Analysis of Progression-Free Survival by Prior Chemotherapy Use and Updated Safety in RADIANT-3: A Randomized, Double-Blind, Placebo-Controlled, Multicenter, Phase III Trial of Everolimus in Patients With Advanced Low- or Intermediate-Grade Pancreatic Neuroendocrine Tumors

Timothy Hobday,[1] Rodney Pommier,[2] Eric Van Cutsem,[3] Ashok Panneerselvam,[4] Stephen Saletan,[4] James C. Yao[5]

[1]Mayo Clinic, Rochester, Minnesota; [2]Oregon Health & Science University, Portland, Oregon; [3]University Hospital Gasthuisberg/Leuven, Leuven, Belgium; [4]Novartis Oncology, Florham Park, New Jersey; [5]The University of Texas MD Anderson Cancer Center, Houston, Texas

BACKGROUND

- Pancreatic neuroendocrine tumors (pNET) account for approximately 1.3% of the incidence of all pancreatic cancers and 4% to 7% of the incidence of all NET, although the incidences of both are increasing[1-4]
- At diagnosis, nearly 65% of patients with pNET have advanced metastatic disease, which is associated with a median survival time of 24 months[4]
- Until recently, the only US Food and Drug Administration (FDA)–approved treatment option for patients with pNET had been streptozotocin, though its usefulness in advanced pNET is unclear[5,6]
- Although the molecular pathogenesis of sporadic pNET is unknown, data suggest that the phosphatidylinositol 3-kinase/Akt/mammalian target of rapamycin (mTOR) pathway, which is important for the regulation of cell growth, proliferation, and angiogenesis, is involved[7-10]
- Everolimus, an oral mTOR inhibitor, showed promising antitumor activity in a phase II study in patients with pNET[11]
- In the large, prospective, international, multicenter, randomized, double-blind, placebo-controlled, phase III RADIANT-3 trial involving 410 patients with progressing low- or intermediate-grade advanced pNET, everolimus 10 mg/d + best supportive care (BSC) compared with placebo + BSC demonstrated a significant 6.4-month prolongation of median progression-free survival (PFS), as assessed by the local investigator (11.0 months vs 4.6 months; hazard ratio [HR], 0.35; 95% confidence interval [CI], 0.27-0.45; $P < 0.001$)[12]
 - The benefit provided by everolimus was durable, as evidenced by the 34% of patients who remained free of disease progression at 18 months[12]
- Everolimus was approved by the FDA in May 2011 and by the European Union Committee for Medicinal Products for Human Use in September 2011 for the management of progressive pNET that is unresectable, locally advanced, or metastatic

OBJECTIVES

- To perform an exploratory analysis of the effect of everolimus on PFS in patients who did or did not receive previous chemotherapy and to evaluate an updated overall survival (OS) analysis (cutoff date, February 23, 2011) and the 90-day safety update (cutoff date, June 3, 2010) of the RADIANT-3 trial

METHODS

Study Design and Patient Population

- RADIANT-3 was an international, multicenter, double-blind, placebo-controlled, phase III study[12] (**Figure 1**)

Figure 1. RADIANT-3 trial design.

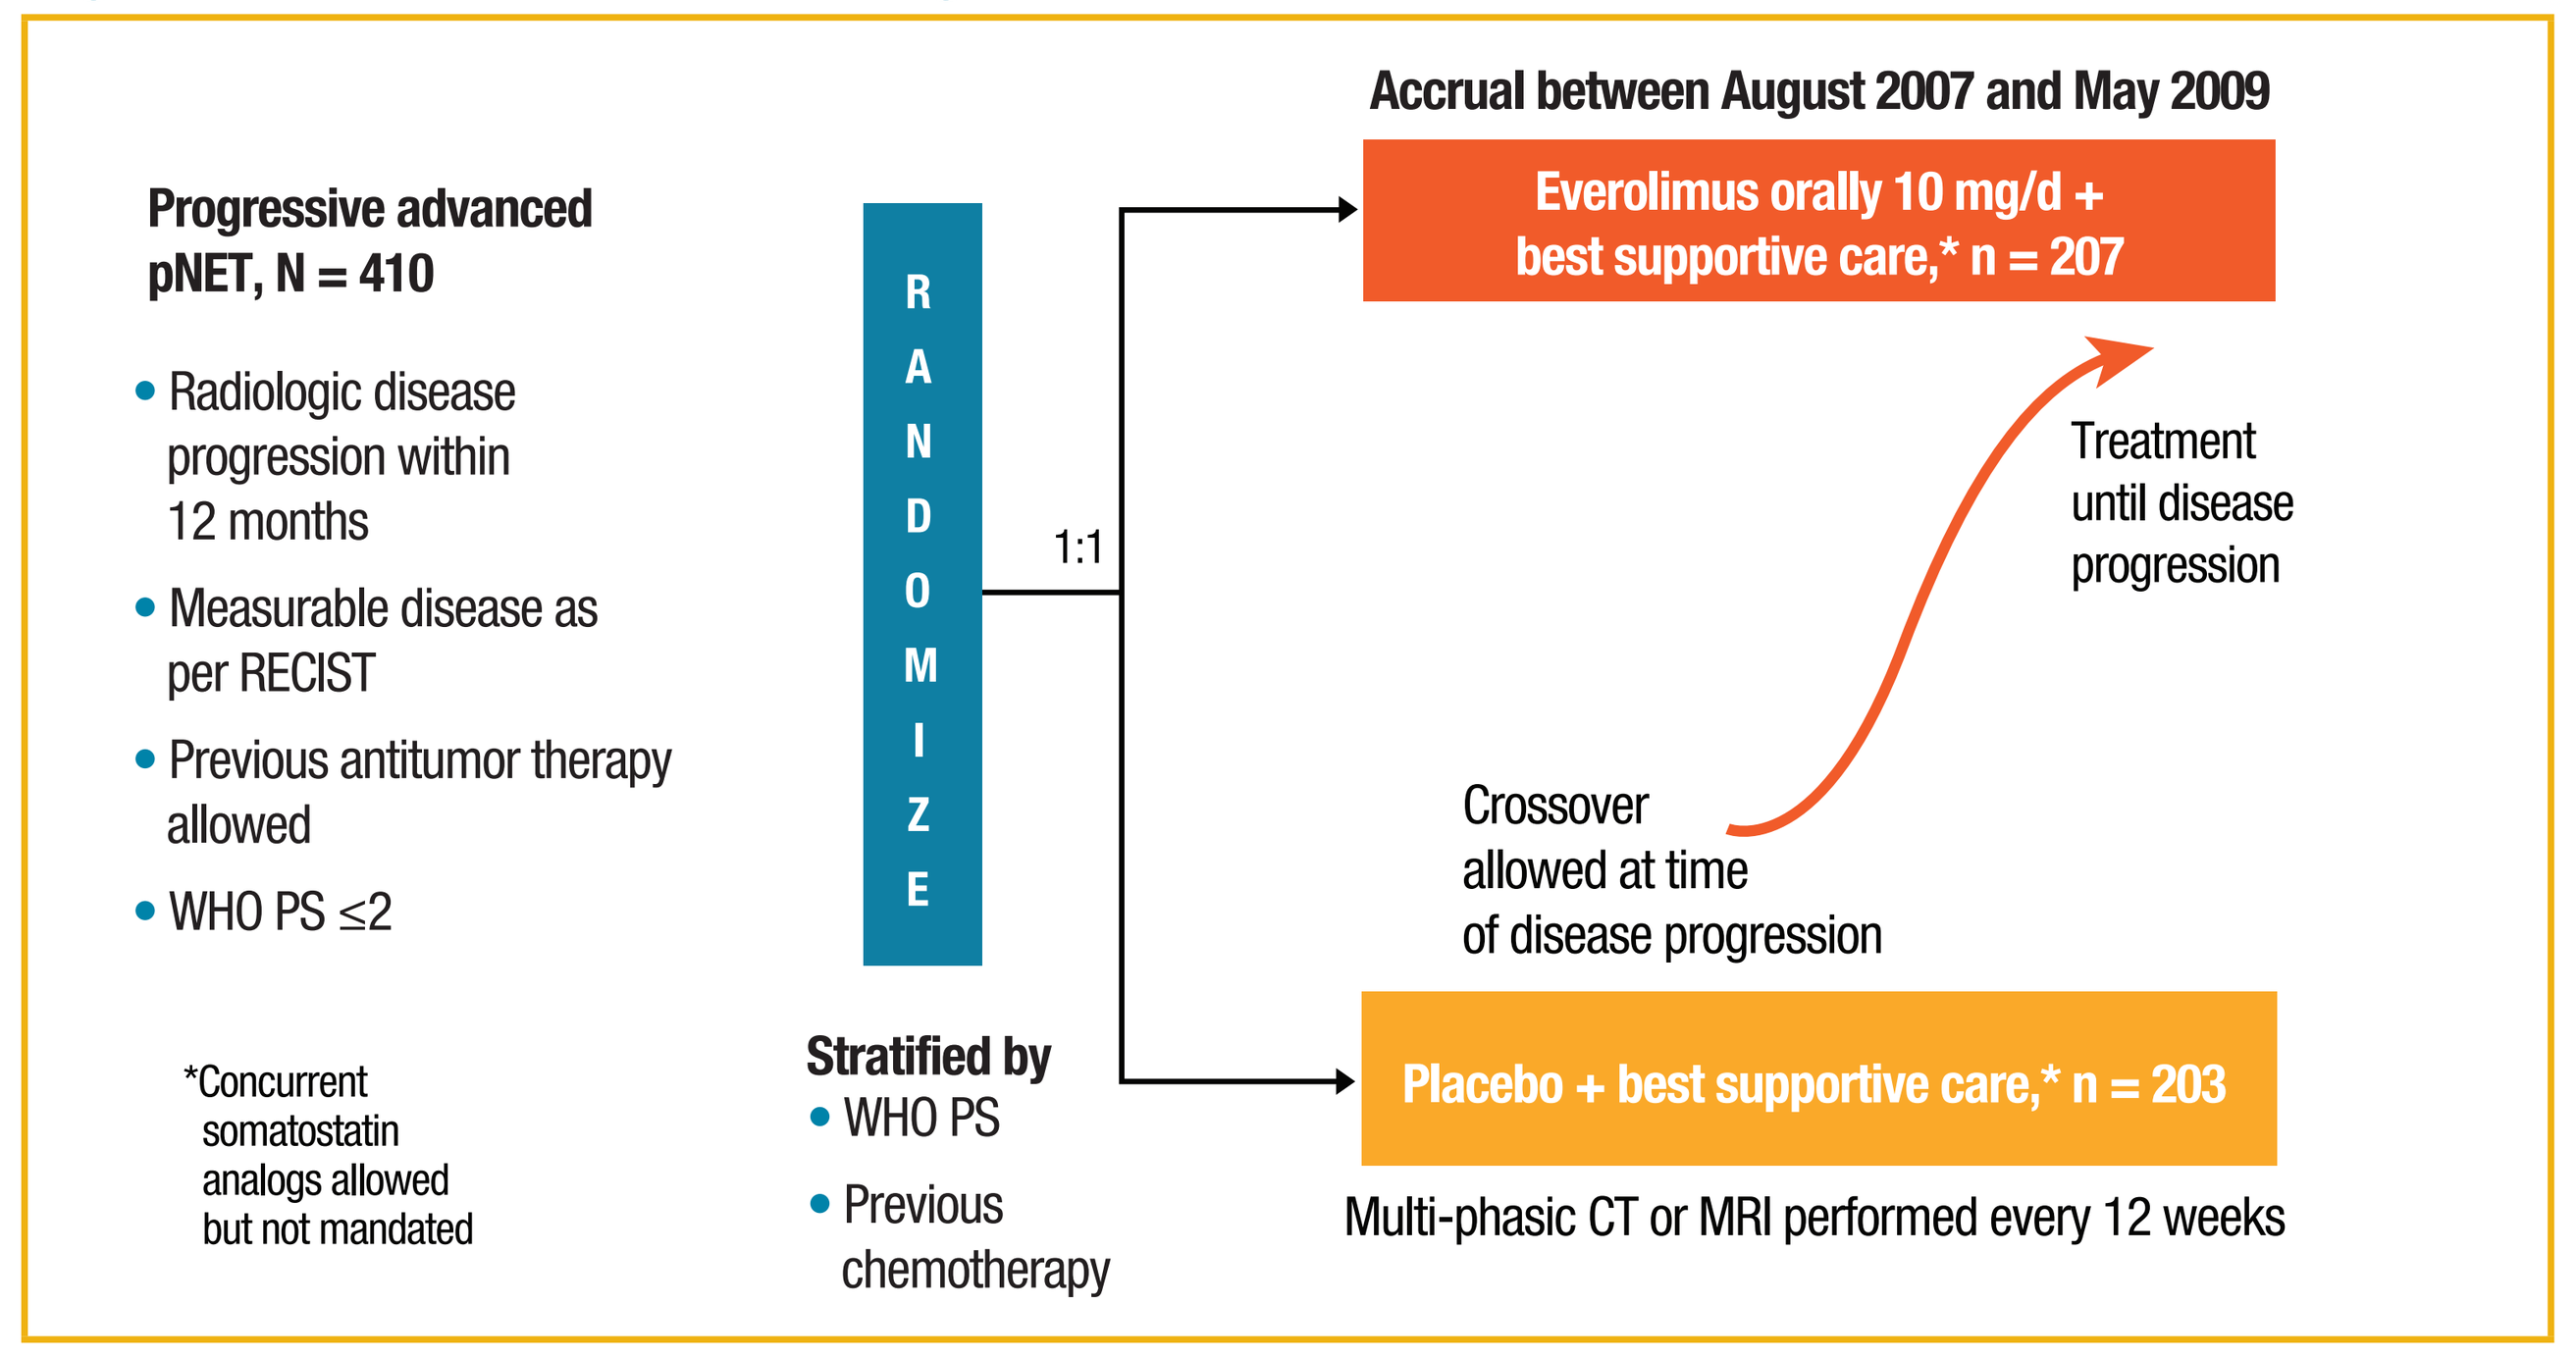
Accrual between August 2007 and May 2009
Progressive advanced pNET, N = 410
• Radiologic disease progression within 12 months
• Measurable disease as per RECIST
• Previous antitumor therapy allowed
• WHO PS ≤2
R A N D O M I Z E
1:1
Everolimus orally 10 mg/d + best supportive care,* n = 207
Treatment until disease progression
Crossover allowed at time of disease progression
Placebo + best supportive care,* n = 203
Stratified by
• WHO PS
• Previous chemotherapy
Multi-phasic CT or MRI performed every 12 weeks
*Concurrent somatostatin analogs allowed but not mandated

WHO PS, World Health Organization performance status.

- Crossover was allowed for patients randomly assigned to placebo at the time of disease progression
- Primary endpoint was PFS by local investigator assessment (Response Evaluation Criteria In Solid Tumors [RECIST], version 1.0). Secondary study endpoints included safety, tumor response, overall survival, and biomarker changes
- Patients were stratified by previous chemotherapy to evaluate the association between previous chemotherapy and PFS by local investigator assessment
- Adverse events (AEs) were collected and coded to a preferred term using the Medical Dictionary for Regulatory Activities and were assessed according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 3.0

RESULTS

Patient Demographics and Baseline Characteristics

- Baseline characteristics were well balanced between treatment arms (**Table 1**). Of the 410 patients with advanced pNET enrolled in the study, 206 (50.2%) had received previous chemotherapy

Table 1. Baseline Demographics and Disease Characteristics

	Everolimus n = 207	Placebo n = 203
Median age, years (range)	58 (23-87)	57 (20-82)
Gender, %		
Male	53	58
Female	47	42
Age group, %		
<65 years	71	75
≥65 years	29	25
Race, %		
Caucasian	75	82
Asian	19	17
Black	4	1
Other	1	1
WHO PS, %		
0	67	66
1	30	32
2	3	3
No. organs involved, %		
1	25	31
2	41	32
≥3	34	38
Histologic grade, %		
Well differentiated	82	84
Moderately differentiated	17	15
Other	1	1
Chemotherapy, %	50	50

WHO PS, World Health Organization performance status.

Patient Disposition

- Median follow-up in the overall population was 17 months; median exposure was 37.8 weeks in the everolimus group and 16.1 weeks in the placebo group
- Median follow-up for the 90-day safety analysis (cutoff date, June 3, 2010) was 20.1 months; with the additional follow-up, disease progression remained the primary reason for treatment discontinuation in both treatment arms

Exploratory Analysis of PFS by Previous Chemotherapy Use

- In the overall population, at the primary analysis data cutoff of February 28, 2010, treatment with everolimus significantly increased median PFS 2.4-fold versus placebo (11.0 months vs 4.6 months; $P < 0.001$) (**Table 2**)
- Everolimus improved median PFS by 7.8 months in patients who received previous chemotherapy and by 6.0 months in patients who did not receive any previous chemotherapy compared with placebo (**Table 2**)
- The increased PFS associated with everolimus was statistically significant irrespective of previous chemotherapy ($P < 0.001$) and similar to that observed in the overall population (**Table 2**)

Table 2. Median PFS by Previous Chemotherapy Use

	Everolimus + BSC n = 207	Placebo + BSC n = 203	HR (95% CI)[a]	P^b
	Months (95% CI)			
Overall N = 410	11.04 (8.41-13.86)	4.60 (3.06-5.39)	0.35 (0.27-0.45)	<0.001
Previous chemotherapy n = 206	11.01 (8.15-13.87) n = 104	3.19 (2.83-5.09) n = 102	0.35 (0.25-0.49)	<0.001
No previous chemotherapy n = 204	11.40 (8.31-15.38) n = 103	5.39 (3.19-5.62) n = 101	0.42 (0.29-0.60)	<0.001

[a]Stratified Cox model used to obtain HRs for overall population. Unstratified Cox model used to obtain HRs for patients who did and did not receive previous chemotherapy.
[b]1-sided *P* value for overall population; 2-sided *P* value for patients who did and did not receive previous chemotherapy.

- Within the everolimus treatment group, median PFS did not significantly differ in patients who did or did not receive previous chemotherapy (**Table 3**)
- Patients in the placebo arm who received previous chemotherapy had a trend toward shorter median PFS than those who received no previous chemotherapy

Table 3. Median PFS by Treatment Group

	Previous Chemotherapy	No Previous Chemotherapy	HR (95% CI)[a]	P^b
	Months (95% CI)			
Everolimus n = 207	11.01 (8.15-13.87) n = 104	11.40 (8.31-15.38) n = 103	1.12 (0.77-1.63)	0.560
Placebo n = 203	3.19 (2.83-5.09) n = 102	5.39 (3.19-5.62) n = 101	1.35 (0.99-1.83)	0.054

[a]Unstratified Cox model used to obtain HRs.
[b]2-sided *P* value.

Overall Survival

- At the updated analysis cutoff date of February 23, 2011, at 40 months of follow-up, median overall survival (OS) was 36.63 months in the placebo arm and had not been reached in the everolimus arm (HR, 0.89; 95% CI, 0.64-1.23) (**Figure 2**)
 - In total, 146 deaths occurred: 68 in the everolimus arm and 78 in the placebo arm
- 172 of 203 patients (84.7%) initially randomly assigned to placebo crossed over to open-label everolimus
 - These patients achieved a median OS of 36.63 months, which was likely the result of treatment with everolimus after crossover

Figure 2. Kaplan-Meier plot of overall survival.

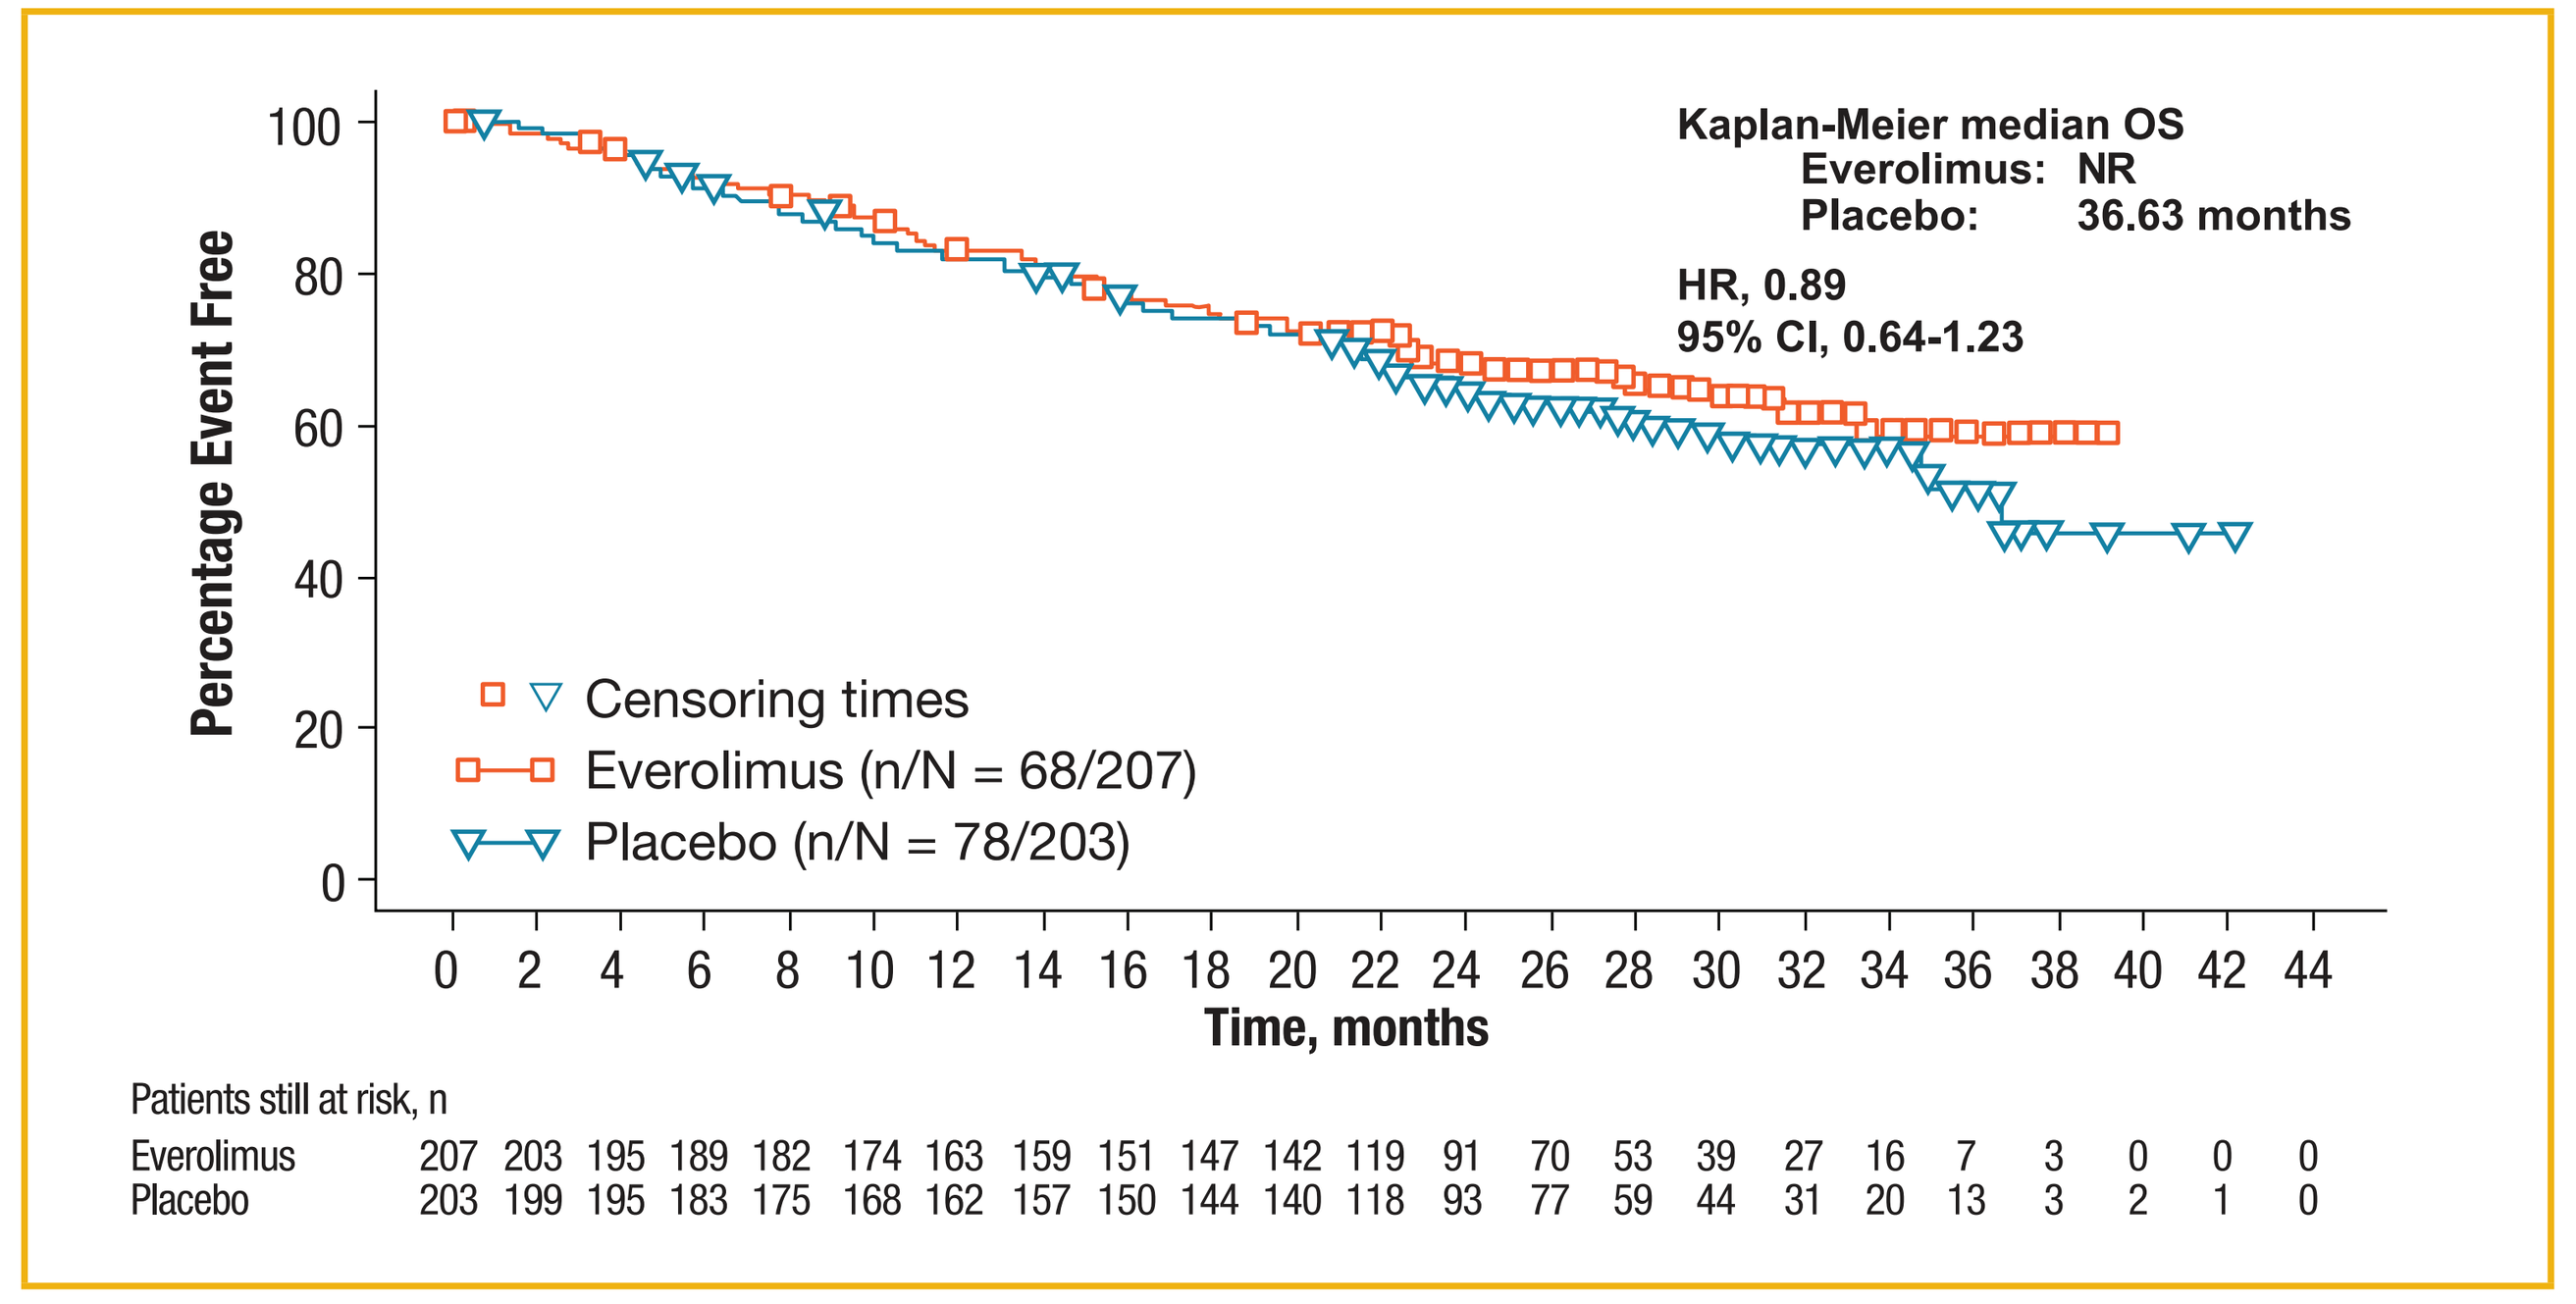
Kaplan-Meier median OS
Everolimus: NR
Placebo: 36.63 months
HR, 0.89
95% CI, 0.64-1.23
Percentage Event Free
100
80
60
40
20
0
Censoring times
Everolimus (n/N = 68/207)
Placebo (n/N = 78/203)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44
Time, months
Patients still at risk, n
Everolimus 207 203 195 189 182 174 163 159 151 147 142 119 91 70 53 39 27 16 7 3 0 0 0
Placebo 203 199 195 183 175 168 162 157 150 144 140 118 93 77 59 44 31 20 13 3 2 1 0

CI, confidence interval; HR, hazard ratio; NR, not reached.

Safety

- At the 90-day safety analysis (cutoff date, June 3, 2010), median follow-up was 20.1 months
- At the time of the extended safety analysis, 2.3-fold more patients allocated to everolimus remained on therapy compared with those assigned to placebo (24.0% and 10.3%, respectively)
- Treatment-related AE profiles remained unchanged in the 90-day safety analysis (**Table 4**)
- The most common nonhematologic treatment-related AEs with everolimus versus placebo were stomatitis (52.9% vs 12.3%), rash (48.5% vs 10.3%), diarrhea (34.3% vs 10.3%), and fatigue (32.4% vs 14.3%)

Table 4. Treatment-Related AEs Occurring in ≥5% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Stomatitis	52.9	11.3	52.9	12.3
Rash	48.5	10.3	48.5	10.3
Diarrhea	33.8	9.9	34.3	10.3
Fatigue	31.4	14.3	32.4	14.3
Infections and infestations	22.5	5.9	24.0	5.9
Peripheral edema	20.1	3.4	22.1	3.4
Headache	19.1	6.4	19.6	6.4
Nausea	20.1	18.2	20.1	18.2
Decreased appetite	19.6	6.9	20.1	6.9
Epistaxis	17.2	0	17.6	0
Dysgeusia	17.2	3.9	17.2	3.9
Decreased weight	15.7	4.4	16.7	4.4
Pulmonary events	16.7	0	16.7	0
Vomiting	15.2	6.4	15.2	6.4
Pruritus	14.7	8.9	14.7	8.9
Hyperglycemia	13.2	4.4	13.7	4.9
Asthenia	12.7	8.4	12.7	8.4
Pneumonitis	12.3	0	12.3	0
Nail disorder	11.8	1.0	12.7	1.0
Pyrexia	10.8	0	11.8	0
Cough	10.8	1.5	11.3	1.5
Aphthous stomatitis	10.8	3.4	10.8	3.4
Dry skin	10.3	4.4	10.3	4.4
Hypercholesterolemia	8.3	0	10.3	0
Hematologic, %				
Anemia	17.2	3.0	17.2	3.4
Thrombocytopenia	13.2	0.5	13.2	0.5
Neutropenia	6.4	2.0	6.4	2.0

- During the extended safety follow-up, most frequently reported nonhematologic grade 3/4 AEs related to everolimus treatment remained hyperglycemia, stomatitis, and diarrhea (**Table 5**)
- Low rates of grade 3 hyperglycemia were seen with both everolimus and placebo (**Table 4**)
 - No occurrences of grade 4 nonhematologic events were observed in the everolimus arm; grade 4 hyperglycemia occurred in 1% of patients in the placebo arm
- There were isolated treatment-related occurrences of grade 4 anemia (1.0%) and thrombocytopenia (<1%)

Table 5. Treatment-Related Grade 3/Grade 4 AEs Occurring in ≥3% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Hyperglycemia	5.4/0	1.5/0.5	5.9/0	1.5/1.0
Stomatitis	4.9/0	0/0	4.9/0	0/0
Diarrhea	3.4/0	0/0	3.4/0	0/0
Hematologic, %				
Anemia	4.9/1.0	0/0	4.9/1.0	0/0
Thrombocytopenia	3.4/0.5	0/0	3.4/0.5	0/0

Data are percentages of grade 3/grade 4 AEs.

- Most commonly reported AEs leading to the discontinuation of everolimus therapy were pneumonitis (3.4%), pyrexia (1.5%), fatigue, respiratory failure, interstitial lung disease, increased aspartate aminotransferase, and pneumonia (1.0% each)
- Most commonly reported AEs leading to dose interruptions or reductions of everolimus therapy were stomatitis (10.3%), thrombocytopenia (7.8%), and pneumonitis (6.9%)

CONCLUSIONS

- **In the large, randomized, double-blind, placebo-controlled, phase III RADIANT-3 trial of patients with advanced pNET, treatment with everolimus resulted in a clinically and statistically significant prolongation of PFS by 7.8 months in patients who received previous chemotherapy and by 6.0 months in patients who did not receive previous chemotherapy**
 - **The median PFS did not significantly differ in patients who did or did not receive previous chemotherapy and who received everolimus**
- **Results of the updated survival analysis (1 year after the primary analysis) at 40 months of follow-up show that median OS has not been reached in the everolimus arm; 146 deaths occurred, and final analysis will be performed once approximately 250 deaths have occurred[11]**
 - **The median OS of 36.6 months in the placebo arm, in which substantial crossover occurred that benefited these patients, exceeded the median OS of 24 months historically observed for patients with metastatic pNET[4]**
- **At a median follow-up of 20.1 months, updated safety results were consistent with those of the primary analysis and the known safety profile of everolimus in patients with cancer**
 - **No increase in AEs was observed with longer exposure to everolimus, demonstrating that the agent is tolerated long term**
- **These results suggest that everolimus may be considered a first-line treatment option before chemotherapy for patients with advanced pNET**

REFERENCES

1. Ries LAG et al. National Cancer Institute; 2000:1-66. NIH Pub. No. 00-2789.
2. Yao JC et al. *Ann Surg Oncol.* 2007;14:3492-3500.
3. Hauso O et al. *Cancer.* 2008;113:2655-2664.
4. Yao JC et al. *J Clin Oncol.* 2008;26:3063-3072.
5. Cheng PN et al. *Cancer*. 1999;86:944-948.
6. Delaunoit T et al. *Eur J Cancer*. 2004;40:515-520.
7. Jensen RT et al. *Cancer.* 2008;113(suppl 7):1807-1843.
8. von Wichert G et al. *Cancer Res.* 2000;60:4573-4581.
9. Missiaglia E et al. *J Clin Oncol.* 2010;28:245-255.
10. Jiao Y et al. *Science*. 2011;331:1199-1203.
11. Yao JC et al. *J Clin Oncol.* 2010;28:69-76
12. Yao JC et al. *N Engl J Med.* 2011;364:514-523.

The authors wish to acknowledge the contribution of Robert E. Winkler to the study.

Presented at the 4th Annual Neuroendocrine Tumor Symposium; October 20–22, 2011; Minneapolis, Minnesota

This study was sponsored by Novartis Pharmaceuticals Corporation.