Tumor growth rate in intestinal/pancreatic neuroendocrine tumors: *post hoc* exploratory analysis of data from the CLARINET study

Martyn Caplin,[1] Marianne Pavel,[2] Philippe Ruszniewski,[3] Nilani Liyanage,[4] Christine Massien,[4] Clarisse Dromain,[5] on behalf of the CLARINET Study Group

[1]Royal Free Hospital, London, UK; [2]Charité University Medicine Berlin, Berlin, Germany; [3]Beaujon Hospital, Clichy, France; [4]Ipsen, Boulogne-Billancourt, France; [5]Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland.

Background

- The slow-growing nature of neuroendocrine tumors (NETs) can confound accurate and early assessment of disease status with the widely used response evaluation criteria in solid tumors (RECIST).[1,2]
- New objective measures of tumor response that detect subtle changes in tumor growth or stabilization over a shorter treatment period are needed to complement RECIST.
 - Tumor growth rate (TGR) is one promising candidate.
- The CLARINET study demonstrated anti-tumor efficacy of lanreotide depot (also known as autogel) in patients with intestinal and pancreatic NETs.[3]
 - Centrally assessed tumor measurements (based on RECIST v1.0) were used to determine radiologic progression.
- In these *post hoc* analyses, tumor measurements from CLARINET were re-evaluated to explore the clinical utility of TGR as a:
 - Measure of tumor progression before and in response to treatment.
 - Prognostic factor for tumor progression before treatment.

Methods

Overview of CLARINET

- CLARINET was an international randomized double-blind placebo-controlled phase 3 study (EudraCT: 2005-004904-35; ClinicalTrials.gov: NCT00353496).[3]
- Patients received lanreotide depot 120 mg or placebo every 28 days for 96 weeks or until death or progressive disease (PD; assessed centrally using RECIST v1.0).
- Study visits were scheduled during screening and at weeks 1 (baseline), 12, 24, 36, 48, 72, and 96.
- A computed tomography (CT) scan was performed twice during screening to determine pre-treatment disease-progression status. Single CT scans were obtained thereafter.

Assessments and analyses

- TGR was expressed as the percentage change in tumor volume over 1 month:

$$\mathbf{TGR = 100 \times (exp\ (TG)-1)}$$

$$\mathbf{TG = \frac{3 \times log\ (D2/D1)}{time\ (months)}}$$

D1=tumor size at date 1; D2=tumor size at date 2; and time (months) = (date 2 – date 1 + 1)/30.44.

- Tumor size was determined using the sum of longest diameters (SLD) of target lesions; non-target and new lesions were not considered.
- TGR as a measure of tumor progression was assessed by:
 - Calculating pre-treatment TGR (TGR at screening vs. TGR at baseline).
 - Change in TGR between consecutive treatment visits.
- To investigate TGR as a prognostic factor for tumor progression:
 - The optimum pre-treatment TGR cut-off value (%/month) was determined.
 - Progression-free survival (PFS) was calculated for the optimum pre-treatment TGR cut-off (≤/>4%/month) using RECIST v1.0.
 - Exploratory analyses were conducted to identify potential prognostic factors for PFS.

Statistical analyses

- TGRs prior and in response to treatment (*post hoc* analyses; intention-to-treat [ITT] population)
 - Least square (LS) means [95% confidence intervals, CI] were estimated using mixed-model regression analyses.
 - Pairwise comparisons using the Wilcoxon signed-rank test were performed to test differences in TGR means during treatment.

Figure 1. Distribution of TGRs among individual patients during the screening period according to RECIST classification.

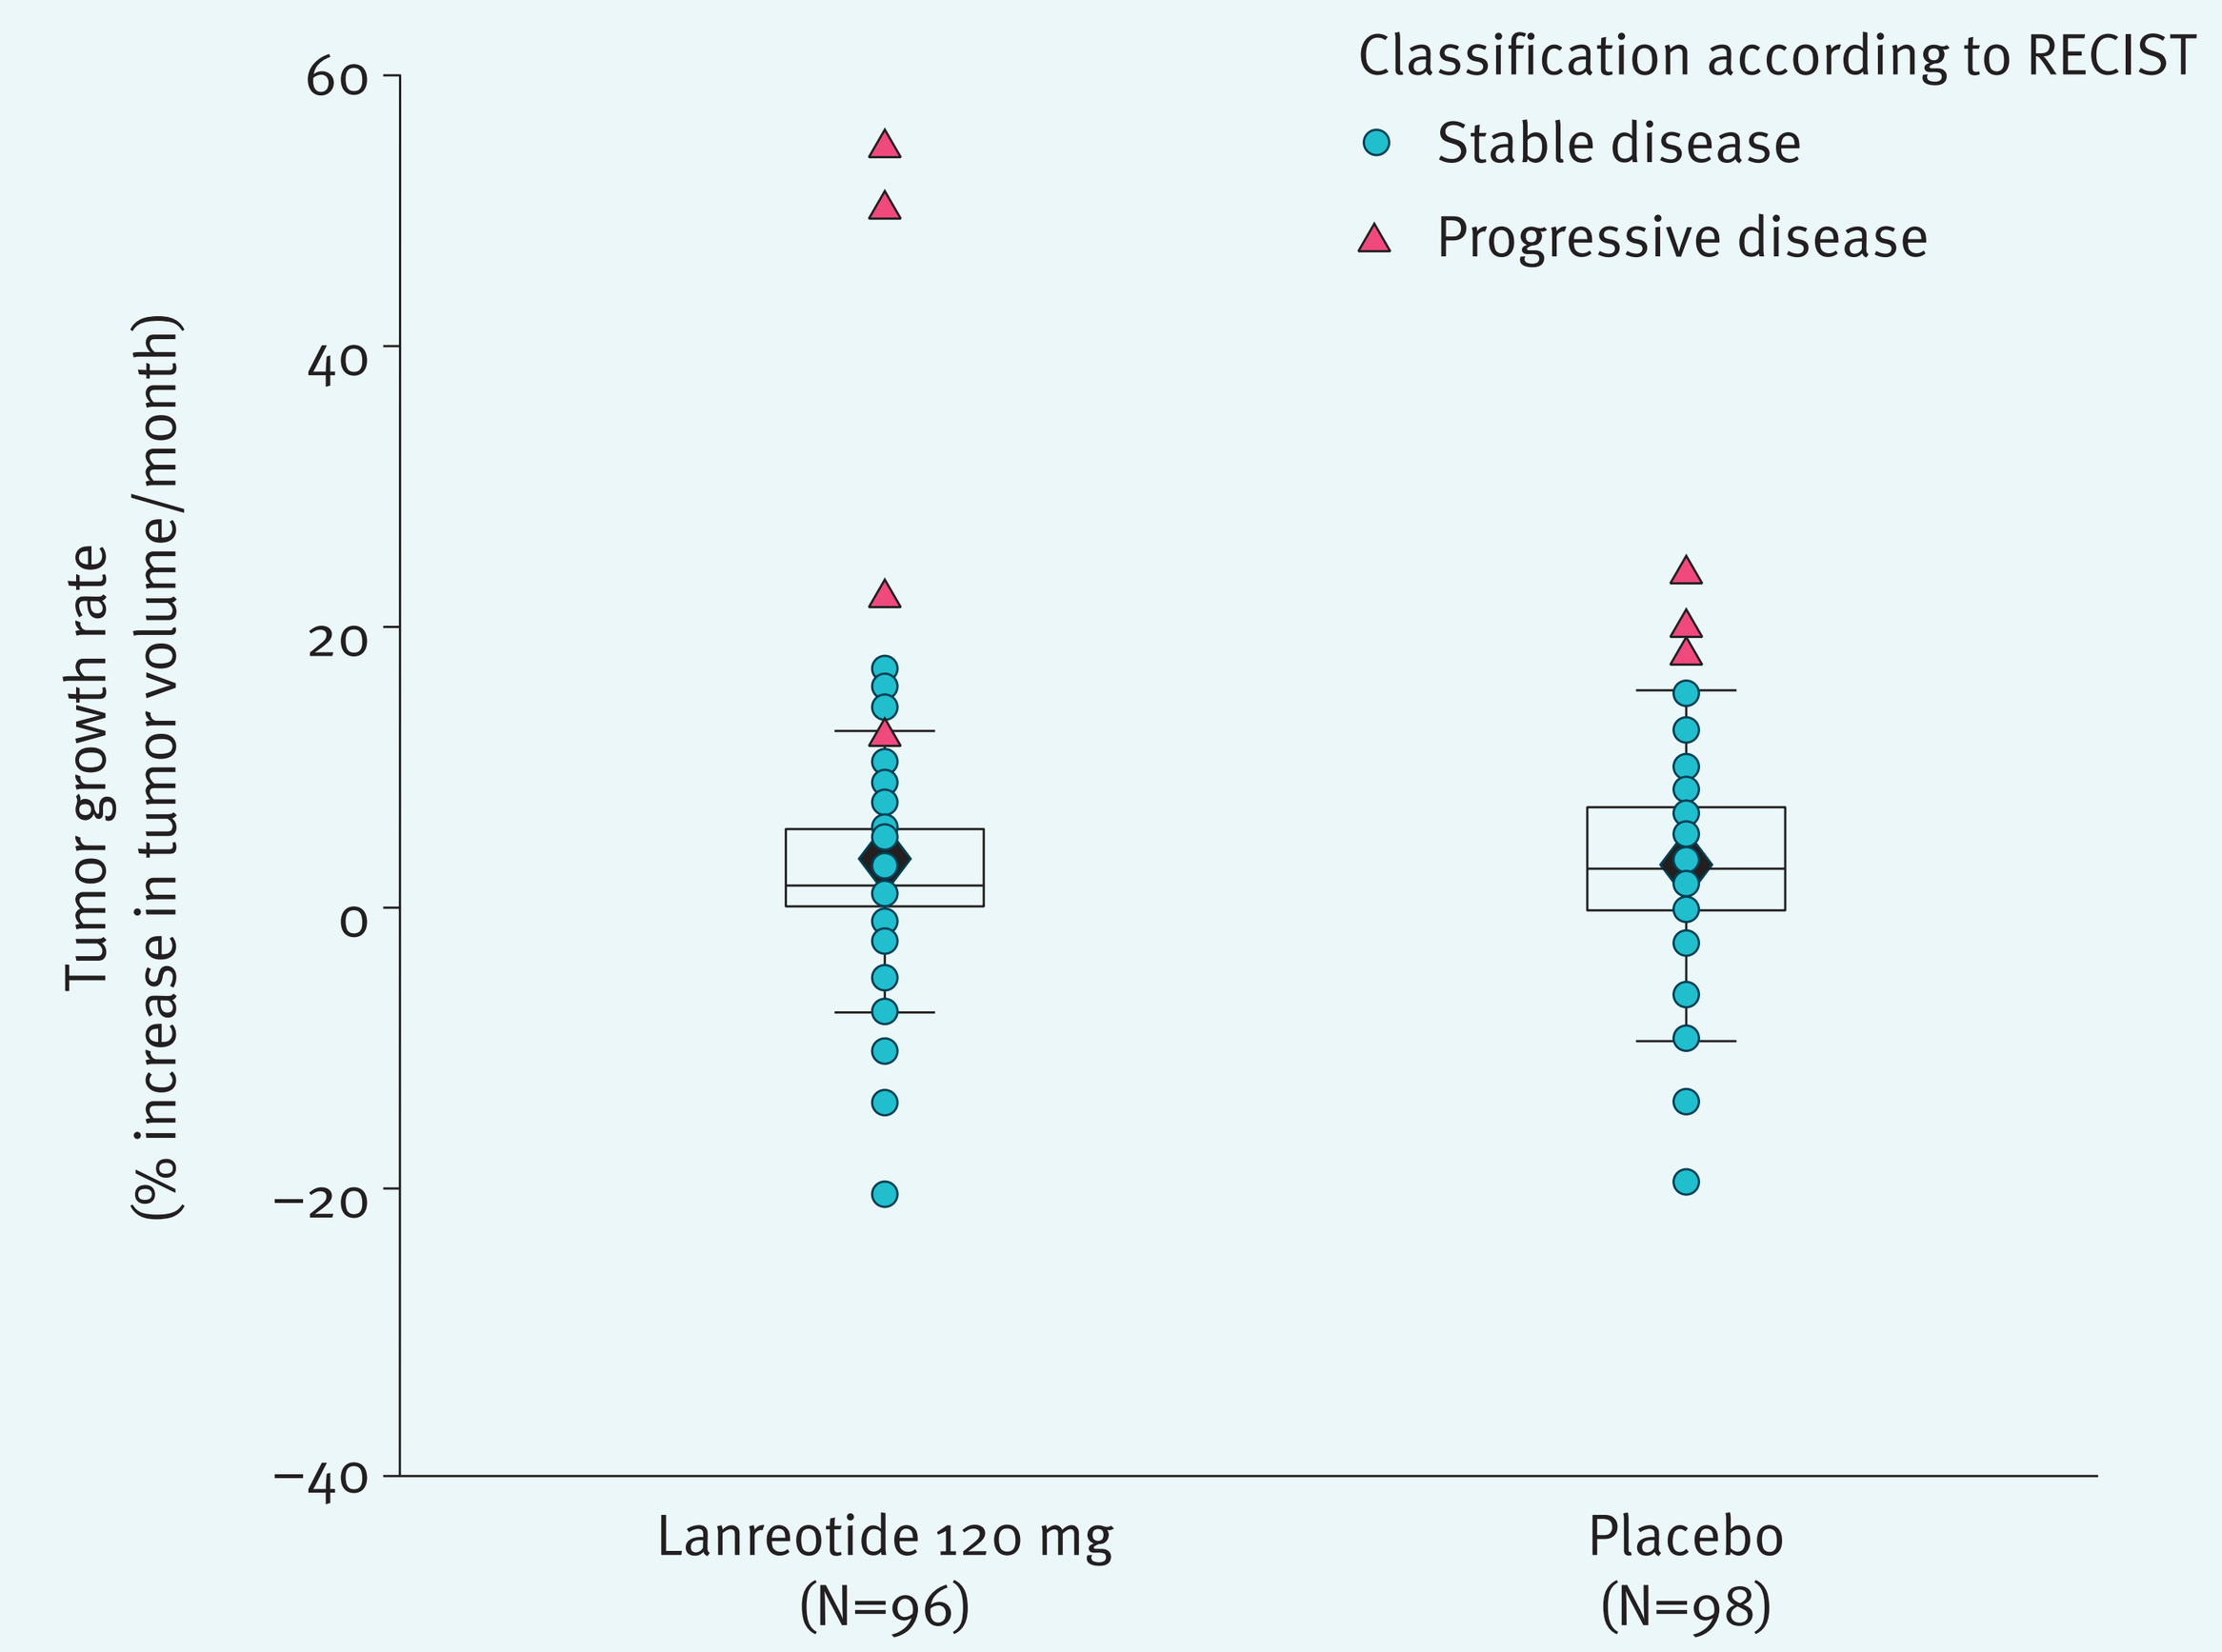
Classification according to RECIST
Stable disease
Progressive disease
Tumor growth rate
(% increase in tumor volume/month)
60
40
20
0
−20
−40
Lanreotide 120 mg
(N=96)
Placebo
(N=98)

Individual data points represent the TGR (% increase in tumor volume in 1 month) in individual patients (ITT population) during screening, according to their classification by RECIST v1.0. Boxes represent median and interquartile range (Q1–Q3), whiskers represent the minimum observation still within 1.5 IQR of lower quartile (Q1), and the maximum observation still within 1.5 IQR of the upper quartile (Q3). IQR; interquartile range; ITT, intention-to-treat; TGR, tumor growth rate.

- TGR as a prognostic factor (*post hoc* analyses; ITT population):
 - Receiver operating characteristic (ROC) analysis was conducted to determine the optimum pre-treatment TGR cut-off (%/month) for predicting the risk of progression.
 - Kaplan–Meier plots were used to describe PFS based on pre-treatment TGR.
 - Hazard ratios (HRs) were estimated from Cox regression models.
 - Additional prognostic factors for PFS were estimated from Cox regression models.

Figure 2. Estimated LS means TGRs calculated between consecutive visits.

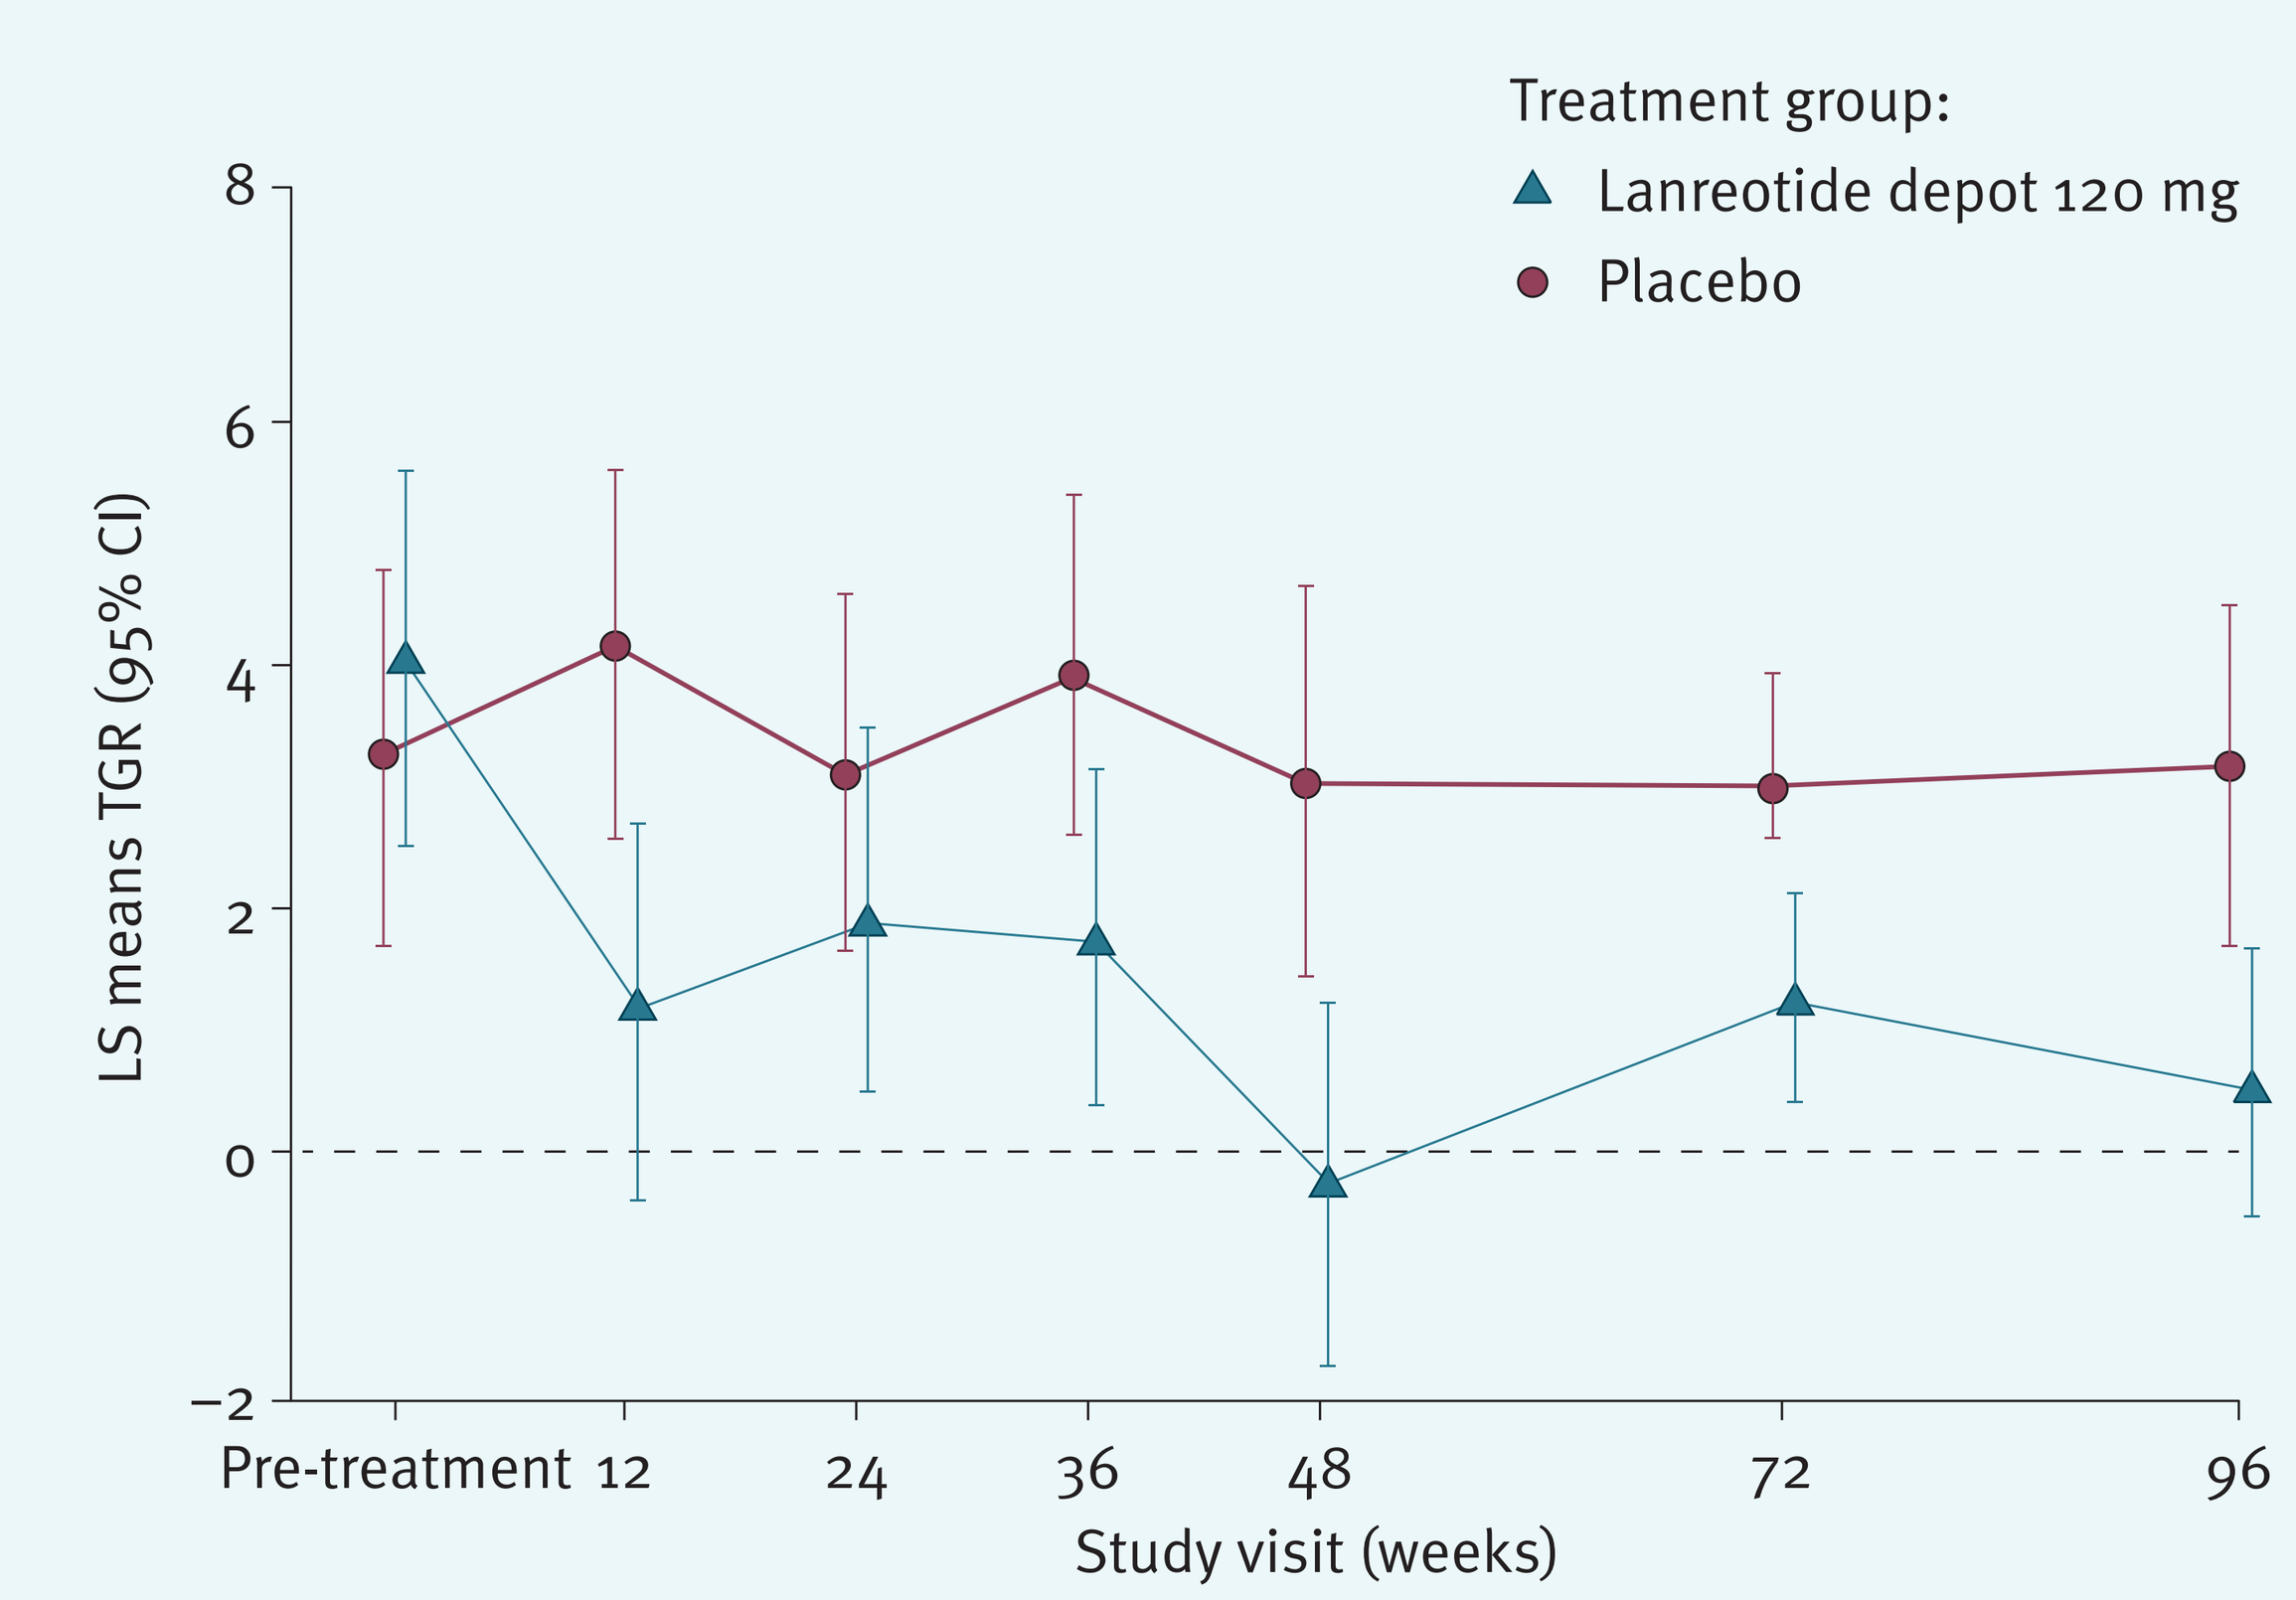
Treatment group:
Lanreotide depot 120 mg
Placebo
LS means TGR (95% CI)
8
6
4
2
0
−2
Pre-treatment
12
24
36
48
72
96
Study visit (weeks)

	Pre-treatment	12	24	36	48	72	96
N=	200	196	174	154	132	106	90
p-value =	0.46	0.008	0.28	0.03	0.003	0.009	0.007

*Pre-treatment TGR is calculated from the screening scan and baseline scan (week 1). CI, confidence interval; LS, least squares; TGR, tumor growth rate.

Figure 3. Kaplan–Meier plot of PFS in patient with TGRs (a) ≤4%/month and (b) >4%/month during the pre-treatment period.

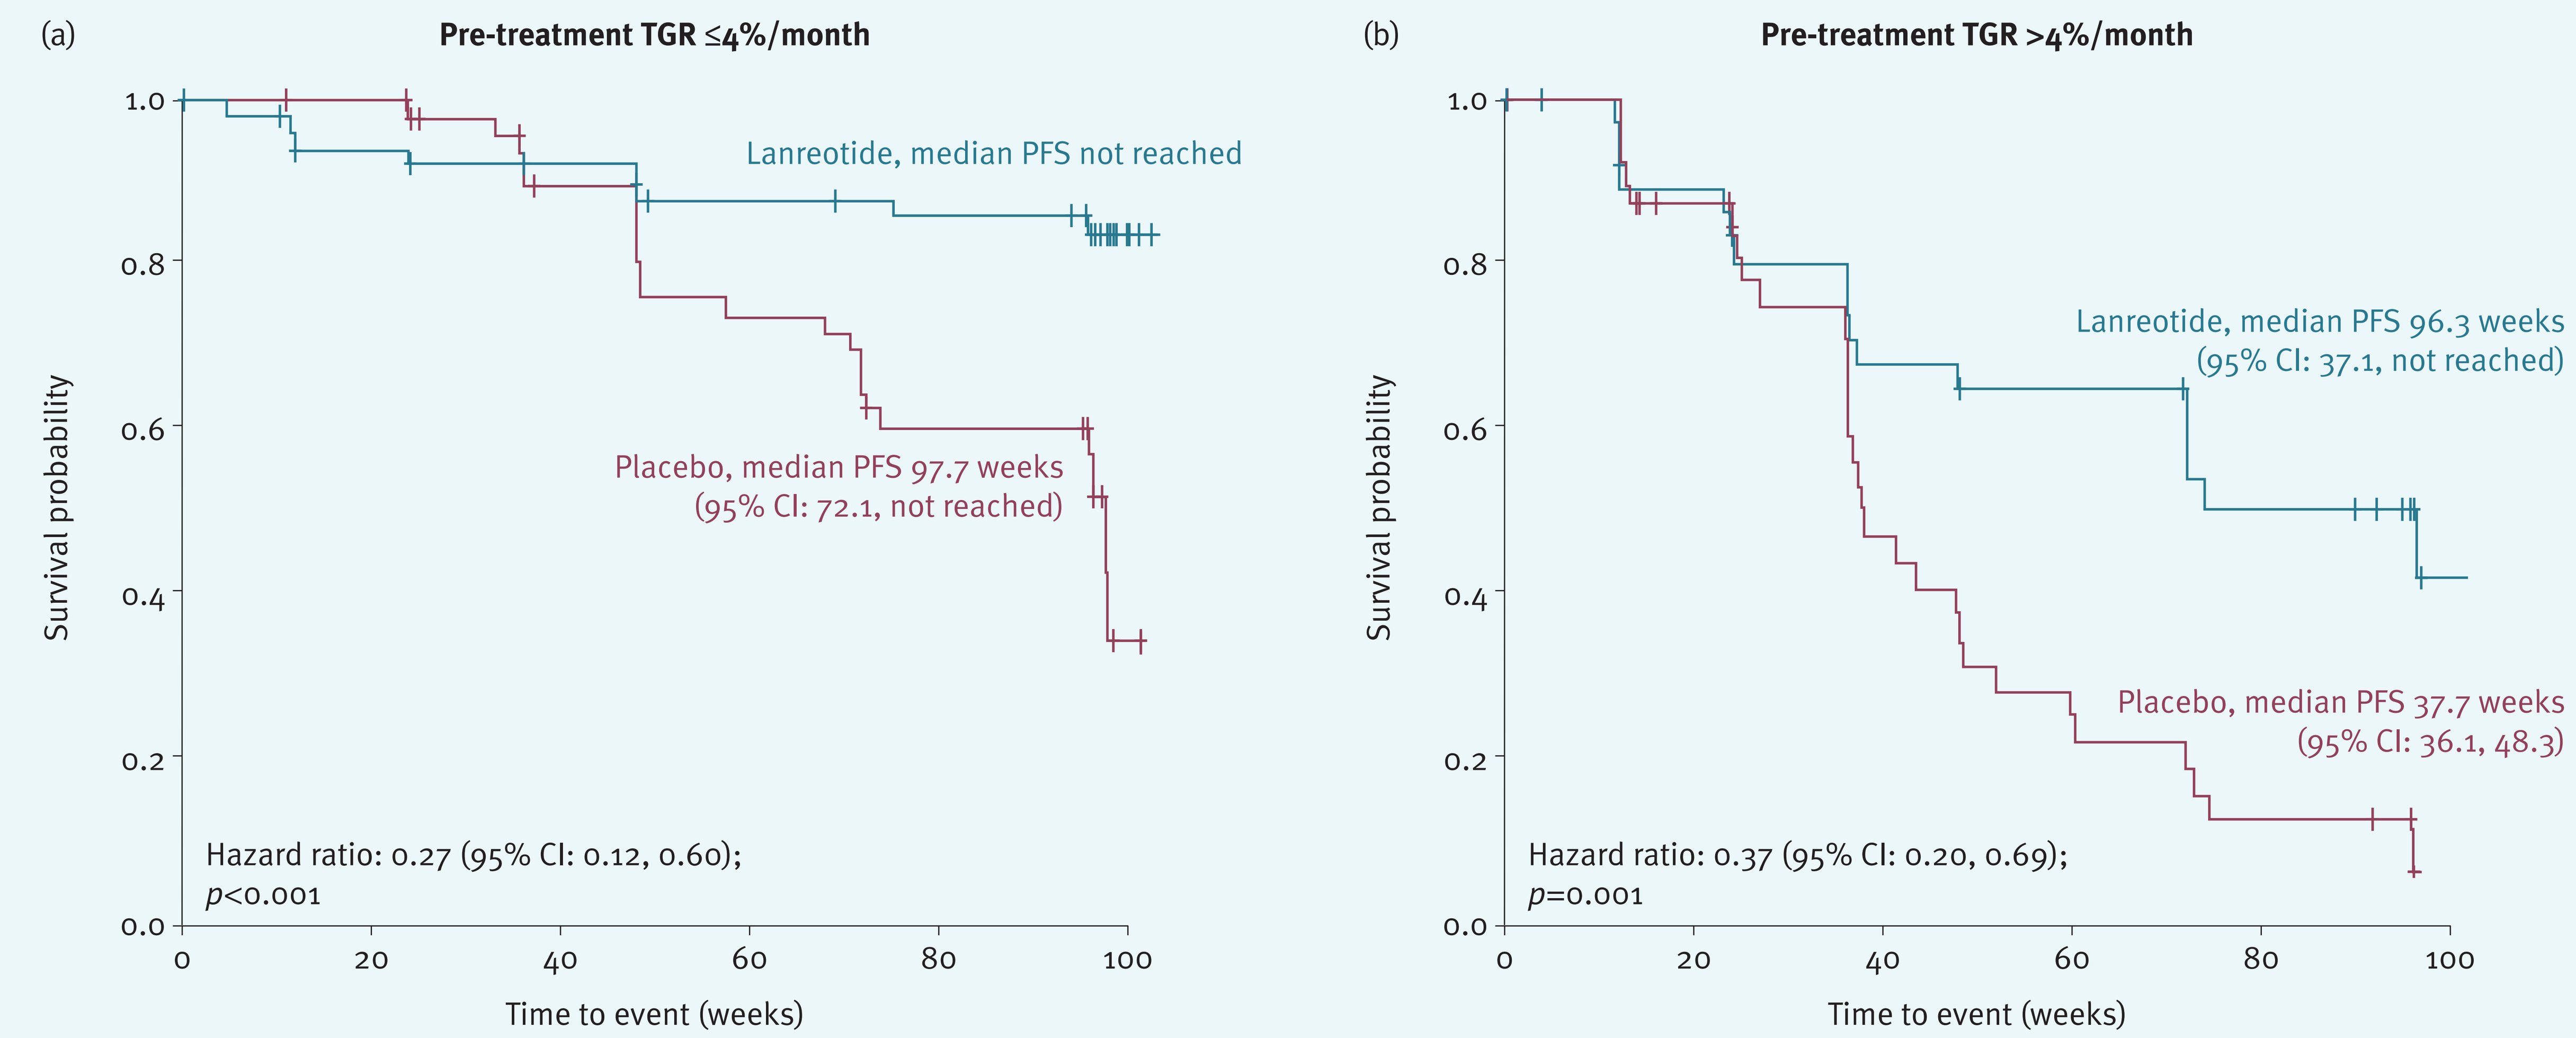
(a)
Pre-treatment TGR ≤4%/month
Lanreotide, median PFS not reached
Placebo, median PFS 97.7 weeks
(95% CI: 72.1, not reached)
Hazard ratio: 0.27 (95% CI: 0.12, 0.60);
p<0.001
Survival probability
Time to event (weeks)
(b)
Pre-treatment TGR >4%/month
Lanreotide, median PFS 96.3 weeks
(95% CI: 37.1, not reached)
Placebo, median PFS 37.7 weeks
(95% CI: 36.1, 48.3)
Hazard ratio: 0.37 (95% CI: 0.20, 0.69);
p=0.001
Survival probability
Time to event (weeks)

Analysis of PFS considers centrally assessed disease progressions (using RECIST v1.0 criteria) and any deaths reported during the study as events. Patients in progression due to non-target or new lesions were excluded. Pre-treatment TGR ≤4%, lanreotide n=56; placebo n=53; pre-treatment TGR >4%, lanreotide n=38; placebo n=40. CI, confidence interval; PFS, progression-free survival; TGR, tumor growth rate.

Results

Baseline characteristics

- A total of 204 patients were randomized (ITT population) to lanreotide depot (n=101) or placebo (n=103).
- Of these, 69% (141) had grade 1 tumors (Ki-67, <2%), 30% (61) had low grade 2 tumors (Ki-67, 3–10%); 33% (67) had hepatic tumor loads >25%; 45% (91) had primary tumors of the pancreas and 36% (73) of the midgut.
- A total of 200 evaluable patients with SLD measurements were included in these *post hoc* analyses, unless otherwise stated.

TGR to measure tumor progression

- Although 97% (193) of evaluable patients had stable disease (SD) before treatment according to RECIST v1.0, TGR analyses showed that a large proportion of tumors were growing (Figure 1).
- A reduction in TGR after only 12 weeks with lanreotide depot vs. placebo, resulted in a significant difference between treatment groups that was maintained throughout the treatment period (Figure 2).

TGR as a prognostic factor

- Of the pre-treatment TGR thresholds assessed by ROC analysis, a value of 4%/month was optimal for predicting the risk of progression independently of treatment group.
 - Pre-treatment TGR >4%/month had a 4.1-fold greater risk of progression than TGR ≤4%/month in the overall population (HR 4.1 [95% CI 2.5, 6.5]; *p*<0.001, n=187).
- Lanreotide depot was significantly more effective than placebo at reducing the risk of PD/death, by 73% and 63% in those with pre-treatment TGR ≤4% and >4%/month, respectively (Figure 3).
- Exploratory analysis of potential prognostic factors for PFS (HR [95% CI]) showed pre-treatment TGR >4%/month (3.26 [2.04–5.21]), prior therapy (2.33 [1.25–4.34]), hepatic tumor load >25%–≤50% (2.96 [1.44–6.04]) and >50% (4.23 [2.00–8.91]), and a pancreatic primary tumor (midgut vs. pancreatic primary tumor: 0.54 [0.32–0.90]; other/unknown vs. pancreatic primary tumor: 0.38 [0.17–0.84]) were independent prognostic factors for worse PFS.

Limitations

- This is a *post hoc* analysis, albeit from a large homogenous study population.
- Target lesions were assumed to be spherical, which tends to be the case for liver metastases.

Conclusions

- Compared with RECIST, TGRs provided more precise and rapid information on tumor kinetics before treatment, revealing the anti-tumor effects of lanreotide depot/autogel as early as 12 weeks.
- Pre-treatment TGR (irrespective of treatment) was a prognostic factor for later PFS outcomes. Lanreotide was more effective than placebo in delaying progression, irrespective of pre-treatment TGR.
- These findings suggest TGR has potential clinical utility as a novel outcome measure for tumor progression.

Acknowledgments

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield company, for medical writing support in preparing this poster. This study was sponsored by Ipsen.

References

1. Therasse P, *et al. J Natl Cancer Inst* 2000;92:205–16.
2. Eisenhauer EA, *et al. Eur J Cancer* 2009;45:228–47.
3. Caplin ME, *et al. N Engl J Med* 2014;371:224–33.

CLARINET Study Group

Austria M. Raderer; **Belgium** I. Borbath, D. Ysebaert; **Czech Republic** E. Sedláčková, P. Vítek; **Denmark** H. Grønbaek; **France** A. Adenis, L. Buscail, G. Cadiot, S. Dominguez, M. Ducreux, C. Lombard-Bohas, E. Mitry, P. Ruszniewski, J.F. Seitz; **Germany** N. Begum, I. Harsch, M. Pavel, C. Schöfl, M. Weber, B. Wiedenmann; **India** M. Mallath, P. Patil, K. Sambasivaiah, R. Saxena; **Italy** E. Bajetta, A. Buonadonna, R. Buzzoni, R. Cannizzaro, A. Colao, C. De Angelis, P. Tomassetti; **Poland** J. Ćwikła, B. Kos-Kudła; **Slovakia** T. Salek; **Spain** J. Capdevila, G. Soler, J.M. Tabernero; **Sweden** H. Ahlman, M. Kjellman; **UK** G. Aithal, A. Anthoney, M. Caplin, A. Grossman, J. Newell-Price, J. Ramage, N. Reed, A. Rees, W. Steward, L. Wall; **USA** M. Choti, A.T. Phan, E.M. Wolin.

Presented at the 2016 NANETS Annual Symposium, 30 September–1 October 2016, Jackson, WY, USA.

This study is sponsored by Ipsen