The antiproliferative effect of octreotide in gastroenteropancreatic neuroendocrine tumors (GEP-NETs)

J Strosberg and L Kvols

H Lee Moffitt Cancer Center and Research Institute, Tampa, FL

Introduction

- Gastroenteropancreatic neuroendocrine tumors (GEP-NETs) are a genetically diverse group of malignant solid tumors that arise on the neuroendocrine cells throughout the body.
- Although GEP-NETs have historically been referred to as 'carcinoid tumors' (meaning cancer-like), GEP-NETs have the potential to become metastatic and malignant.
- Like other malignant solid tumors, survival time in patients with GEP-NETs is associated with tumor differentiation and metastatic spread.
- A medical therapy with an antiproliferative effect and an established well-tolerated safety profile would be a valuable management option compared with other more invasive/aggressive treatments for patients with metastatic GEP-NETs.
- This poster reviews the antiproliferative effect of octreotide sc and LAR from early uncontrolled studies to the definitive results of the PROMID trial with octreotide LAR 30 mg. Potential future medical therapies and ongoing studies are also listed.

Background

Epidemiology of GEP-NETs

- In the US there has been a significant increase in the incidence of GEP-NETs from 1973–2004 (Figure 1).[1]
- 50% of patients with GEP-NETs have regional or distant metastasis at diagnosis.[1]
- Patients with poorly-differentiated tumors and distant metastasis have a worse prognosis than patients with well/moderately-differentiated histology and localized/regional disease, respectively.[1]

Figure 1. Annual incidence of NETs in the US between 1973 and 2004[1]

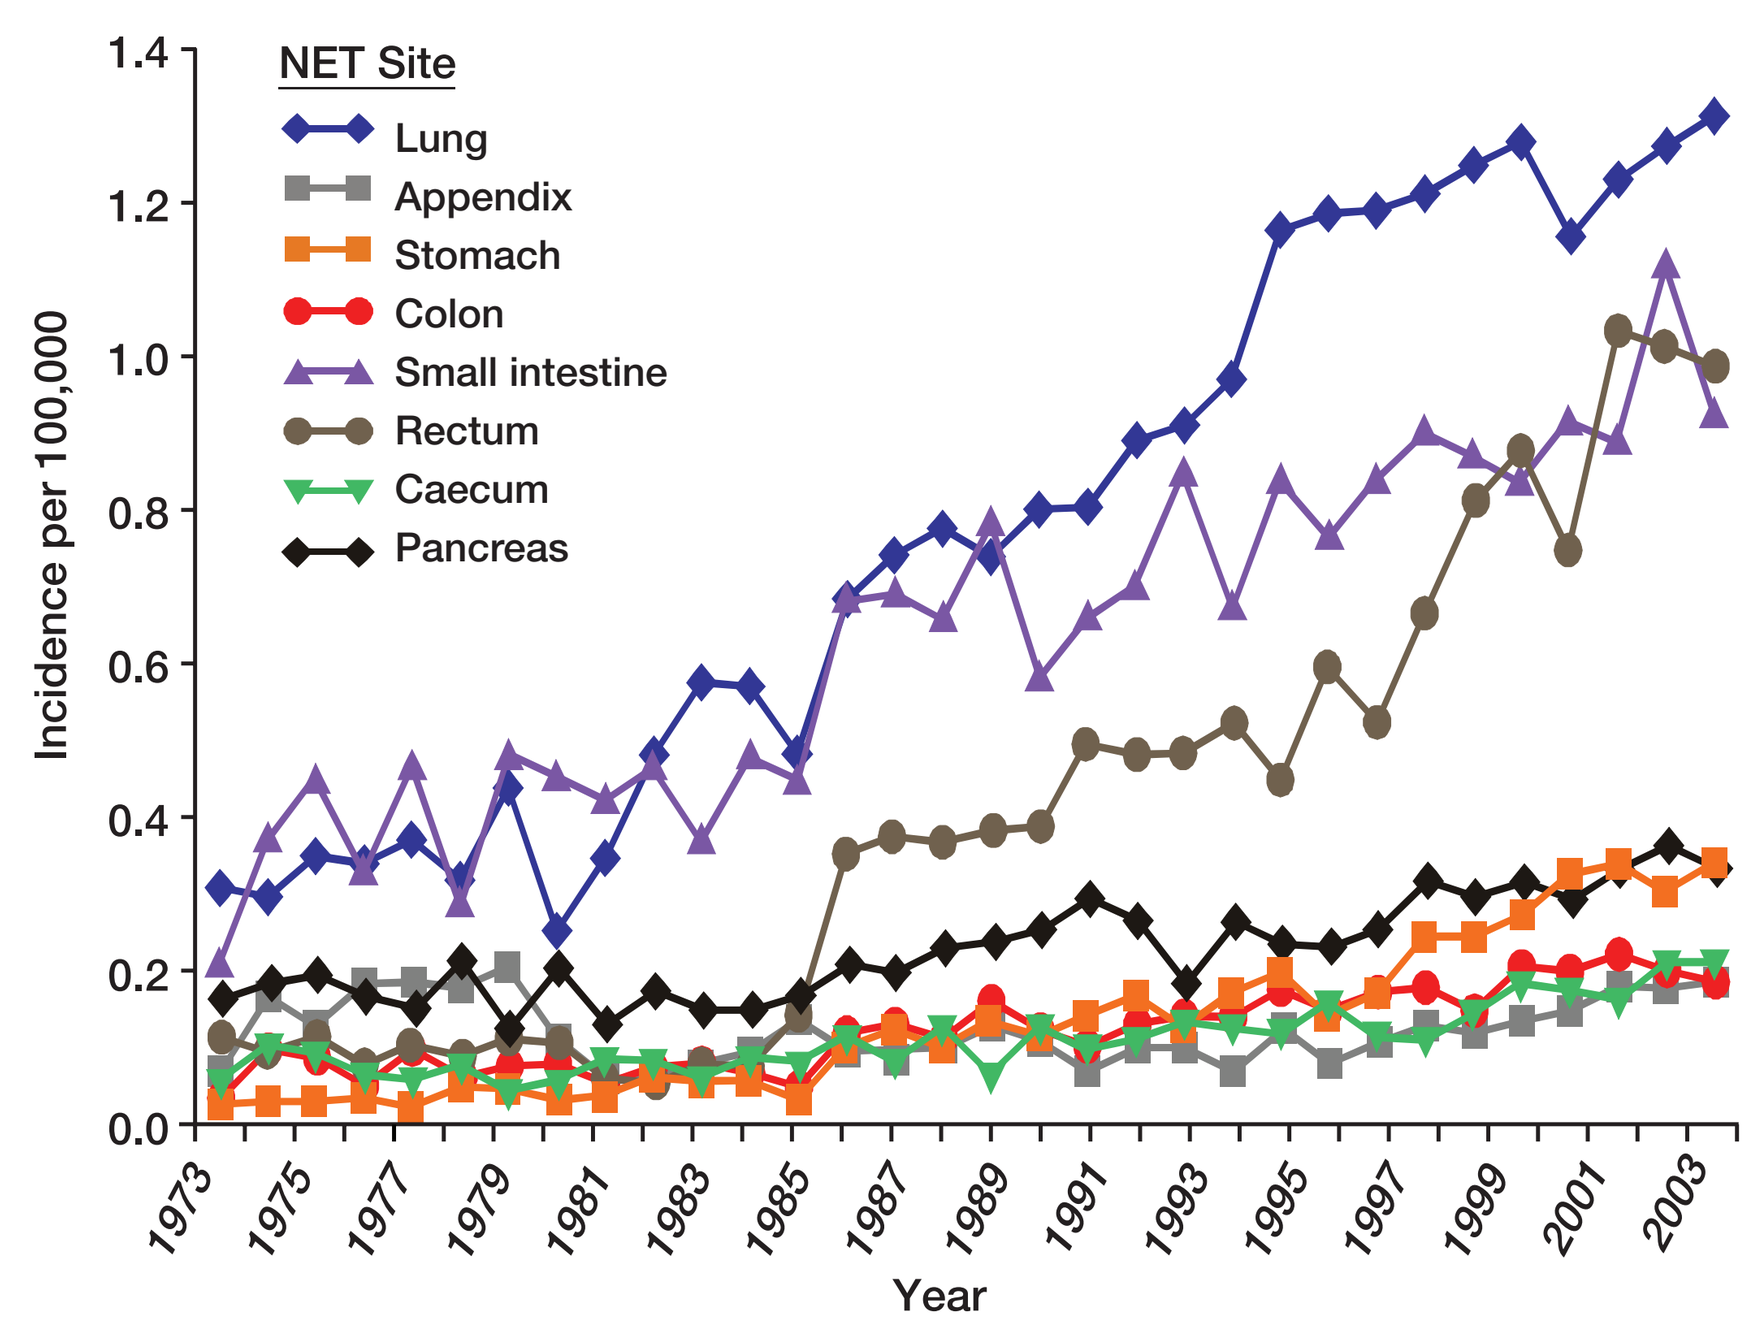
NET Site
Lung
Appendix
Stomach
Colon
Small intestine
Rectum
Caecum
Pancreas
Incidence per 100,000
Year

Reprinted with permission. © 2008 American Society of Clinical Oncology. All rights reserved.

Treatment options for patients with metastatic GEP-NETs

- Treatment goals in patients with GEP-NETs are total eradication of the tumor and/or abrogation of tumor growth. Where applicable, additional goals include amelioration of clinical symptoms and improving or maintaining quality of life.
- Surgical resection is the first-line therapy in patients with GEP-NETs, although most patients present with metastasis and will be unlikely to achieve surgical cure.
- Somatostatin analogues are the mainstay of medical therapy for the symptomatic relief of clinical syndromes associated with GEP-NETs (eg, carcinoid syndrome), which typically arise following liver metastasis.[2] Patients with progressive GEP-NETs are administered more aggressive/invasive procedures, including chemotherapy.
- A significant improvement in survival duration was seen in patients with metastatic NETs diagnosed 1988–2004 compared with those diagnosed 1973–1987, coinciding with the introduction of octreotide (Figure 2).[1]

Figure 2. Median survival time significantly increased in patients with GEP-NETs and distant metastasis diagnosed 1988–2004 compared with those diagnosed 1973–1987[1]

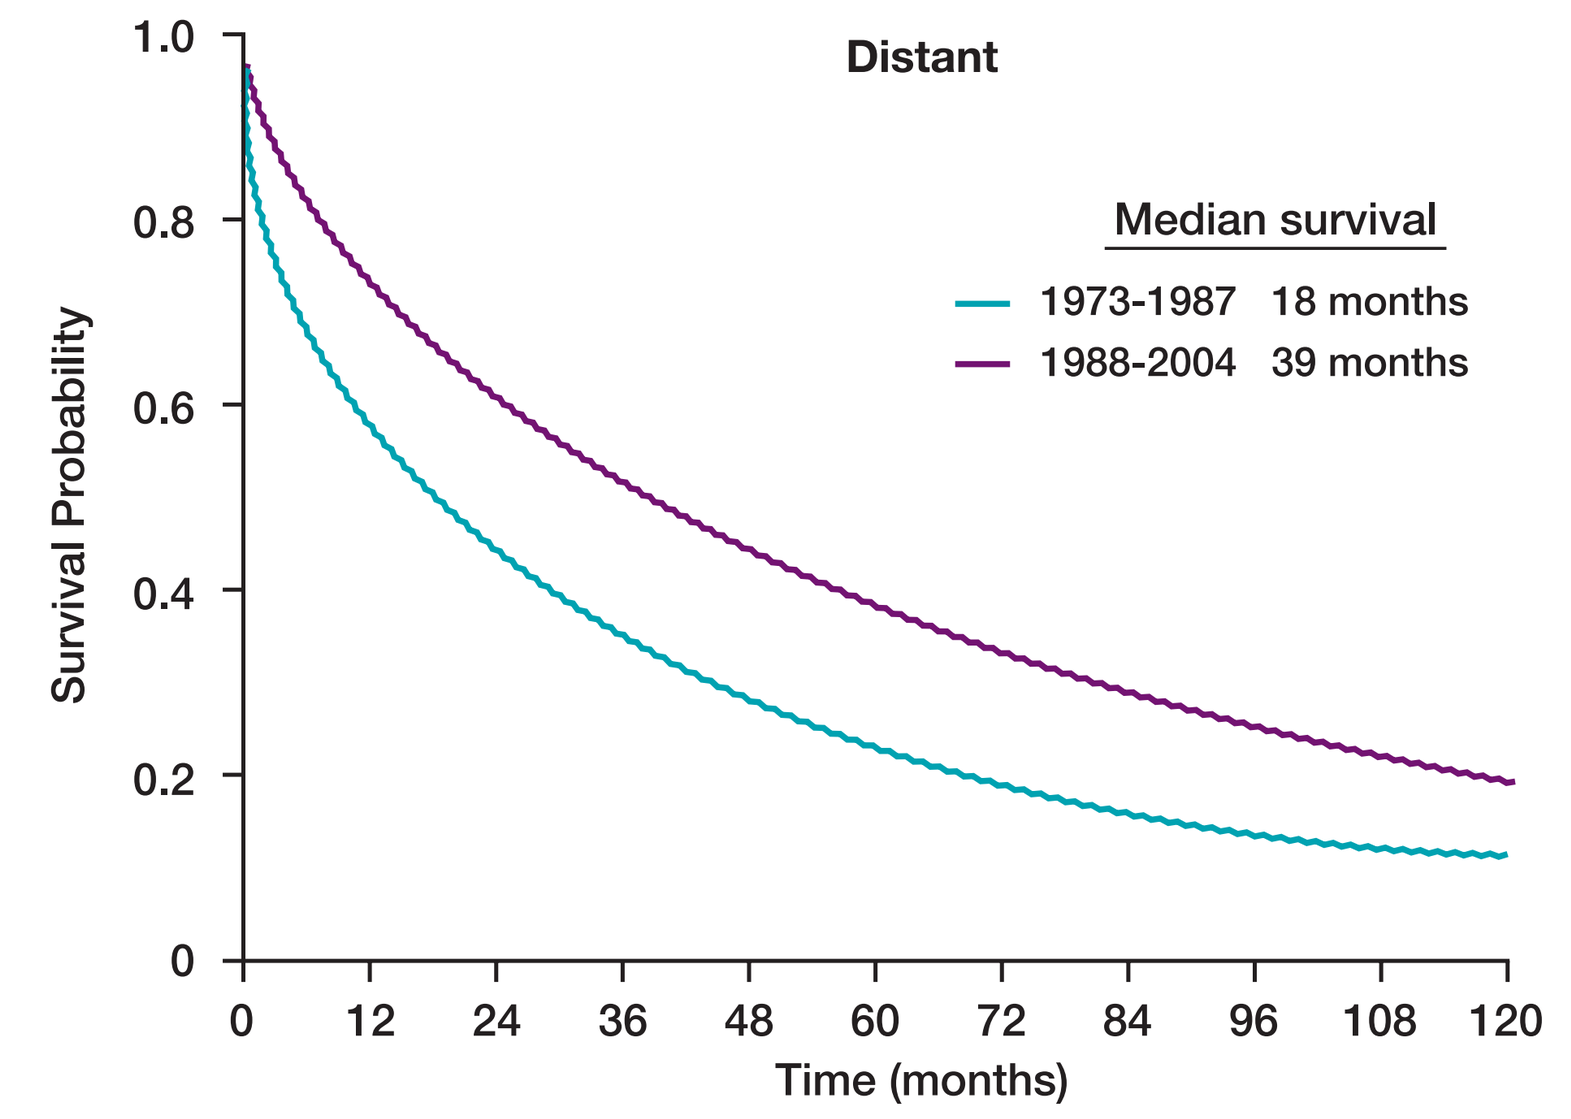
Distant
Median survival
1973-1987 18 months
1988-2004 39 months
Survival Probability
Time (months)

Reprinted with permission. © 2008 American Society of Clinical Oncology. All rights reserved.

Rationale for an antiproliferative effect of octreotide sc and LAR

The biological basis for the antisecretory and antiproliferative effect of octreotide LAR

- Octreotide is a somatostatin analogue with high-affinity binding to sst_2 and sst_5, somatostatin receptors subtypes which are expressed on the majority of GEP-NETs (Table 1).
- Octreotide LAR has antisecretory and direct antitumor effects through multiple receptor-mediated signal transduction pathways; indirect antitumor effects are mediated via inhibition of angiogenesis, suppression of growth factor and growth-promoting hormone synthesis/secretion or immunomodulatory effects[3] (Table 1).

Table 1. Somatostatin receptor subtypes, sst_{1-5}
A. Receptor binding affinities of somatostatin, octreotide, lanreotide and pasireotide; B. Prevalence on GEP-NETs; C. Mediation of exocrine secretion; D. Mediation of cell proliferation

	sst_1	sst_2	sst_3	sst_4	sst_5
A. Receptor binding affinity (IC_{50} nmol/L)					
Somatostatin[4-6]	0.93	0.15	0.56	1.50	0.29
Octreotide[4-6]	**280**	**0.38**	**7.10**	**>1000**	**6.30**
Lanreotide[7]	>1000	0.8	107	>1000	5.2
Pasireotide[4-6]	9.3	1.0	1.5	>1000	0.16
B. Prevalence on GEP-NETs [% NETs expressing sst_{1-5} (n sampled)][7]					
Gastrointestinal	76% (59)	80% (84)	43% (58)	68% (47)	77% (44)
Gastrinoma	79% (28)	93% (28)	36% (28)	61% (23)	93% (28)
Insulinoma	76% (21)	81% (21)	38% (21)	58% (19)	57% (21)
Nonfunctioning pancreatic NET	58% (24)	88% (24)	42% (24)	48% (21)	50% (24)
C. Mediation of exocrine secretion[3,8]					
Gastric acid		+			
Amylase					+
Intestinal GI secretion	+	+			
D. Mediation of cell proliferation[3,8]					
Induction of G_1 cell cycle arrest	+	+		+	+
Induction of apoptosis		+	+		

Early clinical evidence for the antiproliferative effect of octreotide sc and LAR

- Somatostatin analogues achieve tumor stabilization in up to 57% of patients with GEP-NETs and documented tumor progression. Partial or complete response is achieved in <10% of patients.[2]
- Octreotide sc and LAR have been associated with tumor stabilization in many studies, and partial tumor response in some patients (Table 2).
- Although results from these studies suggest that octreotide stabilizes tumor growth in a subset of patients, these studies were not designed to definitely determine the antiproliferative effect of octreotide. The studies were not placebo controlled, included a heterogeneous patient population with GEP-NETs of different origin and biological behavior and, in most cases, did not include treatment-naïve patients.

Table 2. Summary of tumor responses associated with octreotide sc and LAR in patients with GEP-NETs. Data from early and/or uncontrolled studies

Reference	Regimen	Response
Octreotide sc		
Arnold R *et al. Metabolism* 1992;41:116–118	Octreotide sc 200 µg tid (n=68)	SD in 50% and PR in 4.4% at 3 months; SR or PR in 38% at 12 months
Saltz L *et al. Cancer* 1993;72:244–248	Octreotide sc 250 µg tid (n=34)	SD in 50% of patients for 2–27 months
Arnold R *et al. World J Surg* 1993;17:511–519	Octreotide sc 200 µg tid (n=12)	SD in 42% of patients for 6–28 months
Arnold R *et al. Gut* 1996;38:430–438	Octreotide sc 200 µg tid (n=52)	SD in 37% of patients for 3–>42 months
di Bartolomeo M *et al. Cancer* 1996;77:402–408	Octreotide sc 500 µg tid (n=23)	SD in 47% of patients for 6–>32 months
	Octreotide sc 1000 µg tid (n=35)	PR in 3% of patients
Arnold R *et al. Clin Gastroenterol Hepatol* 2005;3:761–771	Octreotide sc 200 µg tid (n=51)	No significant difference between treatment arms
	OR	
	Octreotide sc 200 µg tid + Interferon-α 4.5 x 106 IU tiw (n=54)	SD or PR in 48% for 3 months; 29% for 6 months; 21% for 12 months
Octreotide LAR		
Ricci S *et al. Ann Oncol* 2000;11:1127–1130	Octreotide LAR 20 mg/28d (n=15)	SD in 40% and PR in 7% of patients for 6–>12 months
Shojamanesh H *et al. Cancer* 2002;94:331–343	Octreotide LAR 20–30 mg/28d (n=15)	SD in 47% and PR in 7% of patients for 5.5–54 months
Panzuto F *et al. Ann Oncol* 2006;17:461–466	Octreotide LAR 30 mg/28d (n=20)	SD in 45.2% of patients for median 6–60 months
	Lanreotide SR 60 mg/28d (n=11)	

Octreotide LAR demonstrates a significant antiproliferative effect

Results of the PROMID trial

- Recently, the first randomized, double-blind, placebo-controlled trial (PROMID*) evaluated the antiproliferative effect of octreotide LAR 30 mg in treatment-naïve patients with well-differentiated, metastatic or locally advanced, functioning or nonfunctioning, midgut NETs.[9]
- In this patient group, octreotide LAR 30 mg significantly lengthened time to tumor progression (TTP) *vs.* placebo, regardless of functional status (14.3 *vs.* 6.0 months; P=0.000072)[9] (Figure 3).
- At 6 months, partial response or stable disease was achieved in 29/42 (69.0%) patients treated with octreotide LAR 30 mg and in 17/43 (39.5%) placebo recipients (octreotide LAR 30 mg vs placebo; P=0.0079).[9]
- Significantly extended TTP with octreotide LAR 30 mg was seen in patients with either functioning or nonfunctioning tumors (patients with or without carcinoid syndrome)[9] (Figure 4).
- Octreotide LAR 30 mg significantly extended TTP regardless of elevated or non-elevated chromogranin A, or age <63 or ≥63 years[9] (Figure 5).

** Placebo-Controlled, Double-Blind, Prospective, Randomized Study on the Effect of Octreotide LAR in the Control of Tumor Growth in Patients with Metastatic Neuroendocrine MIDgut Tumors.*

Figure 3. Octreotide LAR 30 mg significantly increases TTP in treatment-naïve patients with well-differentiated, metastatic, midgut NETs[9]

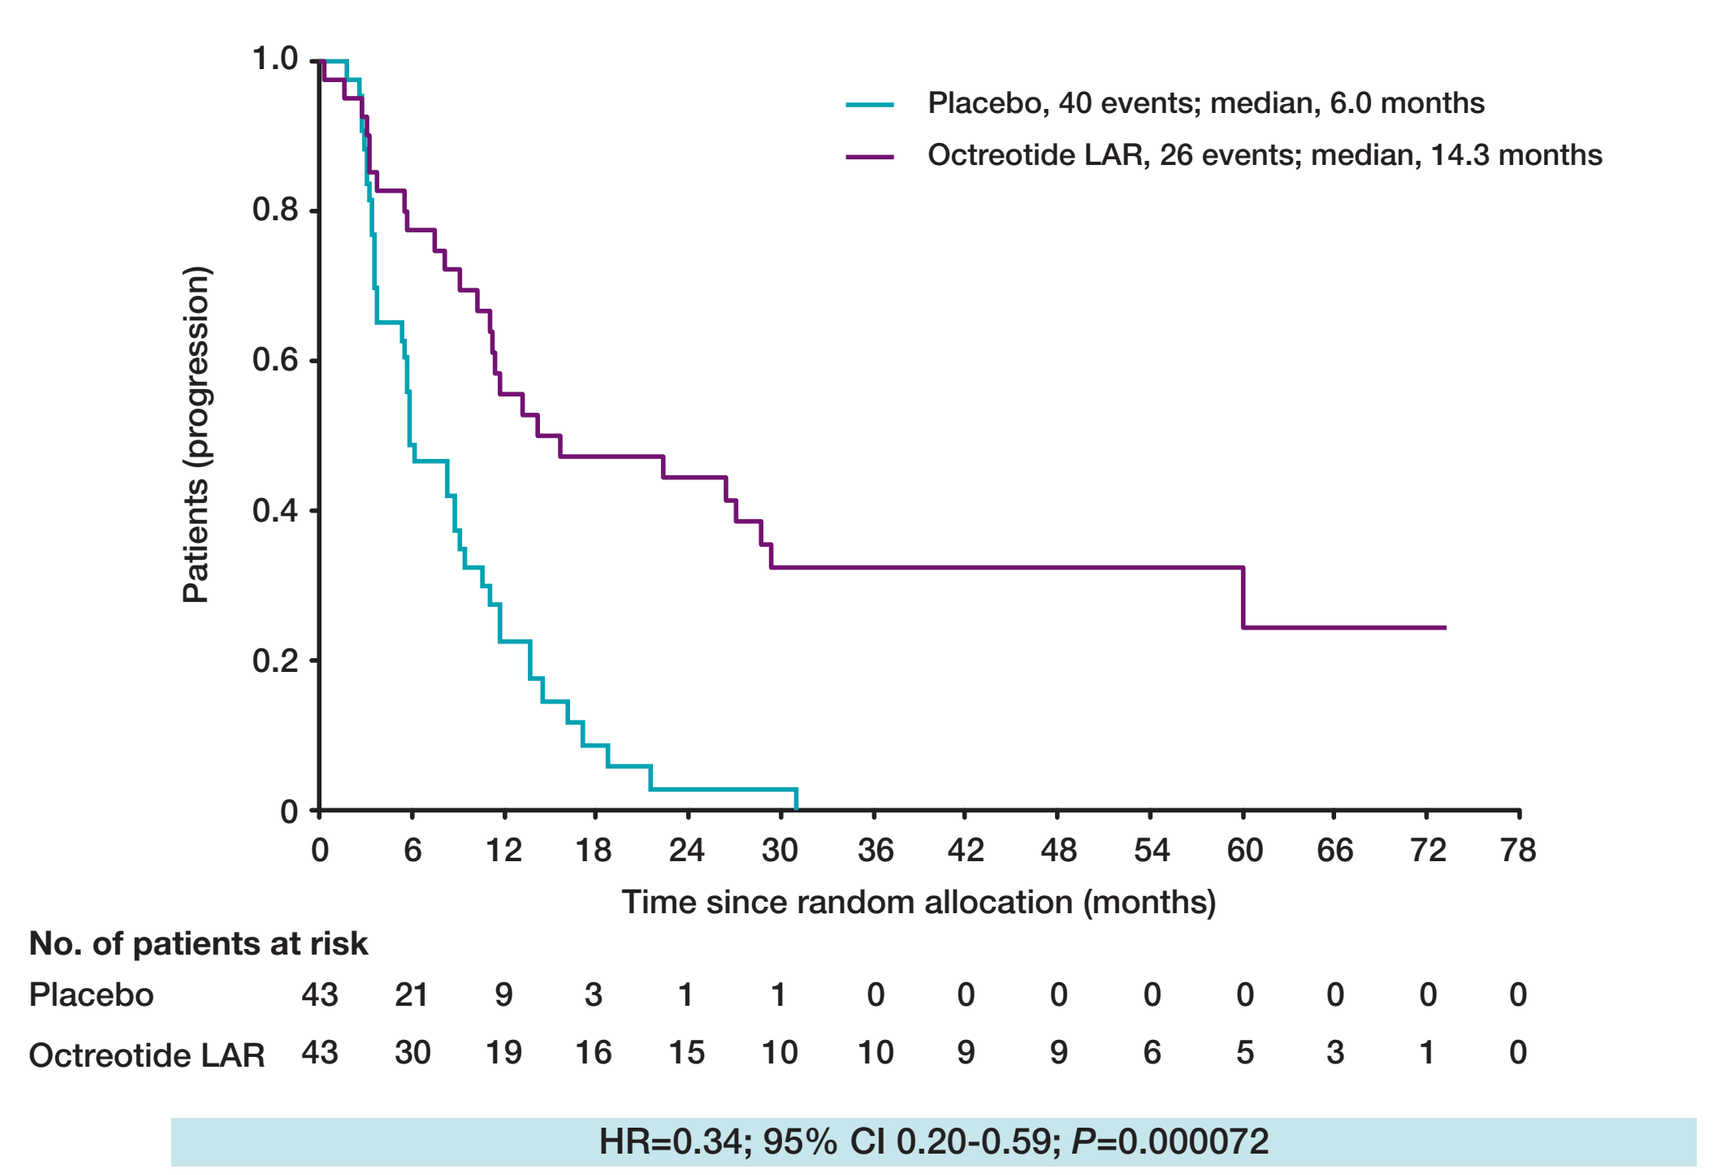
Placebo, 40 events; median, 6.0 months
Octreotide LAR, 26 events; median, 14.3 months
Patients (progression)
Time since random allocation (months)
No. of patients at risk
Placebo 43 21 9 3 1 1 0 0 0 0 0 0 0 0
Octreotide LAR 43 30 19 16 15 10 10 9 9 6 5 3 1 0
HR=0.34; 95% CI 0.20-0.59; P=0.000072

Reprinted with permission. © 2009 American Society of Clinical Oncology. All rights reserved.

Figure 4. Octreotide LAR 30 mg significantly increases TTP in patients with well-differentiated, metastatic, midgut NETs, regardless of functional status[9,10]

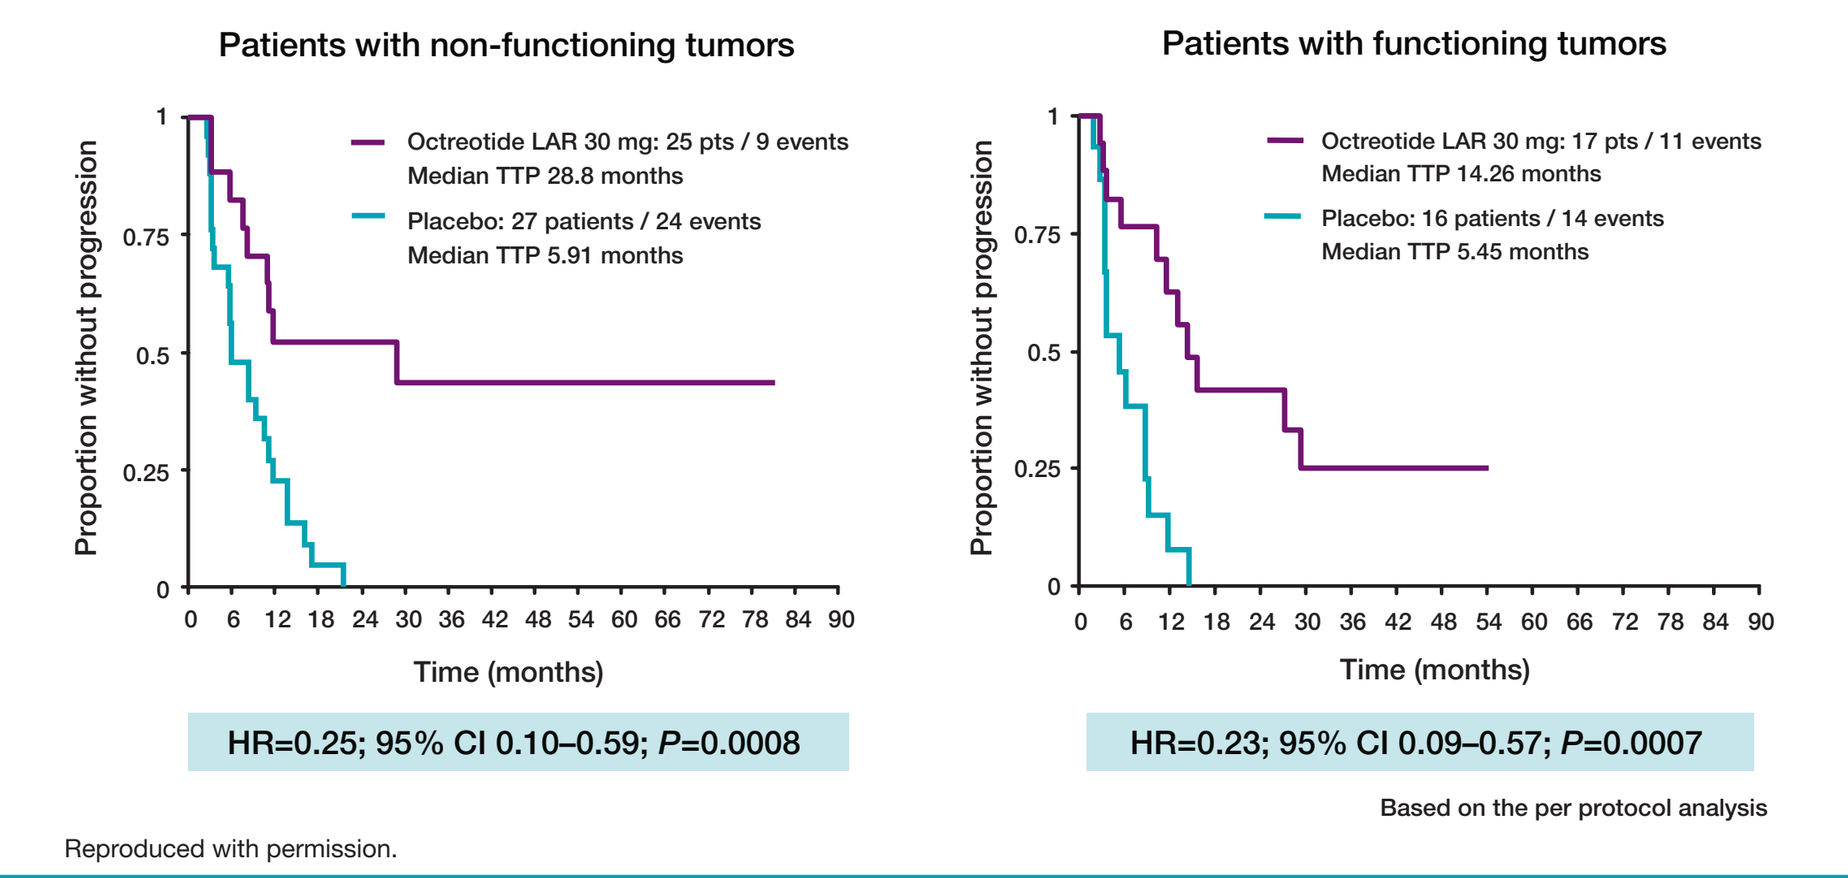
Patients with non-functioning tumors
Octreotide LAR 30 mg: 25 pts / 9 events
Median TTP 28.8 months
Placebo: 27 patients / 24 events
Median TTP 5.91 months
Proportion without progression
Time (months)
HR=0.25; 95% CI 0.10–0.59; P=0.0008
Patients with functioning tumors
Octreotide LAR 30 mg: 17 pts / 11 events
Median TTP 14.26 months
Placebo: 16 patients / 14 events
Median TTP 5.45 months
Proportion without progression
Time (months)
HR=0.23; 95% CI 0.09–0.57; P=0.0007
Based on the per protocol analysis

Reproduced with permission.

Figure 5. PROMID subgroup analysis of TTP in patients treated with octreotide LAR 30 mg or placebo.[9] Four of the five subgroups with confidence intervals crossing 1 had small patient numbers (≤14 patients)

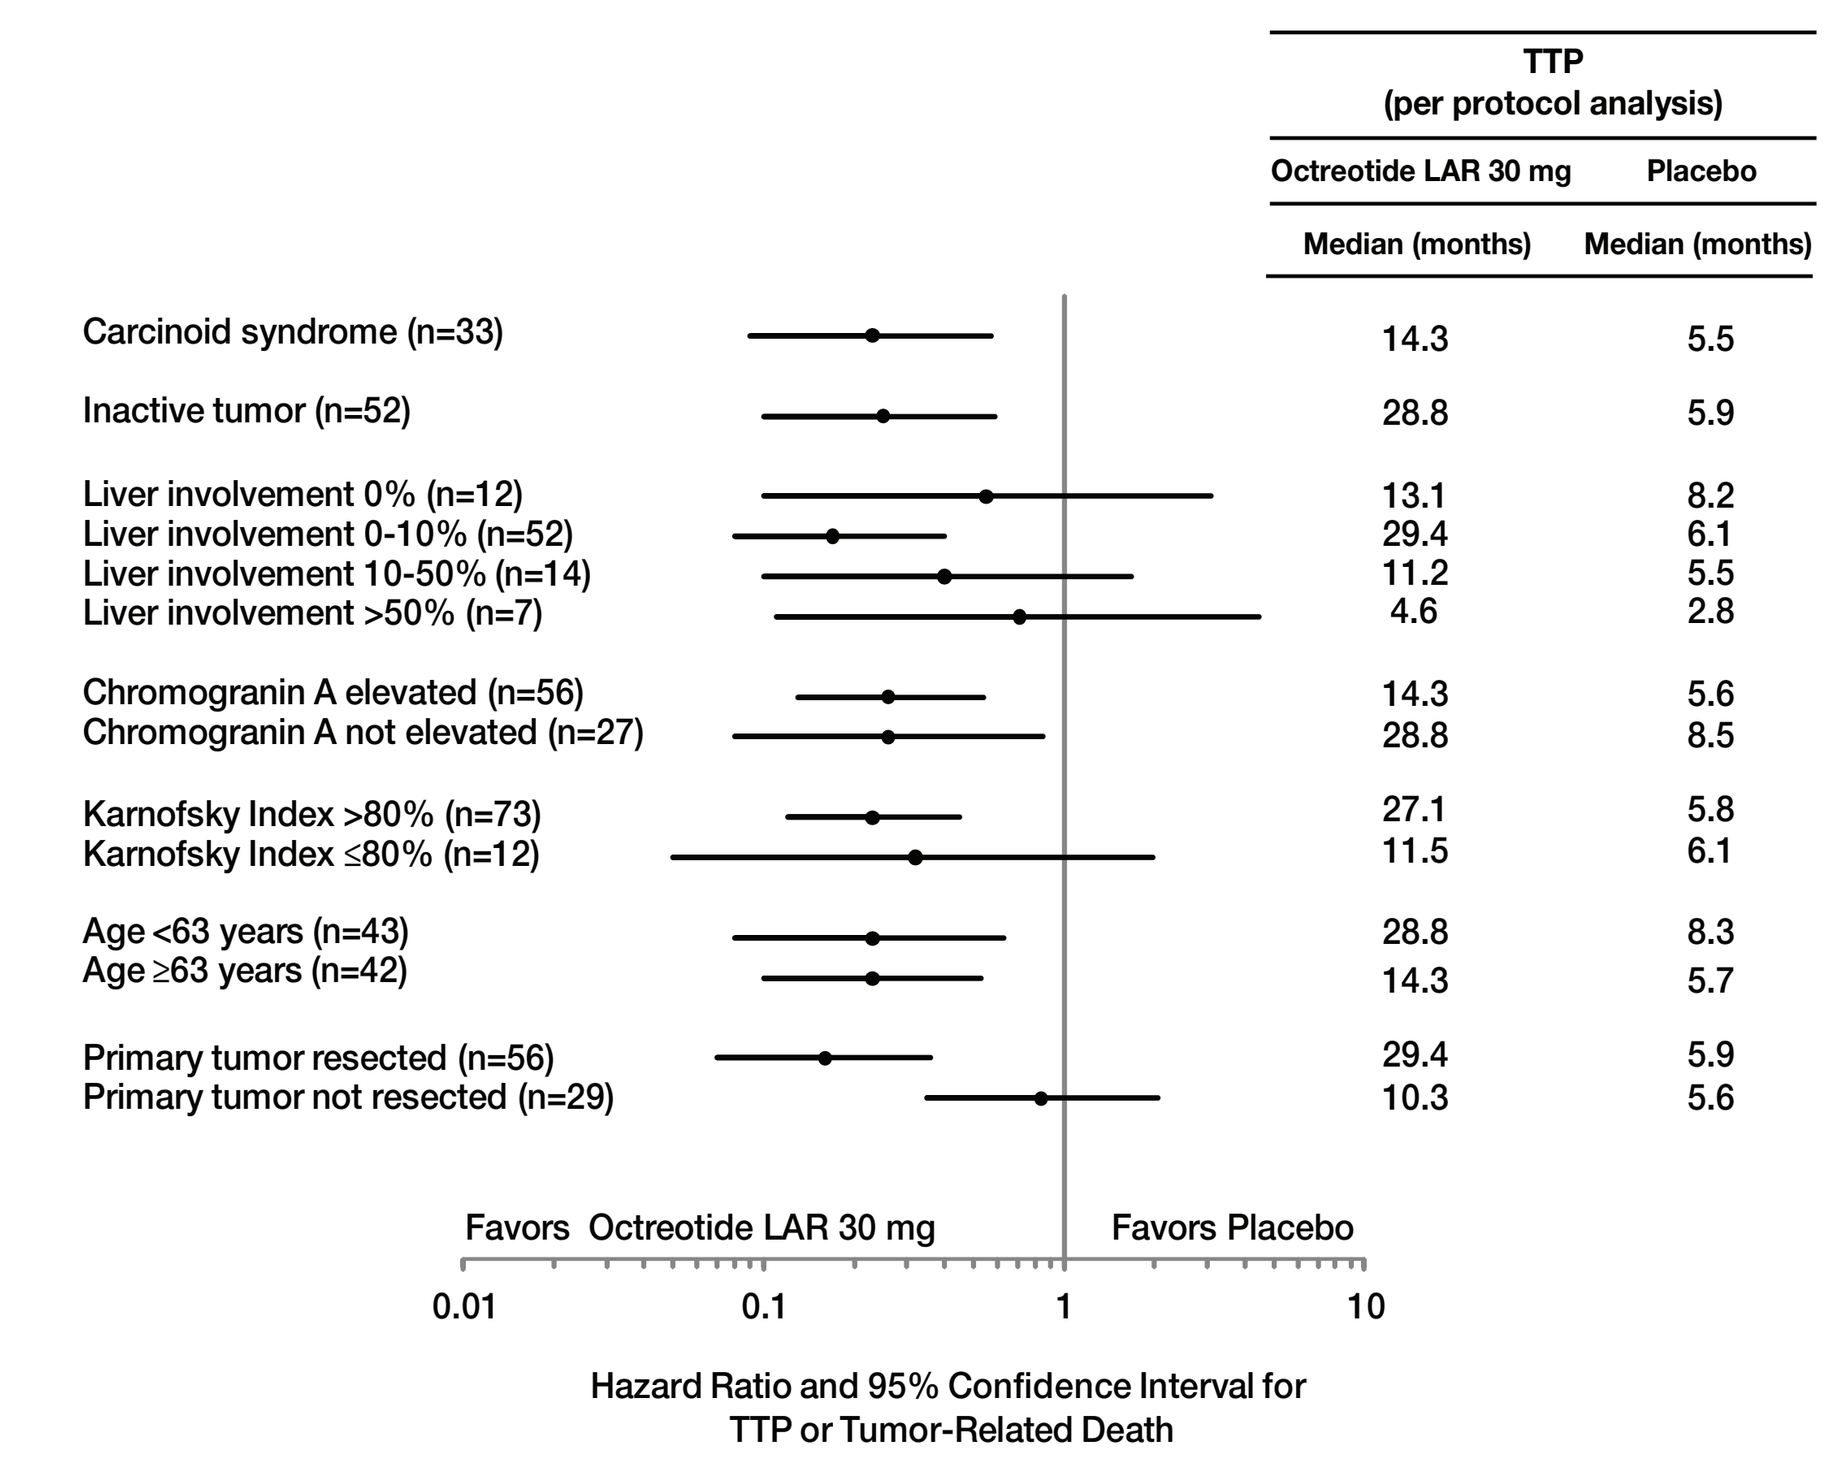
TTP
(per protocol analysis)
Octreotide LAR 30 mg Placebo
Median (months) Median (months)
Carcinoid syndrome (n=33) 14.3 5.5
Inactive tumor (n=52) 28.8 5.9
Liver involvement 0% (n=12) 13.1 8.2
Liver involvement 0-10% (n=52) 29.4 6.1
Liver involvement 10-50% (n=14) 11.2 5.5
Liver involvement >50% (n=7) 4.6 2.8
Chromogranin A elevated (n=56) 14.3 5.6
Chromogranin A not elevated (n=27) 28.8 8.5
Karnofsky Index >80% (n=73) 27.1 5.8
Karnofsky Index ≤80% (n=12) 11.5 6.1
Age <63 years (n=43) 28.8 8.3
Age ≥63 years (n=42) 14.3 5.7
Primary tumor resected (n=56) 29.4 5.9
Primary tumor not resected (n=29) 10.3 5.6
Favors Octreotide LAR 30 mg
Favors Placebo
0.01 0.1 1 10
Hazard Ratio and 95% Confidence Interval for TTP or Tumor-Related Death

Impact of the PROMID trial on clinical practice

- The findings of the PROMID trial will widen the accepted use of octreotide LAR to include both functioning and nonfunctioning NETs.[11]
- The US National Comprehensive Cancer Network (NCCN) has recently published updated treatment guidelines for foregut, midgut and hindgut NETs, expanding the use of octreotide LAR. Octreotide LAR is now a treatment option for asymptomatic, unresectable, metastatic carcinoid tumors, irrespective of the site of the primary tumor, as well as for symptomatic tumors with significant tumor burden or significant tumor progression.[12]
- The use of octreotide LAR in nonfunctioning NETs and as an antitumor agent may be reflected in updated recommendations from international NETs societies, such as those from Europe (ENETS)[11] and North America (NANETS).

Potential future medical therapies for patients with GEP-NETs

- Phase III trials of octreotide LAR in combination with oral, once-daily everolimus (RAD001), an inhibitor of mTOR (a central regulator of cell growth, metabolism and angiogenesis) are ongoing in patients with gastrointestinal (GI) and pancreatic NETs (Table 3). A potential synergistic effect may be seen when these two agents are administered in combination.
- Pasireotide is a novel multi-receptor ligand somatostatin analogue with high affinity for $sst_{1,2,3}$ and sst_5, and approximately 30-, 11- and 158-fold higher functional activity than octreotide on sst_1, sst_3 and sst_5, respectively, and 7-fold lower on sst_2.[5,6] Pasireotide may have an antiproliferative effect similar to or greater than that achieved by octreotide LAR 30 mg by acting at multiple somatostatin receptors.

Table 3. Recent and future trials in patients with metastatic GEP-NETs administered somatostatin analogues alone or in combination with other agents for antiproliferative therapy

Trial	Patients	Endpoint/Outcome	Key Points
Data available			
PROMID **Rinke A *et al.* *J Clin Oncol* 2009;27:4656-4663** Octreotide LAR 30 mg/month *vs.* placebo **Phase III**	Functioning or nonfunctioning metastatic, well-differentiated NETs of the midgut	Median TTP: 14.3 months with octreotide LAR *vs.* 6.0 months with placebo (P<0.000072) SD or PR: 69.0% octreotide LAR; 39.5% placebo (P=0.0079)	Octreotide LAR 30 mg can now be considered as a treatment option in patients with nonfunctioning and functioning NETs
Yao JC *et al.* J Clin Oncol 2008; 26:4311–4318 Everolimus 5 or 10 mg/day plus octreotide LAR 30 mg/month **Phase II**	Metastatic low- to intermediate-grade NETs* of pancreatic or GI origin	SD in 70% of patients PR in 22% Median PFS 60 weeks	Median PFS compared favorably with rates recently reported in Phase II and III studies with chemotherapy, bortezomib, imatinib, sorafenib, sunitinib and bevacizumab
RADIANT-1 **Yao JC *et al.* World GI 2009** Everolimus 10 mg/day with or without octreotide ≤30 mg/month **Phase II**	Advanced pancreatic NETs following chemotherapy failure	Combination therapy: SD in 80% PR in 4% Median PFS 16.7 months	Combination octreotide LAR and everolimus may offer a novel treatment approach for disease and symptom control in patients with advanced pancreatic NETs
Awaiting data			
RADIANT-2 Everolimus 10 mg/day + octreotide LAR 30 mg/month *vs.* everolimus 10 mg/day + placebo **Phase III**	Progressive, functioning GI-NETs of low-intermediate grade*	PFS, tumor response	Octreotide LAR in combination with everolimus may have a synergistic effect
Pasireotide LAR *vs.* octreotide LAR **Phase III**	Patients with metastatic well-differentiated GEP-NETs whose symptoms are inadequately controlled by the maximum dose of currently available somatostatin analogues	Symptom response, tumor response	By acting at multiple somatostatin receptors, pasireotide may have an antiproliferative effect similar to or greater than that achieved by octreotide
Lanreotide Autogel 120 mg/month *vs.* placebo **Phase III**	Patients with locally advanced or metastatic, nonfunctioning, GEP-NETs	PFS, tumor response	Initiated: 2006 Estimated completion date: 2012 Estimated enrollment: 200

** 'Low- to intermediate-grade NETs' are referred to as 'well-differentiated neuroendocrine tumors' and 'well-differentiated neuroendocrine carcinomas' using the WHO classification.*
PFS=progression-free survival; PR=partial response; SD=stable disease

Conclusions

Widening use of octreotide LAR beyond symptom control to tumor control

- The incidence of GEP-NETs is rising. Most patients present with metastatic GEP-NETs and are unable to achieve cure after surgery.
- The PROMID trial confirmed the antiproliferative benefit of octreotide LAR 30 mg in treatment-naïve patients with well-differentiated, metastatic midgut NETs, regardless of tumor functional status.
- Patients with functioning or nonfunctioning metastatic NETs of the midgut should now be considered for treatment with octreotide LAR 30 mg.
- Octreotide LAR in combination with everolimus has shown promising results to date and may prove to be a treatment option for patients with GEP-NETs.
- Evaluation of pasireotide in patients with GEP-NETs is ongoing.

References

1. Yao JC *et al. J Clin Oncol* 2008;26:3063-3072.
2. Eriksson B *et al. Neuroendocrinology* 2008;87:8-19.
3. Susini C & Buscail L. *Ann Oncol* 2006;17:1733-1742.
4. Bruns C *et al. Eur J Endocrinol* 2002;146:707-716.
5. Schmid HA & Schoeffter P. *Neuroendocrinology* 2004;80(Suppl 1):47-50.
6. Schmid HA. *Mol Cell Endocrinol* 2008;286:69-74.
7. Hofland LJ & Lamberts SW. *Endocr Rev* 2003;24:28-47.
8. Weckbecker G *et al. Nat Rev Drug Discov* 2003;2:999-1017.
9. Rinke A *et al. J Clin Oncol* 2009;27:4656-4663.
10. Arnold R. Oral presentation at ASCO 2009.
11. Oberg KE. *J Clin Oncol* 2009;[Epub ahead of print].
12. Clark OH *et al. J Natl Compr Canc Netw* 2009;7:712-747.

NANETS; October 2 & 3, 2009; Charlotte, North Carolina, USA.