Simulated-Use Study of a Single-Use Delivery Device for Lanreotide Depot in Untrained Health Care Professionals

Daphne T. Adelman[1]; Beloo Mirakhur[2]; Alexandria T. Phan[3]

[1]Feinberg School of Medicine, Northwestern University, Chicago, IL, USA; [2]Ipsen Biopharmaceuticals, Inc., Basking Ridge, NJ, USA; [3]Division of Hematology-Oncology, University of New Mexico Comprehensive Cancer Center, Albuquerque, NM, USA

BACKGROUND

- Somatuline® Depot (lanreotide) Injection is a long-acting somatostatin analog formulation approved in the United States for use in[1]:
 - Long-term treatment of patients with acromegaly who have had an inadequate response to or cannot be treated with surgery and/or radiation therapy
 - Treatment of patients with unresectable, well- or moderately-differentiated, locally advanced or metastatic gastroenteropancreatic neuroendocrine tumors to improve progression-free survival
- Lanreotide, available in 60 mg, 90 mg, and 120 mg doses, is provided in a unique, prefilled, single-use syringe with an integrated sharps injury prevention feature
- The new lanreotide injection device (**Figure**) was designed to improve treatment administration for healthcare providers (HCPs)

Figure. Graphic illustration of the lanreotide device

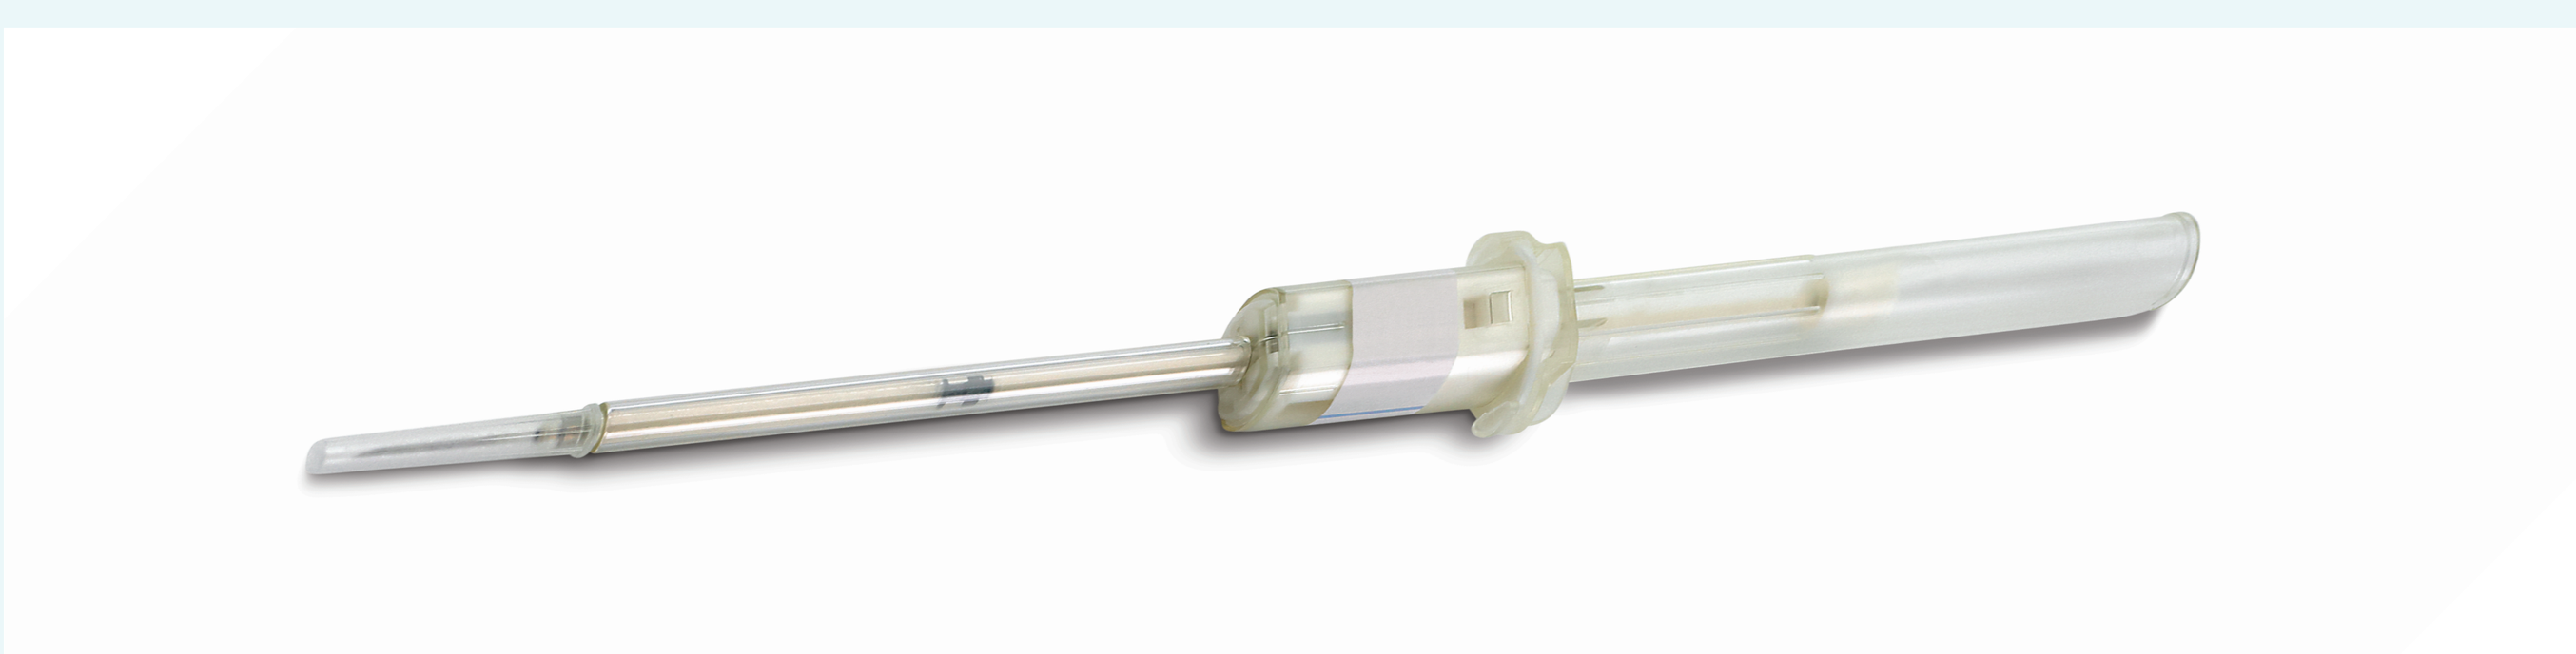

- The design of the lanreotide device was improved to enhance user protection from accidental needle sticks through:
 - Addition of a needle guard using a rigid plastic needle cap
 - Sharps injury prevention feature that automatically retracts the needle once the complete dose has been administered

Human Factors in Medical Devices

- Simulated-use design studies were conducted to identify and mitigate potential risks a user may experience when handling the device[2,3]
- A previous simulated-use study of the lanreotide device observed use events (ie, use errors, near misses, and operational difficulties) in the tasks:[2]
 - Twist and pull to remove plunger protector
 - Continue compressing plunger to bottom
 - Maintain pressure on plunger after delivery of the dose
 - Allow the needle to retract
- Based on the results of the previous simulated-use study[2] and requested changes from the Food and Drug Administration (FDA), modifications were made, primarily to the instructions for use (IFU), to enhance the usability of the lanreotide device (**Table 1**)

Table 1. Changes Made in Device and IFU Based on Previous Study Findings

Device-Related Changes
Label added on outside of plunger protector prompting users to remove the plunger protector before use
IFU-Related Changes
Specified that the user has to check the dose/expiration date on at least 1 of 3 locations: the primary container closure (ie, actual syringe), aluminum sachet (ie, the pouch), or external packaging (the box/carton)
Specified to insert the needle at a 90° angle
Included safety information emphasizing the importance of:
Selecting the correct injection site
Inserting the needle to full depth
Increased importance of removal of plunger protector before attempting to inject
Increased emphasis on continuing to compress plunger to bottom for full dose
Increased importance of maintaining pressure on plunger to activate needle retraction
Increased emphasis on allowing needle to retract
Instruction added stating Somatuline® Depot should be administered by healthcare professionals

OBJECTIVES

- A simulated-use design validation study was conducted to evaluate the effects of the changes in the IFU on usability of the lanreotide device in a group of HCPs who did not receive training in the use of the device

Aims

- To determine if the changes made to the lanreotide device and IFU enhance the "usability" for the intended users, uses, and use environments

METHODS

- This study adhered to FDA guidance[4] and included the following elements:
 - Identified and recruited representative HCP users who were untrained in the use of the lanreotide device
 - Testing was performed in an environment that mimicked actual use
 - Participants were provided the IFU and proceeded with performing tasks without assistance
 - Performance was monitored for evidence of use errors and near misses
 - Participants were engaged in assessment-of-use issues

Subjects

- Participants were HCPs who did not receive training in the use of the lanreotide device

Study Design

- Participants performed a series of tasks while their behavior was recorded
- Participants were given the product packaging materials, prefilled device, and the IFU (**Table 2**)

Table 2. Structure of the Testing Session

Participants completed a one-on-one testing session up to 30 minutes in duration
Participants received the materials they would receive upon delivery of the product:
The prefilled syringe with integrated sharps injury prevention feature
Written and illustrated IFU
Outer packaging (box)
Inner packaging (pouch)
The drug facts information
HCPs were asked to read the IFU and perform one injection with the device (filled with gel to stimulate injection characteristics of the actual product) using a mannequin and injection pad
Before injecting, participants were asked how they would position the mannequin for the injection and the mannequin was placed into position
Participants were asked to place the injection pad on the mannequin at their intended injection site
This allowed the assessment of participants' understanding of proper positioning and injection location

Data Collection

- Direct observation of behavior and subjective participant assessments
- The percentage of participants who successfully completed each task related to specific changes in the IFU and device was reported

RESULTS

- 81% of participants were female, 63% were right handed, and age ranged from 26 to 68 years old (**Table 3**)
- 16 HCPs of various disciplines, ranging from medical assistant to physician, were enrolled and completed the study (**Table 4**)

Demographics

Table 3. Participant Demographics

Gender, n (%)	
Female	13 (81)
Male	3 (19)
Handedness, n (%)	
Right handed	10 (63)
Left handed	5 (31)
Ambidextrous	1 (6)
Age in years, n (%)	
18-30	2 (12)
31-40	3 (19)
41-50	4 (25)
51-60	4 (25)
61-70	3 (19)

Table 4. Participant Employment Role and Educational Level

Employment Role	N (%)	Educational Level
Medical assistant	2 (13)	Associate degree or trade school
Licensed practical nurse	1 (6)	Some college/university
Nurse	9 (56)	Associate degree or trade school (2) College/university degree (4) Master's degree (3)
Physician assistant	1 (6)	College/university degree
Physician	3 (19)	MD/DO degree

Task Completion

Table 5. Percentage of Untrained HCPs With Successful Completion of Device Critical Tasks

Task	Percentage of HCPs With Successful Completion % (n)
1. Choose injection site	100 (16)
2. Verify dose/date	100 (16)
3. Twist and pull to remove plunger protector	100 (16)
4. Insert needle at 90°	100 (16)
5. Penetrate to full length of needle	100 (16)
6. Continue compressing plunger to bottom	100 (16)
7. Maintain pressure on plunger after delivery of the dose	88 (14)
8. Allow the needle to retract	100 (16)

Tasks 1 – 6 and Task 8

- All of the participants successfully completed these tasks (**Table 5**)
 - Checked the dose/date in at least 1 location
 - Selected the correct injection site
 - Removed the plunger protector
 - Inserted the needle at 90°, penetrated to the full length of the needle, and compressed the plunger to the bottom in order to inject all of the medication
 - Allowed the needle to retract, placing the needle in a safe state

Task 7 Maintain Pressure on Plunger

- Most participants (88% [14/16]) correctly maintained pressure on the plunger after delivery of a full dose, which keeps the needle extended until it is removed from the patient's body (**Table 5**)
 - Not maintaining pressure after delivery of a full dose has no measurable clinical impact
 - Results in automatic retraction of the needle, placing the device in a safe state automatically
 - It is not possible to activate the safety mechanism prior to delivery of the full dose

CONCLUSIONS

- Changes to the IFU proposed by the FDA and addition of the label to the plunger protector successfully mitigated previously observed use events and improved use of the prefilled lanreotide injection device by untrained HCPs
- Untrained HCPs were able to successfully use the prefilled lanreotide depot injection device, with no observed events with measurable clinical impact
- In this simulated-use study, lanreotide injection device with the integrated sharps injury-prevention feature was shown to have acceptable "usability" for the intended users, uses, and use environments

ACKNOWLEDGMENTS

Funding for development, editorial, design and production was provided by Ipsen Biopharmaceuticals, Inc., to MedVal Scientific Information Services, LLC, Skillman, NJ, USA.

Corresponding author: Daphne T. Adelman, Feinberg School of Medicine, Northwestern University, Chicago, IL; d-adelman@northwestern.edu

REFERENCES

1. Somatuline® Depot (lanreotide) Injection [prescribing information]. Basking Ridge, NJ. Ipsen Biopharmaceuticals, Inc.; 2014.
2. Smith SL, et al. ENDO 95th Annual Meeting, June 15-18, 2013, San Francisco, CA. Poster SUN-288.
3. Smith SL, et al. ENDO 95th Annual Meeting, June 15-18, 2013, San Francisco, CA. Poster SUN-287.
4. US Department of Health and Human Services. Center for Devices and Radiological Health. Applying Human Factors and Usability Engineering to Medical Devices: Guidance for Industry and Food and Drug Administration Staff. February 3, 2016. Available at: http://www.fda.gov/downloads/MedicalDevices/DeviceRegulationandGuidance/GuidanceDocuments/UCM259760. Accessed March 11, 2016

DISCLOSURES

DTA: Consultant, Chiasma, Ipsen, Novartis, Pfizer; Speakers bureau, Pfizer

BM: Employee, Ipsen

AP: Consultant, Ipsen, Novartis; Speakers bureau, Celgene, Eli Lilly, Genentech, Ipsen, Novartis; Research support, Incyte, Ipsen, Lexicon, Novartis, Sanofi

Poster presented at the 2016 NANETS Symposium, September 30–October 1, 2016, Jackson, WY