LX1032: A Novel Agent to Reduce Peripheral Serotonin as a Potential Treatment for Carcinoid Syndrome

Philip Brown, Kenny S. Frazier, Shanna Jackson, Anne Turnage, William E. Heydorn, Qi Yang, Paddy Yalamanchili, Qingyun Liu
Lexicon Pharmaceuticals, Inc., The Woodlands, TX, United States 77381

INTRODUCTION

LX1032 is a novel, orally delivered, inhibitor of tryptophan hydroxylase (TPH), the rate-limiting enzyme in the biosynthesis of serotonin (5-HT). Elevated 5-HT is a hallmark finding in carcinoid tumors, and in advanced forms of the disease 5-HT is thought to contribute to symptoms associated with carcinoid syndrome (CS), including diarrhea, abdominal pain and cramping, as well as longer-term disease sequelae such as mesenteric and cardiac fibrosis. In advanced tumors, morbidity and mortality relate as much to the secretion of 5-HT and peptide hormones, as to tumor growth and metastasis.

In humans, as well as in other mammals, there are two distinct TPH genes that encode two isoforms of TPH:[1-3] TPH1, found predominantly in the enterochromaffin (EC) cells of the GI tract, and TPH2, which is expressed in neuronal cell types and is the predominant isoform in the brain. The vast majority (95%) of 5-HT is produced in the periphery, principally by EC cells.

Prior experience in the 1960's with another TPH inhibitor, para-chlorophenylalanine (pCPA), illustrated that reduction in 5-HT synthesis could provide a significant improvement in symptoms of patients with CS.[4] Further development of pCPA was ultimately curtailed because it crossed the blood-brain-barrier, resulting in depletion of brain 5-HT and subsequent neuro-psychiatric side effects such as depression. We therefore developed an agent capable of inhibiting 5-HT production in the periphery without affecting brain 5-HT production that could potentially exert a therapeutic effect on multiple CS symptoms without causing undesired effects in the CNS. Consistent with this approach *Tph1* knockout mice, which have almost no intestinal 5-HT but normal levels of brain 5-HT (Figure 1), displayed no abnormalities in any neurobehavioral screens.

In preclinical studies, LX1032 was shown to reduce peripheral 5-HT in a dose-dependent fashion without affecting brain 5-HT, recapitulating the *Tph1* knockout mouse phenotype (Figure 2). These important preclinical observations have now translated into favorable results in Phase 1 clinical studies involving 87 normal healthy volunteers. In Phase 1 studies, LX1032 was well tolerated and produced a reduction in peripheral 5-HT levels, suggesting that LX1032 may provide a new approach for the management of many of the symptoms and morbidity experienced by patients with CS, by altering peripheral 5-HT levels without impacting brain 5-HT.

PRECLINICAL DATA

- *Tph1* knockout mice have almost no 5-HT in the GI tract and blood, but maintain normal levels of 5-HT in the brain (Figure 1).
- *Tph1* knockout mice are generally healthy and GI function is preserved, indicating that loss of TPH1 function is not deleterious.
- The effect of LX1032 in wild-type mice recapitulates the *Tph1* knockout mouse phenotype by reducing 5-HT levels in the intestines but not in the brain (Figure 2). On the contrary, pCPA significantly reduces brain 5-HT (Figure 3).
- In carcinoid tumor cell lines, LX1032 inhibits serotonin production with a greater potency than pCPA (Figure 4).
- LX1032 demonstrated a good safety profile in a standard battery of pharmacologic safety and genetic toxicology studies, indicating that dosing in humans could proceed. These studies yielded the following results:
 - *In vivo*, LX1032 is rapidly and efficiently converted in all species examined through esterase activity to its active metabolite.
 - The active metabolite of LX1032 had no effect on the hERG channel (*in vitro*).
 - All electrocardiograms (in dog) were normal up to the highest dose tested (200 mg/kg) (i.e., no effect on QT interval, QTc, heart rate, or blood pressure).
 - There were no effects on CNS activity or respiratory system up to the highest dose tested (2000 mg/kg).
 - In the GI system LX1032 produced a dose-related and statistically significant decrease in GI transit and stomach emptying, most likely due to the known pharmacology of the compound.

REFERENCES:
1. Gingrich JA, Ansorge MS, Merker R, Weisstaub N and Zhou M, New lessons from knockout mice: The role of serotonin during development and its possible contribution to the origins of neuropsychiatric disorders. CNS Spectr 2003, Aug; 8(8);572-7.
2. Walther, D., et. al., Synthesis of Serotonin by a Second Tryptophan Hydroxylase Isoform. Science 2003, Jan 3;299(5603):76.
3. Veenstra-VanderWeele J., Cook EH., Knockout Mouse Points to Second Form of Tryptophan Hydroxylase. Mol Interv. 2003 Mar;3(2):72-5, 50.
4. Engelman K, Lovenberg W, and Sjoerdsma A, Inhibition of Sertonin Synthesis by Para-chlorophenylalanine in Patients with the Carcinoid Syndrome. NEJM 1967, Nov; 277(21)l; 1103-1108.

ACKNOWLEDGMENT: We sincerely appreciate the help of D. Wade Walke, Jay L. Mitchell, and Penny Logan in producing this poster.

PRECLINICAL DATA (continued)

Figure 1. *Tph1* knockout mice reveal a new mechanism to target the serotonin pathway.

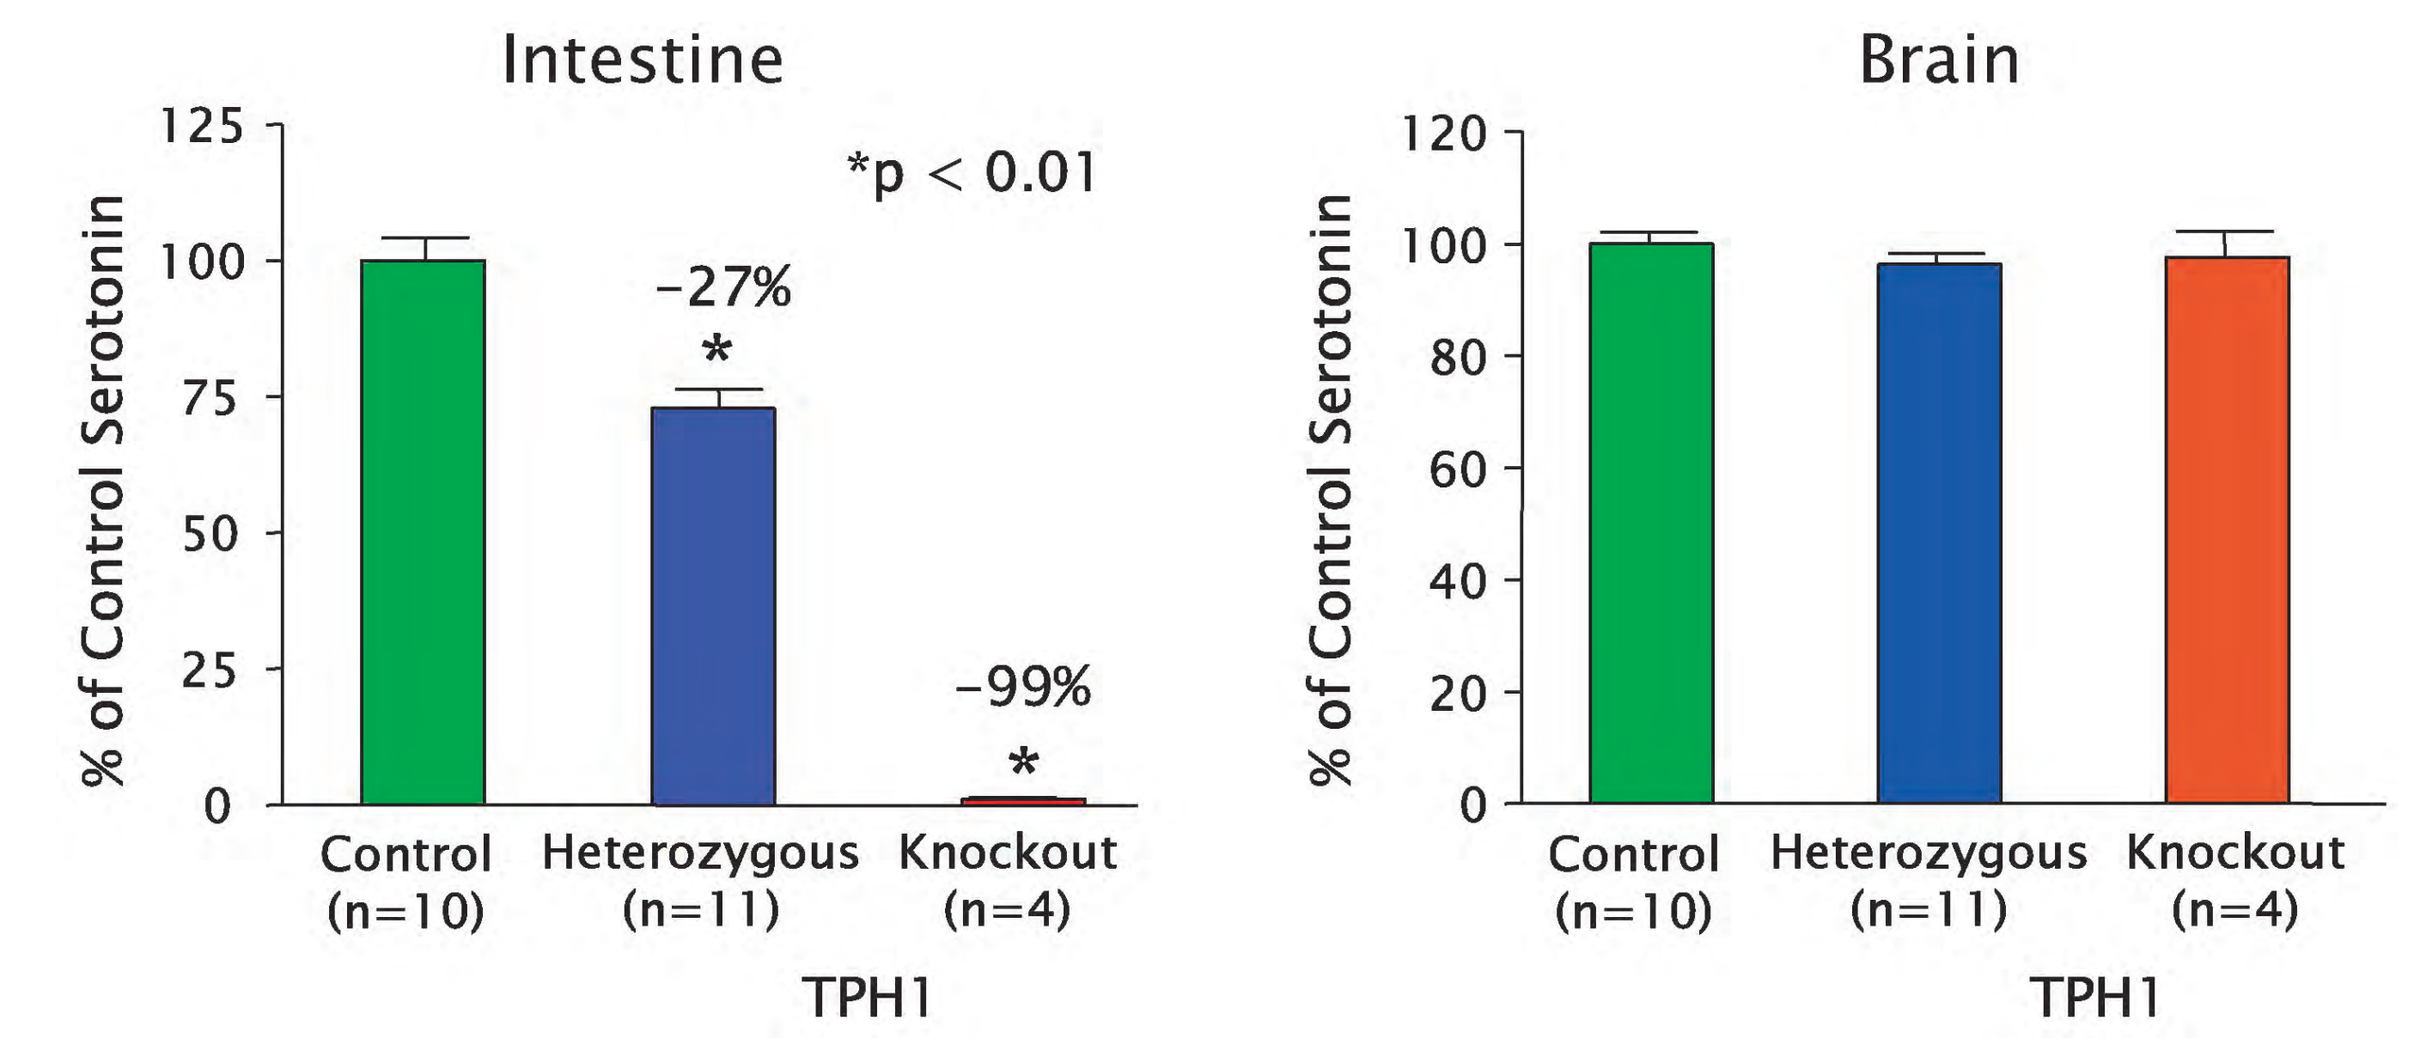
Intestine
*p < 0.01
% of Control Serotonin
-27%
-99%
Control (n=10)
Heterozygous (n=11)
Knockout (n=4)
TPH1
Brain
% of Control Serotonin
Control (n=10)
Heterozygous (n=11)
Knockout (n=4)
TPH1

Figure 2. LX1032 reduces serotonin levels in the GI tract but does not affect brain serotonin levels of mice (day 4).

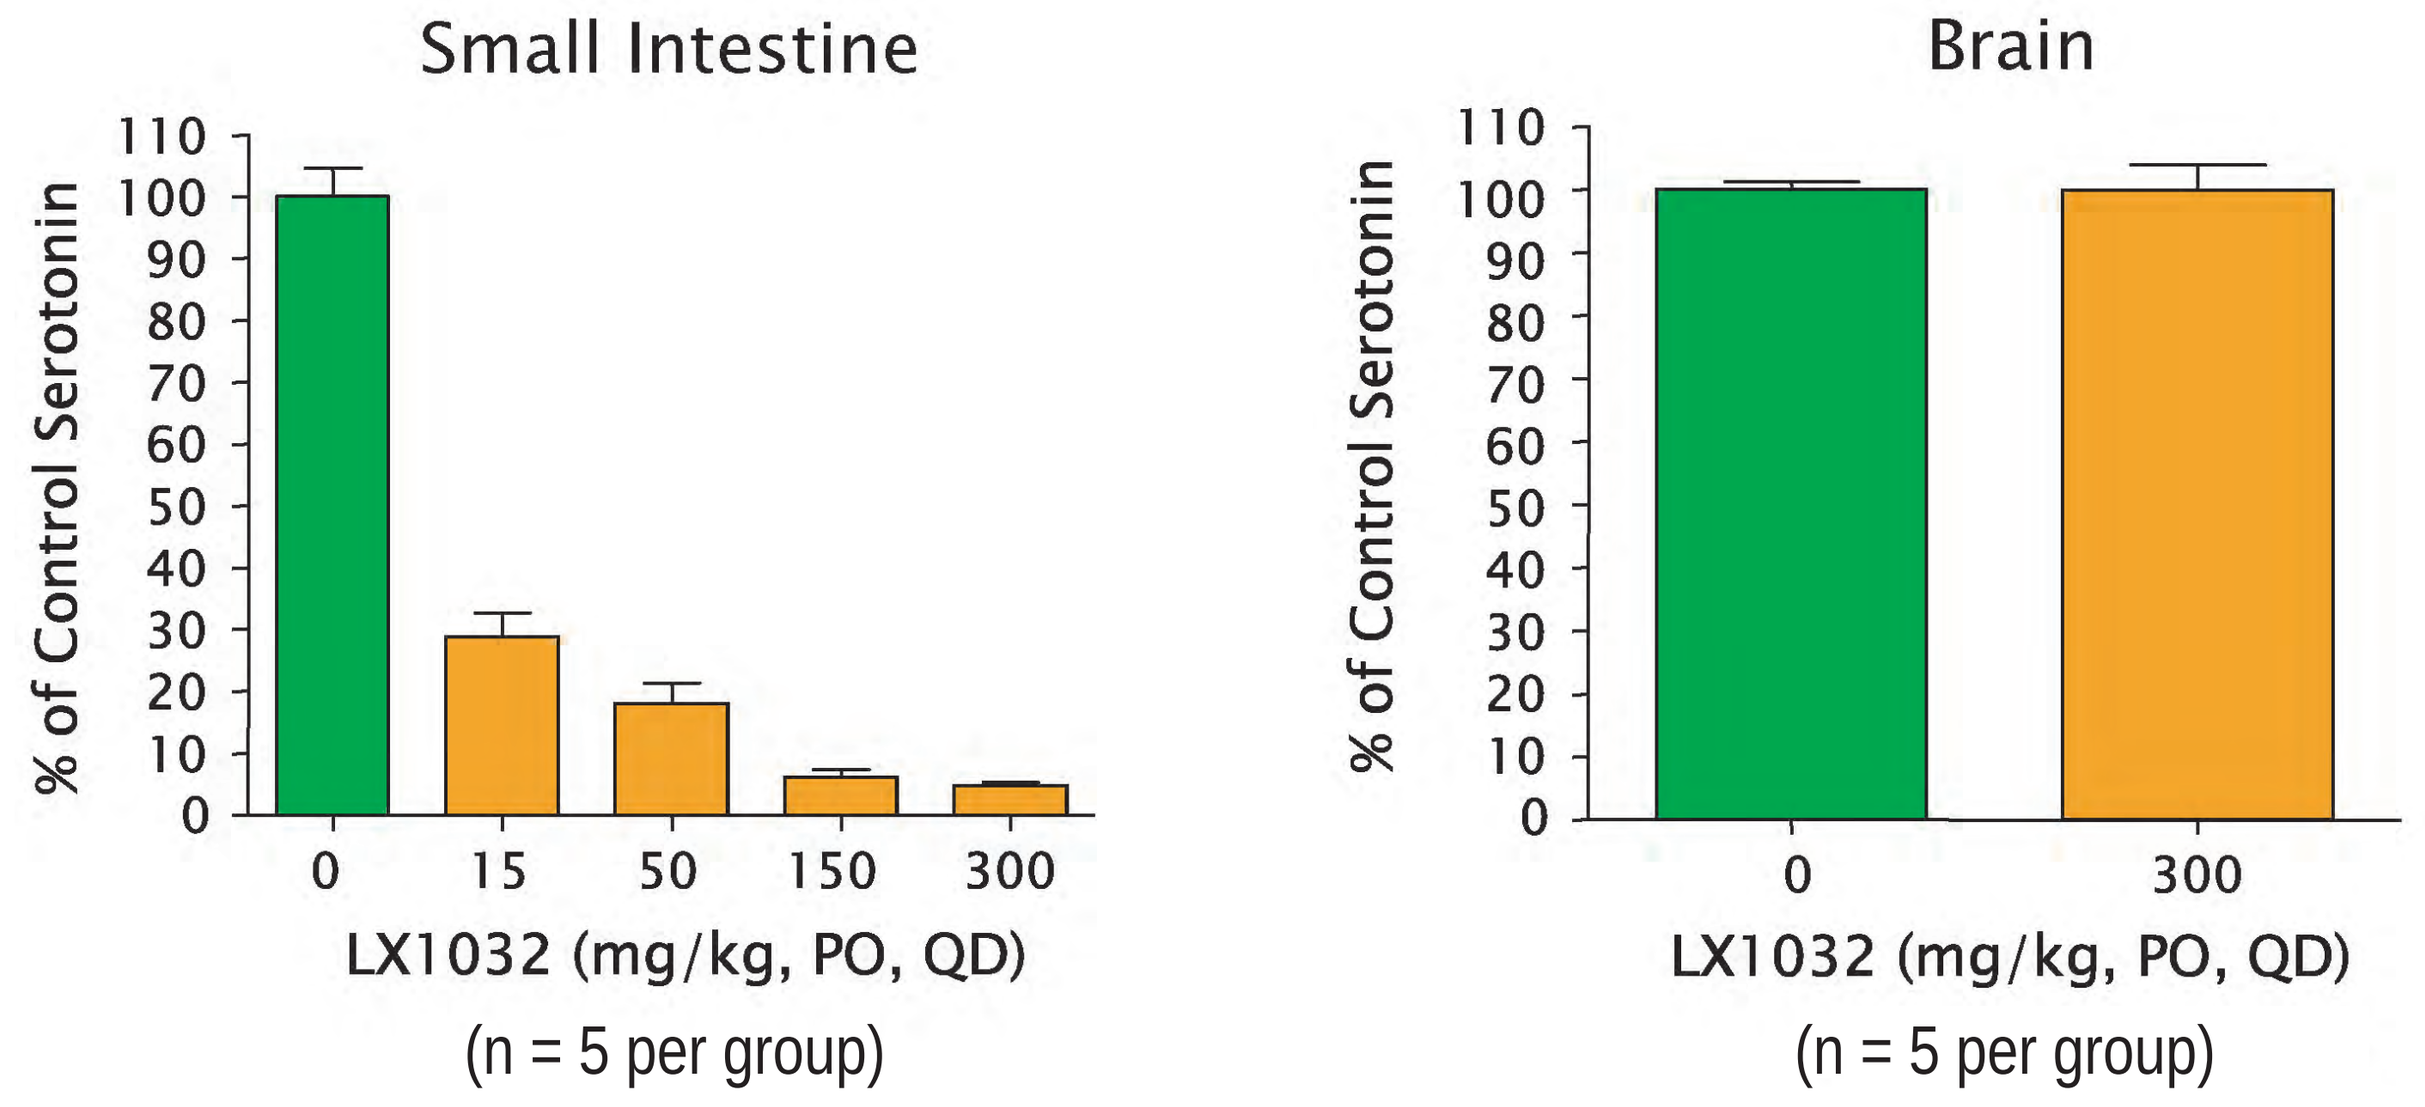
Small Intestine
% of Control Serotonin
0
15
50
150
300
LX1032 (mg/kg, PO, QD)
(n = 5 per group)
Brain
% of Control Serotonin
0
300
LX1032 (mg/kg, PO, QD)
(n = 5 per group)

Figure 3. LX1032 has no effect on brain serotonin levels, unlike pCPA (days 4-5).

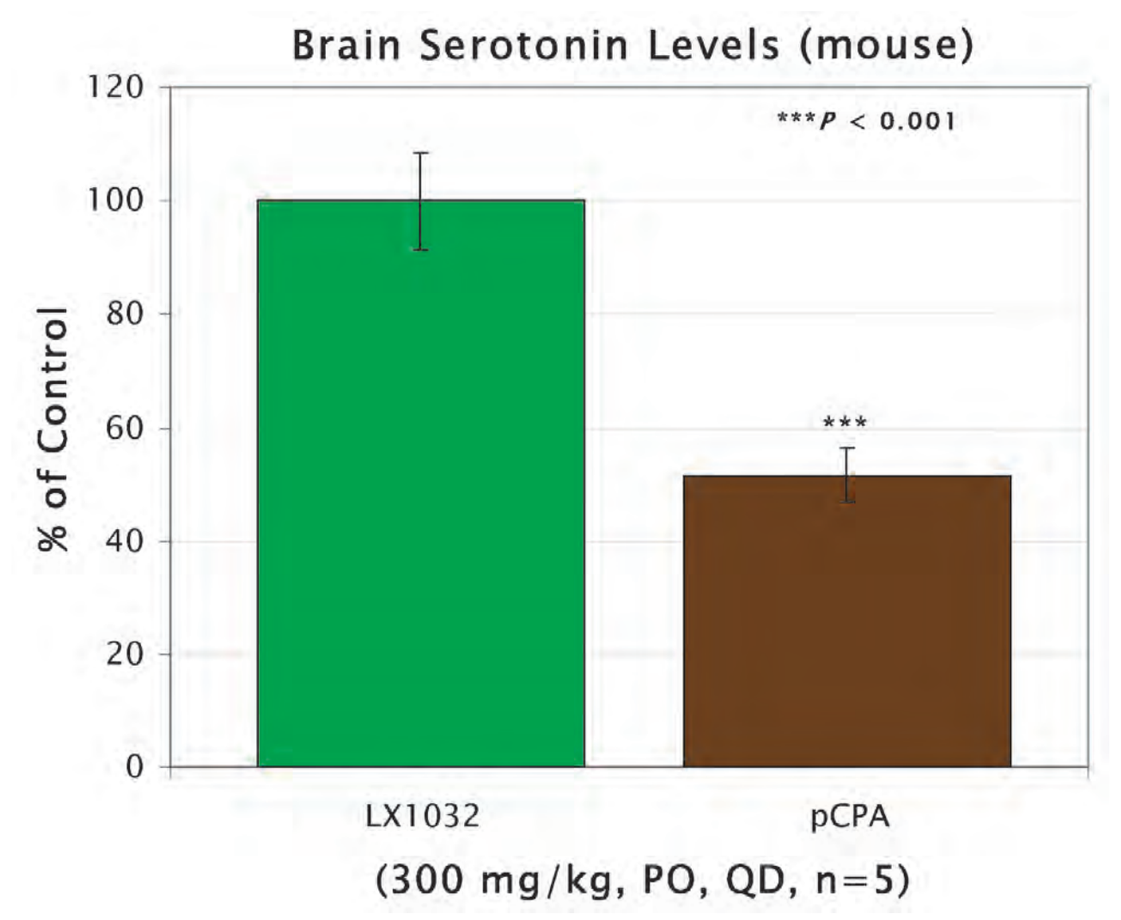
Brain Serotonin Levels (mouse)
***P < 0.001
% of Control

LX1032
pCPA
(300 mg/kg, PO, QD, n=5)

Figure 4. LX1032 is a potent inhibitor of serotonin production in a carcinoid tumor cell line.

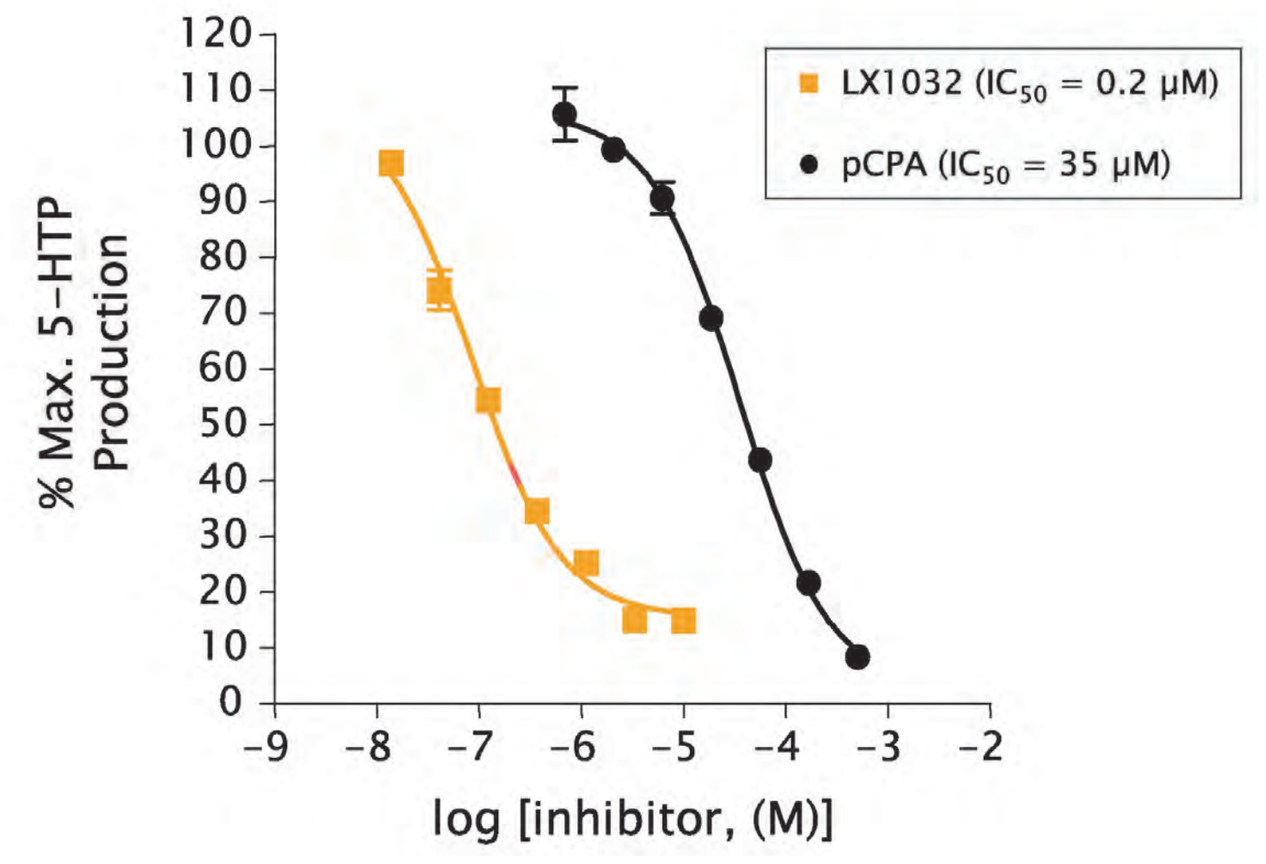
LX1032 (IC50 = 0.2 μM)
pCPA (IC50 = 35 μM)
% Max. 5-HTP Production
log [inhibitor, (M)]

METHODS

- Objective: To evaluate the safety, tolerability, pharmacokinetics, and pharmacodynamics of LX1032 over a range of multiple oral doses in healthy subjects.
- Following the completion of a single ascending dose tolerance trial in healthy volunteers, where single doses up to 1500 mg were tolerated, a separate standard Phase 1b (serial ascending, multiple-dose) double-blind, randomized, placebo-controlled, clinical trial in healthy subjects was conducted.
- Study design for the multiple ascending dose (MAD) study:
 - 8 subjects per dose, randomized 3:1 (active: placebo)
 - Dosing occurred daily for 14 days with regular assessment for safety and tolerability.
 - Diet was controlled in order to avoid foods high in 5-HT.
 - Blood 5-HT and 5-HIAA (a major metabolite of serotonin, commonly used as a urinary bio-marker in the assessment of patients with carcinoid tumor) were measured over the 14-day exposure period in this study to determine the level of serotonin production in the GI tract.
 - Subjects remained sequestered over the entire treatment period and discharged from the unit 48 hours following the completion of a 14-day dosing regimen.
 - The following dose levels were evaluated: 100 mg QD, 250 mg QD, 500 mg QD, 500 mg BID, and 500 mg TID vs. placebo.

RESULTS

MAD Study Summary:

- A Phase 1 multiple ascending dose study was conducted to evaluate the safety and tolerability of LX1032 in healthy volunteers.
- Escalating doses of LX1032 were well tolerated with no serious adverse events or dose-limiting toxicities observed up to 1500 mg (administered as 500 mg TID).
- Significant reductions in 24-hour urinary 5-HIAA (Figure 5), as well as whole-blood 5-HT (Figure 6), in the mid to high dose levels indicate inhibition of the target enzyme in the GI tract. These changes in urinary 5-HIAA and blood 5-HT provide initial confirmation of human pharmacodynamics.
- Rapid absorption was observed as evidenced by the detectable levels of the active moiety, which was readily and almost completely converted from its prodrug, LX1032.
- Plasma concentrations of the active moiety were detectable within 15 minutes of administration of the highest doses (500 mg QD, BID, and TID) and within 30 minutes of administration of the lower doses (100 and 250 mg QD).
- There was no accumulation of the active moiety after multiple doses of LX1032 from 100 mg QD through 500 mg TID.

Adverse Events (AEs):

- Multiple doses up to 500 mg TID were well tolerated over 14 days.
- All AEs were of mild or moderate intensity.
- The most commonly occurring AEs were: nausea (17%), headache (15%), constipation (10%), diarrhea (10%), and abdominal pain (10%) (Table 1).
- No clinically significant changes were observed in vital signs, electrocardiogram (ECG), or physical findings. Laboratory data, including chemistry, hematology, and urinalysis, were evaluated throughout the study The only finding of note was a dose-dependent mild increase (generally ≤ 2 X ULN) in hepatic transaminase levels.
- No significant nervous system disorders or psychiatric disorders were observed.

Figure 5. Urinary 5-HIAA Levels Over 14 Days

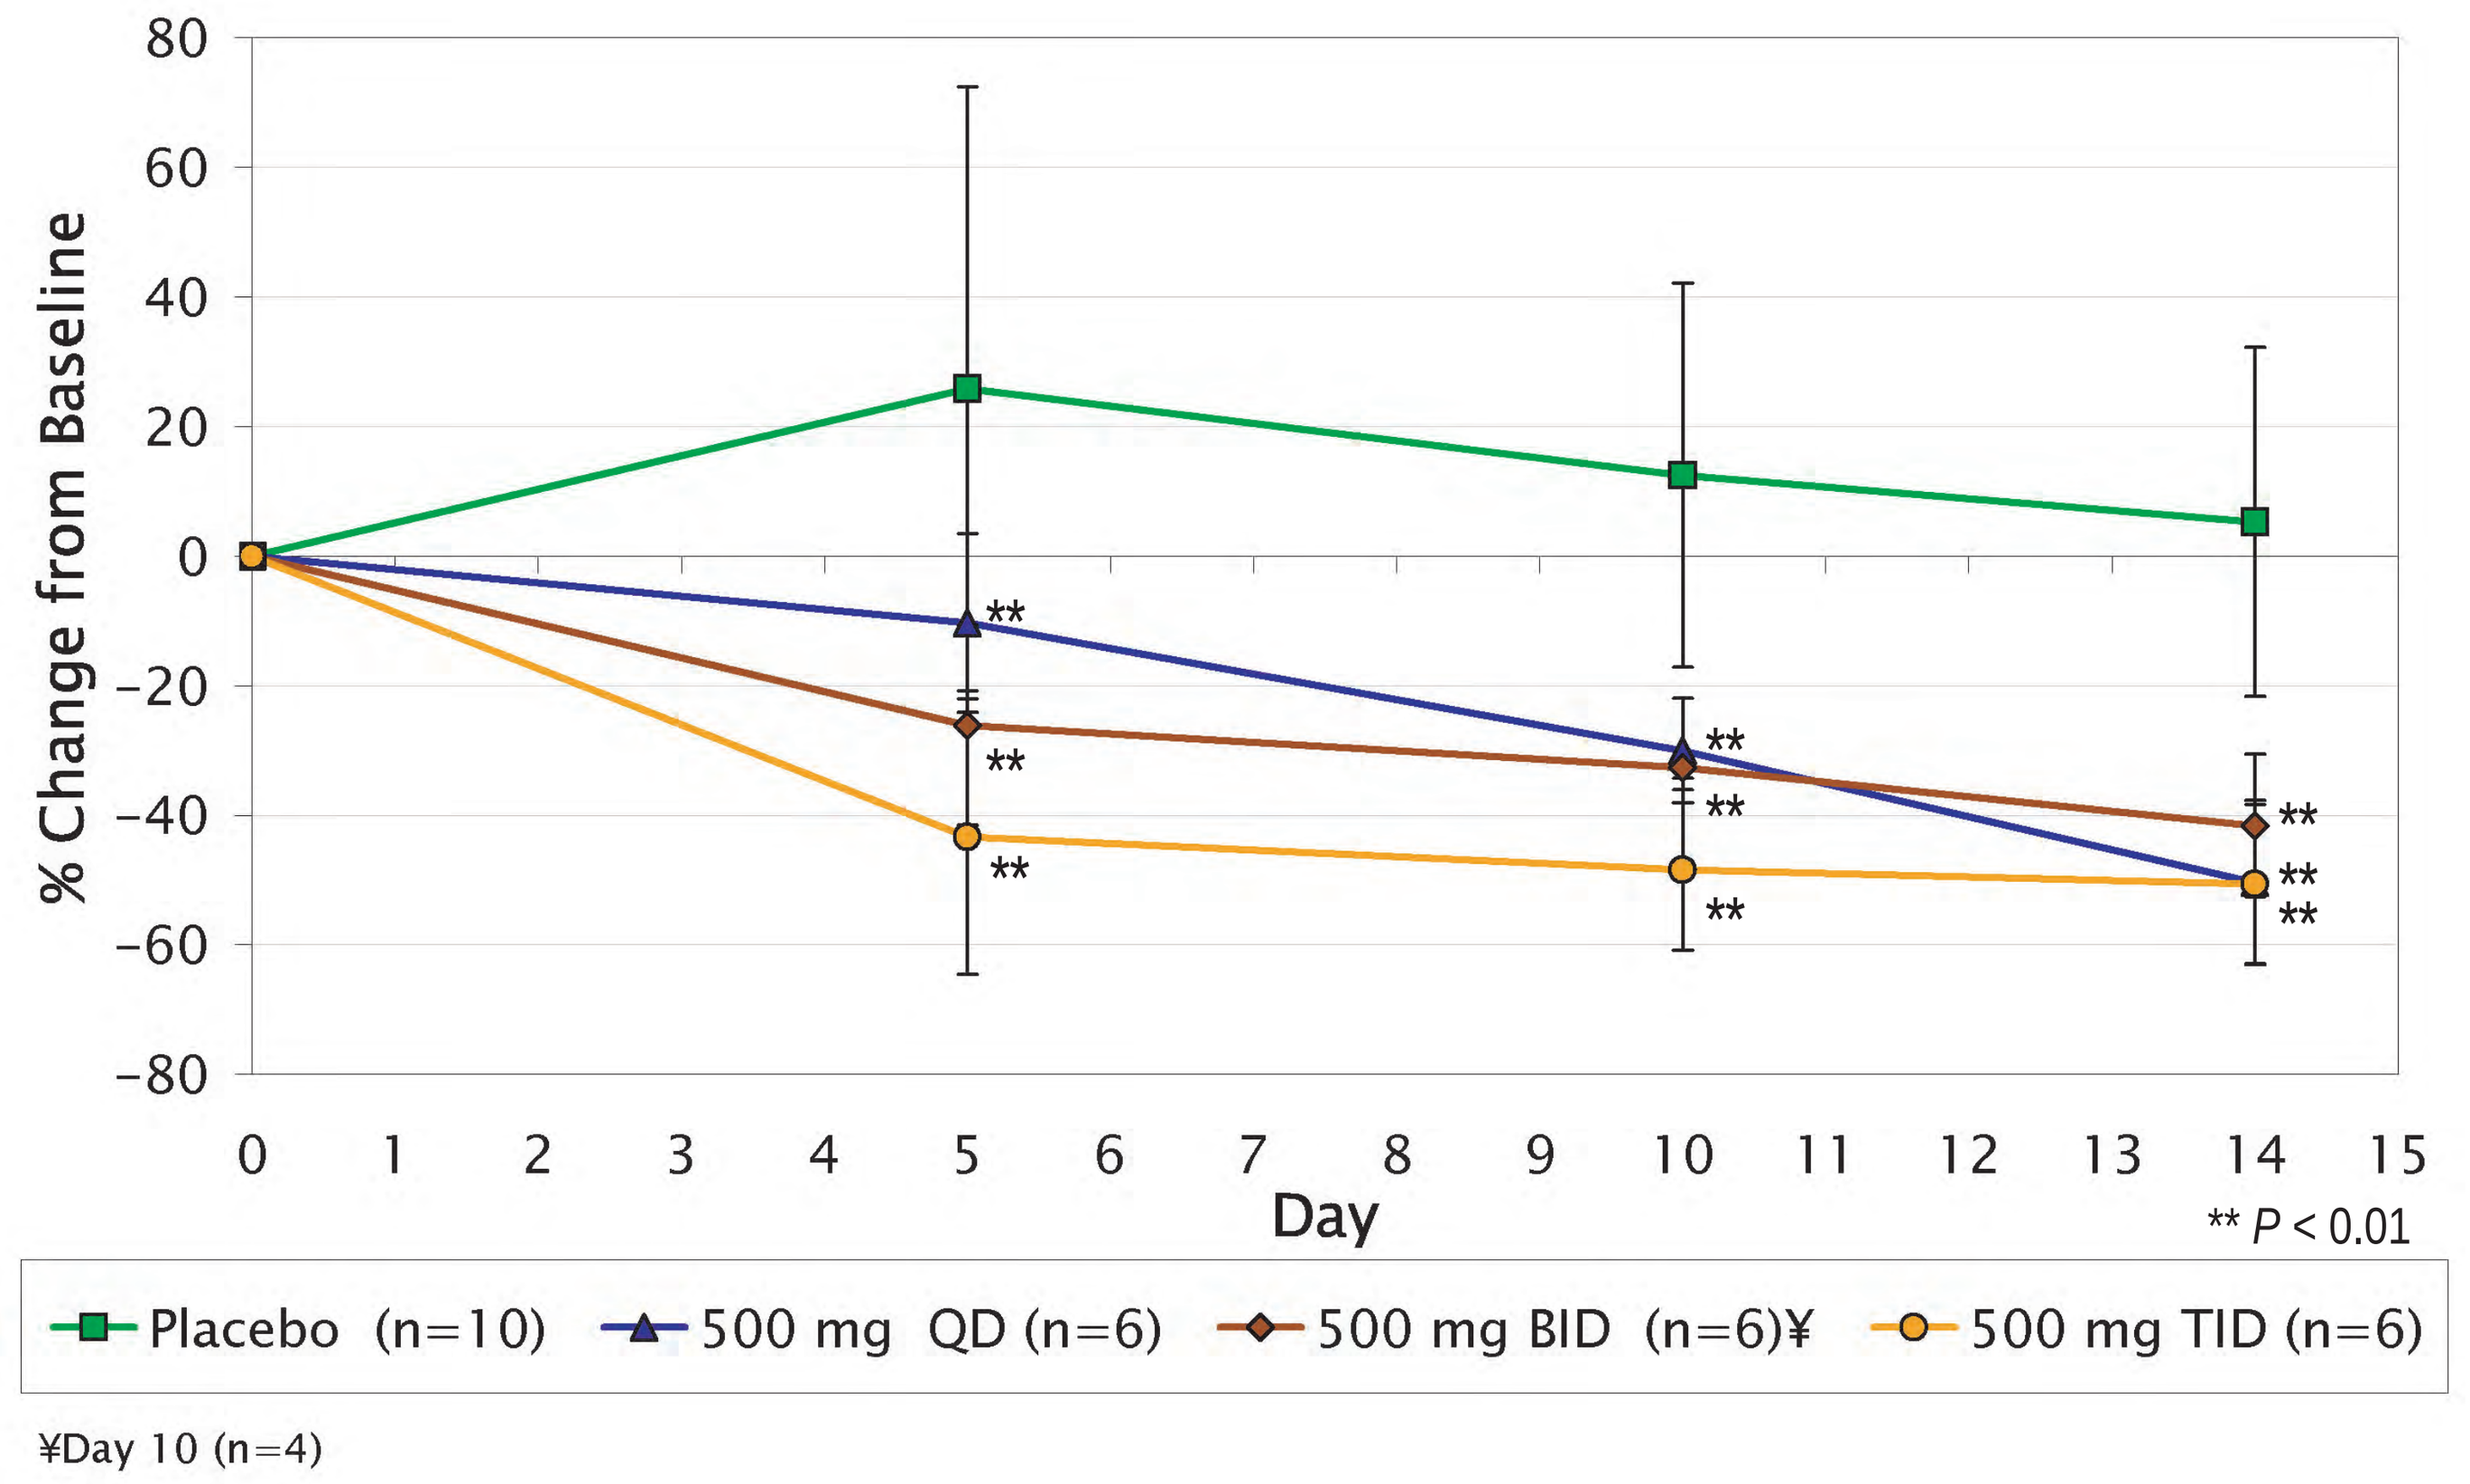
% Change from Baseline
Day
** P < 0.01
Placebo (n=10)
500 mg QD (n=6)
500 mg BID (n=6)¥
500 mg TID (n=6)
¥Day 10 (n=4)

RESULTS (continued)

Table 1. LX1032 MAD Study AE Summary

System Organ Class	Pooled Placebo n=10	LX1032 n=30
Eye Disorders	0	1
Gastrointestinal Disorders	4	9
General Disorders and Administration Site Conditions	0	3
Infections and Infestations	0	3
Investigations (lab value or physical finding)	0	2
Nervous System Disorders	2	6
Respiratory, Thoracic and Mediastinal Disorders	0	2
Total subjects with an AE	6 (60%)	26 (87%)

Figure 6. Blood 5-HT Levels Over 16 Days

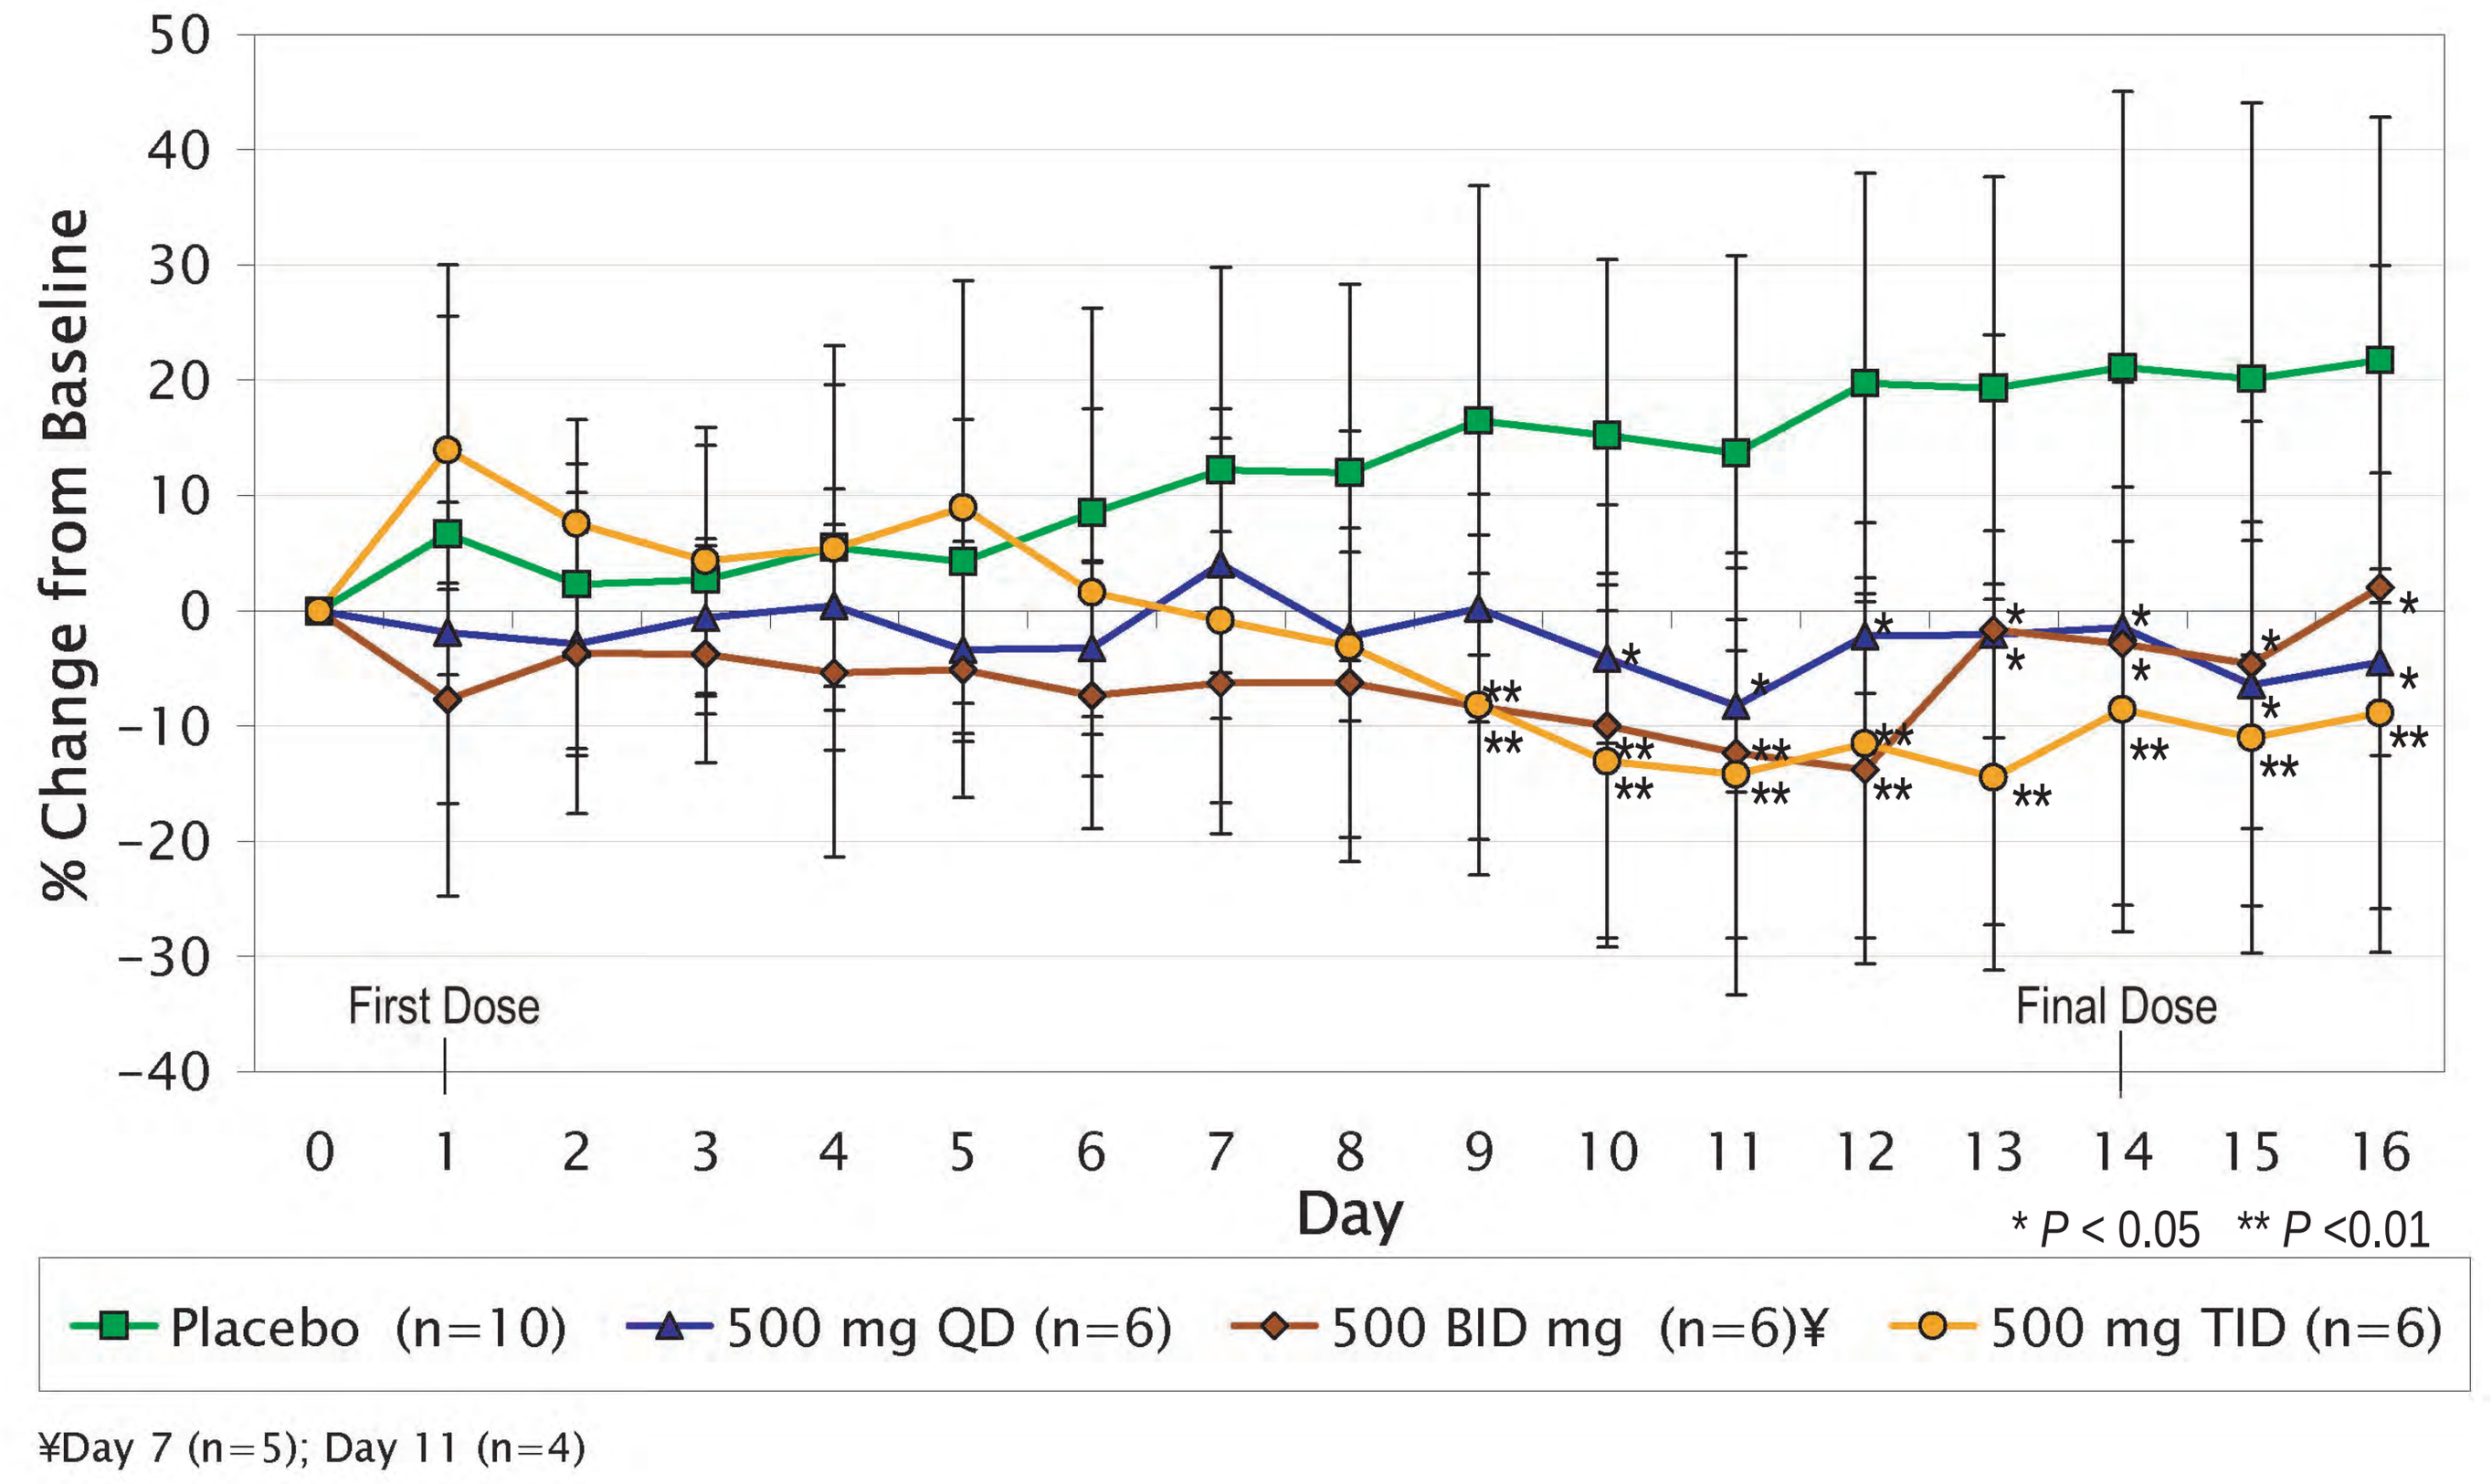
% Change from Baseline
First Dose
Final Dose
Day
* P < 0.05 ** P <0.01
Placebo (n=10)
500 mg QD (n=6)
500 BID mg (n=6)¥
500 mg TID (n=6)
¥Day 7 (n=5); Day 11 (n=4)

CONCLUSIONS

- Modulating peripheral 5-HT production via inhibition of TPH represents a potential new approach to the management of many of the symptoms experienced by patients with carcinoid syndrome.
- LX1032, A novel oral TPH inhibitor, was designed to modulate peripheral serotonin levels without impacting brain serotonin levels.
- Dose levels up to 500 mg TID over 14 days were well tolerated in normal healthy volunteers.
- A statistically significant reduction in 24-hour urinary 5-HIAA output and whole-blood 5-HT levels provides initial confirmation of human pharmacodynamics.
- The pharmacokinetic profile for all dose groups was consistent with preclinical observations; absorption and conversion to the active moiety through esterase activity was rapid.
- The reduction in 5-HT production, coupled with the observed safety profile, supports a potential therapeutic rationale for Phase 2 clinical trials in patients with carcinoid syndrome.
- A study is ongoing in the U.S. evaluating LX1032 in patients experiencing symptomatic carcinoid syndrome despite maximal stable-dose somatostatin analog therapy.

DISCLOSURE: The authors of this poster are employees of, and have received stock options from, Lexicon Pharmaceuticals, Inc.

Presented at the 2009 North American NeuroEndocrine Tumor Society (NANETS) Meeting - Charlotte, NC - October 2nd - 3rd