Pharmacokinetics, Pharmacodynamics, Safety, and Tolerability of DP1038, an Intranasal Formulation of Octreotide Acetate, in Healthy Volunteers

Matteo Levisetti[1], Olivier Laurent[1], Mark Daniels[1], Mark Milad[2], Michelle Mazzoni[1], Joel Martin[1]
[1]Dauntless Pharmaceuticals Incorporated, San Diego, CA; [2]Milad Pharmaceutical Consulting LLC

BACKGROUND

- Octreotide is a synthetic octapeptide analog of naturally occurring somatostatin, with similar pharmacological effects but a longer duration of action, that inhibits the pathological secretion of growth hormone (GH) from pituitary adenomas and of serotonin and other hormones by tumors of the gastroenteropancreatic endocrine system[1,2,3,4].
- Depot formulations of octreotide and the somatostatin analog, lanreotide, which are administered by deep intramuscular and deep subcutaneous (SC) injections, respectively, play a key role in the treatment of neuroendocrine tumors and acromegaly. An immediate release formulation of octreotide for SC administration (Sandostatin® Injection) is also available for the management of diarrhea and flushing associated with carcinoid syndrome and the treatment of acromegaly[5].
- DP1038 is a short-acting, non-invasive, intranasal (NAS) formulation of octreotide acetate that includes the novel excipient 1-O-n-dodecyl-β-D-maltopyranoside (DDM) for enhanced intranasal absorption.
- DP1038 has the potential to provide therapeutic benefit as an essentially painless, needle-free, safe, and efficacious presentation of octreotide.

OBJECTIVE

- The aim of the study was to determine the pharmacokinetics (PK), pharmacodynamics (PD), safety, and tolerability of intranasal octreotide (DP1038) in healthy volunteers.

METHODS

- Two-part, Phase 1, randomized, crossover design study to evaluate varying doses of a novel NAS formulation for octreotide (DP1038) compared to SC octreotide.

Study Part 1 – Safety, Tolerability, and PK

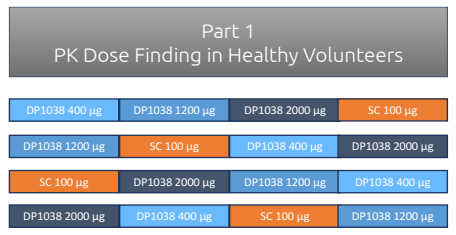
Part 1
PK Dose Finding in Healthy Volunteers
DP1038 400 µg | DP1038 1200 µg | DP1038 2000 µg | SC 100 µg
DP1038 1200 µg | SC 100 µg | DP1038 400 µg | DP1038 2000 µg
SC 100 µg | DP1038 2000 µg | DP1038 1200 µg | DP1038 400 µg
DP1038 2000 µg | DP1038 400 µg | SC 100 µg | DP1038 1200 µg

- **4X4 modified Latin square design**
- **n=3/group (12 total)**
- **Sandostatin® Injection SC 100 µg**
- **Intranasal doses selected to cover range of SC doses of 100 – 500 µg**

- Four-way crossover modified Latin square design, in which each of the 12 subjects enrolled received three single NAS administrations of DP1038 (each at a different dose level: 400, 1200, and 2000 µg), plus a single SC administration of 100 µg Sandostatin Injection, with a one-day washout period between the doses.
- Results from Part 1 were used to identify the NAS DP1038 dose for which the systemic octreotide exposure was similar to that of the 100 µg SC dose of Sandostatin Injection.

Study Part 2 – Safety, Tolerability, PK, and PD

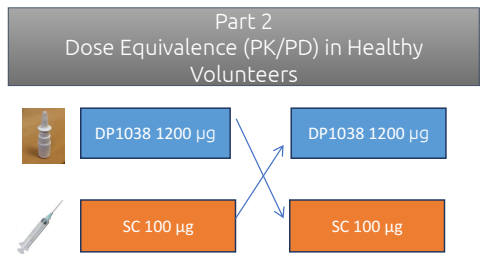
Part 2
Dose Equivalence (PK/PD) in Healthy Volunteers
DP1038 1200 µg
DP1038 1200 µg
SC 100 µg
SC 100 µg

- **Crossover study design between Sandostatin Injection and intranasal dose determined from Part 1**
- **n=10/group (20 total)**
- **PD assessment is GHRH-arginine challenge**

- DP1038 1200 µg dose was further assessed in a new set of 20 healthy subjects in a two-way crossover study design.
 Day 1: Subjects received a standard GH-releasing hormone (GHRH)/arginine challenge to establish baseline values of GH response without any study treatment.
 Days 3 to 5: Half of the subjects were randomized to undergo GHRH/arginine challenge following one intranasal dose of DP1038 on Day 3, washout on Day 4, and GHRH/arginine challenge following one SC dose of Sandostatin Injection on Day 5. The other half of the subjects received drugs in the reverse sequence on the same study days.

RESULTS

DP1038 Demonstrates Rapid Onset and Dose-Proportional PK

Mean (± SE)

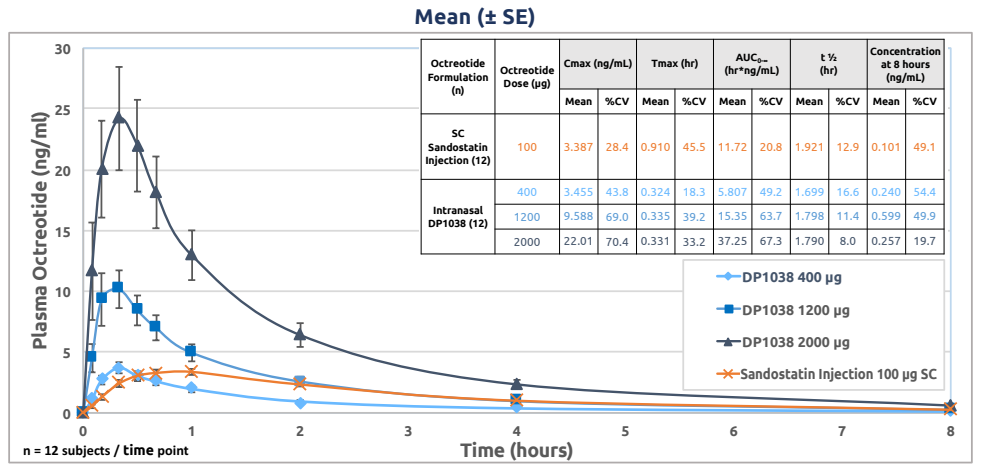
Plasma Octreotide (ng/ml)
Time (hours)
n = 12 subjects / time point
DP1038 400 µg
DP1038 1200 µg
DP1038 2000 µg
Sandostatin Injection 100 µg SC

Octreotide Formulation (n)	Octreotide Dose (µg)	Cmax (ng/mL)		Tmax (hr)		$AUC_{0-\infty}$ (hr*ng/mL)		t ½ (hr)		Concentration at 8 hours (ng/mL)	
		Mean	%CV	Mean	%CV	Mean	%CV	Mean	%CV	Mean	%CV
SC Sandostatin Injection (12)	100	3.387	28.4	0.910	45.5	11.72	20.8	1.921	12.9	0.101	49.1
Intranasal DP1038 (12)	400	3.455	43.8	0.324	18.3	5.807	49.2	1.699	16.6	0.240	54.4
	1200	9.588	69.0	0.335	39.2	15.35	63.7	1.798	11.4	0.599	49.9
	2000	22.01	70.4	0.331	33.2	37.25	67.3	1.790	8.0	0.257	19.7

Pharmacokinetics:

- Plasma octreotide areas under the curve (AUCs) and peak plasma concentration (C_{max}) increased in a dose-proportional manner. Consistently shorter time to peak concentration (T_{max}) was observed for all doses of DP1038 (18 min versus 50 min for SC).
- The plasma octreotide relative bioavailability of 400, 1200, and 2000 µg intranasal doses of DP1038 compared to SC 100 µg Sandostatin Injection was calculated to be between 29.5% and 45.4% for $AUC_{0-\infty}$.

DP1038 Exerts Robust Suppression of GHRH/Arginine Stimulated GH Secretion

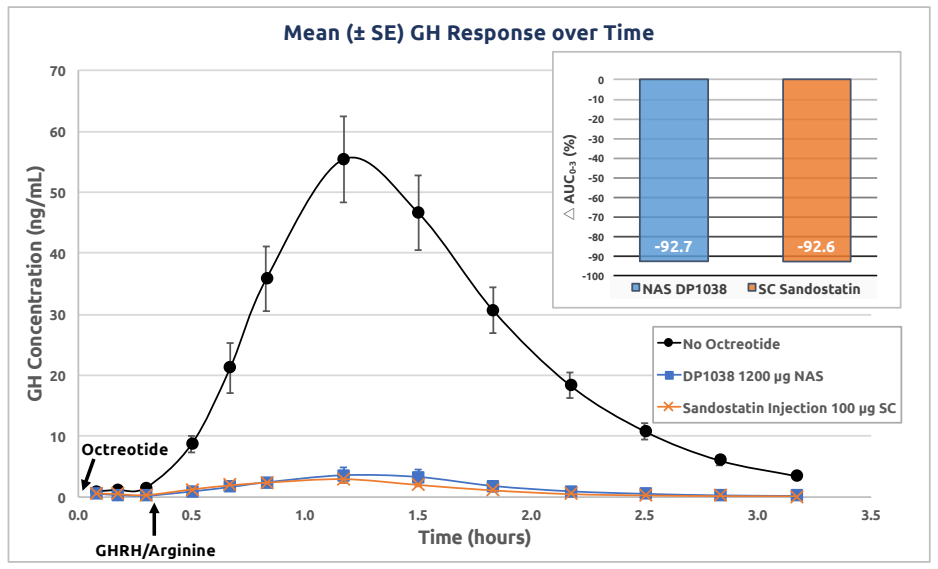
Mean (± SE) GH Response over Time
GH Concentration (ng/mL)
Time (hours)
Octreotide
GHRH/Arginine
△ AUC0-3 (%)
-92.7
-92.6
NAS DP1038
SC Sandostatin
No Octreotide
DP1038 1200 µg NAS
Sandostatin Injection 100 µg SC

DP1038 Suppresses Basal GH

Mean (± SE) GH Response over Time

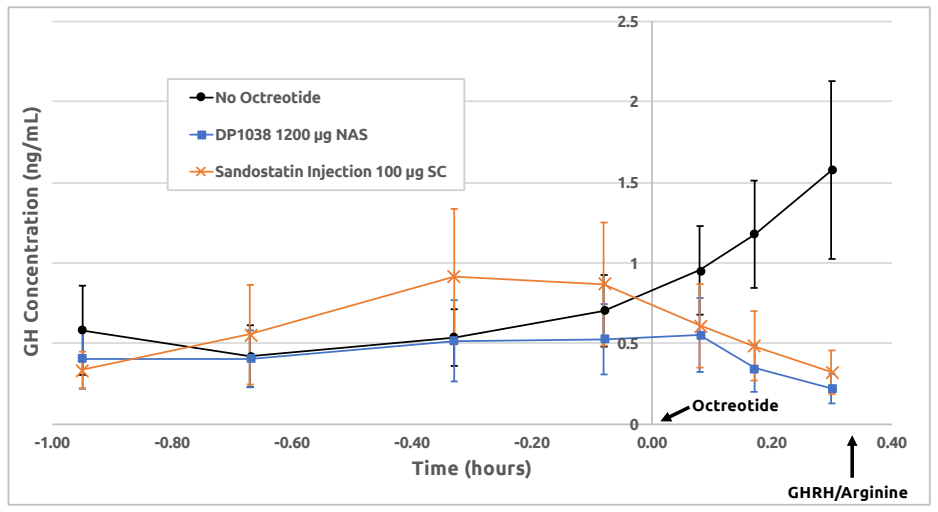
GH Concentration (ng/mL)
Time (hours)
No Octreotide
DP1038 1200 µg NAS
Sandostatin Injection 100 µg SC
Octreotide
GHRH/Arginine

Pharmacodynamics:

- Greater than 90% suppression of GHRH/Arginine stimulated GH production (AUC_{0-3}) was measured with both intranasal DP1038 (1200 µg) and SC Sandostatin (100 µg).
- Inhibition of basal GH levels was also comparable between intranasal DP1038 and SC Sandostatin, as noted during the time points after octreotide administration (Time 0) but prior to GHRH/Arginine stimulation (after 0.3 hours time point).

Safety:

Both Study Parts – Most Common Drug-Related Adverse Events

Adverse Event	DP1038 400 µg N=12		DP1038 1200 µg N=31		DP1038 2000 µg N=12		SC 100 µg N=32		Combined Treatments N=32	
	Grade 1	≥ Grade 2	Grade 1	≥ Grade 2	Grade 1	≥ Grade 2	Grade 1	≥ Grade 2	Grade 1	≥ Grade 2
Sneezing	2 (16.7%)	0 (0%)	12 (38.7%)	0 (0%)	4 (33.3%)	0 (0%)	0 (0%)	0 (0%)	13 (40.6%)	0 (0%)
Nasal Discomfort	3 (25.0%)	0 (0%)	7 (22.3%)	0 (0%)	5 (41.7%)	0 (0%)	0 (0%)	0 (0%)	9 (28.1%)	0 (0%)
Back Pain	0 (0%)	0 (0%)	0 (0%)	1 (3.2%)	3 (25.0%)	0 (0%)	1 (3.1%)	0 (0%)	3 (9.4%)	1 (3.1%)
Diarrhea	1 (8.3%)	0 (0%)	2 (6.5%)	0 (0%)	1 (8.3%)	0 (0%)	1 (3.1%)	0 (0%)	4 (12.5%)	0 (0%)
Feeling Hot	1 (8.3%)	0 (0%)	2 (6.5%)	0 (0%)	0 (0%)	0 (0%)	1 (3.1%)	0 (0%)	4 (12.5%)	0 (0%)
Headache	0 (0%)	0 (0%)	2 (6.5%)	0 (0%)	0 (0%)	0 (0%)	2 (6.3%)	0 (0%)	4 (12.5%)	0 (0%)
Nausea	0 (0%)	0 (0%)	4 (12.9%)	0 (0%)	0 (0%)	0 (0%)	1 (3.1%)	0 (0%)	4 (12.5%)	0 (0%)

- Administration of single doses of 400-2000 µg intranasal DP1038 and 100 µg SC Sandostatin was safe and well tolerated in healthy adult subjects.
- No discontinuations due to adverse events (AEs) or serious AEs.
- Most commonly occurring AEs:
 - Expected and consistent with SC product prescribing information[5] (e.g., loose stools, nausea, and headaches).
 - Minor, local tolerability events.

CONCLUSIONS

- Intranasal delivery of octreotide (DP1038) results in octreotide PK characteristics comparable to SC octreotide injection and achieves drug concentration levels known to be therapeutic.
- DP1038 exerts robust suppression of basal and GHRH/arginine stimulated GH secretion in healthy volunteers.
- DP1038 is well tolerated; no significant safety findings were reported.
- DP1038 is useful for the treatment of patients with neuroendocrine tumors and acromegaly.

REFERENCES

1. Brazeau et al, Science 179(4068), 77-79 (1973)
2. Bauer et al, Life Sci 31(11), 1133-1140 (1982)
3. Lamberts et al, N Engl J Med 313(25), 1576-1580 (1985)
4. Vinik and Moattari, Dig Dis Sci 34(3), 14S-27S (1989)
5. Prescribing Information for Sandostatin® (octreotide acetate) Injection. March 2012.

DAUNTLESS Pharmaceuticals

CONTACT INFORMATION: Matteo Levisetti, MD - matteo@dauntlessph.com