Multi-Center Phase II Trial Of Temsirolimus (TEM) and Bevacizumab (BEV) in Pancreatic Neuroendocrine Tumor (PNET): Results of a Planned Interim Efficacy Analysis

Timothy Hobday MD[1], Rui Qin PhD[1], Diane Reidy MD[2], Malcolm Moore MD[3], Jonathon Strosberg MD[4], Andreas Kaubisch MD[5], Manisha Shah MD[6], Hedy Kindler MD[7], Heinz-Joseph Lenz MD[8], Helen Chen MD[9], Charles Erlichman MD[1]

[1]Mayo Clinic College of Medicine, Rochester, MN (Mayo Phase 2 Consortium [P2C]), [2]Memorial Sloan-Kettering Cancer Center (MMSK), New York, NY (MMSK P2C), [3]Princess Margaret Hospital, Toronto, ON (Princess Margaret P2C), [4]H Lee Moffit Cancer Center, Tampa, Fl (Southeast P2C), [5]Montefiore Medical Center, Bronx, NY (Montefiore P2C), [6]Ohio State University, Columbus, OH (Ohio State P2C), [7]University of Chicago, Chicago, Il (University of Chicago P2C), [8]University of Southern California, Los Angeles, CA (California Cancer P2C), [9]National Cancer Institute, Rockville MD

Abstract

Background: PNET has long had few effective therapies other than chemotherapy. Recent placebo-controlled phase III trials of the mTOR inhibitor everolimus and the VEGF/ PDGF receptor inhibitor sunitinib noted improved progression-free survival (PFS). However, objective response rates (RR) with these agents are still <10%. Preclinical studies suggest enhanced anti-tumor effects with combined mTOR and VEGF targeted therapy.

Methods: We conducted a phase II trial of the mTOR inhibitor TEM (25 mg IV q week) and the VEGF-A monoclonal antibody BEV (10 mg/kg IV q 2 weeks) in patients (pts) with well or moderately differentiated PNET and progressive disease by RECIST within 7 months of study entry. Co-primary endpoints were RR and 6-month PFS. Planned enrollment is 50 patients, with interim analysis for futility after the first 25 evaluable pts. Pts had no prior mTOR or VEGF targeted agents, ECOG PS 0-1, and adequate hematologic and organ function. Prior or continued octreotide was allowed, but not required. Prior interferon, embolization, and ≤ 2 chemotherapy regimens were allowed.

Results: Confirmed PR was documented in 13 of the first 25 (52%) evaluable patients. 21 of 25 (84%) patients were progression-free at 6 months. Both endpoints exceeded the protocol-defined criteria to continue enrollment. For 36 evaluable patients, the most common grade 3-4 adverse events attributed to therapy were hypertension (14%), leukopenia (11%), lymphopenia (11%), hyperglycemia (11%), mucositis (8%), hypokalemia (8%), and fatigue (8%).

Conclusion: The combination of TEM/BEV has substantial activity in a multi-center phase II trial with RR of 52%, well in excess of single targeted agents in PNET. 6-month PFS was a notable 84% in a population of patients with RECIST criteria progression within 7 months of study entry. Accrual is ongoing.

Introduction

- Pancreatic neuroendocrine tumor (PNET) is often diagnosed at an advanced stage, with relentless disease progression despite current therapies. Symptoms may arise from peptide hormone production or disease bulk. Improved medical therapies are needed.
- Everolimus, an inhibitor of mTOR, improved progression free survival (PFS) vs placebo (11.0 months vs 4.6 months, $p < 0.001$) in a randomized phase III trial.[1]
- Sunitinib, a TKI targeting VEGFR-2 and 3 and PDGFRs, improved PFS vs placebo (11.4 months vs 5.5 months, $p < 0.001$), in a randomized phase III trial.[2]
- In both trials, patients had to demonstrate tumor progression over prior 12 months and were candidates for placebo.
- Objective response rates were low (< 10%) in both trials (5% for everolimus and 9% for sunitinib)
- Preclinical studies have suggested that the combination of the mTOR inhibitor rapamycin with a monoclonal antibody against VEGF is associated with enhanced antitumor effects in a pancreatic adenocarcinoma model, compared to each agent alone.[3]
- A phase I/II trial through the Mayo P2C established the safety of full doses of temsirolimus and bevacizumab in combination.[4]
- We hypothesized that the combination of mTOR targeting and VEGF pathway targeting would improve the efficacy of therapy in PNET, with non-overlapping toxicity profiles.

Methods

Patient Eligibility

- Well or moderately differentiated NET with known pancreatic primary
- **Progressive disease within previous 7 months by RECIST criteria**
- Continued concurrent octreotide at stable dose allowed if needed for symptom control
- No prior mTOR or VEGF pathway targeted therapy
- Up to 2 prior chemotherapies, prior interferon, liver-directed therapies and PRRT allowed
- Adequate hematologic reserve (Hb > 9 g/dl, ANC > 1500/mm3, Plts > 75,000/mm^3)
- Creatinine ≤ 1.5 x ULN with < 2 g/24 hour urine protein.
- Liver transaminases < 5x ULN; bilirubin < 1.5 x ULN
- Adequate hypertension, glucose and cholesterol control.
- Normal LVEF
- ECOG PS 0-1
- Willingness to submit archived tumor tissue and blood/serum samples for translational correlates.

Study Regimen

- Temsirolimus 25 mg IV days 1,8,15,22
- Bevacizumab 10 mg/kg IV days 1,15

Repeated every 28 days (1 cycle), with tumor evaluation performed every 2 cycles

Trial design

- Multi-center single arm Phase II trial
- Co-primary endpoints of confirmed response rate and 6-month PFS
- All eligible patients who have signed a consent form and have begun treatment will be evaluable for response.
- A modified two-stage Simon design with fixed sample size of 50 patients is utilized with goal RR of > 20% and/or 6 –month PFS of 80%.
- The trial was activated on 9/8/2009. Data for interim analysis were frozen 11/28/2011.
- **Planned interim efficacy analysis for futility performed after first 25 evaluable patients is presented.** Adverse event data are available for 36 patients.

Results

Patient Characteristics
First 25 Evaluable Patients

Age	
Median (Range)	58.0 (40.0-79.0)
Gender	
Female	16 (64.0%)
Male	9 (36.0%)
Performance Score	
0	11 (44.0%)
1	14 (56.0%)
Ethnicity	
Caucasian	21 (84.0%)
African American	4 (16.0%)
Months From Diagnosis to On Study	
Median (Range)	11.5 (1.0-97.5)

	Total N (%)
Prior Therapy	
Octreotide	6 (24)
1 Prior chemotherapy regimen	5 (20)
2 Prior chemotherapy regimens	4 (16)
Hepatic Artery (Chemo)Embolization	3 (12)
PRRT	1 (4)

Efficacy

- **Primary Endpoints:**
 - **Confirmed partial response rate: 52% (13/25)**
 - **6 month PFS: 84% (21/25)**

 Both co-primary endpoints exceeded the protocol-defined criteria to continue enrollment.
- Secondary Endpoints:
 - Median PFS: 13.0 months
 - Median number of treatment cycles: 12.0
- Patient Disposition:
 - 13 patients have progressed, 1 patient has died.
 - 10 patients remain on therapy.
 - 10 went off of therapy due to progressive disease .
 - 5 patients off-study due to adverse event or patient refusal

Progression Free Survival

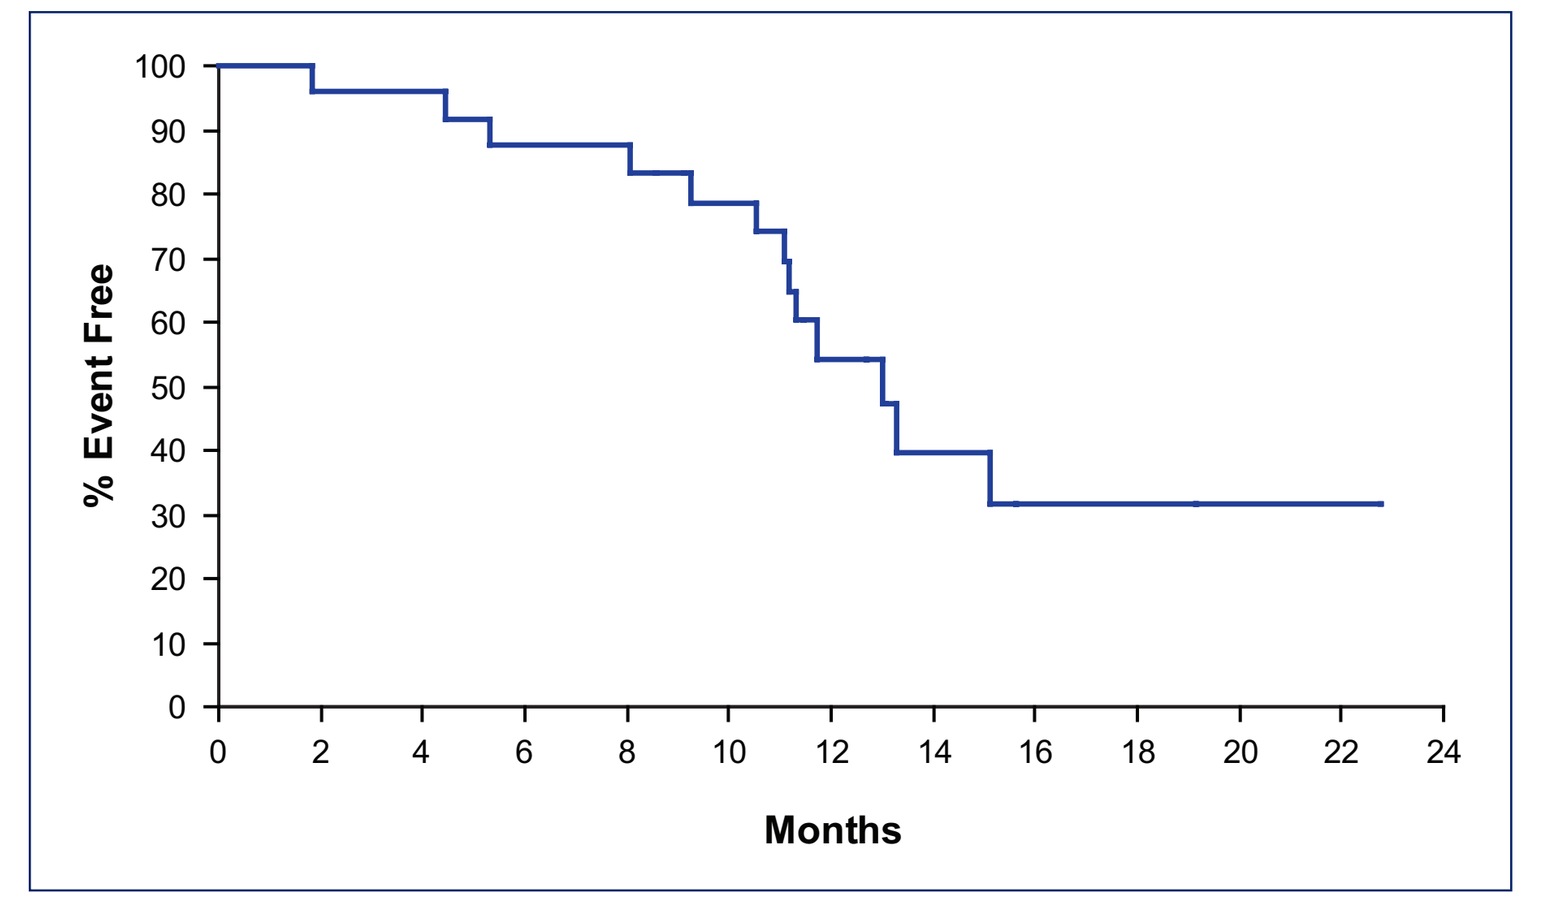
% Event Free
100
90
80
70
60
50
40
30
20
10
0
0
2
4
6
8
10
12
14
16
18
20
22
24
Months

Best Tumor Response

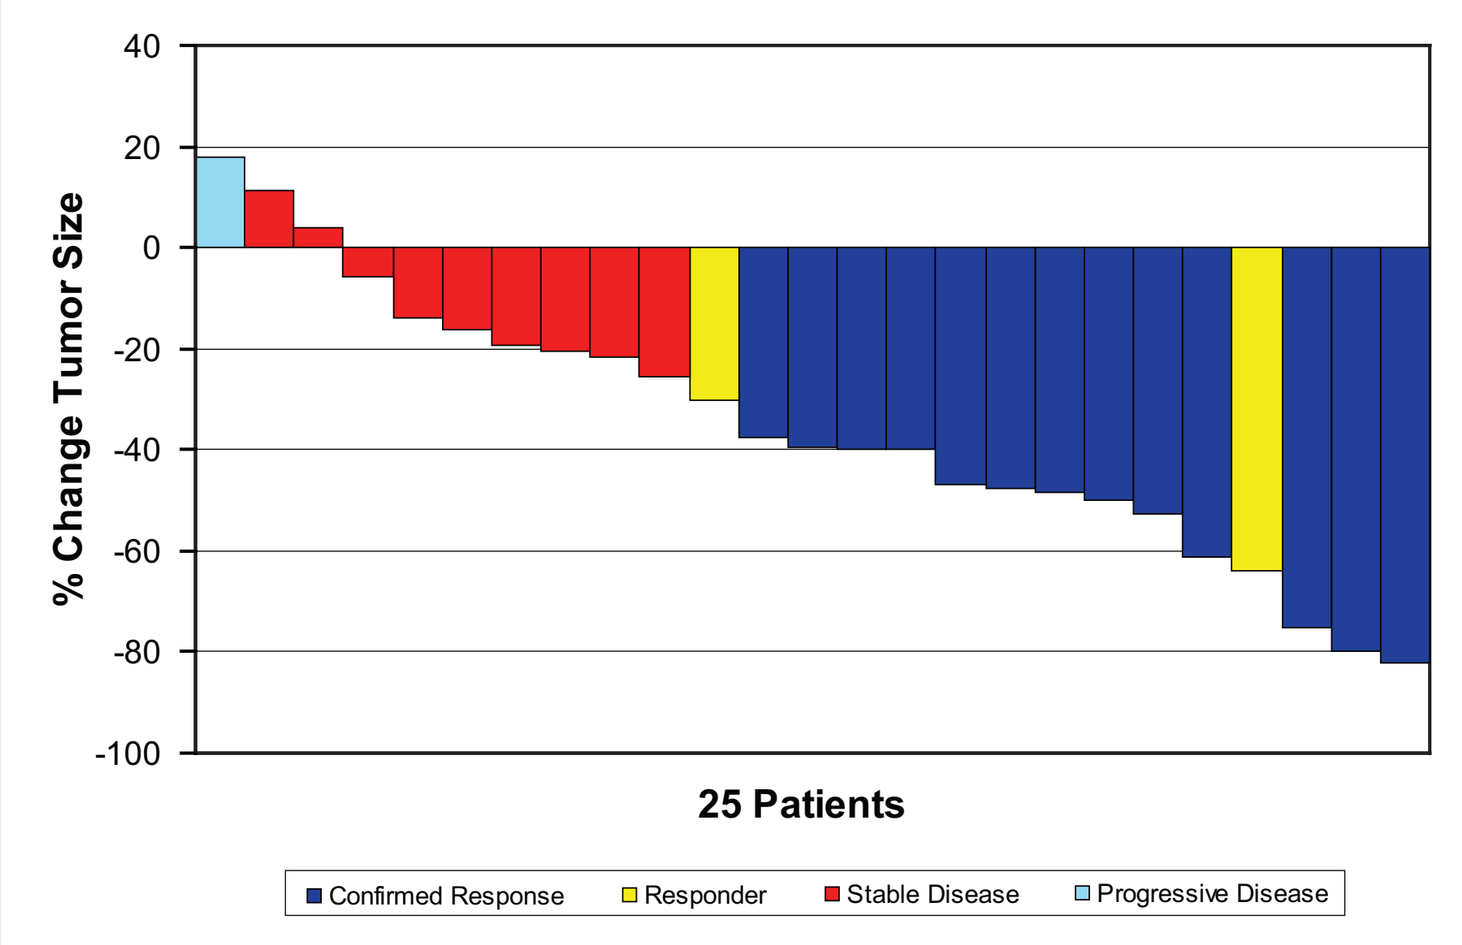
% Change Tumor Size
40
20
0
-20
-40
-60
-80
-100
25 Patients
Confirmed Response
Responder
Stable Disease
Progressive Disease

Adverse Events (N=36)*
Incidence >10% (all grades)

	Grades 1 & 2		Grades 3 & 4	
Hypertension	13	36	5	14
Hyperglycemia	13	36	4	11
Lymphopenia	8	22	4	11
Neutropenia	7	19	4	11
Fatigue	22	61	3	8
Mucositis Oral	19	53	3	8
Hypokalemia	4	11	3	8
Hypercholesterolemia	26	72	2	6
Thrombocytopenia	23	64	2	6
Aspartate Aminotransferase increased	15	42	2	6
Headache	12	33	2	6
Diarrhea	7	19	2	6
Hypophosphatemia	4	11	2	6
Anemia	24	67	1	3
Hypertriglyceridemia	23	64	1	3
Proteinuria	17	47	1	3
Alanine Aminotransferase increased	13	36	1	3
Nausea	11	31	1	3
Alkaline Phosphatase increased	9	25	1	3
Hypocalcemia	5	14	1	3
Elevated Creatinine	3	8	1	3
Anorexia	13	36	0	0
Rash	12	33	0	0
Epistaxis	9	25	0	0
Hemorrage	8	22	0	0
Hypoalbuminemia	8	22	0	0
Vomiting	7	19	0	0
Edema Limbs	6	17	0	0
Bilirubin	4	11	0	0

*Highest grade per patient possibly attributed to therapy

Conclusions

- Planned interim analysis of this phase II trial met criteria for proceeding with further accrual to goal of 50 evaluable patients. Accrual is ongoing.
- Adverse event data are consistent with those expected for each agent
- The remarkable preliminary efficacy data is encouraging and supports the ongoing study of combined targeted therapy in this disease, such as the randomized phase II trial CALGB 80701 evaluating everolimus versus the combination of everolimus and bevacizumab.
- Evaluation of this regimen in the setting of prior mTOR inhibitor and/or VEGFR TKI therapy is warranted.
- Translational investigation of mandatory tumor and blood sample submission will be performed.

References

1. Yao et al, New Eng J Med, 2011, 514-523.
2. Raymond et al, New Eng J Med 2011, 501-513.
3. Stephan, S., et al., Clin. Cancer Res., 2004; 6993-7000.
4. Merchan et al, Proc ASCO, 2007.

Supported by NCI N01 Contracts:

662205, 62203, 62208, 62209, 62206, 62204, 62207, 62201

© 2011 Mayo Foundation for Medical Education and Research