Lung Neuroendocrine Tumor (NET) US Patient-Reported Experience: Results From the First Global NET Patient Survey – A Collaboration Between the International Neuroendocrine Cancer Alliance (INCA) and Novartis

Edward Wolin,[1] Maia Sissons,[2] Grace Goldstein,[3] John Leyden,[4] Teodora Kolarova[5]

[1]University of Kentucky, Lexington, KY, USA; [2]NET Patient Foundation, Hockley Heath, United Kingdom; [3]The Carcinoid Cancer Foundation, Inc., White Plains, NY, USA; [4]The Unicorn Foundation, Mosman, NSW, Australia; [5]APOZ & Friends, Sofia, Bulgaria.

BACKGROUND

- NETs are a rare, heterogeneous group of neoplasms whose incidence has steadily risen over the past few decades[1]
- NETs originating in the lungs (lung NETs) are classified into 1 of 4 subgroups: small cell lung carcinoma (SCLC; 20%), large cell neuroendocrine carcinoma (LCNEC; 3%), typical carcinoid (TC; 2%), and atypical carcinoid (AC; 0.2%)[2,3]
- Lung NET prognosis varies by tumor subtype and corresponding degree of malignancy
 - TC is low grade, AC is intermediate grade, and SCLC and LCNEC are poorly differentiated, highly malignant tumors.[2,3] However, TC and AC are not benign; these tumors are capable of regional or distant metastasis,[4] thus negatively affecting survival[1]
- A large proportion of patients with lung NETs are asymptomatic at diagnosis, and this contributes to treatment delays and reduced survival[5]
- To date, limited data have been published on the experience of patients living with NETs,[6,7] and none have specifically evaluated the burden in patients with lung NETs
- INCA is a network of 18 independent charitable organizations and patient groups from 15 countries around the world. Novartis Pharmaceuticals Corporation collaborated with INCA on the first global survey to gather data about the NET patient experience from multiple countries, with the goals of
 - Capturing individual patient experiences of living with NETs to increase understanding of the NET journey and the needs of patients with NETs
 - Highlighting differences and similarities between countries and regions to help improve NET awareness and care
- We present data on the burden of lung NETs in patients from the United States

AIM

- To describe the burden of lung NETs through patient-reported experiences

METHODS

- From February through May 2014, patients with NETs participated in a 25-minute anonymous survey that captured the NET patient experience, including disease burden, medical resource use, and access to care
 - With the exception of certain demographic information, survey questions were close-ended; participants were provided options from which to choose
- Patients were recruited via use of flyers, Web site postings, e-mails, and social media channels of the INCA member organizations/patient advocacy groups (in 2014, INCA consisted of 17 organizations from 14 countries)
 - Extensive use of online social media sources through local/regional advocacy groups allowed recruitment of a large number of patients with this rare type of cancer
- This survey was primarily conducted online and was available in 8 languages: Bulgarian, Dutch, English, French, German, Japanese, Norwegian, and simplified Chinese
 - Paper surveys were developed in several languages and distributed by patient groups and health care providers (HCPs) to reach patients without Internet access
- Data were analyzed at global, regional, and country levels; here, we present results from the US analyses
 - Statistical differences between comparisons were significant at the 95% confidence level (P <0.05) for all statements within the text
- This survey was conducted as an equal collaboration between INCA and Novartis, and was funded by Novartis. Hall & Partners, a research organization, fielded and analyzed the results

RESULTS

Demographics

- A total of 1928 patients with NETs were recruited from >12 countries, including 758 patients (39%) from the United States (**Figure 1**)

Figure 1. Countries participating in the global NET patient survey.

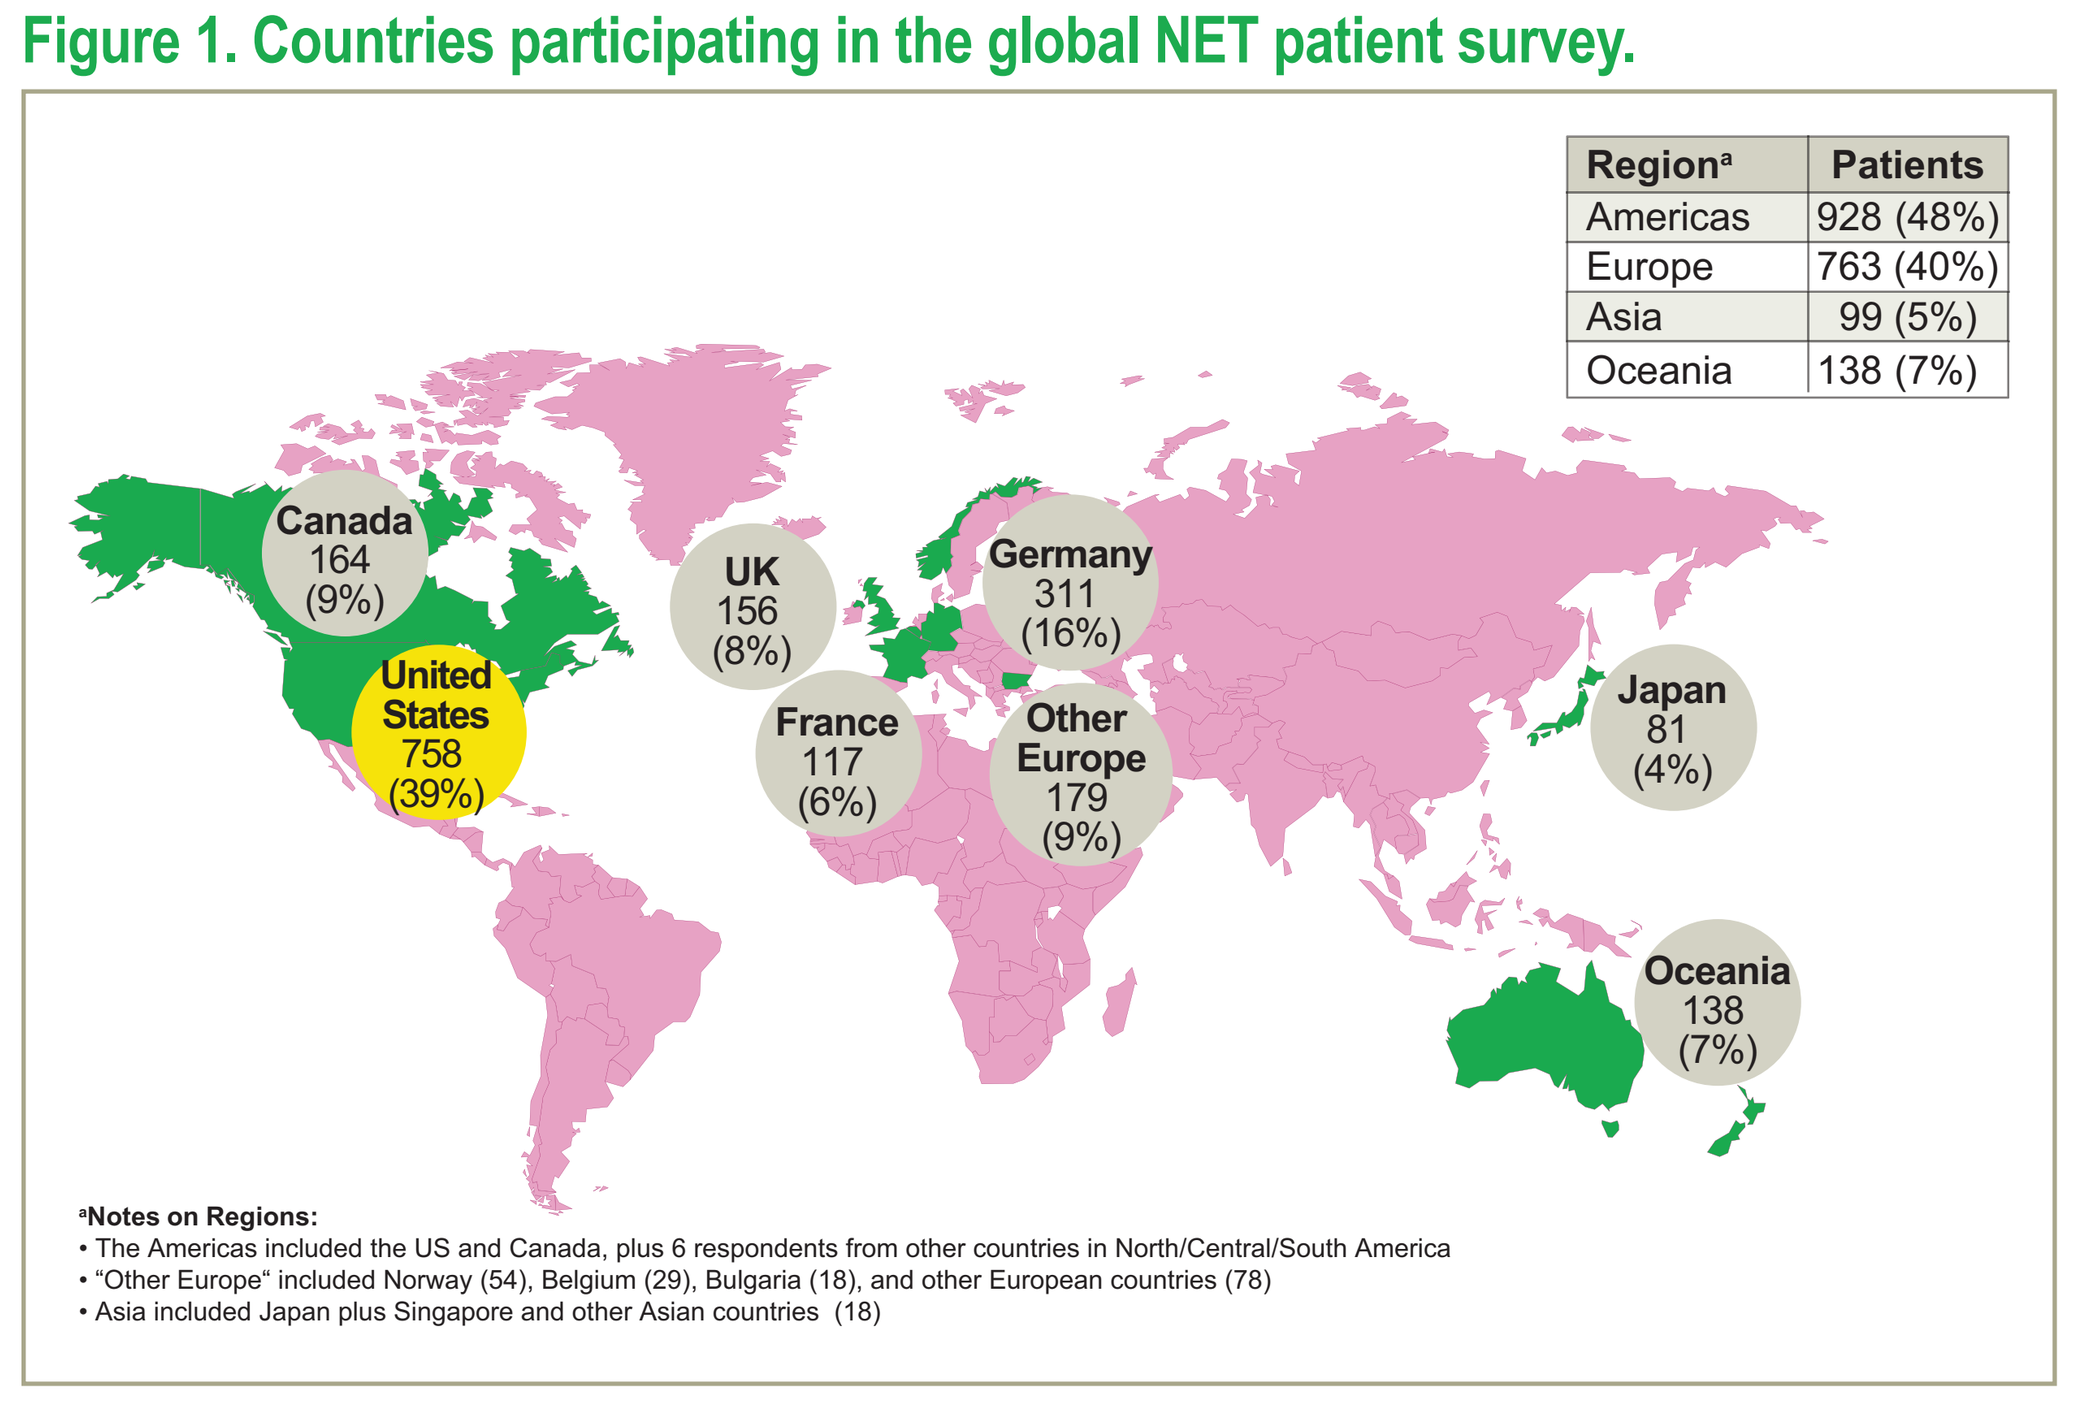
Region[a] | Patients
Americas | 928 (48%)
Europe | 763 (40%)
Asia | 99 (5%)
Oceania | 138 (7%)
Canada 164 (9%)
United States 758 (39%)
UK 156 (8%)
France 117 (6%)
Germany 311 (16%)
Other Europe 179 (9%)
Japan 81 (4%)
Oceania 138 (7%)
[a]Notes on Regions:
• The Americas included the US and Canada, plus 6 respondents from other countries in North/Central/South America
• "Other Europe" included Norway (54), Belgium (29), Bulgaria (18), and other European countries (78)
• Asia included Japan plus Singapore and other Asian countries (18)

Base population: All respondents (N = 1928).
Questions: In which country do you live? In which region do you live?

- 15% of US patients had lung NETs (n = 112)
 - Of patients who knew their status, the majority reported having nonfunctional/asymptomatic (53%, n = 79) and low-grade (G1; 57%, n = 70) lung NETs (**Table 1**)

Table 1. Patient-Reported Functional Status and Grade of NETs[a]

Tumor Type	Functional Status, n (%)			Grade, n (%)			
	Functional[b]	Nonfunctional[c]/ Asymptomatic[d]	Unknown[e]	G1[f]	G2[g]	G3[h]	Unknown[i]
Total US	363 (48)	248 (33)	147 (19)	264 (35)	127 (17)	44 (6)	323 (43)
Lung NET	37 (33)	42 (38)	33 (29)	40 (36)	17 (15)	13 (12)	42 (38)

Base population: All US respondents (n = 758), and all US respondents with lung NETs (n = 112).
Questions: How, if at all, does your physician describe the functional status of your NET? Which of the following, if any, does your physician currently use to describe the grade of your NET?
[a]Some percentages add up to 101% because of rounding.
[b]Produces symptoms caused by the secretion of hormones (eg, flushing, diarrhea, wheezing, cramping).
[c]Do not secrete hormones; however, they may elicit symptoms caused by tumor growth (eg, pain, intestinal blockage, bleeding).
[d]Experience no symptoms.
[e]Physician does not describe/discuss; I don't remember.
[f]NETs are relatively slow growing; Ki-67 index <2%.
[g]NETs have a less predictable, moderately aggressive course; Ki-67 index 3%-20%.
[h]NETs can be highly aggressive; Ki-67 index >20%.
[i]Physician did not give my NET a grade/describe/discuss; I don't remember.

Diagnosis and Management of Lung NETs

- For approximately one-third of the patients surveyed (31%), >5 years elapsed from symptom onset to a diagnosis of lung NET (**Figure 2**)
 - Almost half of all patients (49%) were diagnosed with other conditions prior to receiving a diagnosis of lung NET, with asthma (56%) and pneumonia (35%) the most common initial diagnoses (**Figure 3**)
- The majority of patients had not visited a NET specialist center (64%)
 - Of those who had, only 13% were diagnosed there

Figure 2. Time between first symptom onset and diagnosis.

Diagnosed with NET in the following time period	Don't know/ NA	<6 months	6 months–2 years	2–5 years	>5 years	Mean (months)
Total US	8%	18%	17%	19%	34%	59.0
Lung NET	8%	24%	12%	21%	31%	58.2

NA, not applicable.
Base population: All US respondents (n = 758), and all US respondents with lung NETs (n = 112).
Question: What was the approximate length of time between your first symptom and your NET diagnosis?

Figure 3. Diagnosis received before NET diagnosis.[a]

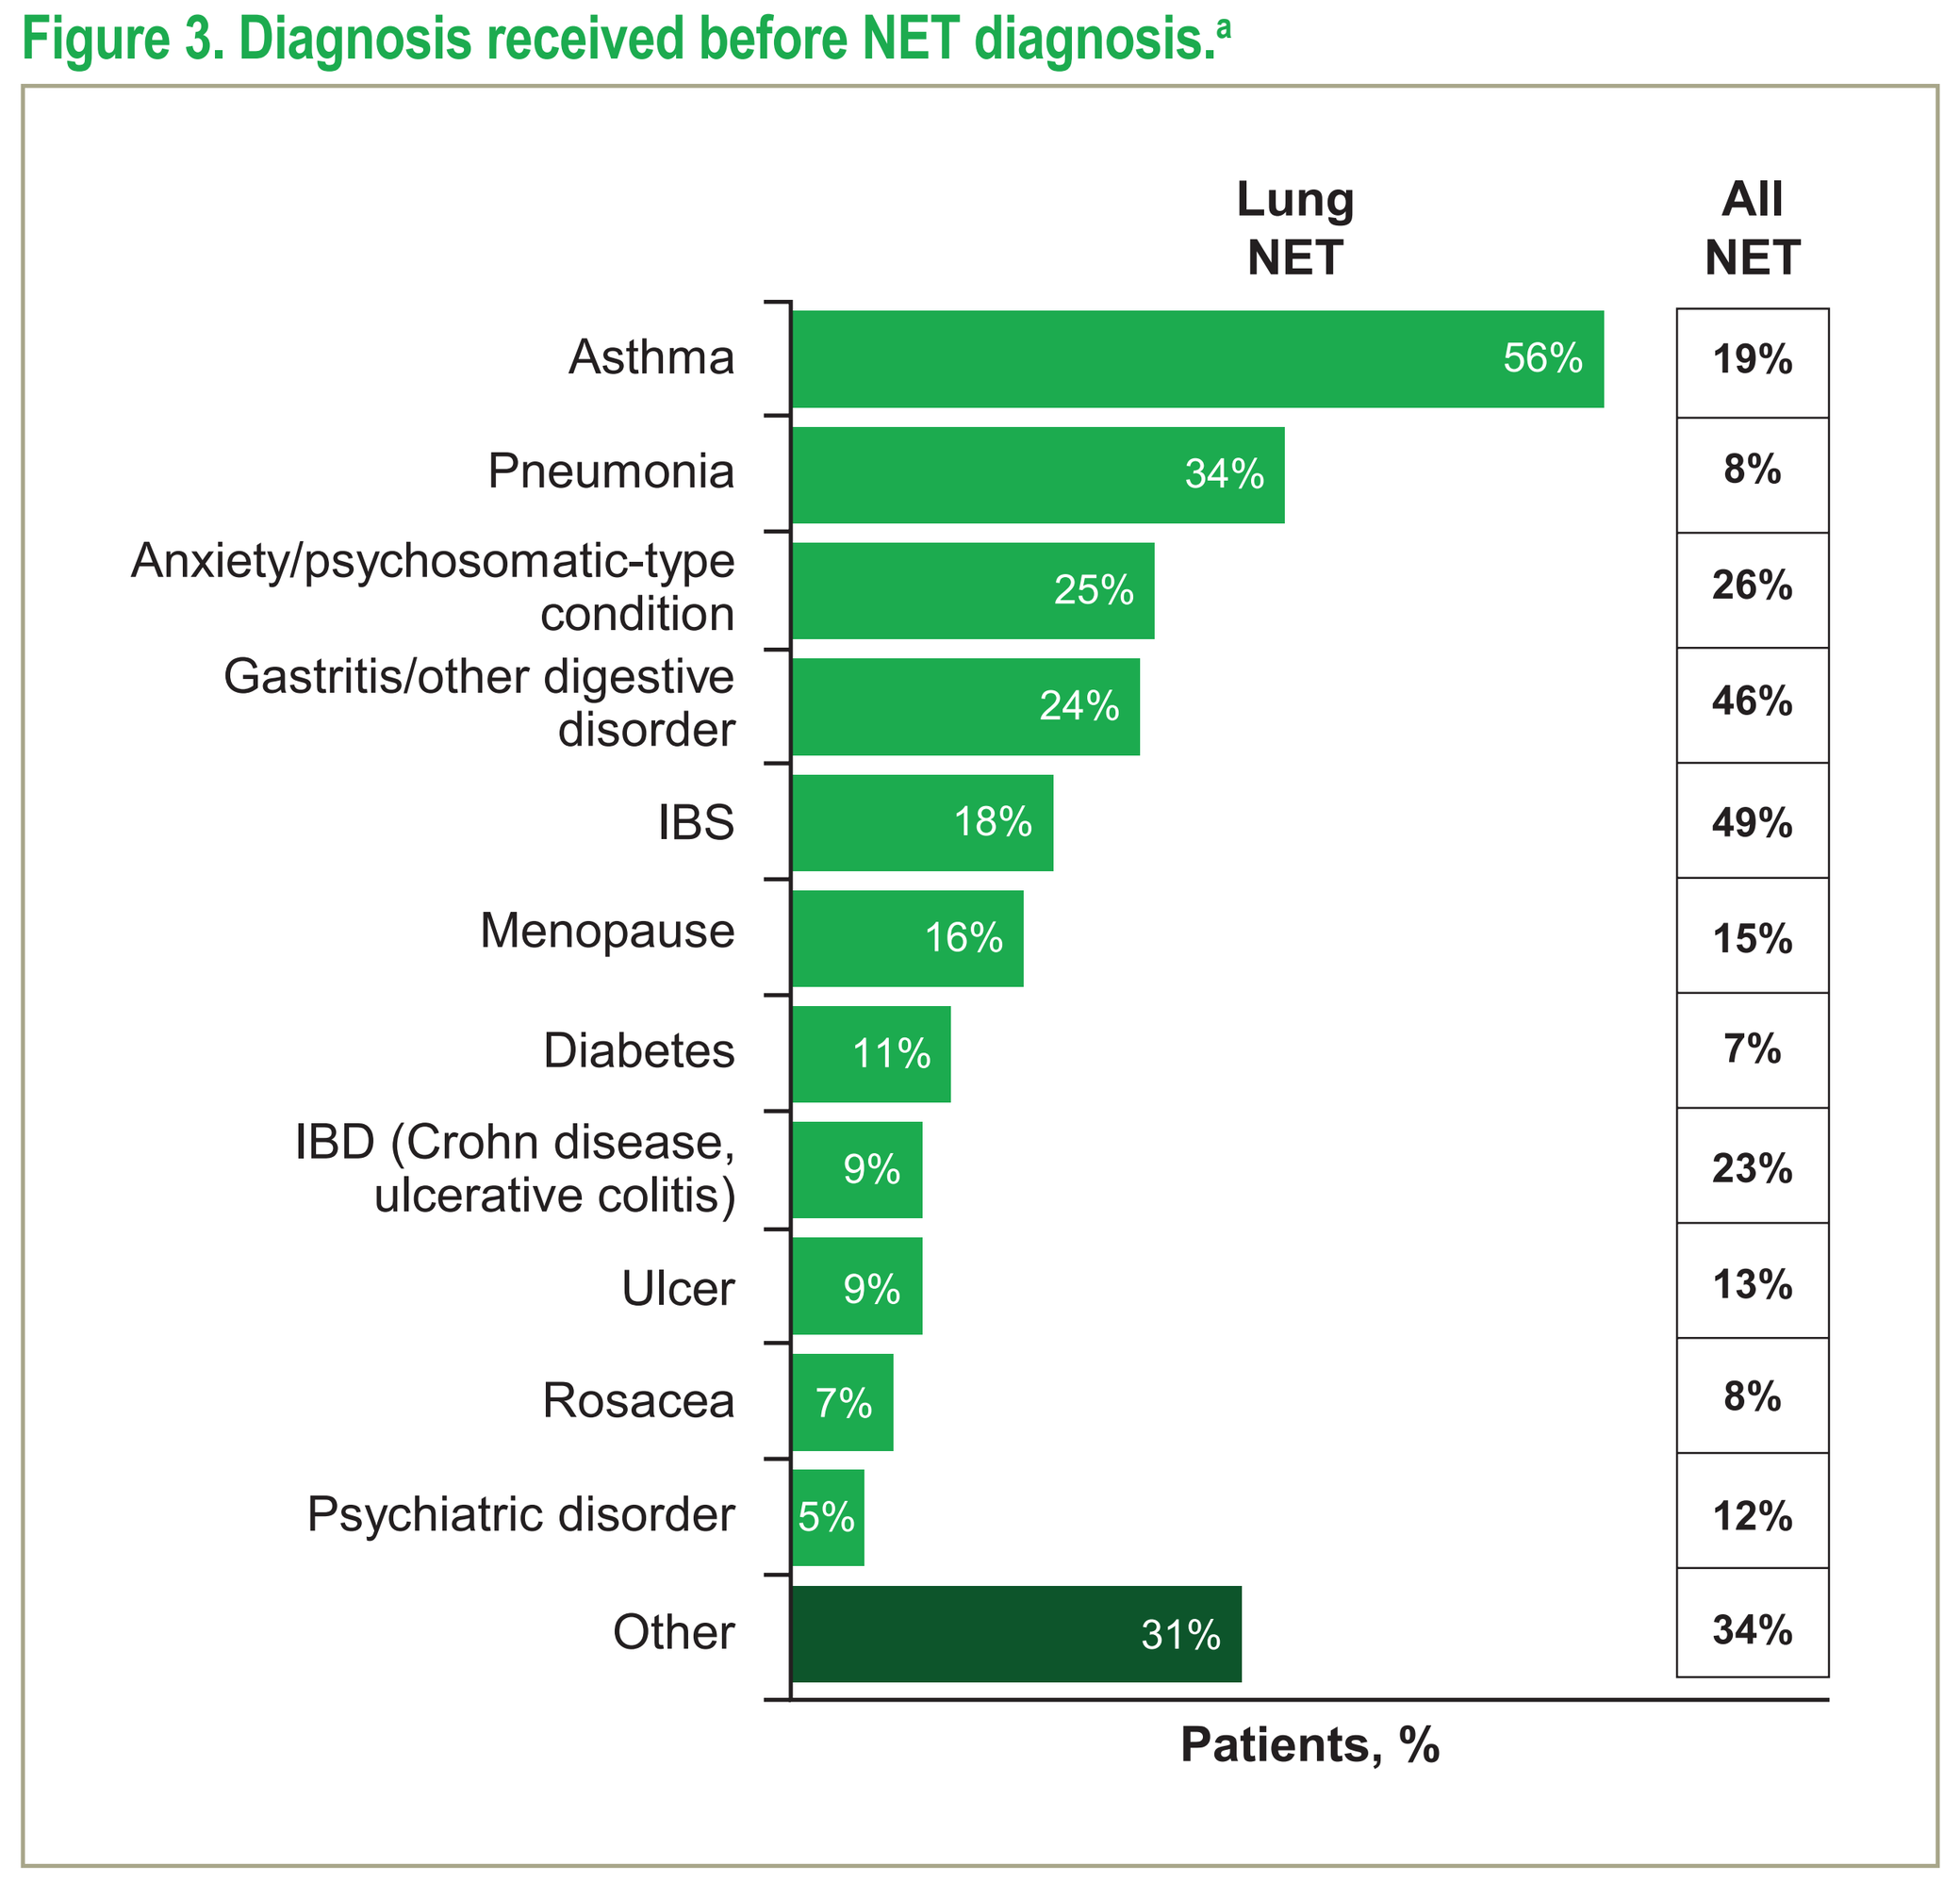
Lung NET
All NET
Asthma 56% 19%
Pneumonia 34% 8%
Anxiety/psychosomatic-type condition 25% 26%
Gastritis/other digestive disorder 24% 46%
IBS 18% 49%
Menopause 16% 15%
Diabetes 11% 7%
IBD (Crohn disease, ulcerative colitis) 9% 23%
Ulcer 9% 13%
Rosacea 7% 8%
Psychiatric disorder 5% 12%
Other 31% 34%
Patients, %

IBD, inflammatory bowel disease; IBS, irritable bowel syndrome.
Base population: All US respondents who were diagnosed with other conditions prior to lung NET (n = 55), and all US respondents diagnosed with other conditions prior to NET (n = 368).
Question: Which of the following conditions were you initially diagnosed with prior to receiving a lung NET diagnosis?
[a]Only includes conditions mentioned by ≥5% of patients.

- Patients with lung NETs were more likely to be given the impression that their NET was curable (32%, vs 9% and 14% for gastrointestinal [GI] NET and pancreatic [p]NET, respectively) and not something to worry about (40%, vs 23% and 14% for GI NET and pNET, respectively)

Impact of NETs on Quality of Life and Work Life

- 67% of patients with lung NETs experienced a moderate to large negative impact on their quality of life (**Figure 4**)
 - This finding was similar to patients with GI NETs and pNETs (73% and 74%, respectively)
- Of patients with lung NETs who were still working at the time of the survey, 31% reported having to stop working temporarily (**Figure 5**)
 - This rate was higher than for the overall patient population (16%), GI NETs (12%), and pNETs (12%)

Figure 4. Negative impact of NETs on different aspects of life.

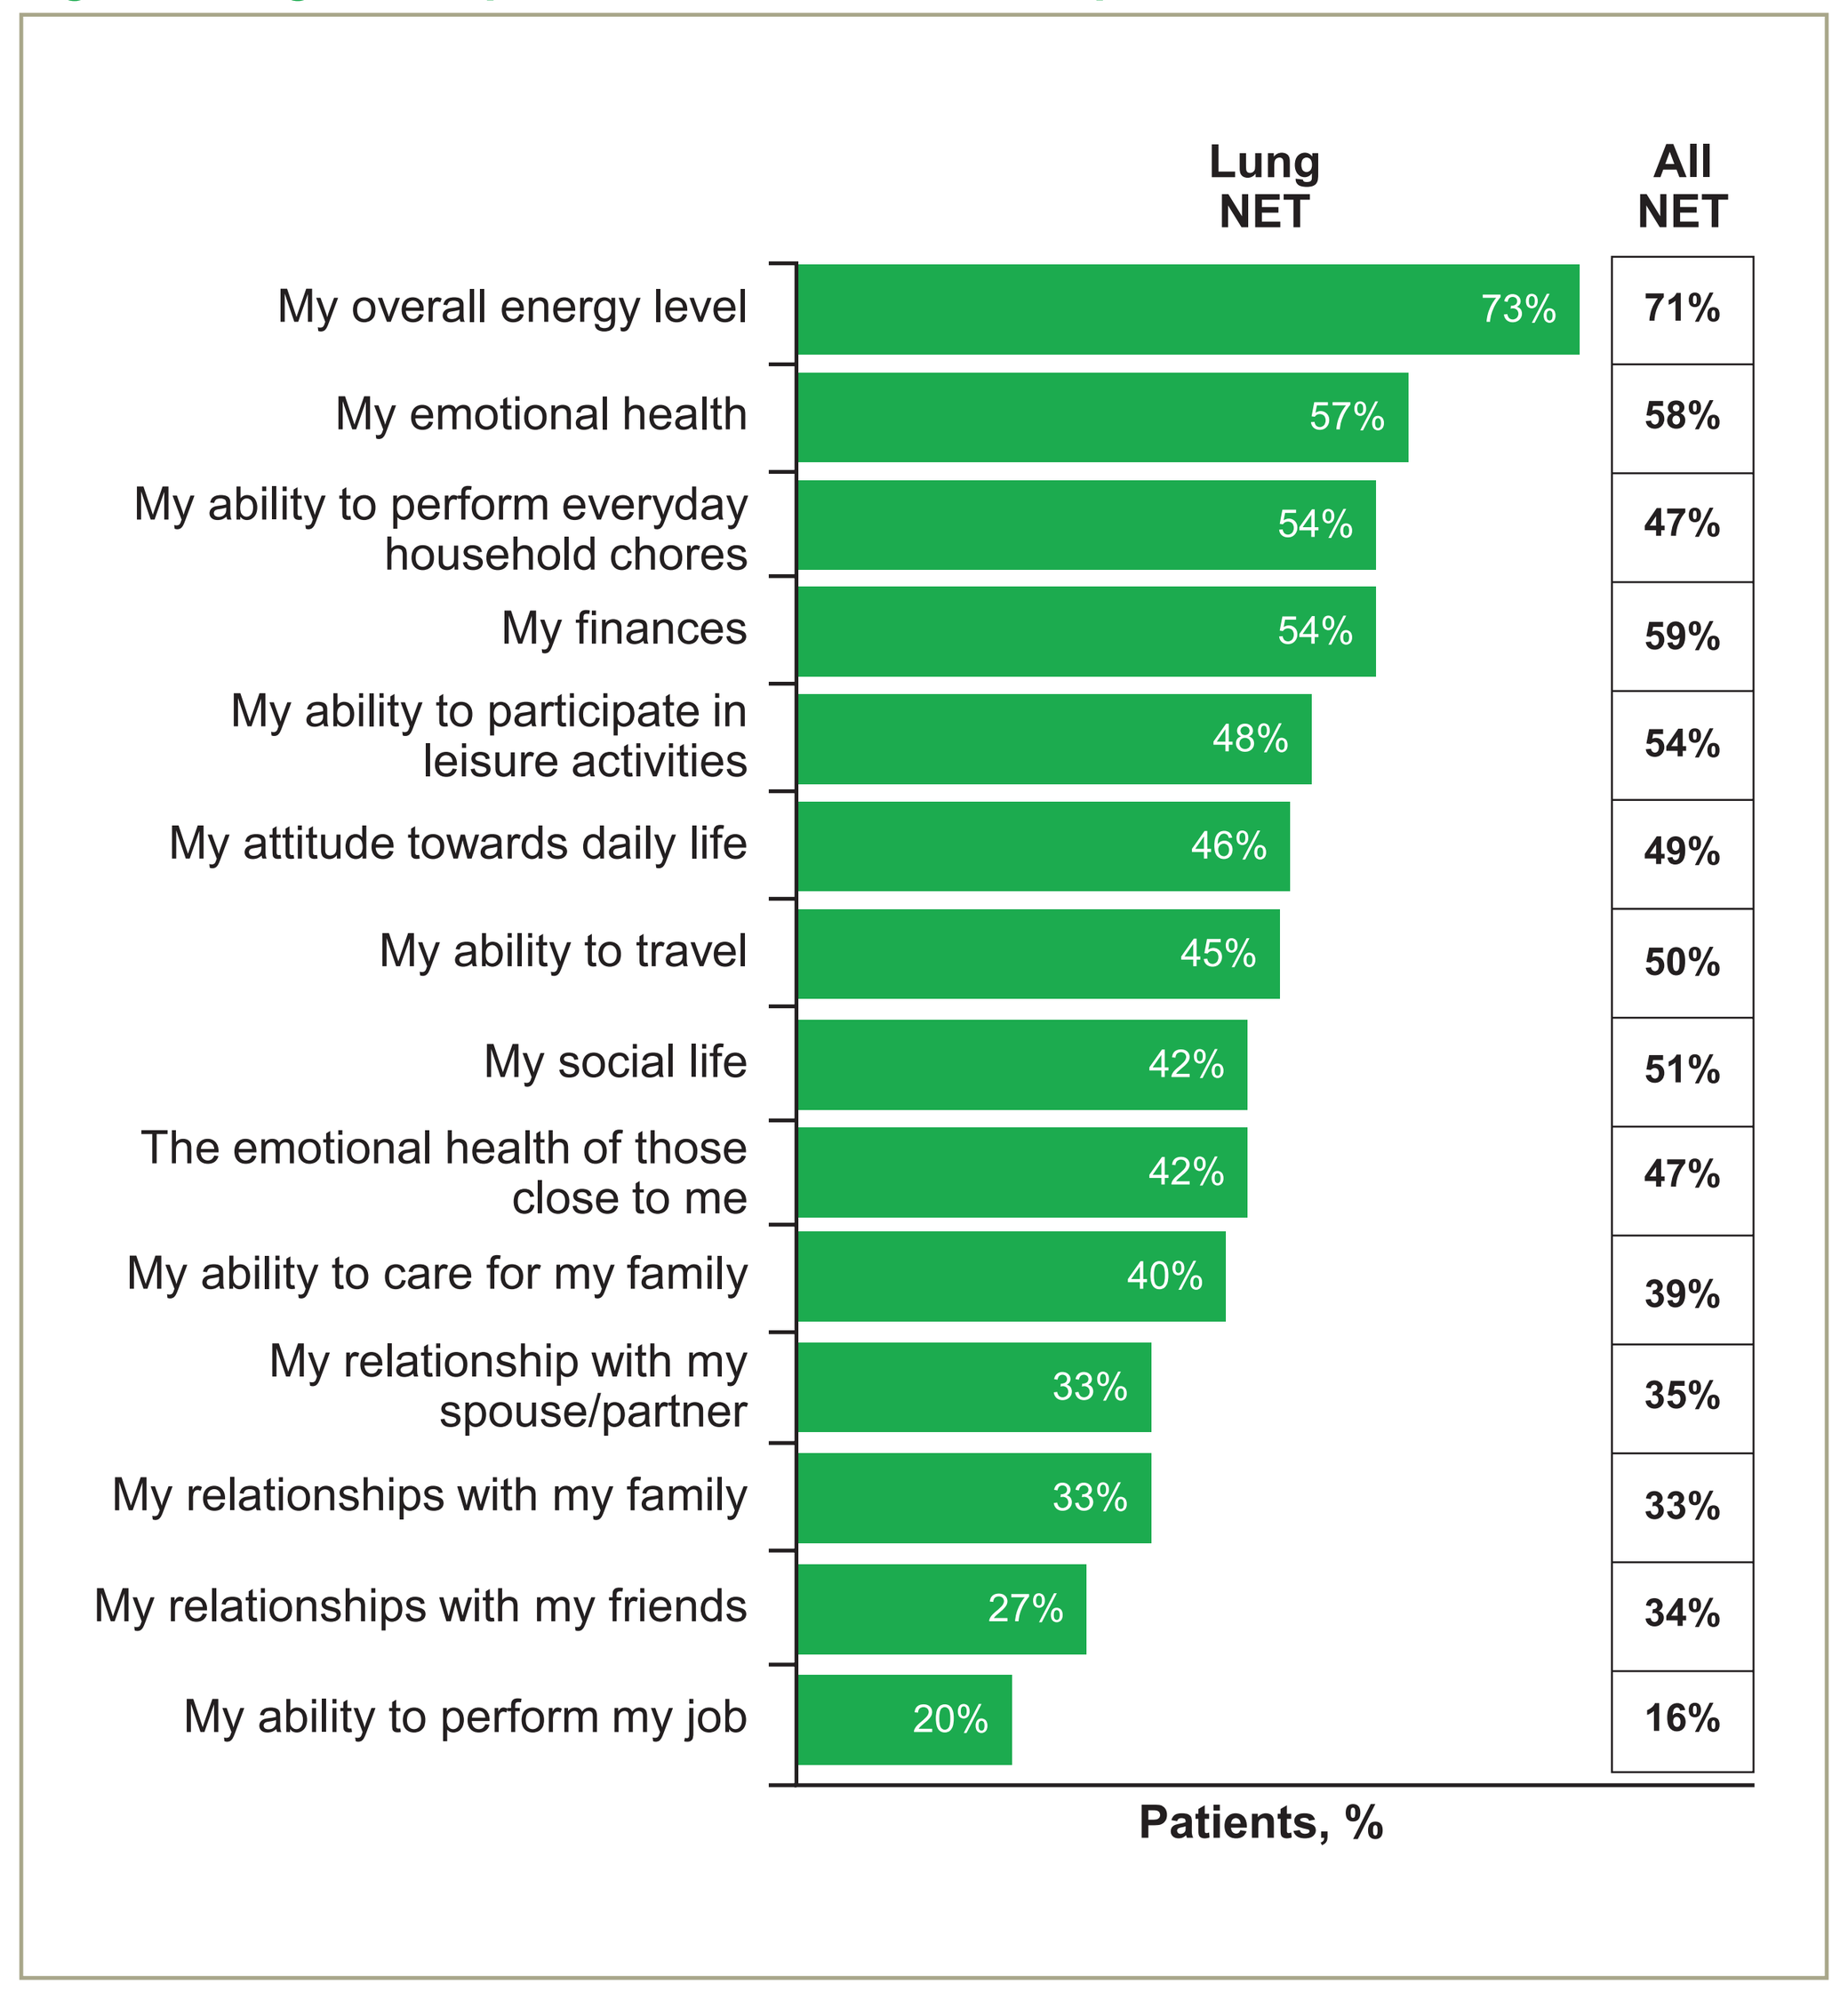
Lung NET
All NET
My overall energy level 73% 71%
My emotional health 57% 58%
My ability to perform everyday household chores 54% 47%
My finances 54% 59%
My ability to participate in leisure activities 48% 54%
My attitude towards daily life 46% 49%
My ability to travel 45% 50%
My social life 42% 51%
The emotional health of those close to me 42% 47%
My ability to care for my family 40% 39%
My relationship with my spouse/partner 33% 35%
My relationships with my family 33% 33%
My relationships with my friends 27% 34%
My ability to perform my job 20% 16%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (n = 316).
Question: How much has each of the following areas of your life been negatively affected, if at all, by your NET?
Top 2 responses shown (a moderate amount/a lot).

Figure 5. Impact of NETs on work life.

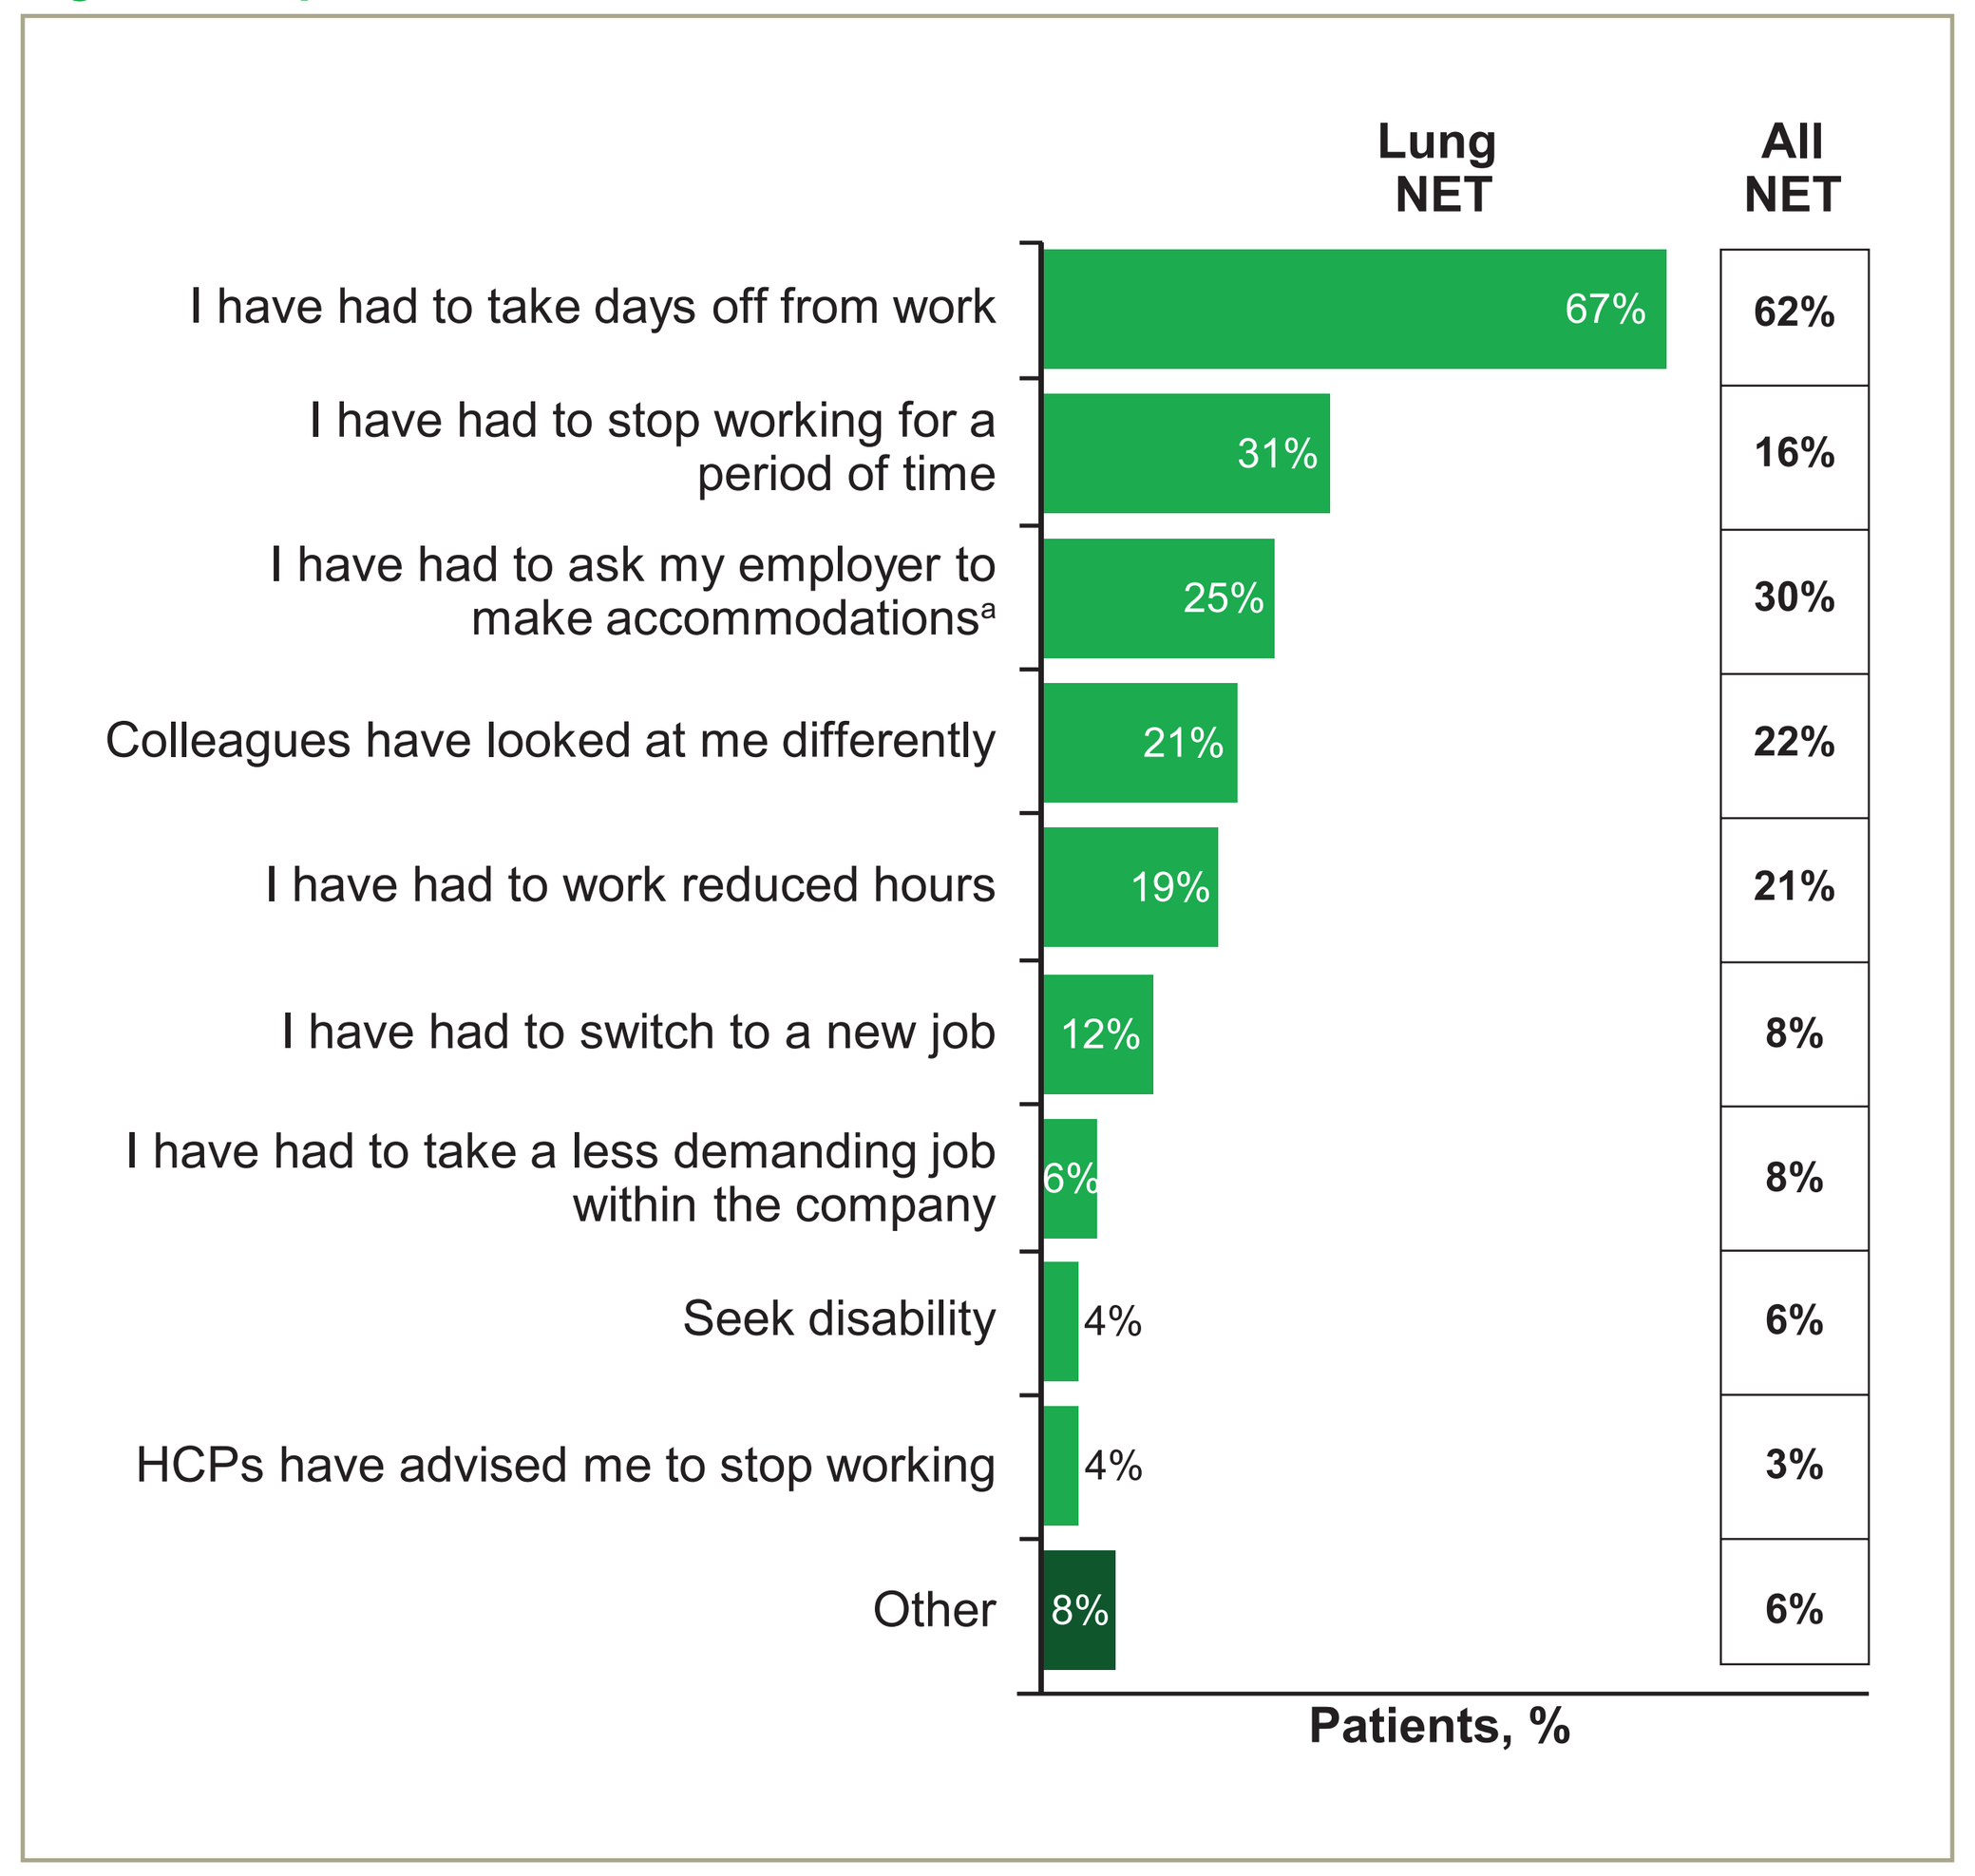
Lung NET
All NET
I have had to take days off from work 67% 62%
I have had to stop working for a period of time 31% 16%
I have had to ask my employer to make accommodations[a] 25% 30%
Colleagues have looked at me differently 21% 22%
I have had to work reduced hours 19% 21%
I have had to switch to a new job 12% 8%
I have had to take a less demanding job within the company 6% 8%
Seek disability 4% 6%
HCPs have advised me to stop working 4% 3%
Other 8% 6%
Patients, %

Base population: All US respondents with lung NETs who were working full time/part time or self-employed (n = 52), and all US respondents who were working full time/part time or self-employed with NETs (N = 316).
Question: Has your NET impacted you at work in any of the following ways? Please select all that apply.
[a]Make accommodations: eg, flexible work schedule, work from home, adaptive devices, opportunities for rest.

Key Improvements Patients Believe Would Help Them Live Better with NETs

- Overall, patients with lung NETs had more negative feelings regarding health care provider expertise/knowledge and quality of care compared with the overall NET population (**Figure 6**)
 - Lung NET patients were more likely to desire better access to NET experts (56%), better access to NET-specific medical treatments (54%; not significant), and a more knowledgeable health care team (52%)
- When asked what would have helped the patient have a better experience following a NET diagnosis, 71% of lung NET patients expressed the need for more immediate access to NET specialists, and 45% desired transfers to NET centers of expertise

Figure 6. Improvements patients believed would help them live better with NETs.

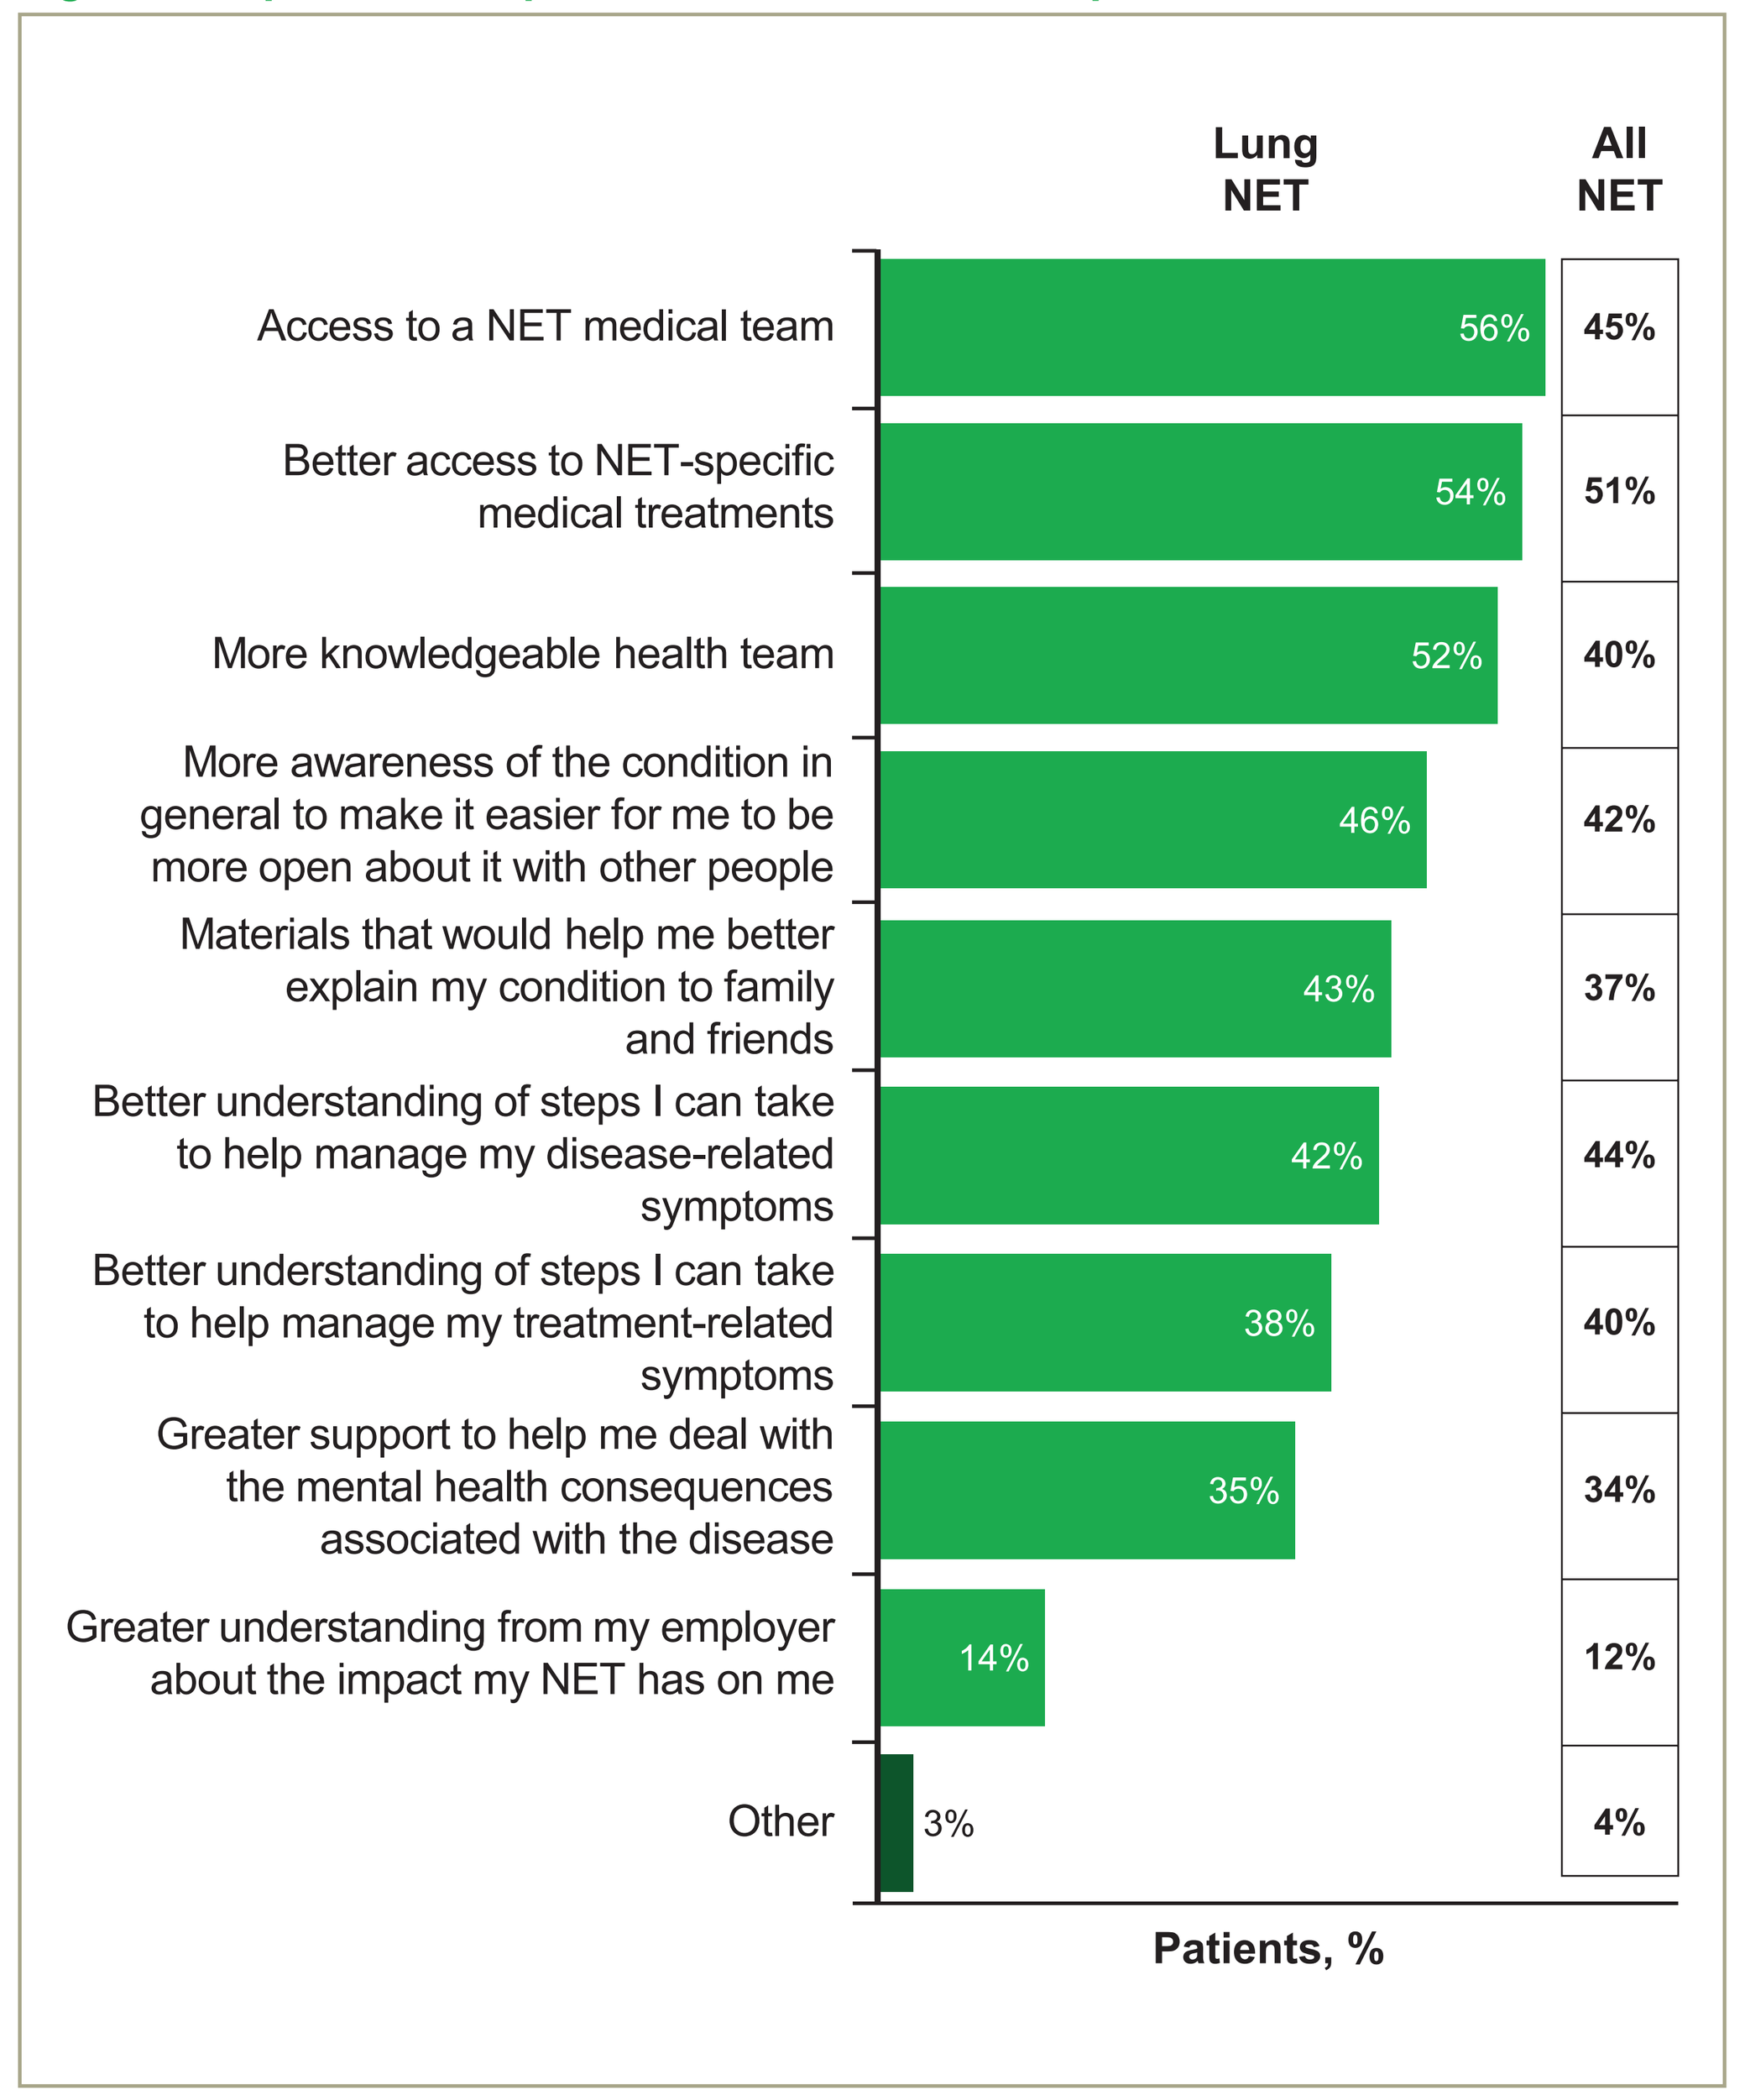
Lung NET
All NET
Access to a NET medical team 56% 45%
Better access to NET-specific medical treatments 54% 51%
More knowledgeable health team 52% 40%
More awareness of the condition in general to make it easier for me to be more open about it with other people 46% 42%
Materials that would help me better explain my condition to family and friends 43% 37%
Better understanding of steps I can take to help manage my disease-related symptoms 42% 44%
Better understanding of steps I can take to help manage my treatment-related symptoms 38% 40%
Greater support to help me deal with the mental health consequences associated with the disease 35% 34%
Greater understanding from my employer about the impact my NET has on me 14% 12%
Other 3% 4%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (N = 758).
Question: Which of the following, if any, would help you living with a NET?

Patient Perception of NET Management and Treatment

- Following diagnosis, fewer lung NET patients were satisfied that they received answers to questions (43%) or sufficient information (33%) compared with GI/pNET patients (62%/59% and 48%/48%, respectively) (**Figure 7**)

Figure 7. Patient perception of NET management.

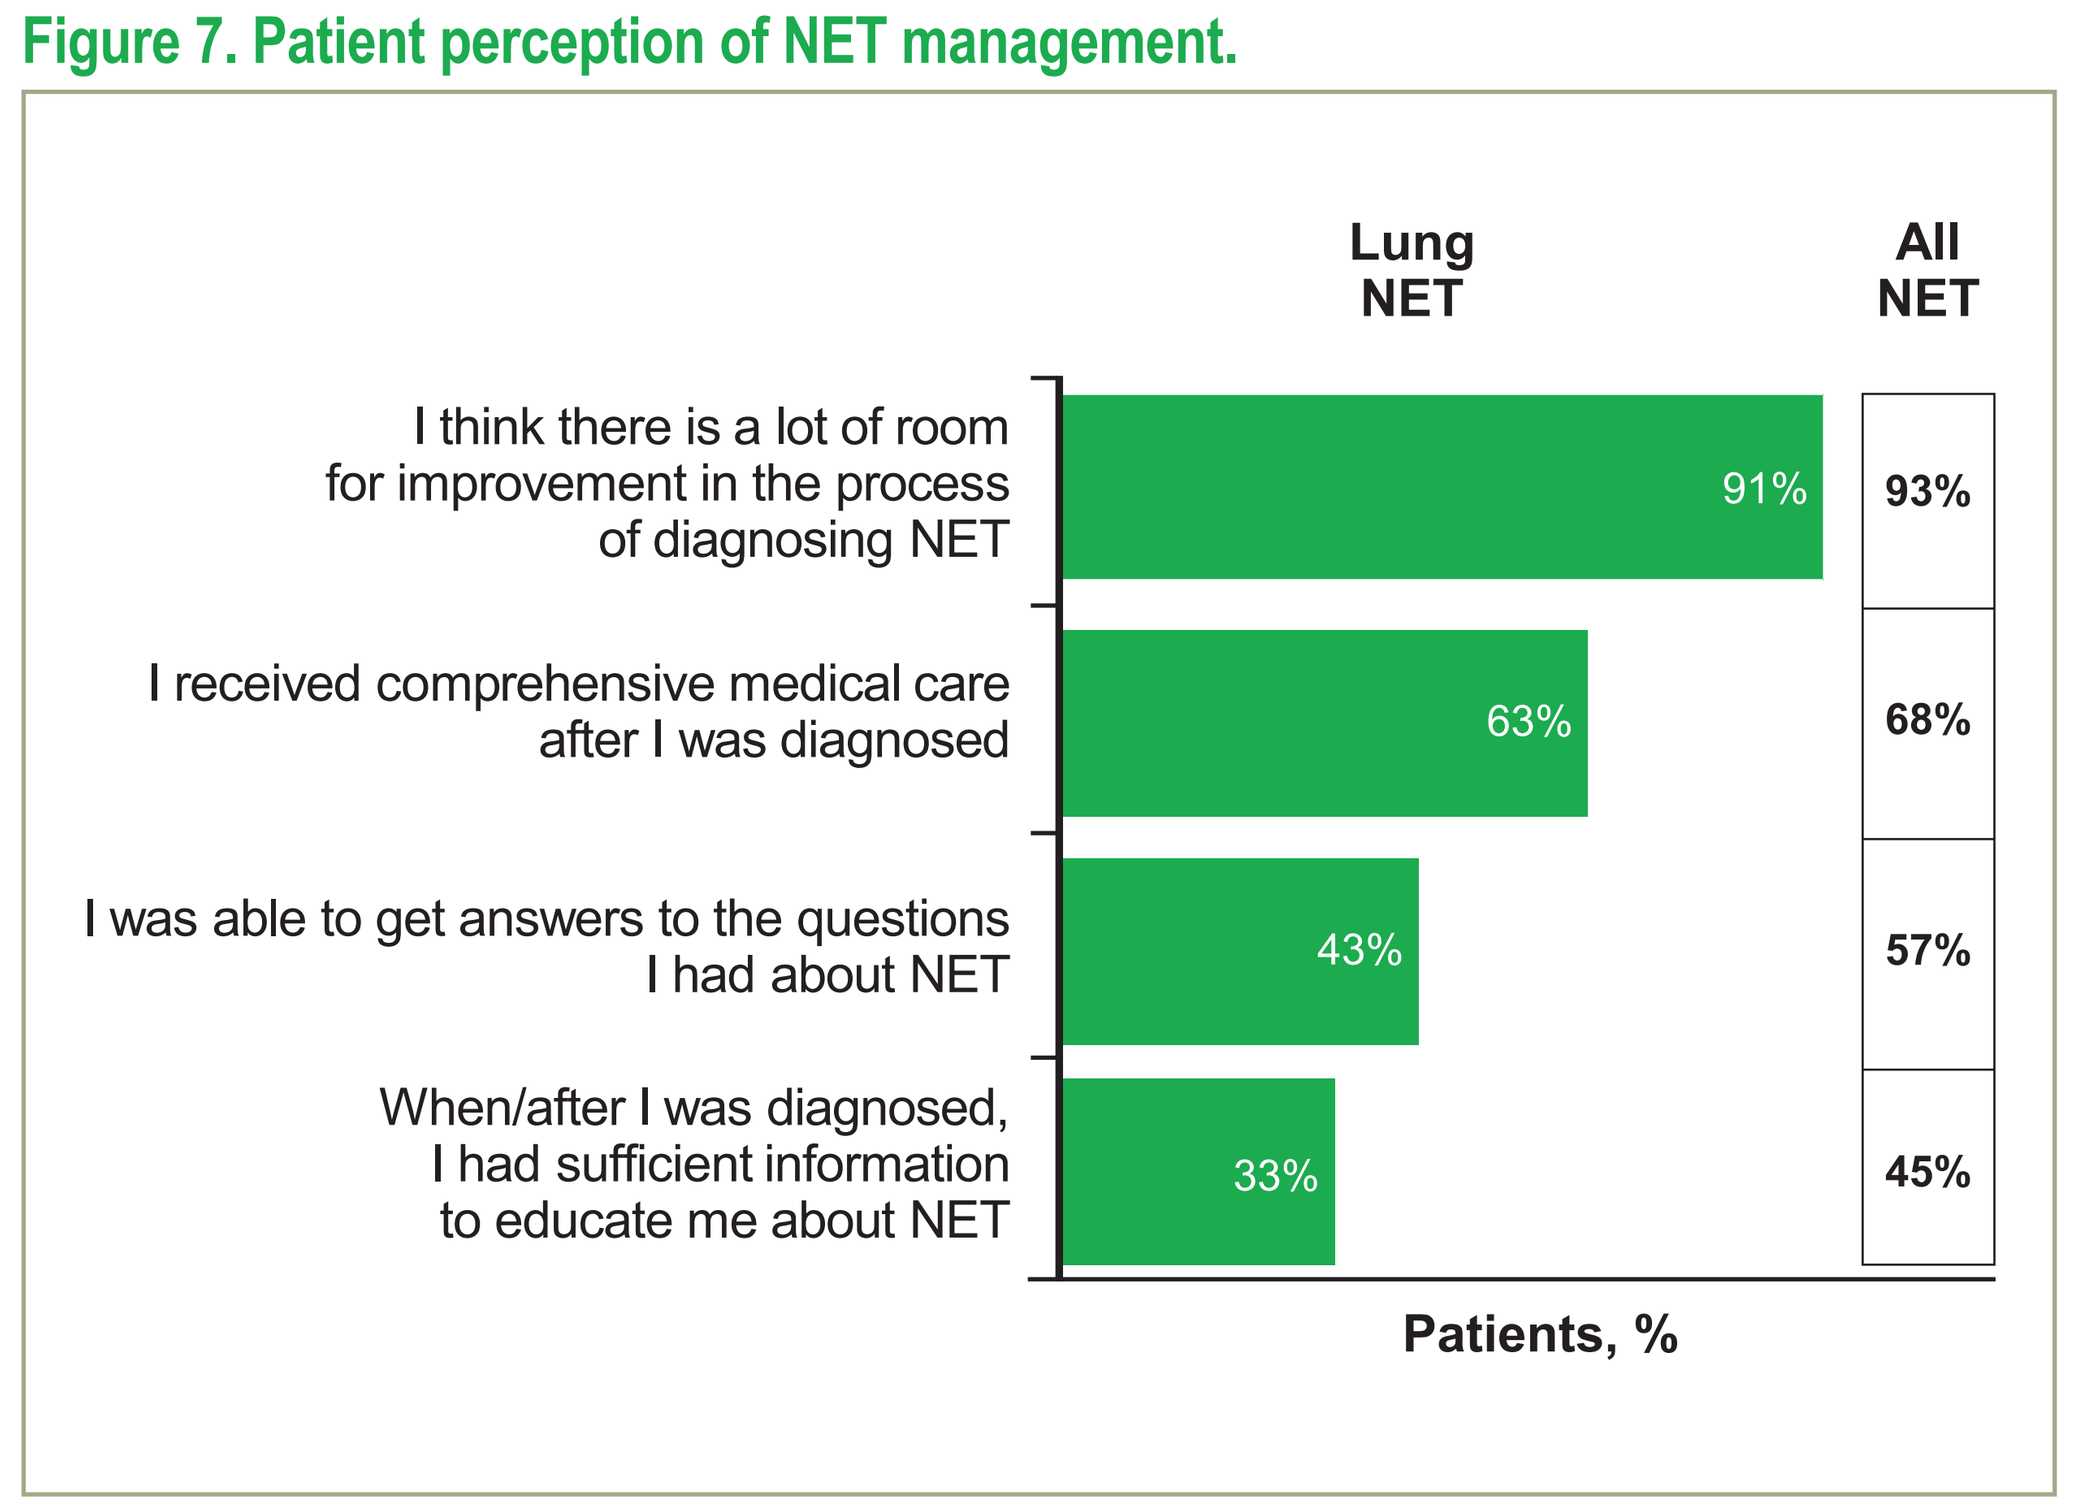
Lung NET
All NET
I think there is a lot of room for improvement in the process of diagnosing NET 91% 93%
I received comprehensive medical care after I was diagnosed 63% 68%
I was able to get answers to the questions I had about NET 43% 57%
When/after I was diagnosed, I had sufficient information to educate me about NET 33% 45%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (N = 758).
Question: To what extent do you agree or disagree with the following statements?
Top 2 responses shown (a moderate amount/ a lot).

- Almost half (45%) of lung NET patients regarded the quality of available treatments to be poor or very poor, compared with 29% of GI NET patients (significant), and 33% of pNET patients (nonsignificant)

LIMITATIONS

- This large NET patient survey had several important limitations that may have impacted results:
 - A patient-reported design was employed without independent verification, leading to potential recall bias
 - This survey did not use standardized, validated quality-of-life assessments
 - Recruitment in the United States was conducted primarily through patient advocacy groups (20%) and online sources (72%), which may have resulted in a potentially biased sample not fully representative of the heterogeneous US NET patient population
 - Respondents were more likely to be highly engaged and motivated care seekers, including female patients and/or those with a poorer prognosis

CONCLUSIONS

- Findings from this large survey provide valuable information on the experience of patients with lung NETs
 - Almost one-third of patients were diagnosed >5 years after symptom presentation
 - The majority of patients experienced a moderate to large negative impact on their daily life, and many report a negative impact on work life
 - Patients with lung NETs were more likely than those with other NET types to have negative feelings regarding various aspects of their care
 - Most patients indicated that improvements should be made in diagnosis, and nearly half indicated that the quality of available treatments was poor or very poor
- A number of suggestions for improving the diagnosis and management of lung NETs were identified, including
 - Better access to health care professionals with NET expertise
 - Immediate transfer to centers of expertise following diagnosis

REFERENCES

1. Yao JC et al. *J Clin Oncol*. 2008;26:3063-3072.
2. Travis WD et al. *J Thorac Oncol*. 2015;10:1243-1260.
3. Rekhtman N. *Arch Pathol Lab Med*. 2010;134:1628-1638.
4. Filosso PL et al. *J Thorac Dis*. 2015;7(suppl 2):S163-S171.
5. Travis WD. *Ann Oncol*. 2010;21(suppl 7):vii65-vii71.
6. Griffiths J et al. *Eur J Oncol Nurs*. 2007;11:434-441.
7. Feinberg Y et al. *Eur J Oncol Nurs*. 2013;17:541-545.

ACKNOWLEDGMENTS

We thank the patients who participated in this survey and our patient advocacy partners in the United States who contributed to this project, The Carcinoid Cancer Foundation and Caring for Carcinoid Foundation

Medical editorial assistance for this poster was provided by the ApotheCom Novartis Oncology team (Yardley, PA) and was funded by Novartis Pharmaceuticals Corporation

Mobile Friendly e-Prints 3 ways to instantly download an electronic copy of this poster to your mobile device or e-mail a copy to your computer or tablet

Scan this QR code

Text Message (SMS)
Text: **Q4d167**
to: 8NOVA (86682) *US Only*
+18324604729 *North, Central and South Americas; Caribbean; China*
+447860024038 *UK, Europe & Russia*
+46737494608 *Sweden, Europe*

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=4d167

Standard data or message rates may apply.

Presented at: North American Neuroendocrine Tumor Society (NANETS) 8th Annual Symposium; October 15–17, 2015; Austin, Texas

Novartis Pharmaceuticals Corporation provided funding for the survey.
© 2015 Novartis Pharmaceuticals Corporation, used under license by INCA and its member organizations.

BACKGROUND

- NETs are a rare, heterogeneous group of neoplasms whose incidence has steadily risen over the past few decades[1]
- NETs originating in the lungs (lung NETs) are classified into 1 of 4 subgroups: small cell lung carcinoma (SCLC; 20%), large cell neuroendocrine carcinoma (LCNEC; 3%), typical carcinoid (TC; 2%), and atypical carcinoid (AC; 0.2%)[2,3]
- Lung NET prognosis varies by tumor subtype and corresponding degree of malignancy
 - TC is low grade, AC is intermediate grade, and SCLC and LCNEC are poorly differentiated, highly malignant tumors.[2,3] However, TC and AC are not benign; these tumors are capable of regional or distant metastasis,[4] thus negatively affecting survival[1]
- A large proportion of patients with lung NETs are asymptomatic at diagnosis, and this contributes to treatment delays and reduced survival[5]
- To date, limited data have been published on the experience of patients living with NETs,[6,7] and none have specifically evaluated the burden in patients with lung NETs
- INCA is a network of 18 independent charitable organizations and patient groups from 15 countries around the world. Novartis Pharmaceuticals Corporation collaborated with INCA on the first global survey to gather data about the NET patient experience from multiple countries, with the goals of
 - Capturing individual patient experiences of living with NETs to increase understanding of the NET journey and the needs of patients with NETs
 - Highlighting differences and similarities between countries and regions to help improve NET awareness and care
- We present data on the burden of lung NETs in patients from the United States

AIM

- To describe the burden of lung NETs through patient-reported experiences

METHODS

- From February through May 2014, patients with NETs participated in a 25-minute anonymous survey that captured the NET patient experience, including disease burden, medical resource use, and access to care
 - With the exception of certain demographic information, survey questions were close-ended; participants were provided options from which to choose
- Patients were recruited via use of flyers, Web site postings, e-mails, and social media channels of the INCA member organizations/patient advocacy groups (in 2014, INCA consisted of 17 organizations from 14 countries)
 - Extensive use of online social media sources through local/regional advocacy groups allowed recruitment of a large number of patients with this rare type of cancer
- This survey was primarily conducted online and was available in 8 languages: Bulgarian, Dutch, English, French, German, Japanese, Norwegian, and simplified Chinese
 - Paper surveys were developed in several languages and distributed by patient groups and health care providers (HCPs) to reach patients without Internet access
- Data were analyzed at global, regional, and country levels; here, we present results from the US analyses
 - Statistical differences between comparisons were significant at the 95% confidence level (P <0.05) for all statements within the text
- This survey was conducted as an equal collaboration between INCA and Novartis, and was funded by Novartis. Hall & Partners, a research organization, fielded and analyzed the results

RESULTS

Demographics

- A total of 1928 patients with NETs were recruited from >12 countries, including 758 patients (39%) from the United States (**Figure 1**)

Figure 1. Countries participating in the global NET patient survey.

Region[a]	Patients
Americas	928 (48%)
Europe	763 (40%)
Asia	99 (5%)
Oceania	138 (7%)

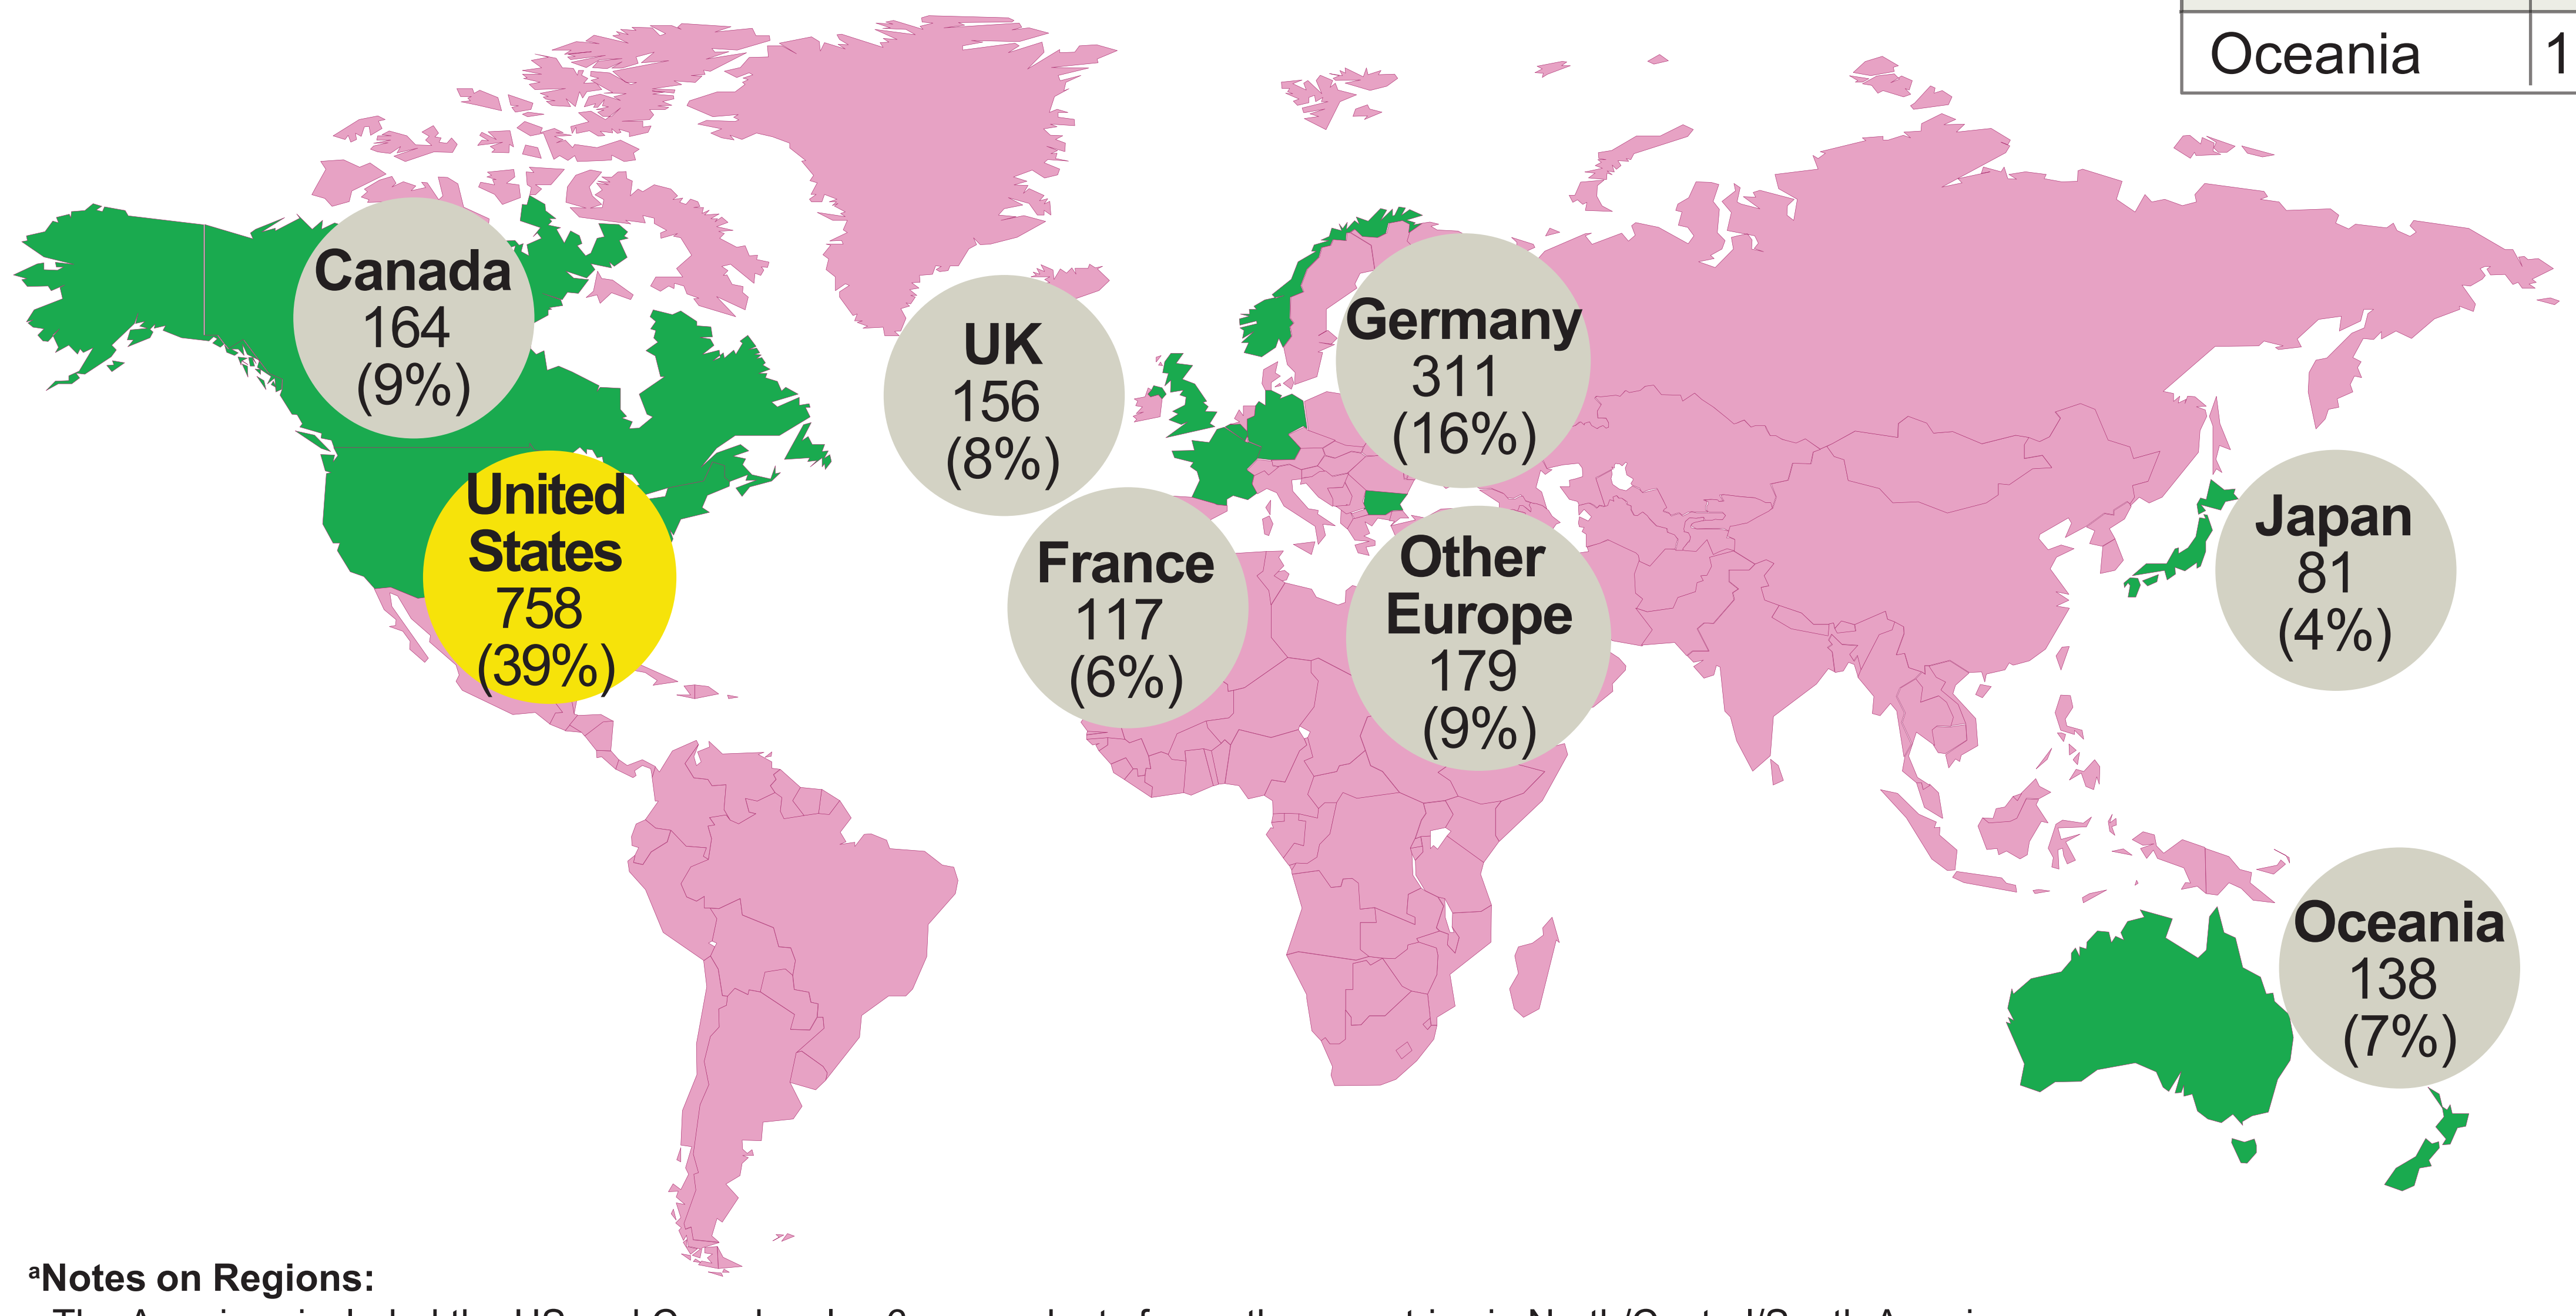

Canada
164
(9%)
United States
758
(39%)
UK
156
(8%)
France
117
(6%)
Germany
311
(16%)
Other Europe
179
(9%)
Japan
81
(4%)
Oceania
138
(7%)

[a]**Notes on Regions:**
- The Americas included the US and Canada, plus 6 respondents from other countries in North/Central/South America
- "Other Europe" included Norway (54), Belgium (29), Bulgaria (18), and other European countries (78)
- Asia included Japan plus Singapore and other Asian countries (18)

Base population: All respondents (N = 1928).
Questions: In which country do you live? In which region do you live?

- 15% of US patients had lung NETs (n = 112)
 - Of patients who knew their status, the majority reported having nonfunctional/asymptomatic (53%, n = 79) and low-grade (G1; 57%, n = 70) lung NETs (**Table 1**)

Table 1. Patient-Reported Functional Status and Grade of NETs[a]

Tumor Type	Functional Status, n (%)			Grade, n (%)			
	Functional[b]	Nonfunctional[c]/ Asymptomatic[d]	Unknown[e]	G1[f]	G2[g]	G3[h]	Unknown[i]
Total US	363 (48)	248 (33)	147 (19)	264 (35)	127 (17)	44 (6)	323 (43)
Lung NET	37 (33)	42 (38)	33 (29)	40 (36)	17 (15)	13 (12)	42 (38)

Base population: All US respondents (n = 758), and all US respondents with lung NETs (n = 112).
Questions: How, if at all, does your physician describe the functional status of your NET? Which of the following, if any, does your physician currently use to describe the grade of your NET?
[a]Some percentages add up to 101% because of rounding.
[b]Produces symptoms caused by the secretion of hormones (eg, flushing, diarrhea, wheezing, cramping).
[c]Do not secrete hormones; however, they may elicit symptoms caused by tumor growth (eg, pain, intestinal blockage, bleeding).
[d]Experience no symptoms.
[e]Physician does not describe/discuss; I don't remember.
[f]NETs are relatively slow growing; Ki-67 index <2%.
[g]NETs have a less predictable, moderately aggressive course; Ki-67 index 3%-20%.
[h]NETs can be highly aggressive; Ki-67 index >20%.
[i]Physician did not give my NET a grade/describe/discuss; I don't remember.

Diagnosis and Management of Lung NETs

- For approximately one-third of the patients surveyed (31%), >5 years elapsed from symptom onset to a diagnosis of lung NET **(Figure 2**)
 - Almost half of all patients (49%) were diagnosed with other conditions prior to receiving a diagnosis of lung NET, with asthma (56%) and pneumonia (35%) the most common initial diagnoses (**Figure 3**)
- The majority of patients had not visited a NET specialist center (64%)
 - Of those who had, only 13% were diagnosed there

Figure 2. Time between first symptom onset and diagnosis.

Diagnosed with NET in the following time period	Don't know/ NA	<6 months	6 months–2 years	2–5 years	>5 years	Mean (months)
Total US	8%	18%	17%	19%	34%	59.0
Lung NET	8%	24%	12%	21%	31%	58.2

NA, not applicable.
Base population: All US respondents (n = 758), and all US respondents with lung NETs (n = 112).
Question: What was the approximate length of time between your first symptom and your NET diagnosis?

Figure 3. Diagnosis received before NET diagnosis.[a]

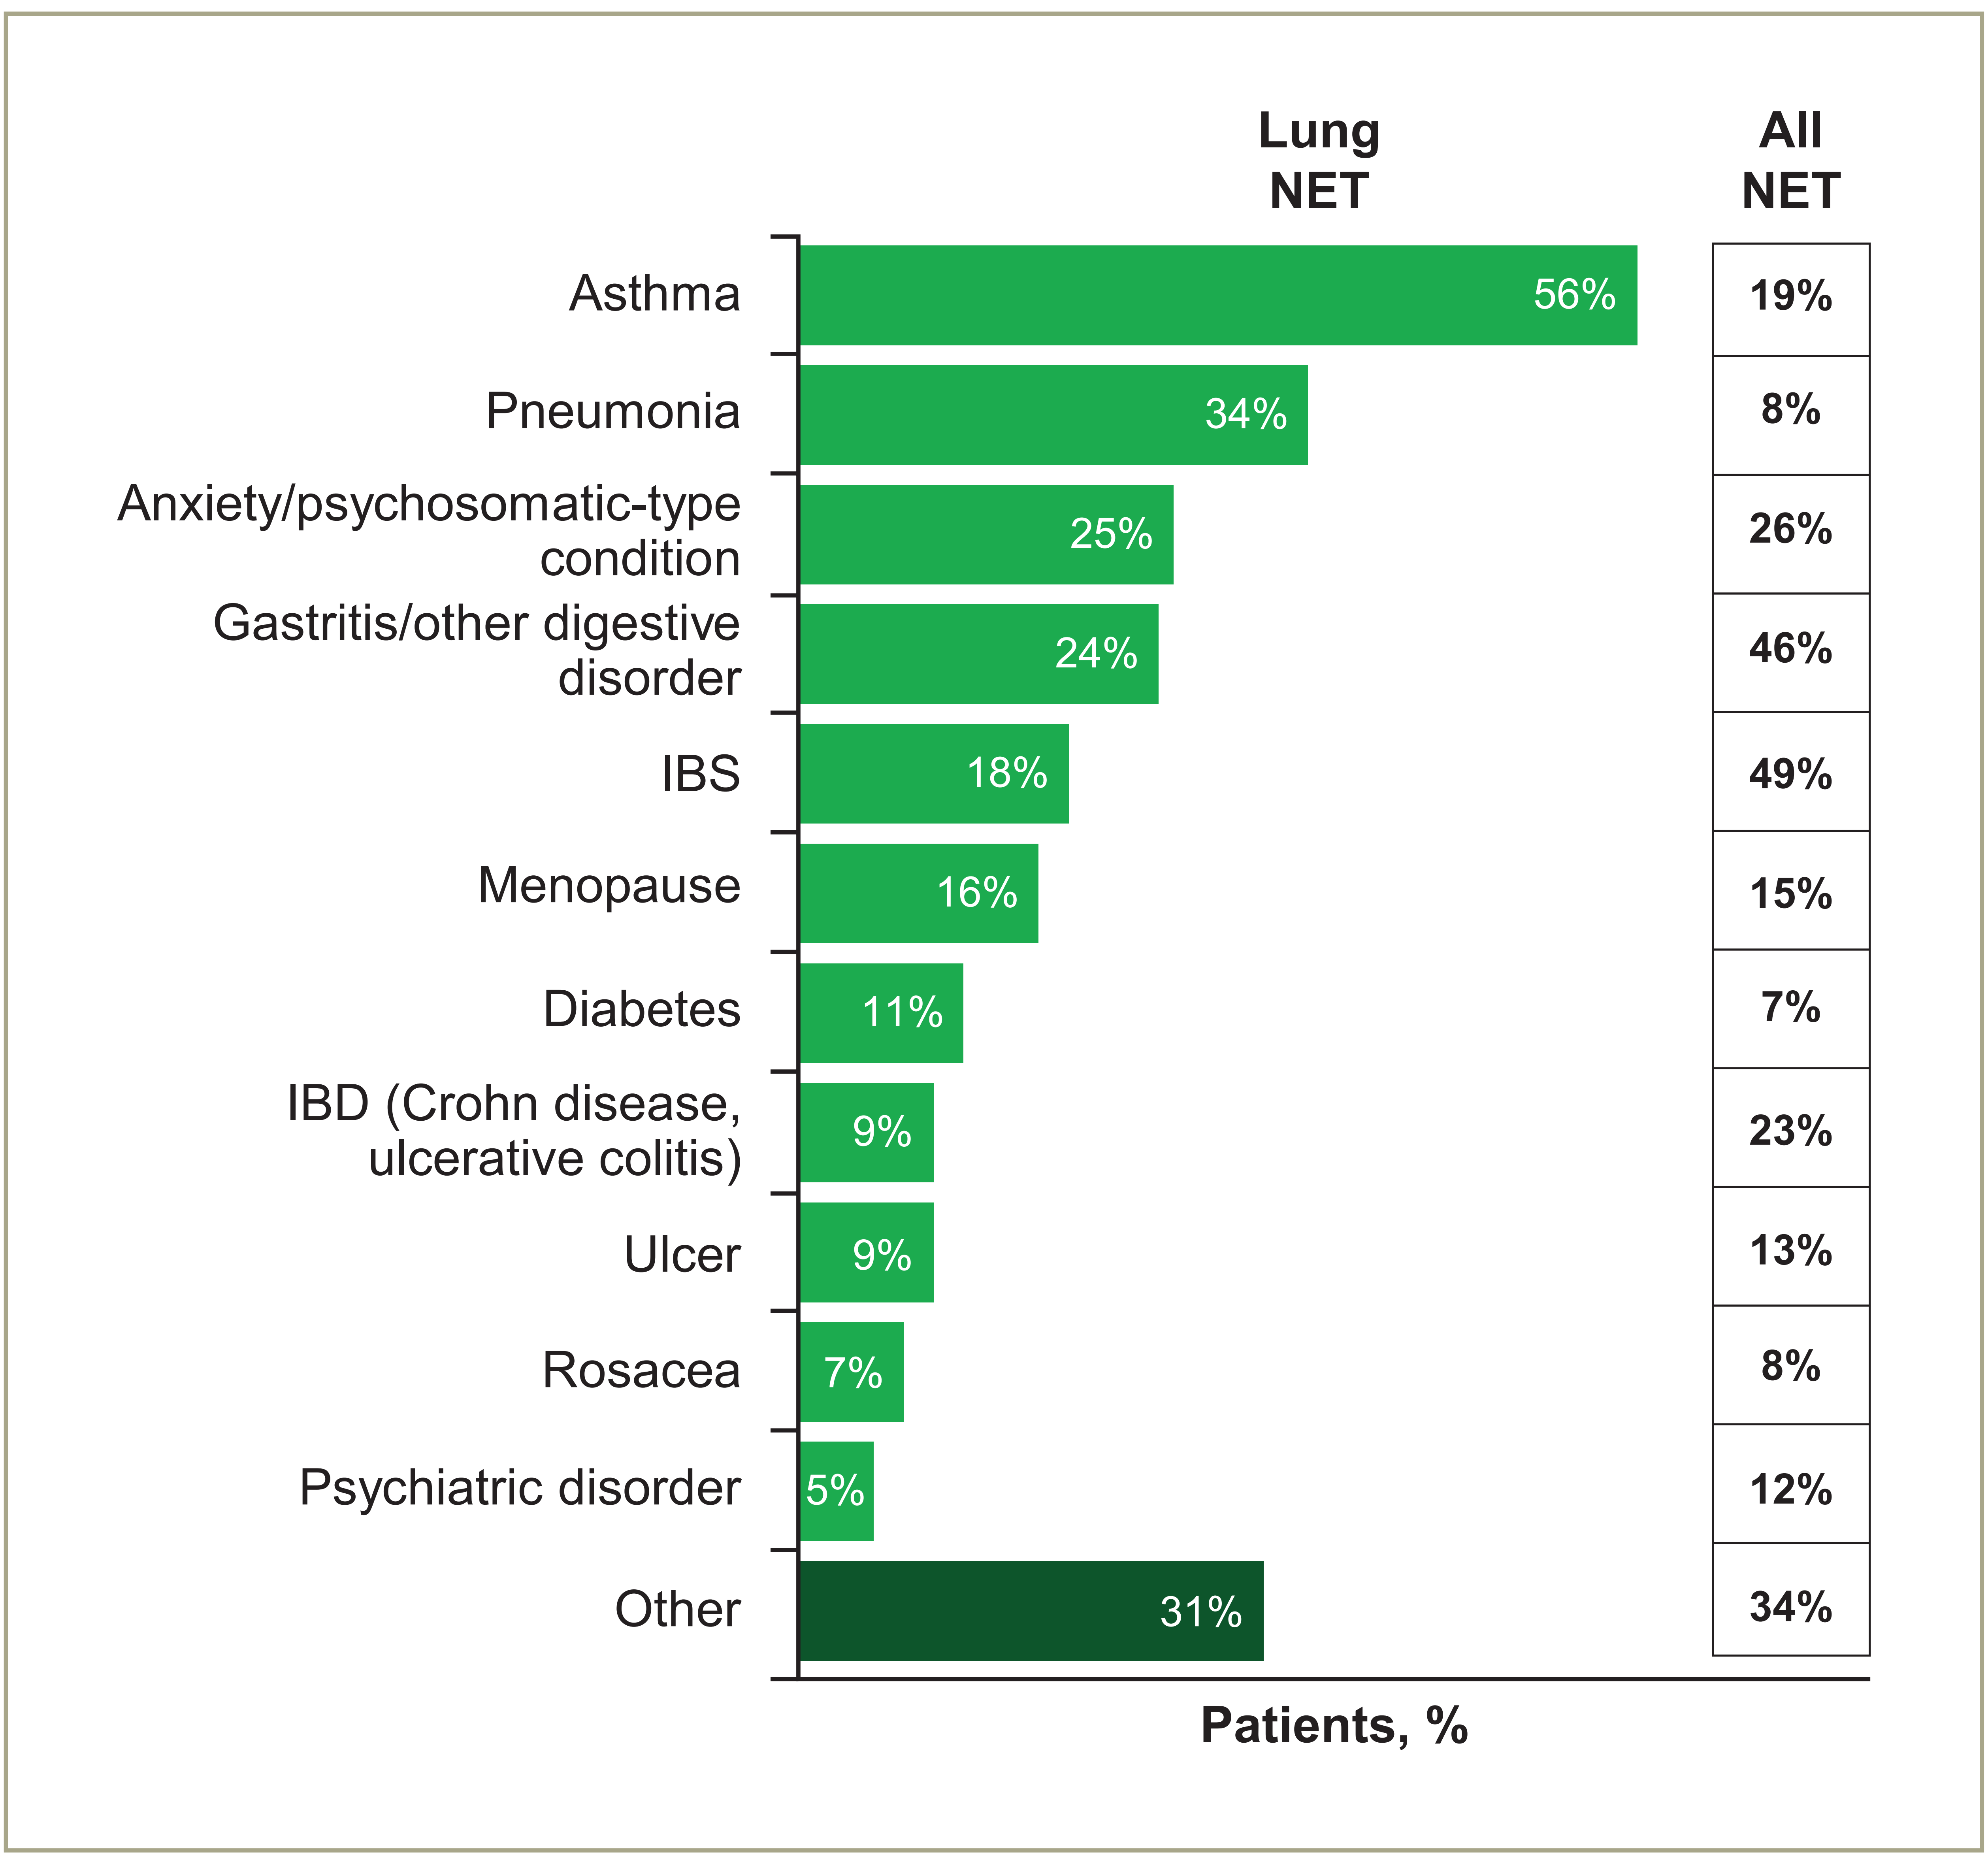

Lung NET
All NET
Asthma
56%
19%
Pneumonia
34%
8%
Anxiety/psychosomatic-type condition
25%
26%
Gastritis/other digestive disorder
24%
46%
IBS
18%
49%
Menopause
16%
15%
Diabetes
11%
7%
IBD (Crohn disease, ulcerative colitis)
9%
23%
Ulcer
9%
13%
Rosacea
7%
8%
Psychiatric disorder
5%
12%
Other
31%
34%
Patients, %

IBD, inflammatory bowel disease; IBS, irritable bowel syndrome.
Base population: All US respondents who were diagnosed with other conditions prior to lung NET (n = 55), and all US respondents diagnosed with other conditions prior to NET (n = 368).
Question: Which of the following conditions were you initially diagnosed with prior to receiving a lung NET diagnosis?
[a]Only includes conditions mentioned by ≥5% of patients.

- Patients with lung NETs were more likely to be given the impression that their NET was curable (32%, vs 9% and 14% for gastrointestinal [GI] NET and pancreatic [p]NET, respectively) and not something to worry about (40%, vs 23% and 14% for GI NET and pNET, respectively)

Impact of NETs on Quality of Life and Work Life

- 67% of patients with lung NETs experienced a moderate to large negative impact on their quality of life (**Figure 4**)
 - This finding was similar to patients with GI NETs and pNETs (73% and 74%, respectively)
- Of patients with lung NETs who were still working at the time of the survey, 31% reported having to stop working temporarily (**Figure 5**)
 - This rate was higher than for the overall patient population (16%), GI NETs (12%), and pNETs (12%)

Figure 4. Negative impact of NETs on different aspects of life.

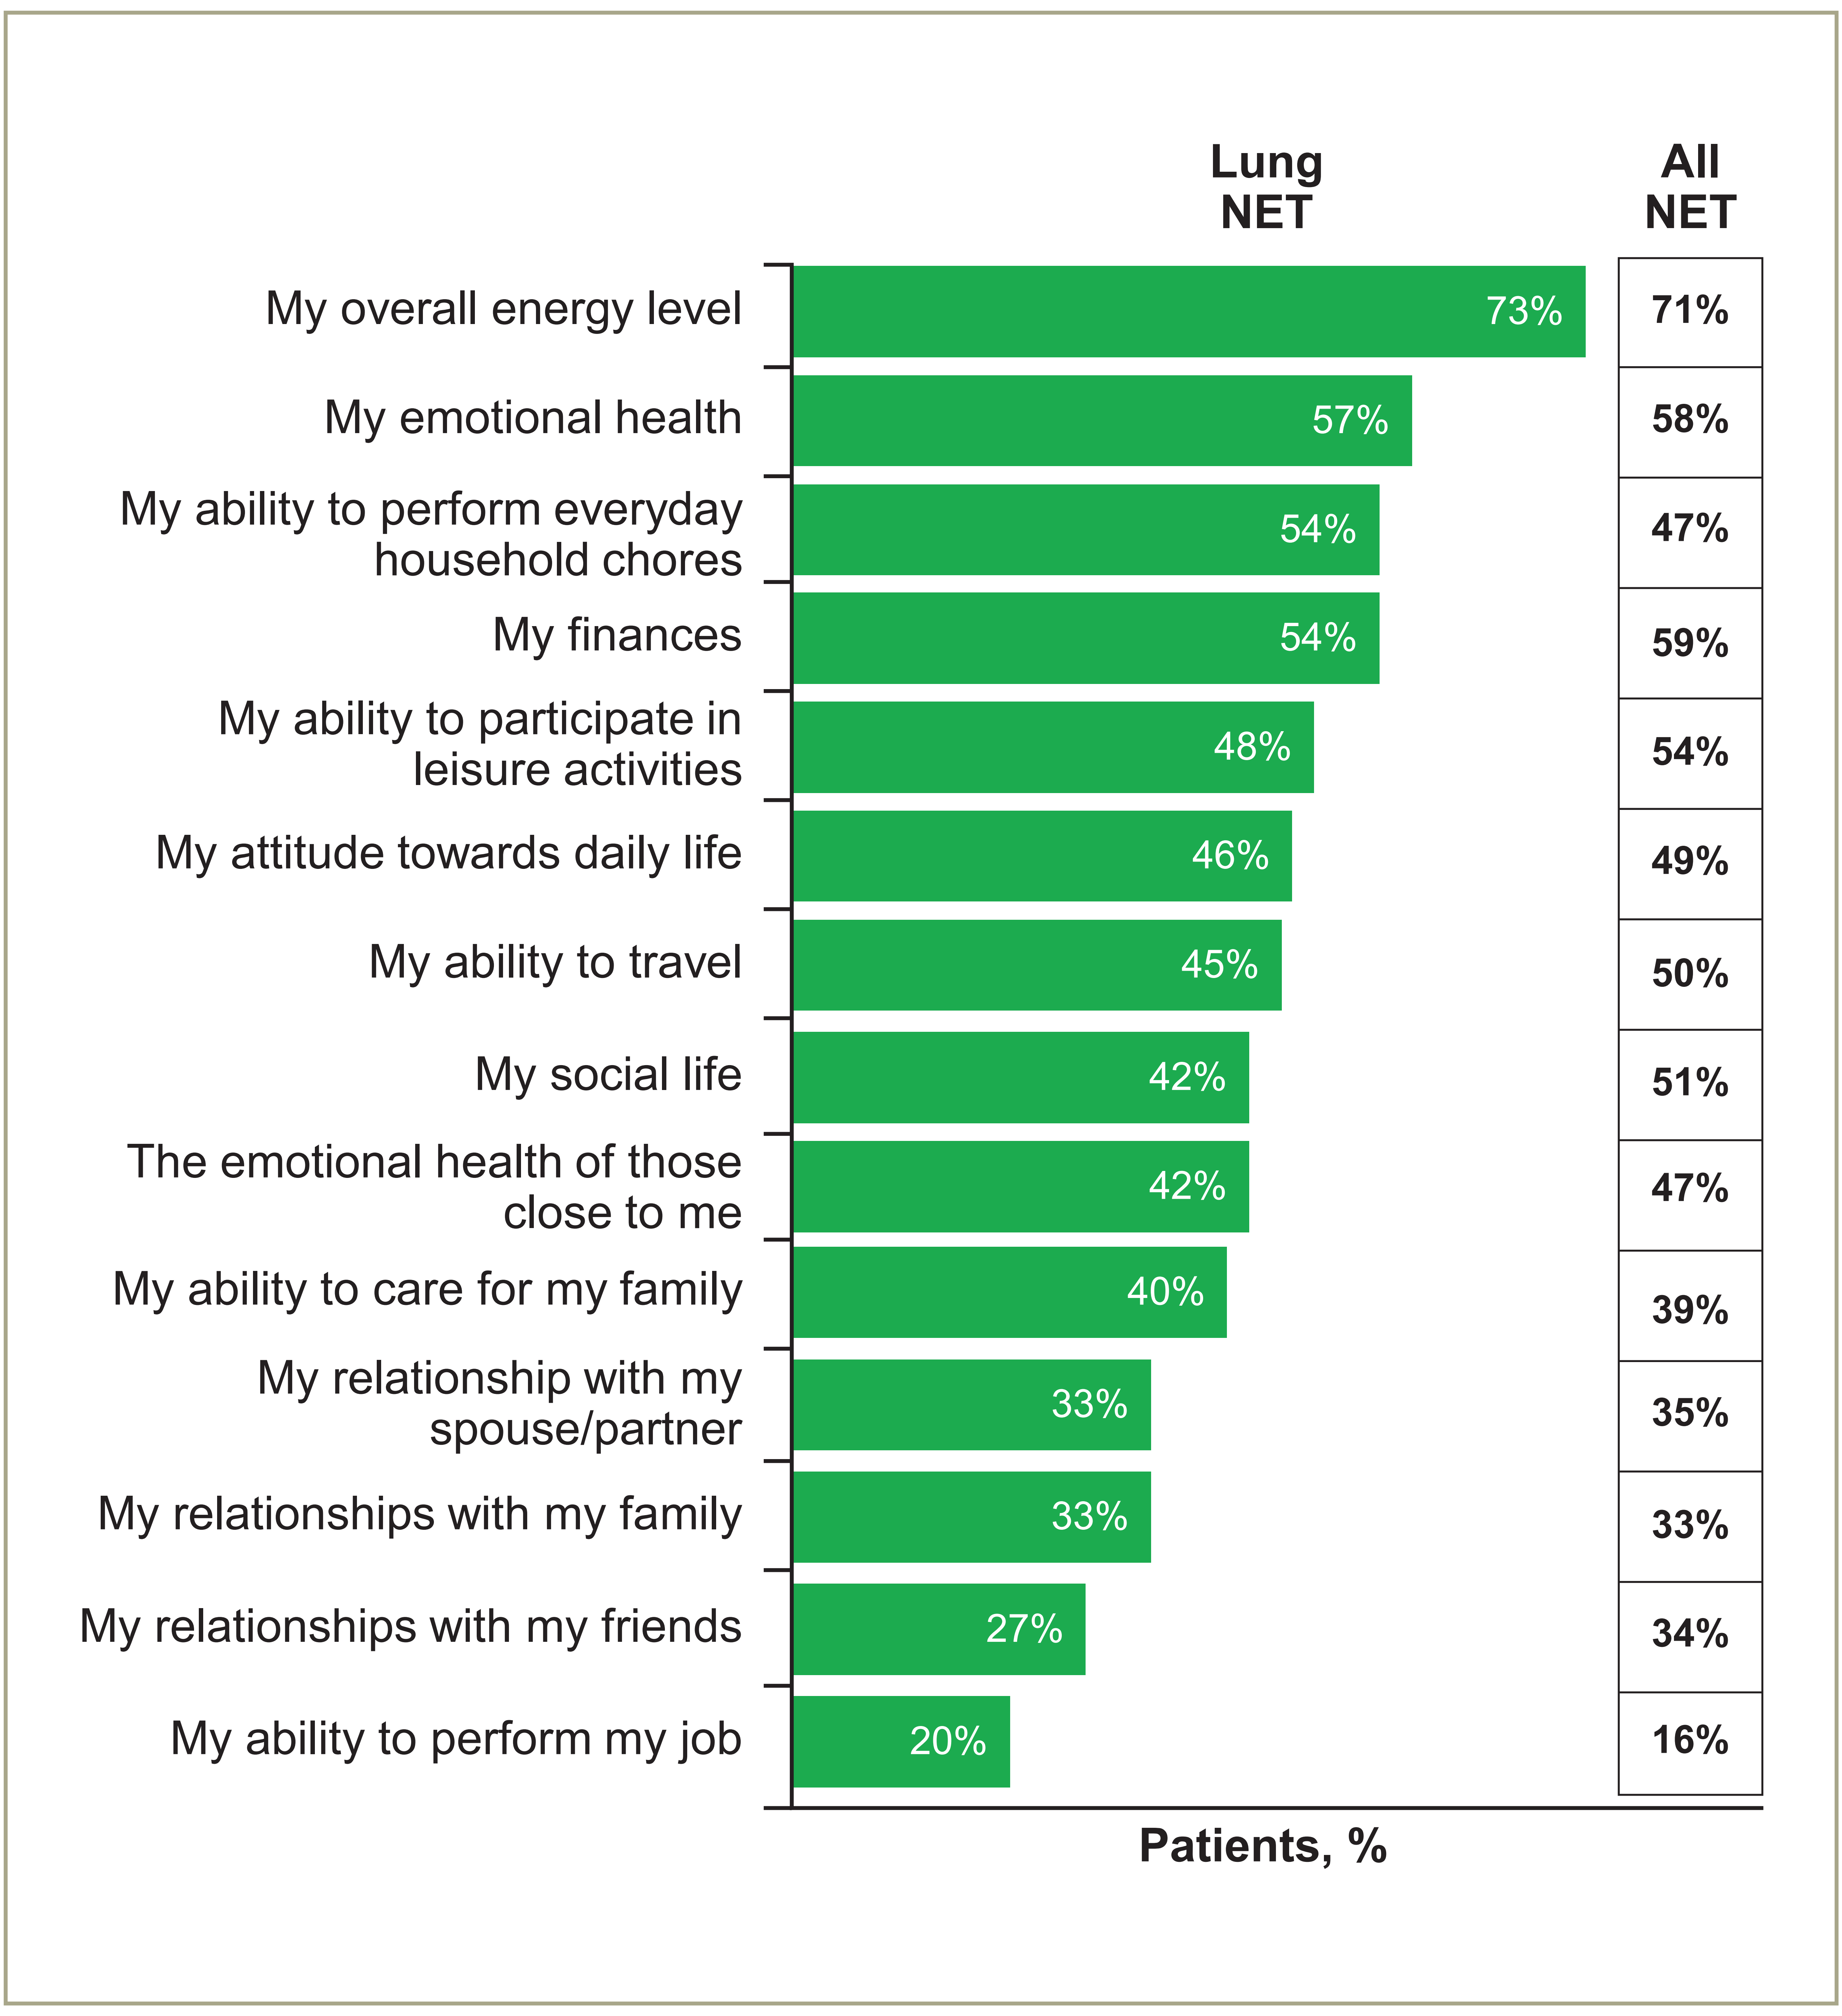

Lung NET
All NET
My overall energy level
73%
71%
My emotional health
57%
58%
My ability to perform everyday household chores
54%
47%
My finances
54%
59%
My ability to participate in leisure activities
48%
54%
My attitude towards daily life
46%
49%
My ability to travel
45%
50%
My social life
42%
51%
The emotional health of those close to me
42%
47%
My ability to care for my family
40%
39%
My relationship with my spouse/partner
33%
35%
My relationships with my family
33%
33%
My relationships with my friends
27%
34%
My ability to perform my job
20%
16%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (n = 316).
Question: How much has each of the following areas of your life been negatively affected, if at all, by your NET?
Top 2 responses shown (a moderate amount/a lot).

Figure 5. Impact of NETs on work life.

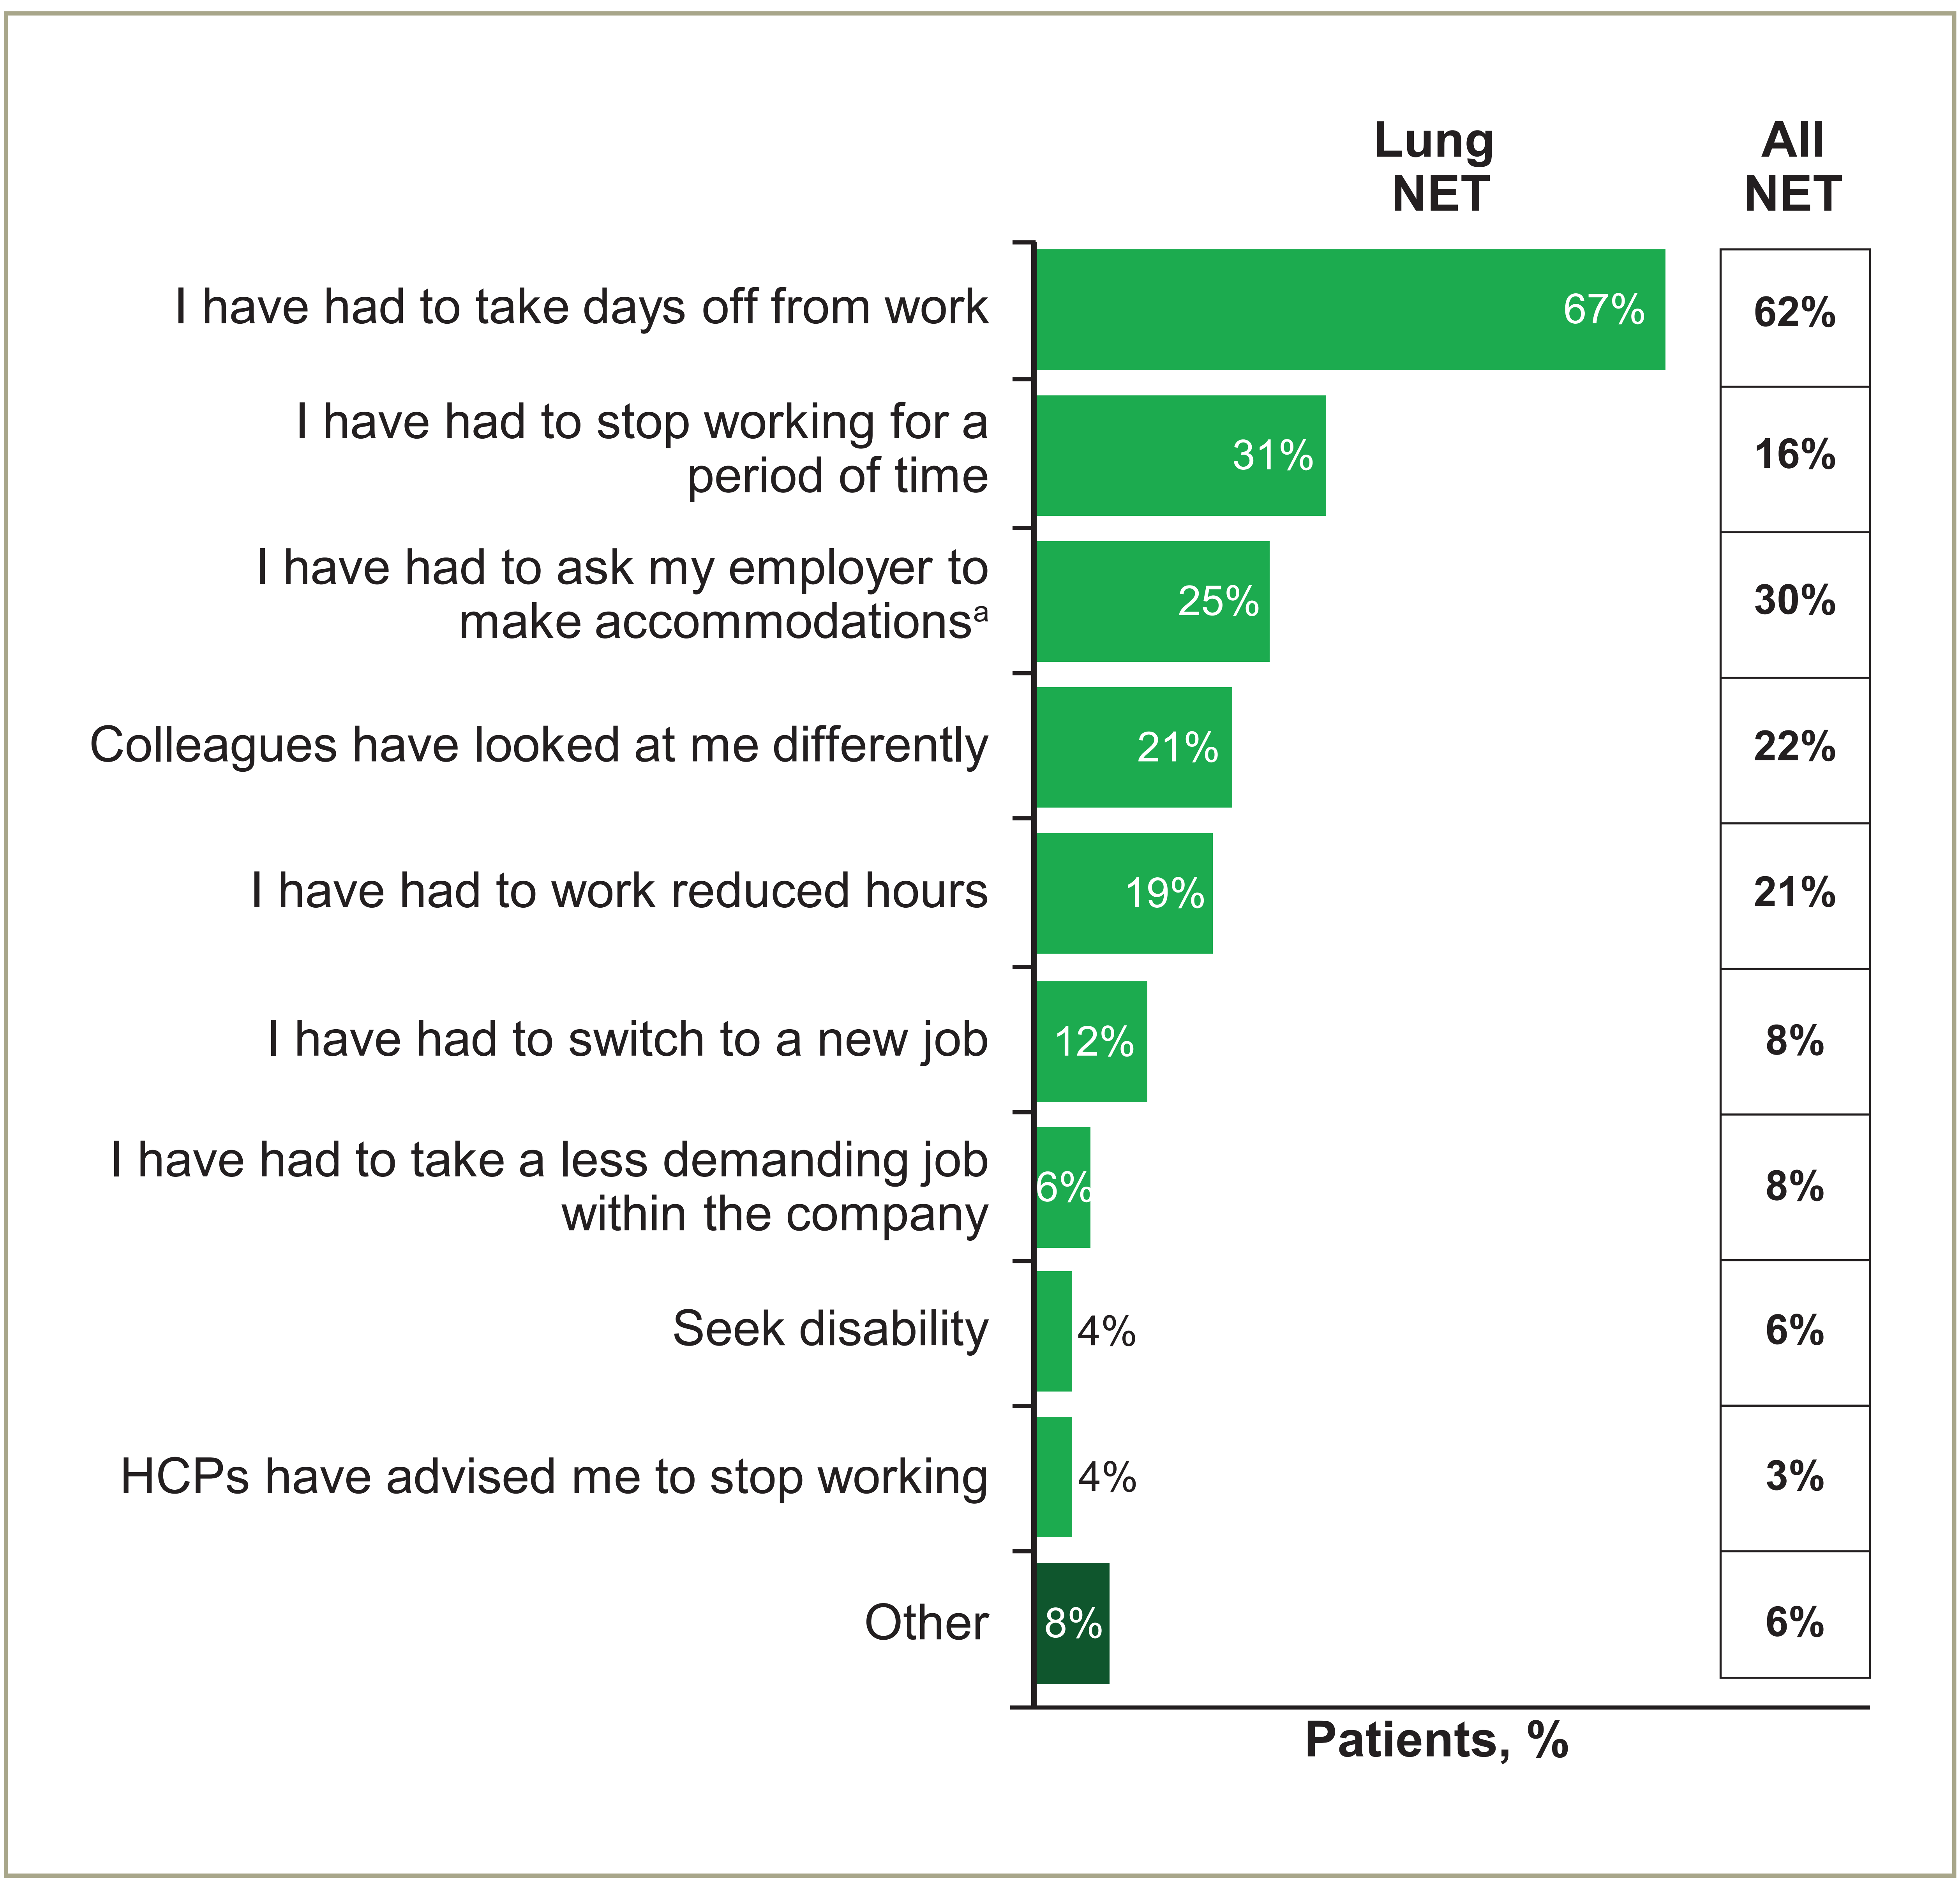

Lung NET
All NET
I have had to take days off from work
67%
62%
I have had to stop working for a period of time
31%
16%
I have had to ask my employer to make accommodations[a]
25%
30%
Colleagues have looked at me differently
21%
22%
I have had to work reduced hours
19%
21%
I have had to switch to a new job
12%
8%
I have had to take a less demanding job within the company
6%
8%
Seek disability
4%
6%
HCPs have advised me to stop working
4%
3%
Other
8%
6%
Patients, %

Base population: All US respondents with lung NETs who were working full time/part time or self-employed (n = 52), and all US respondents who were working full time/part time or self-employed with NETs (N = 316).
Question: Has your NET impacted you at work in any of the following ways? Please select all that apply.
[a]Make accommodations: eg, flexible work schedule, work from home, adaptive devices, opportunities for rest.

Key Improvements Patients Believe Would Help Them Live Better with NETs

- Overall, patients with lung NETs had more negative feelings regarding health care provider expertise/knowledge and quality of care compared with the overall NET population (**Figure 6**)
 - Lung NET patients were more likely to desire better access to NET experts (56%), better access to NET-specific medical treatments (54%; not significant), and a more knowledgeable health care team (52%)
- When asked what would have helped the patient have a better experience following a NET diagnosis, 71% of lung NET patients expressed the need for more immediate access to NET specialists, and 45% desired transfers to NET centers of expertise

Figure 6. Improvements patients believed would help them live better with NETs.

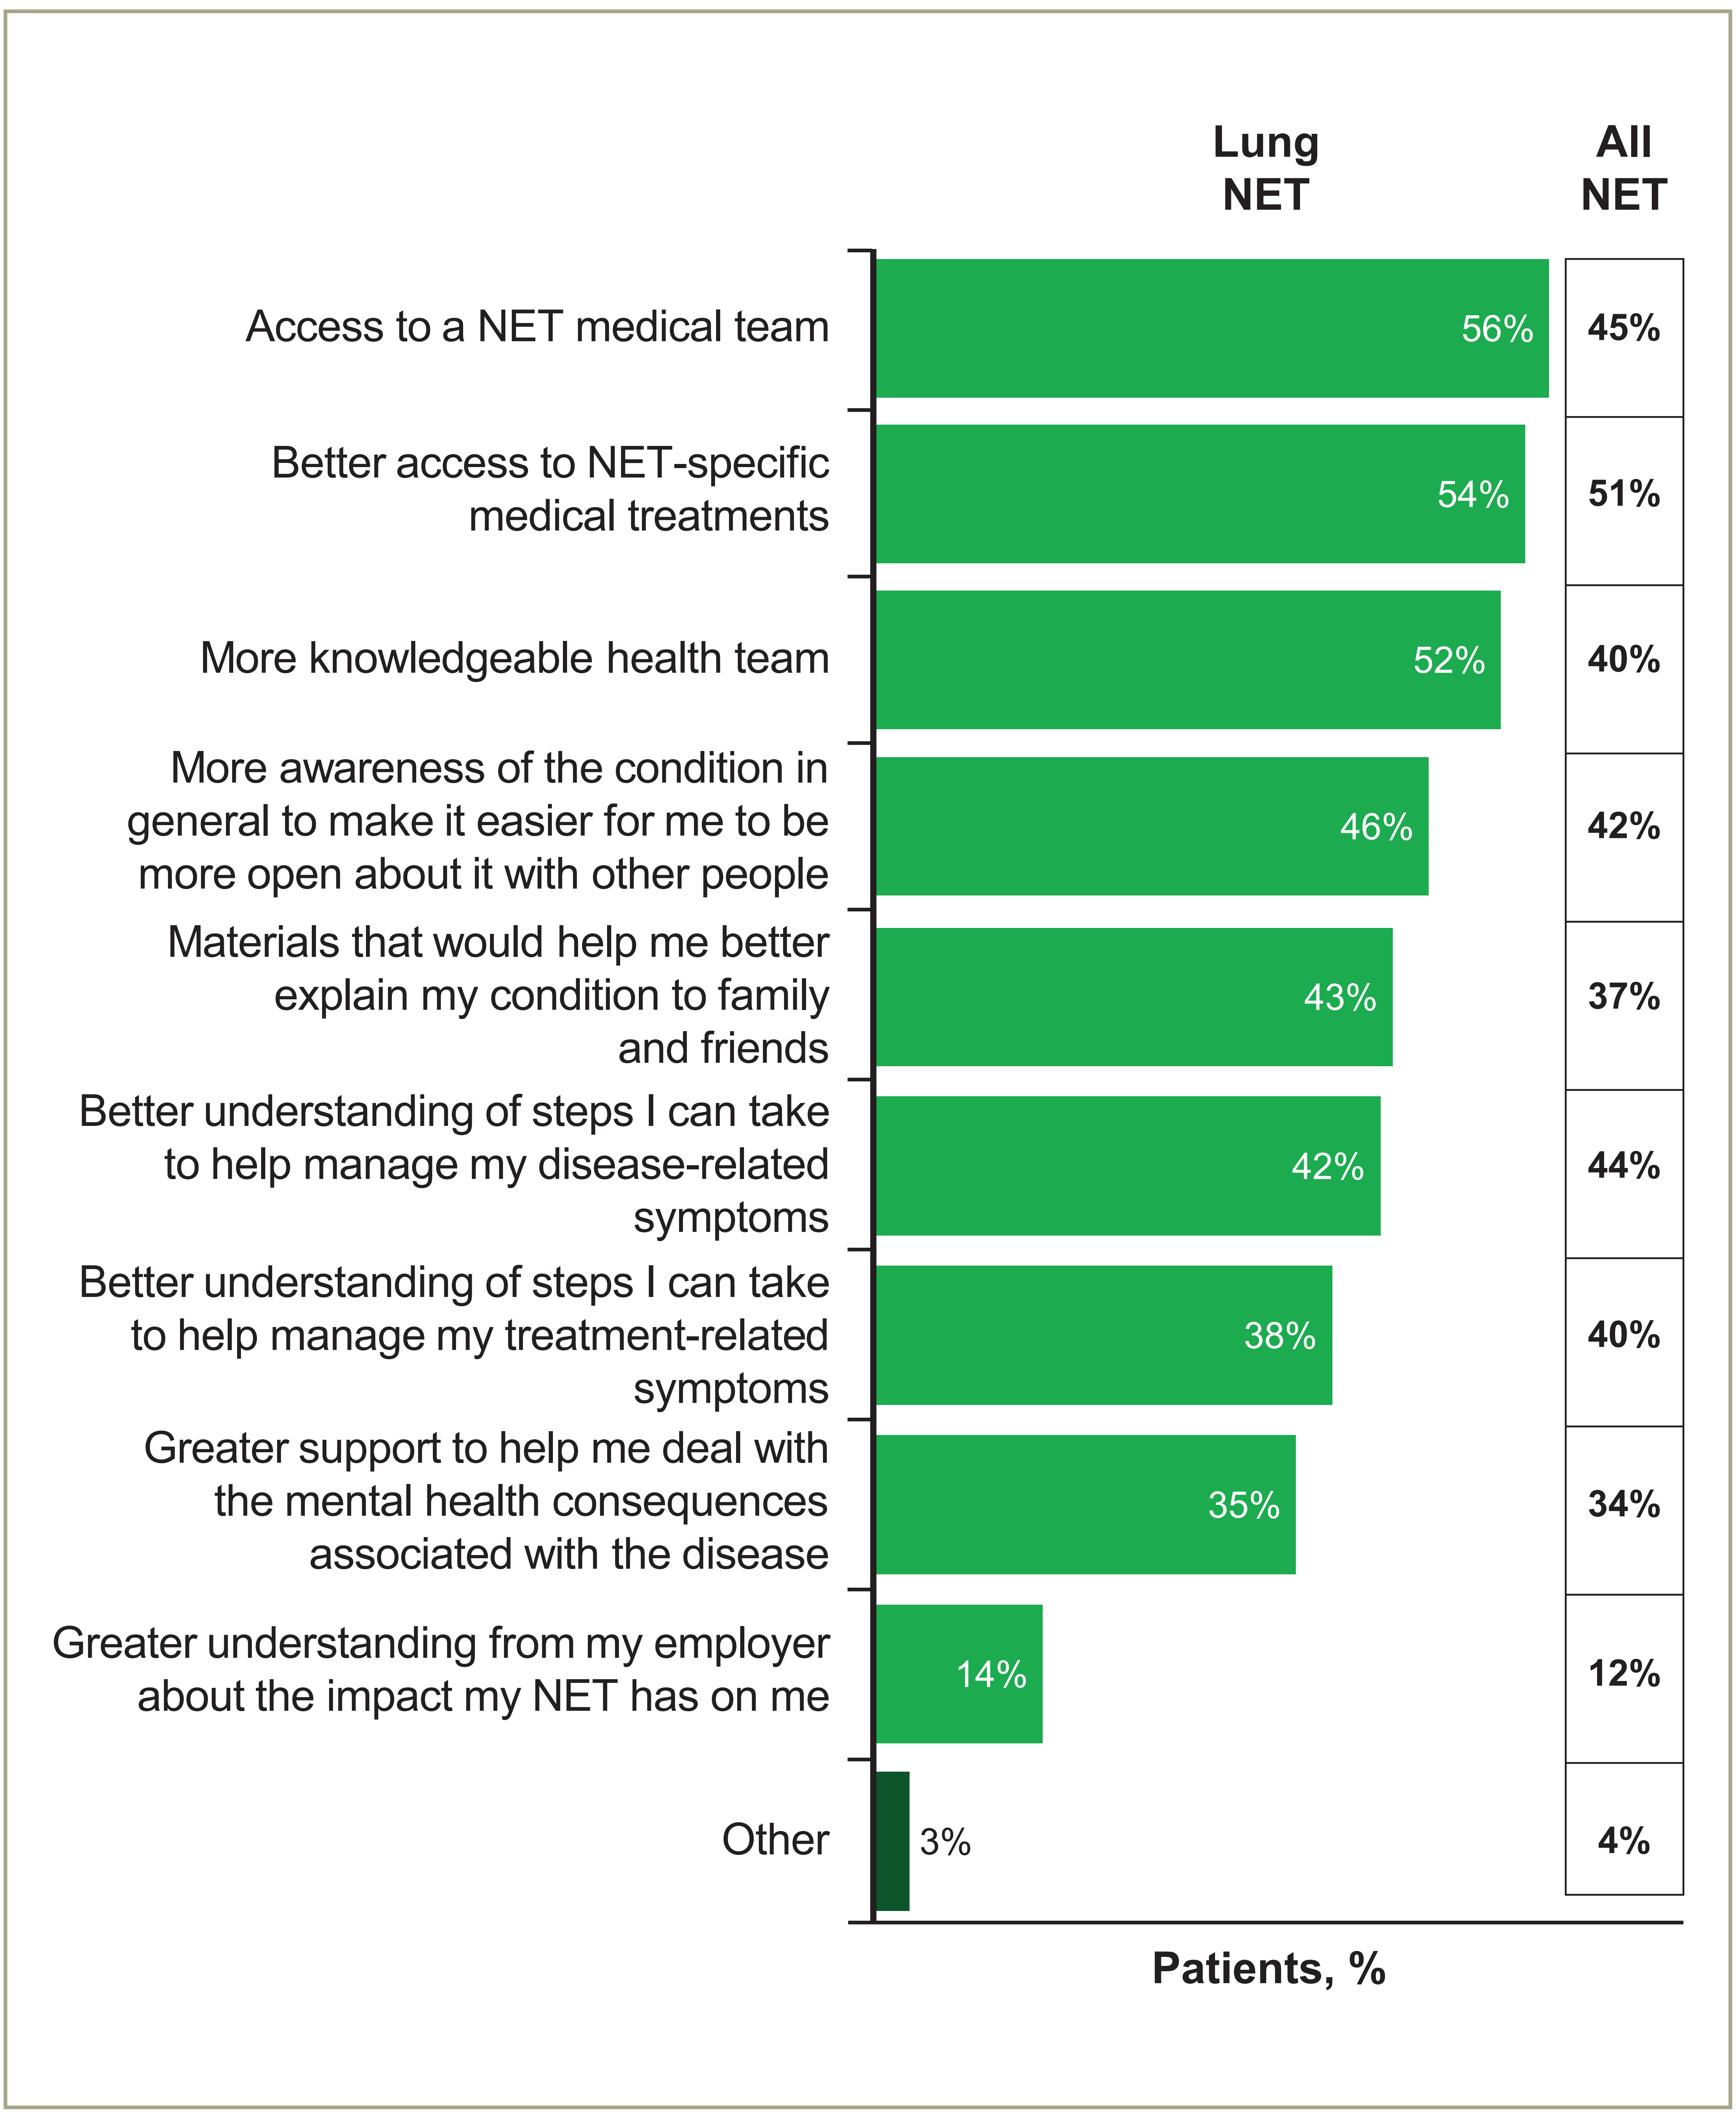
Lung NET
All NET
Access to a NET medical team
56%
45%
Better access to NET-specific medical treatments
54%
51%
More knowledgeable health team
52%
40%
More awareness of the condition in general to make it easier for me to be more open about it with other people
46%
42%
Materials that would help me better explain my condition to family and friends
43%
37%
Better understanding of steps I can take to help manage my disease-related symptoms
42%
44%
Better understanding of steps I can take to help manage my treatment-related symptoms
38%
40%
Greater support to help me deal with the mental health consequences associated with the disease
35%
34%
Greater understanding from my employer about the impact my NET has on me
14%
12%
Other
3%
4%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (N = 758).
Question: Which of the following, if any, would help you living with a NET?

Patient Perception of NET Management and Treatment

- Following diagnosis, fewer lung NET patients were satisfied that they received answers to questions (43%) or sufficient information (33%) compared with GI/pNET patients (62%/59% and 48%/48%, respectively) (**Figure 7**)

Figure 7. Patient perception of NET management.

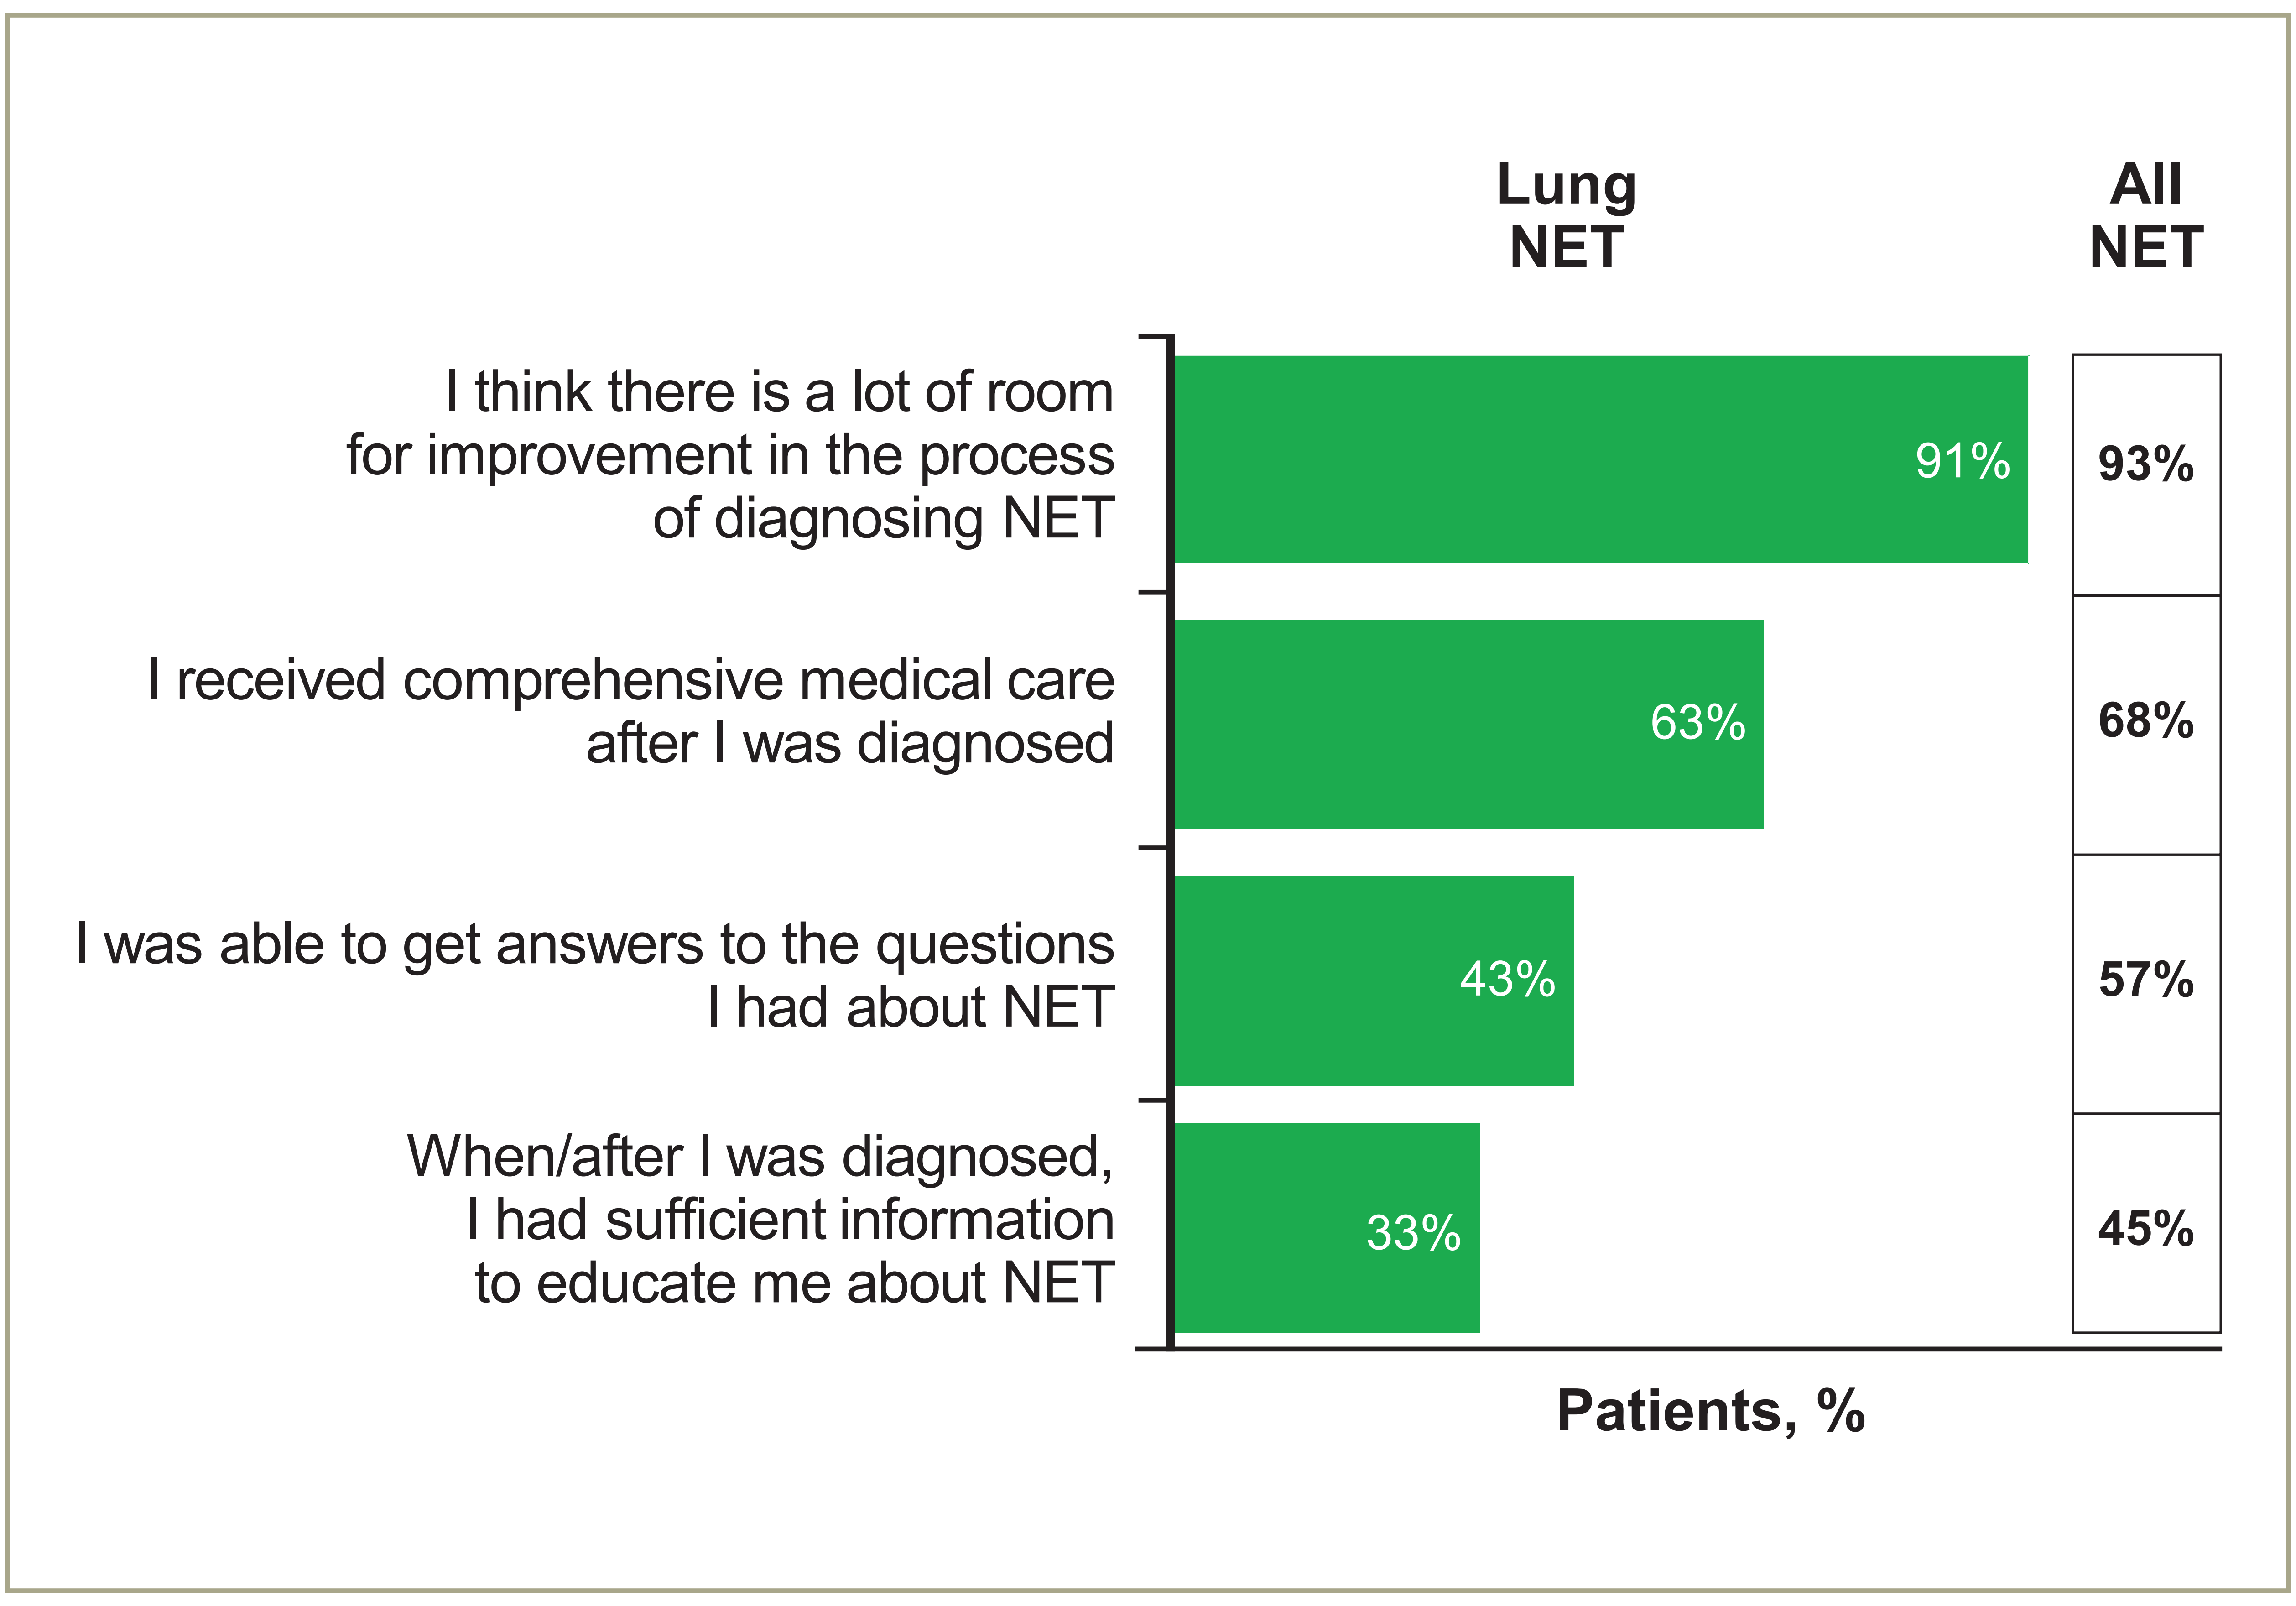
Lung NET
All NET
I think there is a lot of room for improvement in the process of diagnosing NET
91%
93%
I received comprehensive medical care after I was diagnosed
63%
68%
I was able to get answers to the questions I had about NET
43%
57%
When/after I was diagnosed, I had sufficient information to educate me about NET
33%
45%
Patients, %

Base population: All US respondents with lung NETs (n = 112), and all US respondents with NETs (N = 758).
Question: To what extent do you agree or disagree with the following statements?
Top 2 responses shown (a moderate amount/ a lot).

- Almost half (45%) of lung NET patients regarded the quality of available treatments to be poor or very poor, compared with 29% of GI NET patients (significant), and 33% of pNET patients (nonsignificant)

LIMITATIONS

- This large NET patient survey had several important limitations that may have impacted results:
 - A patient-reported design was employed without independent verification, leading to potential recall bias
 - This survey did not use standardized, validated quality-of-life assessments
 - Recruitment in the United States was conducted primarily through patient advocacy groups (20%) and online sources (72%), which may have resulted in a potentially biased sample not fully representative of the heterogeneous US NET patient population
 - Respondents were more likely to be highly engaged and motivated care seekers, including female patients and/or those with a poorer prognosis

CONCLUSIONS

- Findings from this large survey provide valuable information on the experience of patients with lung NETs
 - Almost one-third of patients were diagnosed >5 years after symptom presentation
 - The majority of patients experienced a moderate to large negative impact on their daily life, and many report a negative impact on work life
 - Patients with lung NETs were more likely than those with other NET types to have negative feelings regarding various aspects of their care
 - Most patients indicated that improvements should be made in diagnosis, and nearly half indicated that the quality of available treatments was poor or very poor
- A number of suggestions for improving the diagnosis and management of lung NETs were identified, including
 - Better access to health care professionals with NET expertise
 - Immediate transfer to centers of expertise following diagnosis

REFERENCES

1. Yao JC et al. *J Clin Oncol*. 2008;26:3063-3072.
2. Travis WD et al. *J Thorac Oncol*. 2015;10:1243-1260.
3. Rekhtman N. *Arch Pathol Lab Med*. 2010;134:1628-1638.
4. Filosso PL et al. *J Thorac Dis*. 2015;7(suppl 2):S163-S171.
5. Travis WD. *Ann Oncol*. 2010;21(suppl 7):vii65-vii71.
6. Griffiths J et al. *Eur J Oncol Nurs*. 2007;11:434-441.
7. Feinberg Y et al. *Eur J Oncol Nurs*. 2013;17:541-545.

ACKNOWLEDGMENTS

We thank the patients who participated in this survey and our patient advocacy partners in the United States who contributed to this project, The Carcinoid Cancer Foundation and Caring for Carcinoid Foundation

Medical editorial assistance for this poster was provided by the ApotheCom Novartis Oncology team (Yardley, PA) and was funded by Novartis Pharmaceuticals Corporation

Mobile Friendly e-Prints

3 ways to instantly download an electronic copy of this poster to your mobile device or e-mail a copy to your computer or tablet

Scan this QR code

Text Message (SMS)

Text: **Q4d167**

to: 8NOVA (86682) *US Only*
+18324604729 *North, Central and South Americas; Caribbean; China*
+447860024038 *UK, Europe & Russia*
+46737494608 *Sweden, Europe*

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=4d167

Standard data or message rates may apply.