PXXX

Multivariate analysis of progression-free survival in the CLARINET study of lanreotide depot/autogel vs. placebo identifies prognostic factors in neuroendocrine tumors

Edward M. Wolin MD,[1] Martyn E. Caplin DM,[2] Marianne E. Pavel MD,[3] Jarosław B. Ćwikła MD PhD,[4] Alexandria T. Phan MD,[5] Markus Raderer MD,[6] Eva Sedláčková MD,[7] Guillaume Cadiot MD PhD,[8] Jaume Capdevila MD,[9] Lucy Wall MD,[10] Guido Rindi MD PhD,[11] Nilani Liyanage MSc,[12] Edda Gomez-Panzani MD,[12] Philippe B. Ruszniewski MD PhD,[13] on behalf of the CLARINET Study Group

[1]Markey Cancer Center, Lexington, KY, USA; [2]Royal Free Hospital, London, UK; [3]Charité University Medicine Berlin, Berlin, Germany; [4]University of Varmia and Masuria, Olsztyn, Poland; [5]The Methodist Hospital, Houston, TX, USA; [6]University Hospital, Vienna, Austria; [7]First Faculty of Medicine and General Teaching Hospital, Prague, Czech Republic; [8]Robert-Debré Hospital, Reims, France; [9]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, Barcelona, Spain; [10]Western General Hospital, Edinburgh, UK; [11]Università Cattolica del Sacro Cuore, Rome, Italy; [12]Ipsen, Les Ulis, France; [13]Beaujon Hospital, Clichy, France.

Background

- Data from the CLARINET study of lanreotide depot (also known as autogel) provide robust evidence of antitumor effects that underpin its position as first-line medical therapy in US guidelines for metastatic pancreatic and intestinal neuroendocrine tumors (NETs).[1]
 - The CLARINET study demonstrated that lanreotide significantly prolonged progression-free survival (PFS) vs. placebo: lanreotide, median PFS not reached vs. placebo, 18 months (hazard ratio [HR]: 0.47 [95% CI: 0.30, 0.73]) at 96 weeks.[2]
 - This antitumor treatment response was consistent across subgroups based on key clinical characteristics (tumor location, tumor grade 1 or 2 [Ki-67 up to 10%], and hepatic tumor load).[2]
- Individual patient circumstances are key considerations in the application of treatment guidelines.[1] Determining prognostic factors can assist in understanding baseline characteristics that may have a meaningful influence on clinical outcomes for individual patients.
- Here, we present the results of a preplanned, exploratory, stepwise multivariate analysis to identify prognostic factors for PFS in the CLARINET study, with investigation of potential treatment interactions.

Methods

Overview of CLARINET study

- The design of the CLARINET core study has been reported in detail previously.[2]
 - This was a randomized double-blind placebo-controlled trial of 96 weeks' duration conducted at 48 study centers in 14 countries (ClinicalTrials.gov: NCT00353496; EudraCT 2005-004904-35).
 - Patients had: metastatic and/or locally advanced well/moderately differentiated non-functioning somatostatin receptor-positive NETs with a Ki-67 up to 10%; tumors originating in the pancreas, midgut or hindgut, or of unknown origin.
 - Disease progression status was documented at baseline (RECIST 1.0).
 - Patients received lanreotide depot 120 mg (fixed dose) or placebo by deep subcutaneous injection once every 28 days for 96 weeks or until death/progressive disease (PD; assessed using RECIST 1.0).
- PFS for lanreotide was defined as time from randomization to death/centrally assessed PD. Analyses of PFS were stratified according to PD (yes/no) and prior therapy for NET (yes/no) at baseline.

Exploratory univariate and multivariate analyses

- The effect of treatment (lanreotide vs. placebo) was tested, alongside the baseline stratification factors (PD, prior therapy [both yes vs. no]), in a separate Cox proportional hazards (PH) model for each of several potential prognostic factors at baseline (listed in **Table 1**).
- Any factors found to be significant at $p \leq 0.10$ (Wald Chi-square test statistic) in univariate analyses were considered to be potentially important, and were tested for significance in stepwise multivariate analyses in the presence of the other factors (via the Wald Chi-square test statistic).
- Terms not originally meeting $p \leq 0.10$ were added to the multivariate model one at a time to determine if they had an effect, and were retained if so.
- Any interaction with treatment was investigated for each term remaining in the multivariate model.

Results

Patients

- Of 204 patients in the intent-to-treat population, 101 were randomized to lanreotide and 103 to placebo.[2]
 - Most patients (172/204 [84%]) had not received previous therapy at entry; 16 (16%) patients in each group had received prior therapy.
 - The majority of patients (105/204 [96%]) did not have PD at baseline; 4 (4%) patients in the lanreotide group and 5 (5%) in the placebo group had PD.
- Baseline characteristics were similar in the two treatment groups, with mean (±SD) age: 63.3 (±9.8) and 62.2 (±11.1) years; BMI 27.3 (±4.9) and 26.7 (±5.9) kg/m^2; and time since diagnosis: 32.6 (±46.1) and 34.4 (±41.4) months, in the lanreotide and placebo groups, respectively.

Exploratory univariate and multivariate analyses

- The proportions of patients in each category for the fourteen potential prognostic factors at baseline are shown in **Table 1**.
- Six of the fourteen factors were of potential importance in univariate analyses; four out of these six retained significance in the presence of other factors and were retained in the multivariate model (**Figure 1**).

Table 1. Categories tested in the exploratory univariate and multivariate analyses of potential prognostic factors.

Factor	Category	Lanreotide n=101	Placebo n=103	Total n=204
Location of primary tumor	Pancreas	42 (41.6)	49 (47.6)	91 (44.6)
	Midgut	33 (32.7)	40 (38.8)	73 (35.8)
	Hindgut	11 (10.9)	3 (2.9)	14 (6.9)
	Other / unknown	15 (14.9)	11 (10.7)	26 (12.7)
US/non-US enrolment	US	16 (15.8)	14 (13.6)	30 (14.7)
	Non-US	85 (84.2)	89 (86.4)	174 (85.3)
Region	Western Europe	53 (52.5)	67 (65.0)	120 (58.8)
	Eastern Europe and India	32 (31.7)	22 (21.4)	54 (26.5)
	USA	16 (15.8)	14 (13.6)	30 (14.7)
Sex	Male	53 (52.5)	54 (52.4)	107 (52.5)
	Female	48 (47.5)	49 (47.6)	97 (47.5)
Age	≤65 years	55 (54.5)	60 (58.3)	115 (56.4)
	>65 years	46 (45.5)	43 (41.7)	89 (43.6)
BMI	≤median*	43 (42.6)	55 (53.4)	98 (48.0)
	>median	53 (52.5)	44 (42.7)	97 (47.5)
Ethnicity	Caucasian/white	97 (96.0)	96 (93.2)	193 (94.6)
	Other	4 (4.0)	7 (6.8)	11 (5.4)
Time since diagnosis	≤5 years	85 (84.2)	87 (84.5)	172 (84.3)
	>5 years	16 (15.8)	16 (15.5)	32 (15.7)
Hepatic tumor load	0%	16 (15.8)	18 (17.5)	34 (16.7)
	0% to ≤10%	33 (32.7)	40 (38.8)	73 (35.8)
	>10% to ≤25%	13 (12.9)	17 (16.5)	30 (14.7)
	>25% to ≤50%	23 (22.8)	12 (11.7)	35 (17.2)
	>50%	16 (15.8)	16 (15.5)	32 (15.7)
Baseline Ki-67	≤2%	52 (51.5)	51 (49.5)	103 (50.5)
	>2% to ≤5%	24 (23.8)	19 (18.4)	43 (21.1)
	>5% to <10%	7 (6.9)	10 (9.7)	17 (8.3)
Grade of tumor	G1	69 (68.3)	72 (69.9)	141 (69.1)
	G2	32 (31.7)	29 (28.2)	61 (29.9)
Baseline CgA	≤ULN	33 (32.7)	34 (33.0)	67 (32.8)
	1–2 × ULN	25 (24.8)	18 (17.5)	43 (21.1)
	>2 × ULN	41 (40.6)	48 (46.6)	89 (43.6)
Previous chemotherapy for NF-NET	Yes	14 (13.9)	15 (14.6)	29 (14.2)
	No	87 (86.1)	88 (85.4)	175 (85.8)
Previous surgery of the primary tumor	Yes	40 (39.6)	39 (37.9)	79 (38.7)
	No	61 (60.4)	64 (62.1)	125 (61.3)

Data are n (%) patients (intent-to-treat population; patients with data missing [not shown] are included for percentage calculations). The first category listed for each factor was used as the reference category in the analyses. *Median BMI = 26.2 kg/m^2. BMI, body mass index; CgA, chromogranin A; NF-NET, non-functioning (enteropancreatic) neuroendocrine tumor; ULN, upper limit of normal.

- There was no evidence of significant interaction with treatment for any of the potential prognostic factors (Wald Chi-square test statistic: $p > 0.10$; data not shown).
- Hazard ratios (HRs) and upper limits of 95% confidence intervals from the individual Cox PH models were all consistently well below 1 (**Figure 2**). Data from each univariate model (together with the overall final multivariate Cox PH model) show that the PFS result in favour of lanreotide is not sensitive to the effects of the factors investigated.
- In the final multivariate model:
 - Treatment effect – with adjustment for potential prognostic factors, lanreotide reduced the risk of PD/death by 60% vs. placebo (HR 0.40 [95% CI 0.25, 0.63]; **Table 2**).
 - Of the potential prognostic factors used for stratification – prior therapy did not have a significant effect on PFS after adjustment, whereas PD at baseline was associated with over 4 times higher risk of PD/death compared no PD at baseline (**Table 2**).

Figure 1. Univariate analyses of PFS: significant factors.

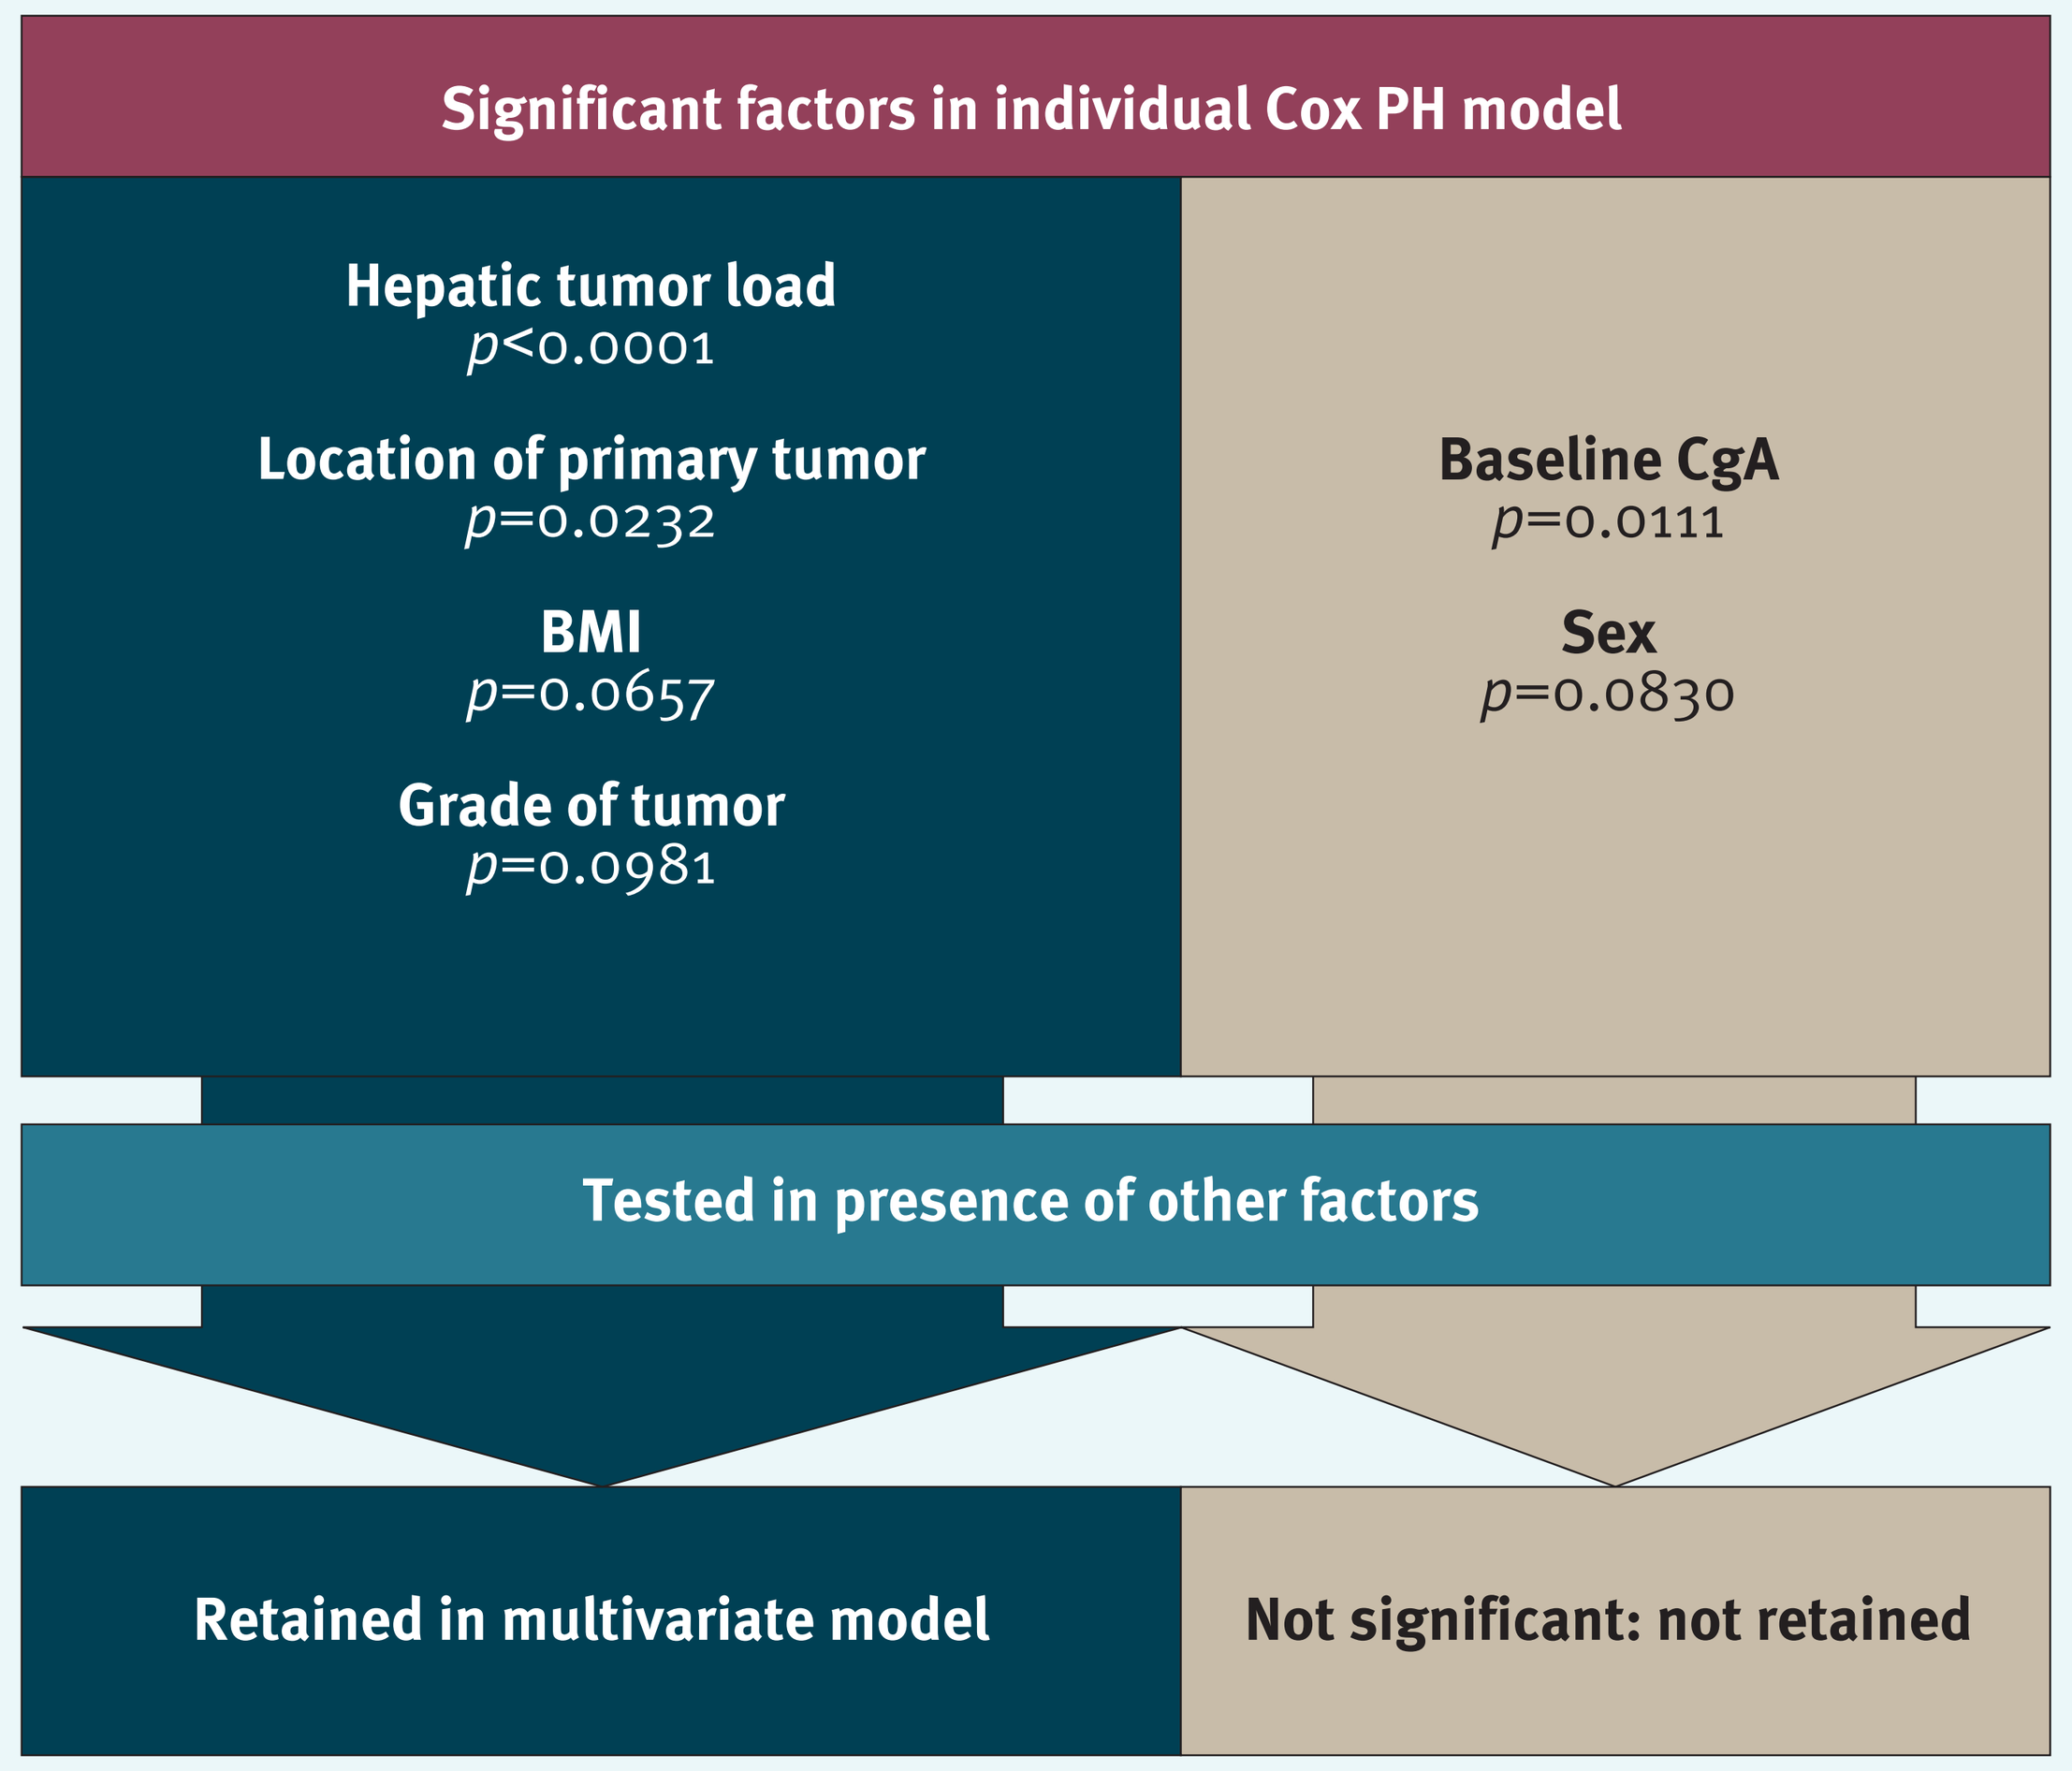
Significant factors in individual Cox PH model
Hepatic tumor load
p<0.0001
Location of primary tumor
p=0.0232
BMI
p=0.0657
Grade of tumor
p=0.0981
Baseline CgA
p=0.0111
Sex
p=0.0830
Tested in presence of other factors
Retained in multivariate model
Not significant: not retained

For categories tested, see Table 1. p-values derived via the Wald Chi-square test statistic; factors significant at $p \leq 0.10$ were considered potentially important. BMI, body mass index; CgA, chromogranin A; PFS, progression-free survival; PH, proportional hazards.

Figure 2. Exploratory analysis of potential prognostic factors for PFS: hazard ratios [95% CI] for treatment from the final multivariate model alongside those from the individual models.

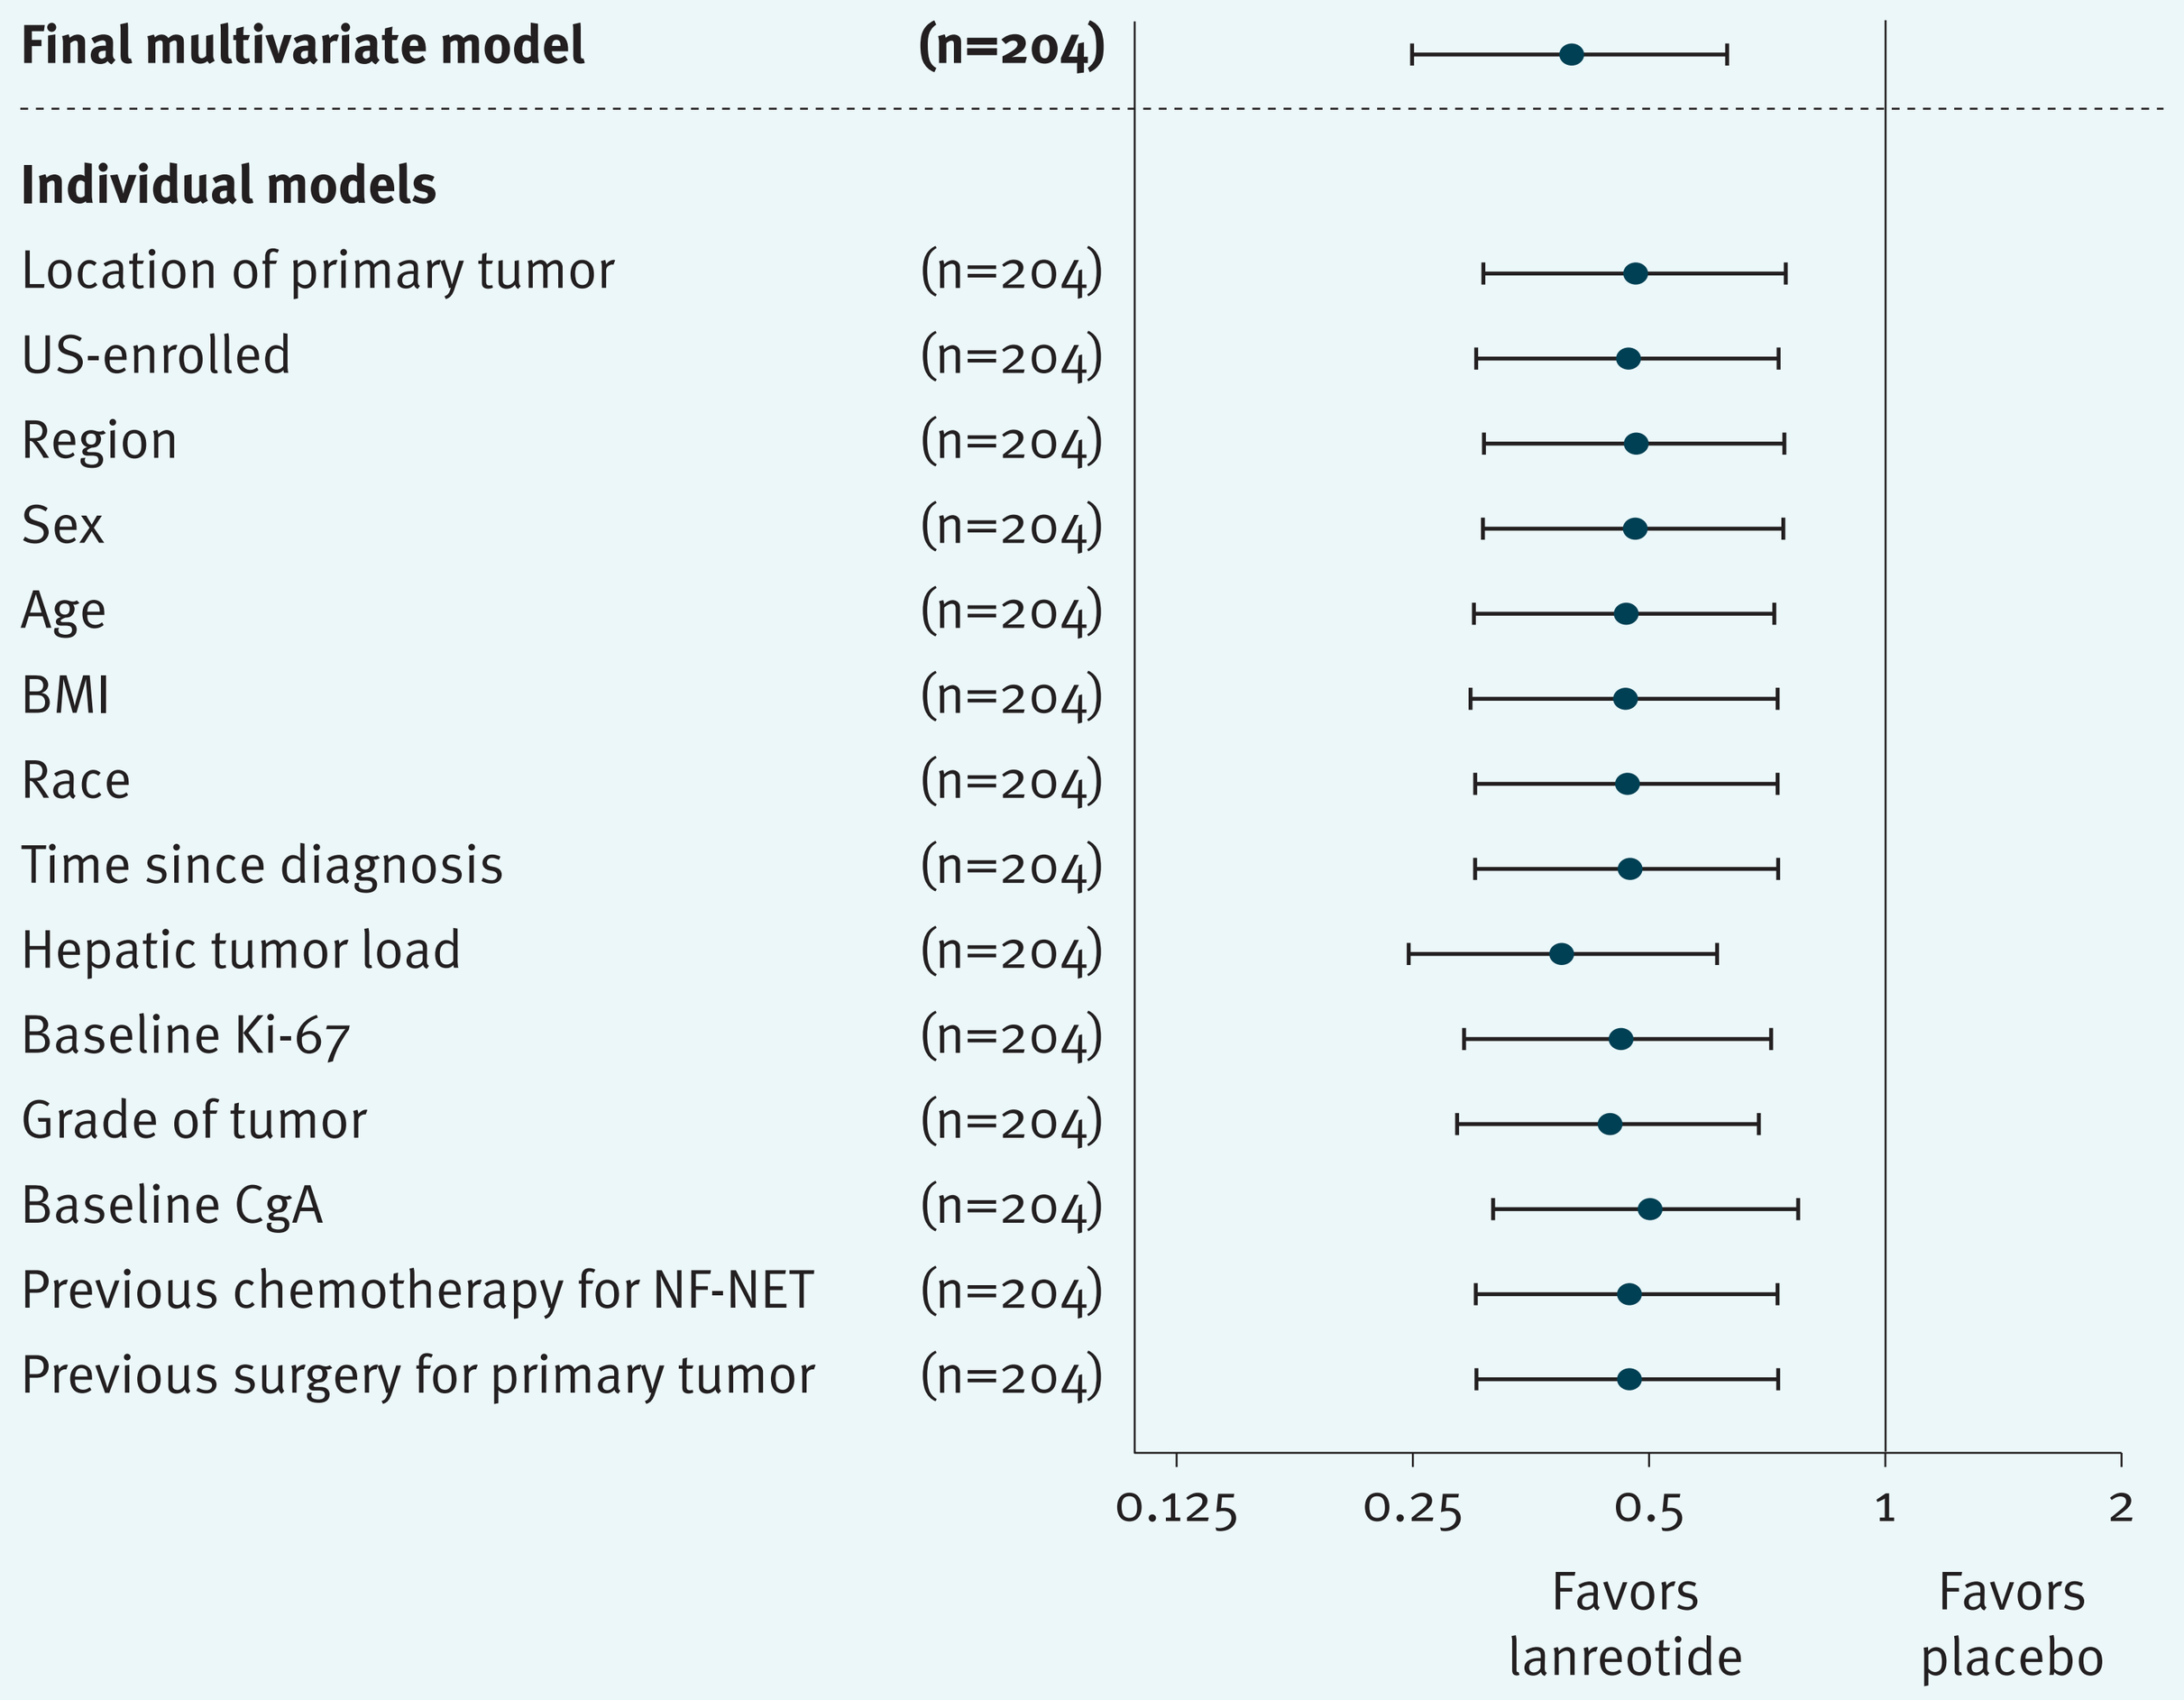
Final multivariate model
(n=204)
Individual models
Location of primary tumor (n=204)
US-enrolled (n=204)
Region (n=204)
Sex (n=204)
Age (n=204)
BMI (n=204)
Race (n=204)
Time since diagnosis (n=204)
Hepatic tumor load (n=204)
Baseline Ki-67 (n=204)
Grade of tumor (n=204)
Baseline CgA (n=204)
Previous chemotherapy for NF-NET (n=204)
Previous surgery for primary tumor (n=204)
0.125
0.25
0.5
1
2
Favors lanreotide
Favors placebo

Analyses conducted with the intent-to-treat population. BMI, body mass index; CgA, chromogranin A; NF-NET, non-functioning (enteropancreatic) neuroendocrine tumor; PFS, progression-free survival.

 - Of the other potential prognostic factors – hepatic tumor load >25% was associated with a 2–3-fold increased risk of PD/death vs. no hepatic tumor load; primary pancreatic tumors increased the risk by 20–60% vs. hindgut, midgut, and other/unknown tumor locations); and below-median BMI increased the risk by 36% vs. above-median BMI (**Table 2**).

Table 2. Exploratory analysis of potential prognostic factors for PFS: final multivariate model.

Term (reference)	Parameter estimate (SE)*	Wald Chi-square	*p* value	HR [95% CI]
Lanreotide (placebo)	−0.92 (0.24)	15.0	<0.0001	0.40 [0.25, 0.63]
Progressive disease (no)	1.52 (0.52)	8.7	0.0032	4.57 [1.67, 12.54]
Prior therapy (no)	0.25 (0.30)	0.7	0.3914	1.29 [0.72, 2.31]
Hepatic tumor load, % (0):		19.9	0.0005	
>0, ≤10	−0.21 (0.34)			0.81 [0.42, 1.59]
>10, ≤25	0.20 (0.37)			1.22 [0.59, 2.52]
>25, ≤50	1.04 (0.35)			2.82 [1.41, 5.63]
>50	0.90 (0.36)			2.47 [1.21, 5.03]
Primary tumor type (pancreas):		9.0	0.0289	
Hindgut	−0.22 (0.45)			0.80 [0.33, 1.94]
Midgut	−0.64 (0.26)			0.53 [0.32, 0.88]
Other/unknown	−0.95 (0.41)			0.39 [0.17, 0.86]
BMI (≤median: 26.2 kg/m^2)	−0.45 (0.23)	3.9	0.0483	0.64 [0.41, 1.00]

*Estimates are derived from a multivariate Cox proportional hazards model relative to the reference level; intent-to-treat population. Analysis of PFS considers as events: centrally assessed disease progressions and any deaths reported during the study. BMI, body mass index; HR, hazard ratio; PFS, progression-free survival.

Conclusions

- In the CLARINET study, lanreotide extended PFS compared with placebo across patient subgroups based on selected demographics and disease characteristics at baseline.
- Factors identified as prognostic for PFS were hepatic tumor load and primary tumor location (although, it should be noted that only 7% of patients had hindgut tumors).
- Several factors evaluated (including tumor grade, prior therapy and time since diagnosis) had no significant impact on outcome.
- The risk of PD or death was lowest among patients with no PD at baseline (nevertheless, only 4% of patients had PD).

Acknowledgments

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical for medical writing support (funded by Ipsen) in preparing this poster. We note with sadness that the author Dr Edda Gomez-Panzani passed away during the preparation of this work.

References

1. NCCN guidelines version 1.2015. Available at: www.NCCN.org.
2. Caplin ME *et al. N Engl J Med* 2014;371:224–33.

Presented at the NANETS Annual Meeting, 15–17 October 2015, Austin, Texas, USA

This study was sponsored by Ipsen