Effect of Open-Label Everolimus in Patients With Advanced Neuroendocrine Tumors After Disease Progression on Somatostatin Analog: A RADIANT-2 Analysis

Edward M. Wolin,[1] John D. Hainsworth,[2] Dieter Hörsch,[3] Gabriele Luppi,[4] Valentine Jehl,[5] and Marc Peeters[6]

[1]Cedars-Sinai Medical Center, Los Angeles, CA, USA; [2]Sarah Cannon Research Institute, Nashville, TN, USA; [3]Klinik für Innere Medizin, Gastroenterologie und Endokrinologie, Zentrum für Neuroendokrine Tumore, Zentralklinik Bad Berka GmbH, Bad Berka, Germany; [4]Azienda Ospedaliero-Universitaria, Modena, Italy; [5]Novartis Pharma AG, Basel, Switzerland; [6]Department of Oncology, Antwerp University Hospital, Edegem, Belgium

BACKGROUND

- Neuroendocrine tumors (NET) are a diverse family of tumors that arise from various primary sites
- NET are difficult to diagnose and have a poor prognosis; most are metastatic at diagnosis, and 65% of patients die within 5 years of diagnosis[1]
- NET often secrete hormones or vasoactive peptides, which leads to hormonal syndromes such as hyperinsulinemia and carcinoid syndrome[2]
- Octreotide is a somatostatin analog that has shown efficacy for the management of hormonal syndromes in patients with NET and has been used in the treatment of patients with syndromic NET for more than 25 years[3,4]
 - More recently, the phase III, placebo-controlled PROMID study showed that octreotide long-acting repeatable (LAR) has antitumor efficacy in advanced, progressive, midgut NET; median time to tumor progression was 14.3 months in the octreotide LAR arm and 6.0 months in the placebo arm (hazard ratio [HR], 0.34; 95% confidence interval [CI], 0.20-0.59, *P* = 0.000072)[5]
- The mammalian target of rapamycin (mTOR) inhibitor everolimus has shown antitumor efficacy in patients with advanced pancreatic NET[6,7] and is approved for use in both the United States and Europe
- In the large, randomized, phase III, placebo-controlled RADIANT-2 trial of everolimus + octreotide LAR in patients with advanced, progressive NET with carcinoid symptoms (flushing, diarrhea, or both), patients receiving everolimus + octreotide LAR had progression-free survival (PFS) of 16.4 months as per adjudicated central review compared with 11.3 months in patients receiving placebo + octreotide LAR (HR, 0.77; 95% CI, 0.59-1.00; 1-sided log-rank test, *P* = 0.026 [not statistically significant because the prespecified boundary for significance was *P* ≤0.0246])[8]
 - More PFS events were captured in the local investigator review (HR, 0.78; 95% CI, 0.62-0.98; *P* = 0.018)[8]
 - Local investigator review supports the efficacy of everolimus + octreotide LAR in this patient population
- RADIANT-2 was designed to allow for crossover to open-label everolimus + octreotide LAR in those patients who experienced disease progression while receiving placebo + octreotide LAR in the double-blind portion of the study (**Figure 1**)[8]
- Here we present an analysis of the PFS of patients who crossed over to open-label everolimus + octreotide LAR upon disease progression

METHODS

Figure 1. RADIANT-2 study design.[8]

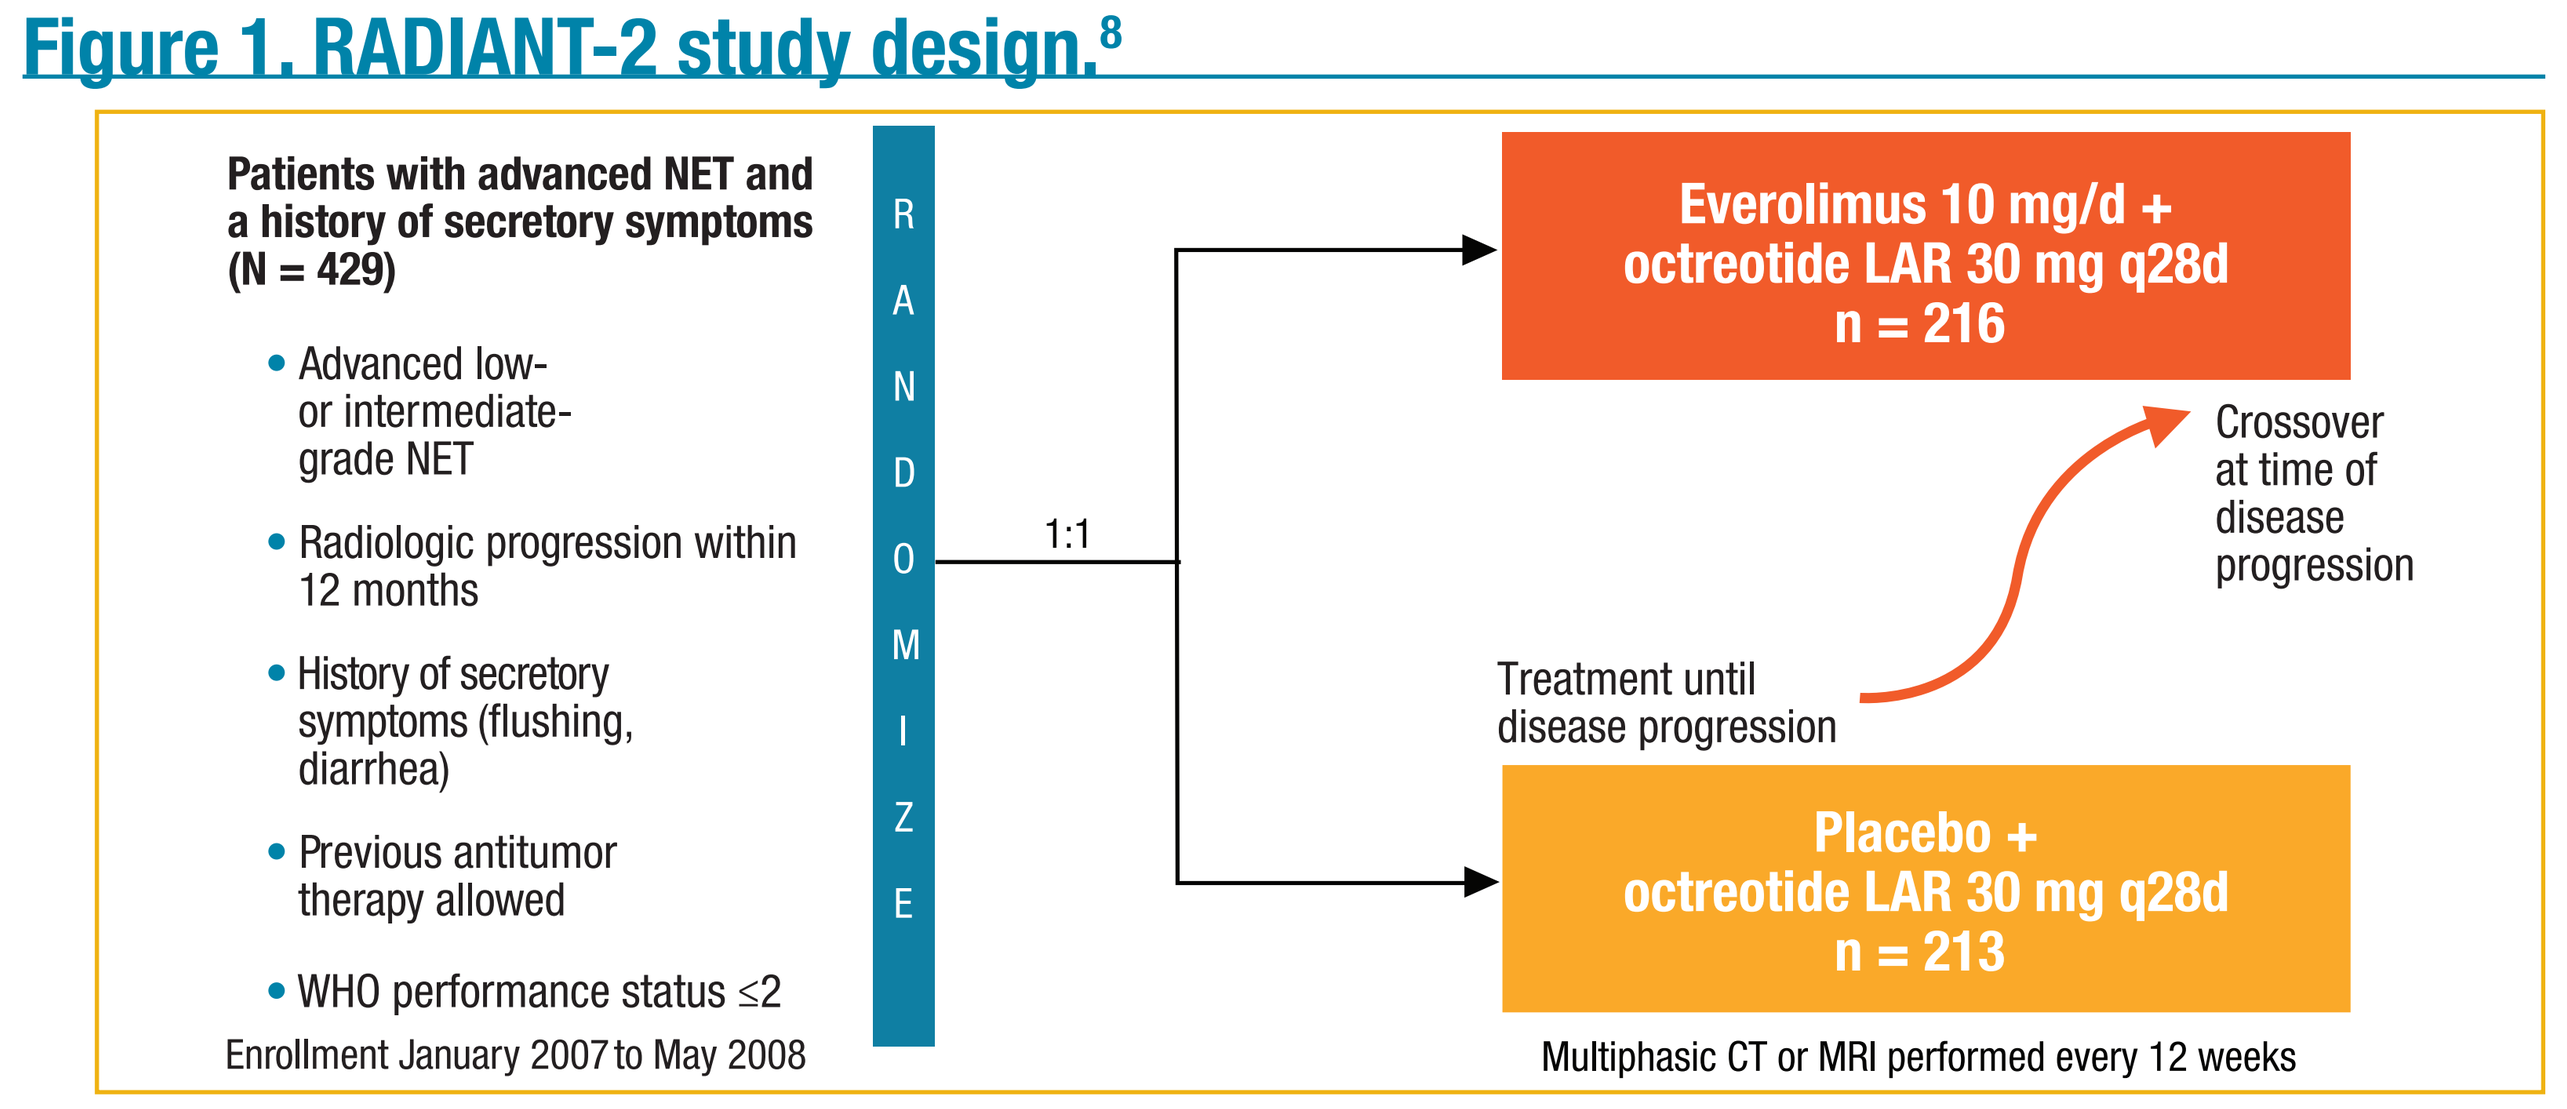

Patients with advanced NET and a history of secretory symptoms (N = 429)
• Advanced low- or intermediate-grade NET
• Radiologic progression within 12 months
• History of secretory symptoms (flushing, diarrhea)
• Previous antitumor therapy allowed
• WHO performance status ≤2
Enrollment January 2007 to May 2008
RANDOMIZE
1:1
Everolimus 10 mg/d + octreotide LAR 30 mg q28d n = 216
Crossover at time of disease progression
Treatment until disease progression
Placebo + octreotide LAR 30 mg q28d n = 213
Multiphasic CT or MRI performed every 12 weeks

CT = computed tomography; LAR, long-acting repeatable; MRI = magnetic resonance imaging; NET = neuroendocrine tumors; q28d = every 28 days; WHO = World Health Organization.

- Patients with advanced NET and a history of symptoms attributed to carcinoid syndrome (eg, flushing, diarrhea, or both) and disease progression (per Response Evaluation Criteria In Solid Tumors [RECIST] version 1.0 criteria) within 12 months of randomization were included in RADIANT-2
- The study was designed for well-differentiated or moderately differentiated NET (low or intermediate grade); poorly differentiated tumors were excluded
- Patients were allowed to cross over from the control arm to receive open-label everolimus + octreotide LAR upon disease progression, as determined by the investigator
 - A new radiologic baseline (per RECIST v1.0) was not required before the initiation of open-label everolimus
 - During the open-label part of the trial, tumors were evaluated only by local investigator; no central review was performed
- Data analyses
 - Investigator-assessed PFS was performed as a key supportive analysis; PFS was censored for additional antitumor therapy and when progression or death occurred after 2 or more missing assessments
 - The open-label data set consisted of all patients who received at least 1 dose of open-label everolimus and who had at least 1 valid safety assessment after the initiation of open-label treatment
 - The time between the start of open-label everolimus and the earliest date between progression, death, or start of further antitumor therapy was evaluated; patients without progression or death were censored at their last available tumor assessment

RESULTS

- 124 of the 213 patients initially randomly assigned to the placebo + octreotide LAR arm crossed over to open-label everolimus + octreotide LAR
- Total patient-years of exposure to open-label everolimus were 82.6; duration of exposure to everolimus in the open-label population is reported in **Table 1**

Table 1. Duration of Exposure to Open-Label Everolimus

	Open-Label Everolimus (n = 124)
Exposure, weeks	
<4	10 (8.1)
4 to <8	16 (12.9)
8 to <12	13 (10.5)
12 to <24	18 (14.5)
24 to <36	19 (15.3)
36 to <52	14 (11.3)
52 to <78	20 (16.1)
78 to <104	12 (9.7)
104 to <130	1 (0.8)
≥130	1 (0.8)
Duration of exposure, weeks	
Mean (standard deviation)	34.8 (30.09)
Median (range)	26.3 (1-133)

Unless otherwise noted, all data are presented as n (%).

- Based on the full analysis set, when censoring patients who never started open-label everolimus, the median time from randomization to crossover to open-label everolimus + octreotide LAR was 14.1 months (Kaplan-Meier estimate)
- Median everolimus relative dose intensity was 0.98 (range, 0.4-1.0)
- 34 patients (27.4%) received open-label everolimus + octreotide LAR for ≥12 months
- Median duration of treatment with open-label everolimus + octreotide LAR was 26.3 weeks (range, 1-133 weeks); median duration in the blinded treatment phase was 37 weeks
- Of 124 patients in the double-blind placebo + octreotide LAR arm, 123 (58%) also had an open-label safety assessment and are included in the PFS analysis of patients who crossed over (**Figure 2**)
 - The open-label safety set included a total of 124 patients; 1 patient was initially randomly assigned to everolimus and continued open-label everolimus

Figure 2. PFS after crossover to open-label everolimus (per local investigator review; patients randomly assigned to placebo only).

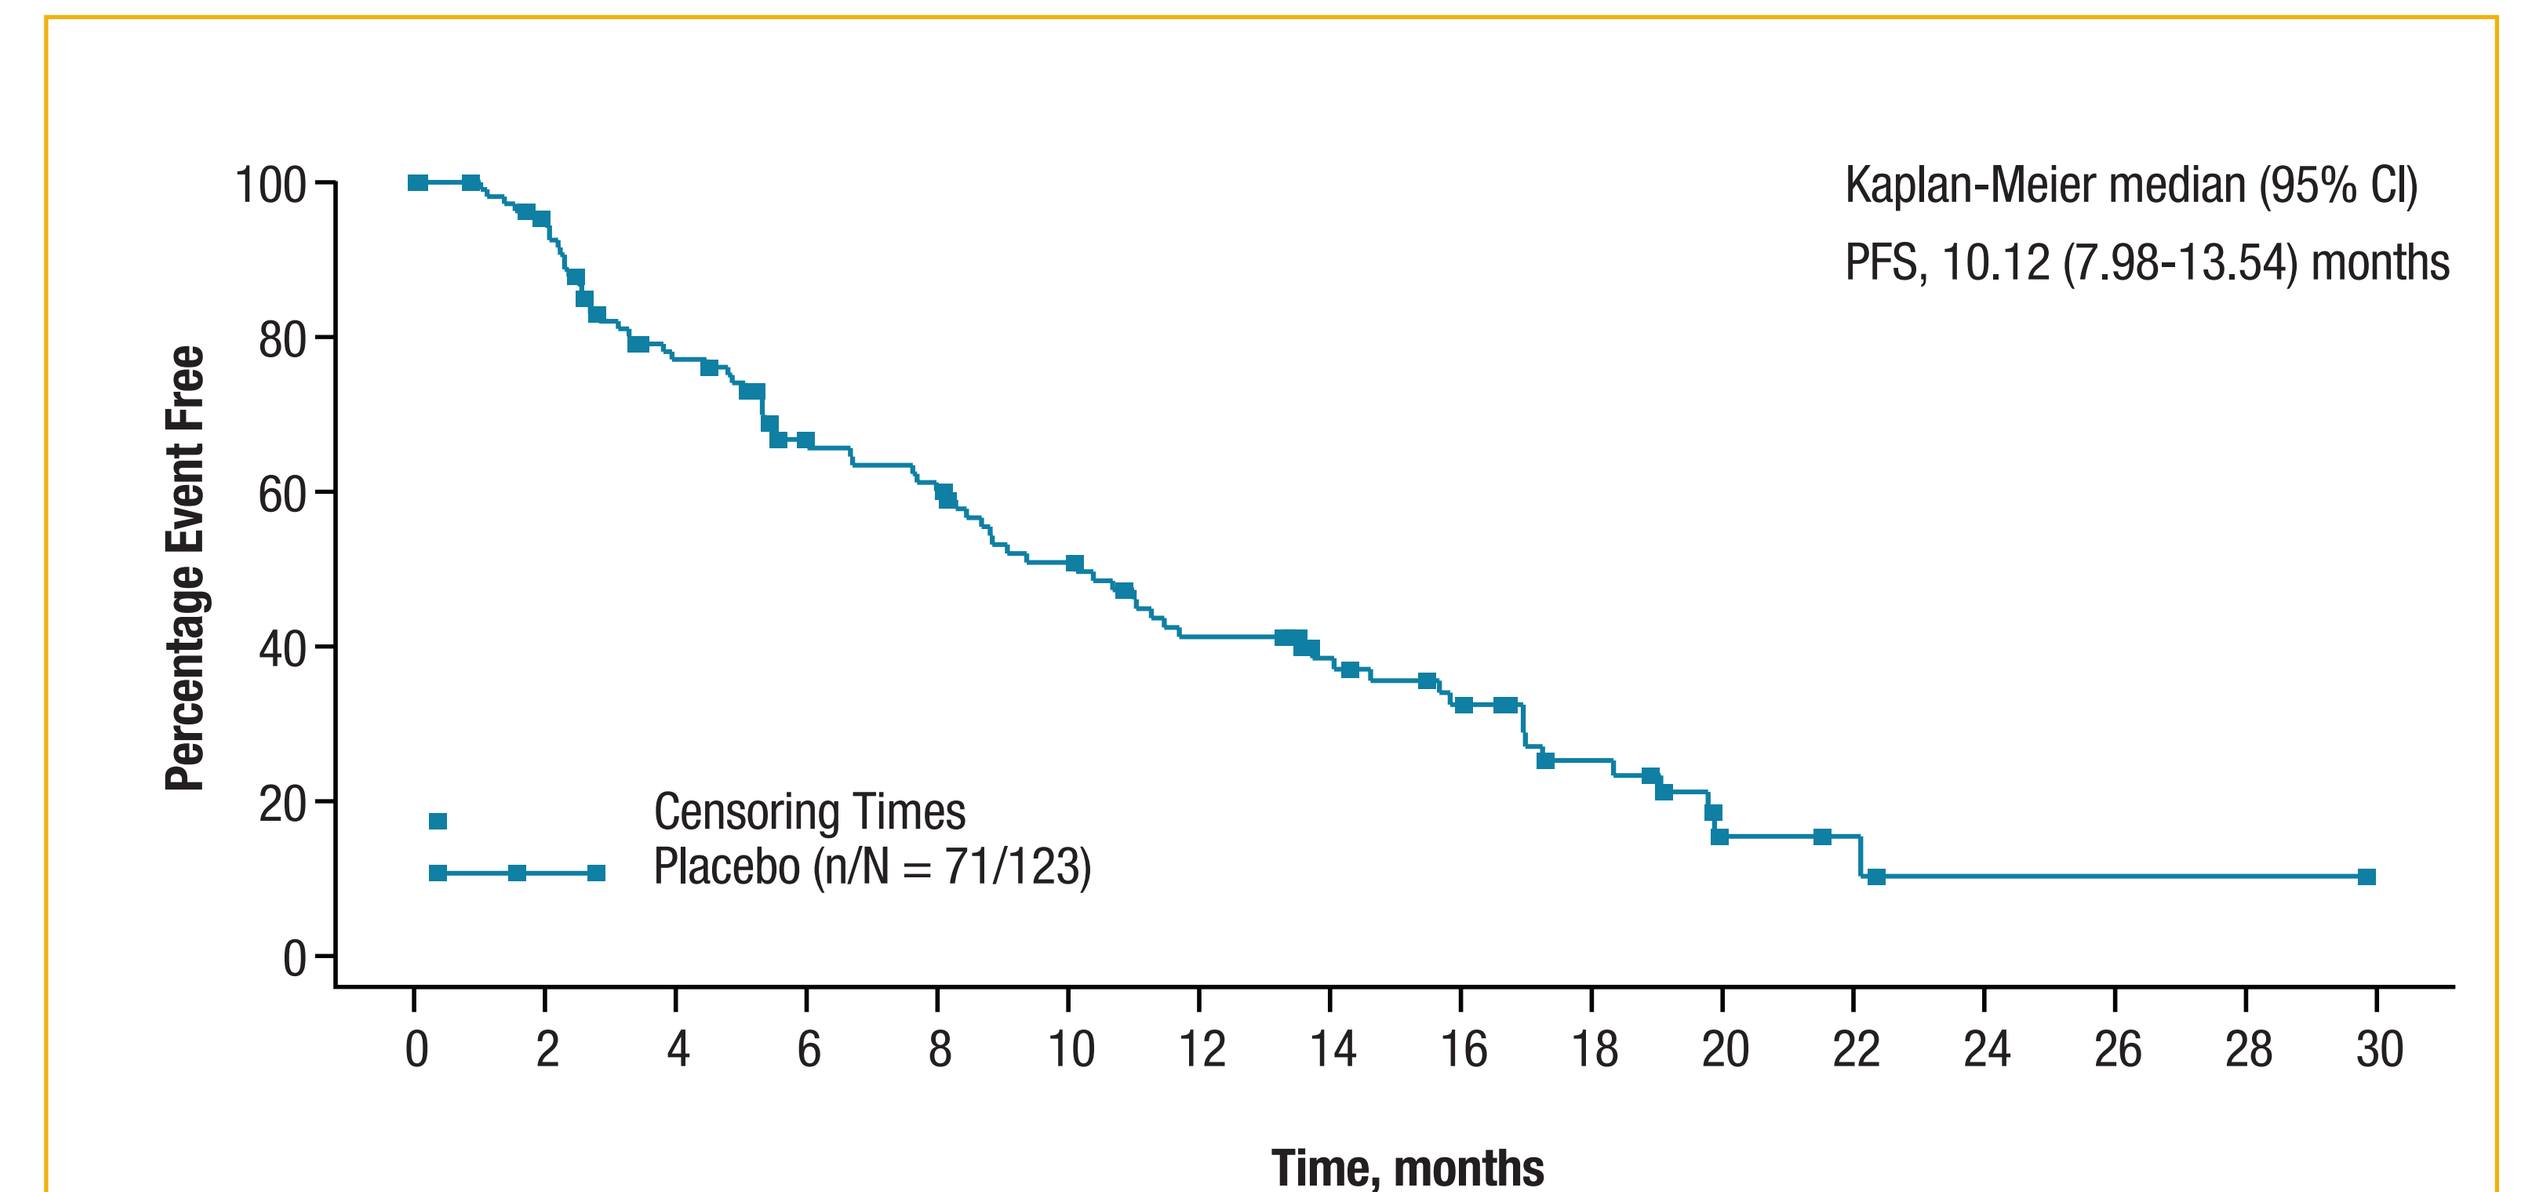

Kaplan-Meier median (95% CI)
PFS, 10.12 (7.98-13.54) months
Percentage Event Free
100
80
60
40
20
0
Censoring Times
Placebo (n/N = 71/123)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Time, months

No. patients still at risk

Time, months	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30
Placebo	123	101	77	60	54	44	34	27	21	13	4	3	1	1	1	0

CI = confidence interval; PFS = progression-free survival.

- PFS was censored for additional antitumor therapy
- Of the 71 PFS events, 45 (36.6%) were attributed to disease progression and 26 (21.1%) to patient death

Safety

- The most common adverse events (AEs) during the open-label phase were diarrhea, stomatitis, rash, and fatigue (**Table 2**); AEs were typically grade 1 or grade 2 (data not shown)
- The type and nature of the treatment-related AEs reported during the open-label phase of the study were consistent with those reported for the everolimus + octreotide LAR treatment group during the blinded treatment phase
- The most common serious AEs were abdominal pain (6.5%), diarrhea (4.8%), pneumonia (4.0%), anemia (3.2%), general physical health deterioration (3.2%), sepsis (3.2%), and vomiting (3.2%)

Table 2. Adverse Events During the Open-Label Period

Adverse Event (AE)	Open-Label Everolimus + Octreotide LAR n = 124
Any AE	122 (98.4)
Treatment-related AE	111 (89.5)
Treatment-related AE grade 3 or 4	44 (35.5)
Serious AE	62 (50.0)
Treatment-related serious AE	13 (10.5)
AE leading to study discontinuation	29 (23.4)
Treatment-related AE leading to study discontinuation	13 (10.5)
AE requiring dose interruption and/or reduction	77 (62.1)
Treatment-related AE requiring dose interruption and/or reduction	55 (44.4)
Grade 3/4 treatment-related AEs by preferred term in ≥5 of patients	
Stomatitis[a]	8 (6.5)
Diarrhea	7 (5.6)
Asthenia	5 (4.0)
Fatigue	5 (4.0)
Rash	5 (4.0)

All data are presented as n (%).
[a]Stomatitis includes stomatitis and aphthous stomatitis.

CONCLUSIONS

- **Patients with NET who were enrolled in the RADIANT-2 study and received open-label everolimus + octreotide LAR after progressing on placebo + octreotide LAR in the double-blind phase of the study had median PFS of 10.1 months**
- **The safety profile was similar to that reported for the double-blind phase of the study**
- **These results suggest a benefit of open-label everolimus + octreotide LAR after disease progression on placebo + octreotide LAR**

REFERENCES

1. Yao JC et al. *J Clin Oncol*. 2008;26:3063-3072.
2. van der Lely AJ et al. *Arq Bras Endocrinol Metabol*. 2005;49:850-860.
3. Kvols LK et al. *N Engl J Med*. 1986;315:663-666.
4. Rubin J et al. *J Clin Oncol*. 1999;17:600-606.
5. Rinke A et al. *J Clin Oncol*. 2009;27:4656-4663.
6. Yao JC et al. *J Clin Oncol*. 2008;26:4311-4318.
7. Yao JC et al. *N Engl J Med*. 2011;364:514-523.
8. Pavel ME et al. *Lancet*. 2011;378:2005-2012.

Mobile Friendly e-Prints — 3 ways to instantly download an electronic copy of this poster to your mobile device or e-mail a copy to your computer or tablet

Scan this QR code

Text Message (SMS)
Text: **Q4bb1f**
to: 8NOVA (86682) *US Only*
+18324604729 *North, Central and South Americas; Caribbean; China*
+447860024038 *UK, Europe & Russia*
+46737494608 *Sweden, Europe*

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=4bb1f

Standard data or message rates may apply.

North American Neuroendocrine Tumor Society Annual Conference; October 11-13, 2012; San Diego, California

This study was sponsored by Novartis Pharmaceuticals Corporation.