Tap to go back to kiosk menu

Netazepide, a gastrin/CCK_2 receptor antagonist, can eradicate type 1 gastric NETs

Boyce M[1], Moore A[2], Parsons B[2], Lloyd K[2], Sagatun L[3], Varro A[2], Fossmark R[3], Waldum H[3], Pritchard M[2]

[1]Trio Medicines Ltd, Hammersmith Medicines Research, Cumberland Avenue, London, NW10 7EW, UNITED KINGDOM. [2]Department of Cellular and Molecular Physiology, Institute of Translational Medicine, University of Liverpool, Crown Street, Liverpool, L69 3GE, UNITED KINGDOM. [3]Departments of Gastroenterology and Pathology, St Olav's Hospital, and the Departments of Cancer Research, Molecular Medicine and Laboratory Medicine, Norwegian University of Science and Technology, Trondheim, NORWAY.

1. Introduction

Background

There are 3 types of gastric neuroendocrine tumour (g-NET). Type 1 arise from gastric mucosal ECL cells, which possess gastrin/CCK_2 receptors, and are found in patients with hypergastrinaemia secondary to achlorhydria caused by autoimmune chronic atrophic gastritis (CAG). 70–80% of g-NETs are type 1. Guidelines recommend regular gastroscopy and surveillance, endoscopic polypectomy or gastric antrectomy, depending on the findings [1]. Recent surveys from tertiary referral centres in Europe and USA report metastasis rates of 8% [2] and 19% [3].

Aims

To find out if netazepide, a gastrin/CCK_2 receptor antagonist [4], can eradicate type 1 g-NETs, and to identify biomarkers to monitor efficacy.

2. Materials and methods

Two centres (Liverpool, UK; Trondheim, Norway) first did a 12-week, open trial of netazepide, with 12-week recovery, in 16 patients with CAG, achlorhydria, hypergastrinaemia, multiple type 1 g-NETs, and raised serum chromogranin A (CgA). After a mean 14 (range 8–19) months off netazepide, 13 patients (Liverpool 8; Trondheim 5) took it for another 52 weeks. Assessments were: gastroscopy, to count the number of tumours and measure the largest one; and assays of CgA and gastrin in blood. Liverpool also assessed histology of tumour biopsies; gene transcript expression in mucosal biopsies; and blood miR-222.

3. Netazepide for 12 weeks reduces number and size of tumours

Netazepide for 12 weeks reduced the number of tumours and size of the largest one (Figure 1), and normalised CgA. The changes partially reversed over the 12 weeks' recovery period. Serum gastrin was unchanged throughout (Figure 2) [5,6].

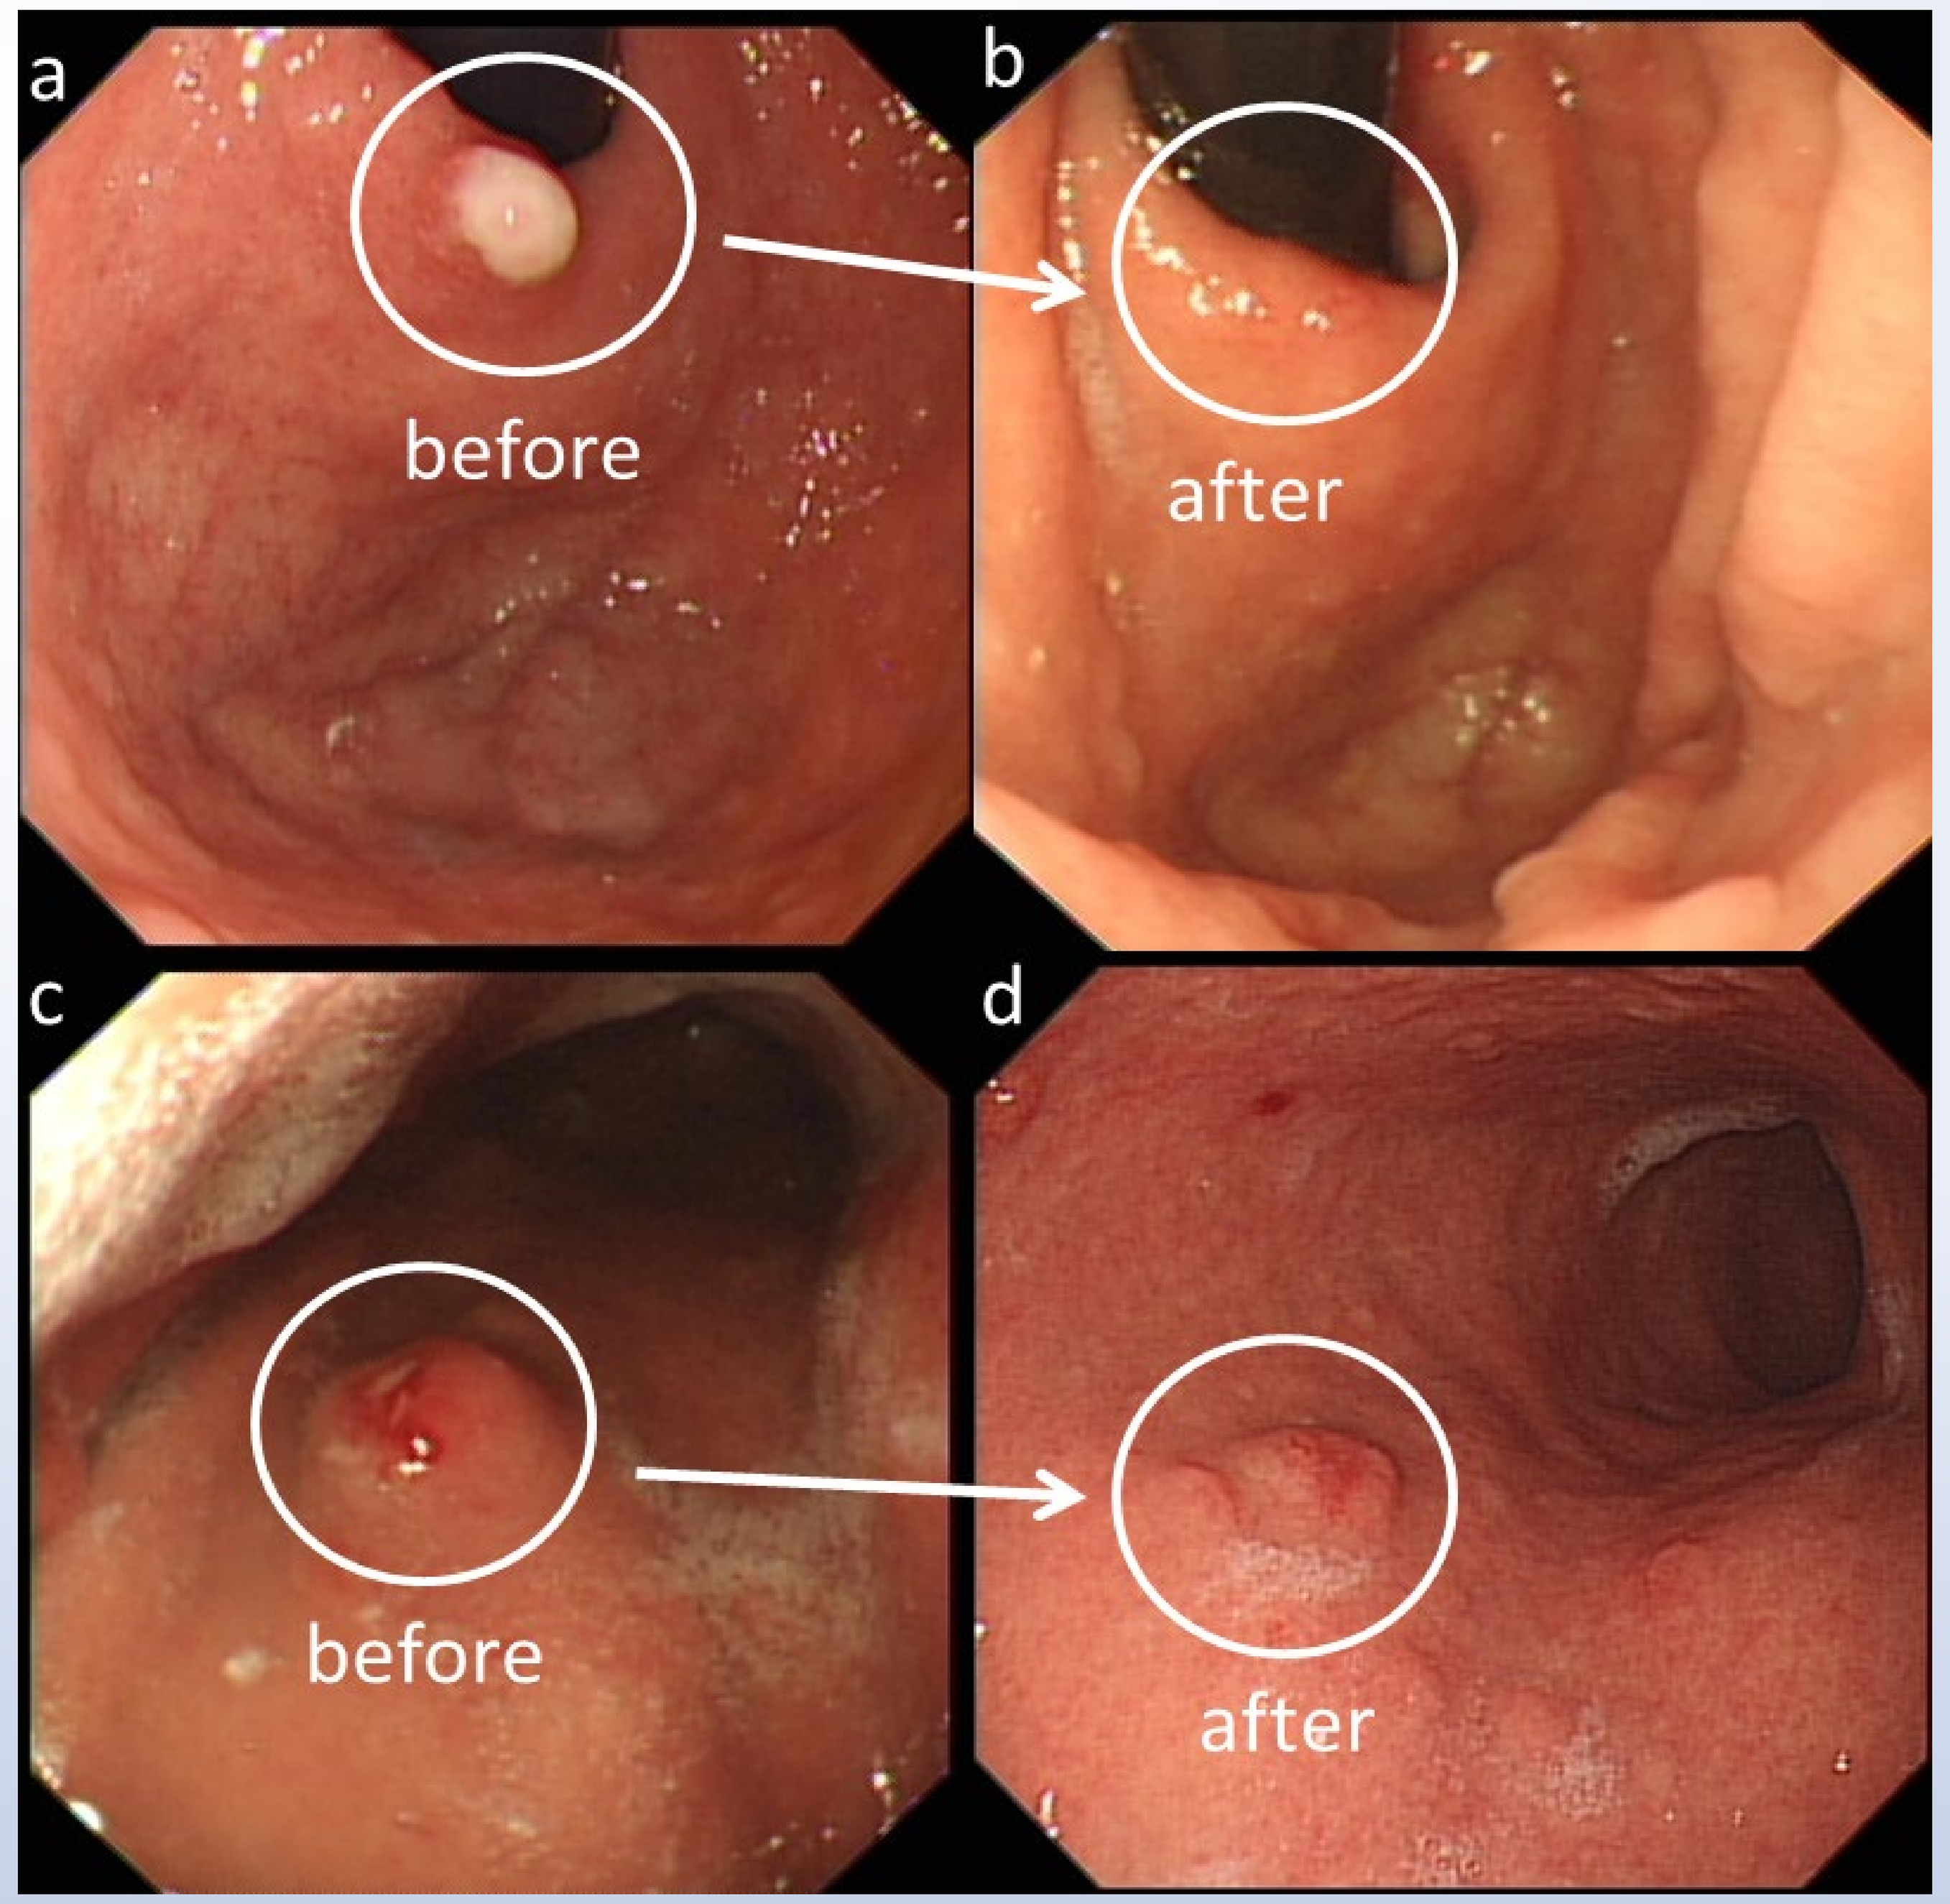

a
before
b
after
c
before
d
after

Figure 1. Endoscopic photos from stomach of Liverpool patients 1 (a, b) and 2 (a, b) before and after netazepide for 12 weeks.

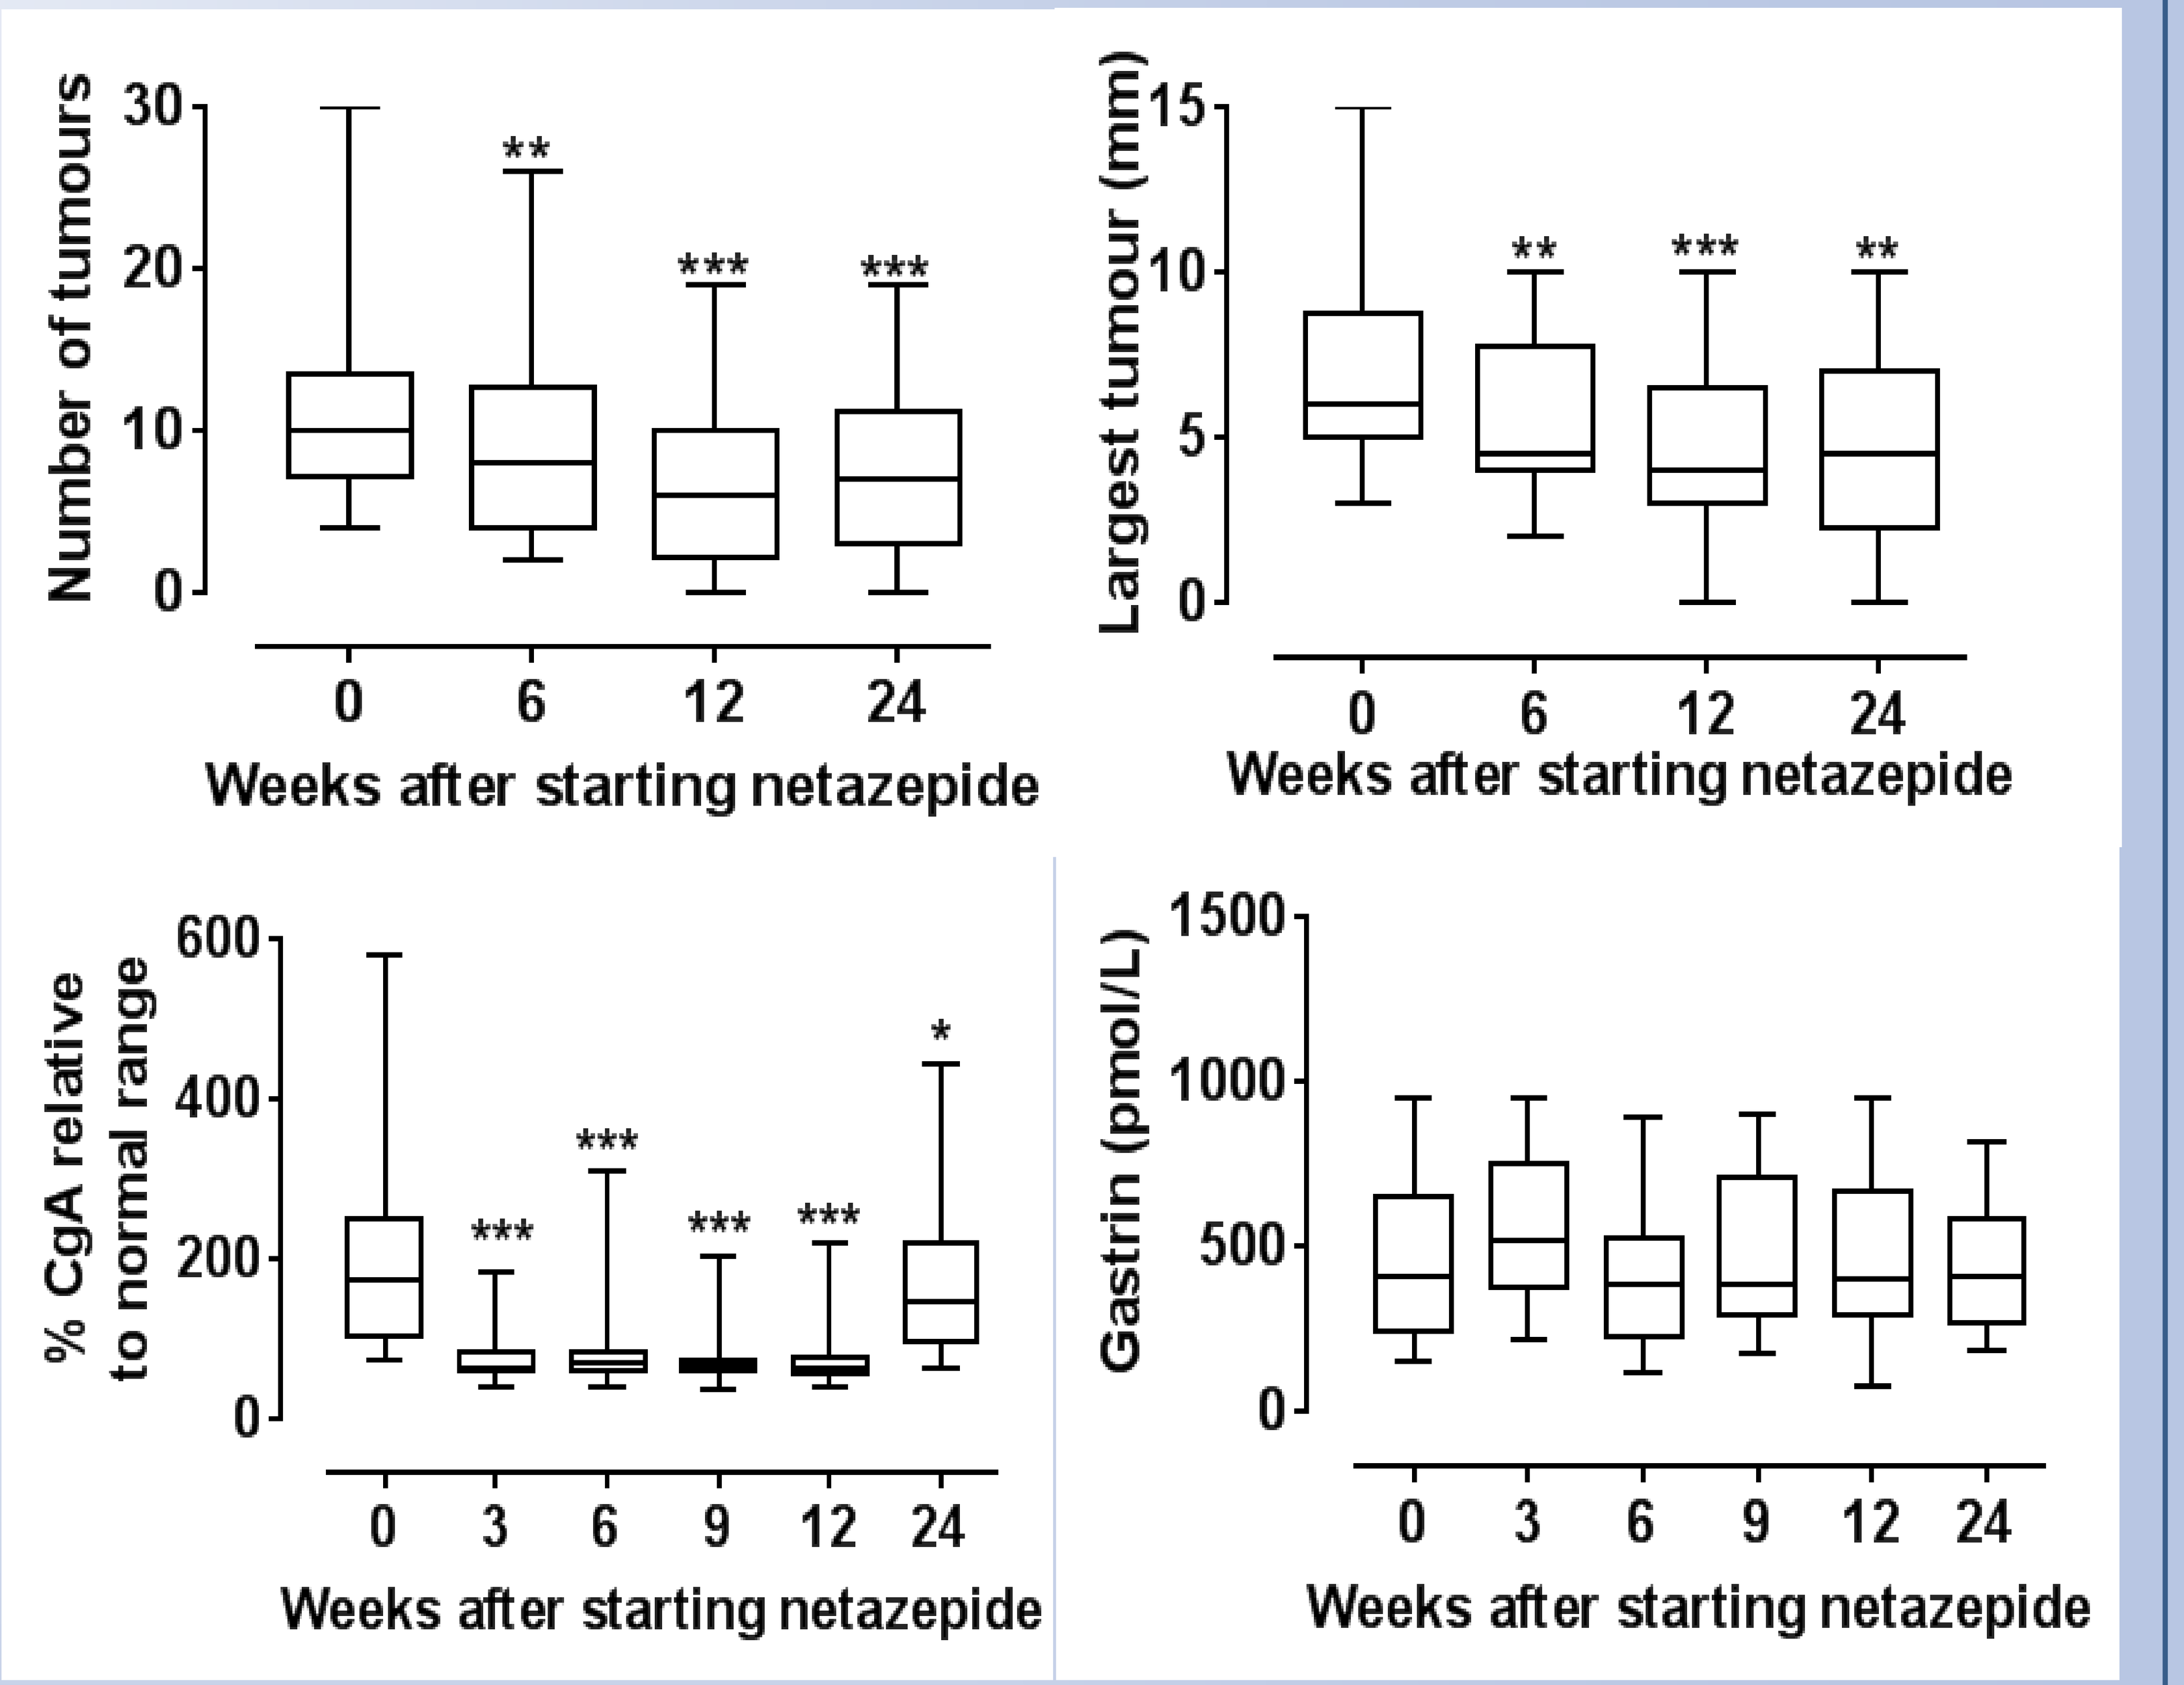

Number of tumours
Weeks after starting netazepide
Largest tumour (mm)
Weeks after starting netazepide
% CgA relative to normal range
Weeks after starting netazepide
Gastrin (pmol/L)
Weeks after starting netazepide

Figure 2. Effect of oral netazepide 50 mg once daily for 12 weeks, followed by recovery for 12 weeks in 16 patients with CAG, multiple type 1 g-NETs, hypergastrinaemia, and raised CgA. *p<0.05; **p<0.01; ***p<0.001.

Netazepide, a gastrin/CCK_2 receptor antagonist, can eradicate type 1 gastric NETs

Boyce M[1], Moore A[2], Parsons B[2], Lloyd K[2], Sagatun L[3], Varro A[2], Fossmark R[3], Waldum H[3], Pritchard M[2]

[1]Trio Medicines Ltd, Hammersmith Medicines Research, Cumberland Avenue, London, NW10 7EW, UNITED KINGDOM. [2]Department of Cellular and Molecular Physiology, Institute of Translational Medicine, University of Liverpool, Crown Street, Liverpool, L69 3GE, UNITED KINGDOM. [3]Departments of Gastroenterology and Pathology, St Olav's Hospital, and the Departments of Cancer Research, Molecular Medicine and Laboratory Medicine, Norwegian University of Science and Technology, Trondheim, NORWAY.

4. Relapse off treatment

While off treatment, the number of tumours, the size of the largest tumour, and CgA all increased again (Figure 3).

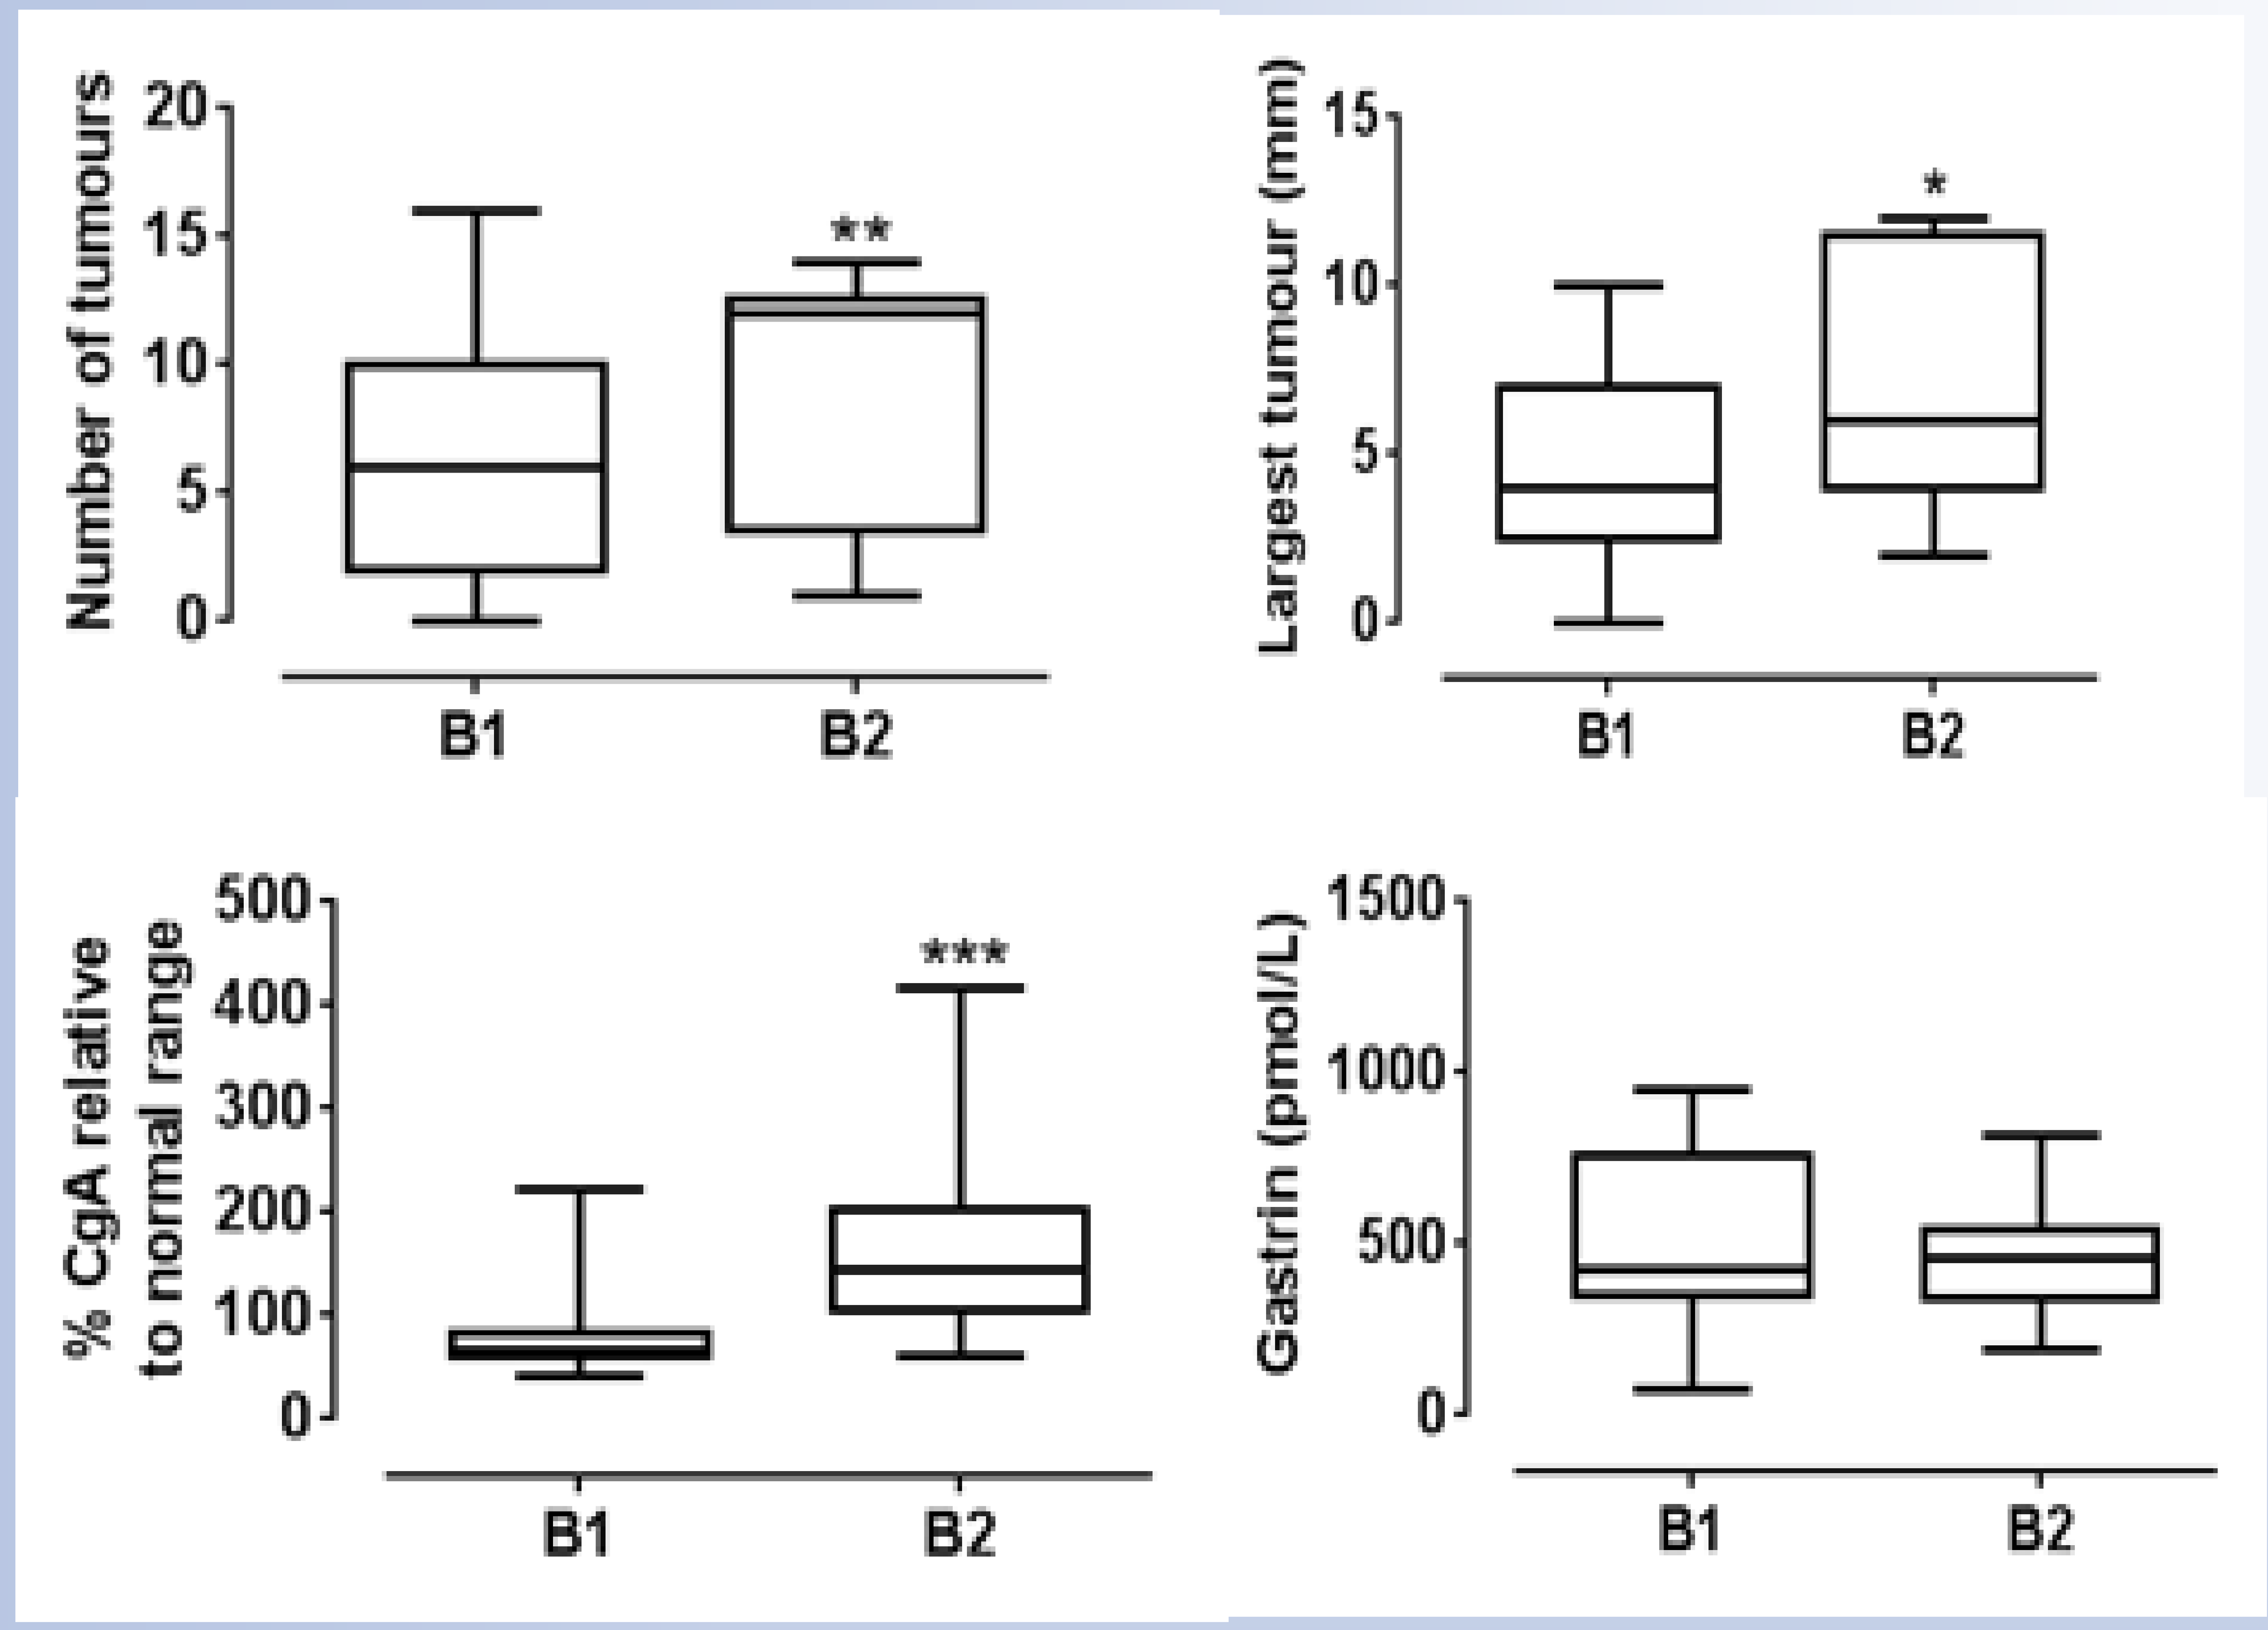
Number of tumours
20
15
10
5
0
**
B1
B2
Largest tumour (mm)
15
10
5
0
*
B1
B2
% CgA relative to normal range
500
400
300
200
100
0

B1
B2
Gastrin (pmol/L)
1500
1000
500
0
B1
B2

Figure 3. Changes in 13 patients during mean 14 (range 8–19) months off netazepide. *p<0.05; **p<0.01; ***p<0.001. B1 = end of 12 weeks' netazepide. B2 = start of 52 weeks' netazepide.

5. Netazepide for 52 weeks eradicates tumours

Netazepide for 52 weeks eradicated all tumours in 5 patients, left one patient with only one tumour, reduced the number and size of tumours in the other patients, and normalised blood CgA (Figure 4). Again, gastrin was unaffected, confirming patients had achlorhydria. Netazepide was safe and well tolerated [7].

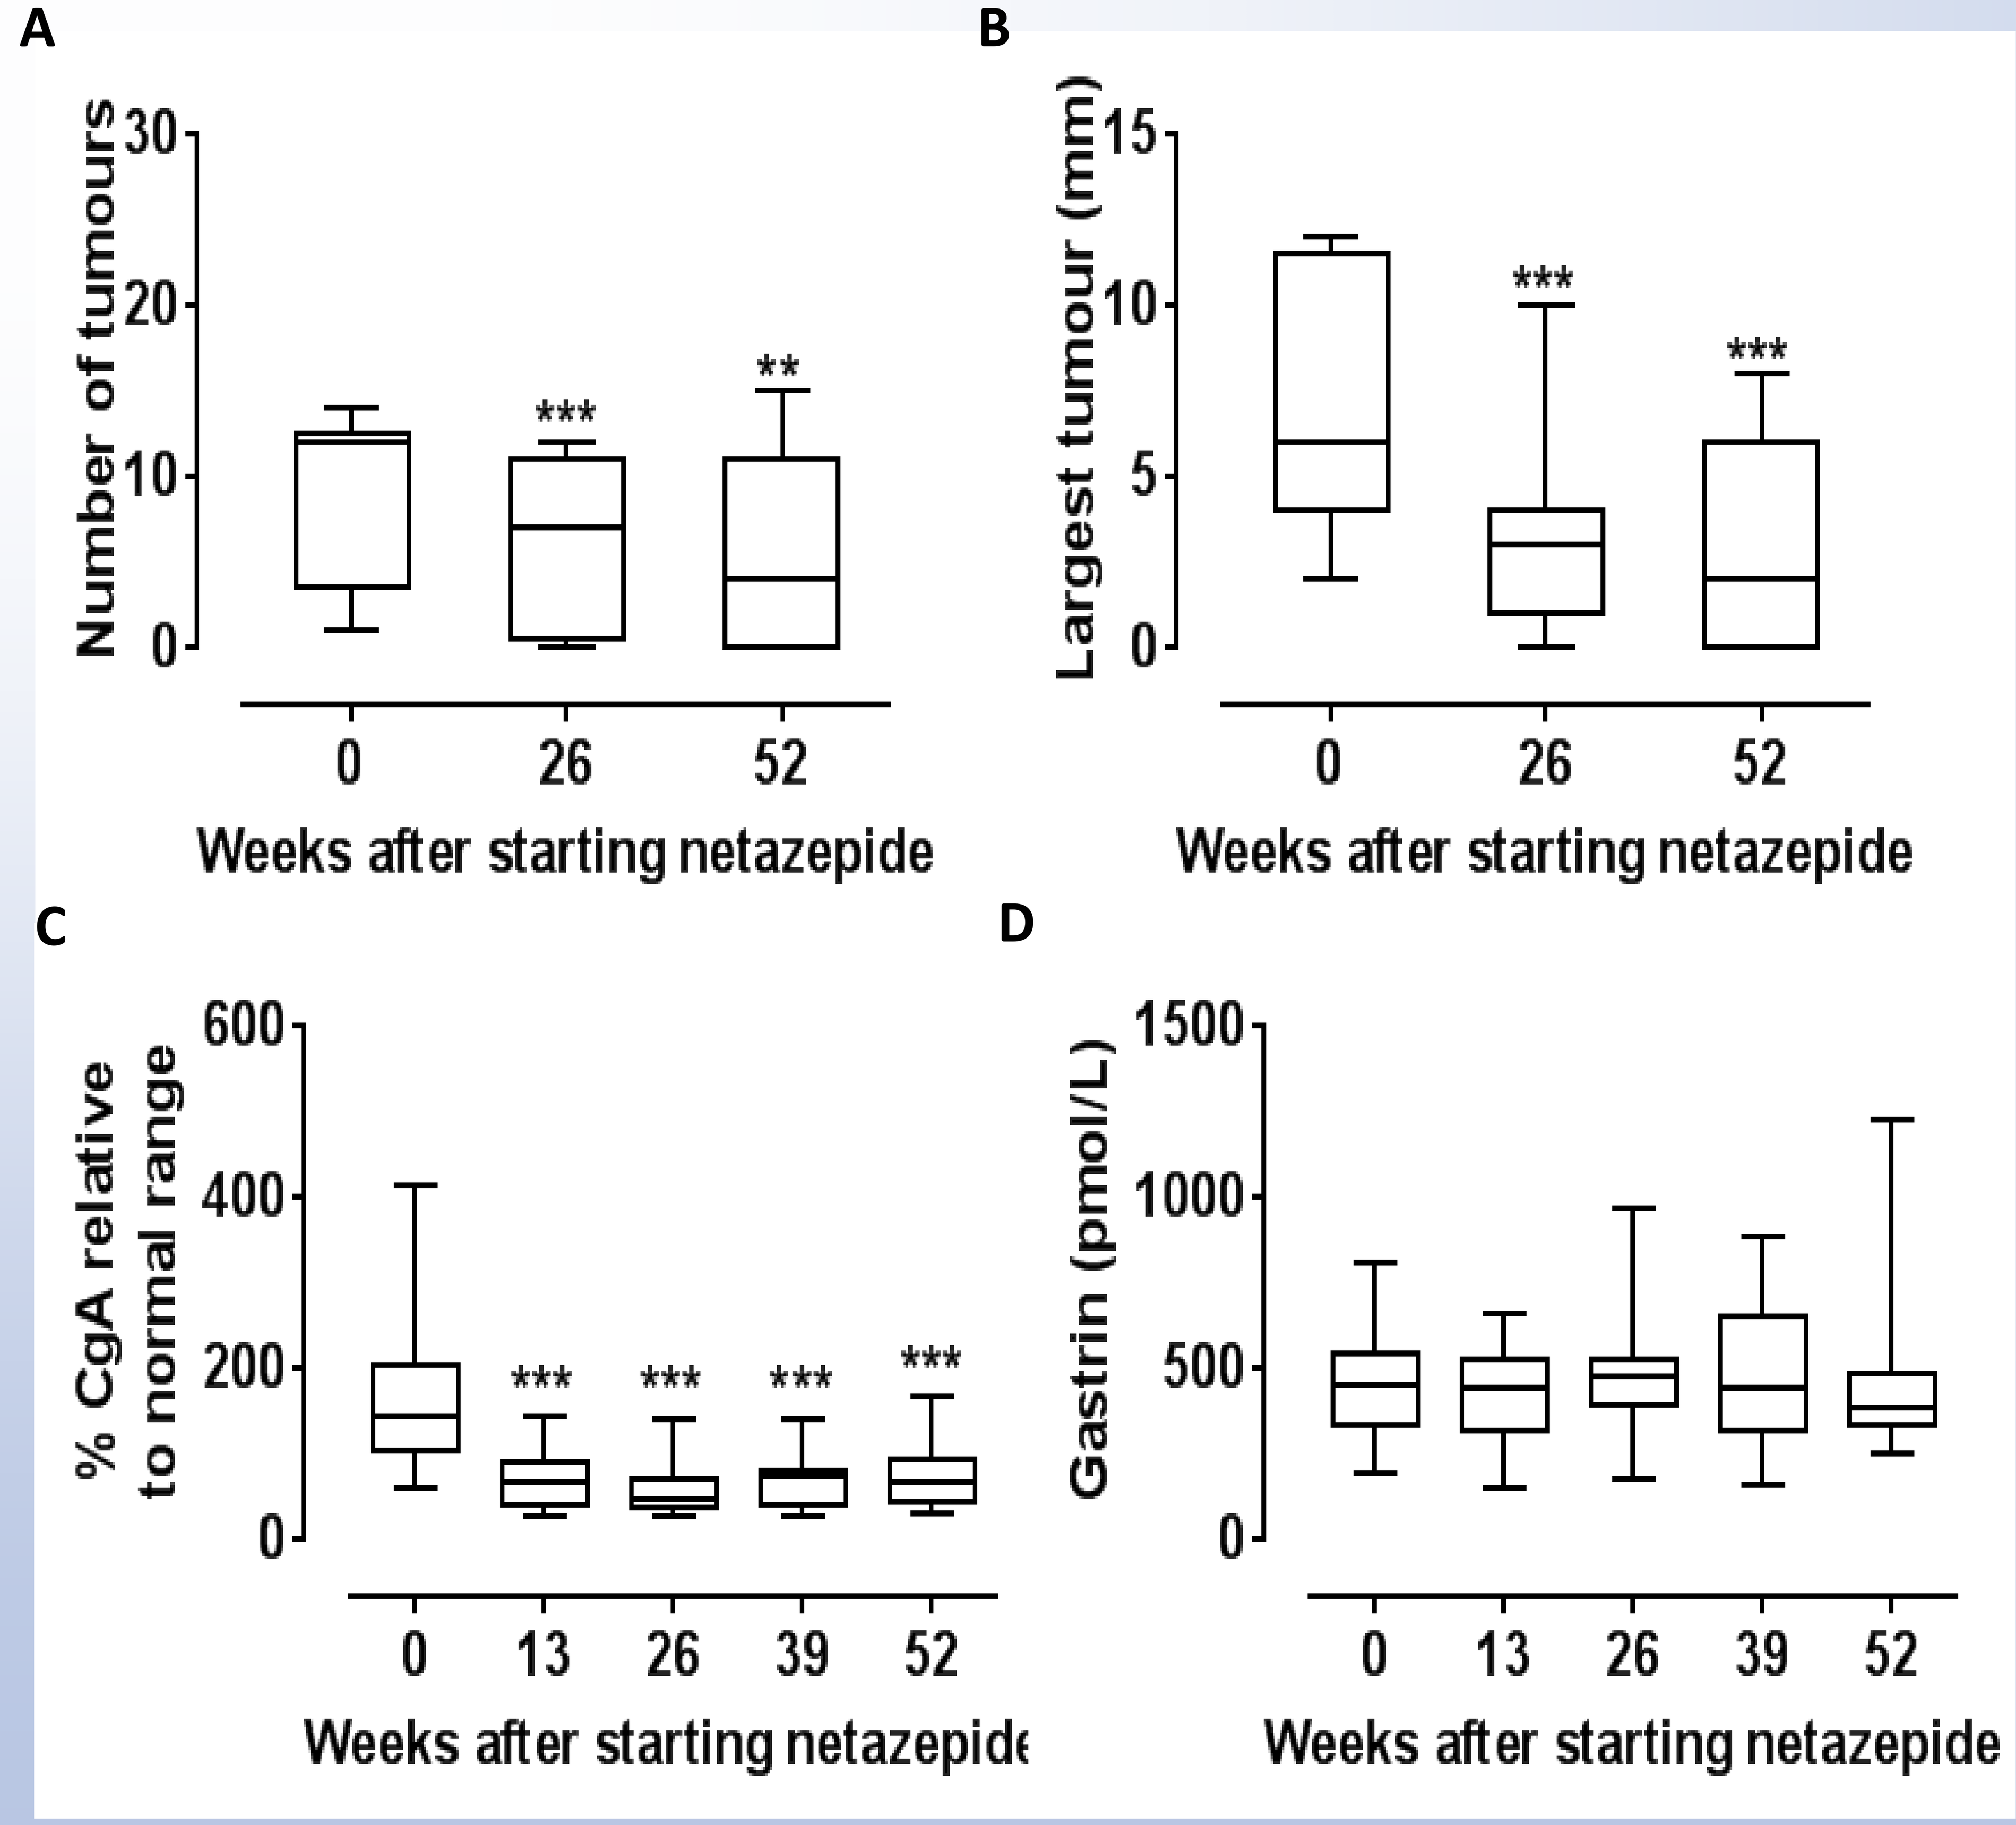
A
Number of tumours
30
20
10
0

**
0
26
52
Weeks after starting netazepide
B
Largest tumour (mm)
15
10
5
0

0
26
52
Weeks after starting netazepide
C
% CgA relative to normal range
600
400
200
0

0
13
26
39
52
Weeks after starting netazepide
D
Gastrin (pmol/L)
1500
1000
500
0
0
13
26
39
52
Weeks after starting netazepide

Figure 4. Effect of oral netazepide 25 or 50 mg once daily for 52 weeks in 13 patients with CAG, multiple type 1 g-NETs, hypergastrinaemia, and raised CgA. **p<0.01; ***p<0.001.

Netazepide, a gastrin/CCK_2 receptor antagonist, can eradicate type 1 gastric NETs

Boyce M[1], Moore A[2], Parsons B[2], Lloyd K[2], Sagatun L[3], Varro A[2], Fossmark R[3], Waldum H[3], Pritchard M[2]

[1]Trio Medicines Ltd, Hammersmith Medicines Research, Cumberland Avenue, London, NW10 7EW, UNITED KINGDOM. [2]Department of Cellular and Molecular Physiology, Institute of Translational Medicine, University of Liverpool, Crown Street, Liverpool, L69 3GE, UNITED KINGDOM. [3]Departments of Gastroenterology and Pathology, St Olav's Hospital, and the Departments of Cancer Research, Molecular Medicine and Laboratory Medicine, Norwegian University of Science and Technology, Trondheim, NORWAY.

6. Effect on biomarkers

All Liverpool patients had NET histology before treatment; only 3 had NET histology after 52 weeks of netazepide. Netazepide reduced mucosal gene expression of CgA and histidine decarboxylase (HDC) (Figure 5), PAPPA2, ERP27, CLC, PAM, SCG2, and MAOB, and increased CLDN10 (Figure 6). Netazepide also reduced mucosal and serum miR-222 (Figure 7). **CLDN10 modulates tight junction permeability** [8]. **miR-222 targets the tumour suppressor and oncogene p27^{kip1}** [9].

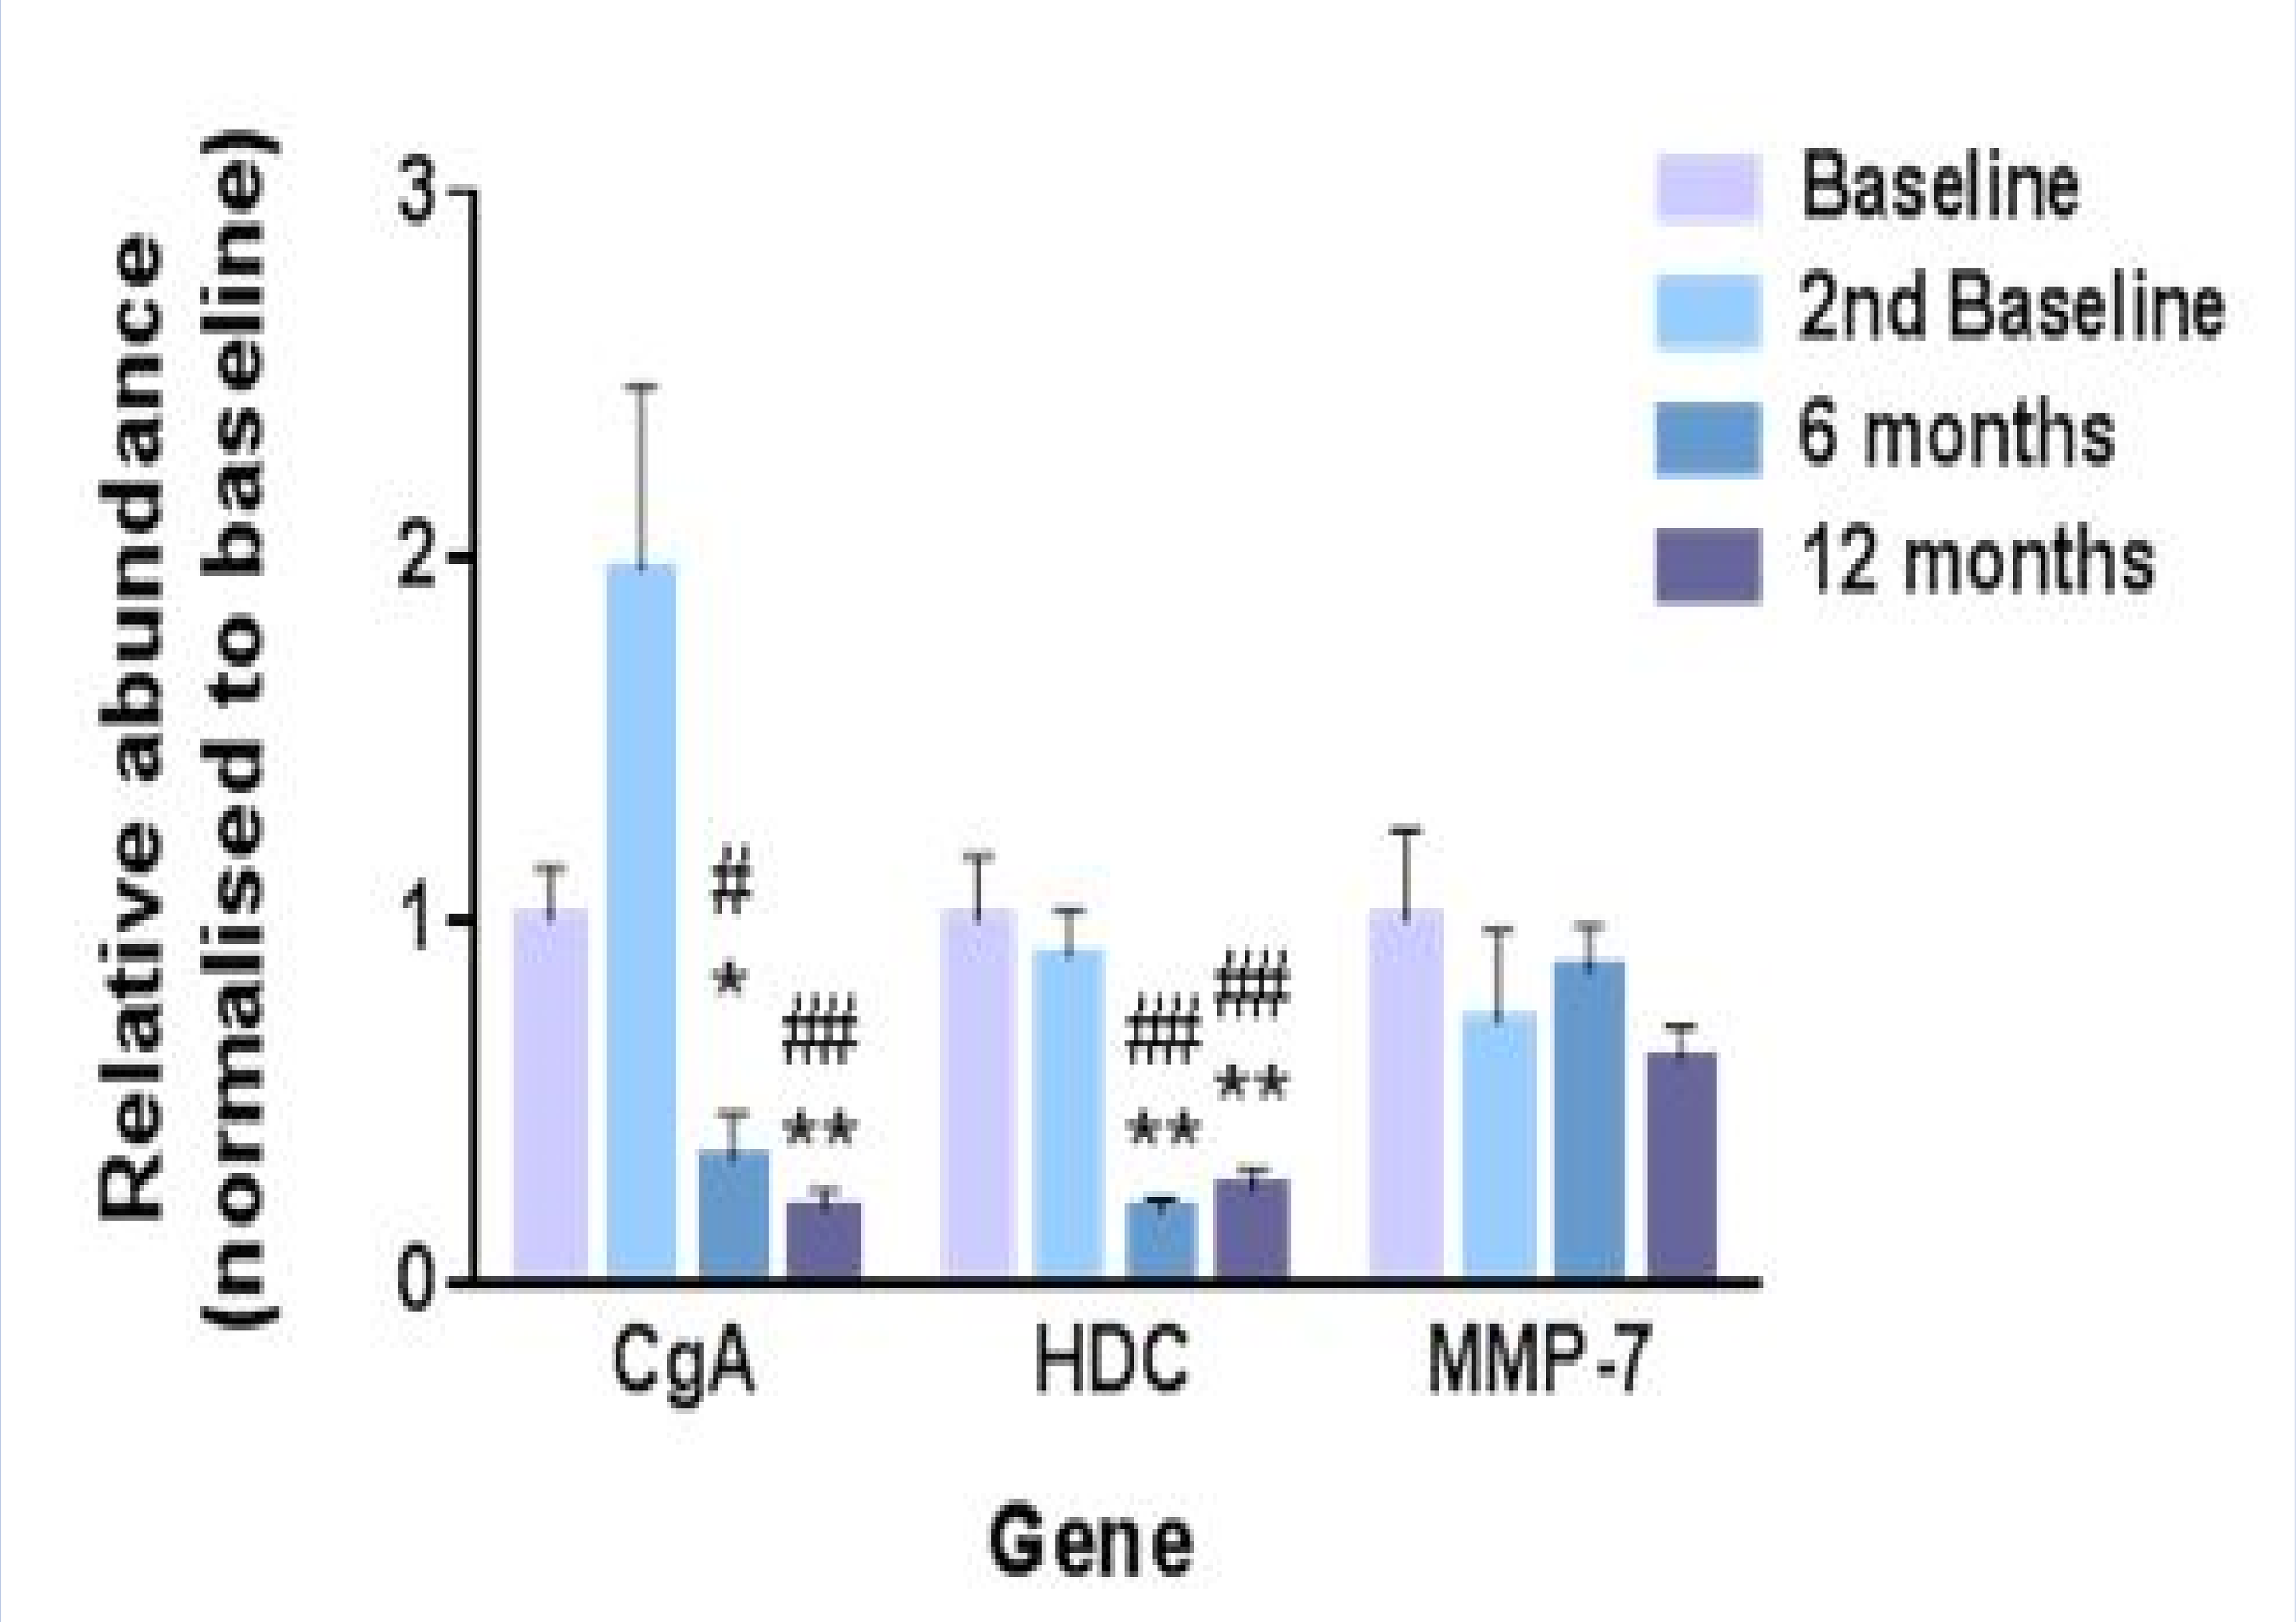
Relative abundance (normalised to baseline)
Baseline
2nd Baseline
6 months
12 months
CgA
HDC
MMP-7
Gene

Figure 5. Abundance of biomarkers in gastric mucosal biopsies (n=8) relative to GAPDH after 52 weeks of netazepide treatment. **p<0.05 compared to baseline. *p=0.055 compared to baseline. #p<0.05 and ##p<0.01 compared to 2nd baseline.

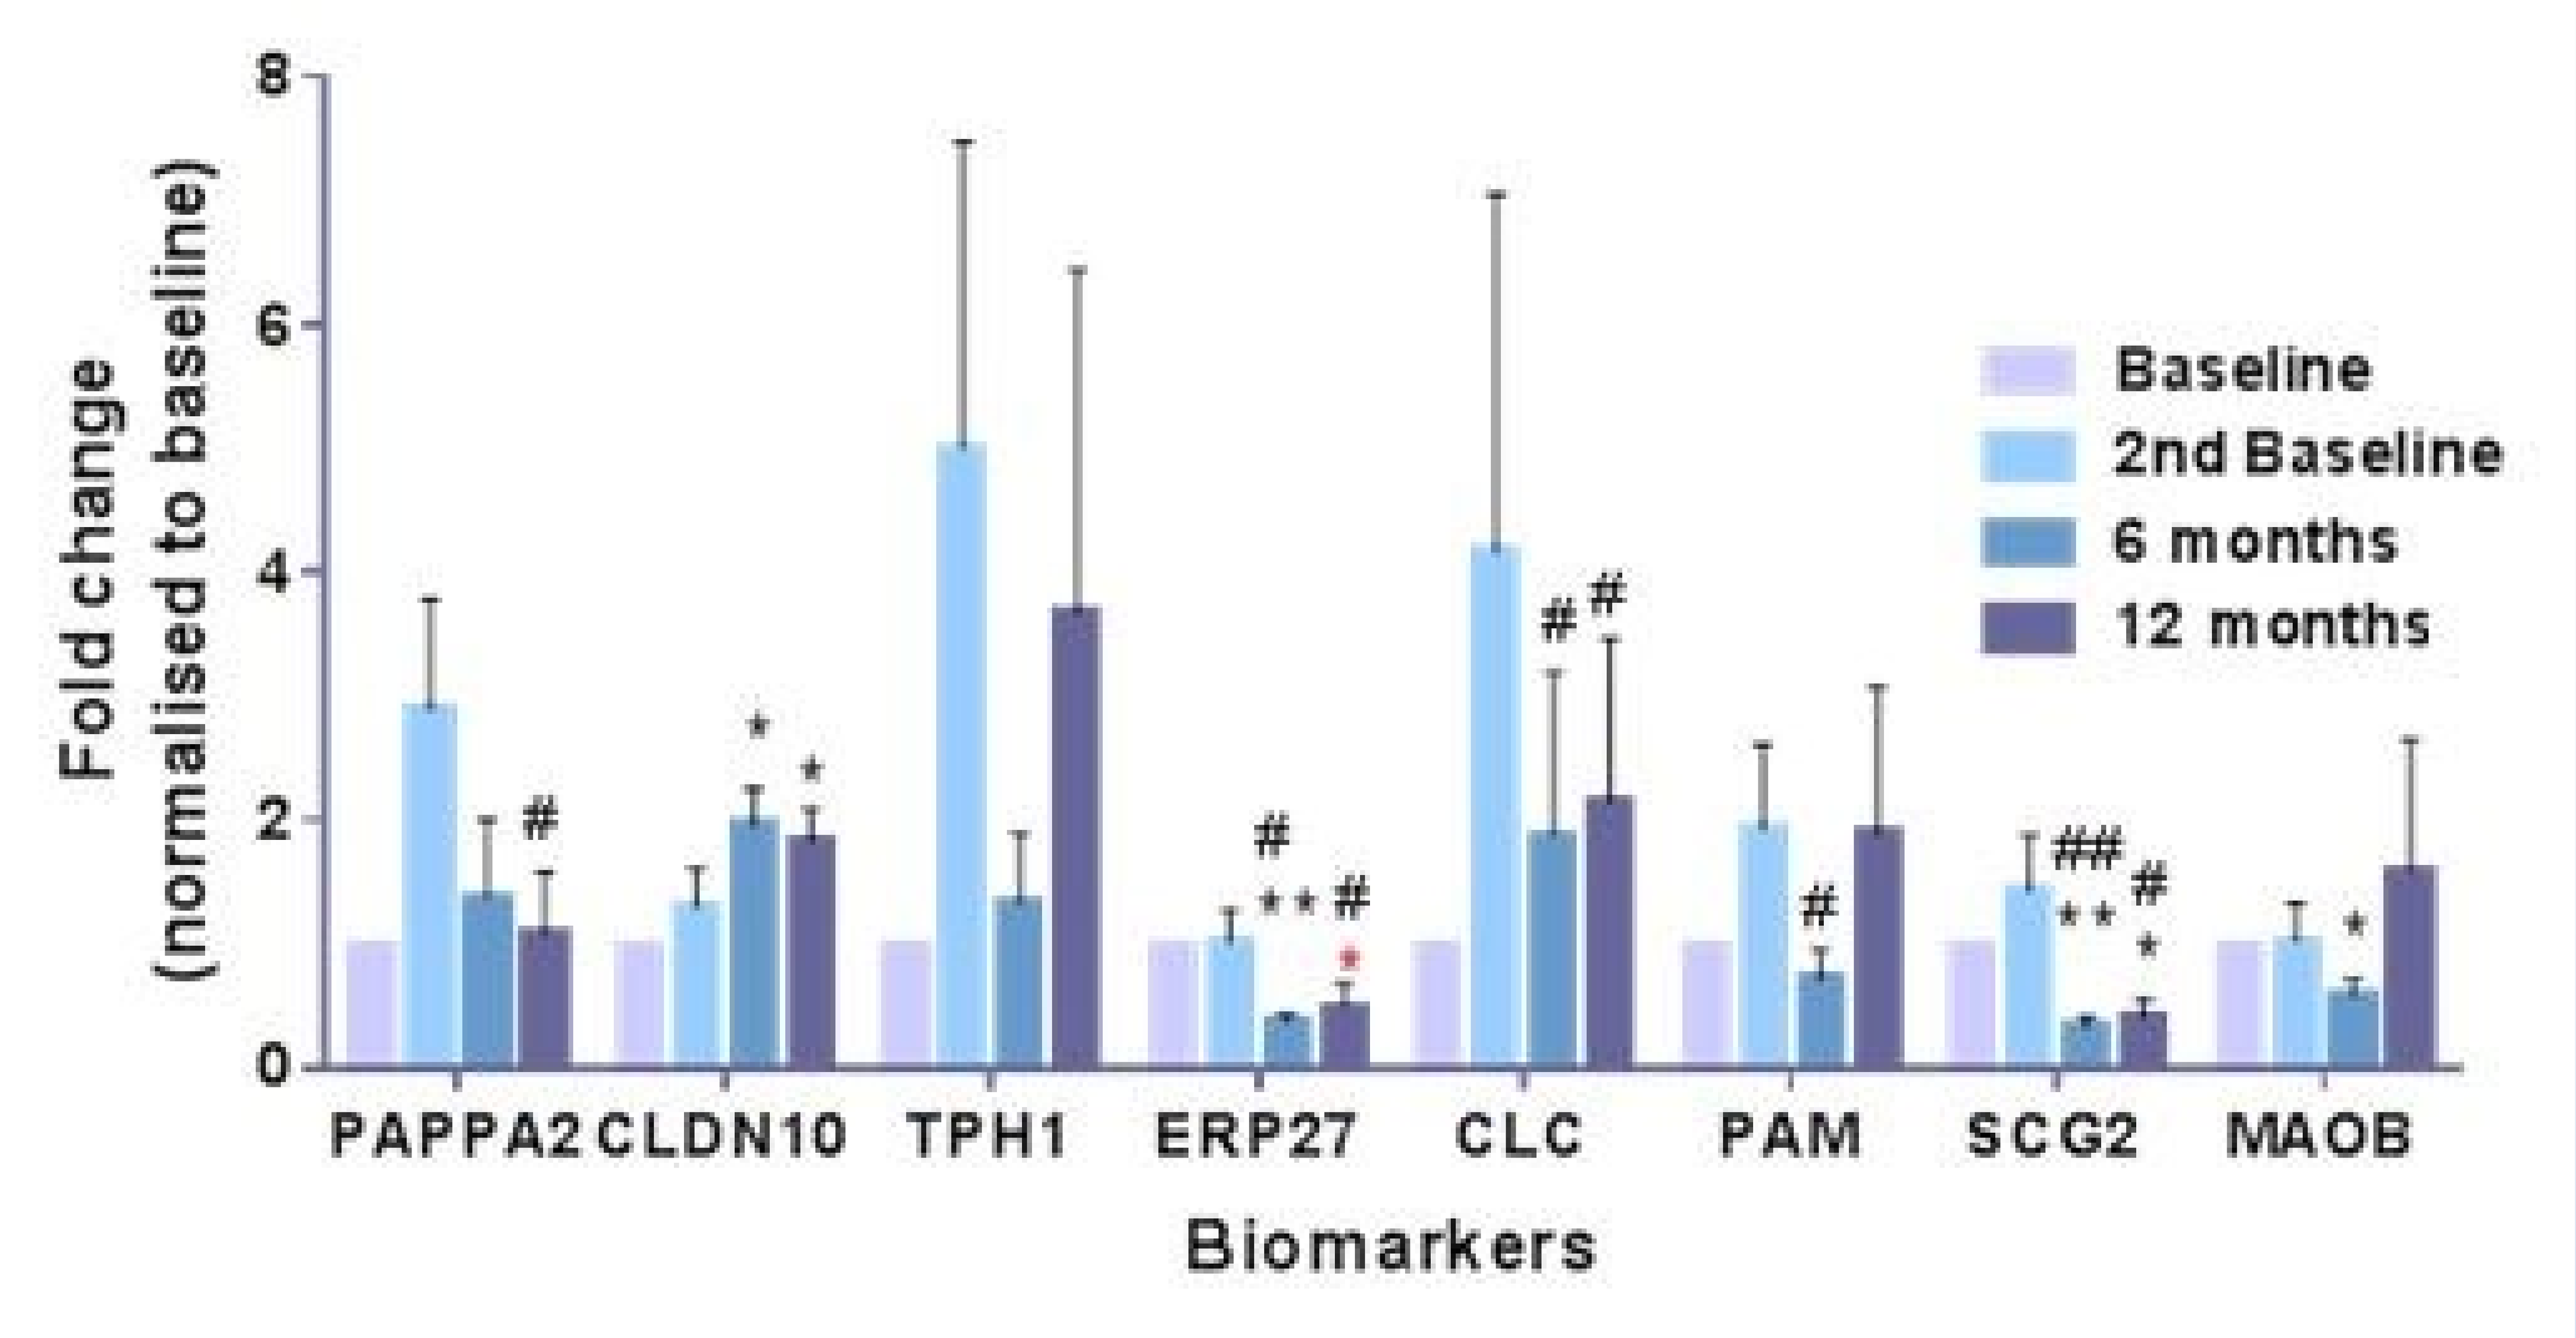
Fold change (normalised to baseline)
Baseline
2nd Baseline
6 months
12 months
PAPPA2
CLDN10
TPH1
ERP27
CLC
PAM
SCG2
MAOB
Biomarkers

Figure 6. Abundance of biomarkers in gastric mucosal biopsies (n=8) relative to GAPDH after 52 weeks of netazepide treatment. Wilcoxon signed rank tests. *p=0.055 and **p<0.05 compared to 1st baseline. # p<0.05 and ## p<0.01 compared to 2nd baseline. Pappalysin 2=PAPPA2; claudin 10=CLDN10; endoplasmic reticulum protein=ERP27; eosinophil lysophospholipase=CLC; peptidyl-glycine alpha-amidating monooxygenase=PAM; secretogranin II=SCG2; monoamine oxidase B=MAOB.

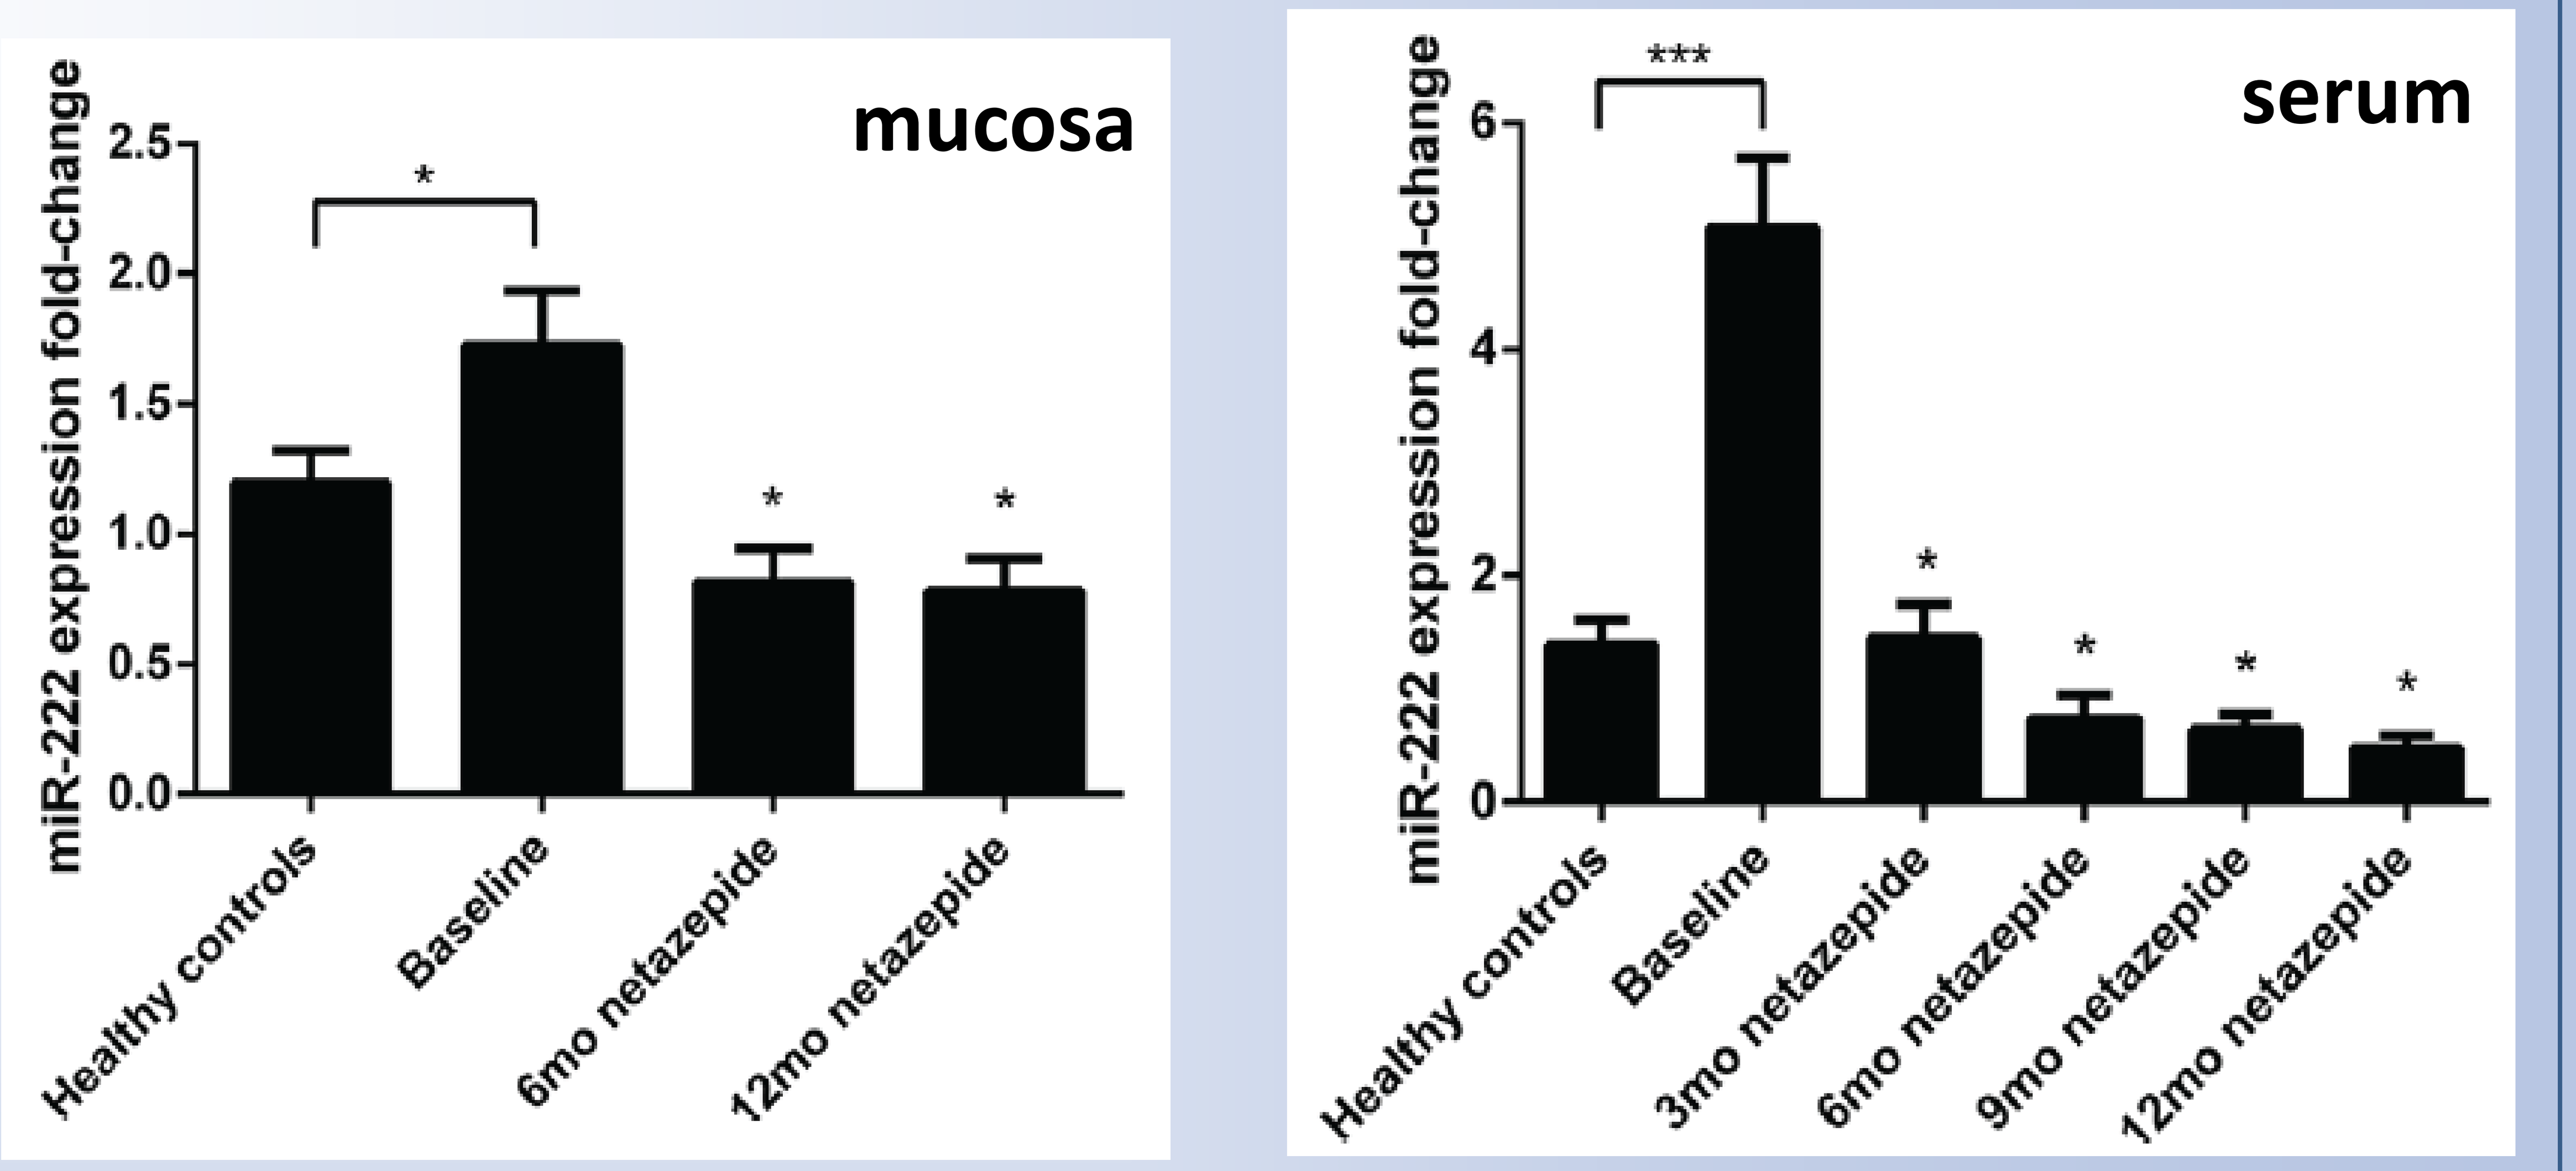
mucosa
miR-222 expression fold-change
Healthy controls
Baseline
6mo netazepide
12mo netazepide
serum
miR-222 expression fold-change
Healthy controls
Baseline
3mo netazepide
6mo netazepide
9mo netazepide
12mo netazepide

Figure 7. miR-222 expression was higher at baseline in mucosa and serum of patients (n=8) compared with healthy controls (n=10). miR-222 decreased during netazepide and returned to baseline after netazepide cessation. *p<0.0125; ***p<0.0001.

Netazepide, a gastrin/CCK_2 receptor antagonist, can eradicate type 1 gastric NETs

Boyce M[1], Moore A[2], Parsons B[2], Lloyd K[2], Sagatun L[3], Varro A[2], Fossmark R[3], Waldum H[3], Pritchard M[2]

[1]Trio Medicines Ltd, Hammersmith Medicines Research, Cumberland Avenue, London, NW10 7EW, UNITED KINGDOM. [2]Department of Cellular and Molecular Physiology, Institute of Translational Medicine, University of Liverpool, Crown Street, Liverpool, L69 3GE, UNITED KINGDOM. [3]Departments of Gastroenterology and Pathology, St Olav's Hospital, and the Departments of Cancer Research, Molecular Medicine and Laboratory Medicine, Norwegian University of Science and Technology, Trondheim, NORWAY.

7. Conclusions

In patients with type 1 g-NETs, netazepide, a gastrin/CCK_2 antagonist:

- can eradicate multiple tumours, or at least reduce their number and size, and, given its effect on CLDN10 and miR-222, may reduce their potential to metastasise;
- is a novel targeted medical treatment, and an alternative to regular gastroscopy and polypectomy, and possibly antrectomy;
- must be taken continuously, otherwise the tumours will regrow;
- reduces hypergastrinaemia-induced increases in blood CgA and miR-222, which can be used to monitor efficacy;
- was safe and well tolerated;
- is a designated orphan product in USA and Europe; and
- merits a multicentre, placebo-controlled trial.

8. References

1. Kulke *et al*. NANETS Guidelines. Pancreas 2010; 39: 735–752.
2. Grozinsky-Glasberg *et al*. World J Gastroenterol 2013; 19: 8687–8695.
3. Sagatun *et al*. Scand J Gastroenterol 2016; 51: 1398–1405.
4. Boyce *et al.* Br J Clin Pharmacol 2015; 79: 744–755.
5. Fossmark *et al*. AP&T 2012; 36: 1067–1075.
6. Moore *et al*. PLoS ONE 2013; 8: e76462.
7. Boyce *et al.* Br J Clin Pharmacol 2017; 83: 466-475.
8. Gunzel and Li. Physiol Rev 2013; 93: 525–569.
9. Lloyd *et al*. Oncotarget 2016; 7: 45462–45478.