Presented at the North American NeuroEndocrine Tumor Symposium, October 20–22, 2011, Minneapolis, Minnesota

Evaluation of the Efficacy and Safety of Lanreotide on Tumor Growth Stabilization in Patients with Progressive NETs who are not Eligible for Treatment with Surgery or Chemotherapy

V. Alonso,[1] M. Marmol,[2] D. Castellano,[3] E. Fonseca,[4] A. Velasco,[5] J.L. García López,[6] E. Pineda,[7] P. Maisonobe,[8] B. Massutí,[9] M. Martín-Richard;[10] TTD Study Group

[1]Medical Oncology, Hospital Miguel Servet, Zaragoza, Spain; [2]Medical Oncology, Hospital Clinic, Barcelona, Spain; [3]Medical Oncology, Hospital Universitario 12 de Octubre, Madrid, Spain; [4]Medical Oncology, Hospital Universitario Salamanca, Salamanca, Spain; [5]Medical Oncology, Hospital Universitario de la Princesa, Madrid, Spain; [6]Medical Oncology, Hospital Ramón y Cajal, Madrid, Spain; [7]Medical Department, IPSEN PHARMA, Barcelona, Spain; [8]Global Medical Affairs, IPSEN PHARMA, Boulogne Billancourt, France; [9]Medical Oncology, Hospital General de Alicante, Alicante, Spain; [10]Medical Oncology, Hospital de la Santa Creu y Sant Pau, Barcelona, Spain

Introduction

- Somatostatin analogs (SSAs) are the treatment of choice for hormonal symptoms associated with neuroendocrine tumors (NETs).
- NETs may exhibit prolonged periods of tumor stabilization. If slow growth of a tumor and/or its metastases is interpreted as treatment-induced stability, this may lead to inaccurate conclusions about treatment efficacy.[1]
- Clinical studies have suggested that SSAs may stabilize or, rarely, induce partial response in NET tumor growth.[2,3]
- This phase II clinical trial (NCT0032646) is the first to examine the anti-tumor activity of monthly SSA monotherapy in patients with documented progressive NETs.

Aim

- To evaluate the efficacy of lanreotide 120 mg in stabilizing tumor growth in patients with progressive gastroenteropancreatic (GEP) or bronchopulmonary (BP) NETs not amenable to surgery or chemotherapy.

Materials and methods

- Caucasian patients (n=30) with advanced and/or metastatic, well-differentiated GEP or BP NETs that had progressed within the last 6 months were treated with deep subcutaneous lanreotide (Somatuline Autogel®*) 120 mg every 28 days for 92 weeks, in 17 Spanish hospitals.
- Sample size was based on the assumption that 25% and 12% of patients with progressive NETs receiving SSA treatment are progression-free after the first and second years of treatment, respectively.[4,5]
- Inclusion criteria were:
 - age ≥18 years
 - histopathologic diagnosis of well-differentiated GEP or BP NET or carcinoma, considered measurable according to Response Evaluation Criteria in Solid Tumors (RECIST) criteria
 - tumor progression according to RECIST criteria over the preceding 6 months
 - 0–2 grading on the Eastern Cooperative Oncology Group (ECOG) general status assessment scale
 - positive (^{111}In-DTPA-D-Phe) octreotide scan.
- Exclusion criteria included SSA treatment during the 6 months prior to study inclusion, and chemotherapy or interferon for 4 weeks prior to study inclusion.
- Radiologic evaluation of change in tumor size (computed tomography, magnetic resonance imaging), according to RECIST criteria, was performed every three treatment cycles by an independent radiologist with expertise in NETs.
- The primary endpoint, efficacy of lanreotide in NET growth stabilization per central blind review, was evaluated by measuring time until disease progression according to RECIST criteria or early death, and the primary efficacy analysis was based on data from the intention-to-treat (ITT) population. Progression-free survival (PFS) was determined using Kaplan–Meier analysis.
- Secondary efficacy endpoints included i) prognostic factors for tumor growth (analyzed using a Cox regression model), ii) percentage of patients who showed partial or complete response according to RECIST criteria (Kaplan–Meier analysis), iii) symptom control and iv) biochemical response (chromogranin A [CgA] and urinary 5-HIAA, analyzed using a parametric or non-parametric signed test).
- Symptom severity was determined using a three-point Likert scale (mild to moderate).
- Quality of life was assessed using the validated European Organisation for the Research and Treatment of Cancer (EORTC) QLQ-C30 questionnaire, and was tested using the paired Student's *t*-test.
- The ITT and safety populations included the 30 patients who received study medication. Two patients were excluded from the ITT population for protocol violation but, as they had no assessments, the per-protocol (PP) and ITT results were identical.

Results

- Baseline demographic and clinical characteristics are presented in Table 1.

Table 1. Baseline demographic and clinical characteristics of patients with progressive neuroendocrine tumors enrolled in a study of the effects of lanreotide on tumor progression.

Characteristic	Value
Age (years)	63.0 (40–78)
Male, n (%)	15 (50)
Caucasian ethnicity, n (%)	30 (100)
Body mass index, kg/m²	24.4 (14.0–38.2)
Time since diagnosis, years	5.5 (0.2–22.2)
Prior treatment for NETs, n (%)	
Surgery	23 (76.7)
Chemotherapy	10 (33.3)
Interferon	7 (23.3)
Somatostatin analogs	6 (20.0)
Radiotherapy	1 (3.3)
Origin of NETs, n (%)	
Gastroenteropancreatic NETs, n=22	
Pancreas	8 (26.7)
Stomach	1 (3.3)
Duodenum	1 (3.3)
Ileum	6 (20.0)
Inferior jejunum	2 (6.7)
Inferior jejunum/ileum	1 (3.3)
Cecum	2 (6.7)
Right colon	1 (3.3)
Bronchopulmonary NETs, n=4	
Bronchus	4 (13.3)
Unknown	4 (13.3)
Tumor functionality, n (%)	
Functional	19 (63.3)
Carcinoid tumor	18 (94.7)
Gastrinoma	1 (3.3)
Non-functional	11 (36.7)
Chromogranin A, µg/L	332.5 (44.1–66056.0)
Urinary 5-HIAA, µmol/day	114.0 (19.9–1684.1)
Ki-67 index	2.0 (0.0–20.0)
ECOG category, n (%)	
Normal life without limitations (grade 0)	19 (63.3)
Limitation in activities (grade 1)	9 (30.0)
Unable to work (grade 2)	2 (6.7)

Data are median (range) unless stated otherwise; ECOG, Eastern Cooperative Oncology Group. NETs, neuroendocrine tumors.

- Median PFS was 12.9 months (95% CI: 7.9, 16.5) in the overall population (Figure 1), such that the probability of being progression-free at 54 weeks was 49.7% (95% CI: 29.4, 67.1), decreasing to 24.8% (95% CI: 10.2, 42.8) at 80 weeks. Median PFS was longer among patients with functioning (15.7 months [95% CI: 10.6, 19.2]) and midgut (16 months [95% CI: 7.4, 19.2]) tumors, although neither association was significant at the 5% level.

Figure 1. Kaplan–Meier plot of progression-free survival among 30 patients with progressive neuroendocrine tumors treated with lanreotide 120 mg every 28 days.

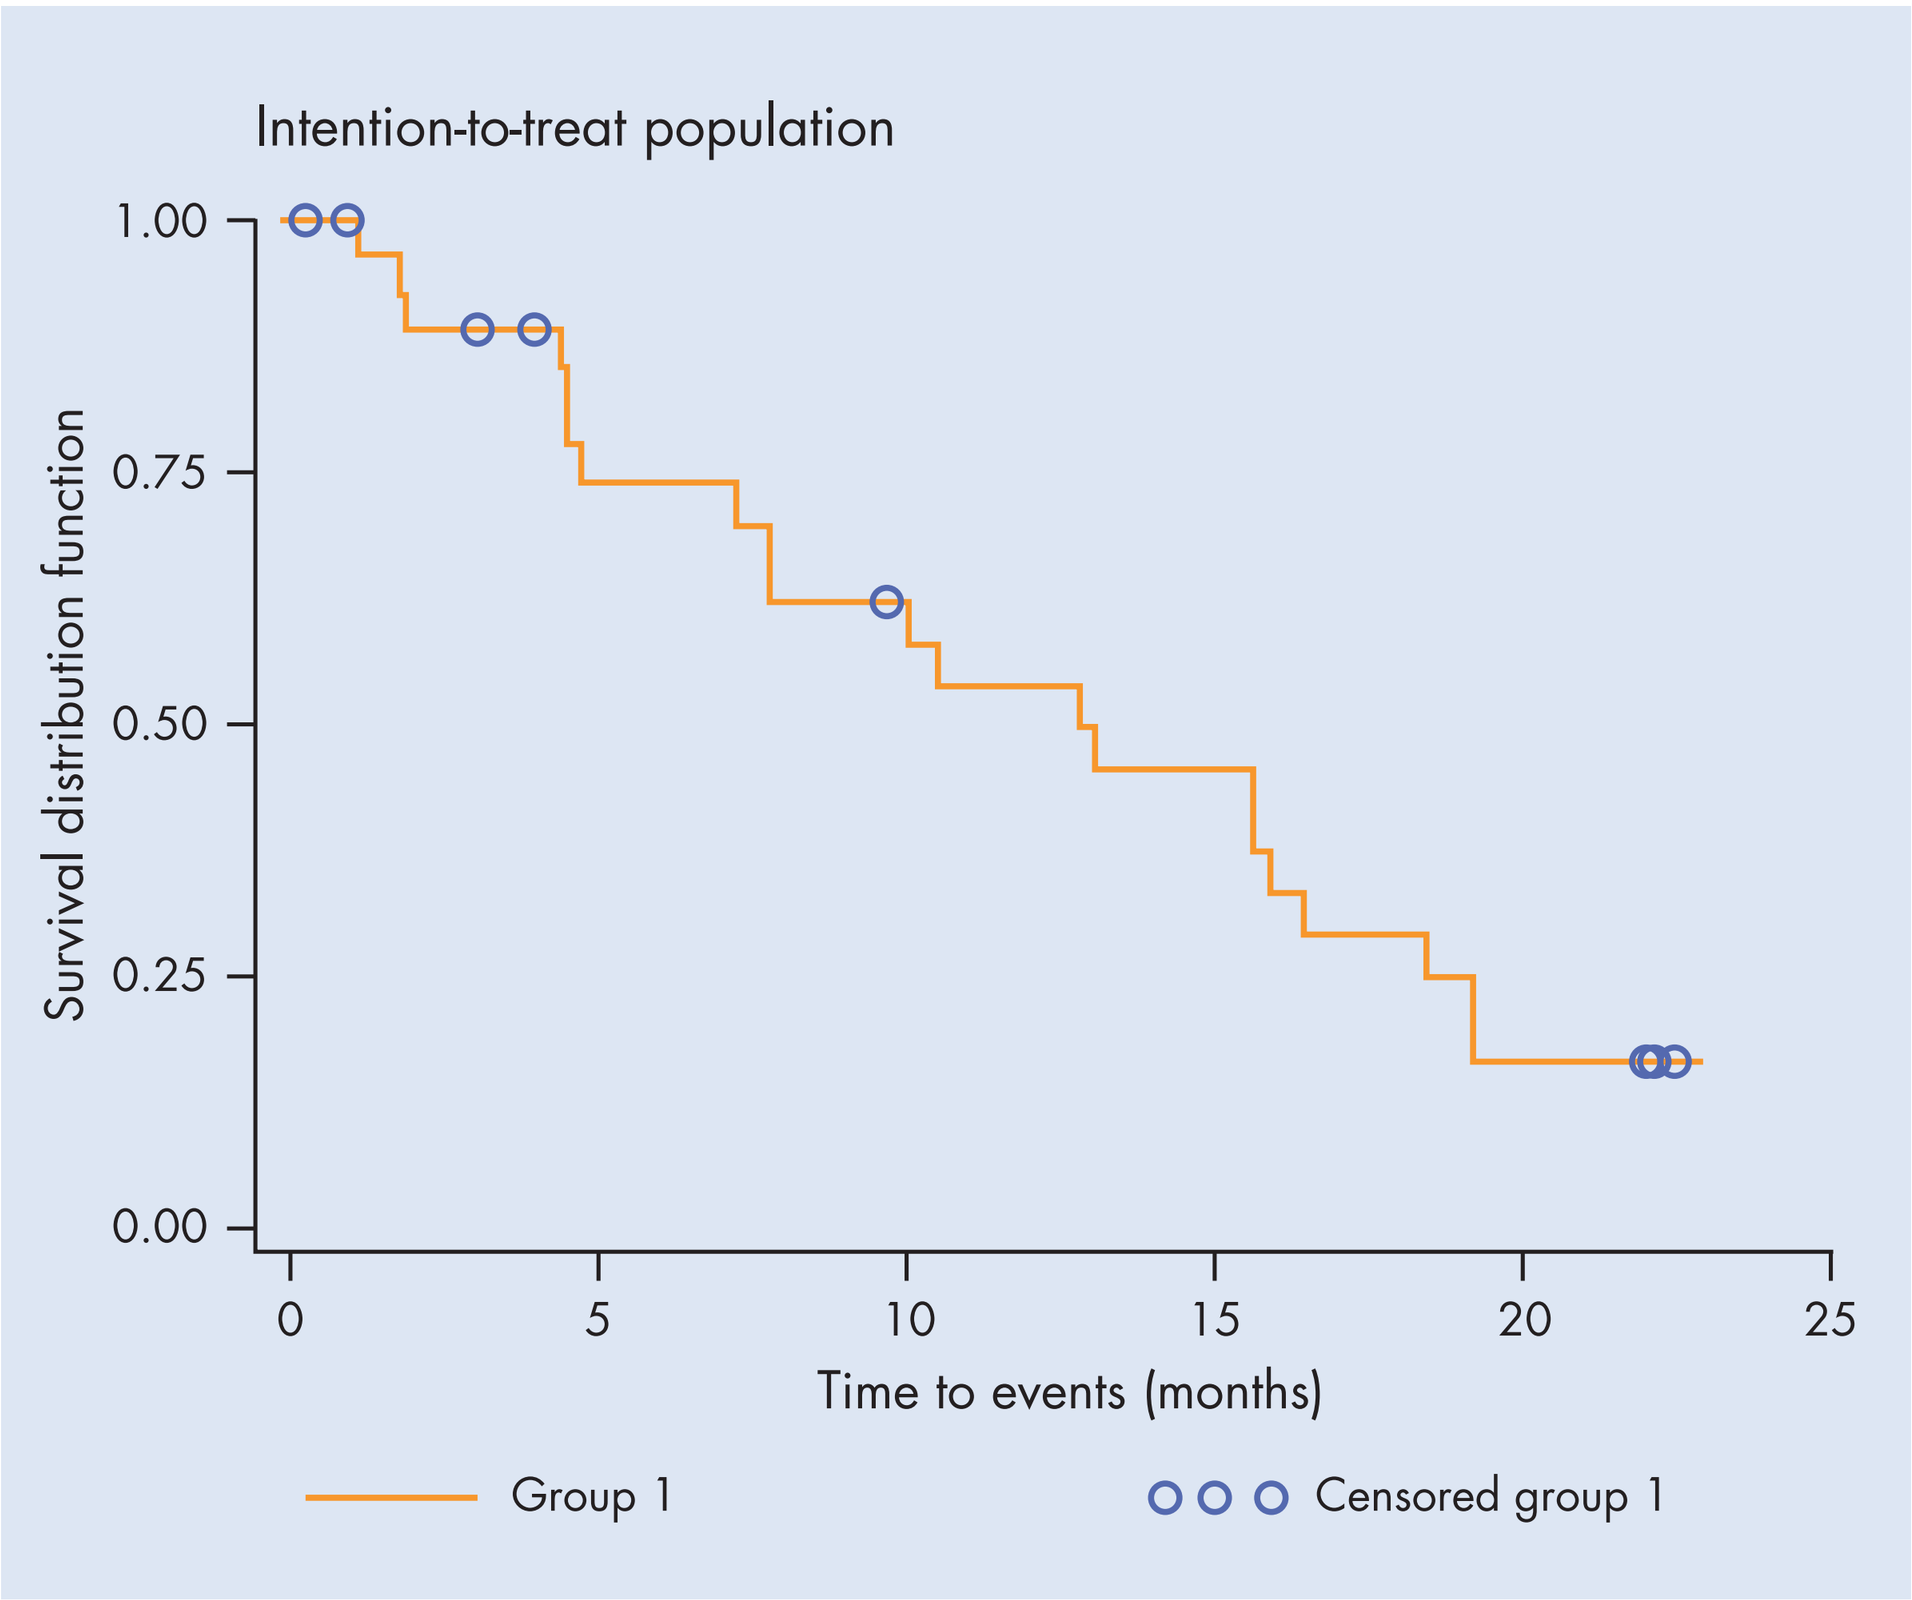
Intention-to-treat population
Survival distribution function
1.00
0.75
0.50
0.25
0.00
0
5
10
15
20
25
Time to events (months)
Group 1
Censored group 1

- Of 27 patients with a first assessment, two (7.4%) experienced disease progression and one (3.7%) had a partial response, with all other patients (n=24; 88.9%) exhibiting stable disease (Figure 2).
- Ki-67 index was the only factor predictive of PFS (n=21; HR 1.17; 95% CI: 1.03,1.33 p=0.018) in the multivariate analysis, such that a lower Ki-67 rank predicted longer PFS.
- After starting treatment, six patients reported 11 symptoms related to NET (diarrhea = 8; asthenia = 1; flushing = 1; respiratory discomfort and tachycardia = 1), all during the first 9 months, suggesting a trend towards symptom improvement. No symptoms were severe.
- By week 8, biochemical responses were:
 - serum CgA:
 - median percentage decrease (change) from baseline = 34.8% (–91.8 to –7.8; Wilcoxon's test: p<0.001), subsequently ranging from –30.6% to –16.8%
 - 70.4% (95% CI: 53.1, 87.6%; McNemar's test: 0.0002) of patients achieved biochemical response (normalization and/or ≥30% decrease in CgA); through the rest of the study this proportion ranged from 53–67%.
 - urinary 5-HIAA:
 - median percentage decrease (change) from baseline = 30.4% (95% CI: –90.4 to 24.7%; Student's *t*-test: p=0.0006), subsequently ranging from –63.5% to –28.5%.

Figure 2. Maximum decrease or minimum increase in sum of maximum diameter of target neuroendocrine tumor lesions (n=27). Each bar represents data for a single patient.

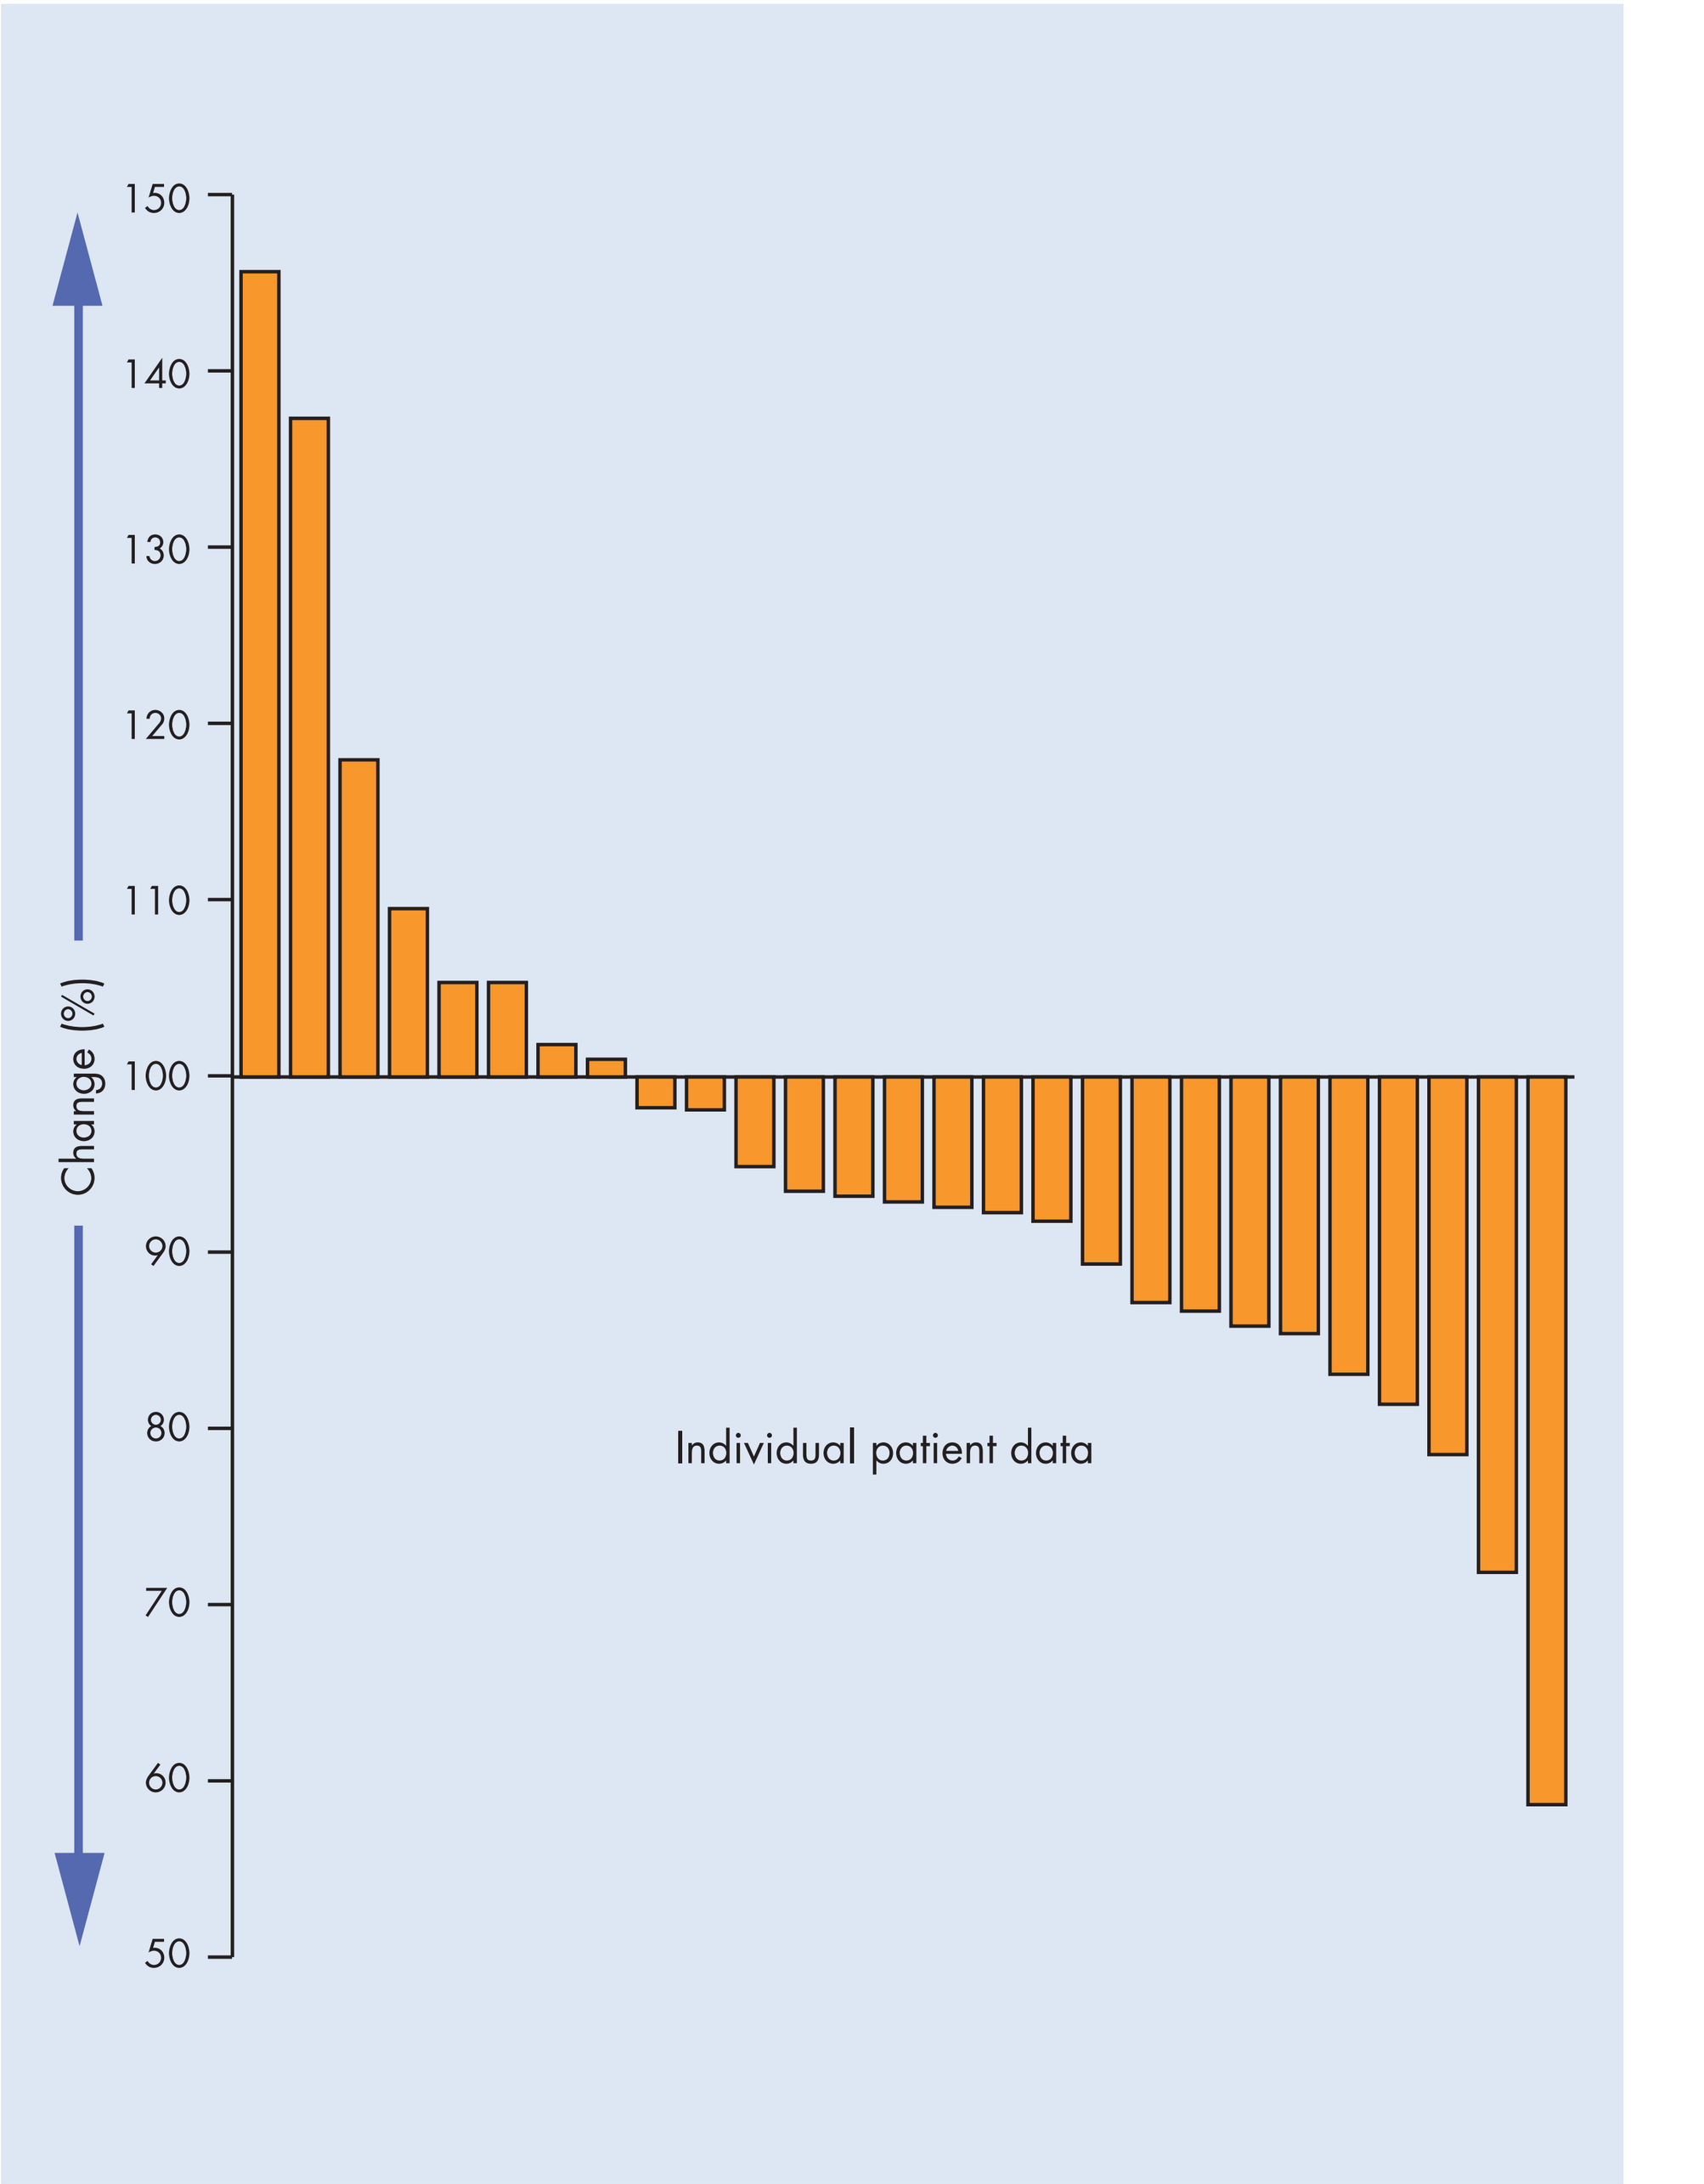
150
140
130
120
110
100
90
80
70
60
50
Change (%)
Individual patient data

- No impairment in quality of life was detected on EORTC QLQ-30 during treatment. Quality of life remained stable, with non-significant changes in most domains of functioning and symptom/side-effects scores.
- In all,169 treatment-emergent adverse events (TEAEs) were reported: 113 mild events in 22 patients (73.3%) and seven severe events in five patients (16.7%). The most frequently reported TEAEs were diarrhea (n=13; 43.3%), abdominal pain (n=4; 13.3%), asthenia (n=8; 26.7%) and nasopharyngitis (n=4; 13.3%).
- Sixty-nine TEAEs (40.8%) were related to treatment, of which only one was severe (aerophagia) (Table 2).
- Six patients reported eight serious adverse events (AEs), the most frequent being intestinal obstruction (three patients [10%]). Two patients (7.4%) withdrew because of AEs: one due to a serious treatment-related AE (moderate acute renal failure), the other from an unrelated cause (gallbladder fistulas).
- Two patients with a serious TEAE died: one withdrew due to disease progression and died during follow-up, the second patient's death, from disease-related intestinal obstruction, led to withdrawal from the study. Neither death was believed to be treatment-related.

Table 2. Number (percentage) of patients reporting treatment-related adverse events during treatment with lanreotide 120 mg every 28 days.

Organ system	Severe	Moderate	Mild	Total
Any	1 (3.3)	8 (26.7)	17 (56.7)	19 (63.3)
Gastrointestinal	1 (3.3)	4 (13.3)	12 (40.0)	13 (43.3)
General and injection site	-	5 (16.7)	9 (30.0)	10 (33.3)
Neurological	-	-	3 (10.0)	3 (10.0)
Metabolic and nutritional	-	1 (3.3)	1 (3.3)	2 (6.7)
Infections	-	1 (3.3)	-	1 (3.3)
Skin and subcutaneous tissue	-	-	1 (3.3)	1 (3.3)
Ear and labyrinth	-	-	1 (3.3)	1 (3.3)
Renal/urinary tract	-	1 (3.3)	-	1 (3.3)
Reproductive system and breast	-	-	1 (3.3)	1 (3.3)
Vascular	-	-	1 (3.3)	1 (3.3)

Conclusions

- In this study, administration of lanreotide 120 mg every 28 days to patients with a progressive GEP or BP NET provided a median PFS of >12 months and was well tolerated.
- Ki-67 index was the only factor predictive of PFS.
- This apparent stabilization of tumor growth should be confirmed in further studies, including an ongoing phase III trial in non-functioning NETs (CLARINET, NCT00842348).

References

1. Arnold R *et al. Digestion* 2000;62(Suppl. 1):84–91.
2. Rinke A *et al.*, for the PROMID Study Group. *J Clin Oncol* 2009;27: 4656–63.
3. Khan M *et al. Aliment Pharmacol Ther* 2011;34:235–42.
4. Aparicio T *et al. Eur J Cancer* 2001;37:1014–9.
5. Faiss S *et al. J Clin Oncol* 2003;21:2689–96.

Disclosures

The study was funded by Ipsen.

Acknowledgements

The authors thank the patients and investigators in this study. The authors also thank Watermeadow Medical for their assistance in preparing this poster.

*Lanreotide Autogel® is known as lanreotide Depot® in the USA.