Phase I Trial of Seneca Valley Virus (NTX-010), a Newly Discovered Systemically Deliverable Oncolytic Picornavirus, in Patients with Solid Tumors with Neuroendocrine Features

Charles M. Rudin[1], Anthony H Williams[2], Neil Senzer[3], Joe Stephenson[4], Kevin Burroughs[2], Seshidhar Reddy[2], Paul Hallenbeck[2]

[1]Johns Hopkins University, Baltimore, MD USA, [2]Neotropix, Inc., Malvern, PA USA, [3] Mary Crowley research Center, Dallas, TX, [4] On Behalf of US Oncology, Houston, TX USA

ABSTRACT

Background SVV-001 is a naturally-occurring replication competent picornavirus, rarely hosted by humans, with potent and selective tropism for NE tumors, including small cell cancers and carcinoid. SVV-001(NTX-010) causes rapid cytolysis in vitro and durable responses following single IV dosing in multiple xenograft models.

Methods A phase I study of intravenous SVV-001 was conducted across 5 log-increment dose cohorts from 10^7 vp/kg to 10^{11} vp/kg, in patients with NE cancers. Study endpoints included toxicity assessment, response assessment, evaluation of viral titers and clearance in blood, sputum, nasal swabs, urine, and stool, and neutralizing antibody development.

Results 30 patients were treated (6 small cell, 24 carcinoid-type). All small cell patients were heavily pretreated (> 3rd line) and received 10^7 vp/kg. In these patients, median PFS was 1.2 months and median OS was 4.1 months with 1 long term (16m+) survivor with prolonged SD despite prior progression.

The 12 carcinoid patients in cohorts 1 to 4 have 70% SD rate and median PFS of 5.4m (95% CI 3.6 to NE); median OS has not been reached. Cohort 5, a 12 patient expansion cohort at 10^{11} vp/kg focused upon carcinoid, shows promising antitumor activity including improvement in carcinoid syndrome symptoms, decline in 5HIAA and other serum markers, minor responses by CT scan, and an objective PET response (>50% decrease in SUV).

There were no DLTs in any cohort. Evidence of intratumoral viral replication was demonstrated with detection of delayed viral titers estimated at >1,000× the administered dose. Viral clearance was documented in all subjects and correlated temporally with development of antiviral antibodies.

Conclusions A single IV dose of 10^{11} vp/kg of SVV-001 is safe with predictable viral kinetics and shows promising activity against NE tumors. Phase II testing of this novel anticancer agent is warranted

BACKGROUND

SVV-001 Is a naturally occurring wild type picornavirus which is highly selective to human neuroendocrine tumor cells

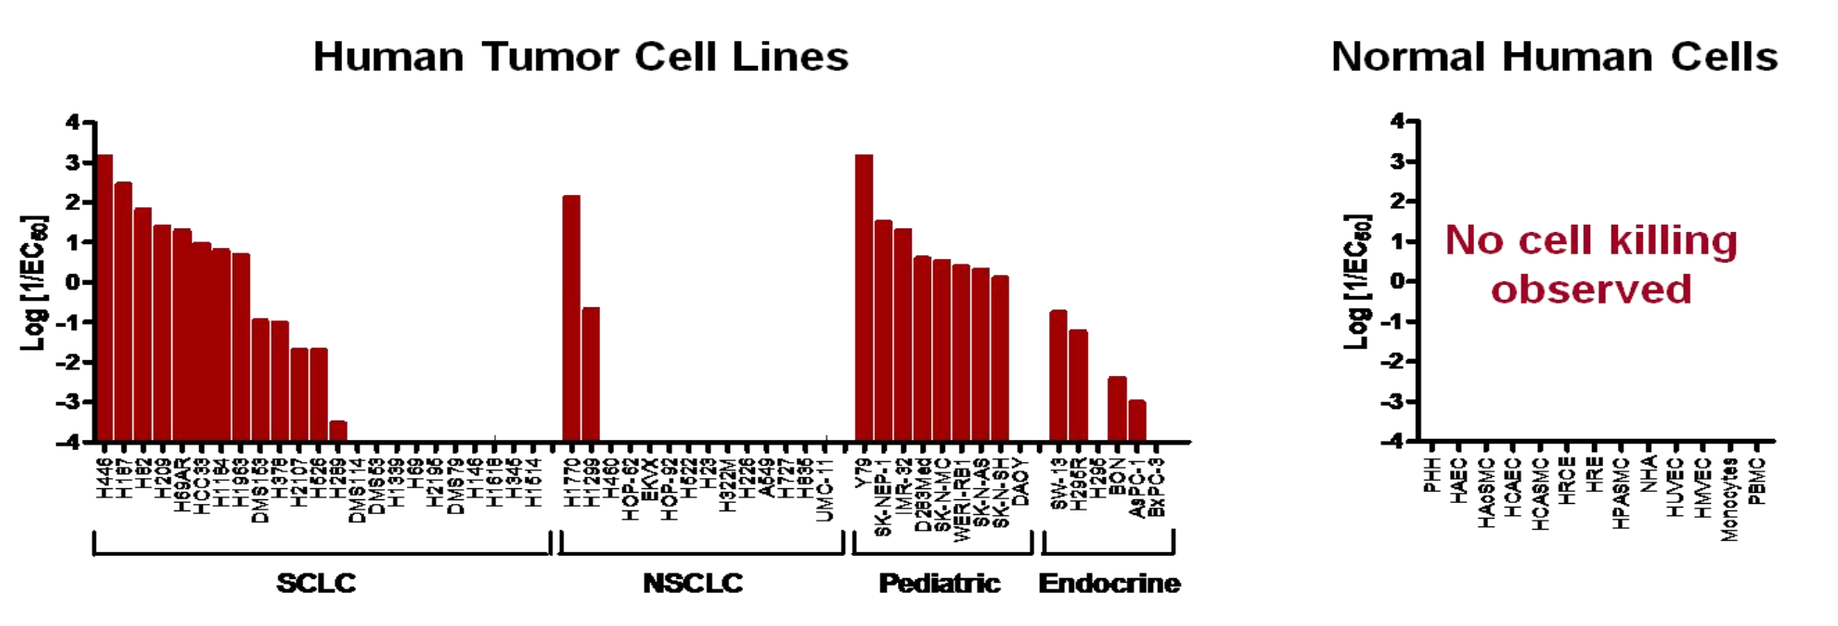

Human Tumor Cell Lines
Normal Human Cells
Log [1/EC50]
No cell killing observed
SCLC
NSCLC
Pediatric
Endocrine

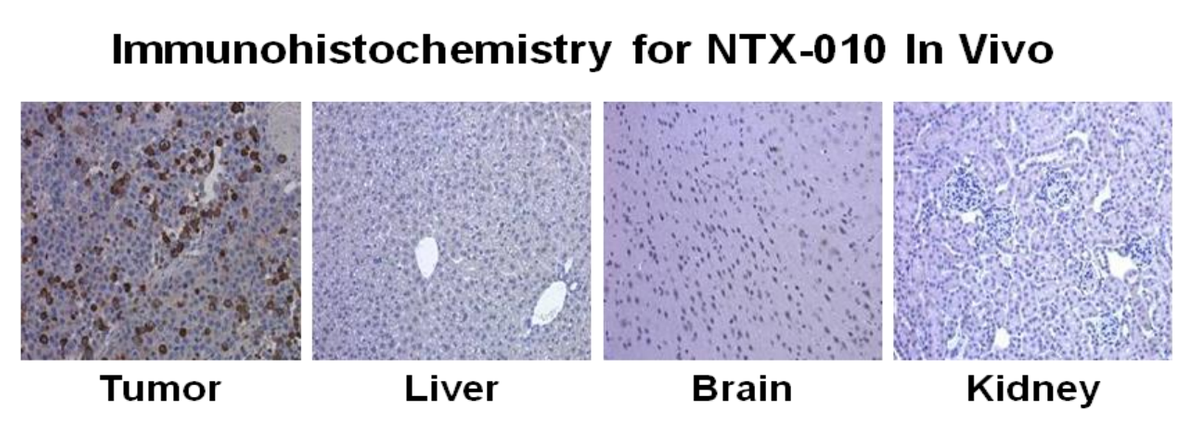

Immunohistochemistry for NTX-010 In Vivo
Tumor
Liver
Brain
Kidney

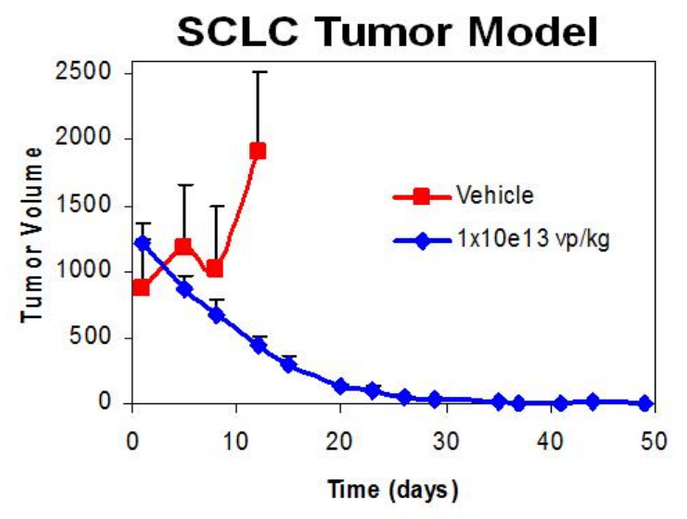

SCLC Tumor Model
Tumor Volume
Vehicle
1x10e13 vp/kg
Time (days)

Possible mechanism of action:
NTX-010 Binds to Integrin Alpha4 (ITGA4) Expressing CHOK1 and H446

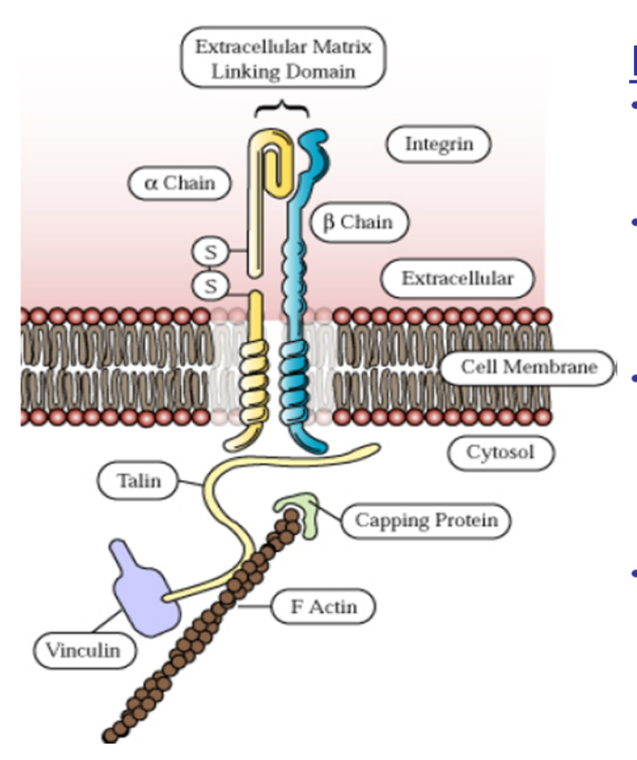

Extracellular Matrix Linking Domain
Integrin
α Chain
β Chain
Extracellular
Cell Membrane
Cytosol
Talin
Capping Protein
F Actin
Vinculin

Integrins
- Integrins link the extracellular space with the inside of the cell
- Integrins serve as cellular receptors for other picornaviruses
- An amino acid motif (LDV) known to bind a4b1 integrin is found in a surface-exposed loop of VP2 - a capsid protein
- Virus mutated in this site could not be rescued from culture

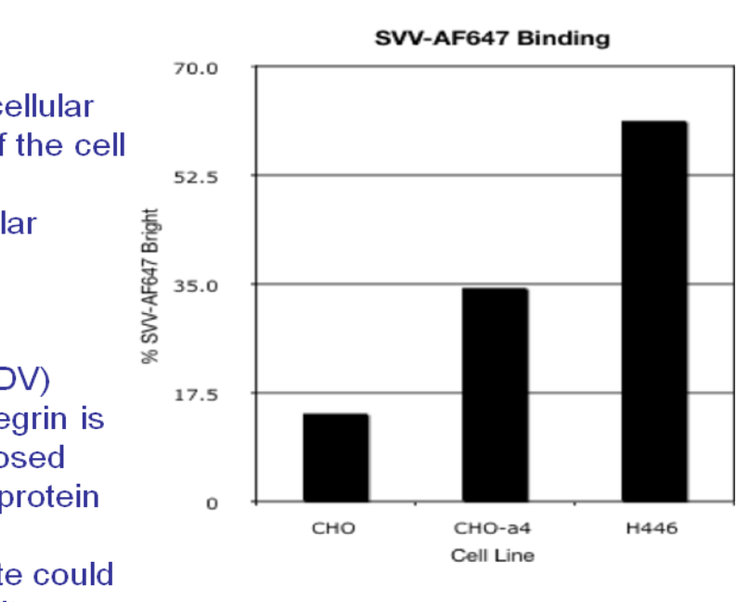

SVV-AF647 Binding
% SVV-AF647 Bright
CHO
CHO-a4
H446
Cell Line

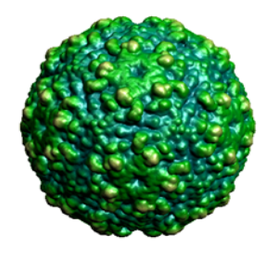

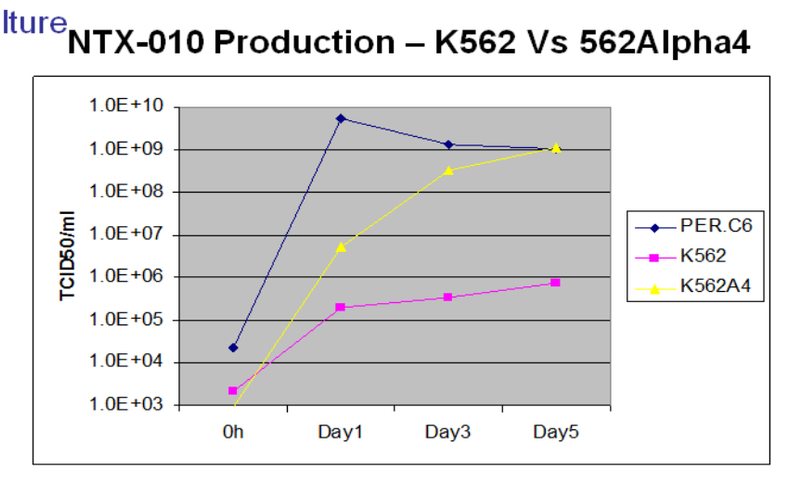

NTX-010 Production – K562 Vs 562Alpha4
TCID50/ml
PER.C6
K562
K562A4
0h
Day1
Day3
Day5

Objectives

Primary

To evaluate safety and tolerability and to define the recommended phase II dose of NTX-010 in patients with advanced solid tumors with neuroendocrine features that were progressing

Secondary

To obtain preliminary information regarding the anti-tumor activity of NTX-010 in this patient population.

To characterize changes in viral titers, and to characterize viral distribution and elimination in blood, urine, stool, sputum, and nasal swab following IV administration of NTX-010.

To evaluate development of neutralizing antibodies to NTX-010 following IV administration of NTX-010.

.

METHODS

Eligibility

Two primary cohorts of patients have been studied to date: a dose-escalation cohort in patients with any NE cancer with estimated survival ≥ 6 months, and an expansion cohort of advanced SCLC treated at 10^7 vp/kg.

ASSESSMENTS

- •Chromogranin A
- •Viral replication assessment in serum, stool, urine and sputum
- •Regular repeat imaging (CT/MRI/PET) on a 6-8 week basis
- •Pre and post therapy assay for neutralizing antibodies
- •Overall Survival and Progression Free Survival
- •Safety

RESULTS

Viral Dynamics

NTX-010 RNA in Serum by qRTPCR

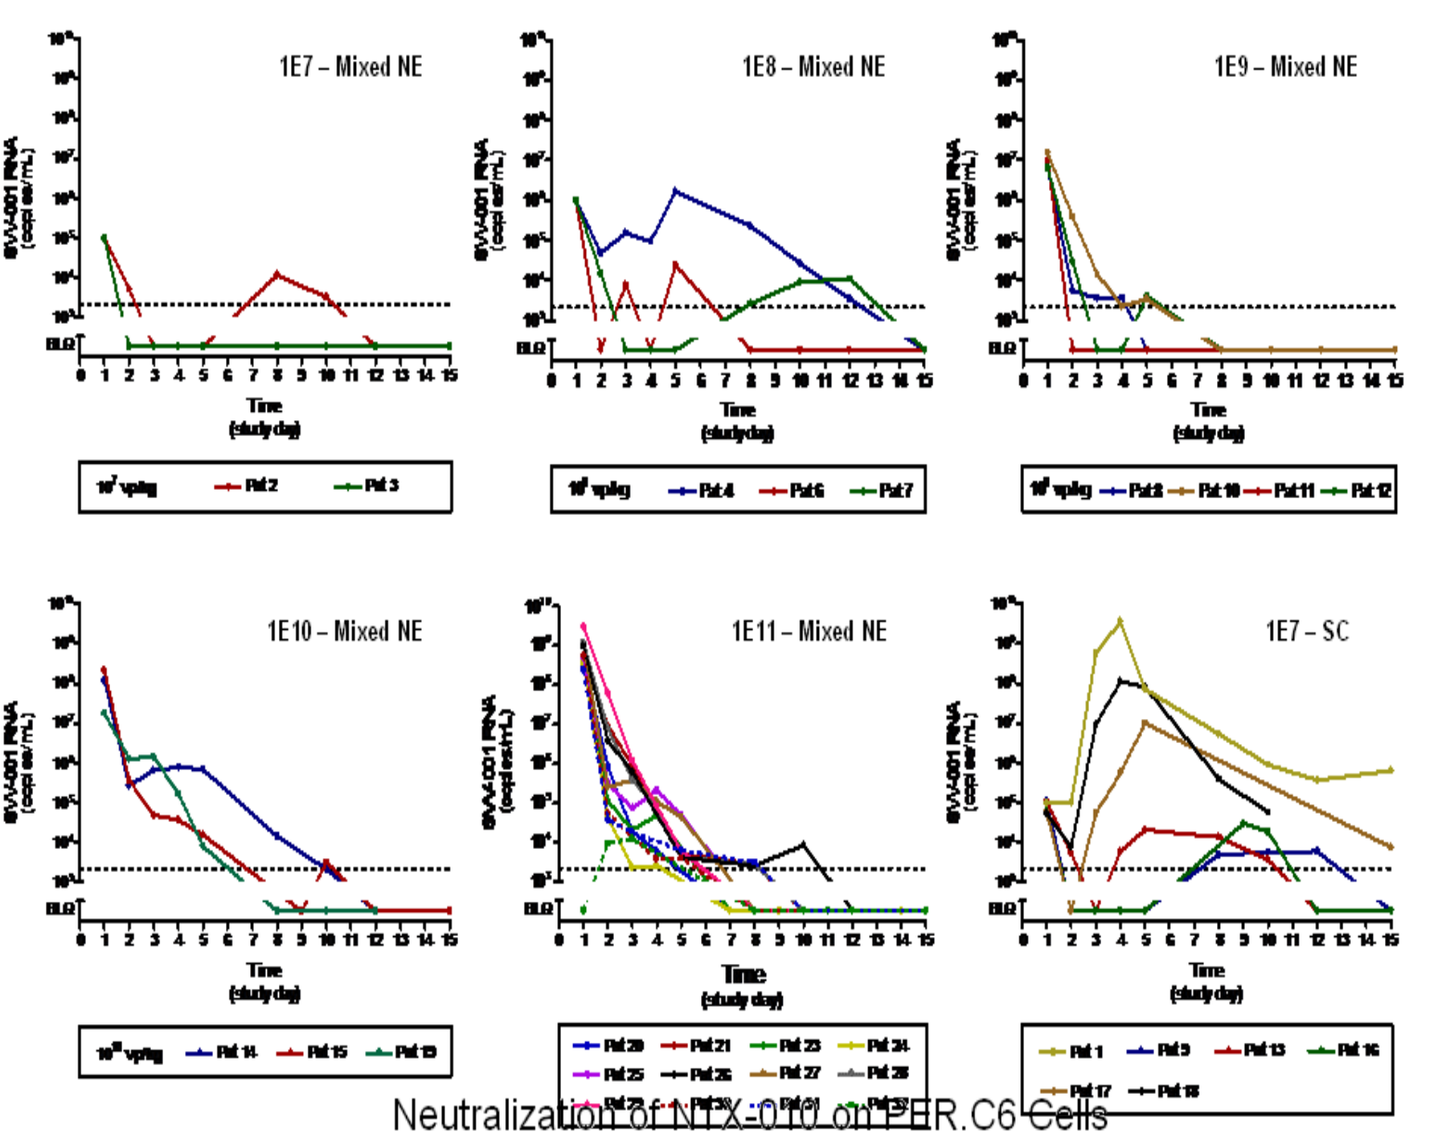

1E7 – Mixed NE
1E8 – Mixed NE
1E9 – Mixed NE
1E10 – Mixed NE
1E11 – Mixed NE
1E7 – SC
Time (study day)

Neutralization of NTX-010 on PER.C6 Cells

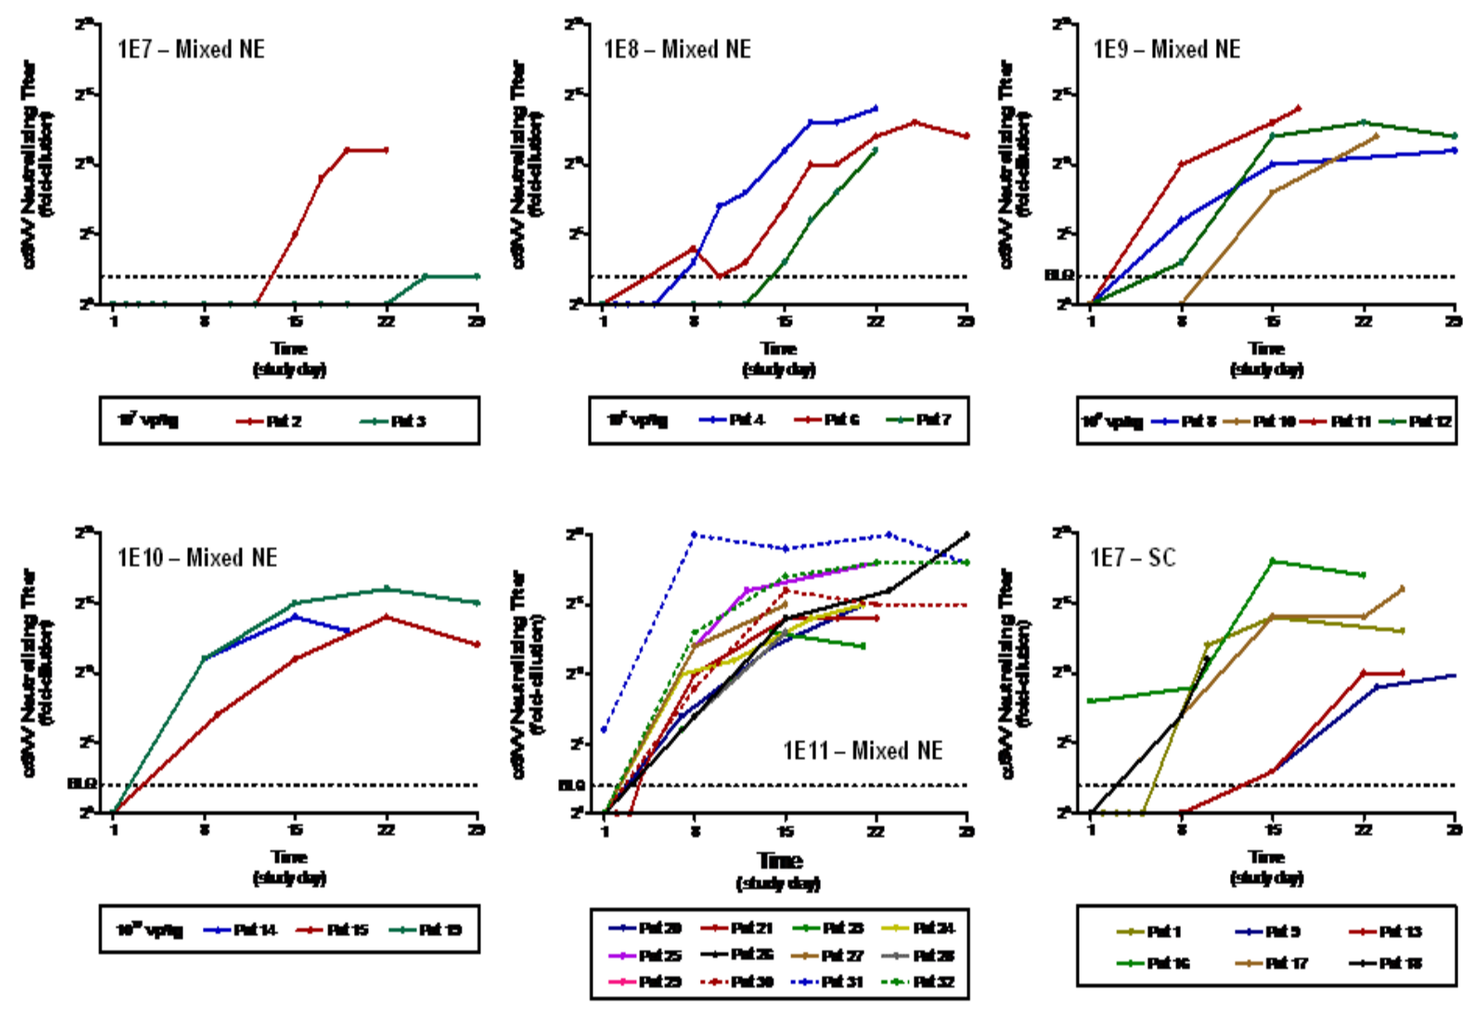

1E7 – Mixed NE
1E8 – Mixed NE
1E9 – Mixed NE
1E10 – Mixed NE
1E11 – Mixed NE
1E7 – SC
Time (study day)

Small Cell Cohort: safety and efficacy

- •Small cell patients in this study had progressed after multiple lines of therapy, typically 3 – 5 lines
- •All small cell patients were dosed at the lowest dose level (10^7 VP/KG)
- •One of the 6 patients in this cohort has experienced long term survival of more than 18 months, and has returned to work.
- •Limited historical data is available on expected PFS and OS beyond third line therapy for small cell.

Overall Survival KM (as of 1/4/09)

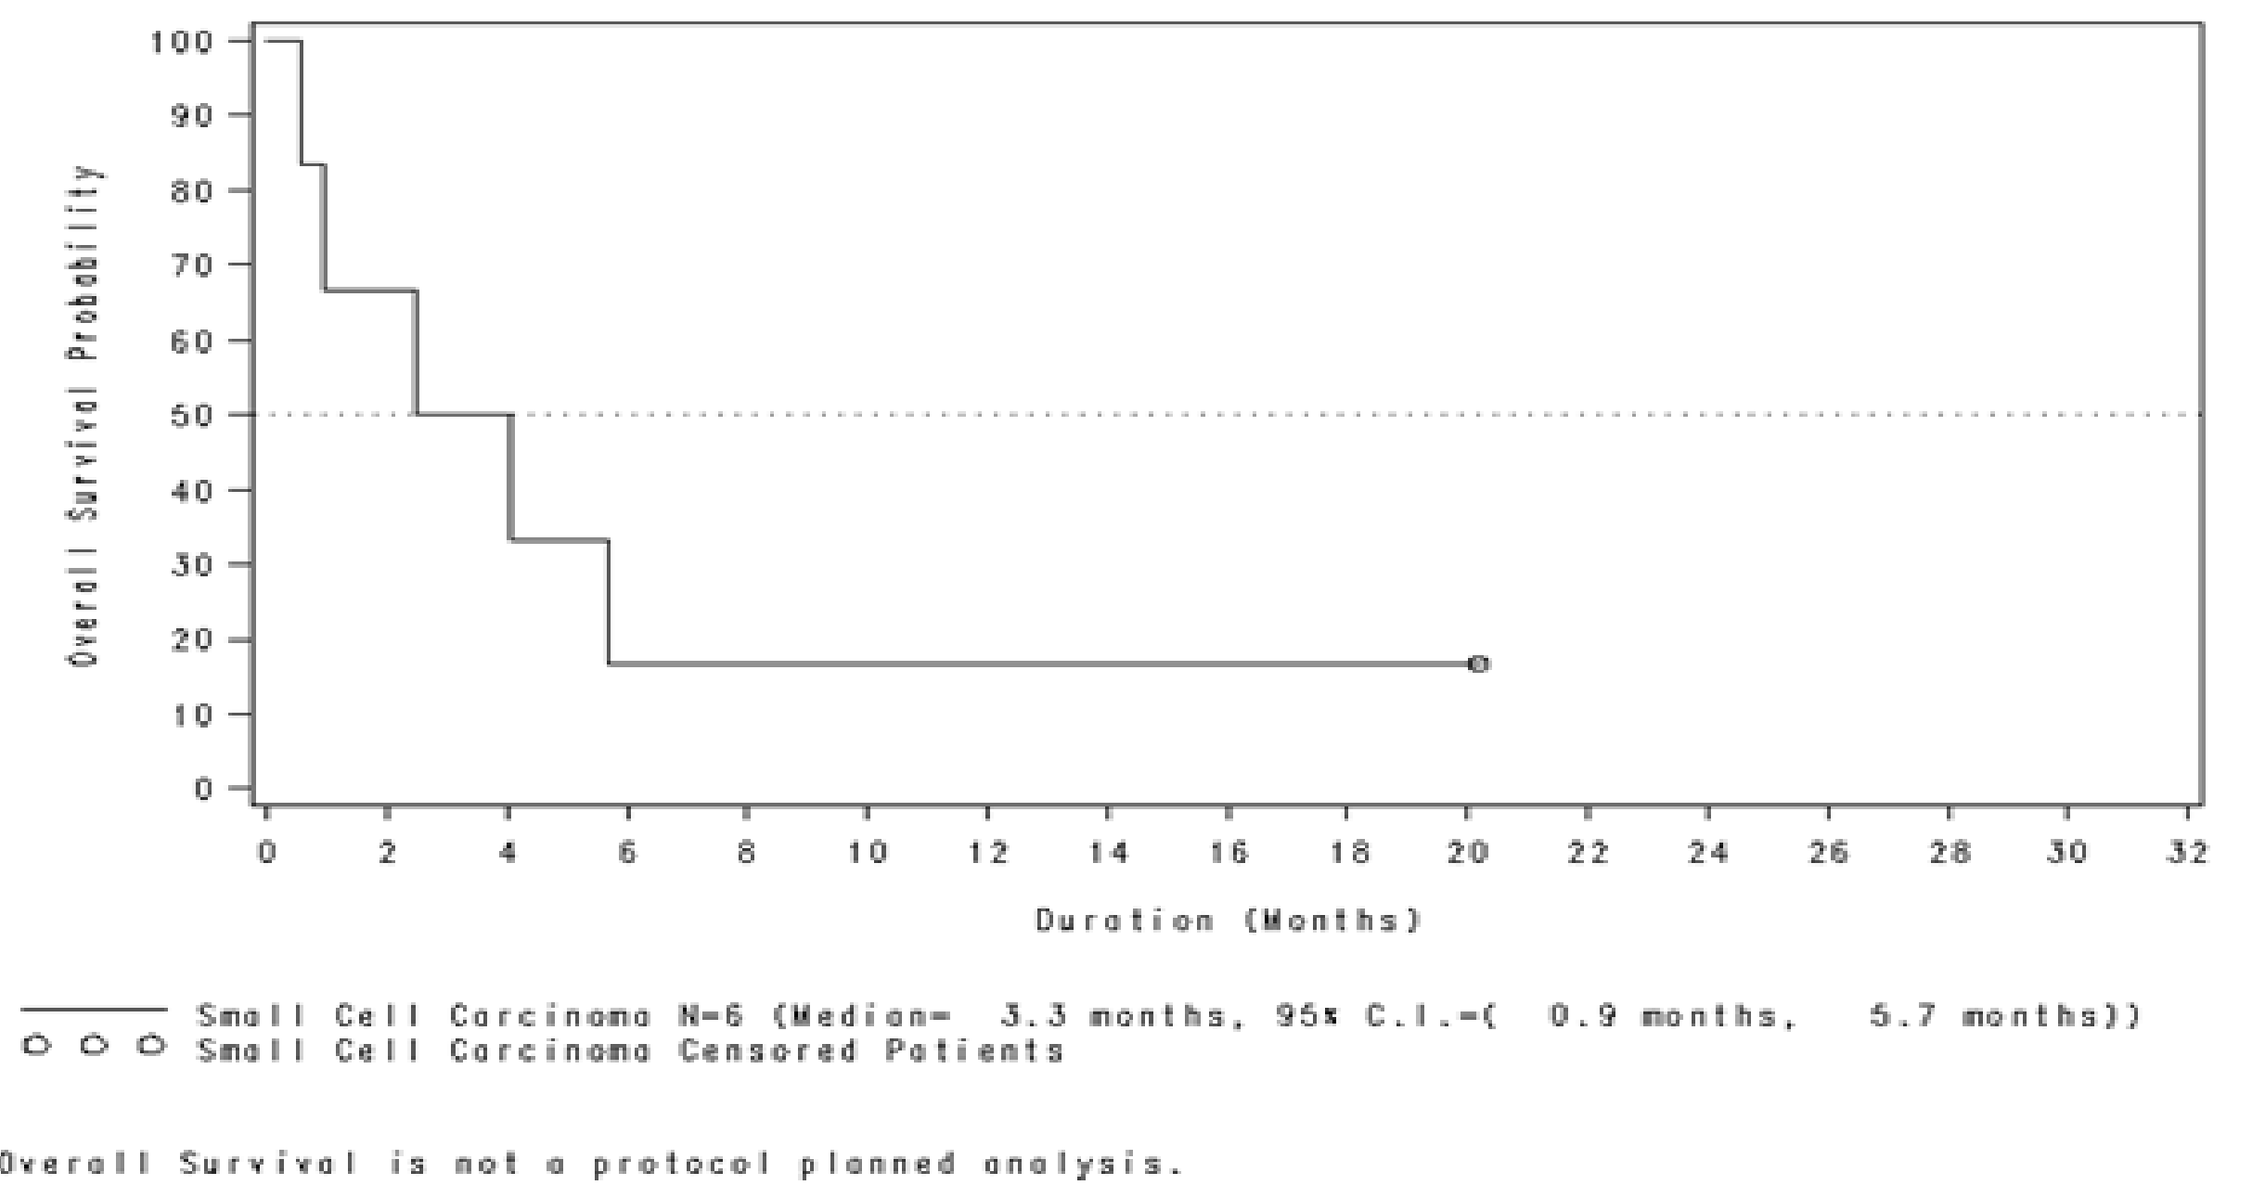

Overall Survival Probability
Duration (Months)
Small Cell Carcinoma N=6 (Median= 3.3 months, 95% C.I.=(0.9 months, 5.7 months))
Small Cell Carcinoma Censored Patients
Overall Survival is not a protocol planned analysis.

RESULTS (Continued)

Summary of Adverse Events by Dose Level, System Organ Class
Patient with Small Cell Carcinoma (1)
Safety Evaluable (N=6)

	Initial Dose Level [1]
System Organ Class [2]	10^7
Number of Patients	6
Patients with Adverse Events	6 (100.0%)
General disorders and administration site conditions	6 (100.0%)
Gastrointestinal disorders	5 (83.3%)
Respiratory, thoracic and mediastinal disorders	5 (83.3%)
Investigations	4 (66.7%)
Metabolism and nutrition disorders	3 (50.0%)
Musculoskeletal and connective tissue disorders	3 (50.0%)
Infections and infestations	3 (50.0%)
Nervous system disorders	2 (40.0%)
Psychiatric disorders	2 (40.0%)
Renal and urinary disorders	3 (50.0%)
Skin and subcutaneous tissue disorders	2 (33.3%)
Blood and lymphatic system disorders	2 (33.3%)
Ear and labyrinth disorders	1 (16.7%)
Reproductive system and breast disorders	1 (16.7%)

[1] Initial Dose Level: SVV-001 10 [7] (vp/kg).
[2] Number of Patients used for denominator in calculation of percentages.

Carcinoid type tumors: safety and efficacy

Low Doses 10^7-10^{10} Response data

1 week prior to NTX-010

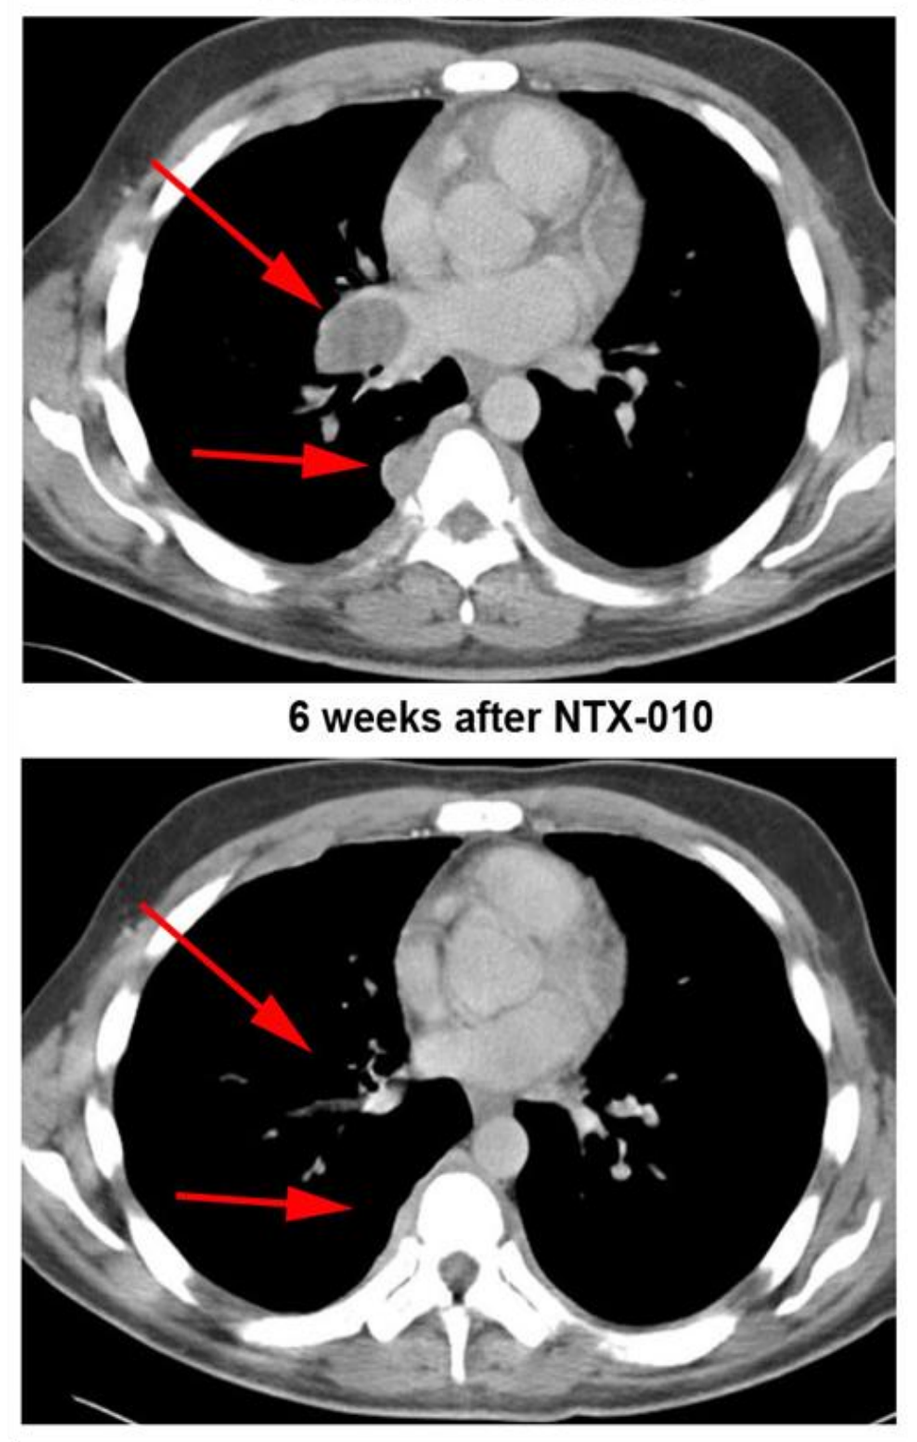

6 weeks after NTX-010

Total evaluable patients 10	
Minor responses by CT	3
SD w/decrease in Chromogranin A	4
No response	2
Death by progressive disease	1

Several patients experienced prolonged stable disease. Additionally, several patients had reduction in reported pain or other symptomatic improvement.

Trend towards better efficacy at 10^{11}

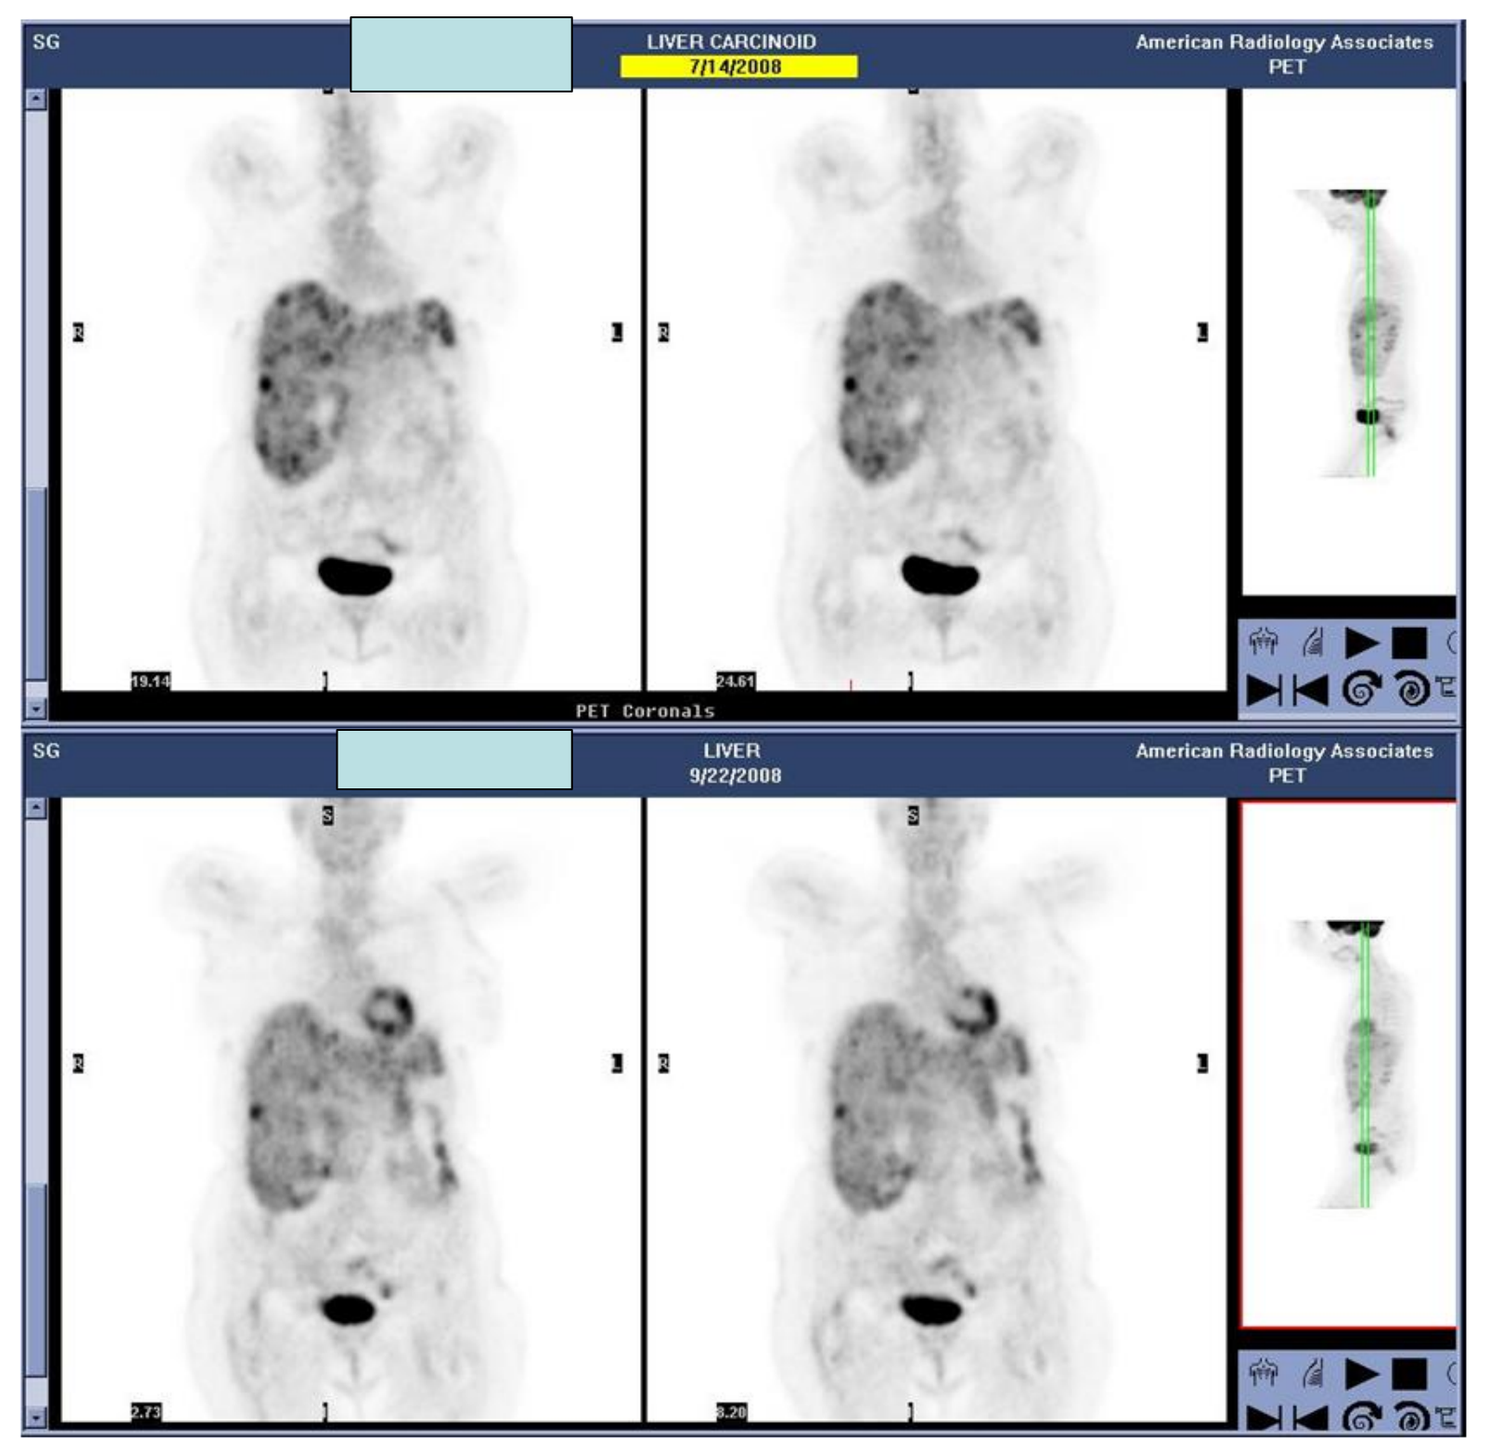

LIVER CARCINOID
7/14/2008
American Radiology Associates
PET
PET Coronals
LIVER
9/22/2008

Patient responses at dose 10^{11}
Total patients treated 12

PR's, or MR (PET or CT)	2
Decrease in Chromogranin A	2
Progression	1
Death by progressive disease	1
Stable disease	6

Clinical Benefit Rate (PR + SD) =10/12

Carcinoid Progression Free Survival (April 2009)

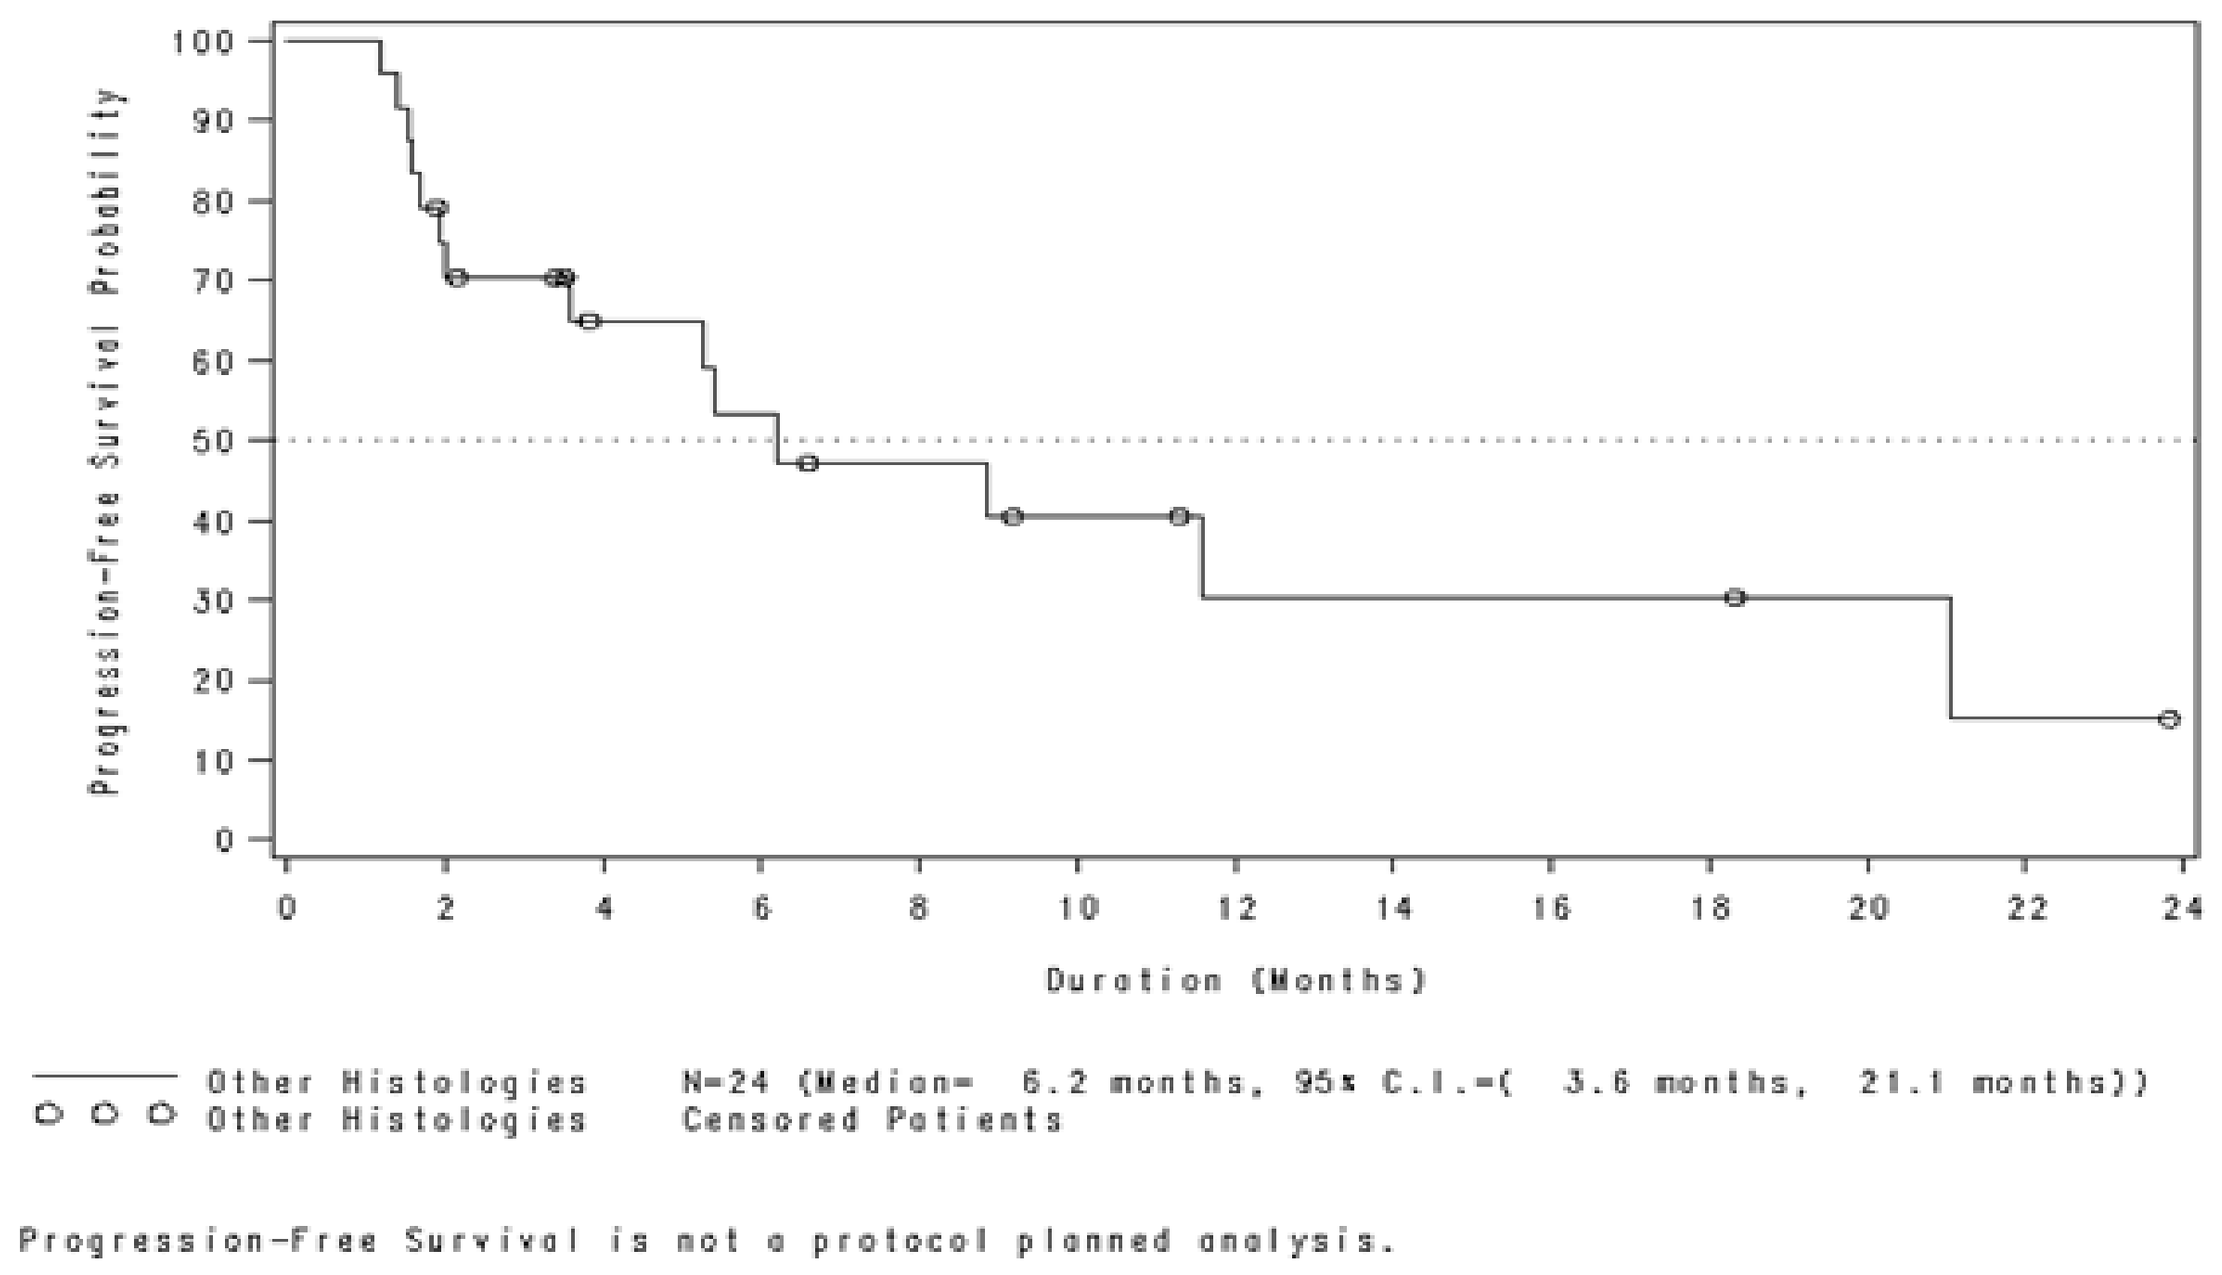

Progression-Free Survival Probability
Duration (Months)
Other Histologies N=24 (Median= 6.2 months, 95% C.I.=(3.6 months, 21.1 months))
Other Histologies Censored Patients
Progression-Free Survival is not a protocol planned analysis.

Carcinoid Overall Survival (April 2009)

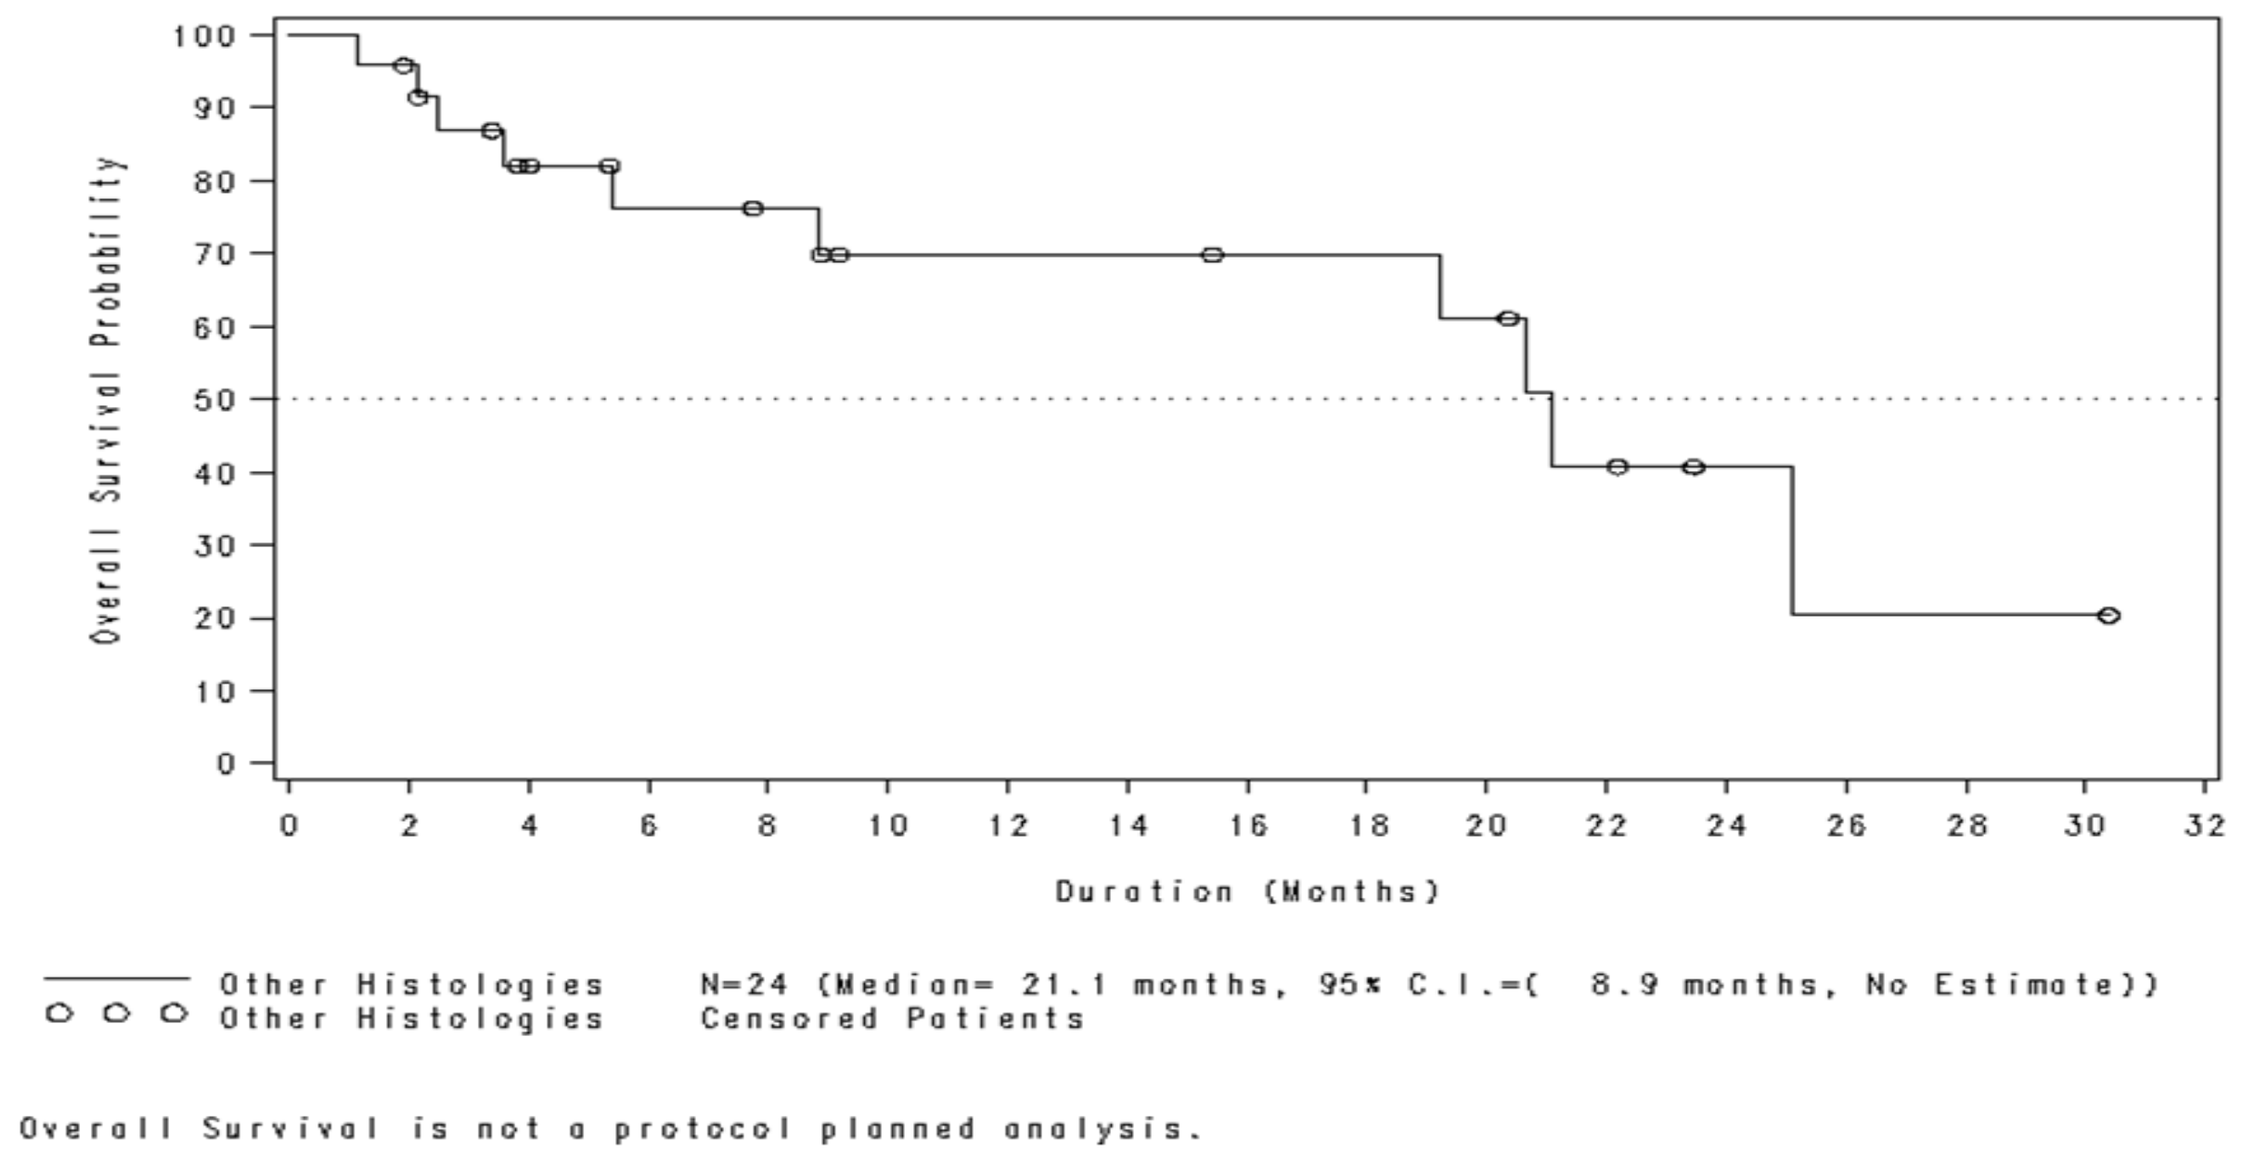

Overall Survival Probability
Duration (Months)
Other Histologies N=24 (Median= 21.1 months, 95% C.I.=(8.9 months, No Estimate))
Other Histologies Censored Patients
Overall Survival is not a protocol planned analysis.

Summary of Adverse Events by Dose Level, System Organ Class and Preferred Term
Patient without Small Cell Carcinoma (1)
Safety Evaluable (N=24)

Initial Dose Level [1]	10^7	10^8	10^9	10^10	10^11	Overall
System Organ Class [2]						
Number of Patients	2	3	4	3	12	24
Patients with Adverse Events	2 (100.0%)	3 (100.0%)	3 (75.0%)	3 (100.0%)	12 (100.0%)	24 (96.0%)
General disorders and administration site conditions	2 (100.0%)	3 (100.0%)	2 (50.0%)	3 (100.0%)	8 (66.7%)	19 (76.0%)
Gastrointestinal disorders	1 (50%)	2 (66.7%)	3 (75.0%)	2 (66.7%)	8 (66.7%)	17 (68.0%)
Musculoskeletal and connective tissue disorders	1 (50%)	1 (33.3%)	1 (25.0%)	2 (66.7%)	6 (50.0%)	11 (44.0%)
Respiratory, thoracic and mediastinal disorders	0	1 (33.3%)	0	3 (100.0%)	5 (41.7%)	10 (40.0%)
Metabolism and nutrition disorders	0	2 (66.7%)	2 (50.0%)	3 (100.0%)	2 (16.7%)	9 (36.0%)
Nervous system disorders	1 (50%)	2 (66.7%)	0	1 (33.3%)	5 (41.7%)	9 (36.0%)
Blood and lymphatic system disorders	0	0	0	1 (33.3%)	4 (33.3%)	6 (24.0%)
Vascular disorders	1 (50%)	0	0	1 (33.3%)	4 (33.3%)	6 (24.0%)
Psychiatric disorders	1 (50%)	1 (33.3%)	0	0	2 (16.7%)	4 (16.0%)
Skin and subcutaneous tissue disorders	0	1 (33.3%)	0	1 (33.3%)	2 (16.7%)	4 (16.0%)
Investigations	0	0	0	1 (33.3%)	1 (8.3%)	(12.0%)
Infections and infestations	0	0	0	0	1 (8.3%)	2 (8.0%)
Renal and urinary disorders	0	0	0	0	1 (8.3%)	2 (8.0%)
Cardiac disorders	0	0	0	0	1 (8.3%)	1 (4.0%)
Eye disorders	0	0	0	1 (33.3%)	0	1 (4.0%)
Injury, poisoning and procedural complications	0	0	0	0	1 (8.3%)	1 (4.0%)

[1] Initial Dose Level: SVV-001 10^7 (vp/kg).
[2] Number of Patients used for denominator in calculation of percentages.

CONCLUSIONS

- ➢ No DLT's at any dose
- ➢ Primary AE: Mild flu like symptoms for 1-2 days
 - ➢ No apparent correlation between viral replication and adverse events
- ➢ Clinical effects apparent from safety study
 - Small Cell Lung Cancer:
 - ➢ Long term survival in 1 of 5 subjects
 - Carcinoid Cancer:
 - ➢ PR, MR and clinical status improvements observed in multiple patients at multiple doses
 - ➢ Durable disease stability observed in several patients
 - ➢ Positive survival trend supports further development
- ➢ Phase II study in SCLC planned with a cooperative group during the watchful waiting period. Trial to start in 2009