Efficacy of lanreotide depot/autogel for symptomatic control of carcinoid syndrome (CS) in neuroendocrine tumor patients: follow-up analysis of the ELECT prospective, randomized, double-blind and open-label phases

155

George A Fisher, Jr,[1] Edward M Wolin,[2] Pamela L Kunz,[1] Nilani Liyanage,[3] Beloo Mirakhur,[4] Susan Pitman Lowenthal,[4] Rodney F Pommier,[5] Montaser Shaheen,[6] Aaron Vinik[7] on behalf of ELECT study investigators

[1]Stanford University School of Medicine, Stanford, CA, USA; [2]Montefiore Einstein Center for Cancer Care, Bronx, NY, USA; [3]Ipsen, Boulogne-Billancourt, France; [4]Ipsen, Basking Ridge, NJ, USA; [5]Oregon Health & Science University, Portland, OR, USA; [6]University of New Mexico Cancer Center, Albuquerque, NM, USA; [7]Eastern Virginia Medical School, Norfolk, VA, USA

INTRODUCTION

- The symptoms of carcinoid syndrome (CS), diarrhea and flushing in particular, have a significant negative impact on patient quality of life[1]
- Long-acting somatostatin analogs (SSAs) are recommended in treatment guidelines as first-line medical therapy for symptomatic control of CS in patients with neuroendocrine tumors (NETs)[2,3]
- Lanreotide depot, known as lanreotide autogel outside of the US, is a long-acting SSA indicated in the US and Europe for antiproliferative treatment in patients with locally advanced or metastatic gastroenteropancreatic NETs (GEP-NETs) and in Europe for the treatment of CS symptoms[4]
- The multinational, phase 3 ELECT (NCT00774930; EudraCT 2010-019066-92) study evaluated the efficacy and safety of subcutaneous (SC) lanreotide depot/autogel 120 mg every 4 weeks vs placebo for symptomatic control of CS in NET patients. The use of short-acting octreotide rescue medication was the primary outcome measure[5]
- ELECT met its primary endpoint demonstrating that lanreotide depot/autogel significantly reduced the percentage of days that short-acting octreotide rescue medication was used for symptomatic control of CS in patients vs placebo (34% vs 49%, P=0.02)[5]
- These additional analyses present the patient-reported symptom outcomes, biochemical response, and safety from the ELECT study

OBJECTIVES

- The objective of this post-hoc analysis was to examine patient-reported symptom data from the 16-week double-blind (DB) phase and 32-week initial open-label (IOL) phase of the ELECT study
- Also included are post-hoc analyses of biochemical response (urinary 5-hydroxyindoleacetic acid [5-HIAA]) as well as safety data during the IOL phase

METHODS

Study design

- ELECT was a phase 3, multinational study consisting of a 1-month screening phase, 16-week DB phase, and 32-week IOL phase followed by a long-term open-label extension (≥2 years) (**Figure 1**)[5]

Figure 1. ELECT study design

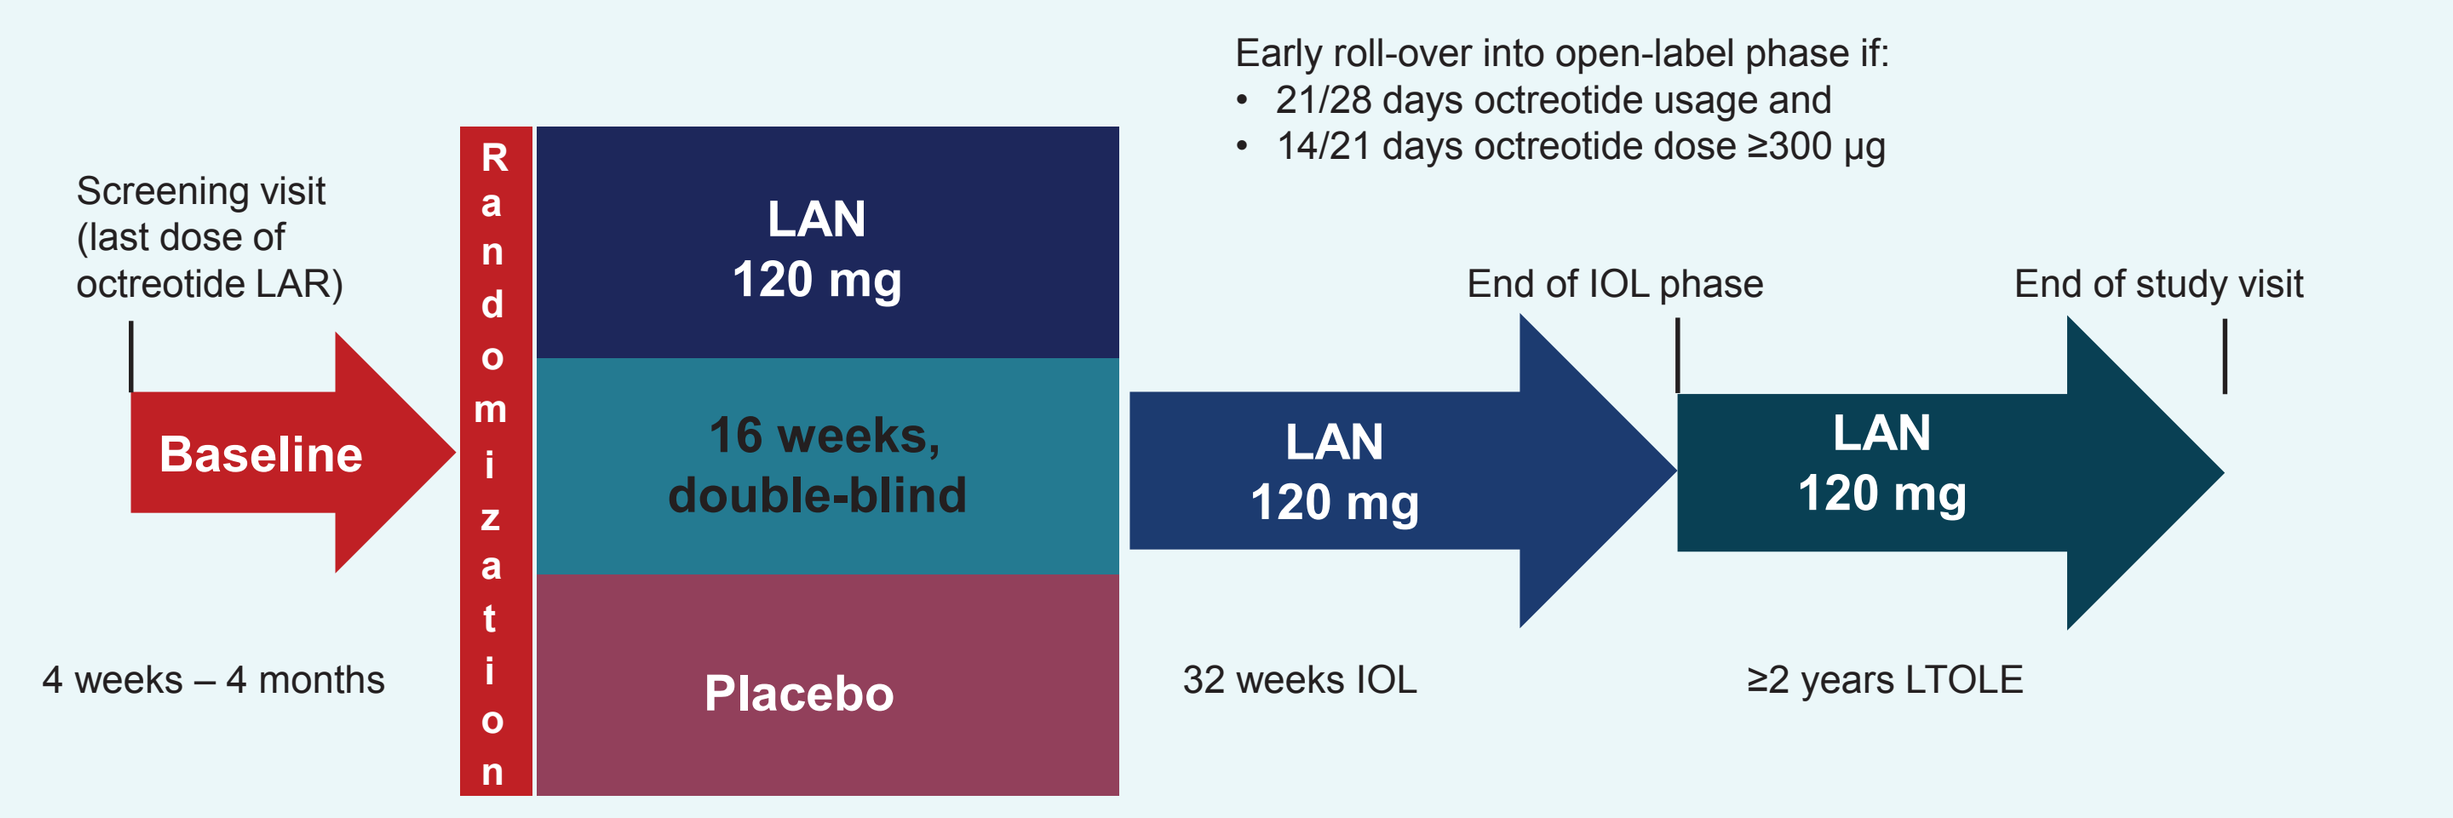
Early roll-over into open-label phase if:
• 21/28 days octreotide usage and
• 14/21 days octreotide dose ≥300 µg
Screening visit (last dose of octreotide LAR)
Baseline
4 weeks – 4 months
Randomization
LAN 120 mg
16 weeks, double-blind
Placebo
LAN 120 mg
32 weeks IOL
End of IOL phase
LAN 120 mg
≥2 years LTOLE
End of study visit

ELECT, Evaluation of Lanreotide Autogel/Depot Efficacy and Safety for Carcinoid Syndrome Treatment; IOL, initial open-label; LAN, lanreotide depot/autogel; LTOLE, long-term open-label extension; LAR, long-acting release.

- Included patients were ≥18 years of age with a histopathologically-confirmed NET or NET of unknown location with liver metastases and history of CS (flushing and/or diarrhea)
- Eligible patients who were either SSA-naïve or responsive to octreotide long-acting release (LAR) (≤30 mg every 4 weeks) or short-acting SC octreotide (≤600 µg daily) were randomized 1:1 to receive SC lanreotide depot/autogel 120 mg or placebo every 4 weeks
- Patients were allowed to self-treat with short-acting SC octreotide as rescue medication for symptom control throughout the DB and open-label phases

Assessments

- In a daily diary, patients recorded the presence (yes/no), frequency (1-50), and severity (mild, moderate, severe) of diarrhea and flushing events using Interactive Voice/Web Response System (IVRS/IWRS) for 1 month pre-randomization and throughout the DB and IOL phases
- Urinary 5-HIAA levels were assessed at baseline and every 12 weeks in the DB and IOL phases for a total of 48 weeks
 - Patients were instructed to avoid a list of food and drugs known to interact with 5-HIAA for 24 hours prior to initiation of the 24-hour urine collection
- Safety assessments during the IOL included recorded adverse events (AEs) and serious AEs (SAEs) by the investigator
 - Diarrhea and flushing events were not captured as AEs as they were already included as study assessments

Data analysis

- The post-hoc efficacy analyses included all patients from the intention-to-treat (ITT) population (randomized and open-label)
- For this analysis, patient-reported symptom data were combined to provide an average daily composite score based on daily frequency and severity of symptoms
 - An average daily symptom score was calculated for each patient per day by multiplying the daily symptom frequency × the daily severity (0=none, 1=mild, 2=moderate, and 3=severe) and dividing by the total number of days that a diary entry was recorded during the DB and IOL phases
- Analysis of covariance (ANCOVA) was used to analyze patient symptom data during the DB phase, with fixed effect terms for treatment group, prior SSA use, and country, and baseline diarrhea and flushing events as continuous variables
- Post-hoc analyses of changes in 5-HIAA from baseline during DB phase were assessed using ANCOVA with fixed effect terms for treatment group, prior SSA use, and country, and 5-HIAA baseline values as a continuous variable
 - Given the skewness of data, 5-HIAA values were log transformed
- Comparison of patient symptoms and 5-HIAA changes between the DB and IOL phases were analyzed using Paired t-tests

RESULTS

Patients

- 115 patients were randomized (n=59 lanreotide depot/autogel, n=56 placebo) in the DB phase, and of those, 101 continued in the IOL phase, including:
 - 56 lanreotide-treated patients (LAN DB/LAN IOL)
 - 45 placebo-treated patients (PBO DB/LAN IOL)
- At baseline (during screening pre-randomization), the percentage of patient-reported days with severe diarrhea (5.13% [95% CI: 1.58, 8.69] vs 3.35% [95% CI: 0.74, 5.96]) and severe flushing (2.21% [95% CI: 0.68, 7.73] vs 4.84% [95% CI: 0.78, 8.90]) was low in both treatment groups (lanreotide vs placebo), with no significant differences between groups
- Demographic and clinical characteristics of patients in the IOL phase by initial treatment group are shown in **Table 1**

Table 1. Demographics and clinical characteristics for patients who entered the IOL phase

	LAN n=56	Placebo n=45
Mean age (SD), years	57.5 (10.7)	59.3 (12.0)
Male	26 (46.4)	18 (40.0)
Region		
US	21 (37.5)	15 (33.3)
Ex-US	35 (62.5)	30 (66.7)
Prior octreotide[a]	33 (58.9)	27 (60.0)
Time since first symptoms to treatment start		
<1 year	13 (23.2)	15 (33.3)
≥1 year	43 (76.8)	30 (66.7)
Time since first diagnosis to treatment start		
<1 year	18 (32.1)	18 (40.0)
≥1 year	38 (67.9)	27 (60.0)
Any prior octreotide[a] **therapy within 3 months before screening**	28 (50.0)	24 (53.3)
Use of short-acting octreotide during screening	28 (50.0)	23 (51.1)

Data are number (%) unless stated otherwise.
[a]Prior octreotide includes octreotide LAR, short-acting octreotide, or both.
SD, standard deviation; LAN, lanreotide depot/autogel; LAR, long-acting release.

Symptoms during DB and IOL phases

- Patients treated with lanreotide depot/autogel showed greater reductions in adjusted average daily composite symptom scores for diarrhea, flushing, and diarrhea and/or flushing compared with patients treated with placebo during DB phase (**Figure 2**)

Figure 2. LS mean (SE) daily composite symptom scores during DB phase (ANCOVA, ITT population)

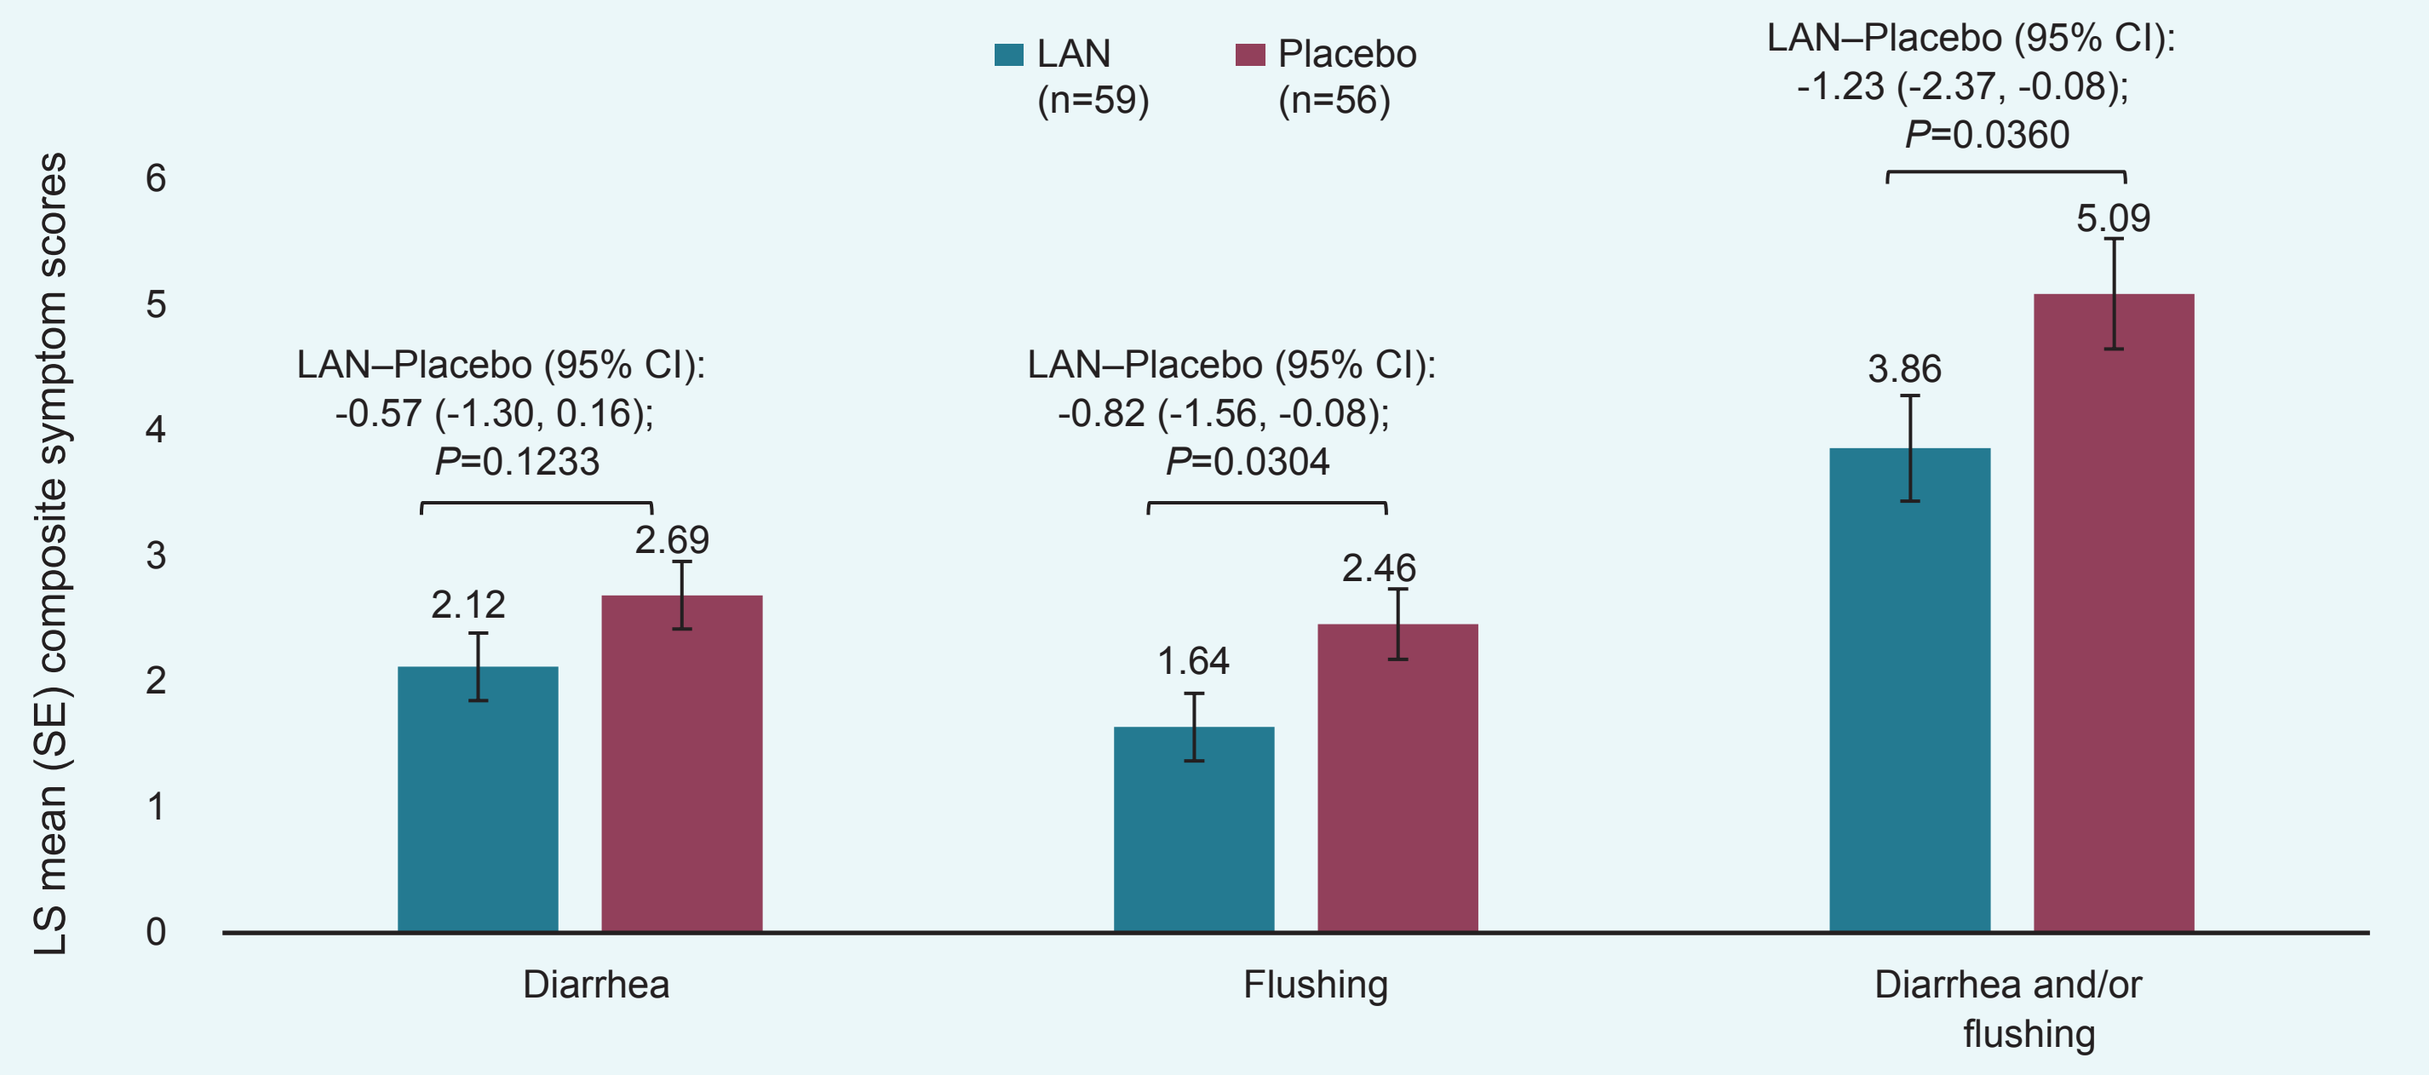
LAN (n=59)
Placebo (n=56)
LS mean (SE) composite symptom scores
0
1
2
3
4
5
6
LAN–Placebo (95% CI): -0.57 (-1.30, 0.16); P=0.1233
2.12
2.69
Diarrhea
LAN–Placebo (95% CI): -0.82 (-1.56, -0.08); P=0.0304
1.64
2.46
Flushing
LAN–Placebo (95% CI): -1.23 (-2.37, -0.08); P=0.0360
3.86
5.09
Diarrhea and/or flushing

ANCOVA, analysis of covariance; CI, confidence interval; DB, double-blind; LAN, lanreotide depot/autogel; ITT, intention-to-treat; LS, least square; SE, standard error.
Note: LS means based on ANCOVA model adjusted for daily average of diarrhea and flushing events at baseline, prior SSA use, and region.

- The adjusted least square (LS) mean percentage of days with moderate/severe diarrhea (15.8% vs 24.6%, P=0.016), moderate/severe flushing (13.2% vs 21.7%, P=0.012), moderate/severe diarrhea and/or flushing (23.4% vs 35.8%, P=0.004), and severe diarrhea and/or flushing (3.9% vs 7.6%, P=0.023) were significantly lower for patients treated with lanreotide depot/autogel vs placebo, respectively (**Figure 3**)

Figure 3. LS mean (SE) percentage of days that A) moderate/severe symptoms and B) severe symptoms were experienced during DB phase (ANCOVA, ITT Population)

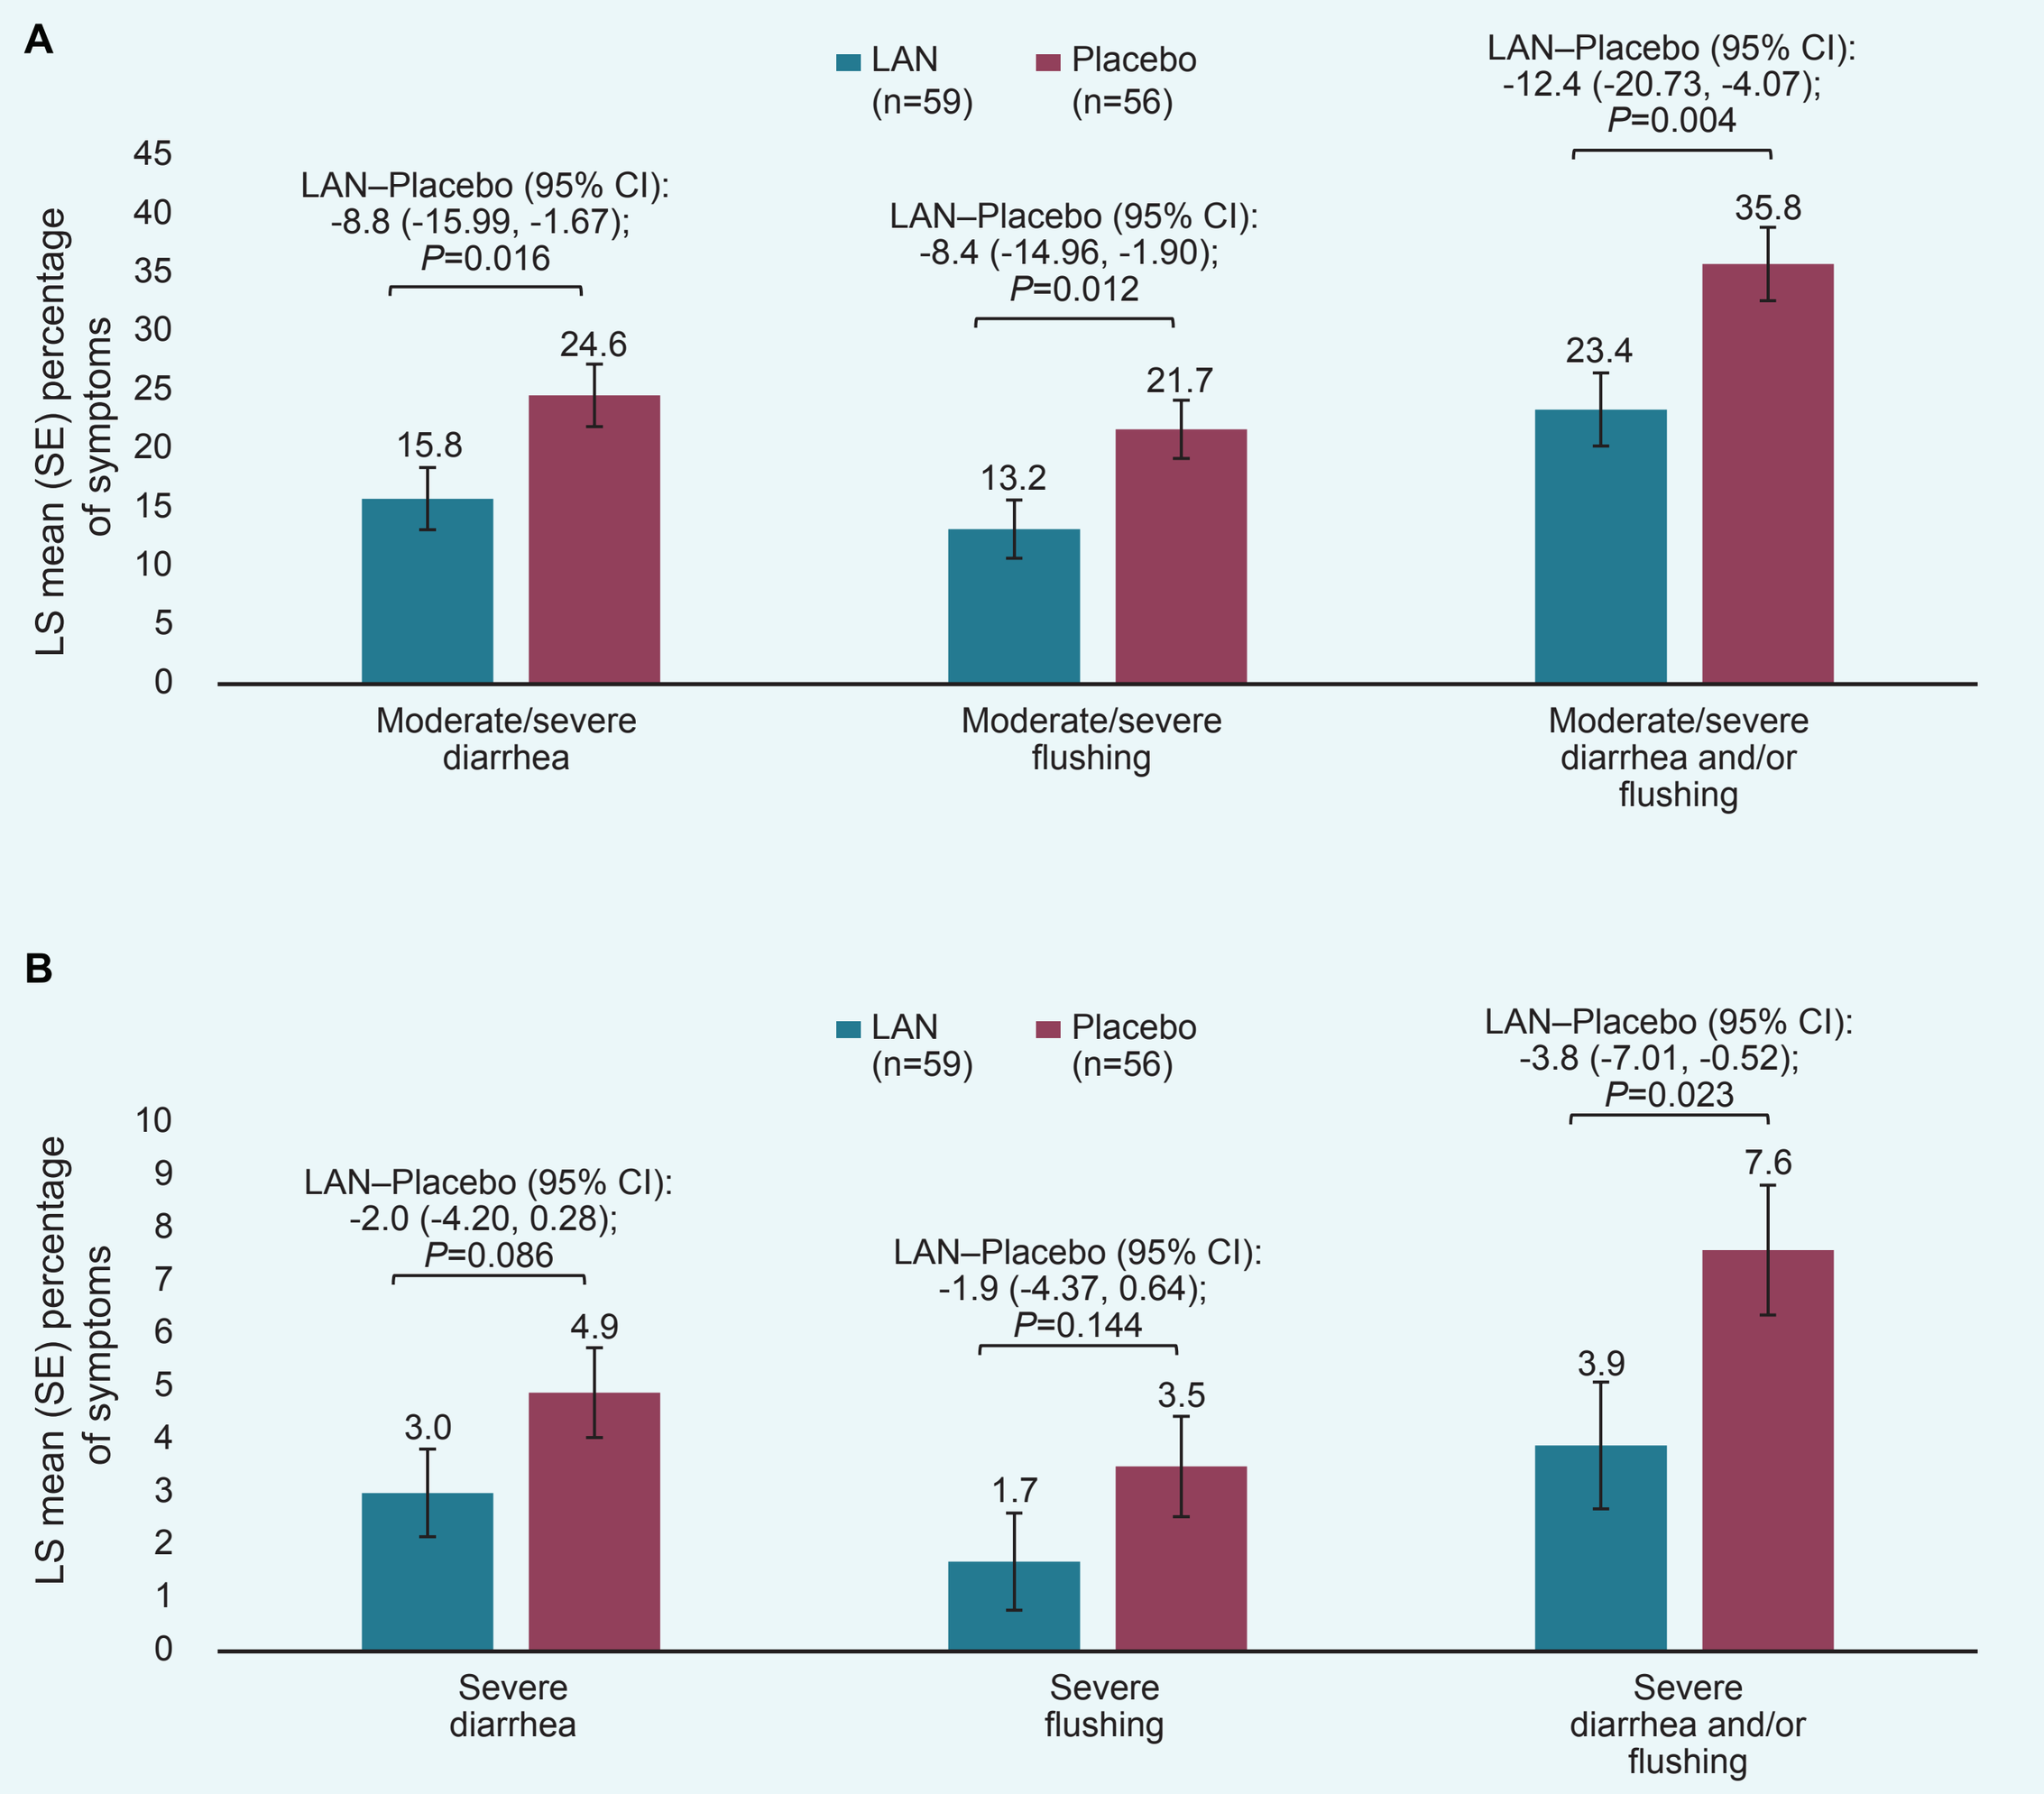
A
LAN (n=59)
Placebo (n=56)
LS mean (SE) percentage of symptoms
0
5
10
15
20
25
30
35
40
45
LAN–Placebo (95% CI): -8.8 (-15.99, -1.67); P=0.016
15.8
24.6
Moderate/severe diarrhea
LAN–Placebo (95% CI): -8.4 (-14.96, -1.90); P=0.012
13.2
21.7
Moderate/severe flushing
LAN–Placebo (95% CI): -12.4 (-20.73, -4.07); P=0.004
23.4
35.8
Moderate/severe diarrhea and/or flushing
B
LAN (n=59)
Placebo (n=56)
LS mean (SE) percentage of symptoms
0
1
2
3
4
5
6
7
8
9
10
LAN–Placebo (95% CI): -2.0 (-4.20, 0.28); P=0.086
3.0
4.9
Severe diarrhea
LAN–Placebo (95% CI): -1.9 (-4.37, 0.64); P=0.144
1.7
3.5
Severe flushing
LAN–Placebo (95% CI): -3.8 (-7.01, -0.52); P=0.023
3.9
7.6
Severe diarrhea and/or flushing

ANCOVA, analysis of covariance; CI, confidence interval; DB, double-blind; LAN, lanreotide depot/autogel; ITT, intention-to-treat; LS, least square; SE, standard error.
Note: LS means are based on ANCOVA model adjusted for prior SSA use, region, and baseline values.

- Among LAN DB/LAN IOL patients, the mean daily composite symptom scores for diarrhea improved significantly from the DB to IOL phase (mean difference -0.71 [95% CI: -1.20, -0.22]; P=0.0054), and the mean composite symptom scores for flushing and diarrhea and/or flushing remained stable from the DB to IOL phase (P-values >0.05) (**Figure 4**)
- Among PBO DB/LAN IOL patients, the mean difference in composite symptom scores for diarrhea, flushing, and diarrhea and/or flushing reflected significant improvement from the DB phase to the end of the IOL phase (P-values <0.05) (**Figure 4**)

Figure 4. Difference in average daily composite symptom scores for patients in DB and IOL phases according to initial DB treatment group (Paired t-test, ITT population, n=101)

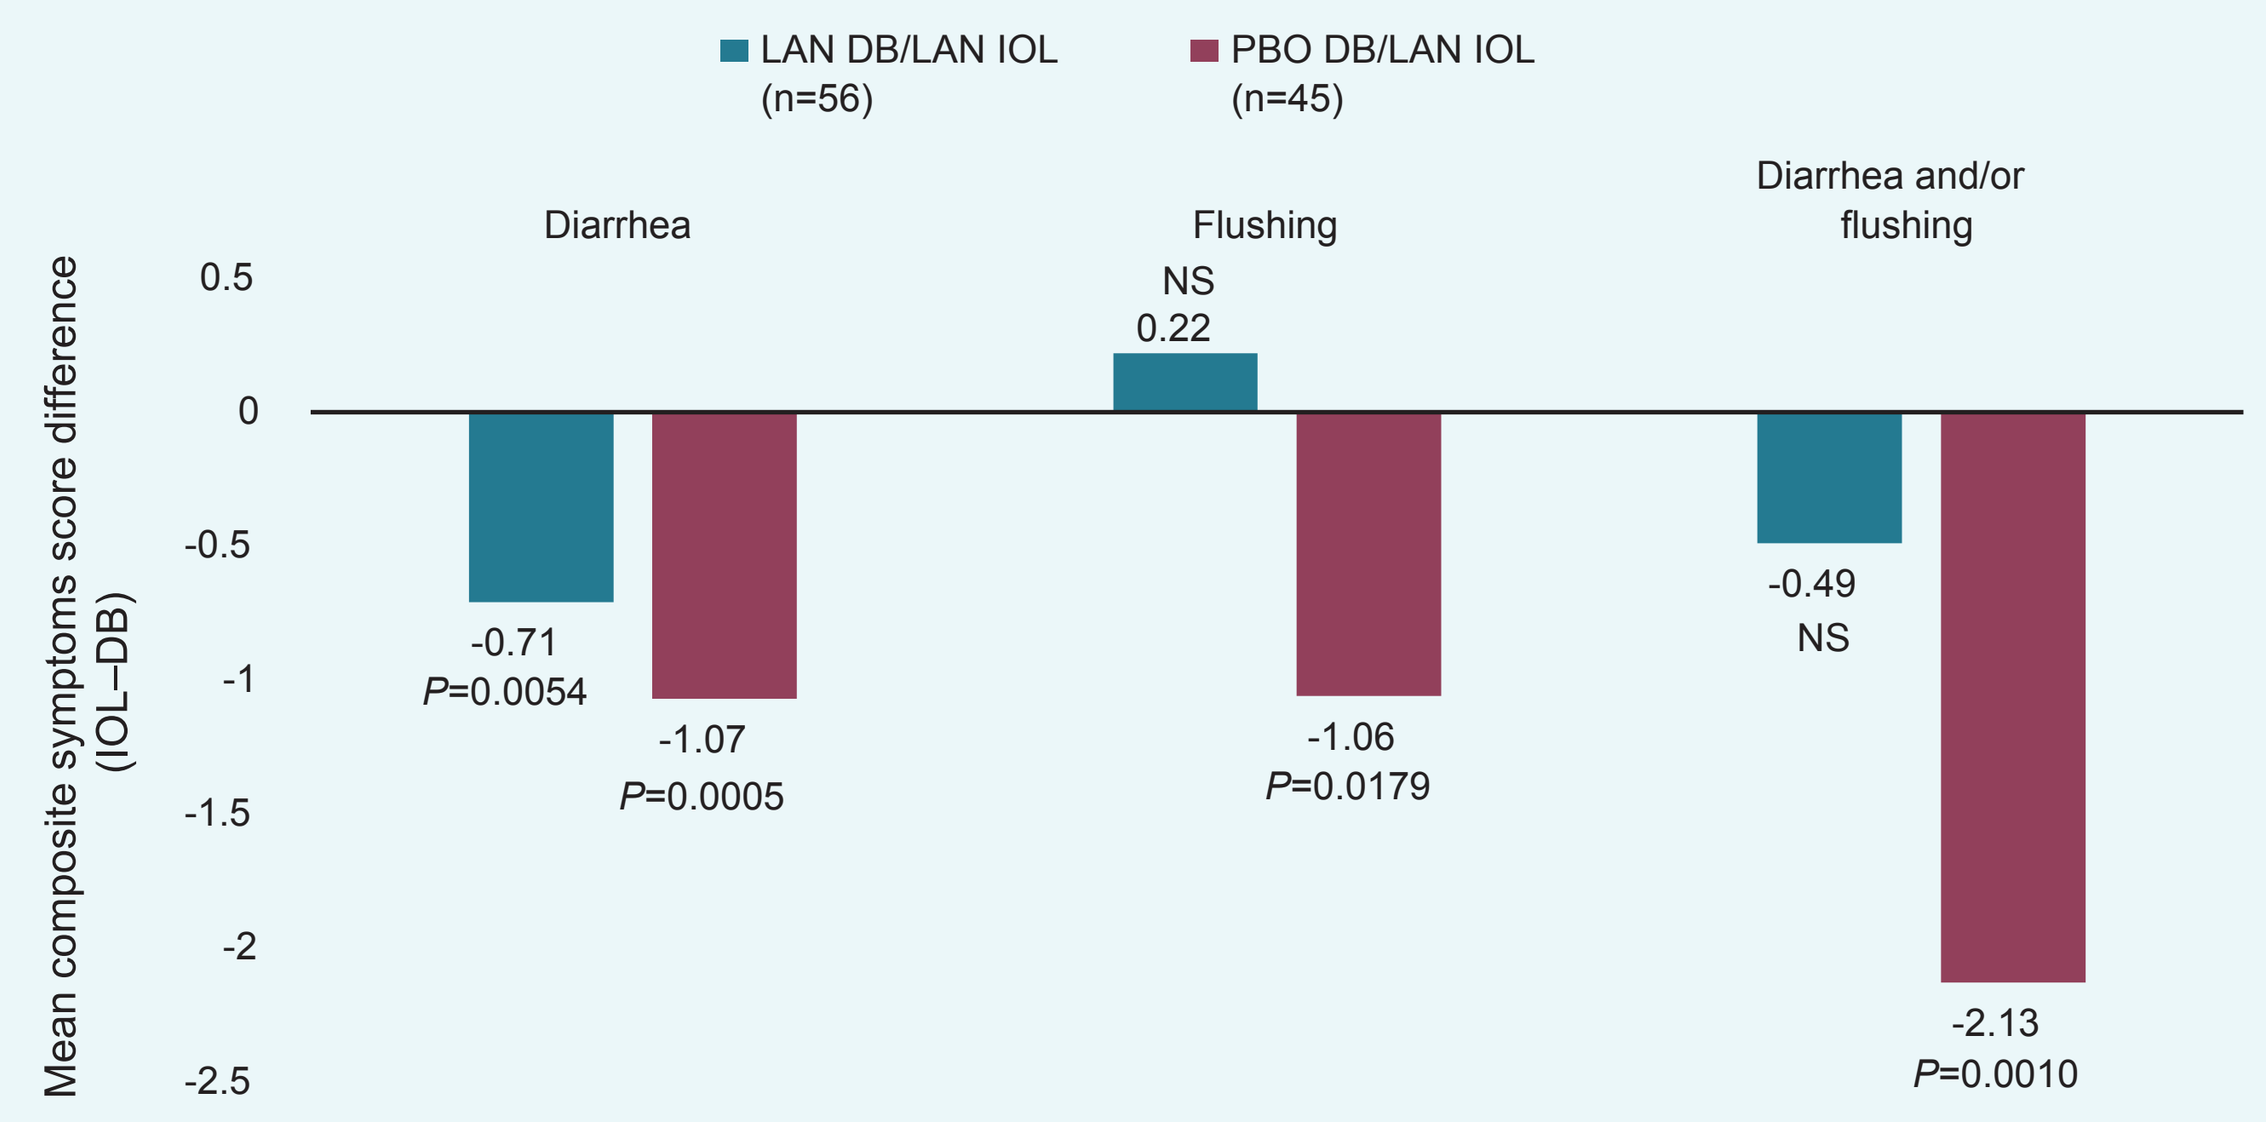
LAN DB/LAN IOL (n=56)
PBO DB/LAN IOL (n=45)
Mean composite symptoms score difference (IOL–DB)
0.5
0
-0.5
-1
-1.5
-2
-2.5
Diarrhea
-0.71 P=0.0054
-1.07 P=0.0005
Flushing
NS 0.22
-1.06 P=0.0179
Diarrhea and/or flushing
-0.49 NS
-2.13 P=0.0010

DB, double-blind; LAN, lanreotide depot/autogel; IOL, initial open-label; ITT, intention-to-treat; PBO, placebo; NS, not significant.

5-HIAA during DB and IOL phases

- During DB treatment, the lanreotide group (n=38) had a 36% greater decrease in adjusted absolute logarithmic urinary 5-HIAA levels at week 12 compared with the placebo group (n=27) (relative mean ratio 0.64 [95% CI: 0.39, 1.04])
- In general, urinary 5-HIAA changes from baseline compared between each IOL visit and DB (week 12) for LAN DB/LAN IOL patients reflected decreases in 5-HIAA along with wide CIs (**Table 2**), which suggests that 5-HIAA levels remained stable during the IOL phase
- In PBO DB/LAN IOL patients, urinary 5-HIAA changes from baseline compared between each IOL visit and DB (week 12) reflected a decrease in 5-HIAA at week 24, which remained comparable at weeks 36 and 48 but with wider CIs

Table 2. Changes from baseline in urinary 5-HIAA in DB and IOL phases

5-HIAA, µmol/d	LAN DB/LAN IOL	PBO DB/LAN IOL
Week 12		
n	43	29
Mean (SD)	179.5 (388.0)	183.9 (228.6)
95% CI for mean	60.1, 298.9	97.0, 270.9
Week 24 (IOL) – Week 12 (DB)		
n	38	25
Mean difference (95% CI)	21.6 (-39.2, 82.4)	-81.3 (-149.7, -13.0)
Week 36 (IOL) – Week 12 (DB)		
n	35	25
Mean difference (95% CI)	14.6 (-21.5, 50.8)	-75.2 (-151.9, 1.5)
Week 48 (IOL) – Week 12 (DB)		
n	33	27
Mean difference (95% CI)	-14.4 (-54.8, 26.0)	-71.3 (-150.4, 7.8)

5-HIAA, 5-hydroxyindoleacetic acid; CI, confidence interval; DB, double-blind; LAN, lanreotide depot/autogel; IOL, initial open-label; PBO, placebo; SD, standard deviation.

Safety

- The frequency of AEs during the IOL phase by initial DB group (LAN DB/LAN IOL vs PBO DB/LAN IOL) was 69.6% vs 71.1%
 - The most common AEs (≥10%) that occurred during the IOL phase among LAN DB/LAN IOL patients were: abdominal pain (14.3% [8/56]), weight decreased (10.7% [6/56]), dyspnea (10.7% [6/56]), and hypertension (10.7% [6/56])
 - The most common AEs (≥10%) that occurred during the IOL phase among PBO DB/LAN IOL patients were: nausea (11.1% [5/45]), fatigue (11.1% [5/45]), and headache (11.1% [5/45])
- The majority of AEs were mild to moderate in severity; severe AEs during the IOL phase that were considered related to treatment were reported for 2 LAN DB/LAN IOL patients
- 7 (12.5%) LAN DB/LAN IOL patients experienced 14 SAEs during the IOL and 1 (2.2%) PBO DB/LAN IOL patient experienced 4 SAEs during the IOL; none were related to treatment

DISCUSSION/CONCLUSIONS

- In the 16-week primary ELECT study, lanreotide depot/autogel (SC 120 mg every 4 weeks) significantly reduced the use of short-acting octreotide for rescue symptom control in patients with CS vs placebo[5]
- This post-hoc analysis of patient-reported symptom data from the ELECT study found improvement in patient-reported diarrhea and flushing during the initial 16 weeks (DB phase) of lanreotide treatment, which was sustained for an additional 32 weeks (through week 48 of the IOL phase)
- As expected, patients initially treated with placebo in the DB phase showed significant improvement in composite symptom scores in the IOL phase, comparable to the improvement reported during randomized treatment with lanreotide depot/autogel in the DB phase
- Additional post-hoc analyses found rapid decreases in urinary 5-HIAA levels at week 12 during DB treatment with lanreotide depot/autogel, which were sustained in the IOL phase
- Lanreotide depot/autogel was generally well-tolerated during the IOL phase and comparable to the AE profile observed during the DB phase, indicating a sustained safety profile with continued use
- Results from the open-label extension study (≥2 years) will provide additional long-term safety data for lanreotide depot/autogel

REFERENCES

1. Beaumont JL, et al. *Pancreas.* 2012;41:461-6.
2. Kunz PL, et al. *Pancreas.* 2013;42:557-77.
3. Oberg K, et al. *Ann Oncol.* 2012;23(suppl 7):vii124-vii130.
4. US Food and Drug Administration. Lanreotide. http://www.fda.gov/Drugs/InformationOnDrugs/ApprovedDrugs/ucm427065.htm. Accessed September 19, 2016.
5. Vinik AI, et al. *Endocr Pract.* 2016 May 23. [Epub ahead of print]

ACKNOWLEDGMENTS

This study was funded by Ipsen Biopharmaceuticals, Inc. Development, editorial, design, and production support was provided by MedVal Scientific Information Services, LLC, and was funded by Ipsen Biopharmaceuticals, Inc.

Presented at the 2016 North American Neuroendocrine Tumor Society Symposium, September 30 – October 1, 2016, Jackson, WY