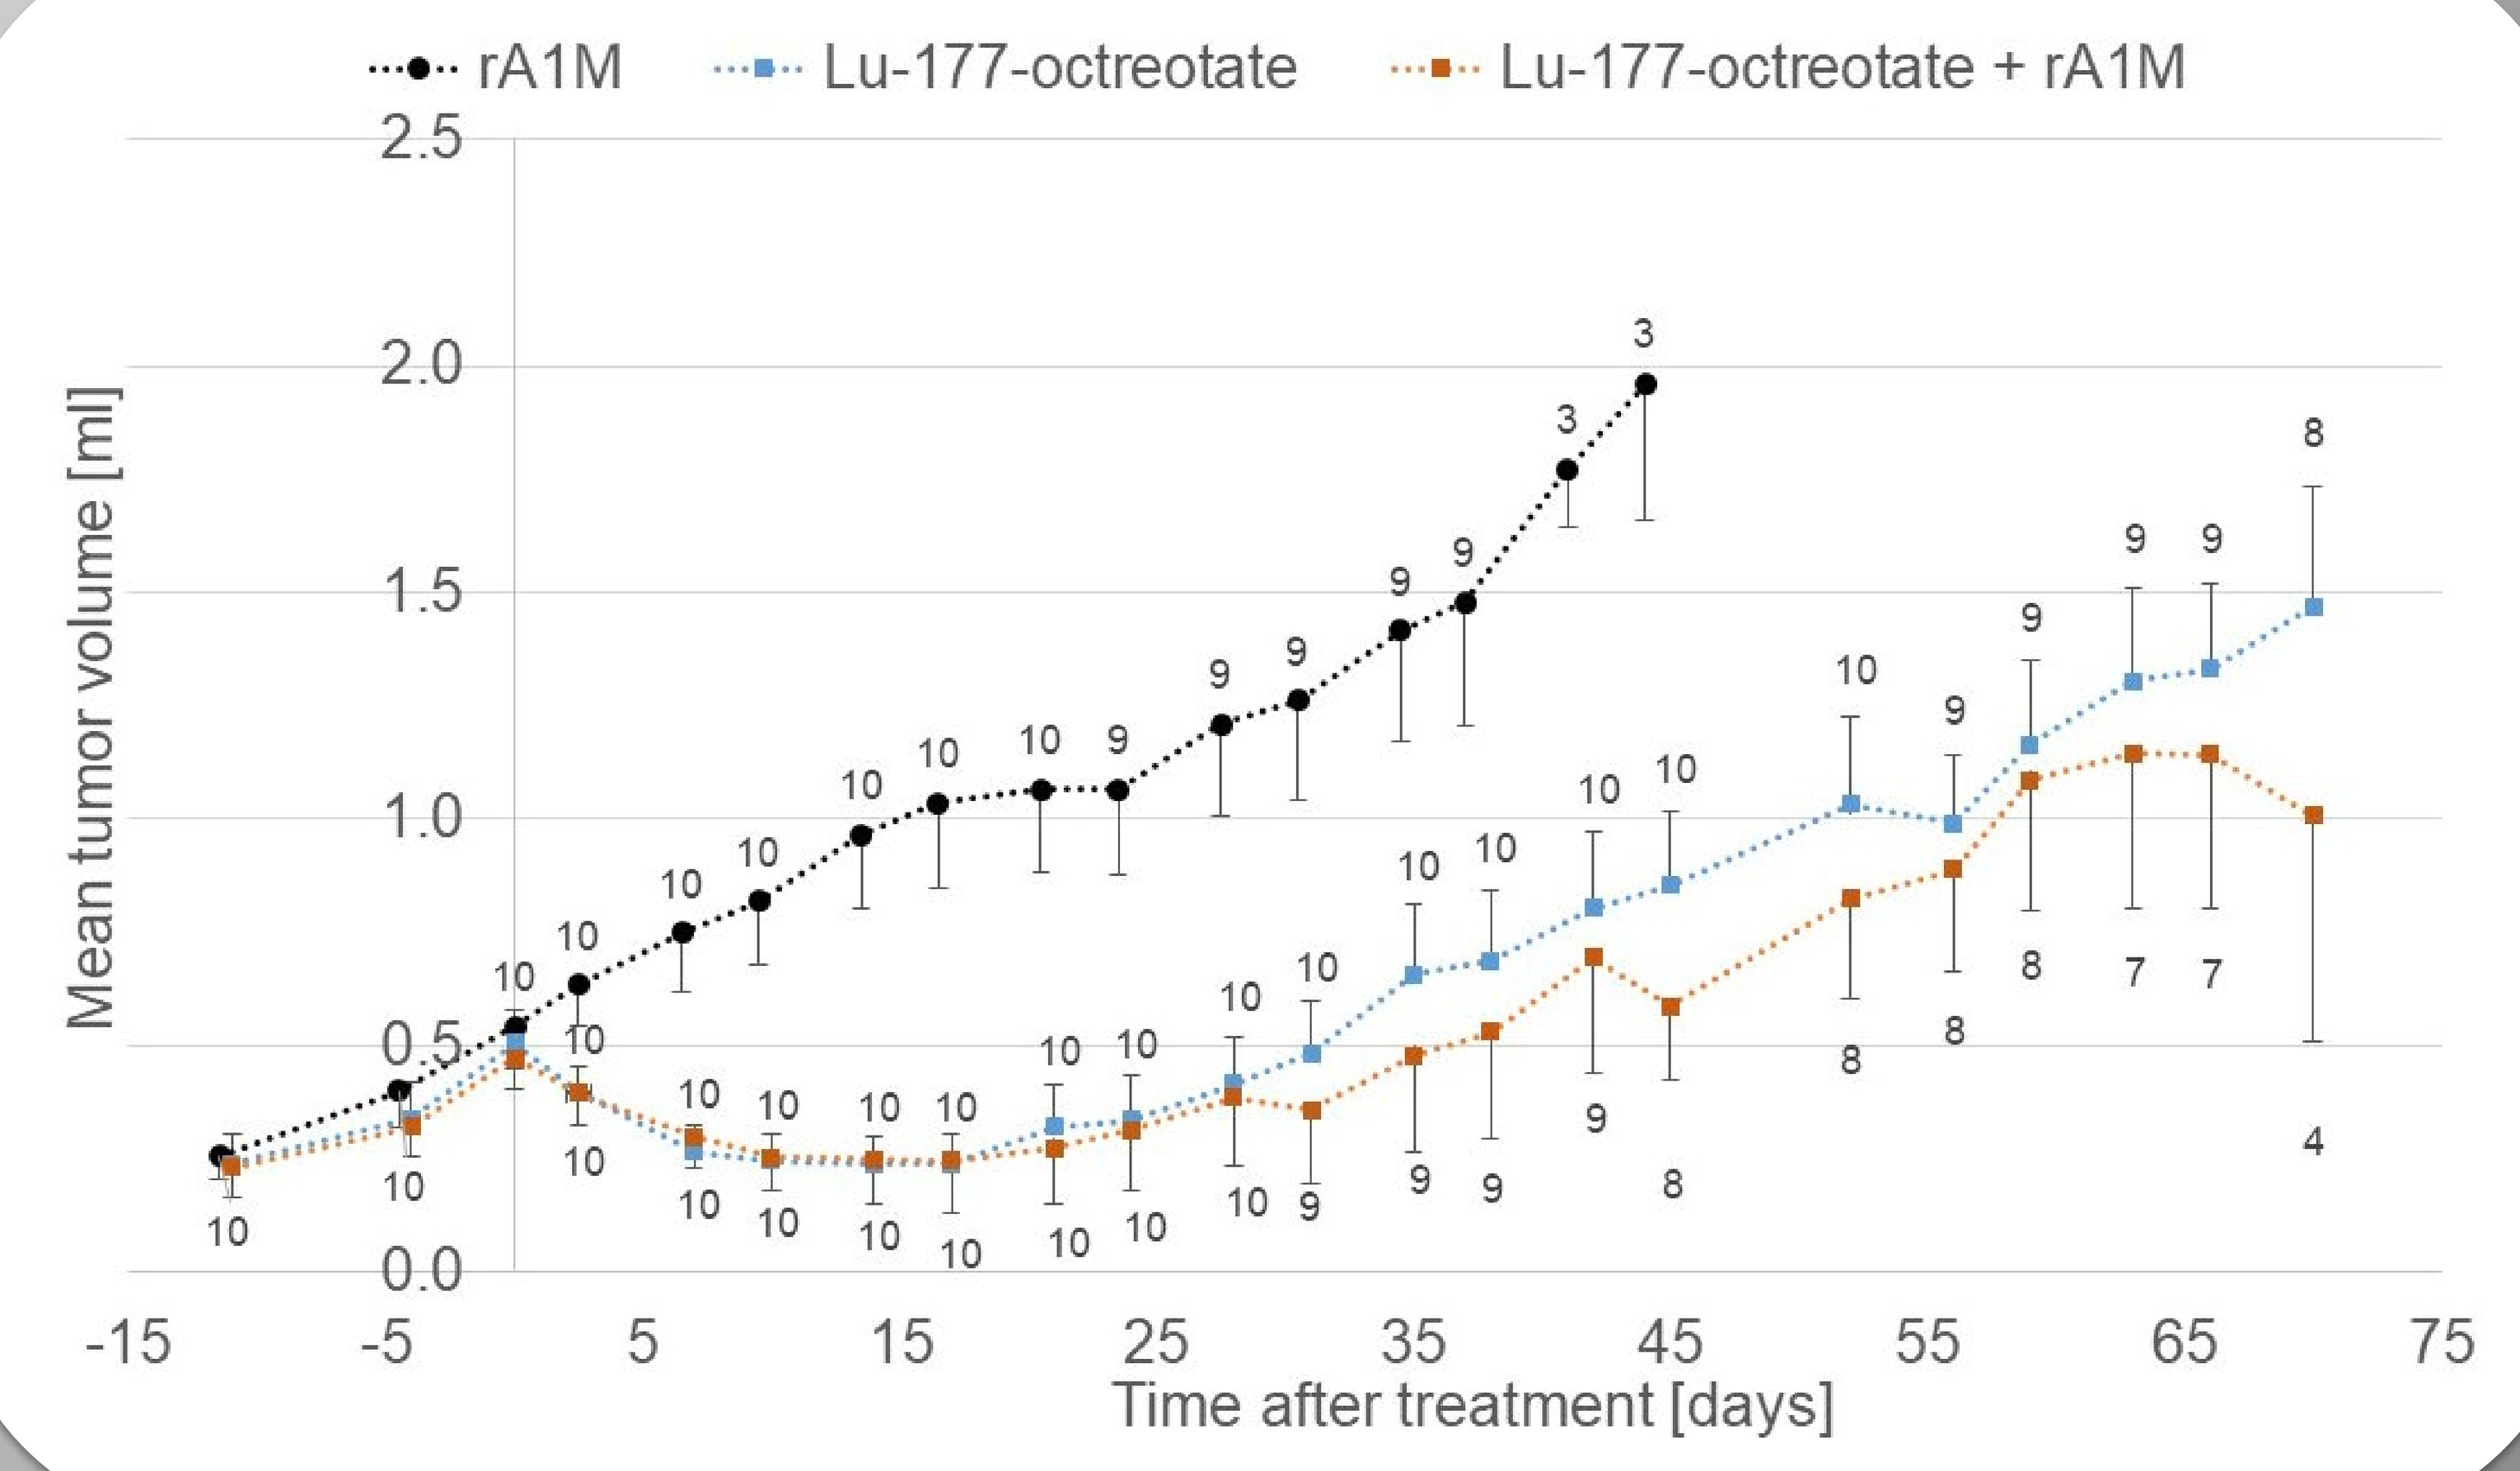
rA1M
Lu-177-octreotate
Lu-177-octreotate + rA1M
Mean tumor volume [ml]
Time after treatment [days]
0.0
0.5
1.0
1.5
2.0
2.5
-15
-5
5
15
25
35
45
55
65
75

Click Headings to View More Information

SAHLGRENSKA CANCER CENTER

GÖTEBORGS UNIVERSITET

Materials and Methods

TAP TO GO BACK TO KIOSK MENU

<u>The biodistribution of ^{177}Lu-octreotate</u> was studied in female nude BALB/c mice bearing human medullary thyroid carcinoma, GOT2. Mice were injected with 5 MBq ^{177}Lu-octreotate or 5 mg/kg rA1M following injection of 5 MBq ^{177}Lu-octreotate. The animals were killed at different time points after administration. Samples of tissues and organs were collected and weighed directly after excision. The ^{177}Lu activity in the samples were measured ex vivo using a gamma counter.

<u>The effect of rA1M on tumor volume</u> alone or in combination with ^{177}Lu-octreotate treatment was studied on female nude BALB/c mice with human small intestine NET, GOT1. A total number of 30 mice were divided into three groups and treated with: 30 MBq ^{177}Lu-octreotate or 5 mg/kg rA1M or both combined (30 MBq ^{177}Lu-octreotate and 5 mg/kg rA1M). The tumor response was followed over time by measuring the tumor volume using a digital slide calipers. Measurement were performed twice a week up to 70 days after administration.

Illustration of experimental setup in the biodistribution study:

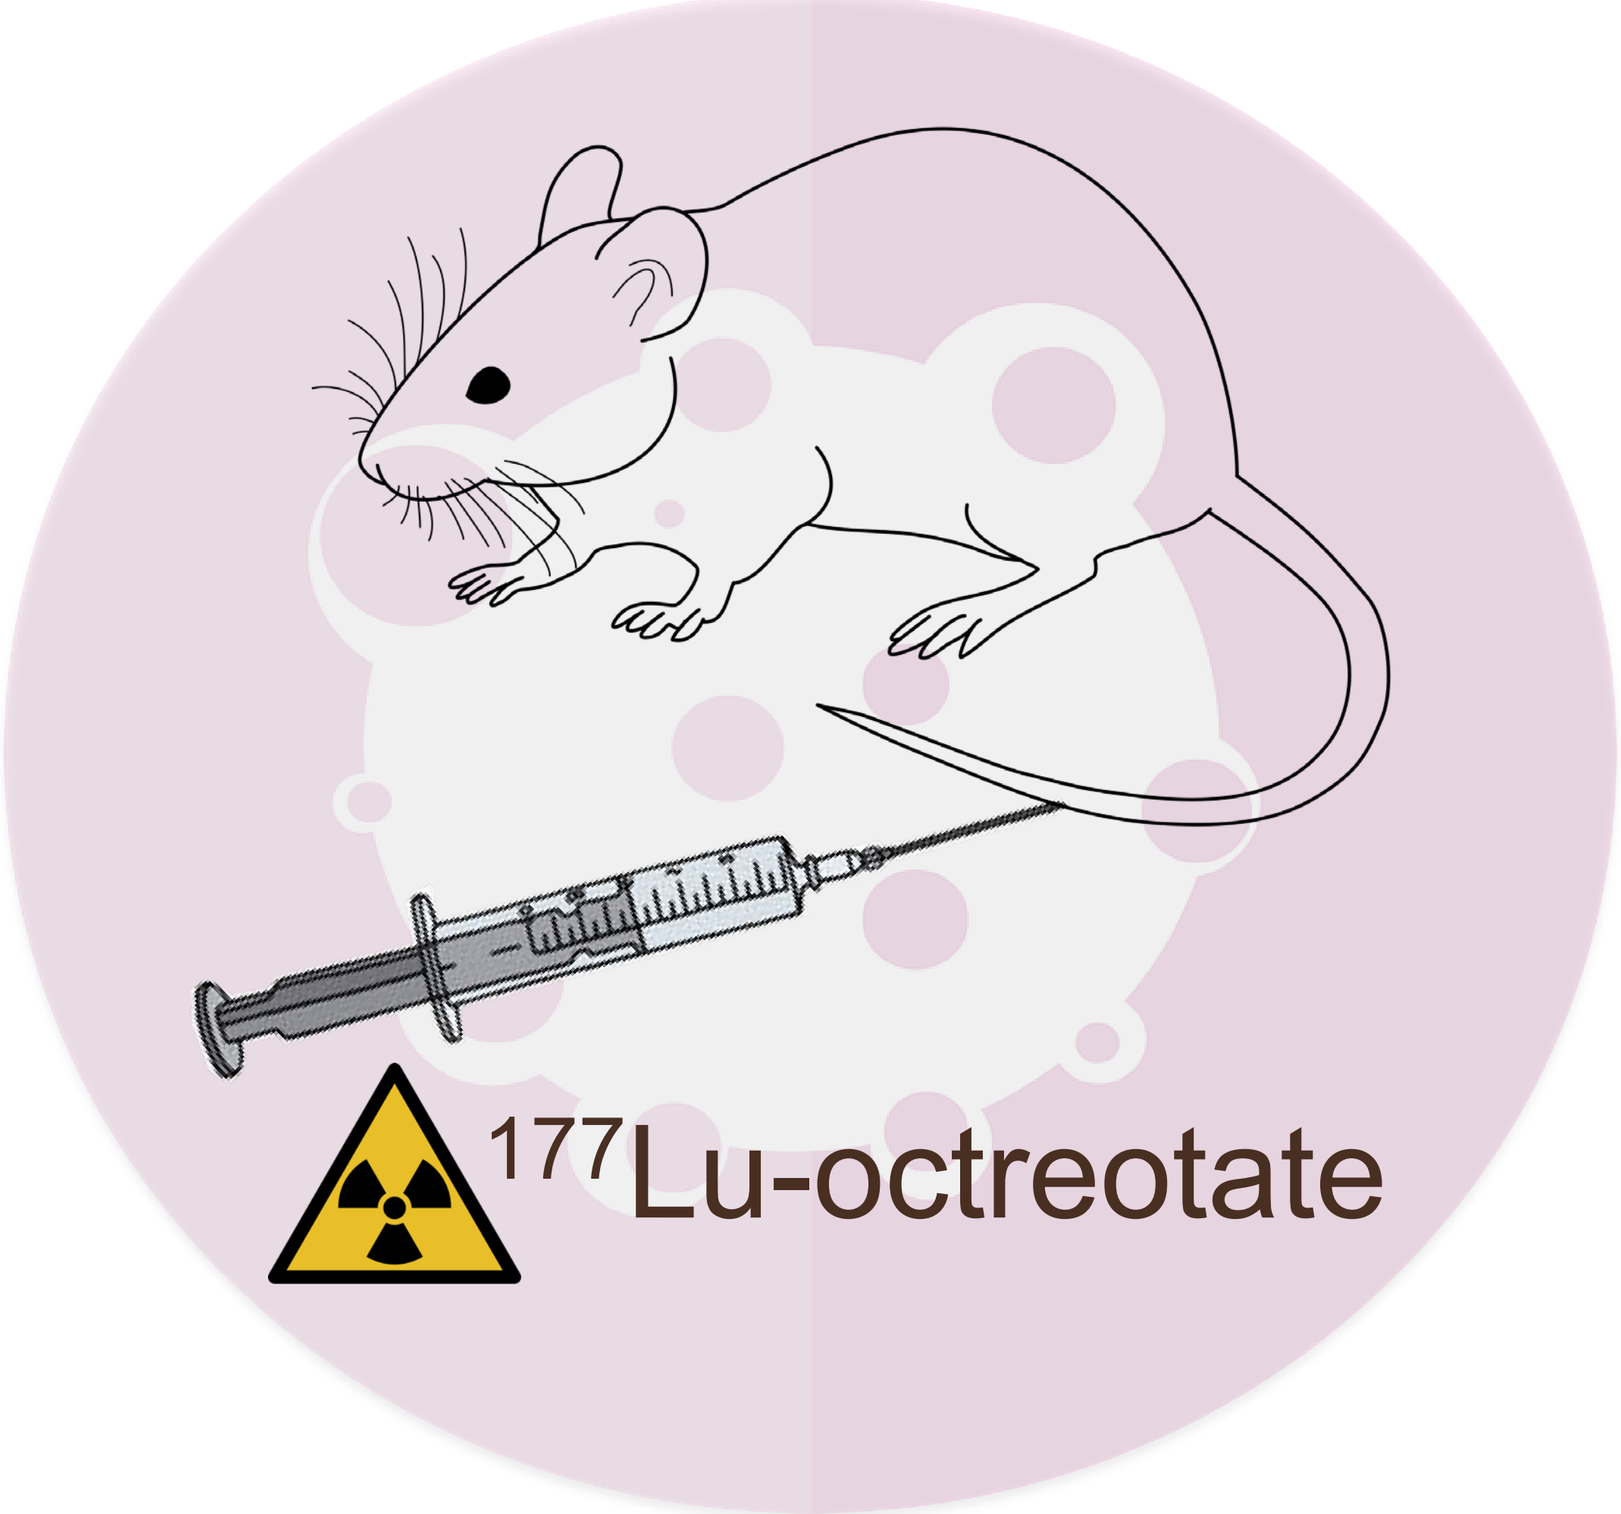

177Lu-octreotate

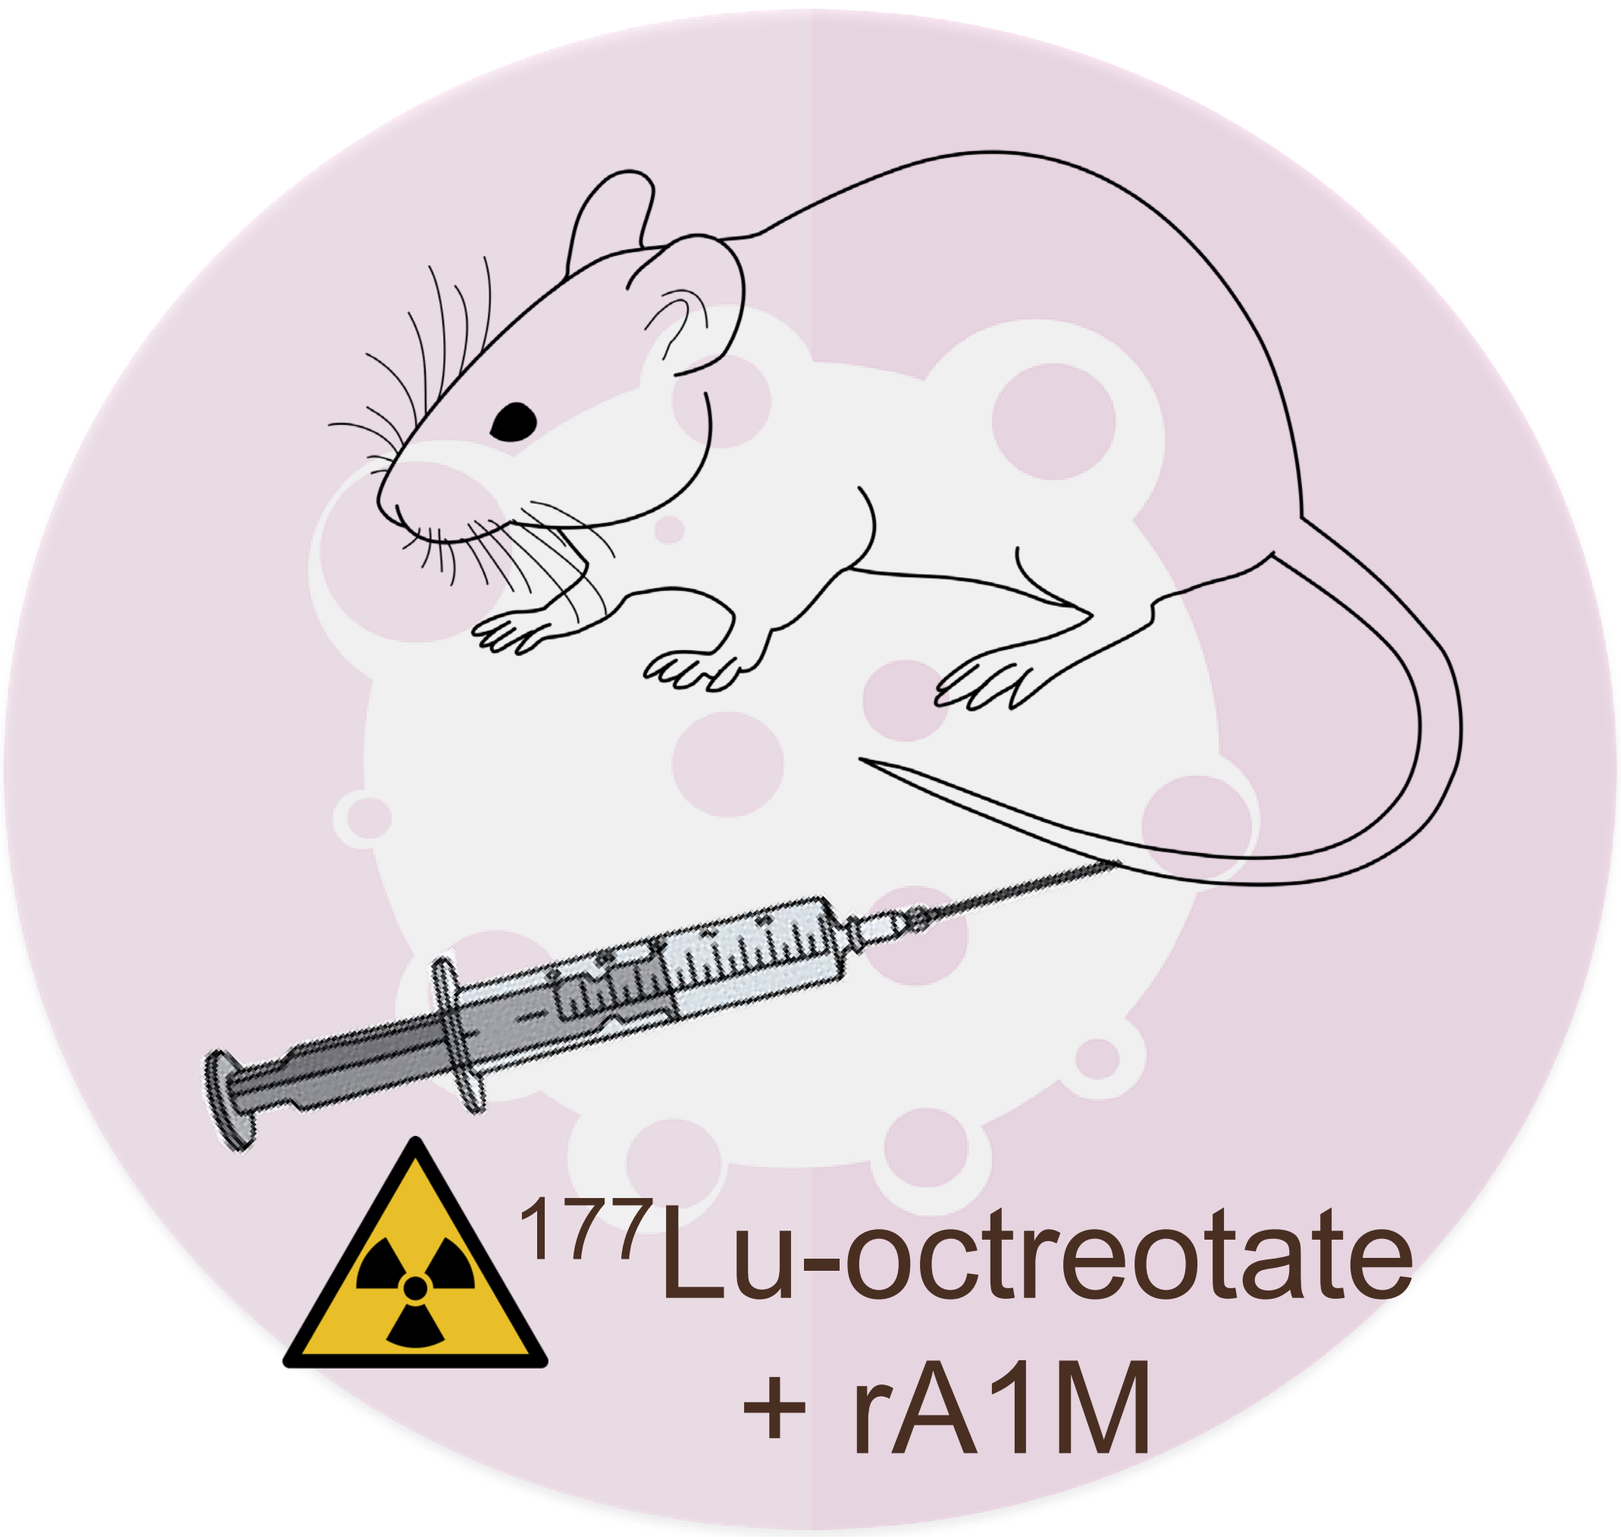

177Lu-octreotate
+ rA1M

Adrenals glands
Bone
Bone marrow
Blood
Liver
Lungs
Pancreas
Spleen
Tumor

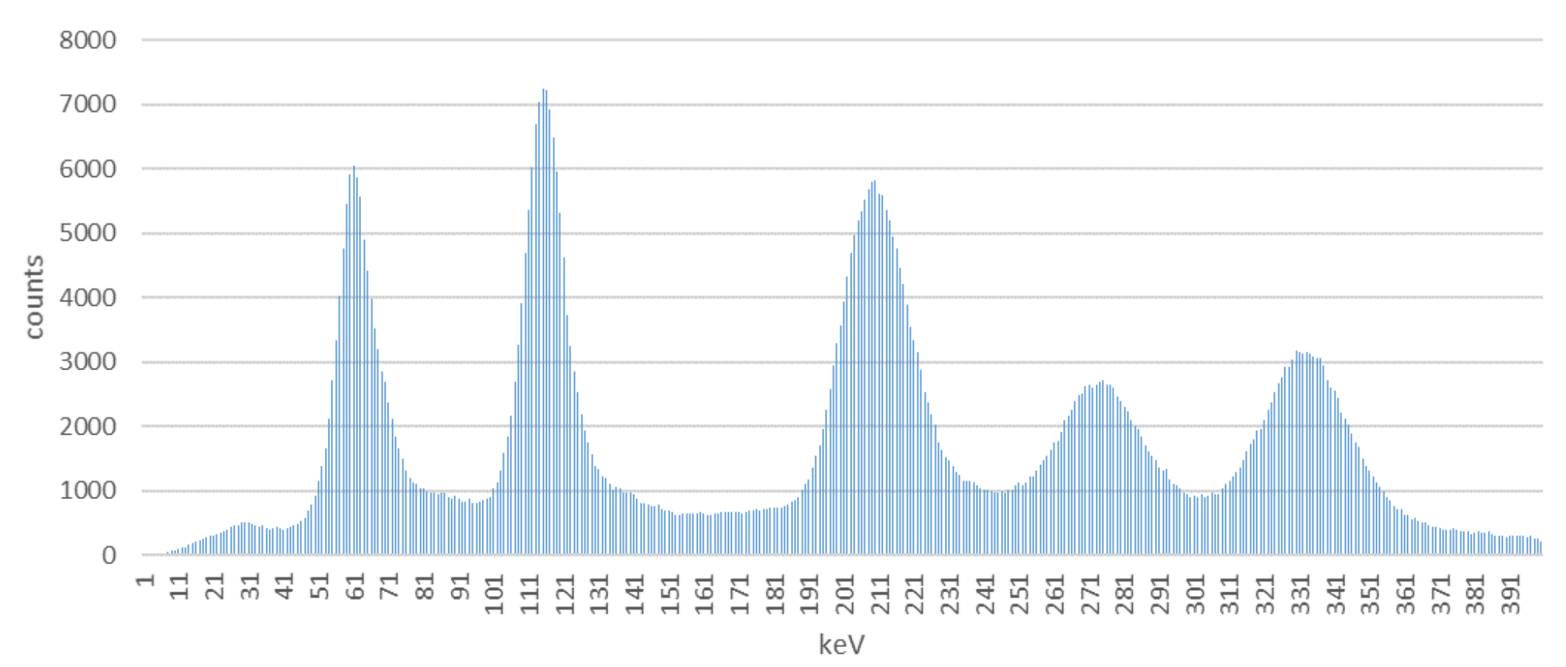

counts
keV

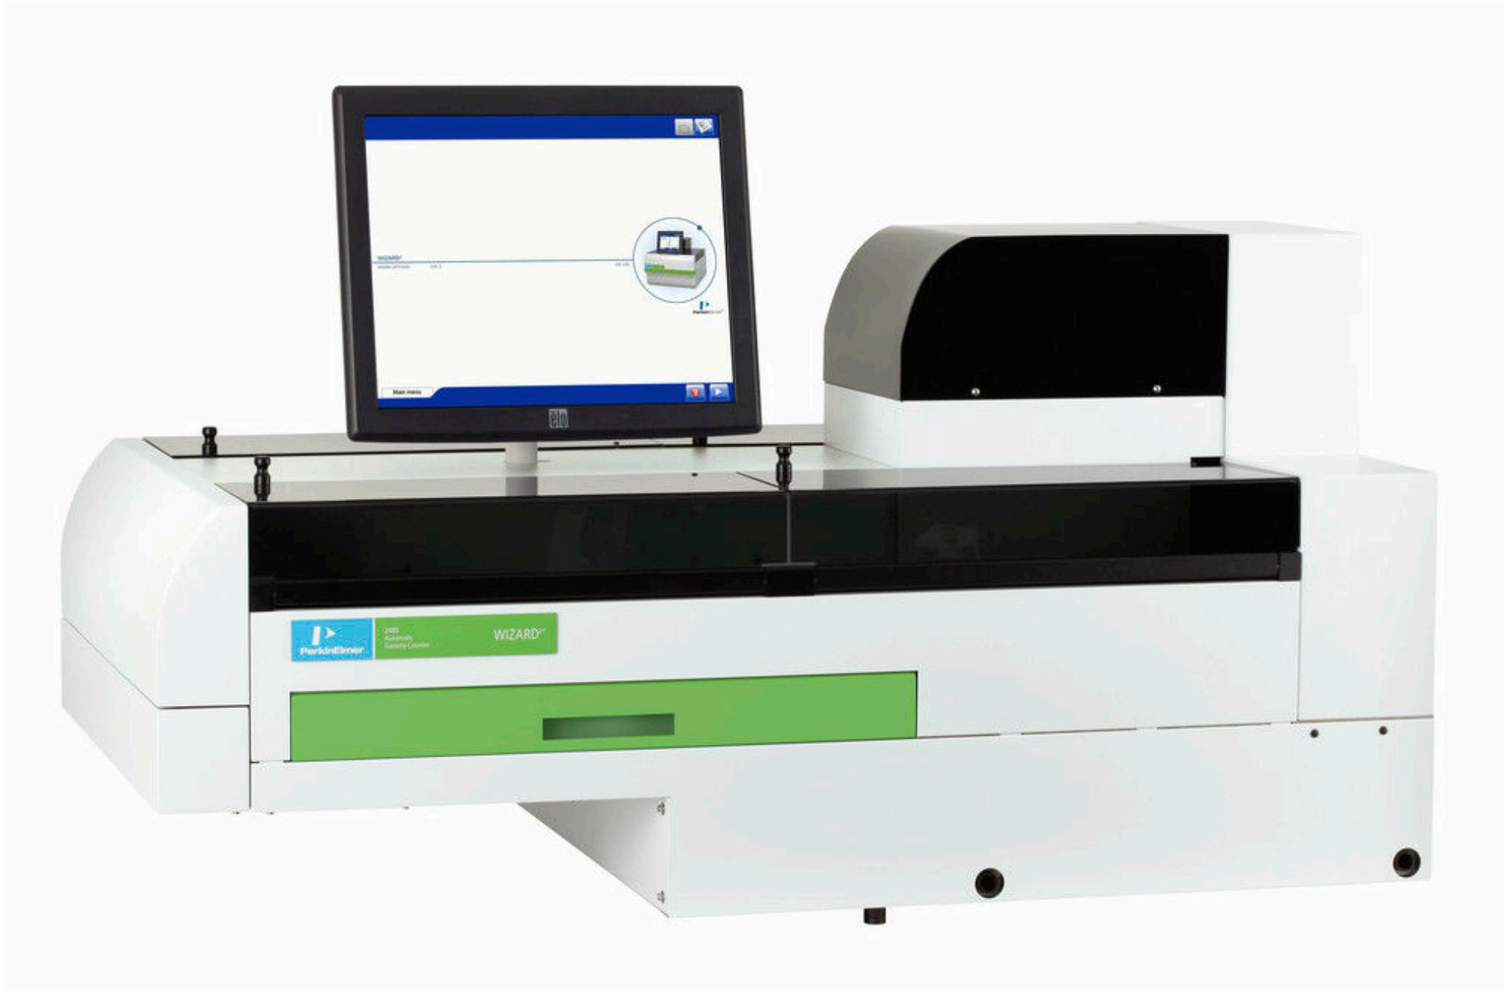

2480 WIZARD 2®
GammaCounter

Illustration of experimental setup in the tumor volume study:

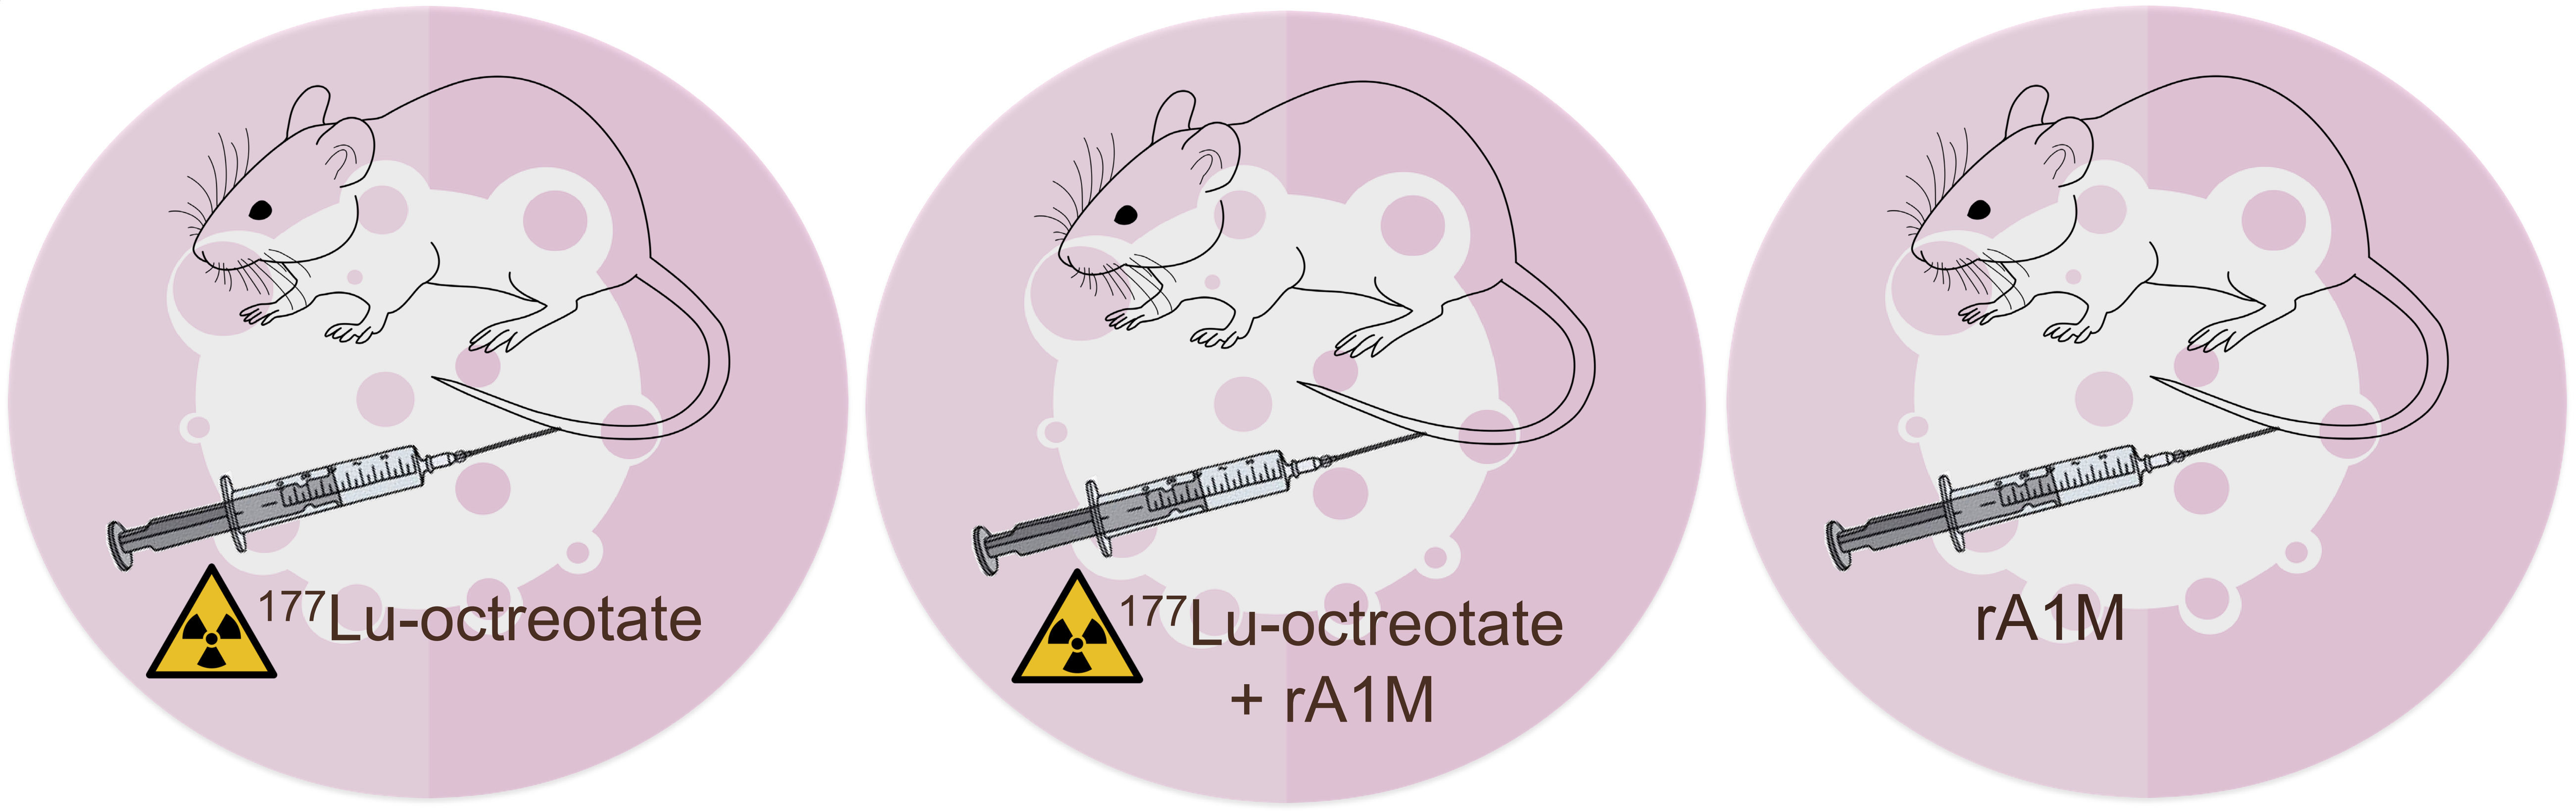

177Lu-octreotate
177Lu-octreotate
+ rA1M
rA1M

$$V = \frac{\pi \cdot a \cdot b \cdot c}{6}$$

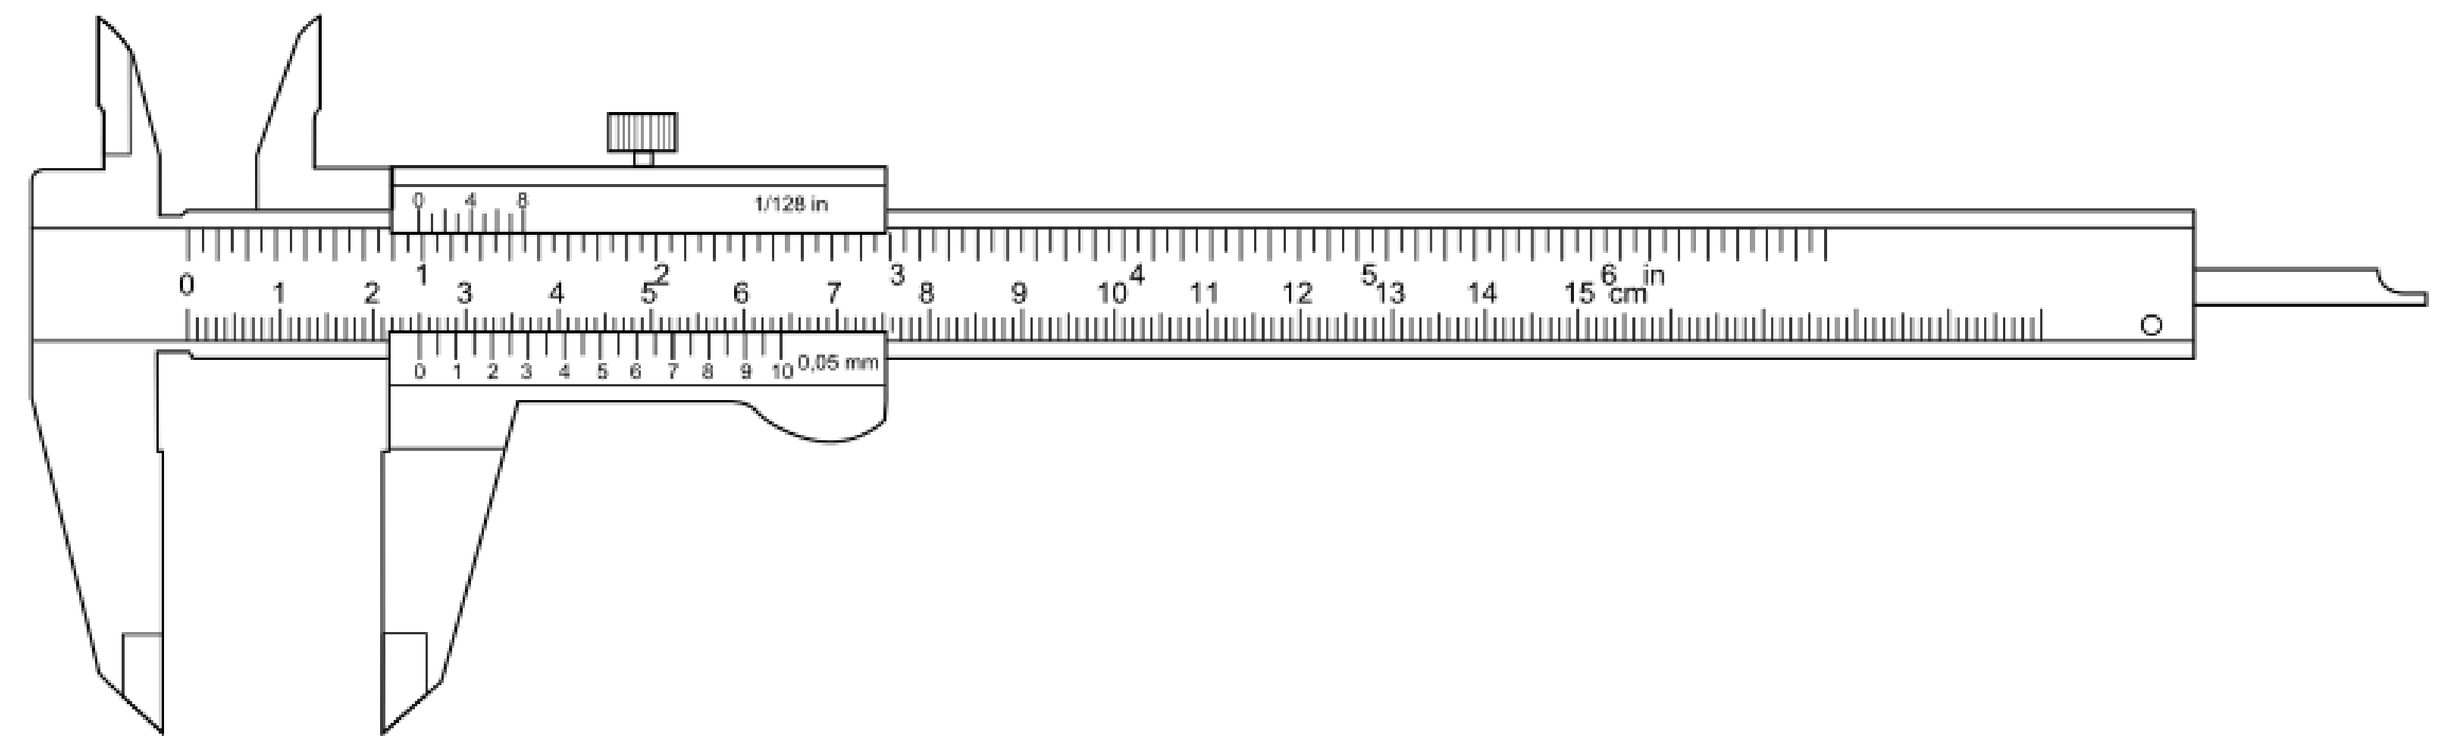

TAP TO GO BACK
TO KIOSK MENU

Results: Biodistribution study

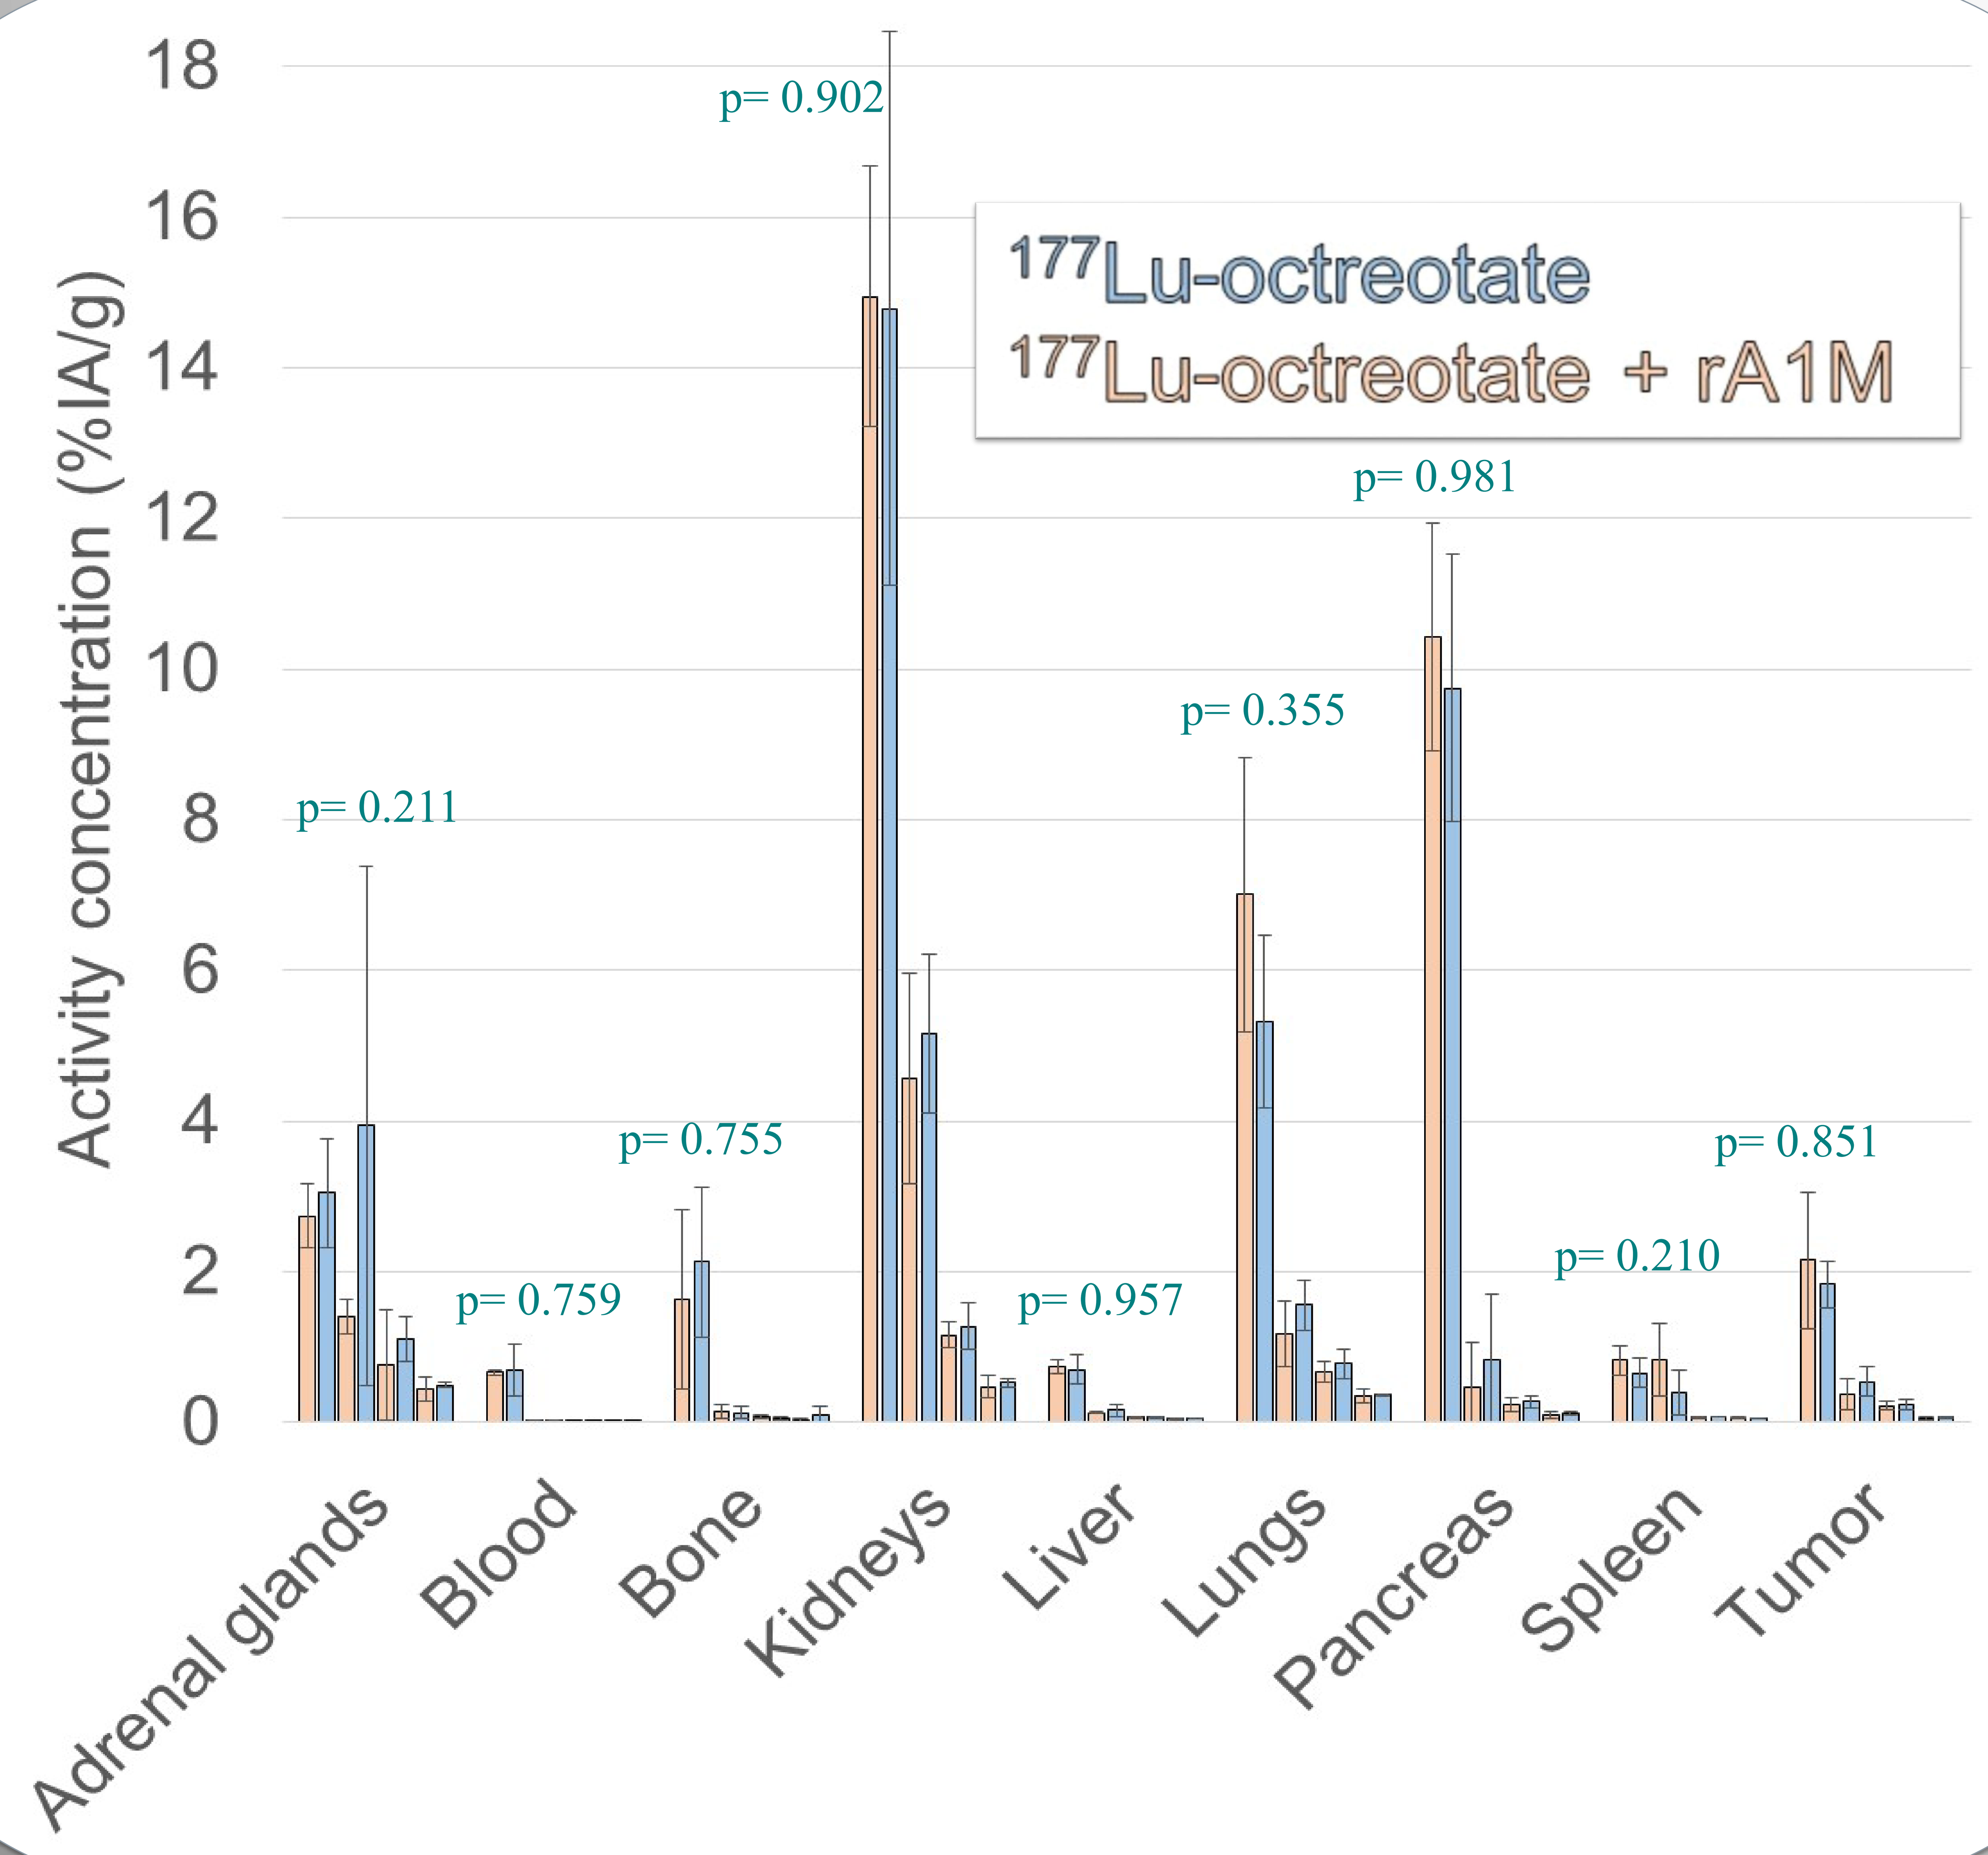
Activity concentration (%IA/g)
18
16
14
12
10
8
6
4
2
0
177Lu-octreotate
177Lu-octreotate + rA1M
p= 0.211
p= 0.759
p= 0.755
p= 0.902
p= 0.957
p= 0.355
p= 0.981
p= 0.210
p= 0.851
Adrenal glands
Blood
Bone
Kidneys
Liver
Lungs
Pancreas
Spleen
Tumor

In almost all investigated organs and tissues the maximum of ^{177}Lu concentration was reached within the first hour after injection for both groups. The activity concentration had a strong time dependence and after maximum the concentration decreased with time. The highest activity concentration of ^{177}Lu was observed in the kidneys.

A two-way ANOVA was used to determine the statistical significance of differences between the different groups. Statistical significance was considered for probabilities higher than 95% ($p<0.05$). The test showed no statistical significance in the activity concentration between the two groups for any tissue at any point-in-time. This demonstrates that co-infusion of rA1M does not change the biokinetics or the biodistribution of ^{177}Lu after ^{177}Lu-octreotate injection.

Figure 1: ^{177}Lu activity concentration, as percent of the injected activity, in mouse organs and tissues over time, at 1 hour, 24, 72 and 168 hours after injection. Pink bars show mean values of 4 mice injected with 5 MBq ^{177}Lu-octreotate and rA1M, blue bars are mean values of 4 mice injected with 5 MBq ^{177}Lu-octreotate and error bars are standard deviations. The p-values show the results from the two-way ANOVA.

Results: Tumor volume study

TAP TO GO BACK
TO KIOSK MENU

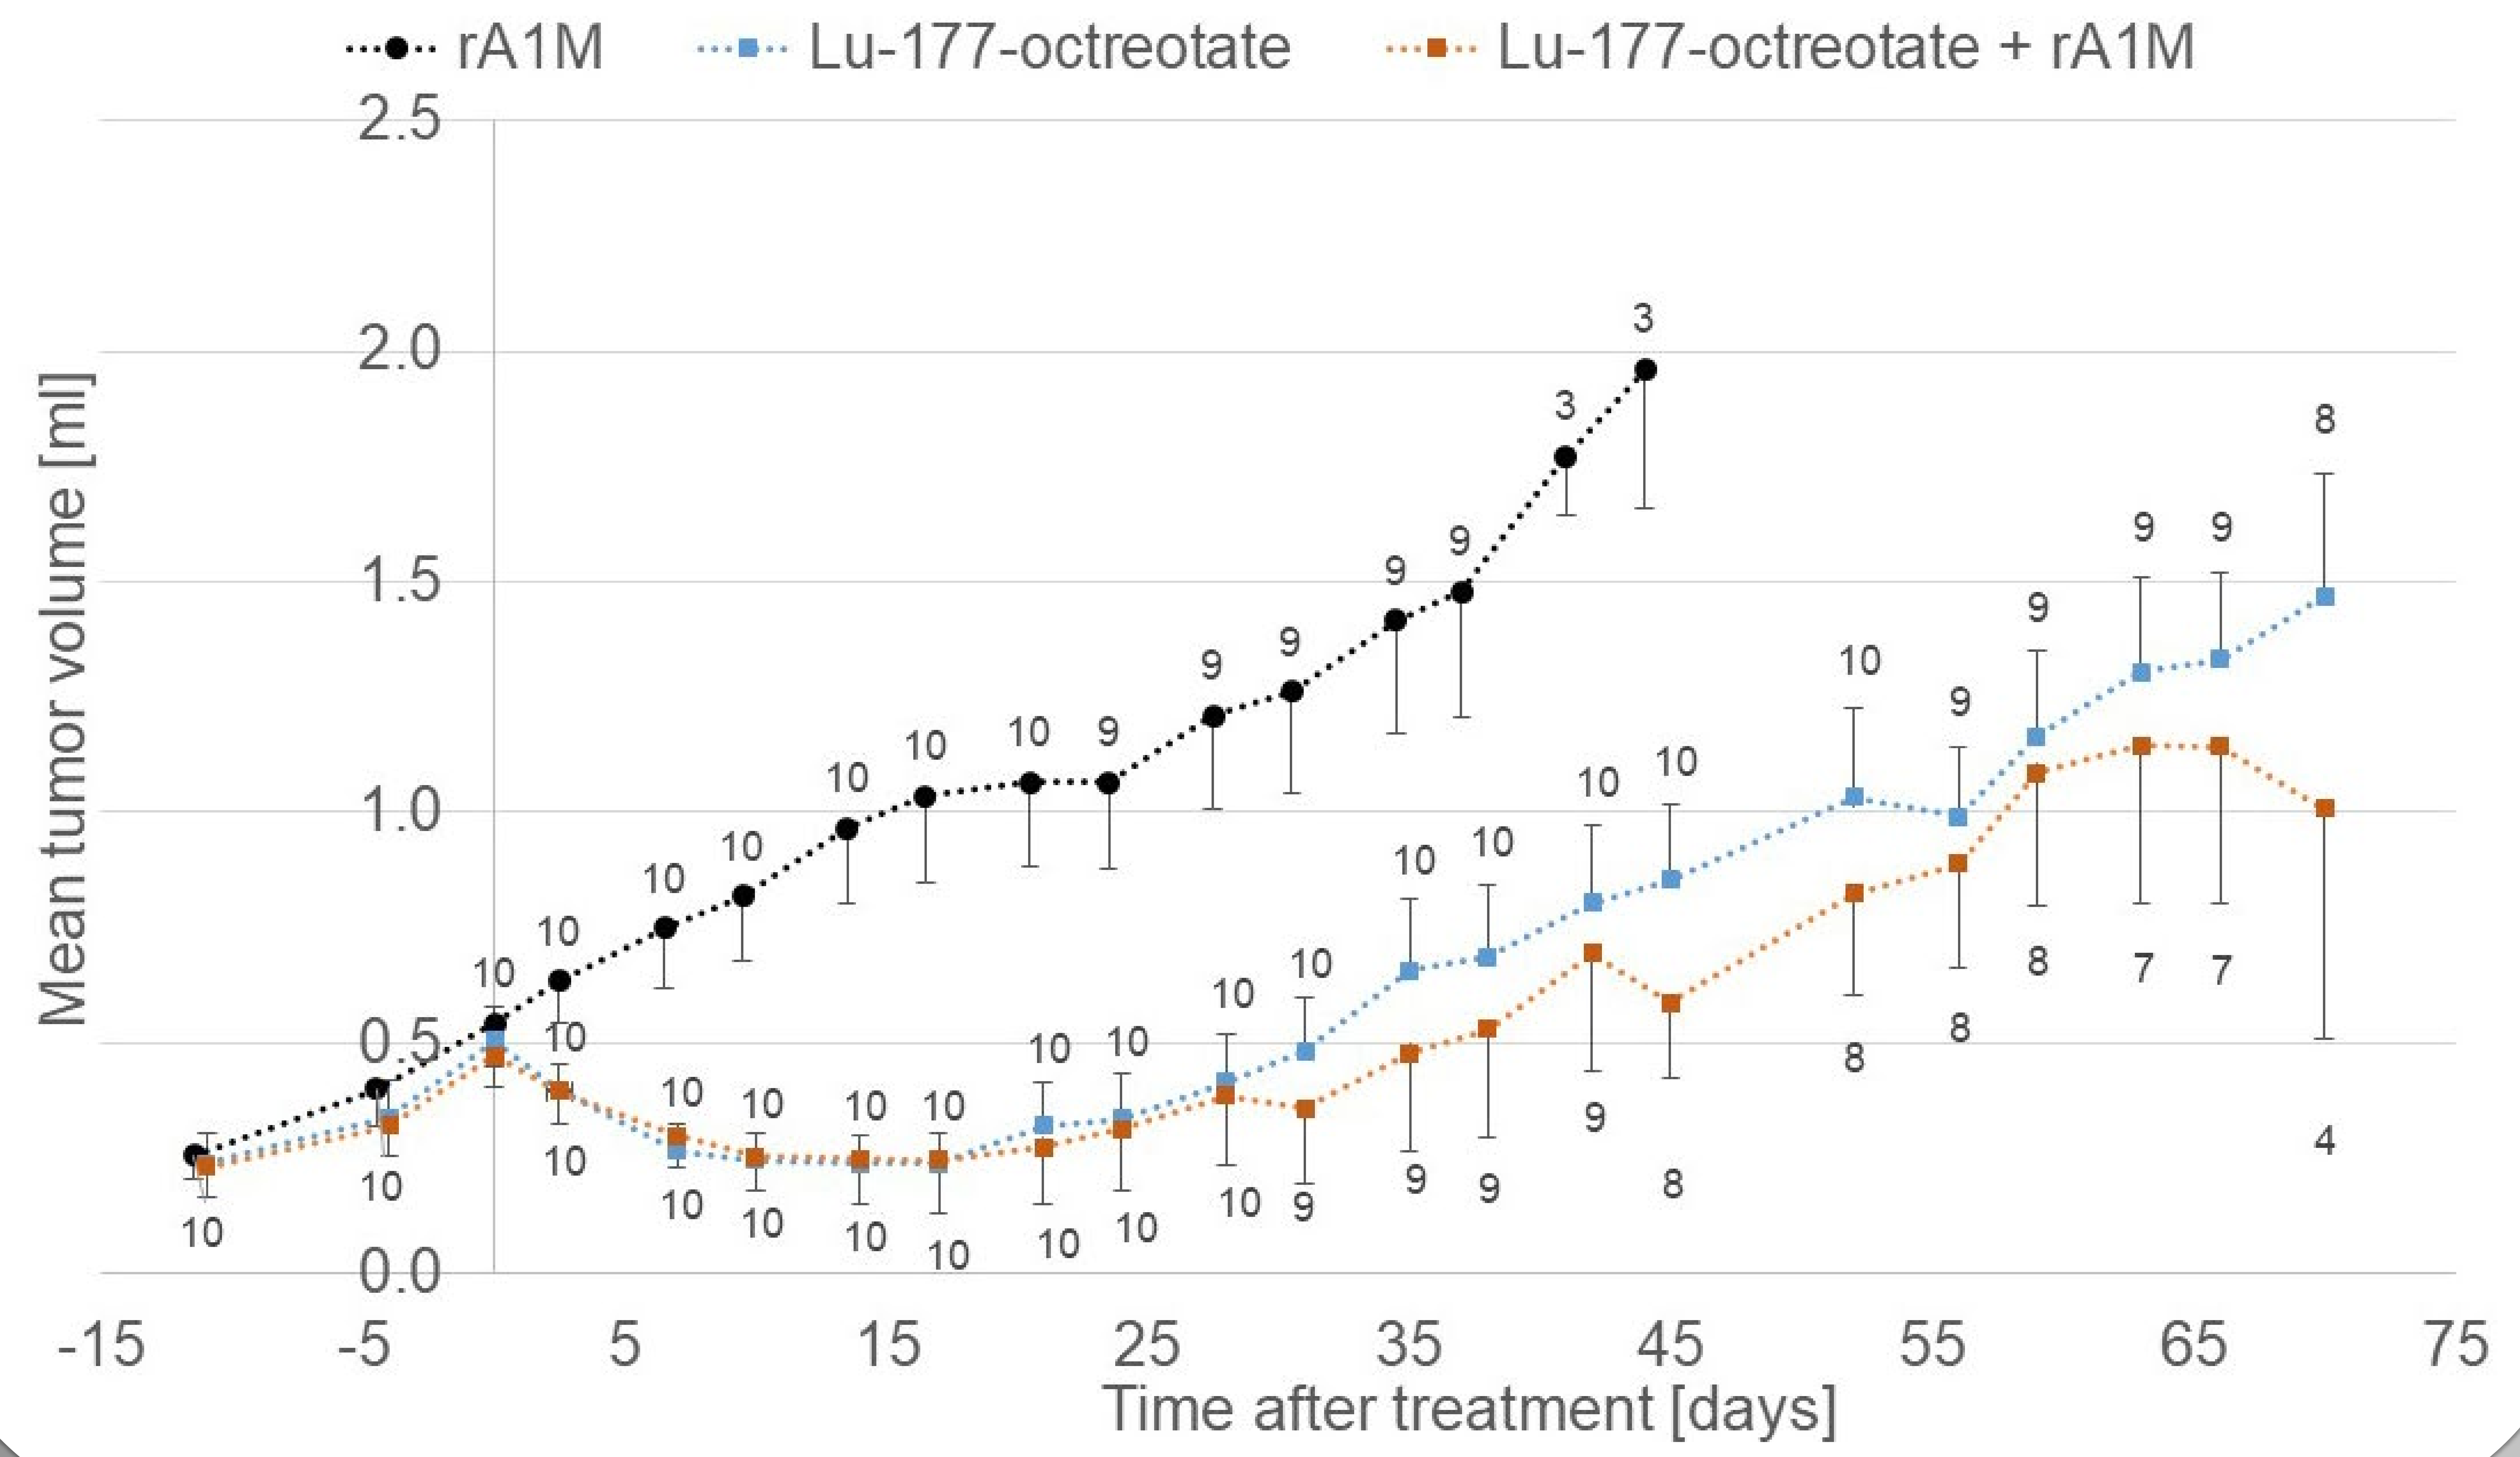
rA1M
Lu-177-octreotate
Lu-177-octreotate + rA1M
Mean tumor volume [ml]
Time after treatment [days]

Figure 2: Mean tumor volume over time for mice treated with rA1M (5 mg/kg), ^{177}Lu-octreotate (30 MBq) or both rA1M (5 mg/kg) and ^{177}Lu-octreotate (30 MBq). Data labels represent the number of animals, error bars indicate SEM.

Tumors in the mice receiving ^{177}Lu-octreotate or co-infusion of rA1M and ^{177}Lu-octreotate had similar therapeutic response; during the two first weeks the mean tumor volume decreased and was reduced to about 50 % of the starting volume. At about 20 days after the treatment the tumors started to regrow. The rA1M group showed no decrease in mean tumor size. Tumor growth of the tumors did not seem to be affected by rA1M.

To assess the tumor response the area under each individual mouse response curve was calculated from day 0 to 21. A one-way ANOVA with Tukey HSD test showed that the mean area for the rA1M group, was statistically significant higher than both the ^{177}Lu-octreotate group and combined treatment group. No statistically significant difference were observed between the ^{177}Lu-octreotate group and the combined treatment group.