Abstract #144

NETTER-1 Phase III in Patients with Midgut Neuroendocrine Tumors Treated with ^{177}Lu-Dotatate: Efficacy, Safety, QoL Results and Subgroup Analysis

Jonathan Strosberg[1], Edward Wolin[2], Beth Chasen[3], Matthew Kulke[4], David Bushnell[5], Martyn Caplin[6], Richard P. Baum[7], Pamela Kunz[8], Timothy Hobday[9], Andrew Hendifar[10], Kjell Oberg[11], Maribel Lopera Sierra[12], Dik Kwekkeboom[13], Philippe Ruszniewski[14], and Eric Krenning[13] on behalf of the NETTER-1 study group

[1] Moffitt Cancer Center, Tampa, FL 33612, USA; [2] Markey Cancer Center, University of Kentucky, Lexington, KY 40536-0093, USA; [3] University of Texas MD Anderson Cancer Center, Houston, TX 77030, USA; [4] Dana-Farber Cancer Institute, Boston, MA 02215, USA; [5] University of Iowa, Iowa City, IA 52242, USA; [6] Royal Free Hospital, London, United Kingdom; [7] Zentralklinik, Bad Berka, Germany; [8] Stanford University Medical Center, Stanford, CA 94305, USA; [9] Mayo Clinic College of Medicine, Rochester, MN 55905, USA; [10] Cedars Sinai Medical Center, Los Angeles, CA 90048, USA; [11] University Hospital, Uppsala University, Uppsala, Sweden; [12] Advanced Accelerator Applications, New York, NY 10118, USA; [13] Erasmus Medical Center, Rotterdam, The Netherlands; [14] Hopital Beaujon, Clichy, France.

Introduction

- There are limited therapeutic options for patients with advanced midgut neuroendocrine tumors (20-45% of NETs) progressing on first-line somatostatin analogue therapy.
- Thousands of patients have been treated with ^{177}Lu-Dotatate, peptide receptors radionuclide therapy (PRRT), with promising results.
- PRRT experience: PFS/OS including ^{177}Lu-Dotatate Phase I-II (Rotterdam)

PFS, progression free survival; OS, overall survival

Center (reference)	Ligand	n	PFS (months)	OS (months)
Multicenter (1)	[^{90}Y-DOTA^0,Tyr^3]octreotide	58	29	37
Multicenter (2)	[^{90}Y-DOTA^0,Tyr^3]octreotide	90	16	27
Copenhagen (3)	[^{90}Y-DOTA^0,Tyr^3]octreotide	53	29	-
Warsaw (4)	[^{90}Y-DOTA^0,Tyr^3]octreotide	58	17	22
Rotterdam (5)	[^{177}Lu-DOTA^0,Tyr^3]octreotate	310	33	46

van der Zwan et al Eur J Endocrinol 2015 Jan; 172(1):R1-8

NETTER -1 Study Objectives and Design

Aim	Evaluate the efficacy and safety of ^{177}Lu-Dotatate + SSAs (symptoms control) compared to Octreotide LAR 60 mg (off-label use)[1] in patients with inoperable, somatostatin receptors positive, midgut NET, progressive under Octreotide LAR 30 mg (label use)
Design	International, multicenter, randomized, comparator-controlled, parallel-group

Treatment and Assessments
Progression free survival (RECIST criteria) every 12 weeks

Dose 1 Dose 2 Dose 3 Dose 4

Baseline and Randomization
- n = 115: 4 administrations of 7.4 GBq of ^{177}Lu-Dotatate every 8 weeks + SSAs (symptoms control)
- n = 115: Octreotide LAR (high dose - 60 mg every 4 weeks[1])

5 years follow up

1. FDA and EMA recommendation

Study Endpoints

Primary objective

Compare Progression Free Survival (PFS) after treatment with ^{177}Lu-Dotatate plus 30 mg octreotide LAR (symptoms control) vs treatment with high dose (60 mg) octreotide LAR

Secondary objectives

- Compare the Objective Response Rate between study arms
- Compare the Overall Survival between study arms
- Compare the Time to Progression between study arms
- Evaluate the safety and tolerability of ^{177}Lu-Dotatate
- Evaluate the health related quality of life (QoL) as measured by the EORTC QLQ-G.I.NET21 questionnaire

Main Inclusion Criteria

- Patients ≥18 years of age
- Metastatic or locally advanced, inoperable, histologically proven, well-differentiated midgut NET
- Ki67 index ≤ 20% (Grade 1-2)
- Progressive disease (RECIST Criteria 1.1 centrally confirmed) on uninterrupted fixed dose of Octreotide LAR (20-30 mg every 3-4 weeks)
- Somatostatin receptors positive disease
- Karnofsky Performance Score ≥ 60
- Including functioning and non-functioning

Population Characteristics at Enrolment (ITT population, N=229)

	^{177}Lu-Dotatate (n=116)	Octreotide LAR 60 mg (n=113)
Gender, n (%)		
Male	53 (46%)	60 (53%)
Female	63 (54%)	53 (47%)
Age (years), mean (SD)	63 (±9)	64 (±10)
BMI (Kg/sqm), mean (SD)	25 (±5)	26 (±7)
Primary tumor site, n (%)		
Jejunum	6 (5%)	9 (8%)
Ileum	86 (74%)	82 (73%)
Appendix	1 (1%)	2 (2%)
Right colon	3 (3%)	1 (1%)
Other*	20 (17%)	19 (17%)
Site of metastasis, n (%)		
Liver	97 (84%)	94 (83%)
Lymph nodes	77 (66%)	65 (58%)
Bones	13 (11%)	12 (11%)
Lungs	11 (10%)	5 (4%)
Other	40 (35%)	37 (33%)
Ki67, n (%)		
G1/G2	76/40 (66/34%)	81/32 (72/28%)
SRS, Krenning scale, n (%)**		
Grade 2	13 (11%)	14 (12%)
Grade 3	34 (29%)	32 (28%)
Grade 4	69 (60%)	67 (59%)
Chromogranin A (μg/L), mean (SD)	649 (420)	670 (422)
5-HIAA (mg/24h), mean (SD)***	100 (183)	77 (83)

(*) mainly peritoneal and mesenteric
(**) highest grade
(***) only available in 98 patients

Progression-Free Survival

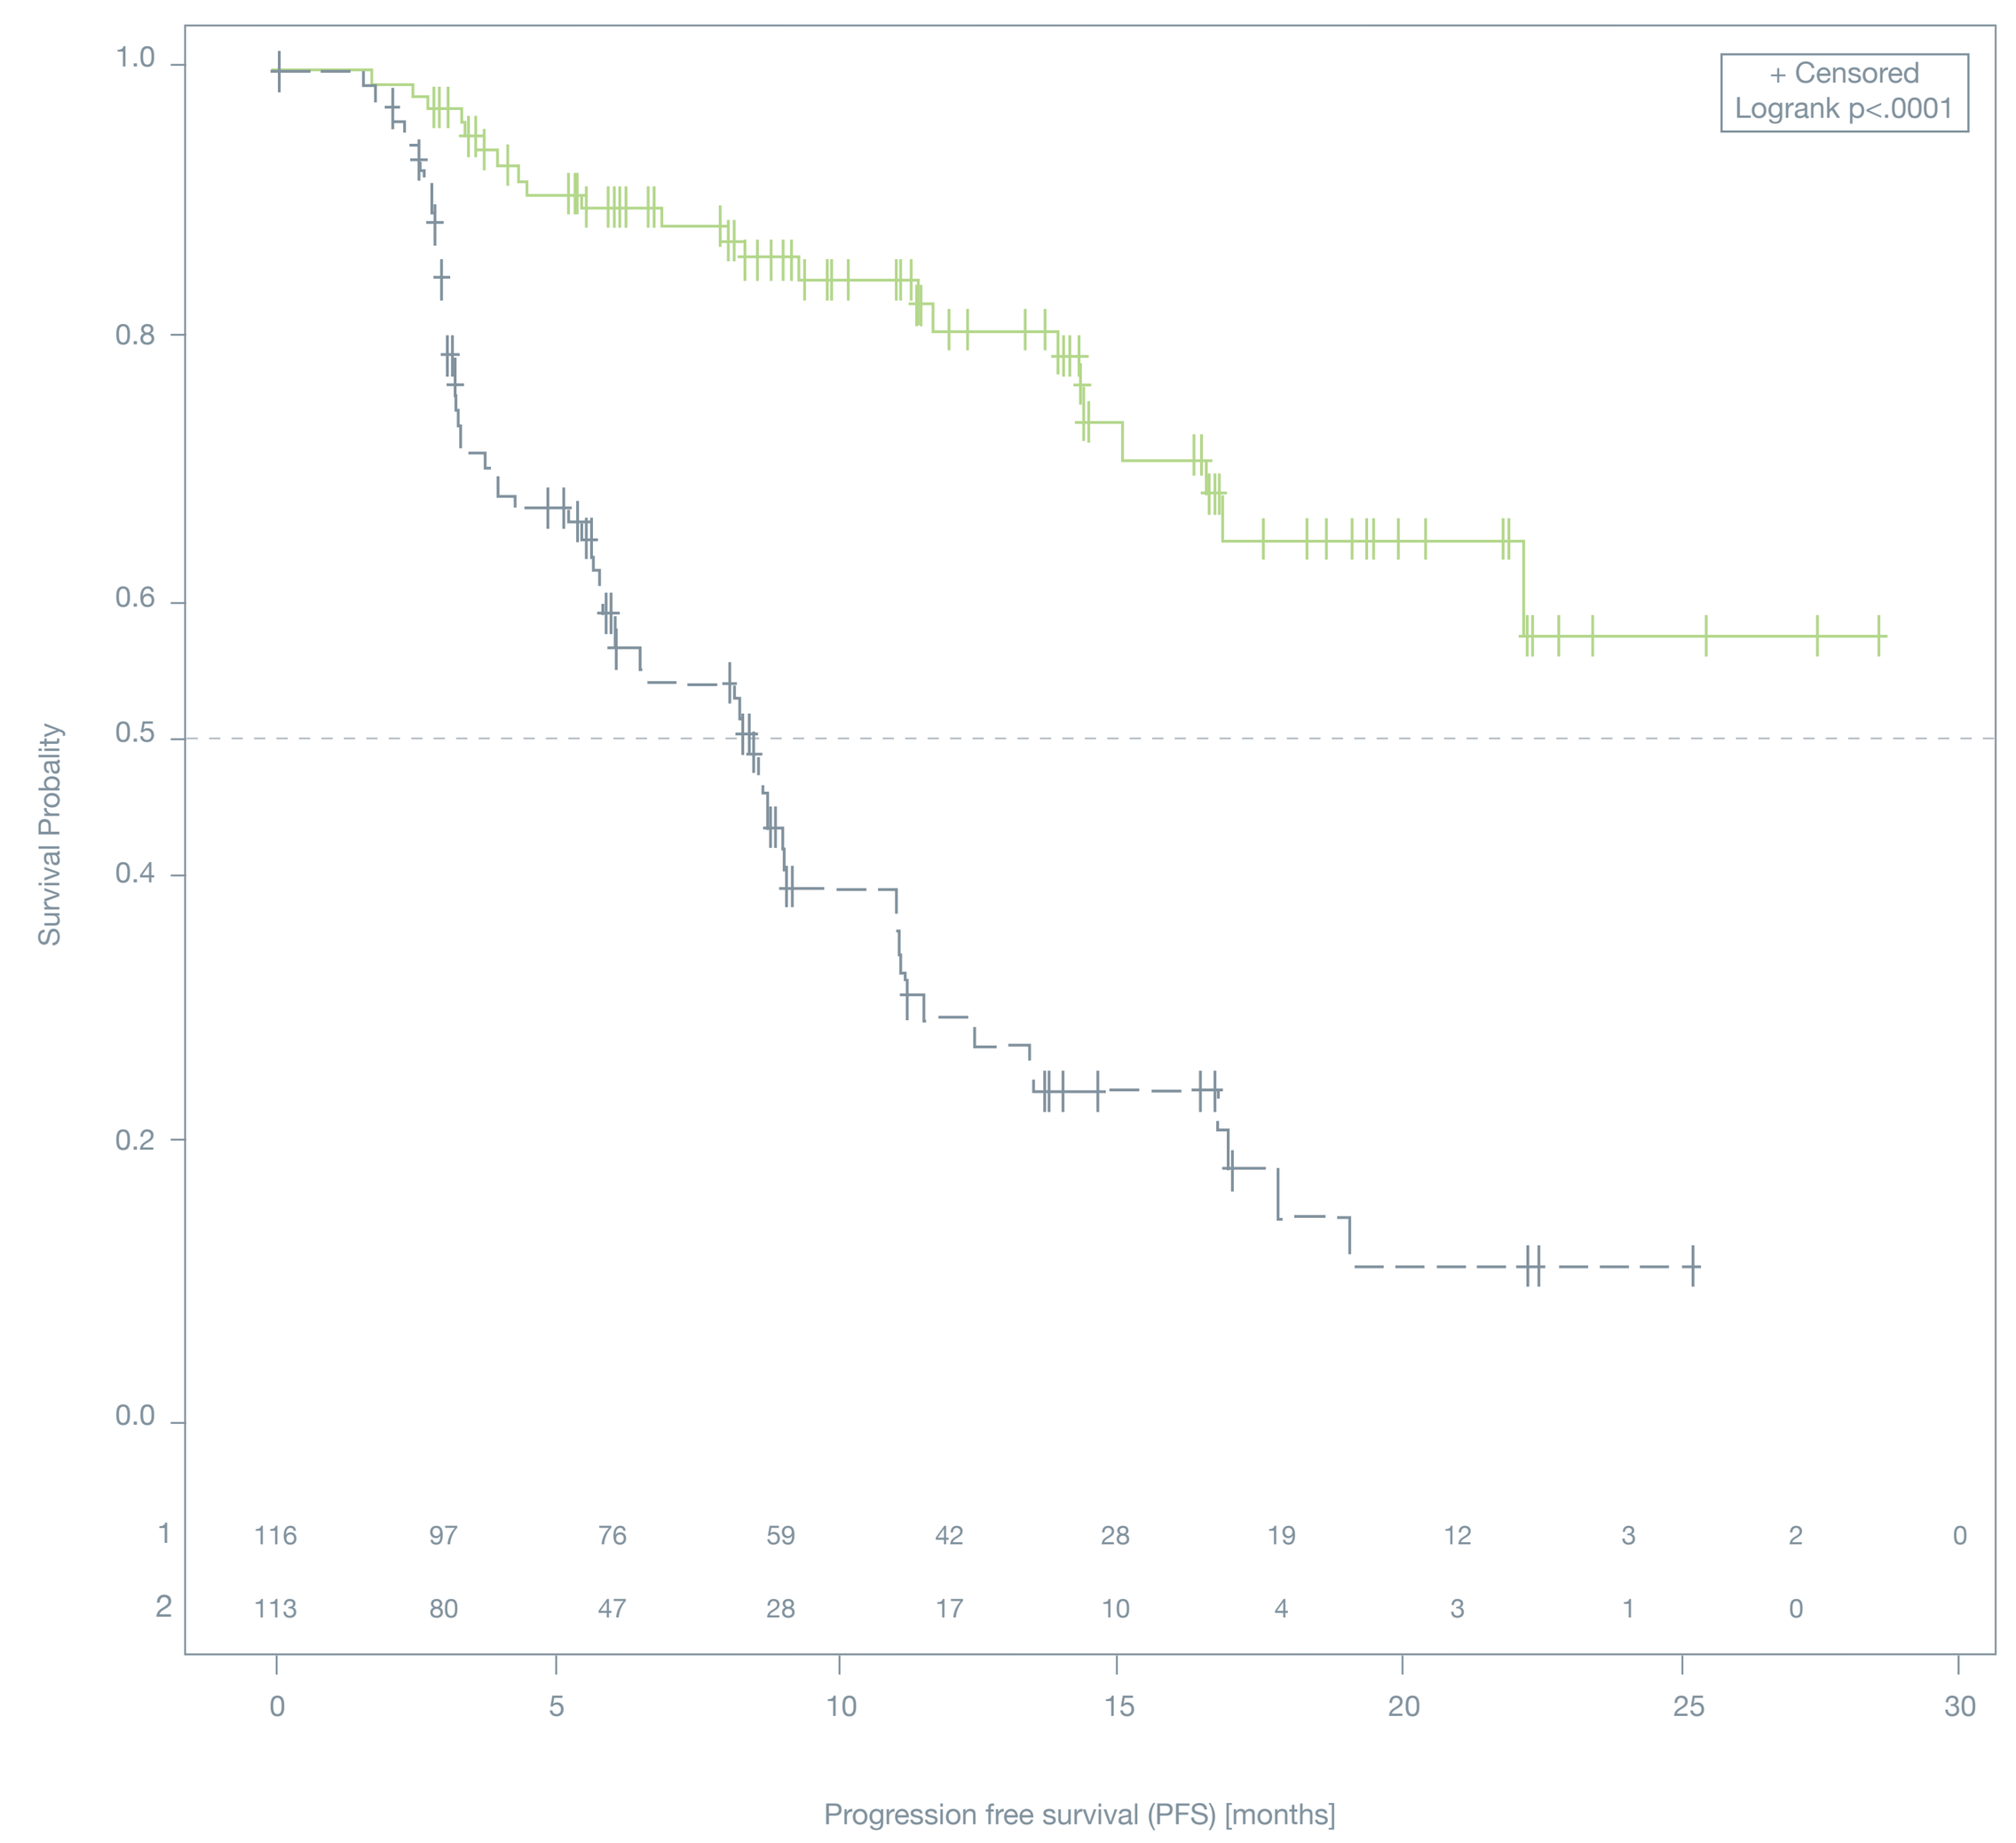
+ Censored
Logrank p<.0001
1.0
0.8
0.6
0.5
0.4
0.2
0.0
Survival Probality
1 116 97 76 59 42 28 19 12 3 2 0
2 113 80 47 28 17 10 4 3 1 0
0 5 10 15 20 25 30
Progression free survival (PFS) [months]

Hazard ratio: **0.21**
[0.13 – 0.33]
p < 0.0001

79% reduction in the risk of disease progression/death

Estimated Median PFS in the ^{177}Lu-Dotatate arm
≈ 40 months

N = 229 (ITT)
Number of events: 91

- ^{177}Lu-Dotatate: 23
 Median PFS: Not reached
- Octreotide LAR 60 mg: 68
 Median PFS: 8.4 months

Treatment
1: ^{177}Lu-DOTA^0-Tyr^3-Octreotate
2: Octreotide LAR 60 mg

All progressions were centrally confirmed and independently reviewed for eligibility (SAP)

- The median PFS was not reached for ^{177}Lu-Dotatate and was 8.4 months with 60 mg Octreotide LAR [95% CI: 5.8 – 9.1 months], p<0.0001, with a hazard ratio of 0.21 [95% CI: 0.13-0.33]. This means a 79% reduction in the risk of disease progression/death could be obtained by treatment with ^{177}Lu-Dotatate.
- Within the currently evaluable patient dataset for tumor responses (n=201), the number of CR + PR was 18 (ORR=18%) in the ^{177}Lu-Dotatate group and 3 (3%) in the Octreotide LAR 60 mg group (p=0.0008).

Forest plot of Hazard ratios by baseline parameter

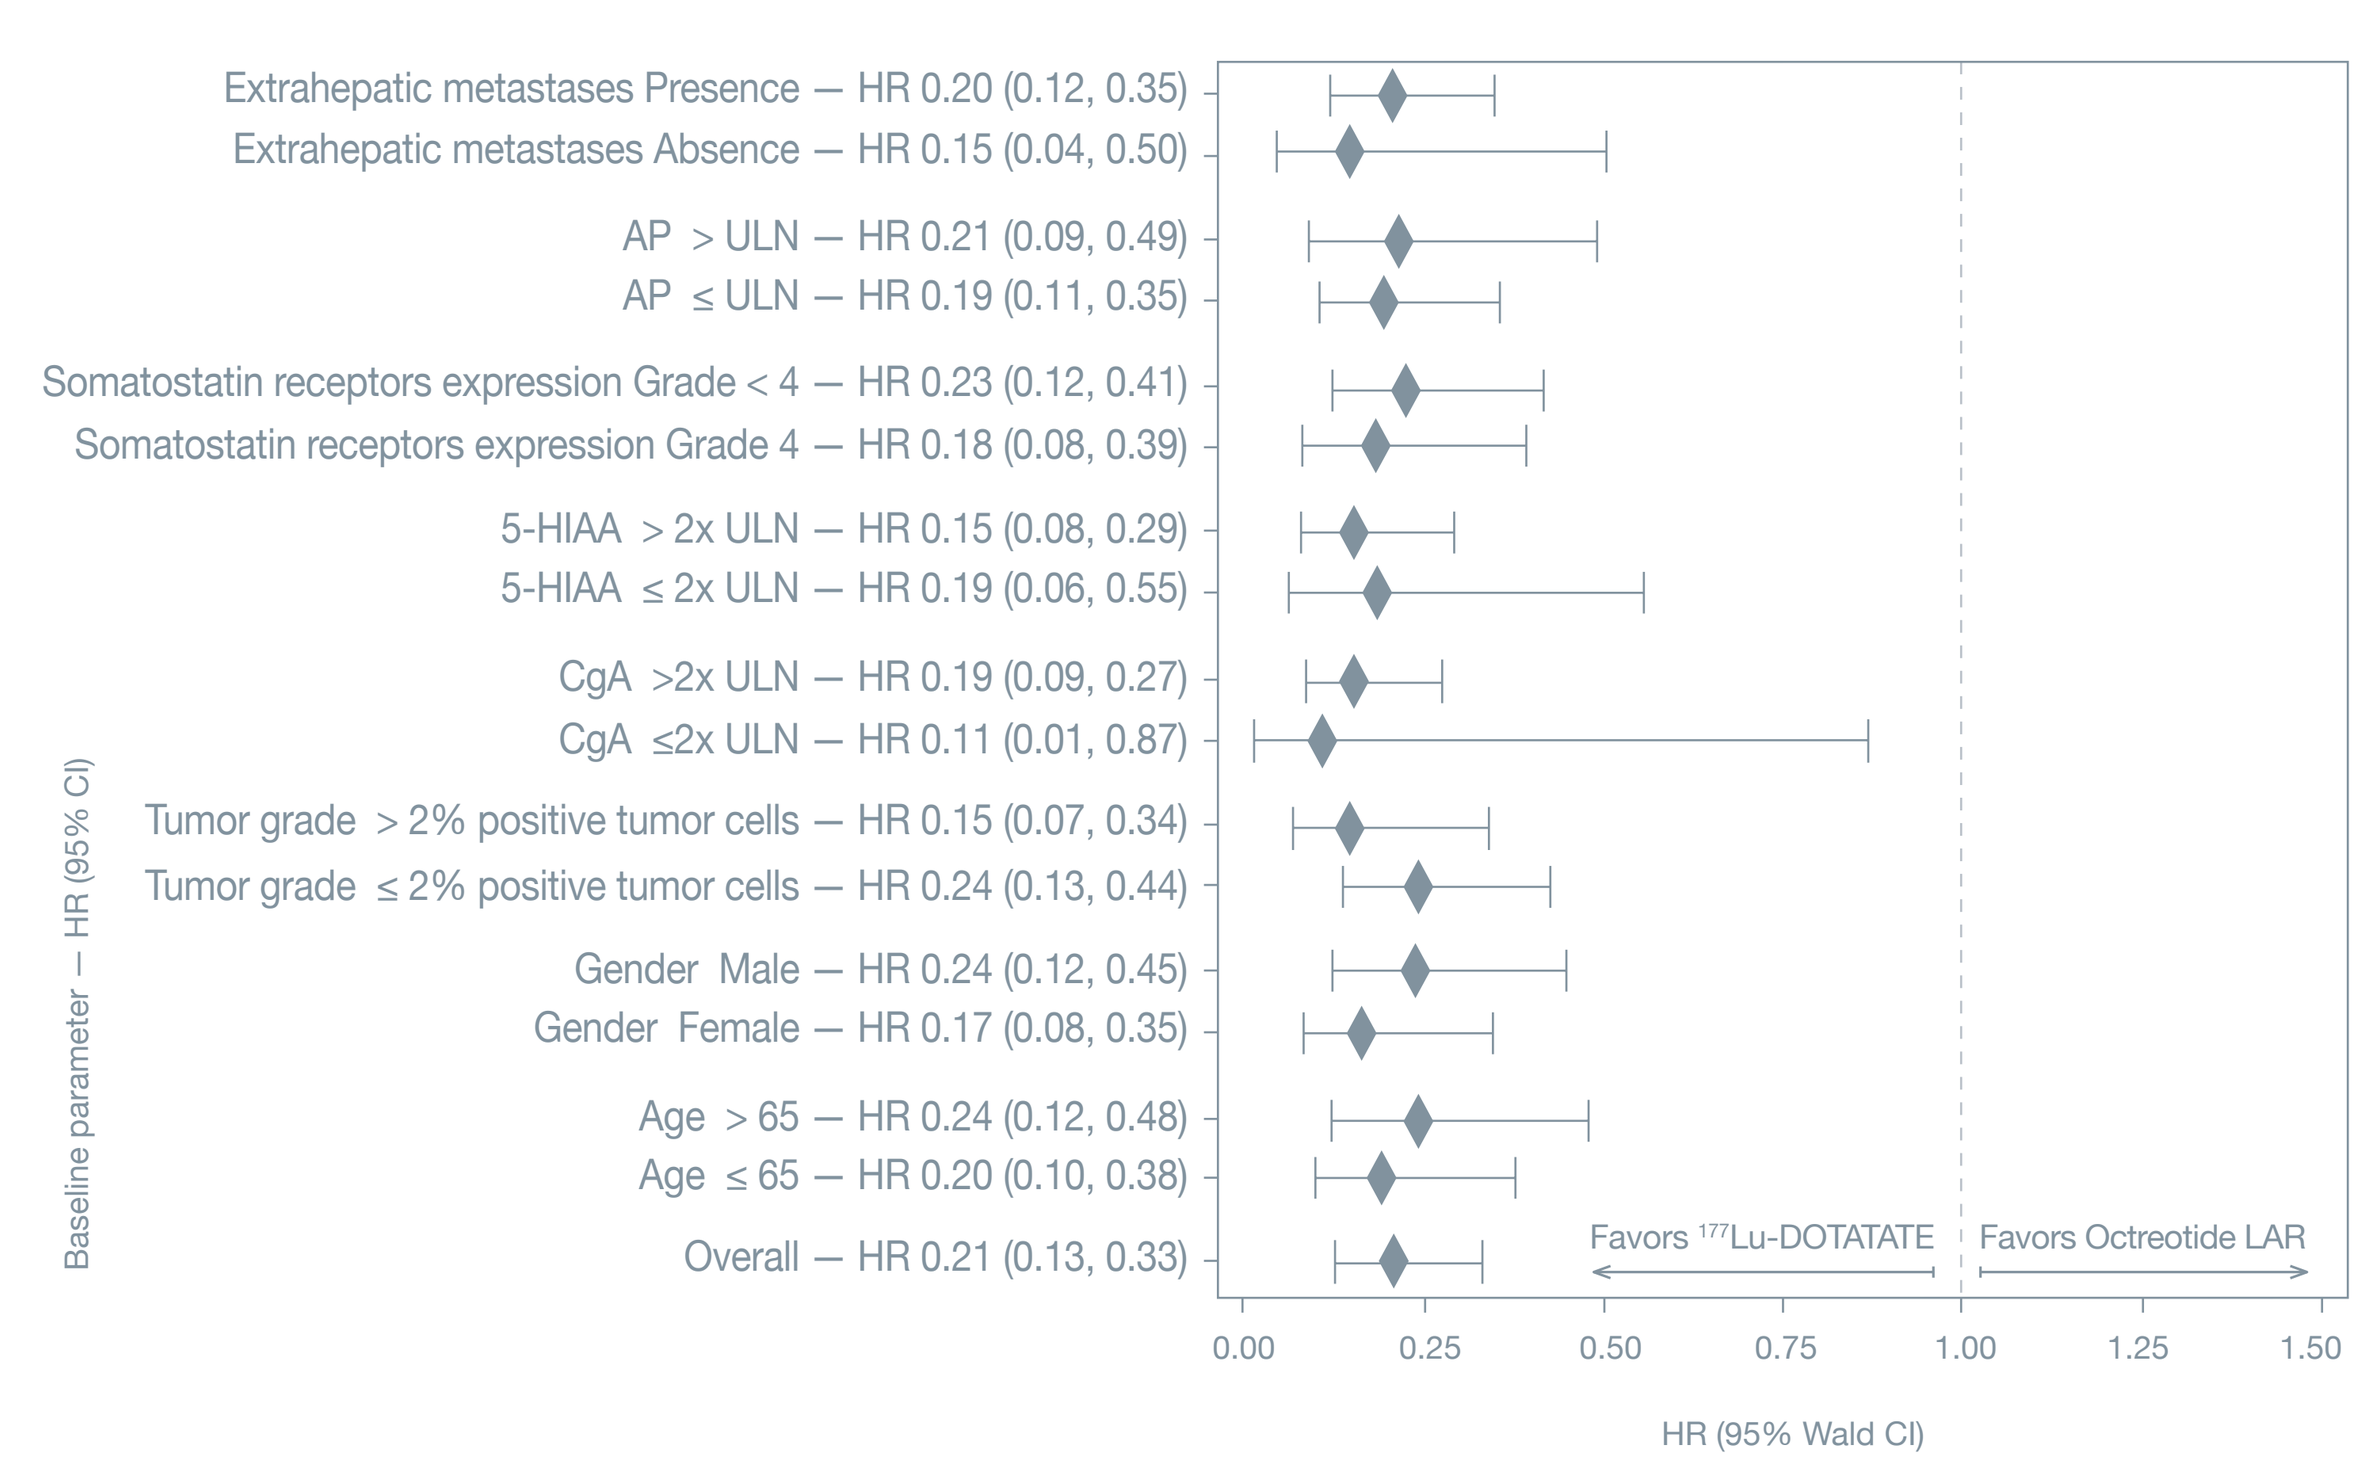
Extrahepatic metastases Presence — HR 0.20 (0.12, 0.35)
Extrahepatic metastases Absence — HR 0.15 (0.04, 0.50)
AP > ULN — HR 0.21 (0.09, 0.49)
AP ≤ ULN — HR 0.19 (0.11, 0.35)
Somatostatin receptors expression Grade < 4 — HR 0.23 (0.12, 0.41)
Somatostatin receptors expression Grade 4 — HR 0.18 (0.08, 0.39)
5-HIAA > 2x ULN — HR 0.15 (0.08, 0.29)
5-HIAA ≤ 2x ULN — HR 0.19 (0.06, 0.55)
CgA >2x ULN — HR 0.19 (0.09, 0.27)
CgA ≤2x ULN — HR 0.11 (0.01, 0.87)
Tumor grade > 2% positive tumor cells — HR 0.15 (0.07, 0.34)
Tumor grade ≤ 2% positive tumor cells — HR 0.24 (0.13, 0.44)
Gender Male — HR 0.24 (0.12, 0.45)
Gender Female — HR 0.17 (0.08, 0.35)
Age > 65 — HR 0.24 (0.12, 0.48)
Age ≤ 65 — HR 0.20 (0.10, 0.38)
Overall — HR 0.21 (0.13, 0.33)
Baseline parameter — HR (95% CI)
Favors 177Lu-DOTATATE
Favors Octreotide LAR
0.00 0.25 0.50 0.75 1.00 1.25 1.50
HR (95% Wald CI)

Note: Somatostatin receptors expression (SSR) is according to Octreoscan tumor uptake score
Note: 5-HIAA and AP assessment are taken at eligibility of baseline visit whichever is the first valid measurement

Objective Responses - currently evaluable patients

	^{177}Lu-Dotatate (n=101)*	Octreotide LAR 60 mg (n=100)*
Complete Response (n)	1	0
Partial Response (n)	17	3
Objective Response Rate (*)	18%	3%
Confidence Interval (95%)	10%-25%	0% - 6%
Statistical Significance	p = 0.0008	

(*) Exclude patients with no post-baseline scan or central response available

Overall Survival (interim analysis)

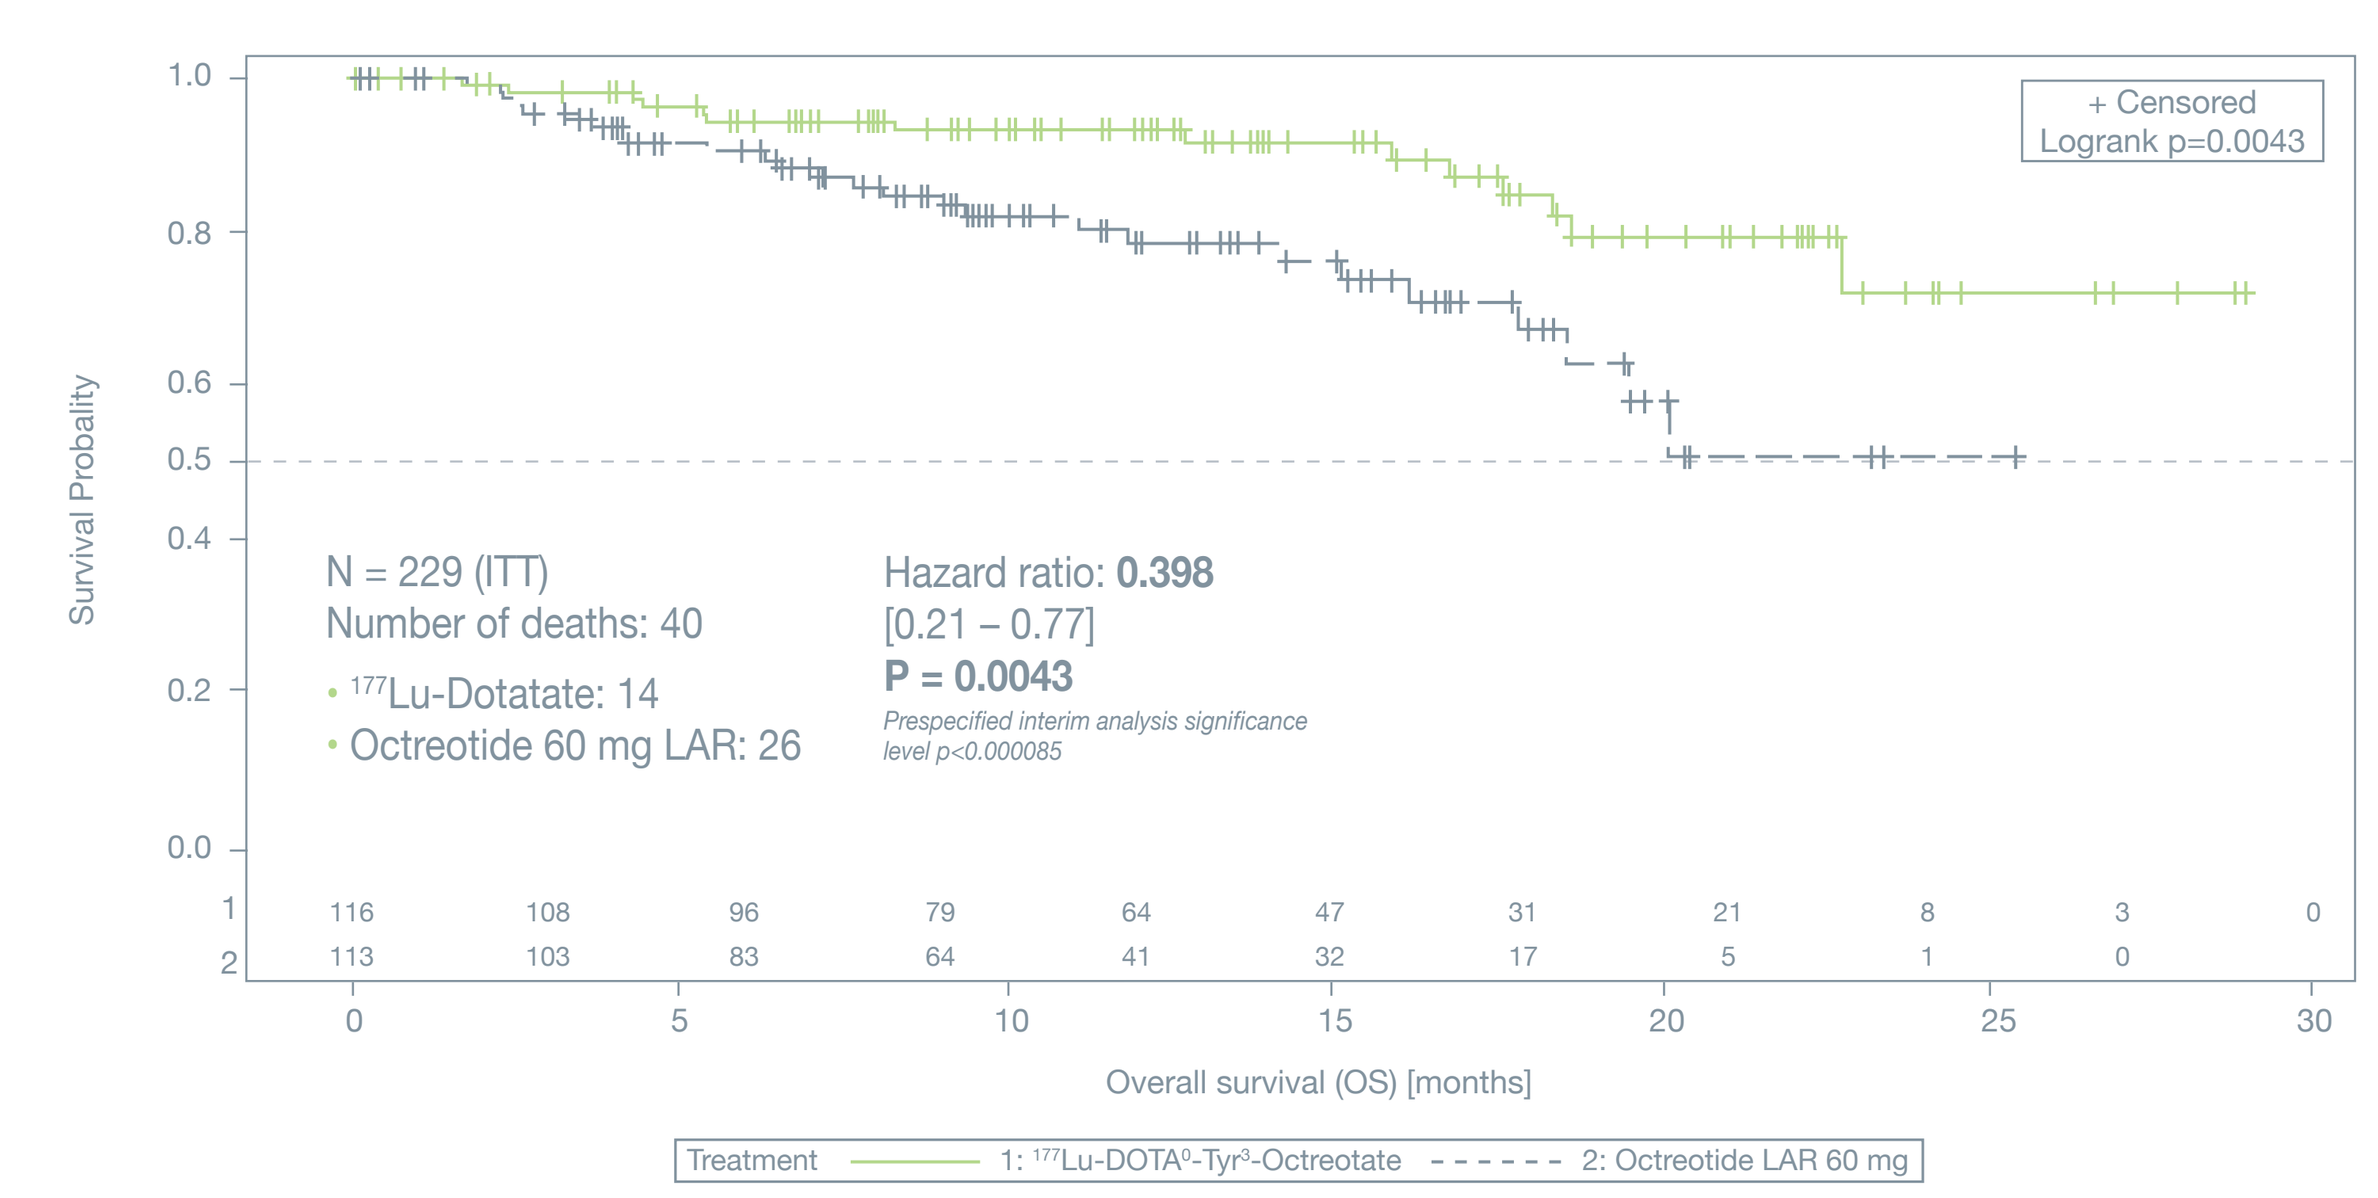
+ Censored
Logrank p=0.0043
1.0
0.8
0.6
0.5
0.4
0.2
0.0
Survival Probality
N = 229 (ITT)
Number of deaths: 40
177Lu-Dotatate: 14
Octreotide 60 mg LAR: 26
Hazard ratio: 0.398
[0.21 – 0.77]
P = 0.0043
Prespecified interim analysis significance level p<0.000085
1 116 108 96 79 64 47 31 21 8 3 0
2 113 103 83 64 41 32 17 5 1 0
0 5 10 15 20 25 30
Overall survival (OS) [months]
Treatment 1: 177Lu-DOTA0-Tyr3-Octreotate 2: Octreotide LAR 60 mg

^{177}Lu-Dotatate Exposure and DMT

Patients who completed trt phase, N=103*	Nb of Patients
Number of administrations, n (%)	
4	79 (77%)
3	6 (6%)
2	12 (12%)
1	5 (5%)
0	1 (1%)
All treated patients – N=111	**Nb of Patients**
Dose modifying toxicity, n (%)	
No DMT	105 (95%)
DMT	6 (5%)

(*) without patients still under treatment (n=8) or no treatment

Safety and Tolerability (Safety Set; n=221)

	^{177}Lu-Dotatate (n=111)	Octreotide LAR 60 mg (n=110)
Any adverse event	106 (96%)	95 (86%)
Related to treatment	95 (86%)	34 (31%)
Serious adverse events	29 (26%)	26 (24%)
Related to treatment	10 (9%)	1 (1%)
Withdrawals due to adverse events	7 (6%)	10 (9%)
Related to treatment	5 (5%)	0 (0%)

Adverse Events: all grades and grades 3-4

		^{177}Lu-Dotatate (n=111)		Octreotide LAR (n=110)	
		All grades	Grade 3 to 4	All grades	Grade 3 to 4
System Organ Class	Preferred Term	%	%	%	%
Gastrointestinal disorders	Nausea	59%	4%	12%	2%
	Vomiting	47%	7%	10%	0%
	Diarrhea	29%	3%	19%	2%
	Abdominal pain	26%	3%	26%	5%
	Abdominal distension	13%	0%	14%	0%
General disorders and administration site conditions	Fatigue / asthenia	40%	2%	25%	2%
	Edema peripheral	14%	0%	7%	0%
Blood and lymphatic system disorders	Thrombocytopenia	25%	2%	1%	0%
	Lymphopenia	18%	9%	2%	0%
	Anemia	14%	0%	5%	0%
	Leukopenia	10%	1%	1%	0%
	Neutropenia	5%	1%	1%	0%

Grade 3/4 Hepatotoxicity

	^{177}Lu-Dotatate (n=111)	Octreotide LAR (n=110)
AST increase	4%	0%
ALT increase	4%	0%
Bilirubin increase	2%	0%
GGT increase (at inclusion respectively 11% and 9%)	18%	13%

(CTCAE V4.0; Safety Set)

Creatinine Clearance
Renal function remains stable over the 2-year observation period

	^{177}Lu-Dotatate (n=111)	Octreotide LAR (n=110)
	Grade 3/4	Grade 3/4
Creatinine increased	0%	0%

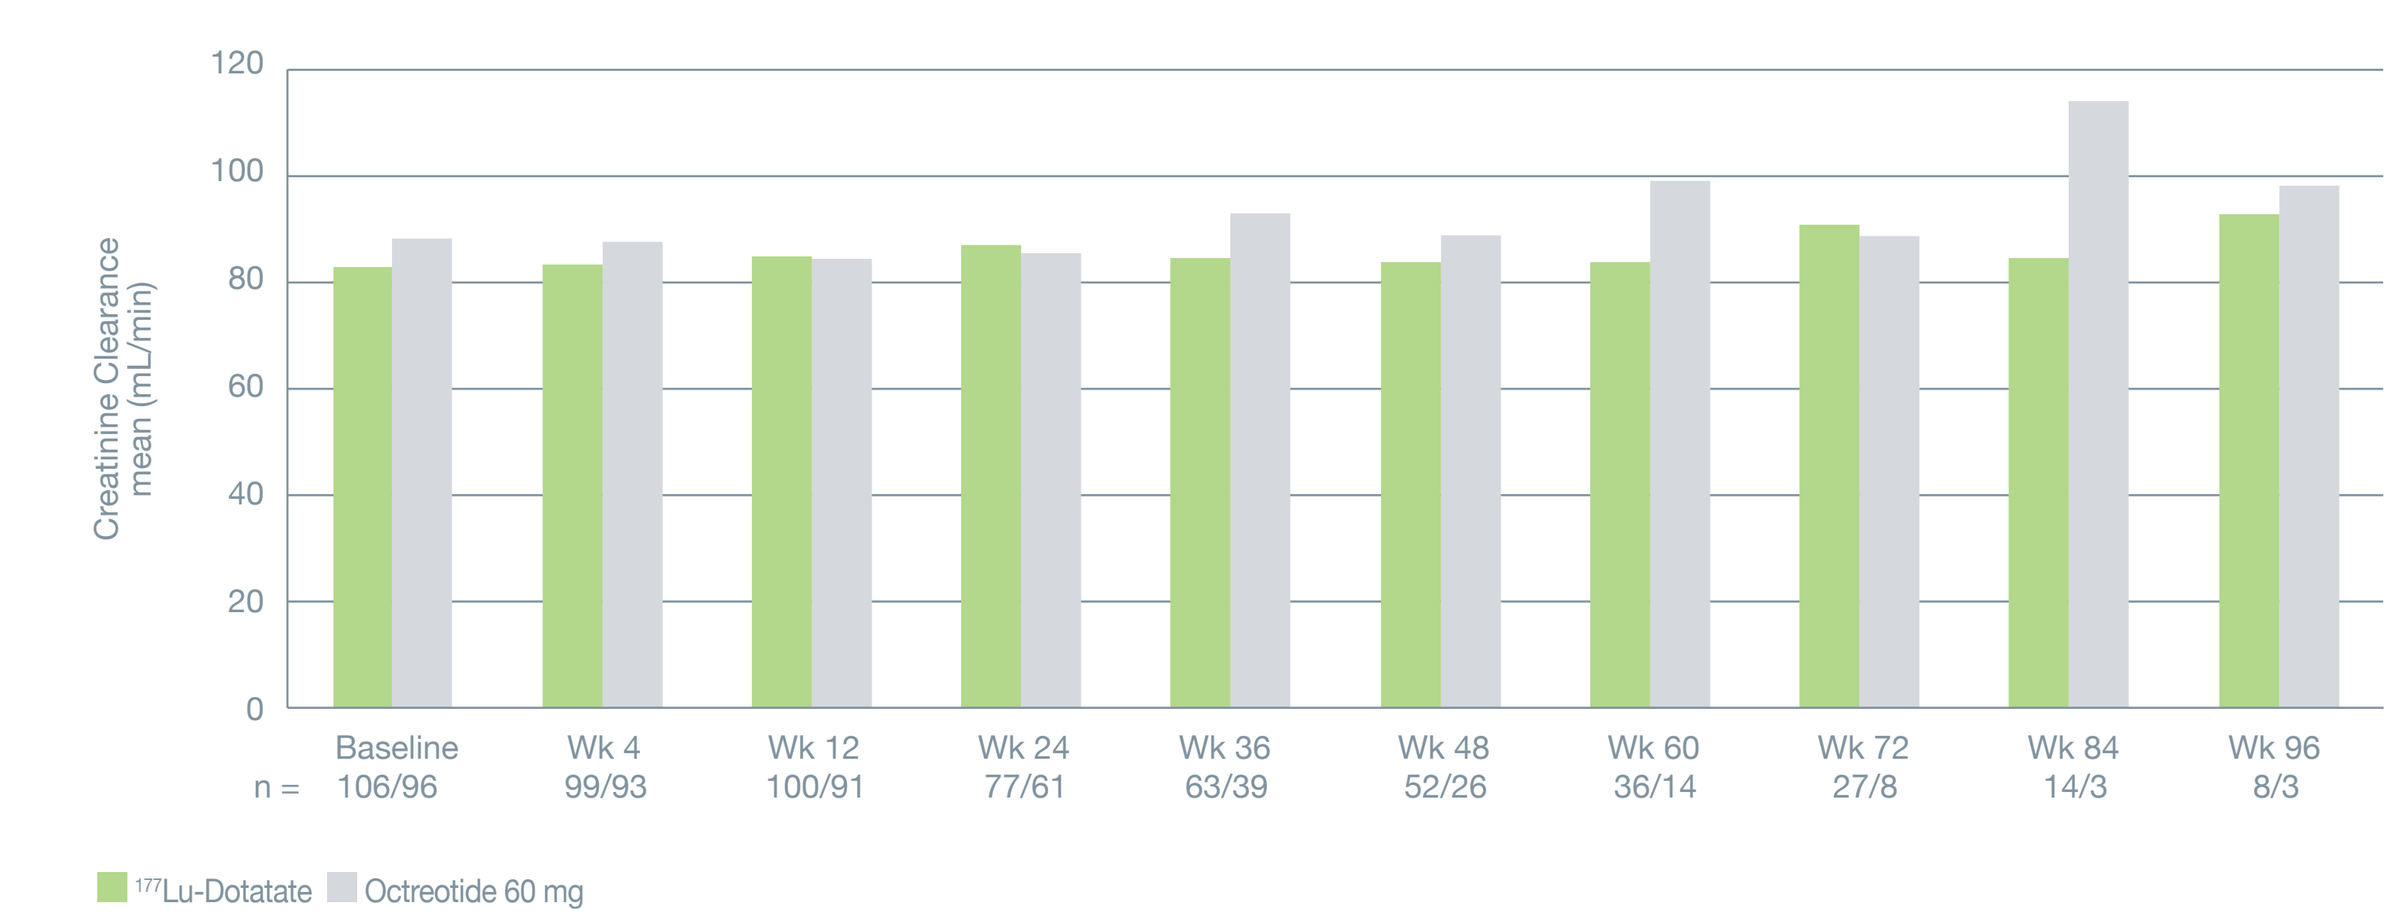
120
100
80
60
40
20
0
Creatinine Clearance mean (mL/min)
Baseline Wk 4 Wk 12 Wk 24 Wk 36 Wk 48 Wk 60 Wk 72 Wk 84 Wk 96
n = 106/96 99/93 100/91 77/61 63/39 52/26 36/14 27/8 14/3 8/3
177Lu-Dotatate
Octreotide 60 mg

Summary and Conclusions

- Final analysis : in this first prospective randomized study in patients with progressive metastatic midgut NETs, ^{177}Lu-Dotatate was superior to Octreotide 60 mg in terms of:
 - PFS (Not Reached vs 8.4 months, p<0.0001)
 - ORR (18% vs 3%, p=0.0008)
- Interim analysis suggests increased OS (14 vs 26 deaths), to be confirmed by final analysis
- ^{177}Lu-Dotatate demonstrates a favorable safety profile, with no clinically relevant findings especially regarding hematological, renal and hepatic parameters
- While few treatment options are available for patients progressing under SSAs, ^{177}Lu-Dotatate has a major therapeutic benefit for this patient population

Acknowledgements

We thank the patients and investigators participating in this trial.

NETTER-1 investigators:
Belgium E Van Cutsem France C Ansquer, E Baudin, F Courbon, F Giammarile, P Ruszniewski, D Taieb Germany R Baum, M Bitzer, S Ezziddin, M Pavel, K Scheidhauer, M Weber Italy E Brianzoni, G Delle Fave, CM Grana, G Mariani, E Seregni, S Severi Portugal I Azevedo Spain P Bello, E Grande, J Mora UK A Al-Nahhas, M Caplin, A Grossman, P Manoharan, N Reed, R Srirajaskanthan, S Vinjamuri USA Al B Benson, D Bushnell, R J Campeau, E Delpassand, A Hendifar, T Hobday, M H Kulke, E H Liu, D Metz, E Mittra, M Morse, M Schipper, J Strosberg, J Yao.

NETTER-1 DSMB: S Garbus, L Anthony, N Freemantle, K Öberg.

NANETs annual conference -September 30 - October 1, 2016 - Jackson, Wyoming

AAA-Lu45-092016